Selective Inhibitors of Nuclear Export (SINETM) Block the Expression of DNA Damage Repair Proteins and Sensitize Cancer Cells to DNA Damage Inducing Therapeutic Agents

26TH EORTC-NCI-AACR SYMPOSIUM ON MOLECULAR TARGETS AND CANCER THERAPEUTICS
18-21 NOVEMBER 2014
BARCELONA, SPAIN

Trinayan Kashyap, Marsha L. Crochiere, Sharon Friedlander, Boris Klebanov, William Senapedis, Erkan Baloglu, Diego del Alamo, Sharon Tamir, Dilara McCauley, Tami Rashal, Robert Carlson, Michael Kauffman, Sharon Shacham, and Yosef Landesman

Karyopharm Therapeutics, Newton, MA, USA

Abstract

Background: SINETM is a family of small-molecule drugs that inhibit Exportin 1 (XPO1/CRM1) mediated nuclear export, resulting in retention of major tumor suppressor proteins (TSPs) such as p53, FOXO, pRb and IκB in the nucleus and subsequently leading to specific cancer cell death. Selinexor is the clinical SINETM compound currently in human phase I/II clinical trials in patients with solid and hematological malignancies. The goal of this study was to evaluate the effects of selinexor on DNA repair mechanisms and to test the cytotoxic effects of combining selinexor with DNA damaging agent on hematological and solid tumors.

Methods: Whole protein cell lysates from solid and hematological cancer cell lines treated with selinexor with or without agents that induce DNA damage were analyzed in Reverse Phase Protein Arrays (RPPA), immunoblots and quantitative PCR. Selinexor treated cells from solid and hematological cancer lines were analyzed by immunofluorescence to evaluate DNA damage. Non-small cell lung cancer A549 Xenografts were treated with selinexor (5 mg/kg or10mg/kg) and radiation (3 Gy) alone or in combination and tumor growth was evaluated for 22 days.

Results: Treatment of solid and hematological cancer cell lines with selinexor did not induce DNA damage but reduced the expression of DNA damage repair (DDR) proteins: MSH2, MSH6, PMS2, MLH1, Rad51 and CHK1. Selinexor regulates the expression of CHK1, RAD51, MSH2, MSH6 and MLH1 on the transcriptional levels and PMS2 expression on the posttranslational level. There was a trend between the degree of DNA-damage-repair-protein reduction to selinexor sensitivity. Knock down of Chk1 alone, induced cytotoxicity whereas silencing of the other DNA repair proteins did not affect cell viability. Selinexor treatment following exposure to the DNA damaging agents doxorubicin and idarubicin inhibited the repair mechanism of DNA damage caused by these agents and resulted in synergistic cell killing as measured by induction of PARP and Caspase 3 cleavage. *In vivo*, selinexor (5 mg/kg) and radiation (3 Gy) decreased xenograft tumor size of non-small cell lung cancer (A549 cell line) by 12% and 30% respectively, relative to vehicle whereas combination of selinexor and radiation resulted in a 86% tumor decrease.

Conclusion: Selinexor inhibits the DNA repair mechanism in solid and hematological cancer cell lines and combination of selinexor with agents that cause DNA damage induces cancer cell death that is superior to each therapy alone. These data suggest that such a combination treatment is predicted to result with synergistic therapeutic outcome in cancer patients.

Selinexor Reduces Levels of DNA Damage Repair Proteins (DDR)

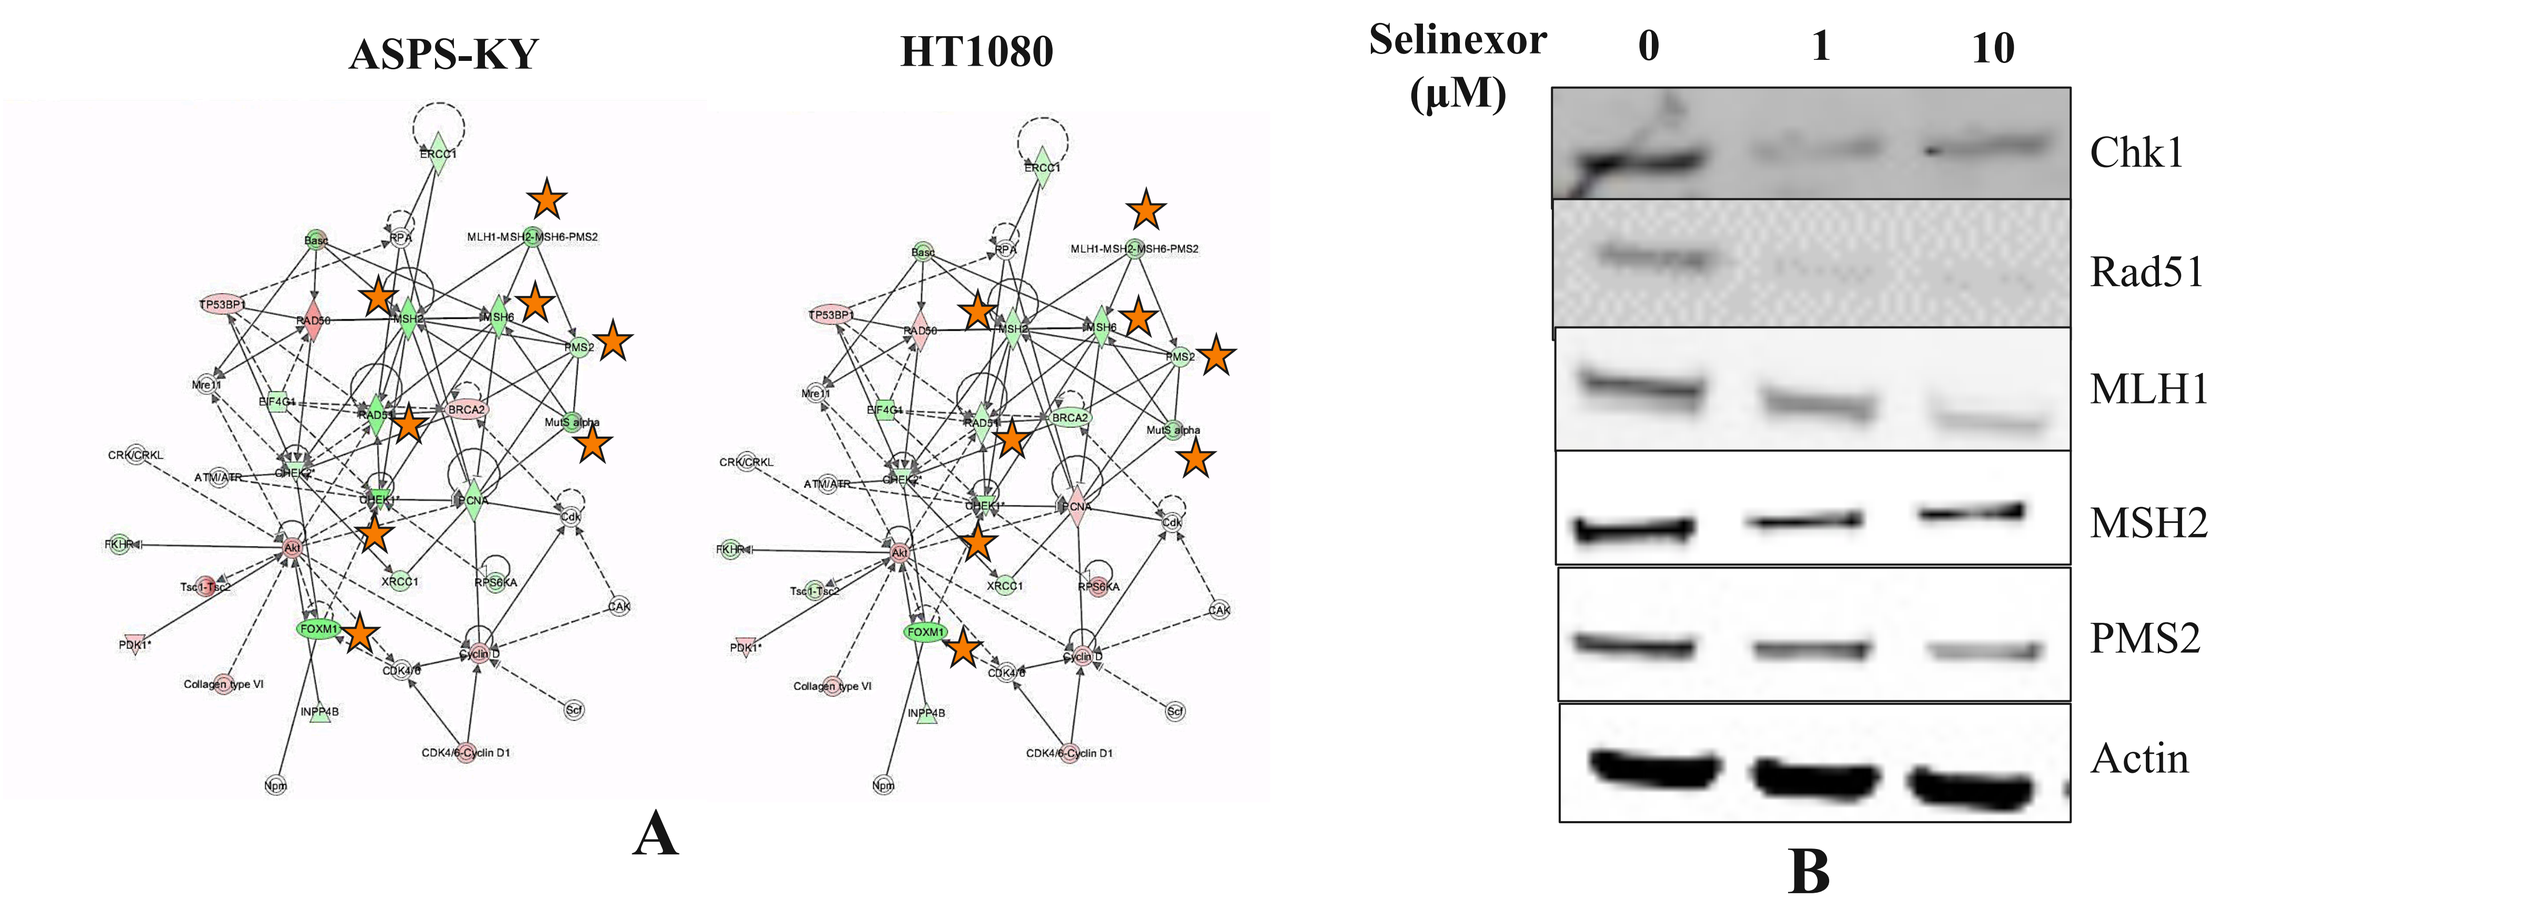
ASPS-KY
HT1080
Selinexor (µM)
0 1 10
Chk1
Rad51
MLH1
MSH2
PMS2
Actin
A
B

