Preclinical and Early Clinical Activity of the Oral Selective Inhibitor of Nuclear Export (SINE™) Exportin 1 (XPO1) Antagonist Selinexor (KPT-330) in Patients (pts) with Platinum Resistant/Refractory Ovarian Cancer (OvCa)

Abstract 7042

John Martignetti[1], Albiruni RA Razak[2], Ying Chen[1], Nashat Y Gabrail[3], John F Gericitano[4], Catalina Camacho[1], Elena Pereira[1], Brad Evans[1], Peter Dottino[1], Sharon Shacham[5], Dilara McCauley[5], Tami Rashal[5], Jean-Richard Saint-Martin[5], Eran Shacham[5], Darcy Vincett[6], Robert Carlson[5], Michael Kauffman[5], Mansoor R Mirza[5], Morten M Sorensen[7]

(1) Genetics & Genomic Sciences, Icahn School of Medicine at Mount Sinai, New York, NY (2) Drug Development Program, Princess Margaret Cancer Center, Toronto, Canada; (3) Gabrail Cancer Center, Canton, OH, USA; (4) Memorial Sloan Kettering Cancer Center, New York, NY, USA; (5) Karyopharm Therapeutics Inc, Newton, MA, USA; (6) Ozmosis Research Inc, Toronto, Canada; (7) Dept. of Oncology, Rigshospitalet, Copenhagen, Denmark;

Mechanism of Action

- Exportin 1 (XPO1) is the major nuclear export protein for tumor suppressors
- SINE™ compounds inhibit XPO1, leading to nuclear retention and reactivation of tumor suppressors leading to tumor cell apoptosis
- Selinexor is a novel, potent, oral SINE™ currently being evaluated in Phase 1/2 studies in solid and hematological malignancies
- Elevated XPO1 expression is associated with invasive ovarian cancers

