Abstract 122902

A Phase 1b/2 Study of Selinexor, Carfilzomib, and Dexamethasone (SKd) in Relapsed / Refractory Multiple Myeloma (RRMM)

Cristina Gasparetto[1], Gary J. Schiller[2], Natalie Scott Callander[3], Suzanne Lentzsch[4], Sascha A Tuchman[5], Nizar Bahlis[6], Darrell J White[7], Christine I Chen[8], Heather J. Sutherland[9], Muhamed Baljevic[10], Rami Kotb[11], Richard Leblanc[12], Michael Sebag[13], Christopher P Venner[14], William I. Bensinger[15], Heidi Sheehan[16], Yi Chai[16], Kazuharu Kai[16], Jatin Shah[16], Sharon Shacham[16], Michael G. Kauffman[16], Brea C. Lipe[17]

(1) Duke Univ. Medical Center, Durham, NC (2) David Geffen School of Medicine at UCLA, Los Angeles, CA (3) Carbone Cancer Center, University of Wisconsin- Madison, Madison, WI (4) Colombia University, New York (5) University of North Carolina, Chapel Hill, NC (6) Southern Alberta Cancer Research Institute, Calgary, AB, Canada (7) Dalhousie University and Queen Elizabeth II Health Sciences Centre, Halifax, Canada (8) Department of Medical Oncology and Hematology, Princess Margaret Cancer Centre, Toronto, ON, Canada (9) Vancouver General Hospital, Vancouver, BC, Canada (10) University of Nebraska Medical Center, Omaha, NE (11) Cancer Care Manitoba, Winnipeg, MB, Canada (12) Hopital Maisonneuve-Rosemont, Montreal, QC, Canada (13) Royal Victoria Hospital, Montreal, QC, Canada (14) Cross Cancer Institute, University of Alberta, Edmonton, AB, Canada (15) Myeloma and Transplant Program, Swedish Cancer Institute, Seattle, WA (16) Karyopharm Therapeutics, Newton, MA (17) University of Rochester Medical College, Rochester, NY