Figure 1: A. Ingenuity Pathway Analysis (IPA) of 150 proteins from cell lysates tested by Reverse Phase Protein Array (RPPA) from selinexor-treated sarcoma cell lines ASPS-KY (Alveolar soft part Sarcoma) and HT1080 (Fibrosarcoma) revealed reduction in the expression of 8 proteins (red stars) with role in DNA damage repair. **B.** Whole protein lysates from ASPS-KY and HT1080 (not shown) on immunoblots confirmed these results.

Selinexor Regulates Expression of DDR Genes on the RNA and Protein Levels

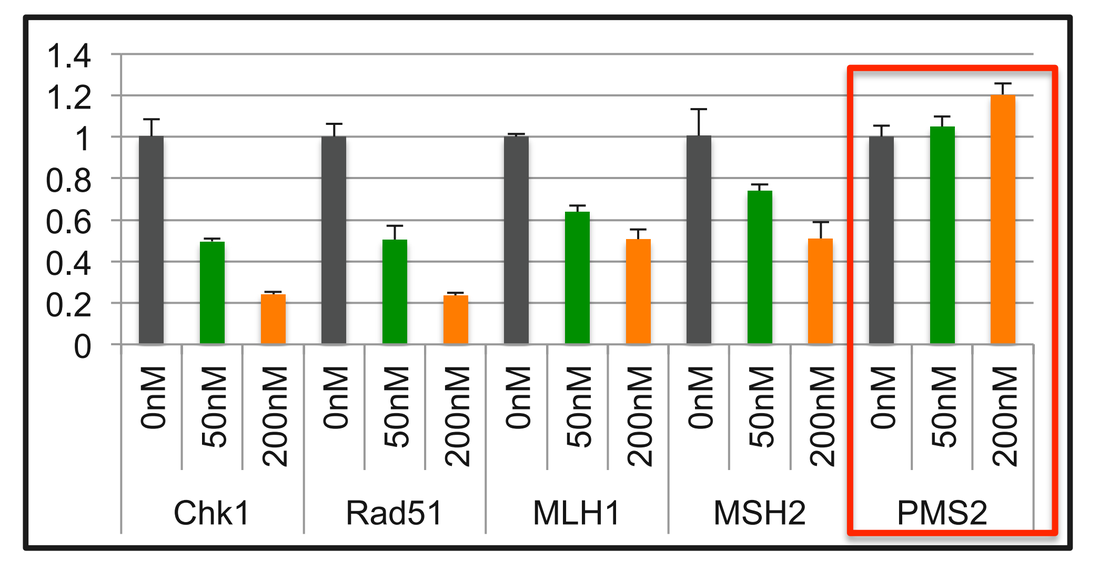
1.4
1.2
1
0.8
0.6
0.4
0.2
0
0nM 50nM 200nM
Chk1
Rad51
MLH1
MSH2
PMS2

Figure 2: MOLM13 (Acute Monocytic Leukemia) cells were treated with selinexor for 12 hr. Real time PCR indicated that selinexor reduced the transcript levels of Chk1, Rad51, MLH1, MSH2 in a dose dependent manner, except for PMS2 (marked by red box).

Selinexor Down Regulates Expression of DDR Proteins in Solid and Hematological Cancer Cells Where More Reduction Observed in More Sensitive Cell Lines