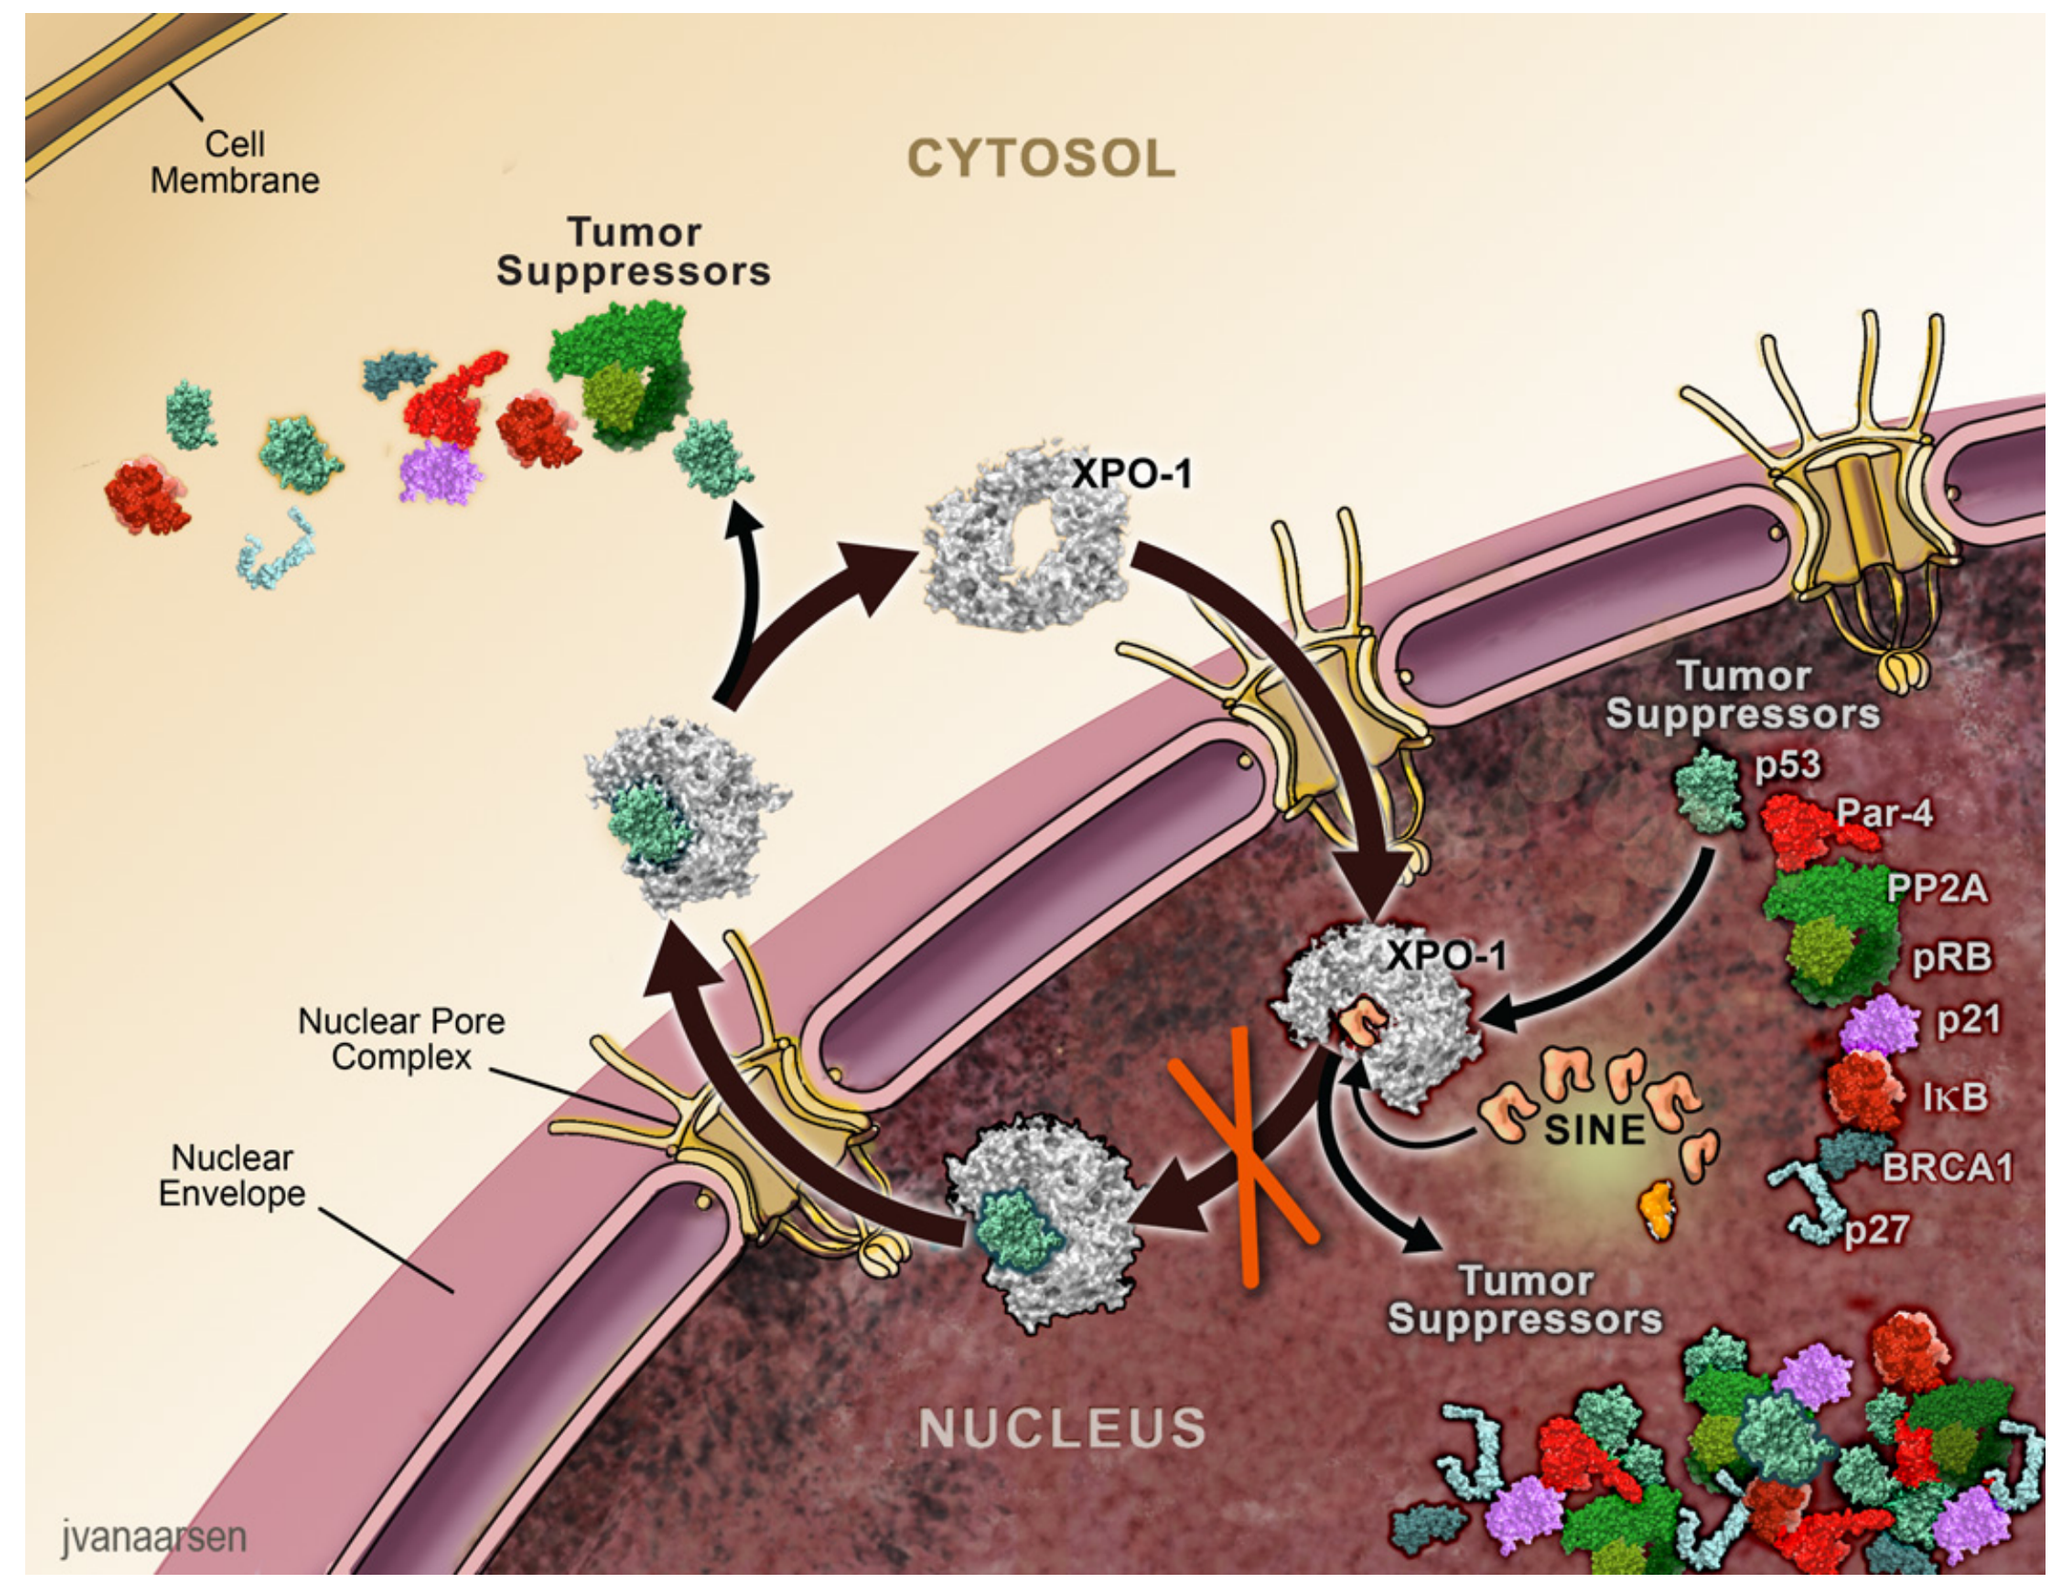
Cell Membrane
CYTOSOL
Tumor Suppressors
XPO-1
Tumor Suppressors
p53
Par-4
PP2A
pRB
p21
IκB
BRCA1
p27
XPO-1
SINE
Nuclear Pore Complex
Nuclear Envelope
Tumor Suppressors
NUCLEUS
jvanaarsen