Selinexor Mechanism of Action

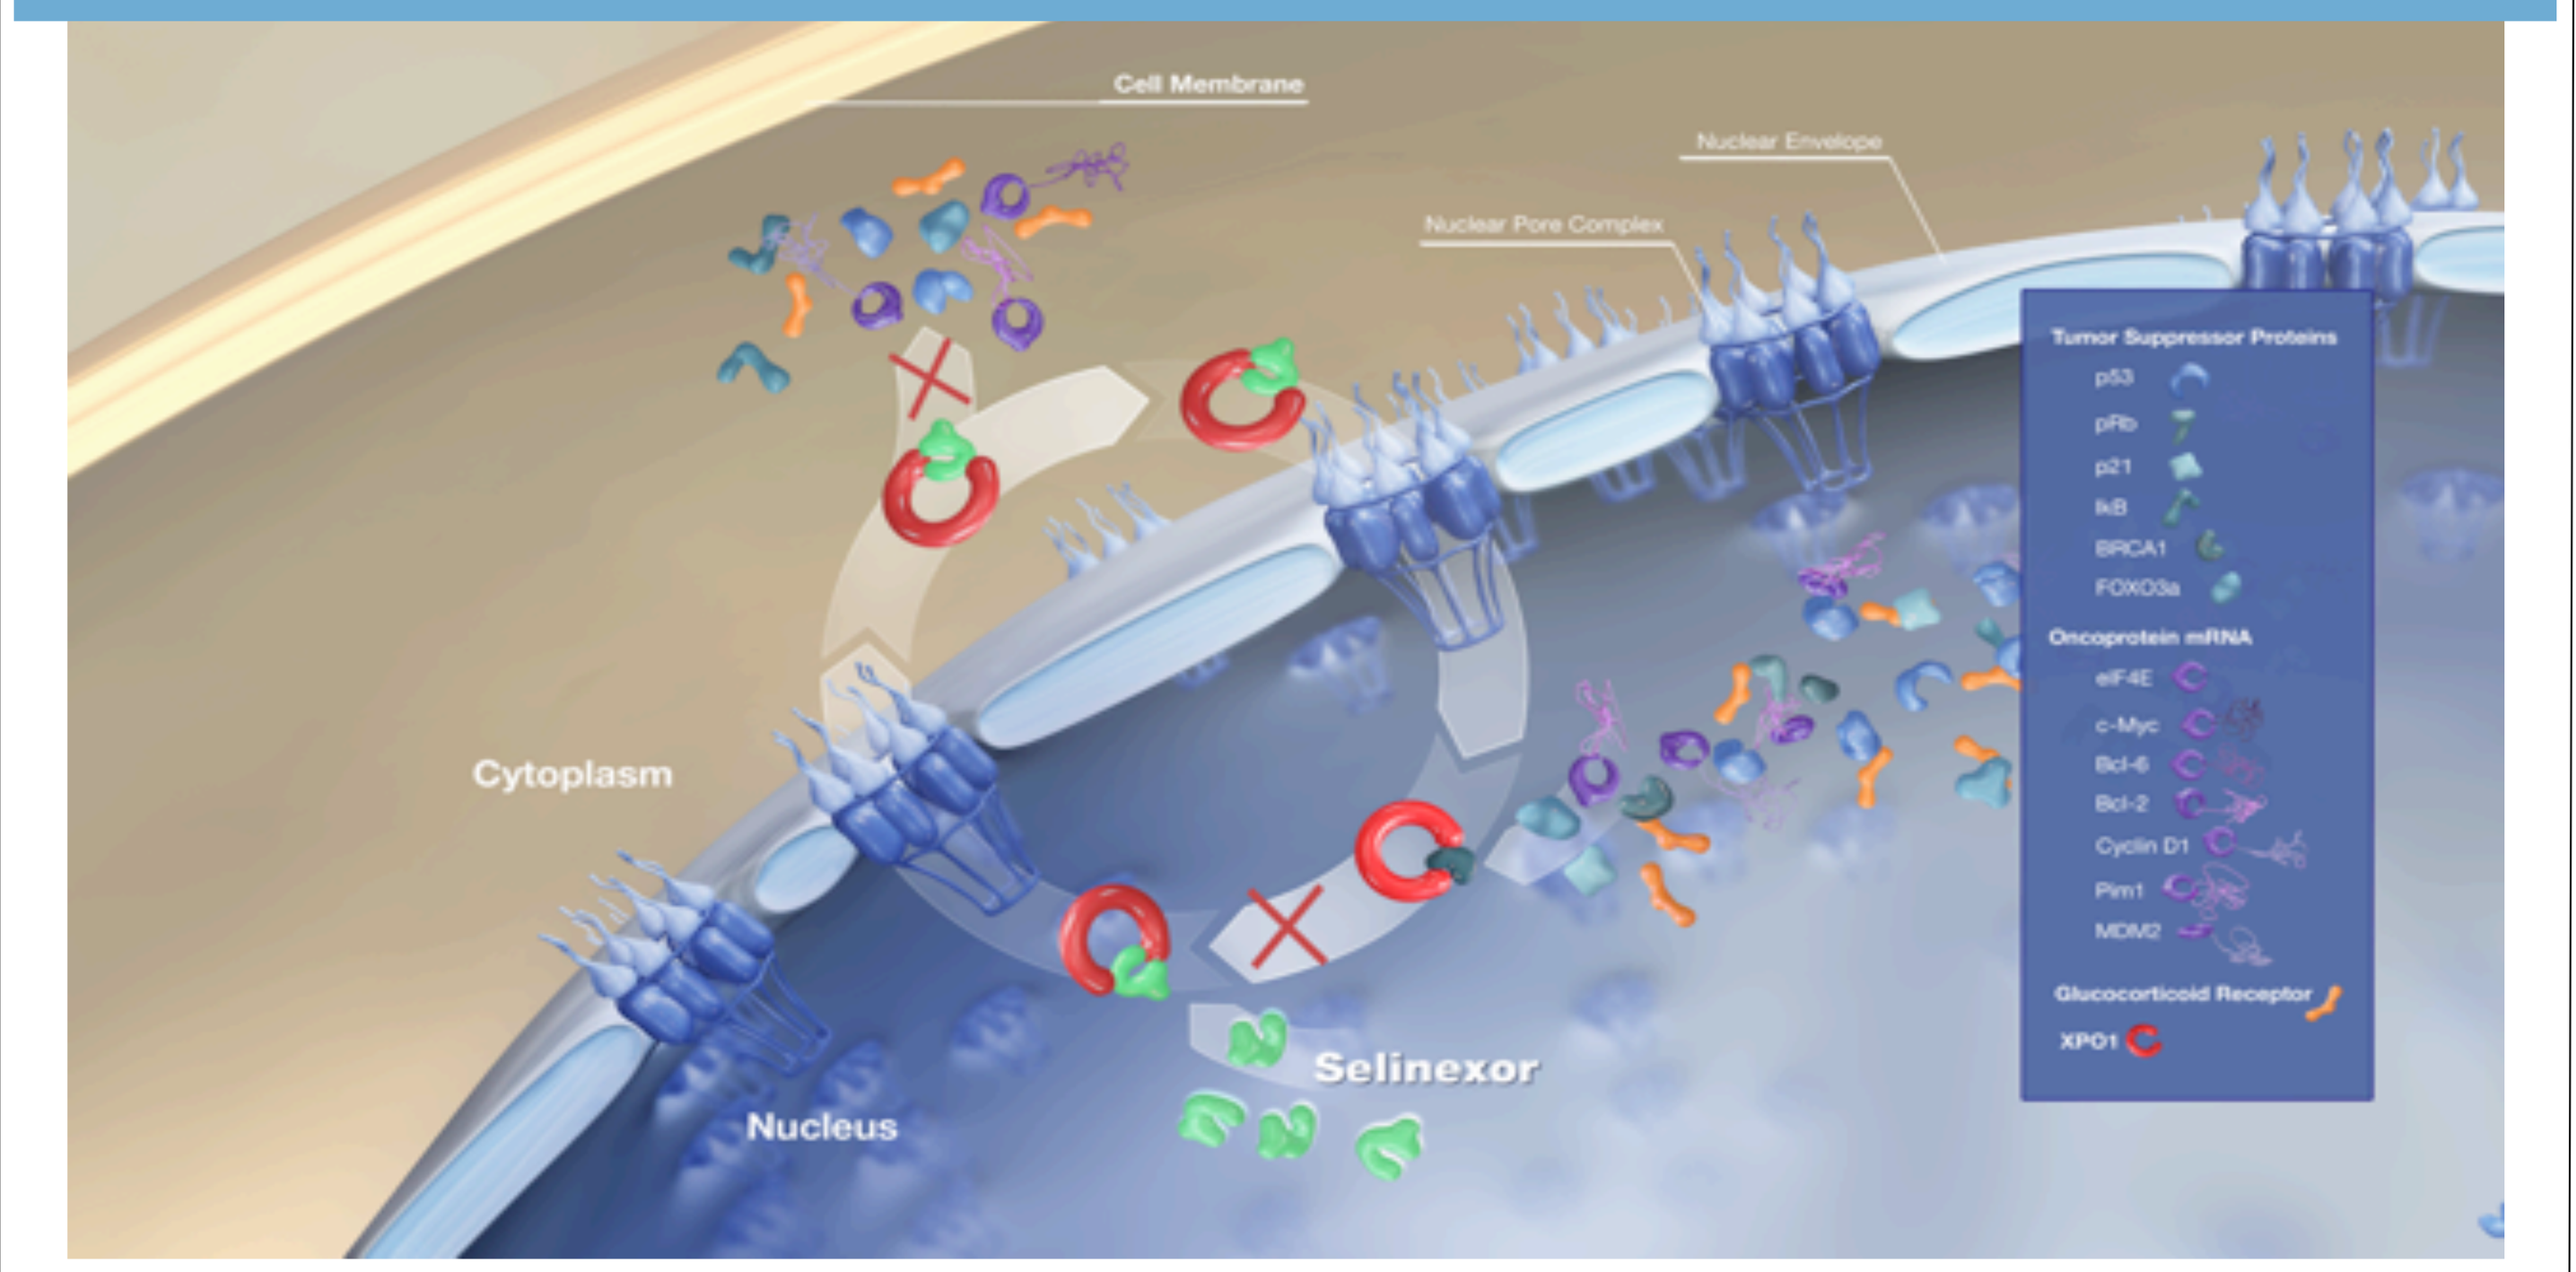

Cell Membrane
Nuclear Envelope
Nuclear Pore Complex
Tumor Suppressor Proteins
p53
pRb
p21
IkB
BRCA1
FOXO3a
Oncoprotein mRNA
eIF4E
c-Myc
Bcl-6
Bcl-2
Cyclin D1
Pim1
MDM2
Glucocorticoid Receptor
XPO1
Cytoplasm
Nucleus
Selinexor

- **Exportin 1 (XPO1)** is the major nuclear export protein for tumor suppressor proteins (TSPs), the glucocorticoid receptor (GR), and eIF4E-bound oncoprotein mRNAs (e.g., c-Myc, Bcl-xL, MDM2, cyclins)
- **XPO1 is overexpressed in multiple myeloma (MM)**
 - High XPO1 levels enable cancer cells to escape TSP mediated cell cycle arrest and induction of apoptosis
 - XPO1 levels correlate with poor prognosis and drug resistance
- **Selinexor** is an oral selective XPO1 inhibitor; preclinical data demonstrate that selinexor
 - Reactivates multiple TSPs relevant to MM, inhibits NF-kB signaling, reduces c-Myc levels and reactivates GR signaling
 - Demonstrates synergistic activity with proteasome inhibitors by forcing nuclear localization of high levels of TSPs

Background / Hypothesis

- **Selinexor (+ dexamethasone)** received accelerated approval from the FDA for patients with RRMM[1]
- Selinexor showed a synergistic anti-MM effect with Carfilzomib in preclinical xenograft MM model[2].
- Twice weekly (BIW) Selinexor + BIW Carfilzomib + Dex achieved 48% ORR and 3.7 months progression-free survival (PFS) in MM patients who received ≥2 prior therapies including PI and IMiD[3]
- Carfilzomib 20/70 mg/m^2 and 20/56/70 mg/ m^2 were not tolerable in combination with lenalidomide 25 mg OD + low dose Dex but maximum tolerable dose (MTD) was Carfilzomib 20/56 mg/m^2

Hypothesis: Once weekly (QW) Carfilzomib and QW selinexor + Dex is tolerable and derives promising responses in Carfilzomib-naïve relapse/refractory multiple myeloma (RRMM) patients