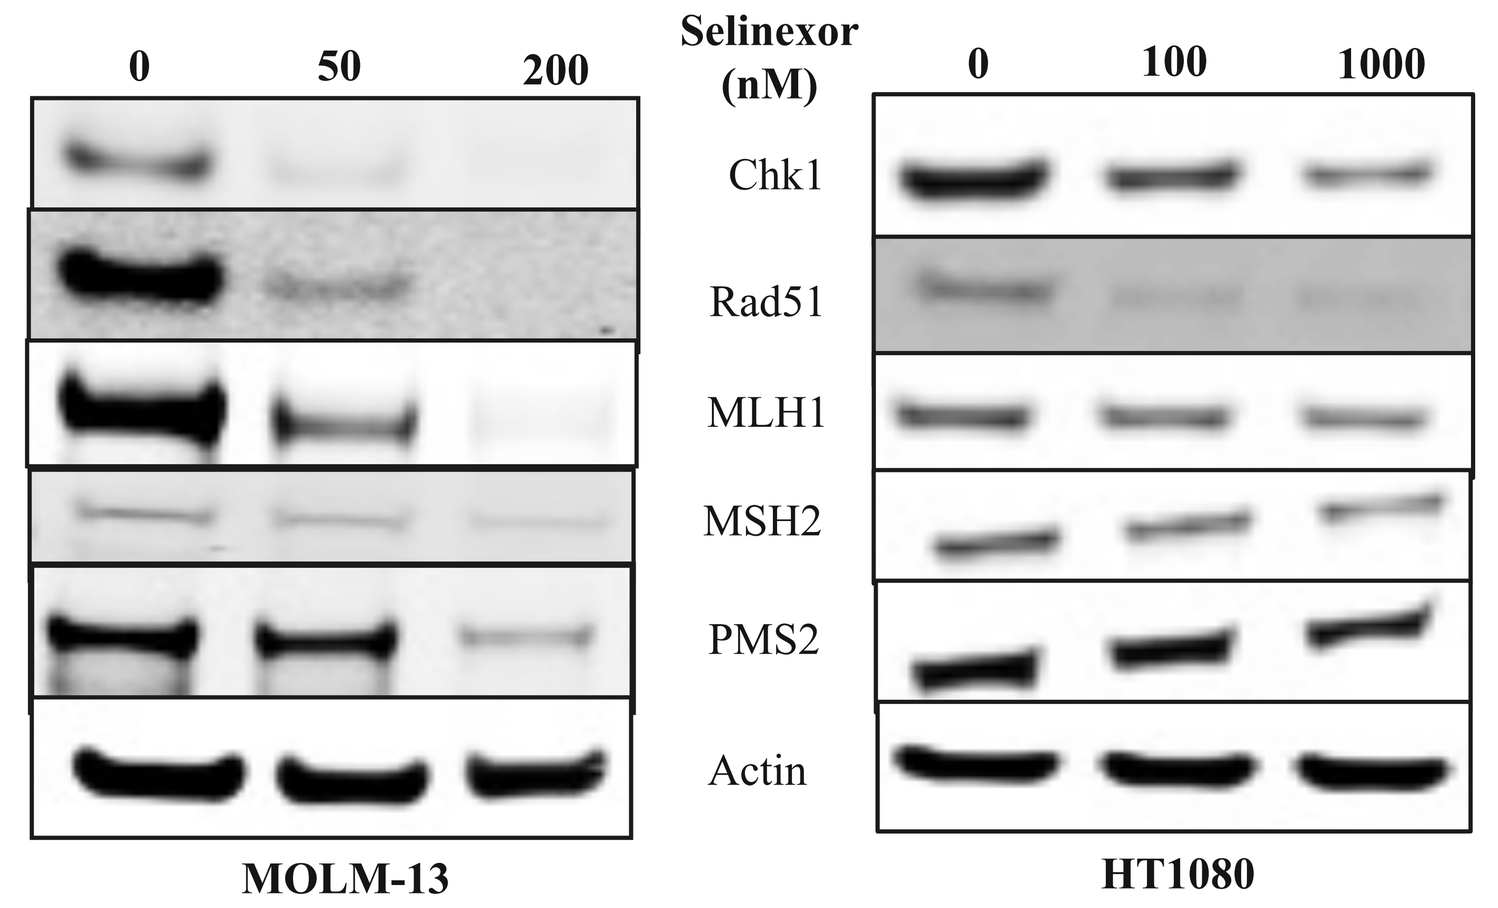
0 50 200
Selinexor (nM)
0 100 1000
Chk1
Rad51
MLH1
MSH2
PMS2
Actin
MOLM-13
HT1080

Figure 3A: Immunoblots of whole cell lysates from MOLM13 (Acute Monocytic Leukemia) and HT1080 (Fibrosarcoma) cells treated with selinexor for 24hrs shows reduction of DNA damage repair proteins Chk1, Rad51, MLH1, MSH2, PMS2 in a dose dependent manner in both hematological and solid cancer cell lines.

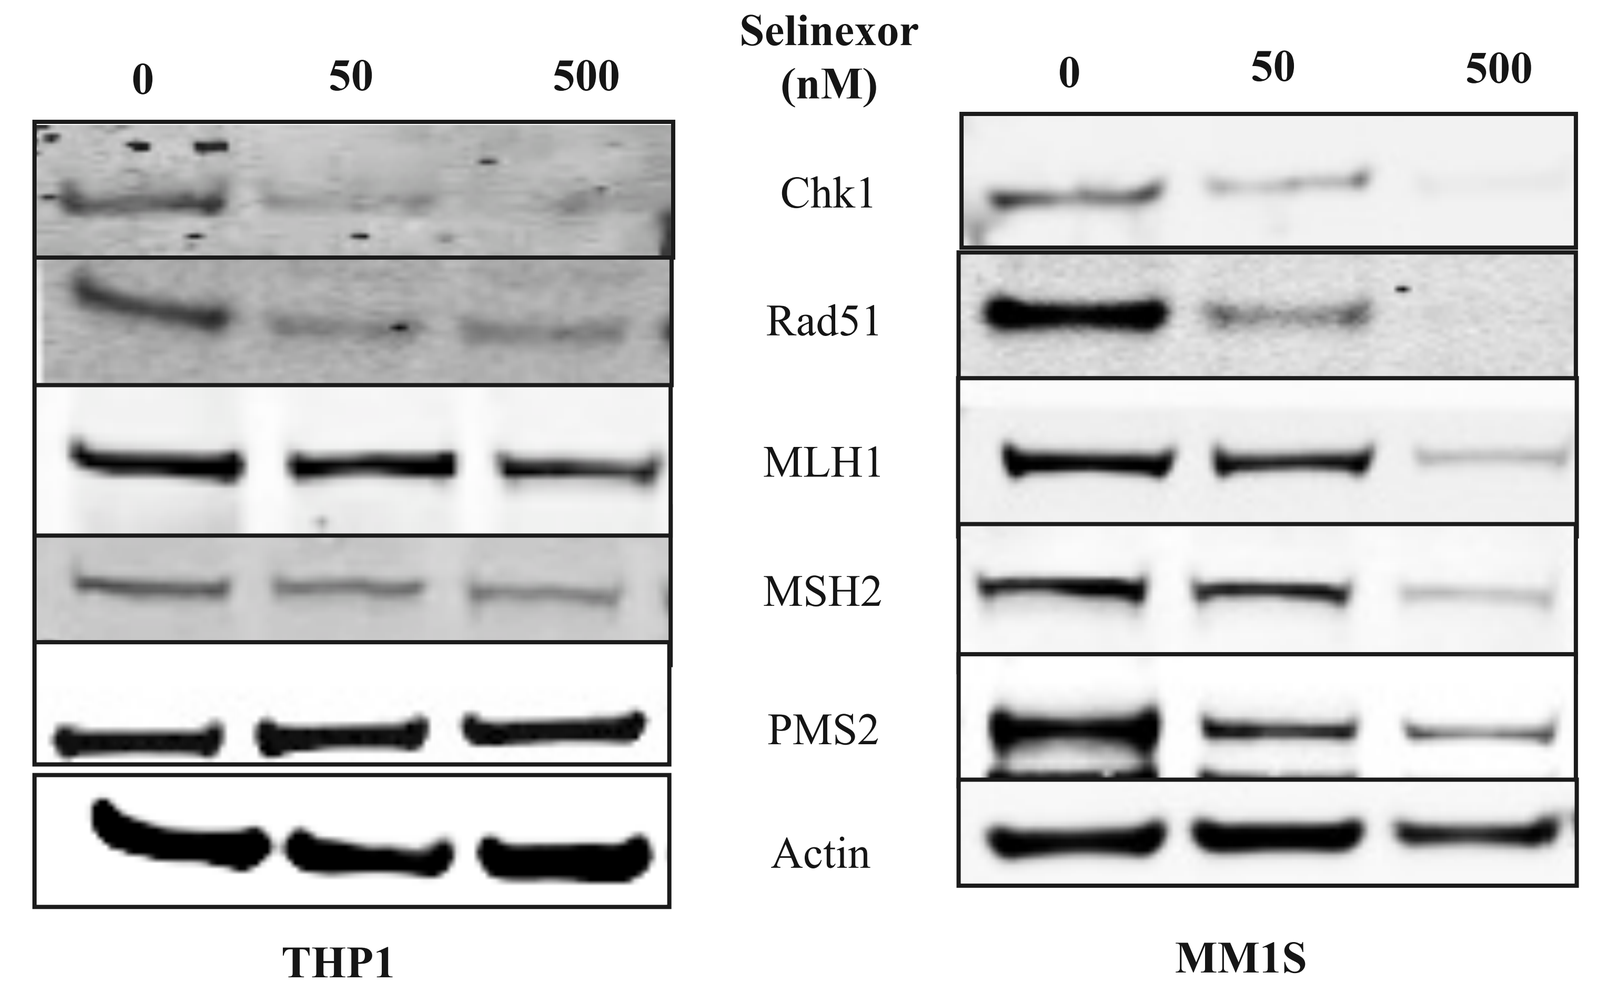
0 50 500
Selinexor (nM)
0 50 500
Chk1
Rad51
MLH1
MSH2
PMS2
Actin
THP1
MM1S

Figure 3B: Immunoblots of whole cell lysates from selinexor resistant (IC_{50} ~ 3µM) THP-1(Acute Monocytic Leukemia) and selinexor sensitive (IC_{50} ~ 100nM) MM.1S (Multiple Myeloma) cells demonstrates that protein level reduction is associated with sensitivity of the cell line to selinexor.