- XPO1 is known to be the sole transporter for a number of proteins implicated in ovarian cancer tumorigenesis
 - Tumor suppressors p53 and BRCA1 are frequently mutated in ovarian cancers (96% and 9% respectively) and KLF6 expression is decreased
 - IκBα, an inhibitor of NF-κB which is frequently activated in ovarian cancers
 - Elevated expression of Topoisomerase IIa and the phosphatase PP2A (CIP2A) are associated with poor prognosis for ovarian cancer
 - Sequestosome 1 (p62), a stress response protein involved in protein shuttling, degradation and aggregation, which may play a role in ovarian cancer cisplatin resistance

SINE™ Induced Nuclear Localization

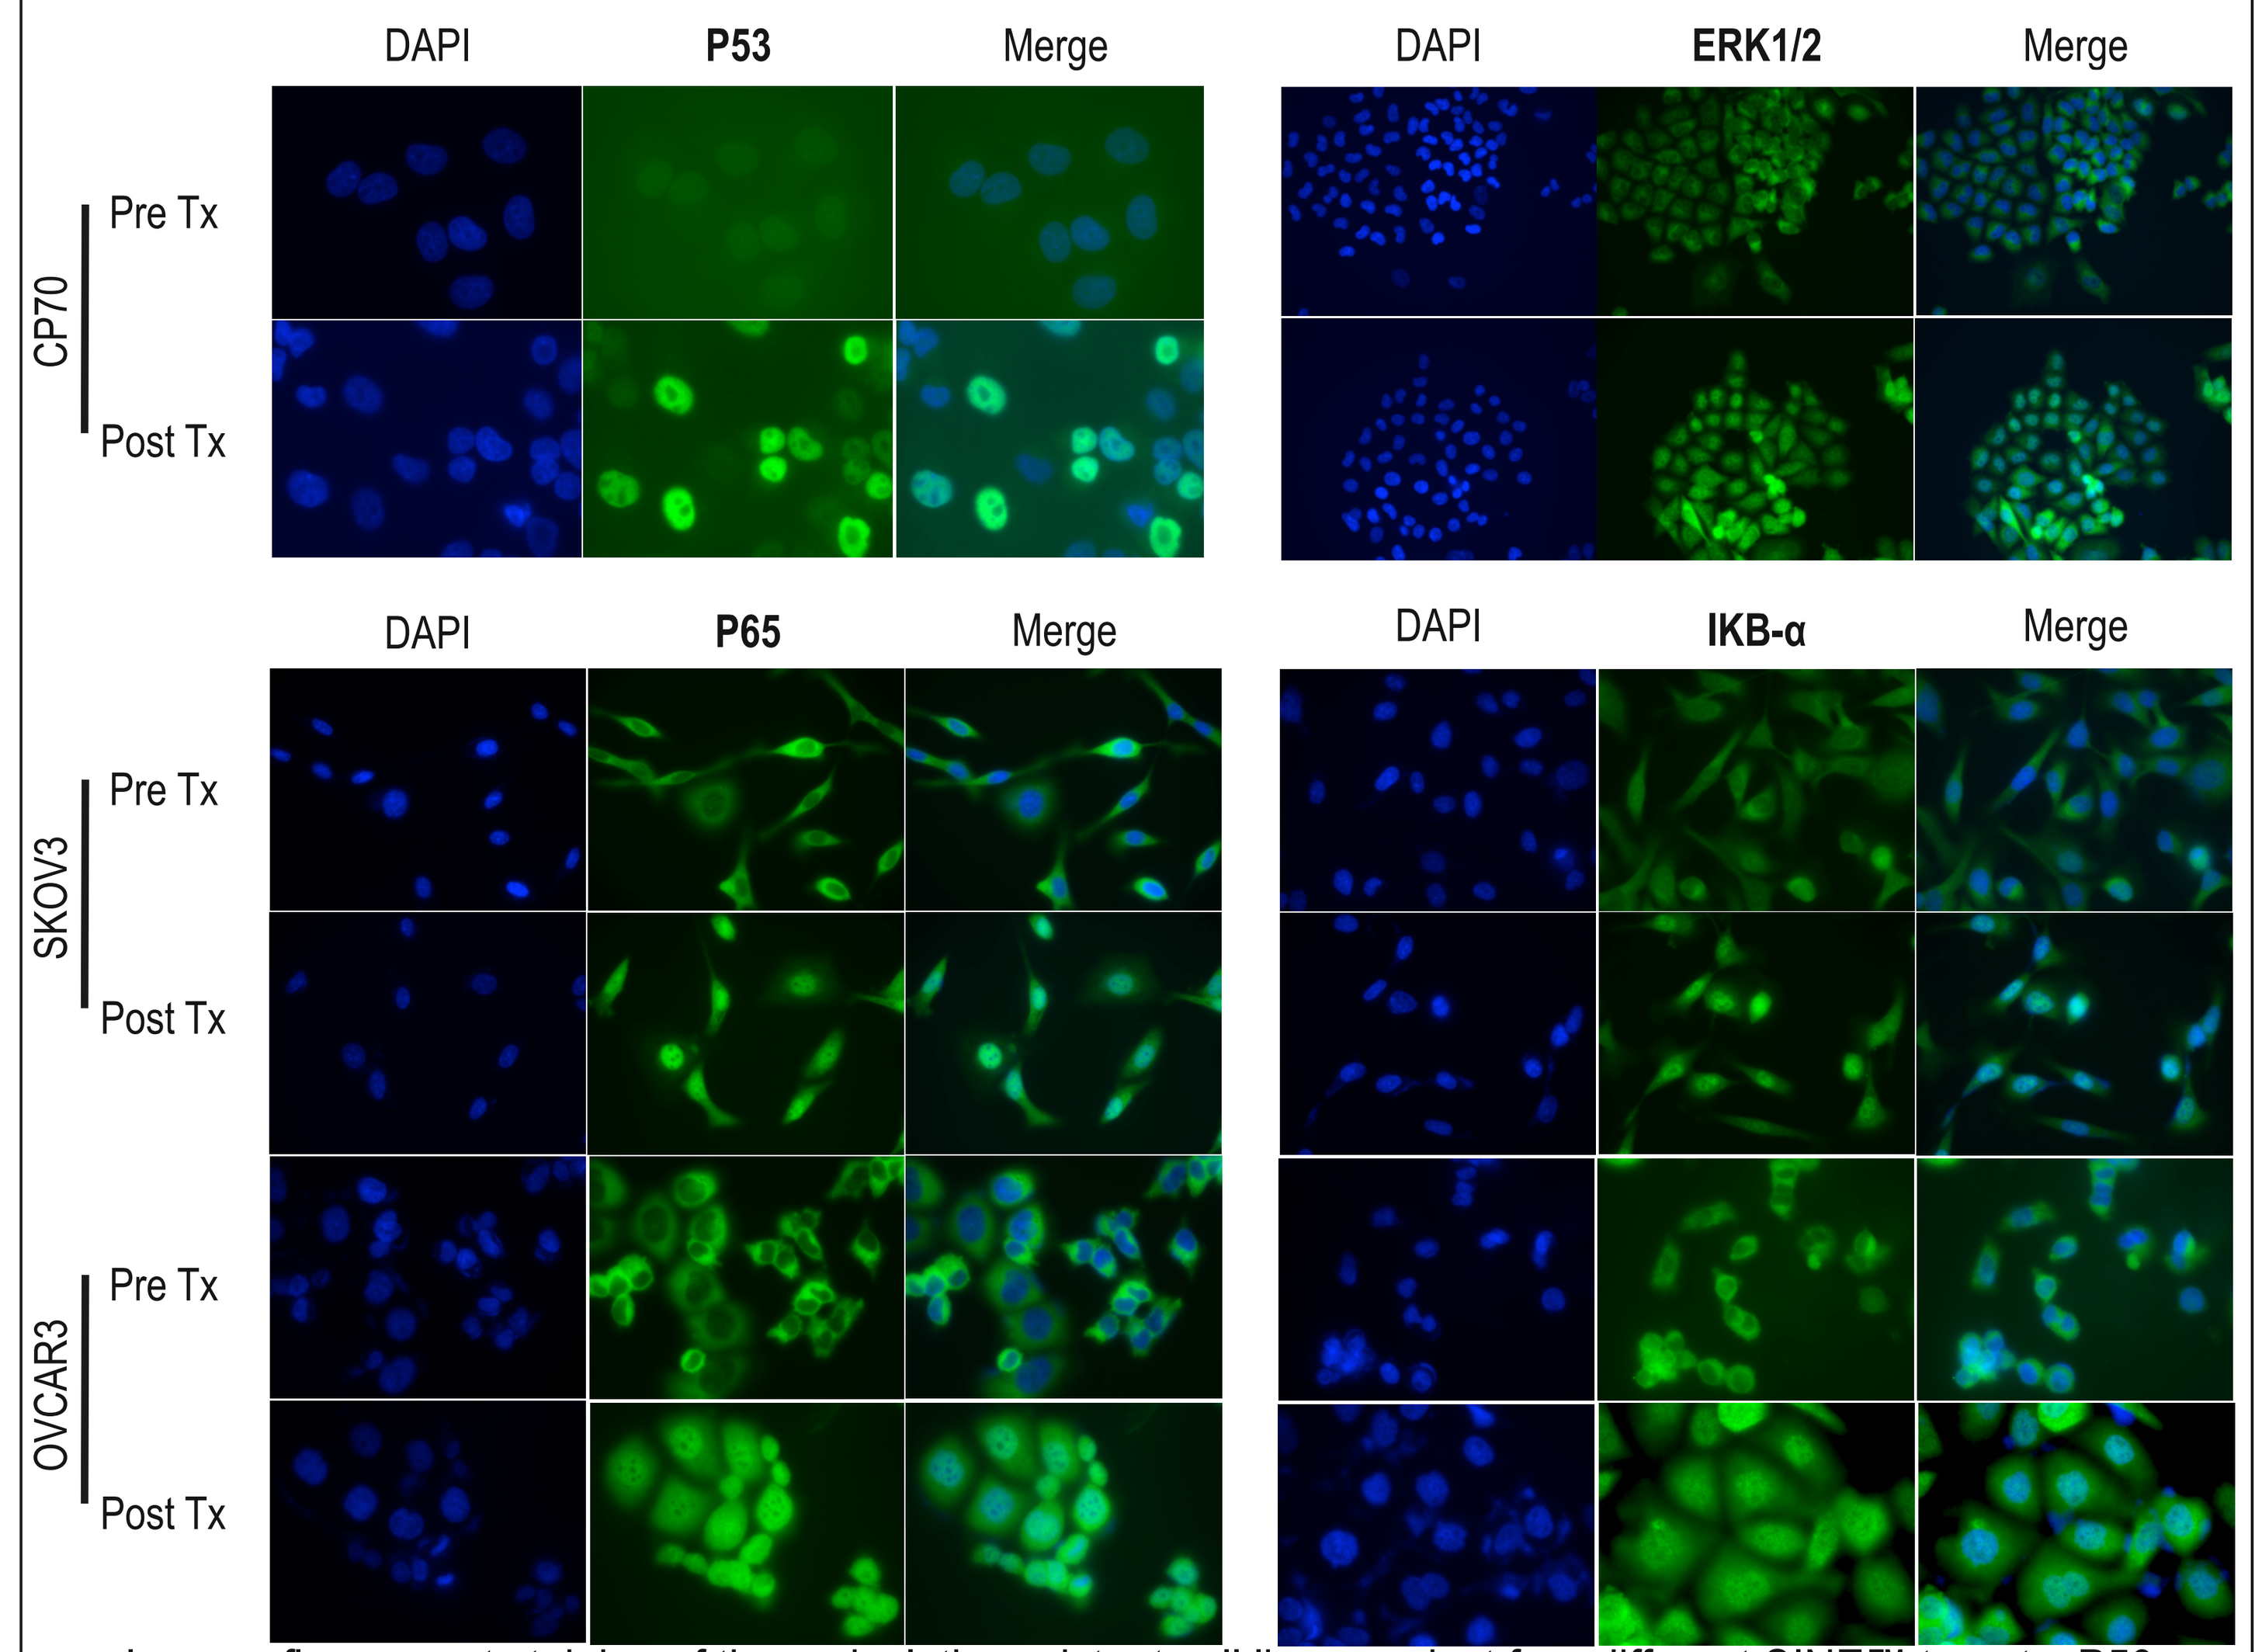
DAPI
P53
Merge
DAPI
ERK1/2
Merge
CP70
Pre Tx
Post Tx
DAPI
P65
Merge
DAPI
IKB-α
Merge
SKOV3
Pre Tx
Post Tx
OVCAR3
Pre Tx
Post Tx