STOMP Study with SKd

Selinexor and Backbone Treatments Of Multiple Myeloma Patients

- Open-label, dose escalation (Phase 1) and expansion (Phase 2) study evaluating selinexor in combination with other anti-myeloma therapies in patients with newly diagnosed and relapsed/refractory multiple myeloma (MM)
- **Objectives**
 - Primary endpoint: maximum tolerated dose (MTD) and recommended phase 2 dose (RP2D)
 - Secondary endpoint: overall response rate (ORR) and duration of response (DOR) for each arm independently
- **Key Inclusion / Exclusion criteria**
 - Age ≥ 18 y.o. at the time of informed consent, ECOG 0-2
 - WBC ≥ 1,500/mm^3 Hb ≥ 8.0 g/dL, platelet count ≥ 75,000/mm^3
 - Progressing or refractory to a previous regimen
 - Prior proteasome inhibitors are allowed, however, patients with refractory to carfilzomib are excluded
 - Smoldering MM, non-secretory MM, active plasma cell leukemia are excluded

Investigation of Once Weekly SKd Doses

SKd Dose Schedule

- Selinexor 80 or 100 mg QW (Day 1, 8, 15, and 22)
- Dexamethasone 40 mg QW (Day 1, 8, 15, and 22)
- Carfilzomib 56 or 70 mg/m^2 QW (Day 1*, 8, and 15)

*Carfilzomib's C1D1 dose is always 20 mg/m^2 per carfilzomib's label

SKd Dose Escalation

Dose Levels	Selinexor	Dexamethasone	Carfilzomib
	Days 1, 8, 15 and 22	Days 1, 8, 15 and 22	Days 1, 8, and 15
2	100 mg PO	40 mg IV or PO	70 mg/m^2 IV
1	100 mg PO	40 mg IV or PO	56 mg/m^2 IV
-1	80 mg PO	40 mg IV or PO	56 mg/m^2 IV
-1a*	80 mg PO	40 mg IV or PO	70 mg/m^2 IV

SKd dose escalation scheme: a standard 3 + 3 design will be used for dose escalations. Starting dose was dose level 1.

BIW=twice-weekly, IV=intravenous, PO=per oral, QD=once-daily, QW=once-weekly, SKd=selinexor-carfilzomib-dexamethasone, SVd=selinexor-bortezomib-dexamethasone

SKd Patient Characteristics

- As of October 1, 2019, 14 patients have been enrolled; enrollment is ongoing with QW selinexor 80 mg + carfilzomib 56 mg/m^2

SKd Patient Characteristics	N
Enrolled as Sep 1, 2019	**14**
- 100 mg selinexor QW + 56 mg/m^2 carfilzomib	3
- 80 mg selinexor QW + 70 mg/m^2 carfilzomib	3
- 80 mg selinexor QW + 56 mg/m^2 carfilzomib	8
Median Age, Years (range)	69.5 (50 – 76)
Males : Females	6 (42.9%) : 8 (57.1%)
Race, White: African American: Other	7 (50%): 4 (29%) : 3 (21%)
ECOG Performance Status, 0 : 1 : 2	3 (21%) : 10 (71%): 1 (7%)
Median Years from Diagnosis to SKd Treatment, Years (range)	4.9 (3 – 11)
Median Prior Regimens (range)	**4 (2–8)***
-Bortezomib (Treated : Refractory)	14 (100%): 9 (64%)
-Carfilzomib Therapy (Treated : Refractory)	0% : 0%
-Lenalidomide (Treated : Refractory)	14 (100%) : 8 (57%)
-Pomalidomide (Treated : Refractory)	9 (64%) : 8 (57%)
-Daratumumab (Treated : Refractory)	9 (64%) : 7 (50%)
-Stem Cell Transplant	9 (64%)

QW=once weekly, SKd=selinexor-carfilzomib-dexamethasone

Selinexor + Carfilzomib + Dex (SKd) Dose-Limiting Toxicity (DLT)

- **Dose limiting toxicity (DLT) was determined in dose escalation cycle 1 only:**
 - >1 missed dose (out of 4 doses – once-weekly selinexor dose schedules), or >2 missed doses (out of 6 doses – twice-weekly dose schedules) of selinexor during a cycle due to study-drug related toxicity
 - Discontinuation of a patient before completing Cycle 1, due to study-drug related toxicity
 - Grade 3 nausea, vomiting, dehydration, diarrhea or fatigue lasting >3 days despite optimal supportive medications
 - Grade 4 neutropenia lasting >7 days or Grade ≥3 thrombocytopenia with clinically significant bleeding, petechiae or purpura

Selinexor Dose	Carfilzomib Dose	Patients Enrolled	Patients DLT-evaluable	Patients with DLT	Dose Limiting Toxicity
100 mg QW	56 mg/m^2 IV	3	*2	2	Selinexor Dose Reduction due to Grade 3 Thrombocytopenia; Selinexor Dose Reduction due to Grade 3 Vomiting
80 mg QW	70 mg/m^2 IV	3	3	2	Grade 4 Thrombocytopenia and Grade 3 Pneumonia; Grade 4 Thrombocytopenia
****80 mg QW**	**56 mg/m^2 IV**	**6**	**6**	**--**	**No DLT and Enrollment is Ongoing**

*One patient was not DLT evaluable because platelet count was <50x10^9/L on C1D1. **Two patients after the first 6 patients were not included in the DLT assessment.

SKd Treatment_related Adverse Events ≥ 2 Patients (as of Oct 1, 2019)

AE Term	100/80 mg Sel QW + 70/56 mg/m^2 Carfil IV			TOTAL (N=14)
	Grade 1/2	Grade 3	Grade 4	
Hematologic				
Thrombocytopenia (%)	2 (14.3)	3 (21.4)	6 (42.9)	**11 (78.6)**
Anemia	6 (42.9)	2 (14.3)	--	**8 (57.1)**
Leukopenia	2 (14.3)	2 (14.3)	--	**4 (28.6)**
Neutropenia	3 (21.4)	1 (7.1)	--	**4 (28.6)**
Gastrointestinal				
Nausea (%)	10 (71.4)	--	--	**10 (71.4)**
Anorexia	4 (28.6)	1 (7.1)	--	**5 (35.7)**
Vomiting	4 (28.6)	1 (7.1)	--	**5 (35.7)**
Constitutional				
Fatigue (%)	4 (28.6)	2 (14.3)	--	**6 (42.9)**
Body weight loss	5 (35.7)	--	--	**5 (35.7)**
Other				
Hyperglycemia (%)	3 (21.4)	1 (7.1)	1 (7.1)	**5 (35.7)**
Insomnia	4 (28.6)	--	--	**4 (28.6)**
Dysgeusia	3 (21.4)	--	--	**3 (21.4)**
Vision blurred	3 (21.4)	--	--	**3 (21.4)**
Diarrhea	2 (14.3)	--	--	**2 (14.3)**
Dyspepsia	2 (14.3)	--	--	**2 (14.3)**
Hyperhidrosis	2 (14.3)	--	--	**2 (14.3)**
Hypoalbuminemia	2 (14.3)	--	--	**2 (14.3)**
Hyponatremia	2 (14.3)	--	--	**2 (14.3)**
Pneumonia	--	2 (14.3)	--	**2 (14.3)**

Adverse Events: The most common treatment-related adverse events were nausea, anemia, anorexia, and fatigue (mainly Grade 1/2). Thrombocytopenia and leukopenia were also common (mainly Grade 3/4). No related Grade 5 events were reported. AE, adverse event; Carfil, carfilzomib; IV, intravenous; QW, Once-Weekly; Sel, selinexor.