Selinexor Inhibits Recovery From Chemotherapeutic Agents Induced DNA Damage

2hrs 10nM Idarubicin	-	+	+	+
48hrs 100nM Selinexor	-	-	-	+
48hrs Recovery	-	-	+	-

A

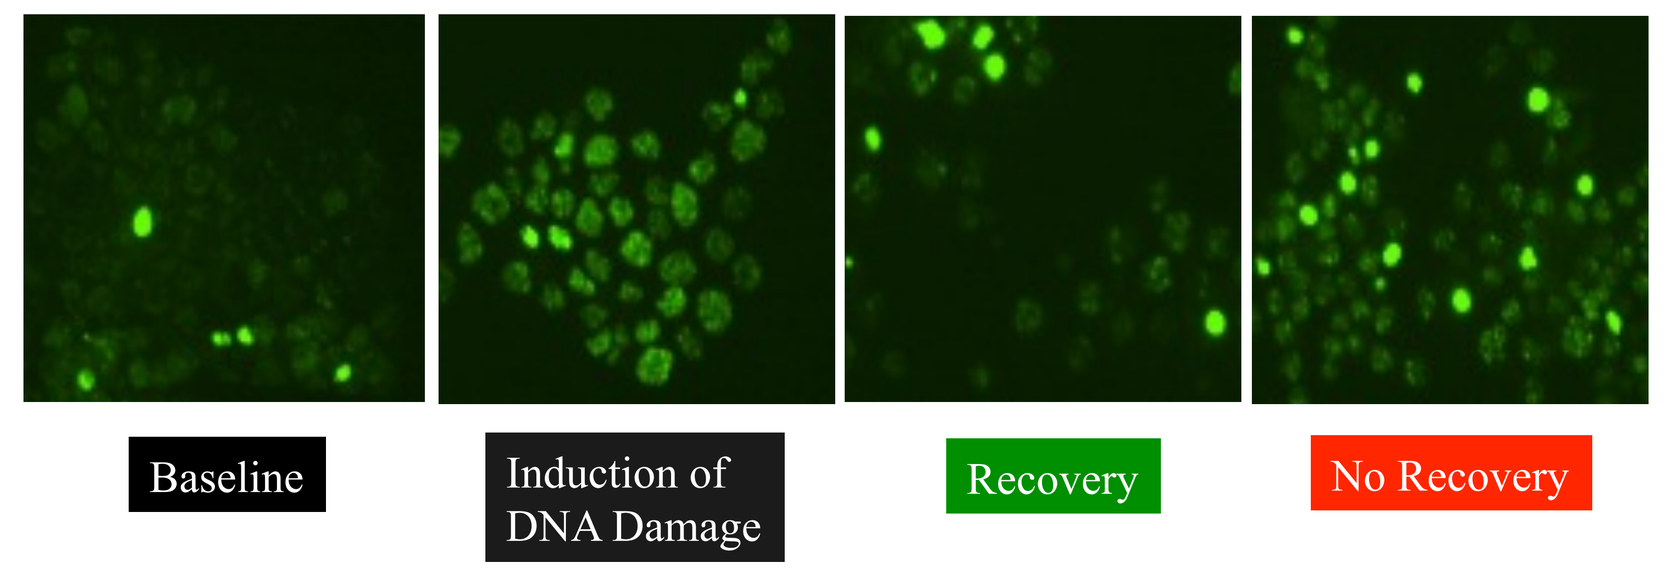
Baseline
Induction of DNA Damage
Recovery
No Recovery

2hrs 0.5µg/mL Doxorubicin	-	+	+	+	-
48hrs 1µM Selinexor	-	-	-	+	+
48hrs Recovery	-	-	+	-	-

B

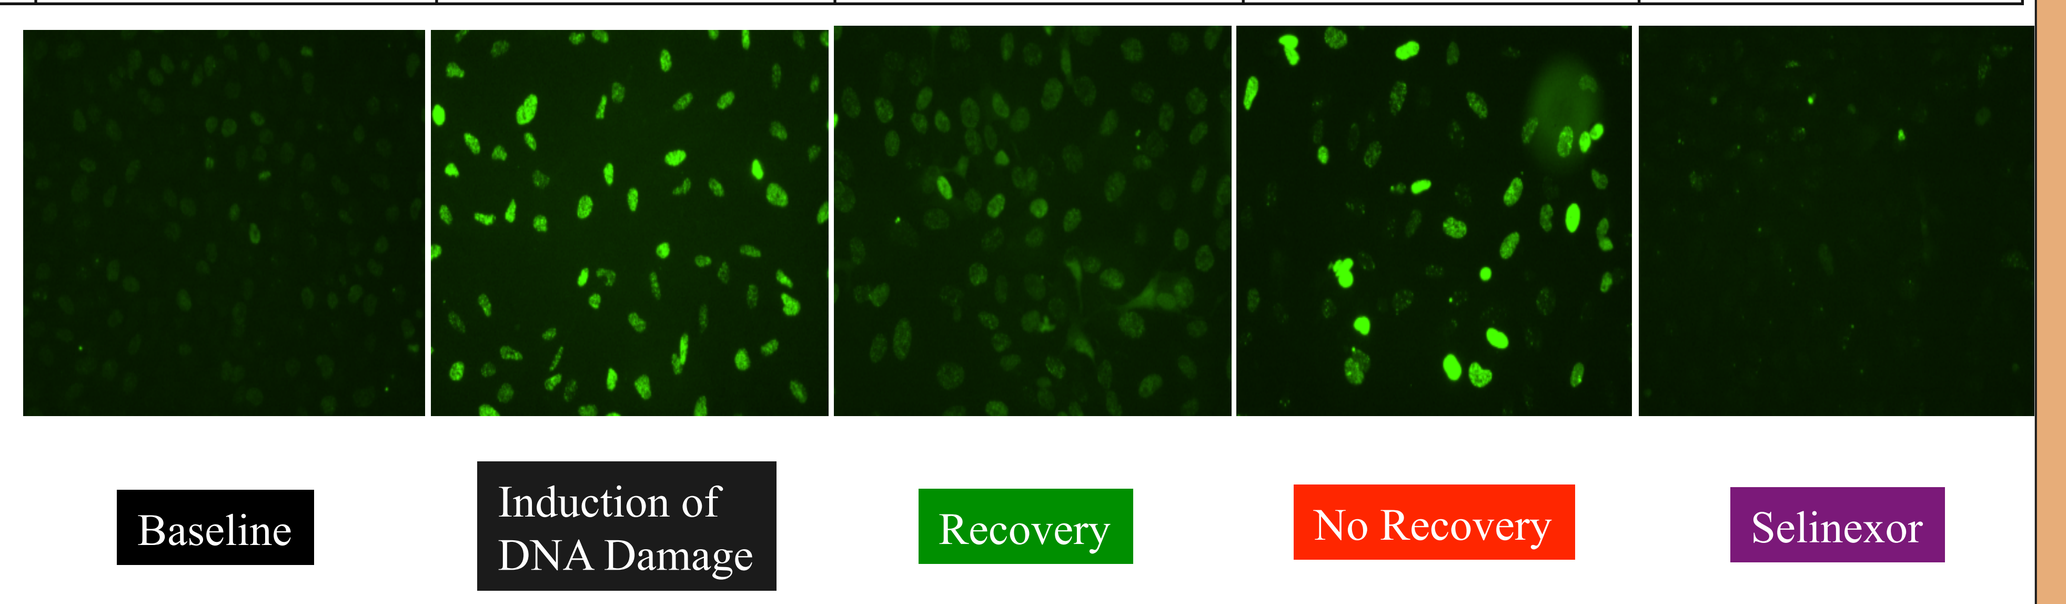
Baseline
Induction of DNA Damage
Recovery
No Recovery
Selinexor

Figure 5: Immunofluorescence of MV-4-11 (Acute Monocytic Leukemia) in **A** and U20S (Osteosarcoma) cells in **B** that were treated for 2hrs with either idarubicin or doxorubicin respectively. The cells were then allowed to recover with or without the presence of selinexor for 48hrs. DNA damage is detected by staining with anti gamma H2A.X. Idarubicin and doxorubicin caused irreversible DNA damage in the presence of selinexor. Importantly, selinexor alone did not induce DNA damage.