Immunofluorescent staining of three cisplatin-resistant cell lines against four different SINE™ targets, P53, ERK1/2, P65 and IκB-α. Cell lines were treated with KPT-185 at their respective IC_{50} doses for 24 hours and harvested for staining. The merged images along with DAPI (blue staining, left of each set of panels) is used to reveal nuclear localization. Magnification is 600X.

Contact Information: Dr. John Martignetti e-mailjohn.martignetti@gmail.com

SINE™ Selectively Kills OvCA Tumor Cells and Acts Synergistically to Overcome Cisplatin Resistance in Both p53 WT and MT Cells

A

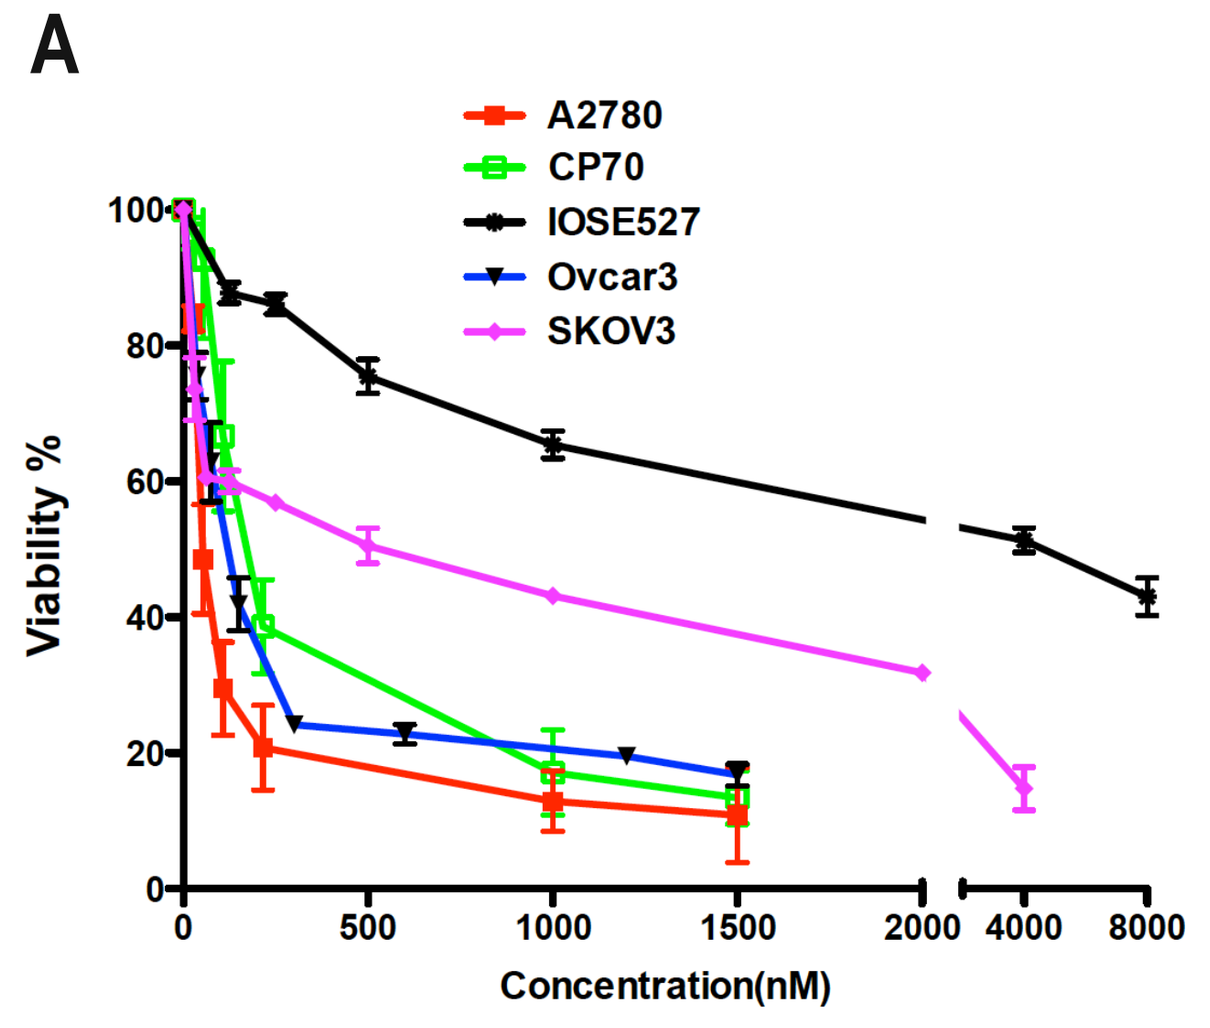
A2780
CP70
IOSE527
Ovcar3
SKOV3
Viability %
Concentration(nM)

IC_{50} of KPT-185 (nM)

IOSE527	A2780	CP70	OVCAR3	SKOV3
4224	46.53	111.7	116.1	328.7

A) Five well-characterized p53 WT/MT ovarian-derived cell lines -- A2780, cisplatin sensitive (WT P53), and its isogenic cisplatin resistant clone CP70, SKOV3 (P53 del) and OVCAR3 (P53 mt) were analyzed for their response to treatment with KPT-185. IOSE527 is an immortalized ovarian surface epithelial cell line. KPT-185 resulted in tumor cell specific cell death – IC_{50} values ranged from 46-330 nm - regardless of cisplatin sensitivity or p53 status. These doses had no toxic effects on the noncancerous IOSE527 cell line.