Dose Intensity of Selinexor and Carfilzomib in SKd

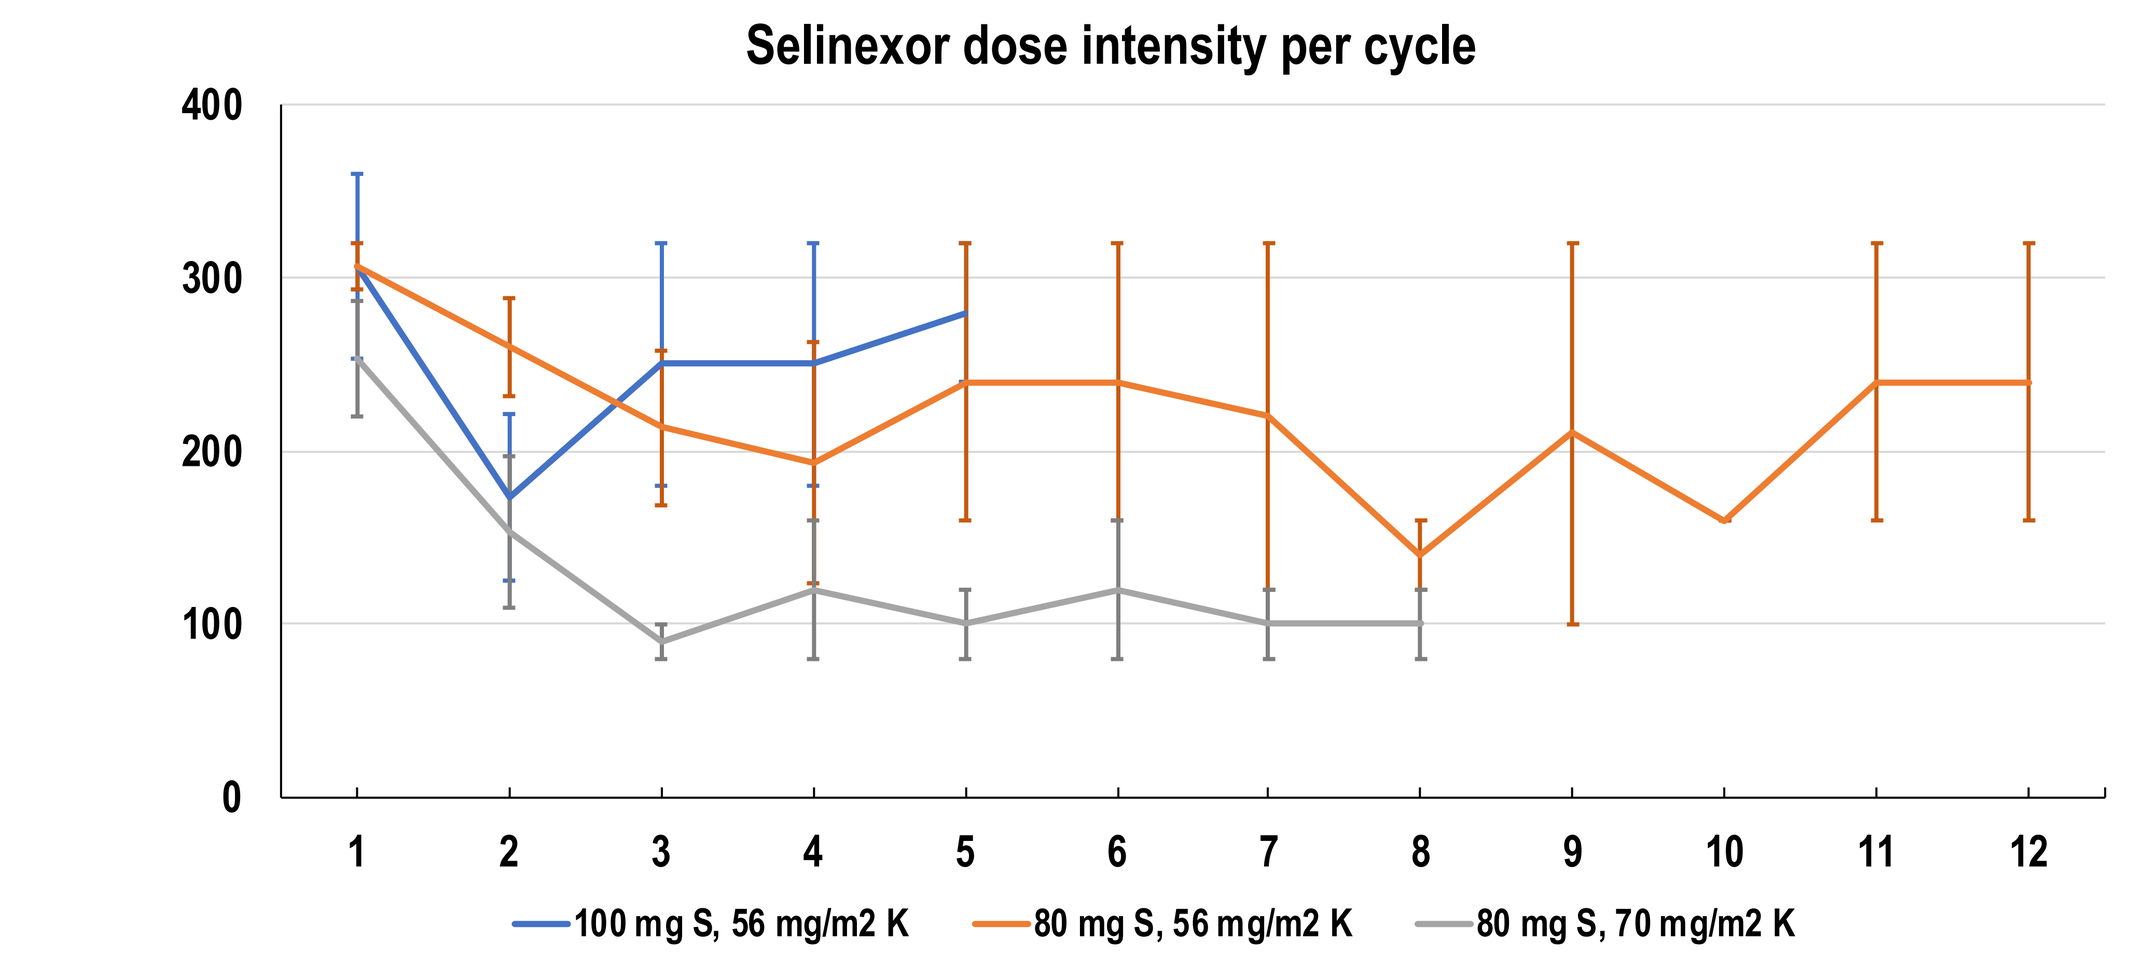

Selinexor dose intensity per cycle
400
300
200
100
0
1 2 3 4 5 6 7 8 9 10 11 12
100 mg S, 56 mg/m2 K
80 mg S, 56 mg/m2 K
80 mg S, 70 mg/m2 K