Selinexor Inhibits Homologous Recombination DNA Repair

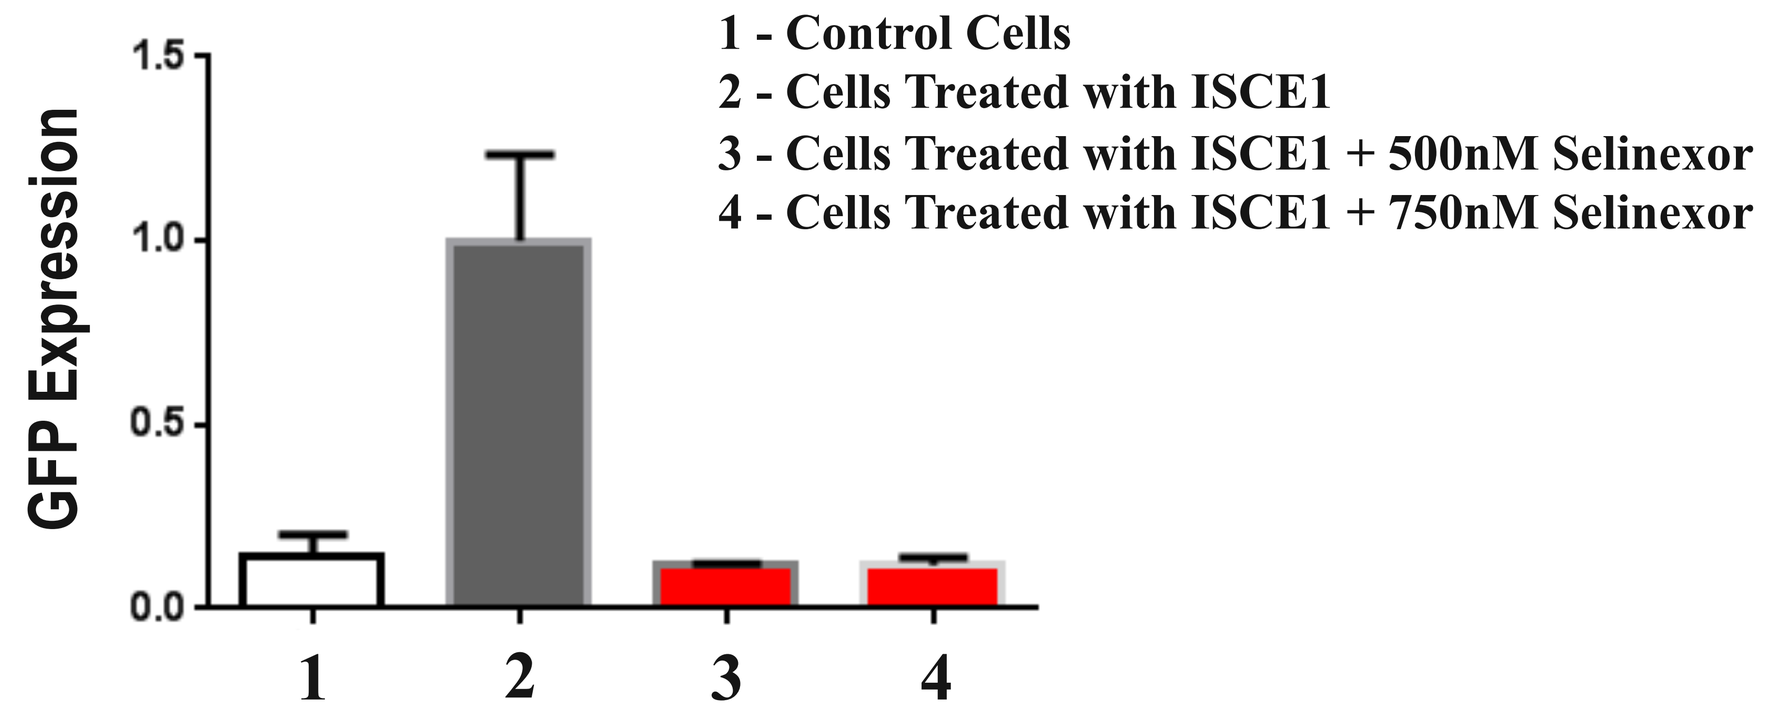
GFP Expression
1.5
1.0
0.5
0.0
1 2 3 4
1 - Control Cells
2 - Cells Treated with ISCE1
3 - Cells Treated with ISCE1 + 500nM Selinexor
4 - Cells Treated with ISCE1 + 750nM Selinexor

Figure 6: ISCE1 treatment of the engineered cells that express the synthetic meganuclease consensus sequence - GFP reporter gene results in DNA homologous recombination and allows GFP expression once DNA strands re-anneal. Selinexor inhibited DNA repair and therefore did not allow GFP expression.

Pre-treatment With DNA Damage Inducing Agents Followed By Selinexor is More Cytotoxic Than Selinexor Pre-treatment

p = Pre-incubation, no washout
w = Pre incubation then Washout

48hrs 8nM Idarubicin	-	-	-	-	-	-	-	+p
48hrs 100nM Selinexor	-	-	-	-	-	-	+p	-
24hrs 8nM Idarubicin	-	-	+	-	+w	+w	+	-
24hrs 100nM Selinexor	-	-	+w	+w	+	-	-	+
48hrs Selinexor + Idarubicin	-	+	-	-	-	-	-	-

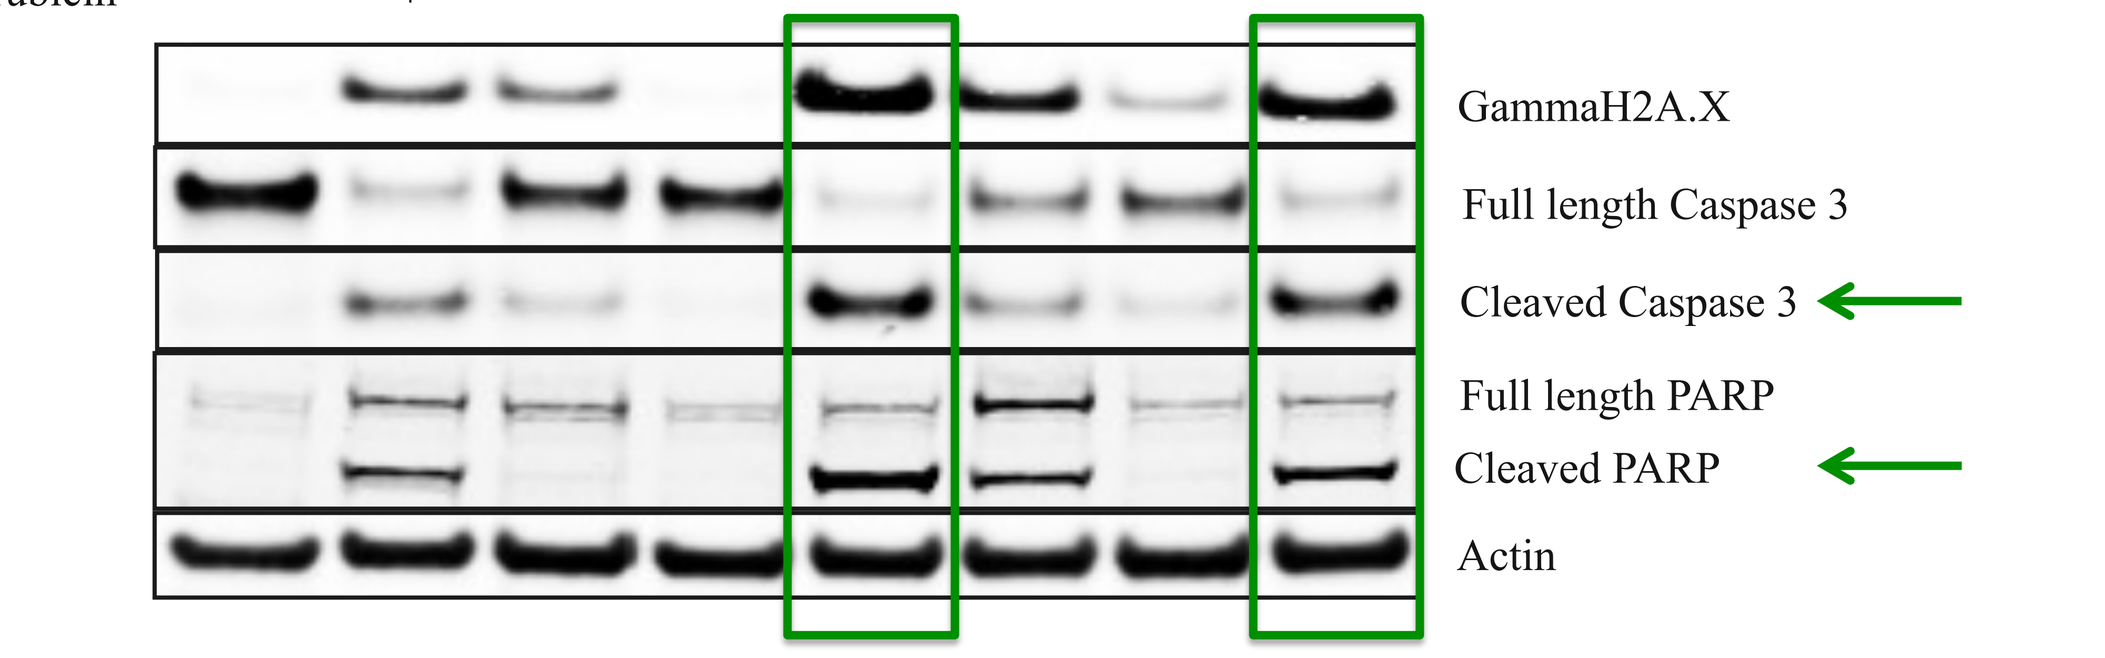
GammaH2A.X
Full length Caspase 3
Cleaved Caspase 3
Full length PARP
Cleaved PARP
Actin

Figure 7: MOLM-13 cells were treated with idarubicin or selinexor for 24 hr and then were incubated with the other drug for additional 24 hr with (+w) or without (+p) washout of the first drug. Pre-treatment with idarubicin followed by exposure to selinexor induced more DNA damage and cell death than pre-treatment with selinexor (Cleaved Caspase 3 and PARP in green boxed immunoblots).

Combination Treatment of Selinexor with Chk1 inhibitors Induced Additive Cytotoxic Effects

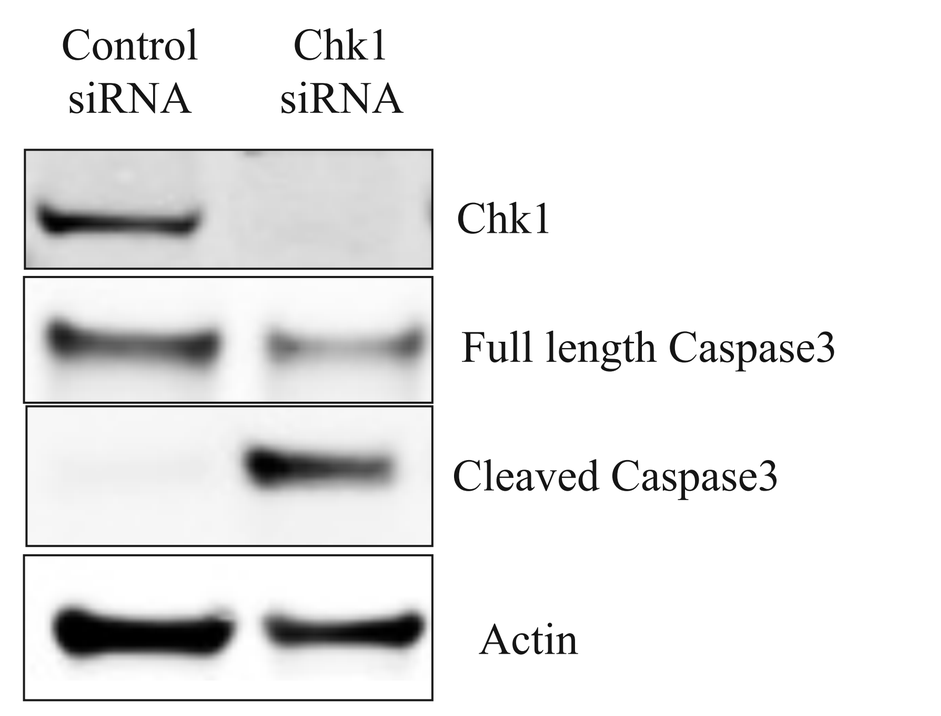
Control siRNA
Chk1 siRNA
Chk1
Full length Caspase3
Cleaved Caspase3
Actin

Figure 8A: Knockdown of Chk1 alone resulted in HT1080 cell death as seen by induction of cleaved Caspase3. However, silencing of the DNA damage repair genes Rad51, MLH1, MSH2 and PMS2 did not affect cell viability (not shown).

	Selinexor IC_{50}		
Concentration of each compound	**UCN-01** (**IC_{50} - 1.71 uM**)	**AZD7762** (**IC_{50} - 680 nM**)	**CHIR-24** (**IC_{50} - 1.1 uM**)
0	**604 ± 70 nM**		
160nM	**148 nM**	**170 nM**	**140 nM**
630nM	**63 nM**	**50nM**	**20 nM**

Figure 8B: HeLa cells (Cervical Adenocarcinoma) treated with various concentrations of selinexor in the presence of Chk1 inhibitors showed additive cytotoxic IC_{50} values.

Combination Treatment of Selinexor with Radiation shows Synergistic Effects in Lung Cancer Xenograft Model