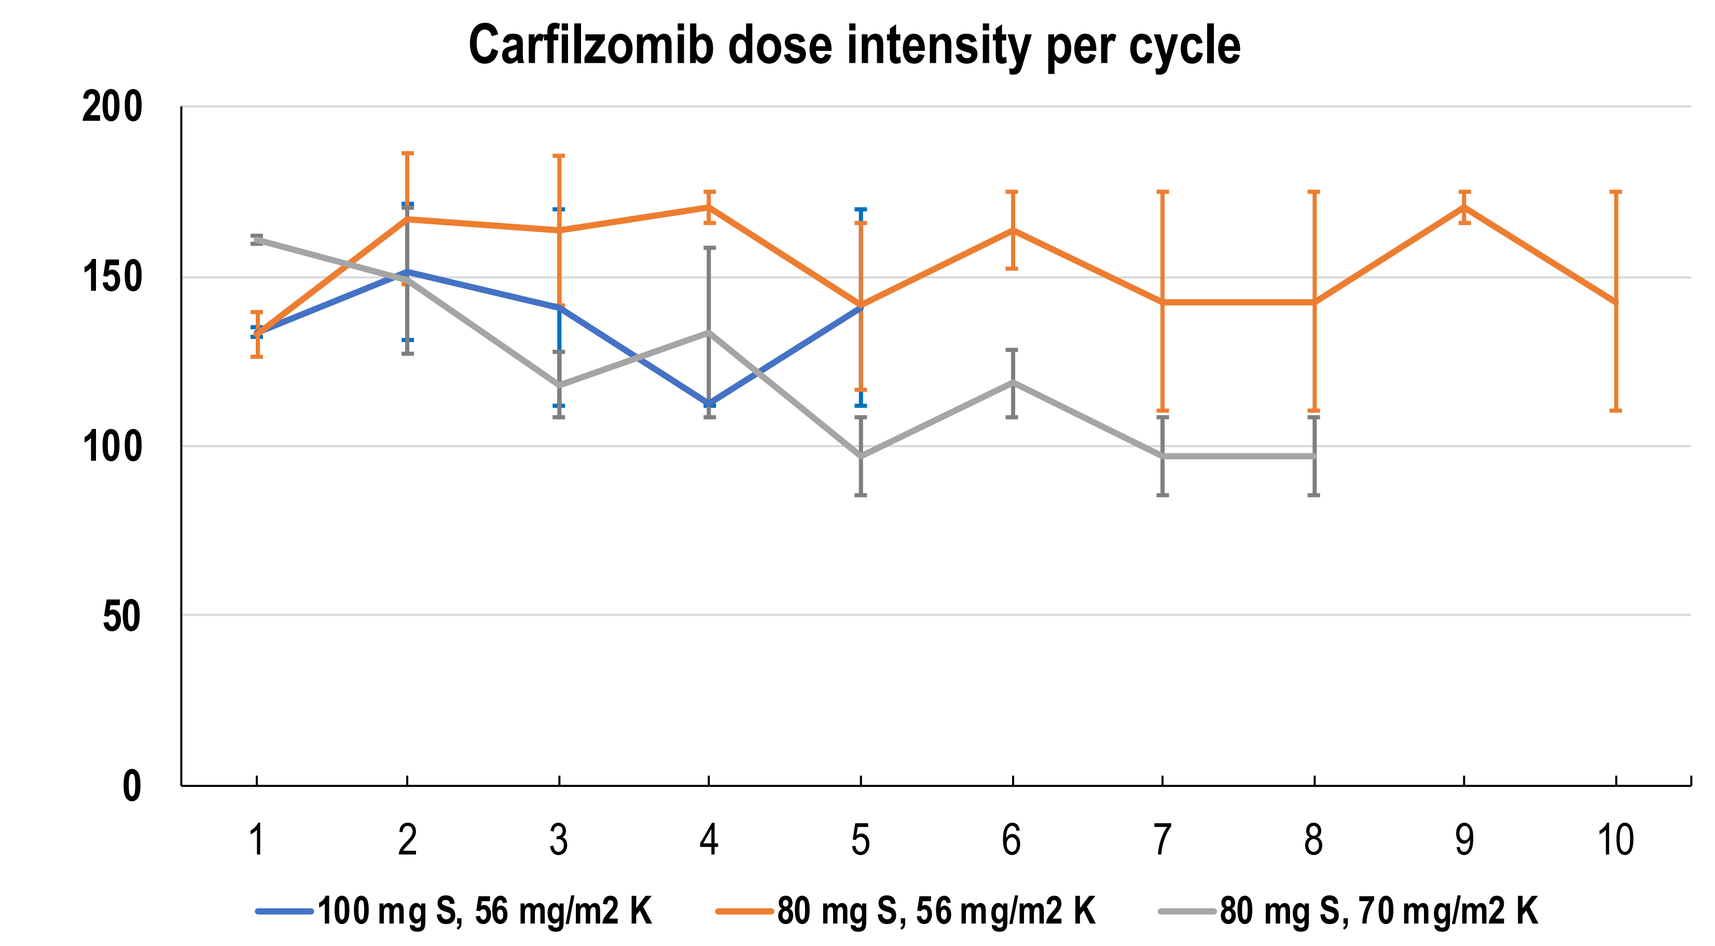

Carfilzomib dose intensity per cycle
200
150
100
50
0
1 2 3 4 5 6 7 8 9 10
100 mg S, 56 mg/m2 K
80 mg S, 56 mg/m2 K
80 mg S, 70 mg/m2 K

Graphs depict cycle points up to 12 and 10 for selinexor and carfilzomib, respectively. At all points, at least 2 patient's data were available. Error bars; s.e.m.

Selinexor + Carfilzomib + Dex (SKd) Efficacy (as of Oct 1, 2019)

Best Responses† in Evaluable SKd Patients

Category	N	ORR (%)	CBR (%)	CR (%)	VGPR (%)	PR (%)	MR (%)	SD (%)	PD (%)
All Efficacy Evaluable Patients	14	10 (71.4%)	11 (78.6%)	3 (21.4%)	7 (50%)	--	1 (7.1%)	3 (21.4%)	--

†Responses were adjudicated according to the International Myeloma Working Group (IMWG) criteria. ORR=Overall Response Rate (CR+VGPR+PR), CBR=Clinical Benefit Rate (ORR+MR), CR=Complete Response, VGPR=Very Good Partial Response, PR=Partial Response, MR=Minimal Response, SD=Stable Disease, PD=Progressive Disease. Responses as of Oct 1, 2019 based on interim unaudited data.