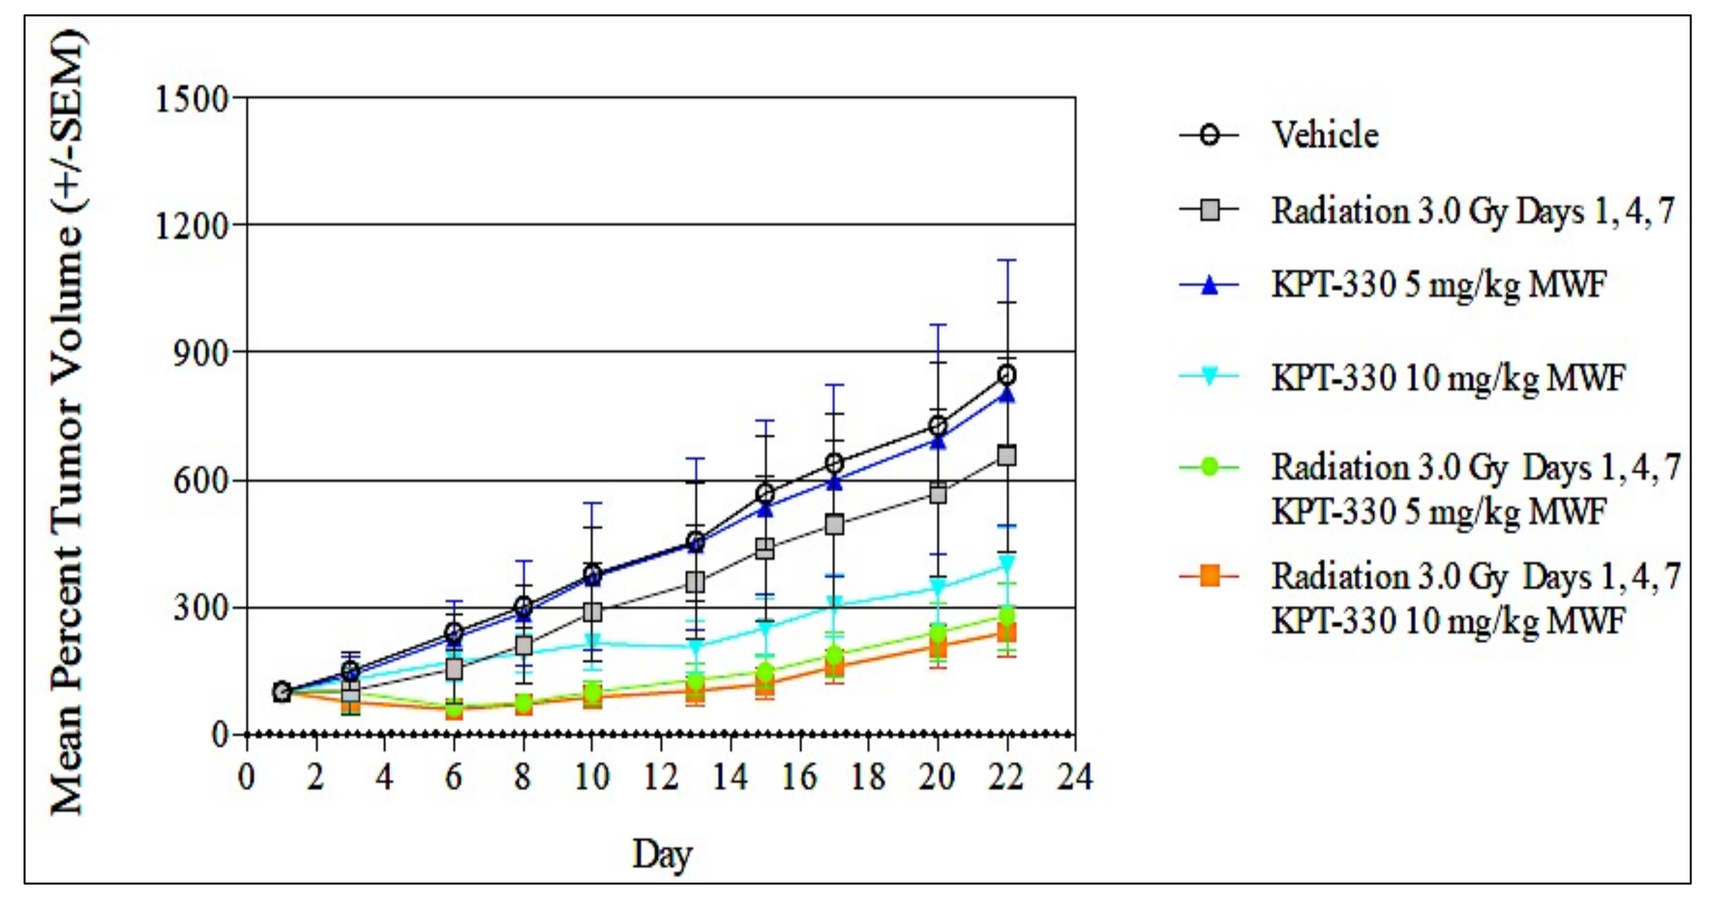
Mean Percent Tumor Volume (+/-SEM)
1500
1200
900
600
300
0
0 2 4 6 8 10 12 14 16 18 20 22 24
Day
Vehicle
Radiation 3.0 Gy Days 1, 4, 7
KPT-330 5 mg/kg MWF
KPT-330 10 mg/kg MWF
Radiation 3.0 Gy Days 1, 4, 7 KPT-330 5 mg/kg MWF
Radiation 3.0 Gy Days 1, 4, 7 KPT-330 10 mg/kg MWF