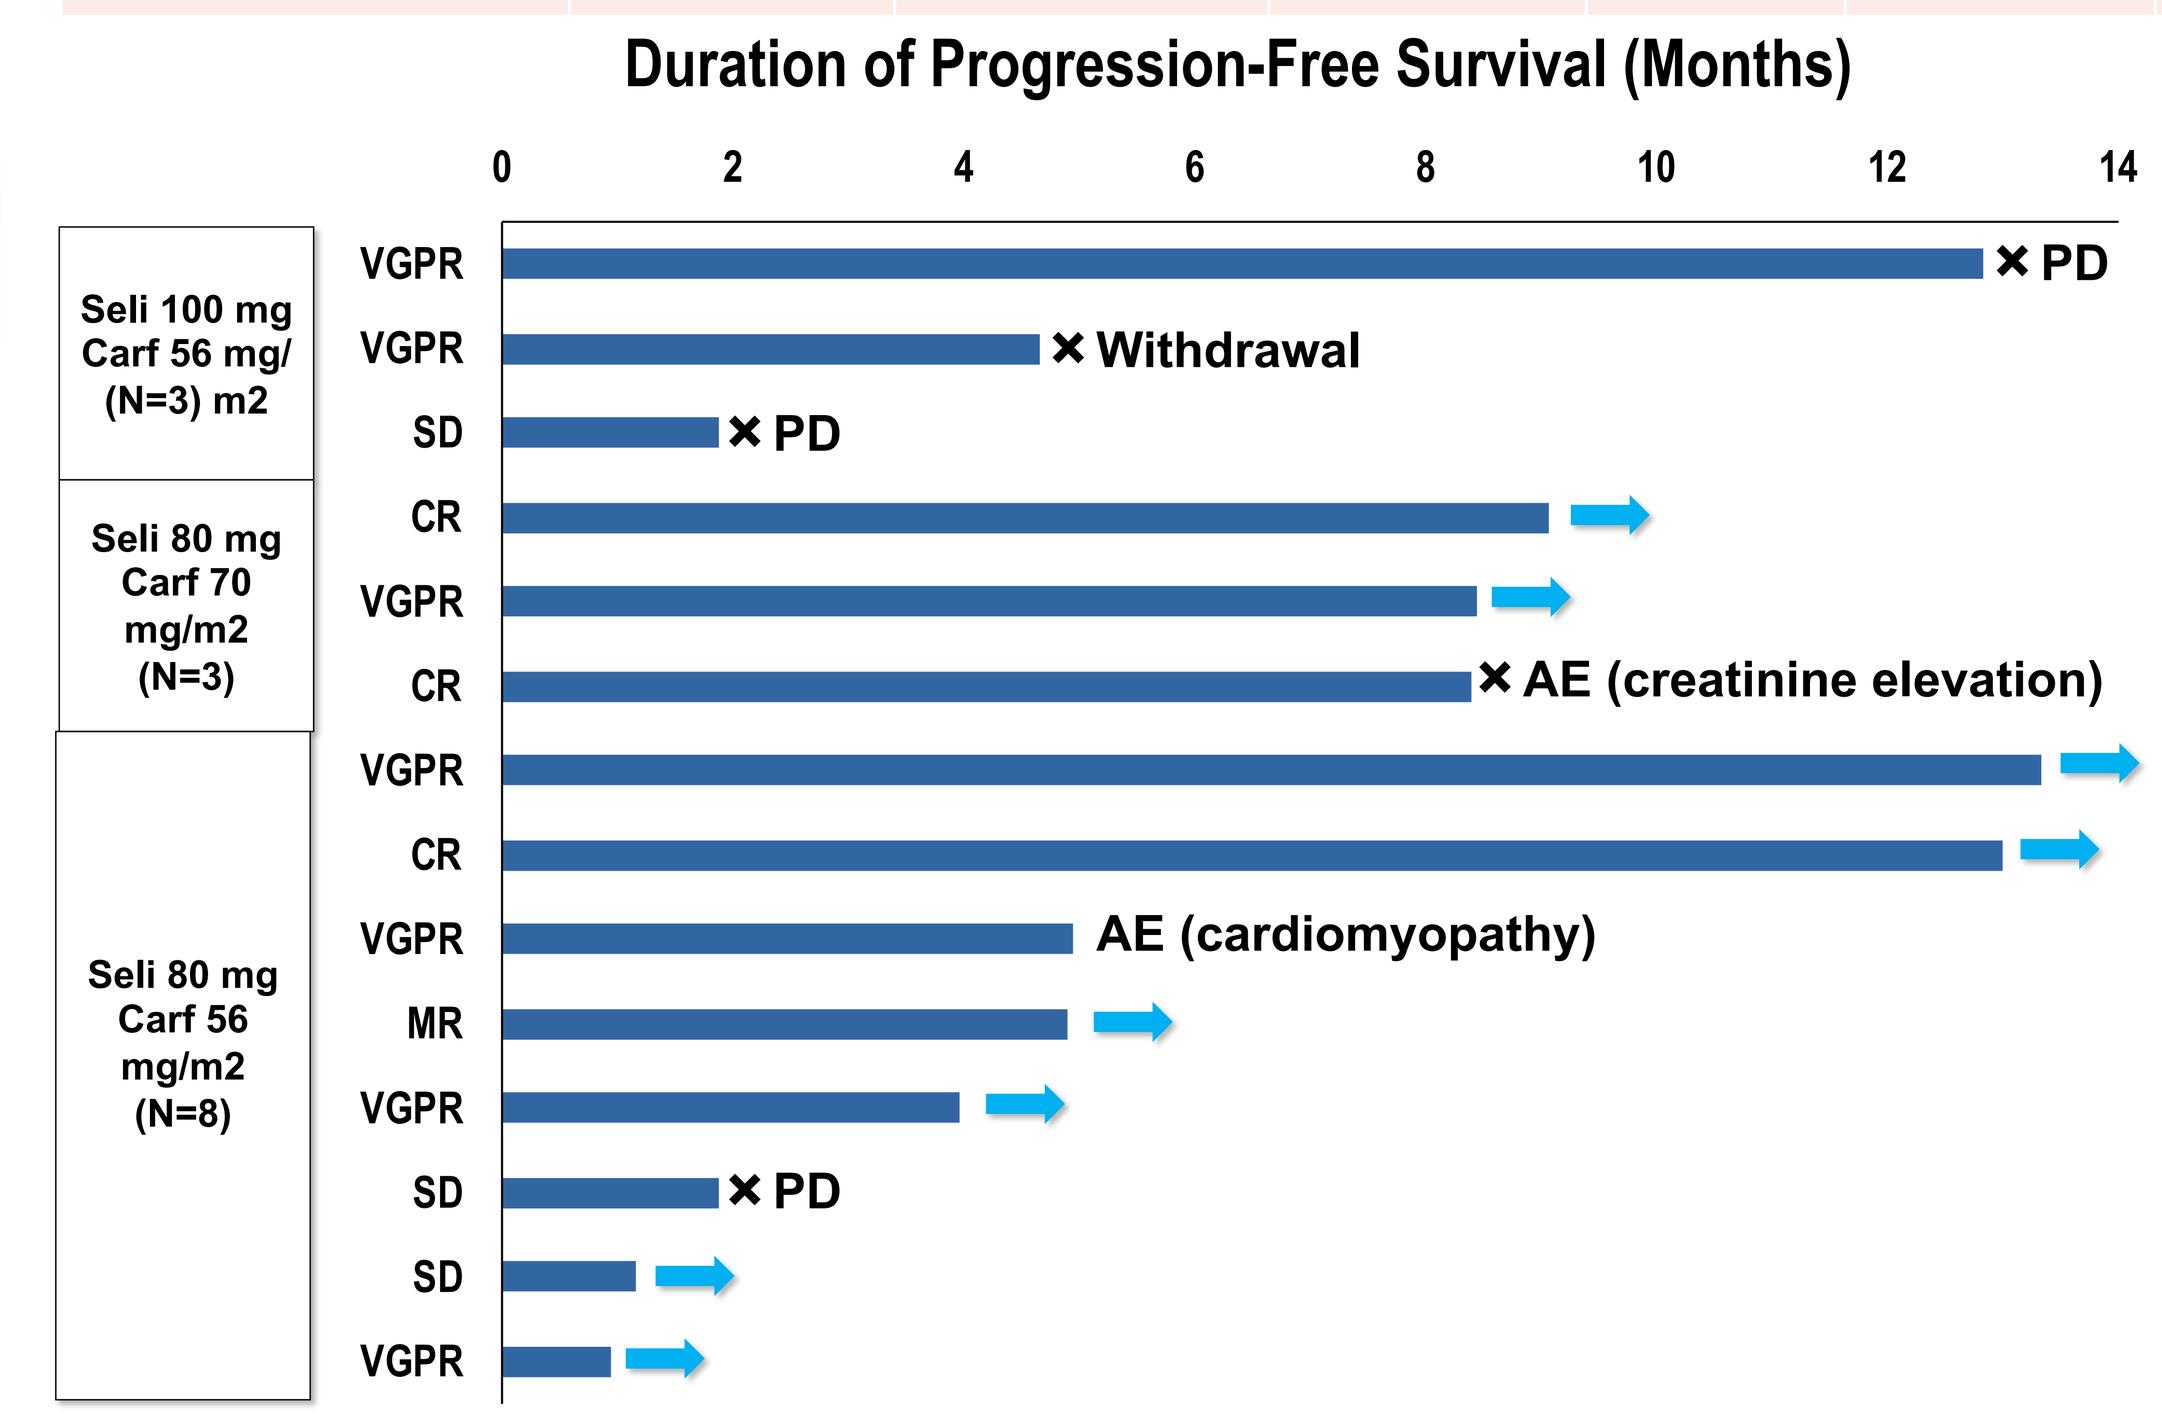

Duration of Progression-Free Survival (Months)
0 2 4 6 8 10 12 14
Seli 100 mg Carf 56 mg/ (N=3) m2
Seli 80 mg Carf 70 mg/m2 (N=3)
Seli 80 mg Carf 56 mg/m2 (N=8)
VGPR
VGPR
SD
CR
VGPR
CR
VGPR
CR
VGPR
MR
VGPR
SD
SD
VGPR
PD
Withdrawal
PD
AE (creatinine elevation)
AE (cardiomyopathy)
PD

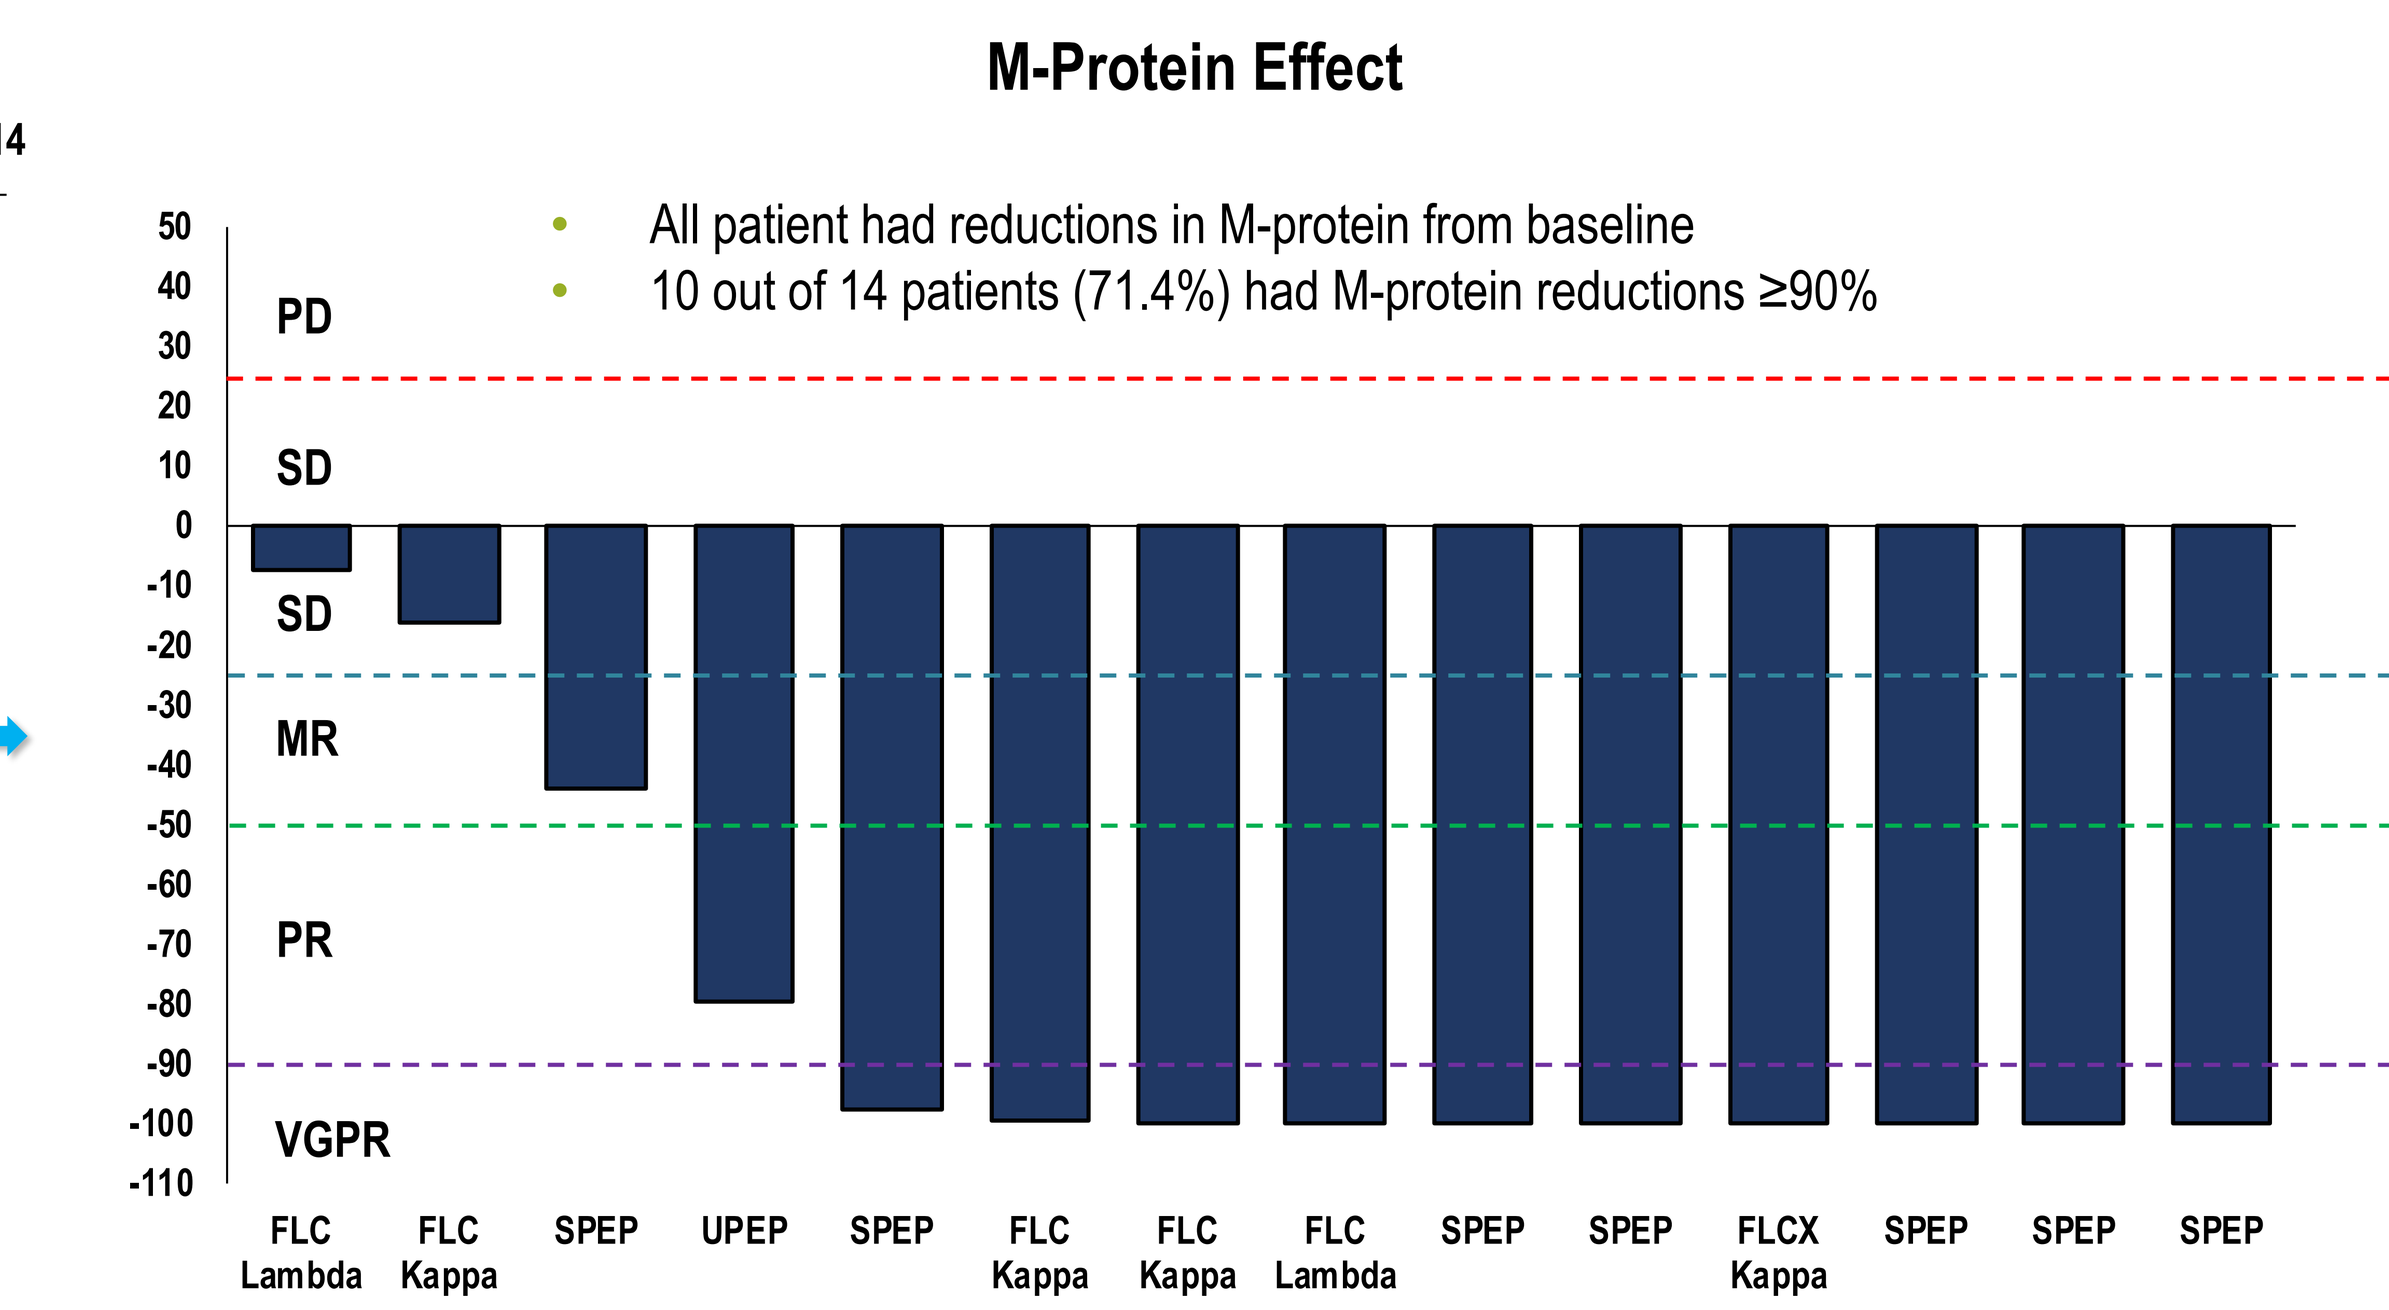

M-Protein Effect
All patient had reductions in M-protein from baseline
10 out of 14 patients (71.4%) had M-protein reductions ≥90%
50
40
30
20
10
0
-10
-20
-30
-40
-50
-60
-70
-80
-90
-100
-110
PD
SD
SD
MR
PR
VGPR
FLC Lambda
FLC Kappa
SPEP
UPEP
SPEP
FLC Kappa
FLC Kappa
FLC Lambda
SPEP
SPEP
FLCX Kappa
SPEP
SPEP
SPEP

Summary and Conclusions

- **Selinexor, once-weekly, can be safely combined with once weekly carfilzomib and low-dose dexamethasone**
- **The RP2D of SKd is once-weekly selinexor 80 mg + carfilzomib 56 mg/m^2**
- **The most common AEs are: nausea, anemia, anorexia, fatigue, and Grade 3/4 thrombocytopenia and leukopenia, which are expected and can be managed with appropriate supportive care and/or dose modifications**
- **The combination is active with a ORR of 71% with deep responses (CR 21 %, VGPR 50%) in the patients who had a median of 4 lines of prior therapy**
- **The trial is ongoing and support further investigation**

References

1. Food and Drug Administration. Selinexor Prescribing Information 2019. Available at: https://www.accessdata.fda.gov/drugsatfda_docs/label/2019/212306s000lbl.pdf [accessed 6 Nov 2019]. 2. Jakubowiak AJ, et al. Br J Haematol 2019;186:549–60. 3. Bahlis NJ, et al. Blood 2018;132(24):2546–54.
2. Rosebeck S *et al.* Mol Cancer Ther. 2016 Jan;15(1):60-71. Synergistic Myeloma Cell Death via Novel Intracellular Activation of Caspase-10 Dependent Apoptosis by Carfilzomib and Selinexor.
3. Jakubowiak AJ *et al.* Br J Haematol. 2019 Aug;186(4):549-560. Phase 1 study of selinexor plus carfilzomib and dexamethasone for the treatment of relapsed/refractory multiple myeloma.
4. Alsina M *et al.* International Myeloma Workshpt (IMW) 2019, Boston MA. OAB-078: A phase 1b study of once-weekly carfilzomib combined with lenalidomide and dexamethasone (wKRd) in patients (pts) with newly diagnosed multiple myeloma (NDMM)

Acknowledgments

Patients, their families, and caregivers, investigators, co-investigators, and study teams at each participating center

This study was supported by Karyopharm Therapeutics.