A

5mg/kg Selinexor	-	+	-	+
3.0 Gy Radiation	-	-	+	+

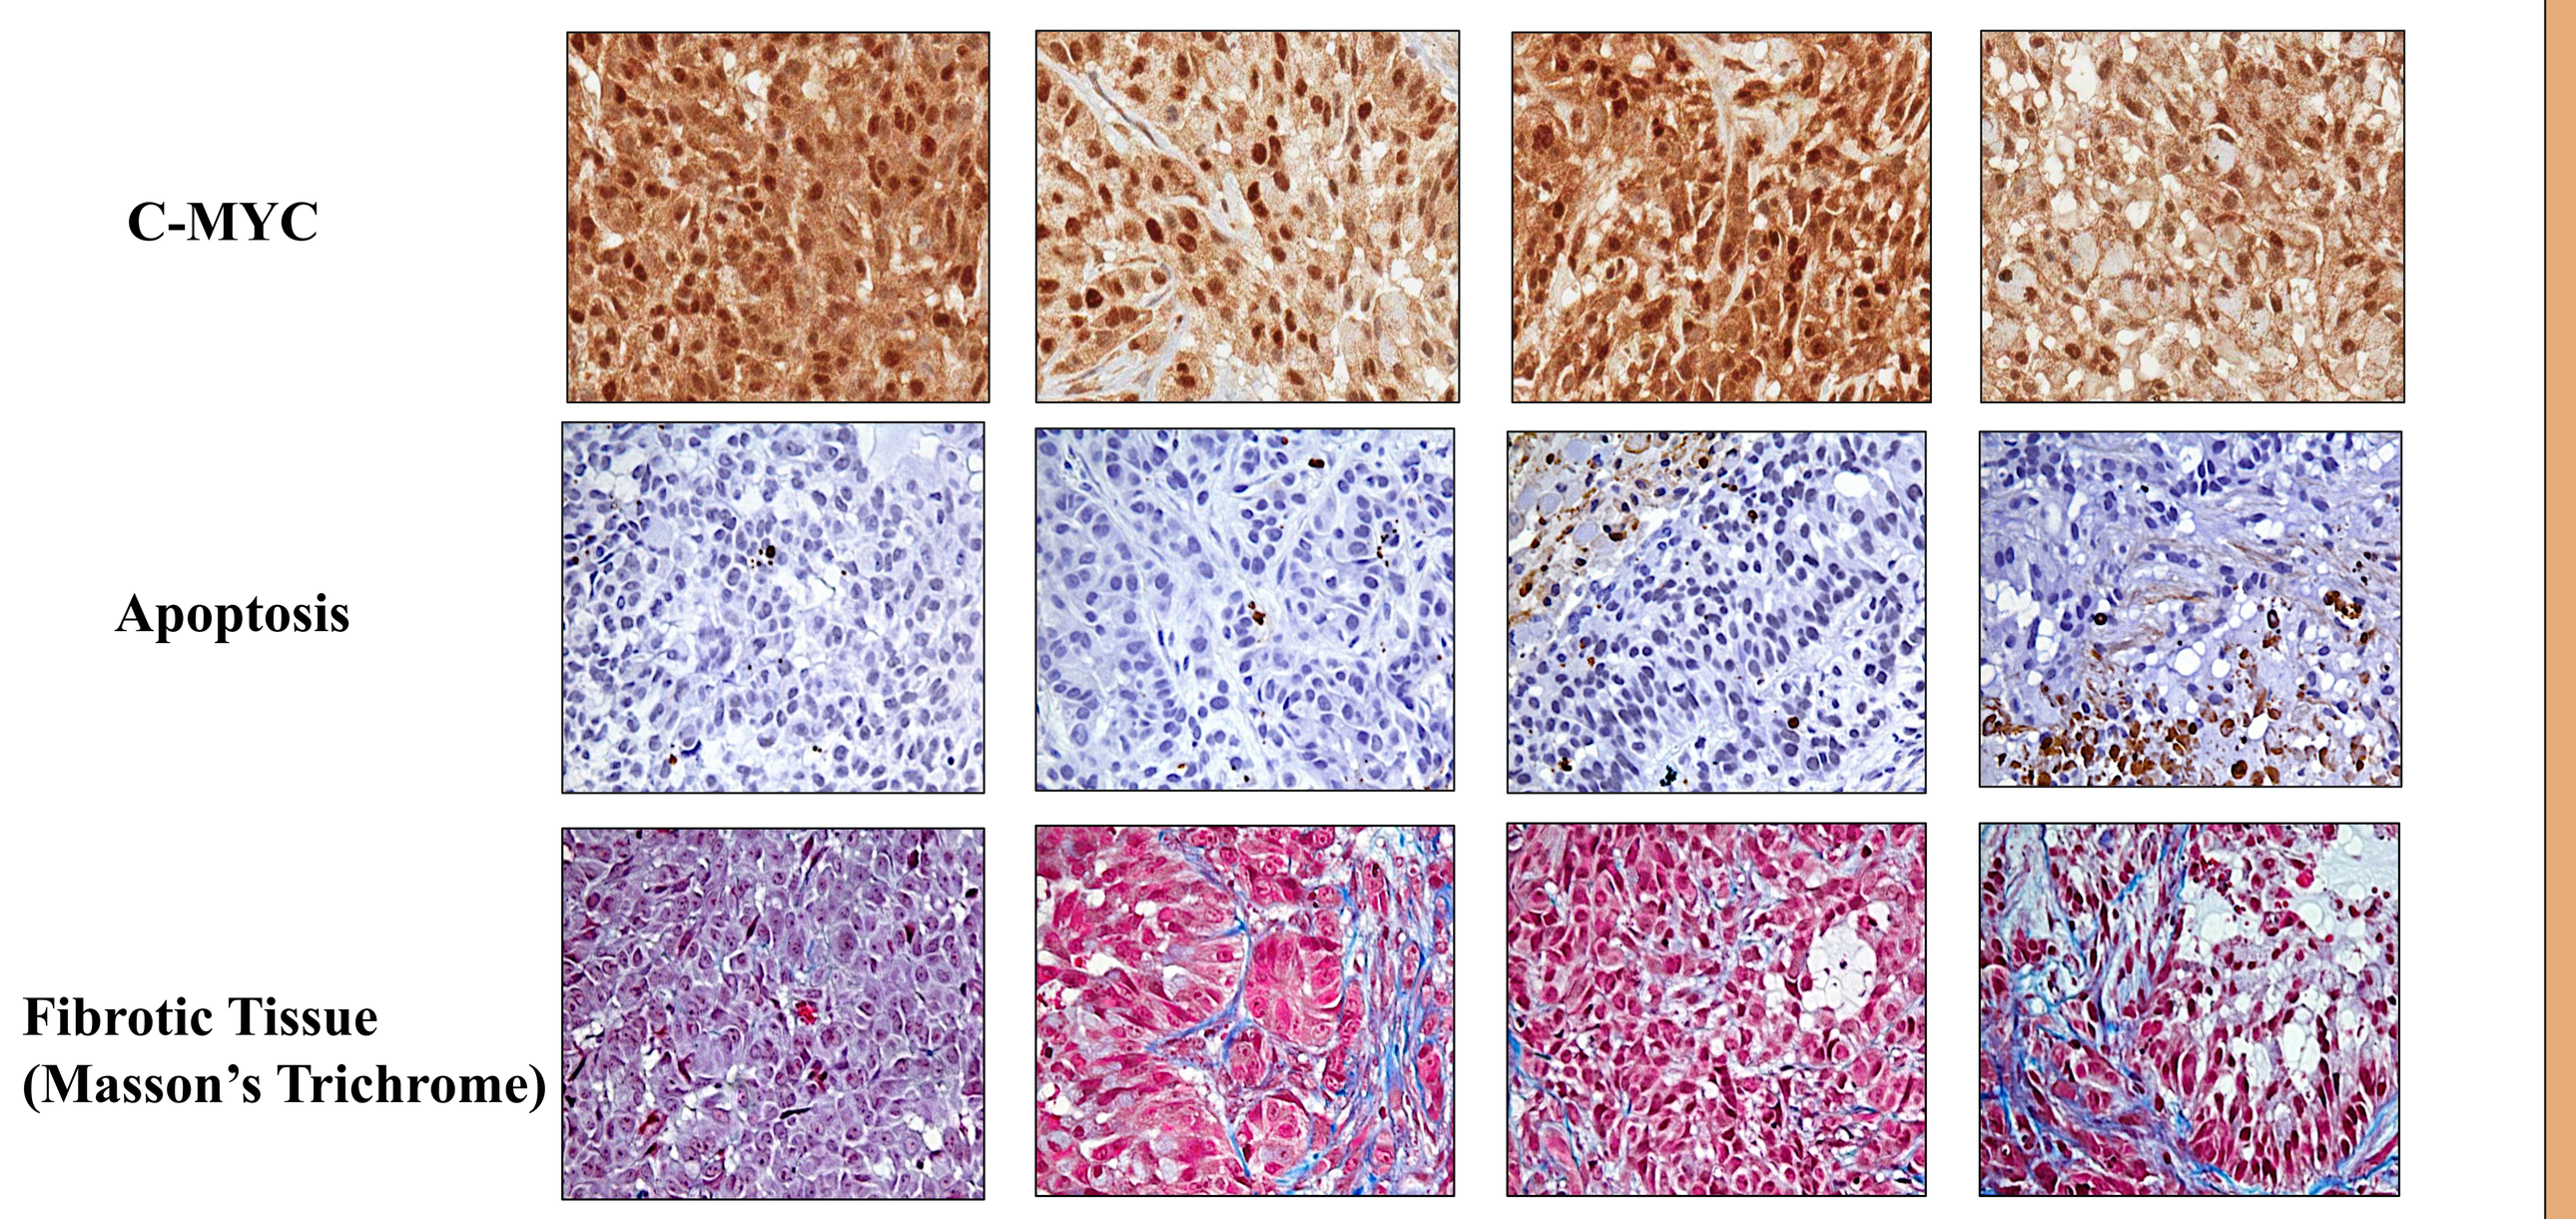
C-MYC
Apoptosis
Fibrotic Tissue
(Masson's Trichrome)

B

Figure 9: A. CB-17 SCID mice inoculated with A549 (Lung Adenocarcinoma) cells were treated with vehicle, radiation (1.5 Gy or 3.0 Gy), selinexor/KPT-330 (5mg/kg or 10mg/kg) or selinexor plus radiation. Vehicle and selinexor were given on a QoD x3/wk schedule beginning on Day 1. Radiation was given on Days 1, 4 and 7. In groups receiving single agent treatment, statistically significant reductions in tumor growth (P<0.05) were seen only at the higher selinexor doses. Statistically significant reductions in tumor growth were seen in all groups treated with selinexor plus radiation (P<0.05) and the reductions were approximately equal for both doses of radiation and selinexor used.
B. Histology sections showed greater extent of cell growth inhibition (c-MYC reduction), more apoptosis and fibrosis in the combination treatment compared with each single agent therapy.

Summary of Results and Conclusions

- Selinexor treatment down-regulated the expression of DDR genes at the transcriptional and the post-translational levels.
- Selinexor down-regulated DDR in both solid and hematological tumor cell lines and the effects appear to correlate with cell sensitivity to Selinexor.
- Inhibition of the DDR gene Chk1 is toxic for cancer cells and selinexor drug combination with Chk1 inhibitors showed additive cytotoxic effects.
- Selinexor treatment did not induce DNA damage, but inhibited cell recovery from DNA damage leading to increased cell death.
- Pre-treatment with DNA damage inducing agents followed by exposure to selinexor was more toxic than the reversed sequential treatment with the two drugs.
- Selinexor treatment in combination with radiation showed synergistic anti-tumor activity in a NSCLC xenograft mouse model.
- The combination treatment of selinexor with DNA damage-inducing treatment is predicted to result in improved therapeutic outcome in cancer patients.

Contact Information: Trinayan Kashyap
e-mail: trinayan@karyopharm.com T: +1 617-658-0559