B

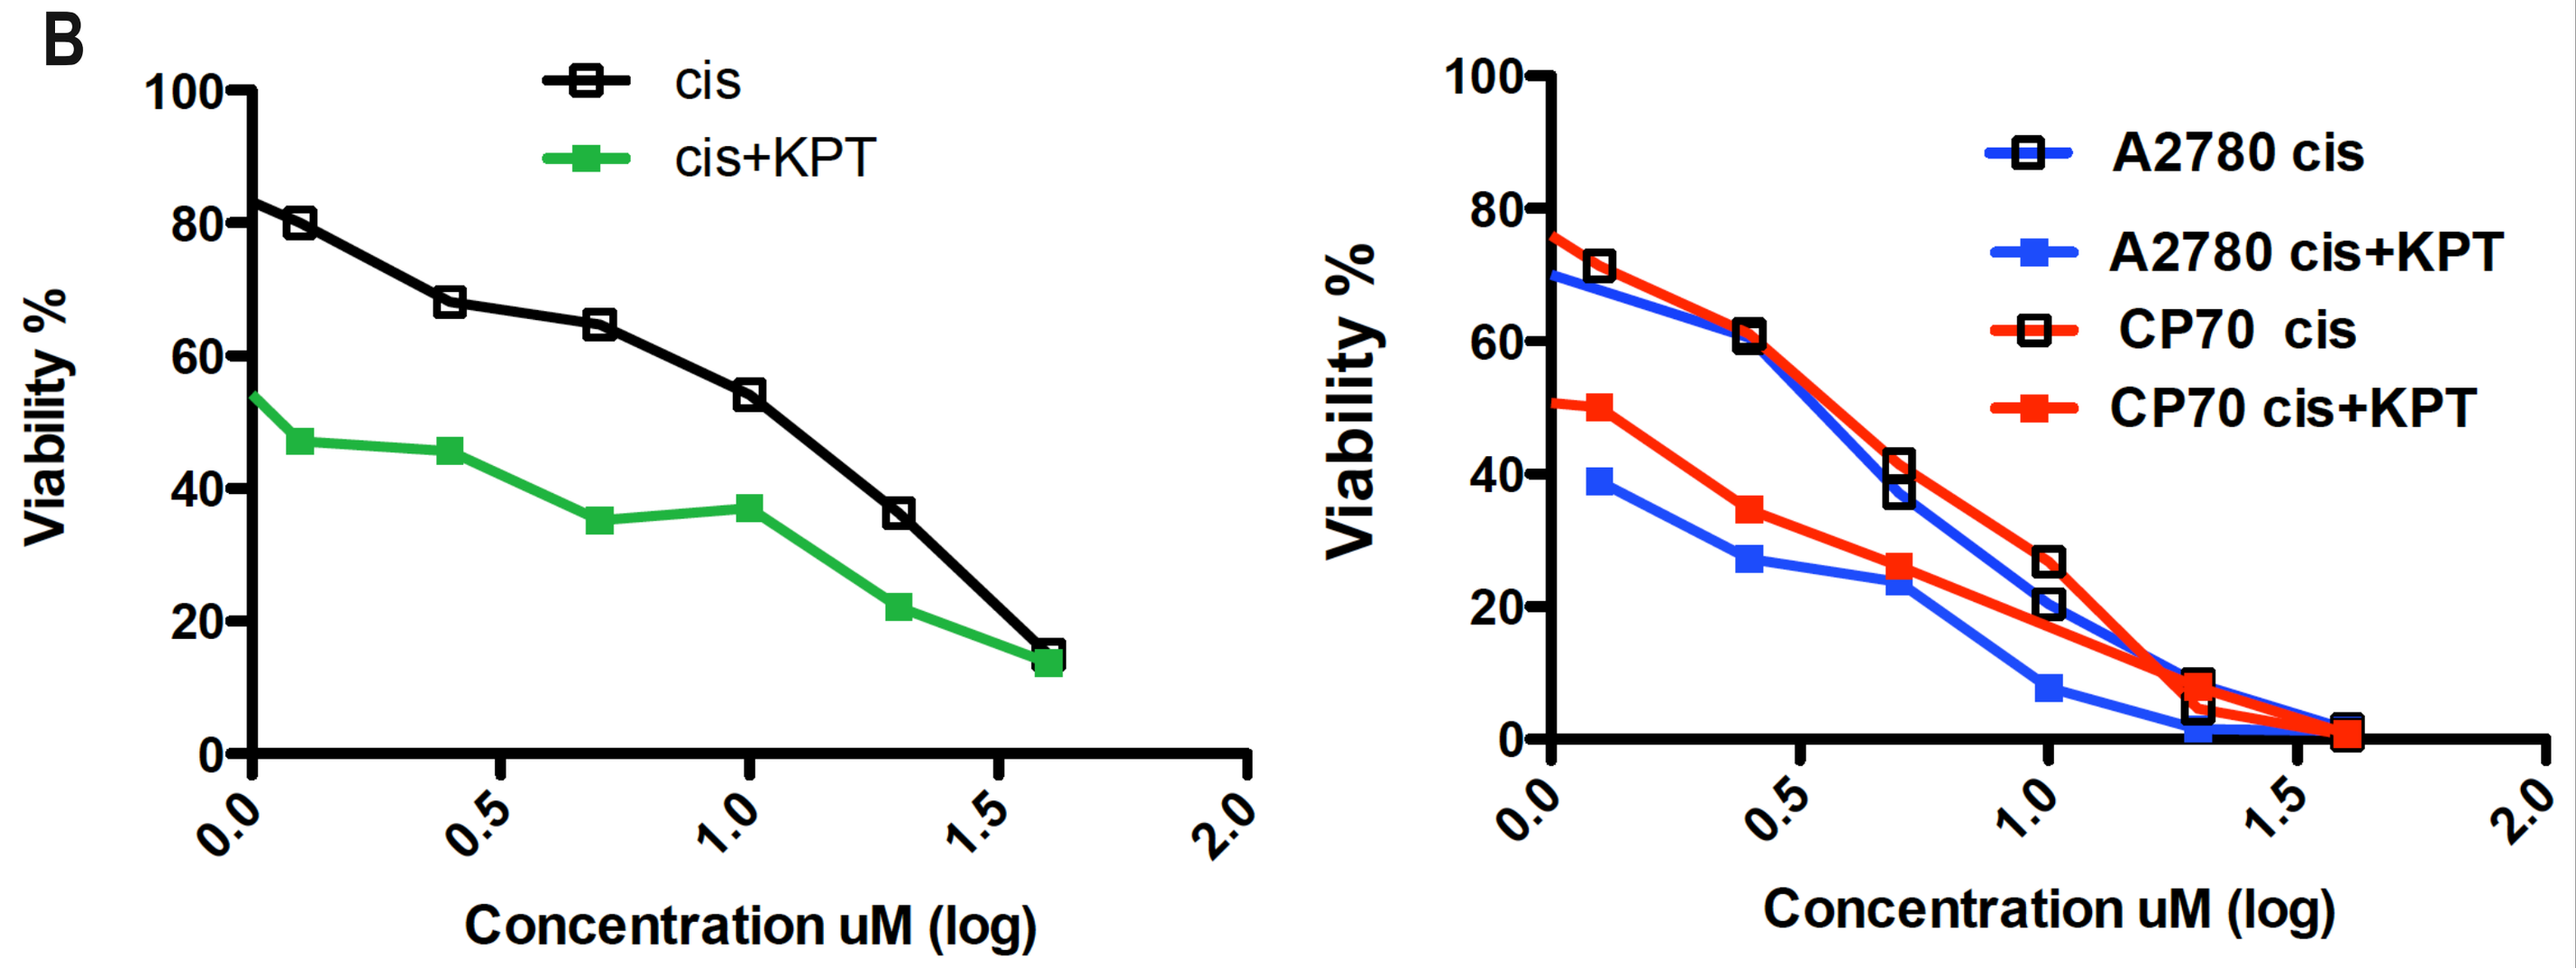
cis
cis+KPT
Viability %
Concentration uM (log)
A2780 cis
A2780 cis+KPT
CP70 cis
CP70 cis+KPT
Viability %
Concentration uM (log)

B) 25 Patient-derived OvCA cell lines (PDOvCA), from both platinum sensitive and resistant tumors, were treated with 500 nM KPT-185 and increasing cisplatin doses. KPT-185 treatment decreased cisplatin IC_{50} levels 5-fold (8.9 μM vs. 1.9 μM, cisplatin vs. combination, left panel). Similar effects were observed with combination treatment using A2780 and CP70 cells, wherein cisplatin IC_{50} is reduced ~ 3.5 fold (A2780: 2.8 μM vs 0.8 μM, CP70: 3.4 μM vs 0.9 μM, right panel).

Phase 1 Study of Selinexor in Solid Tumors

Dose Escalation:
Relapsed Solid Tumors

All Comers
(including OvCa (N=2 @30 mg/m²))

Dose Expansion:
Ovarian Cancer (N=5)

35 mg/m²

Clinicaltrials.gov: NCT01607905

Patient Demographics

Characteristics N=7	
Median Age (Range)	55 (33-75)
Median Regimens (Range)	5 (2-9)
Previously Treated with Platinum (%)	7 Patients (100%)
ECOG PS 0:1	3/4

Selinexor Decreases Tumor Growth and Increases Survival in Two Different Mouse Models: Intraperitoneal and PDX mice

A

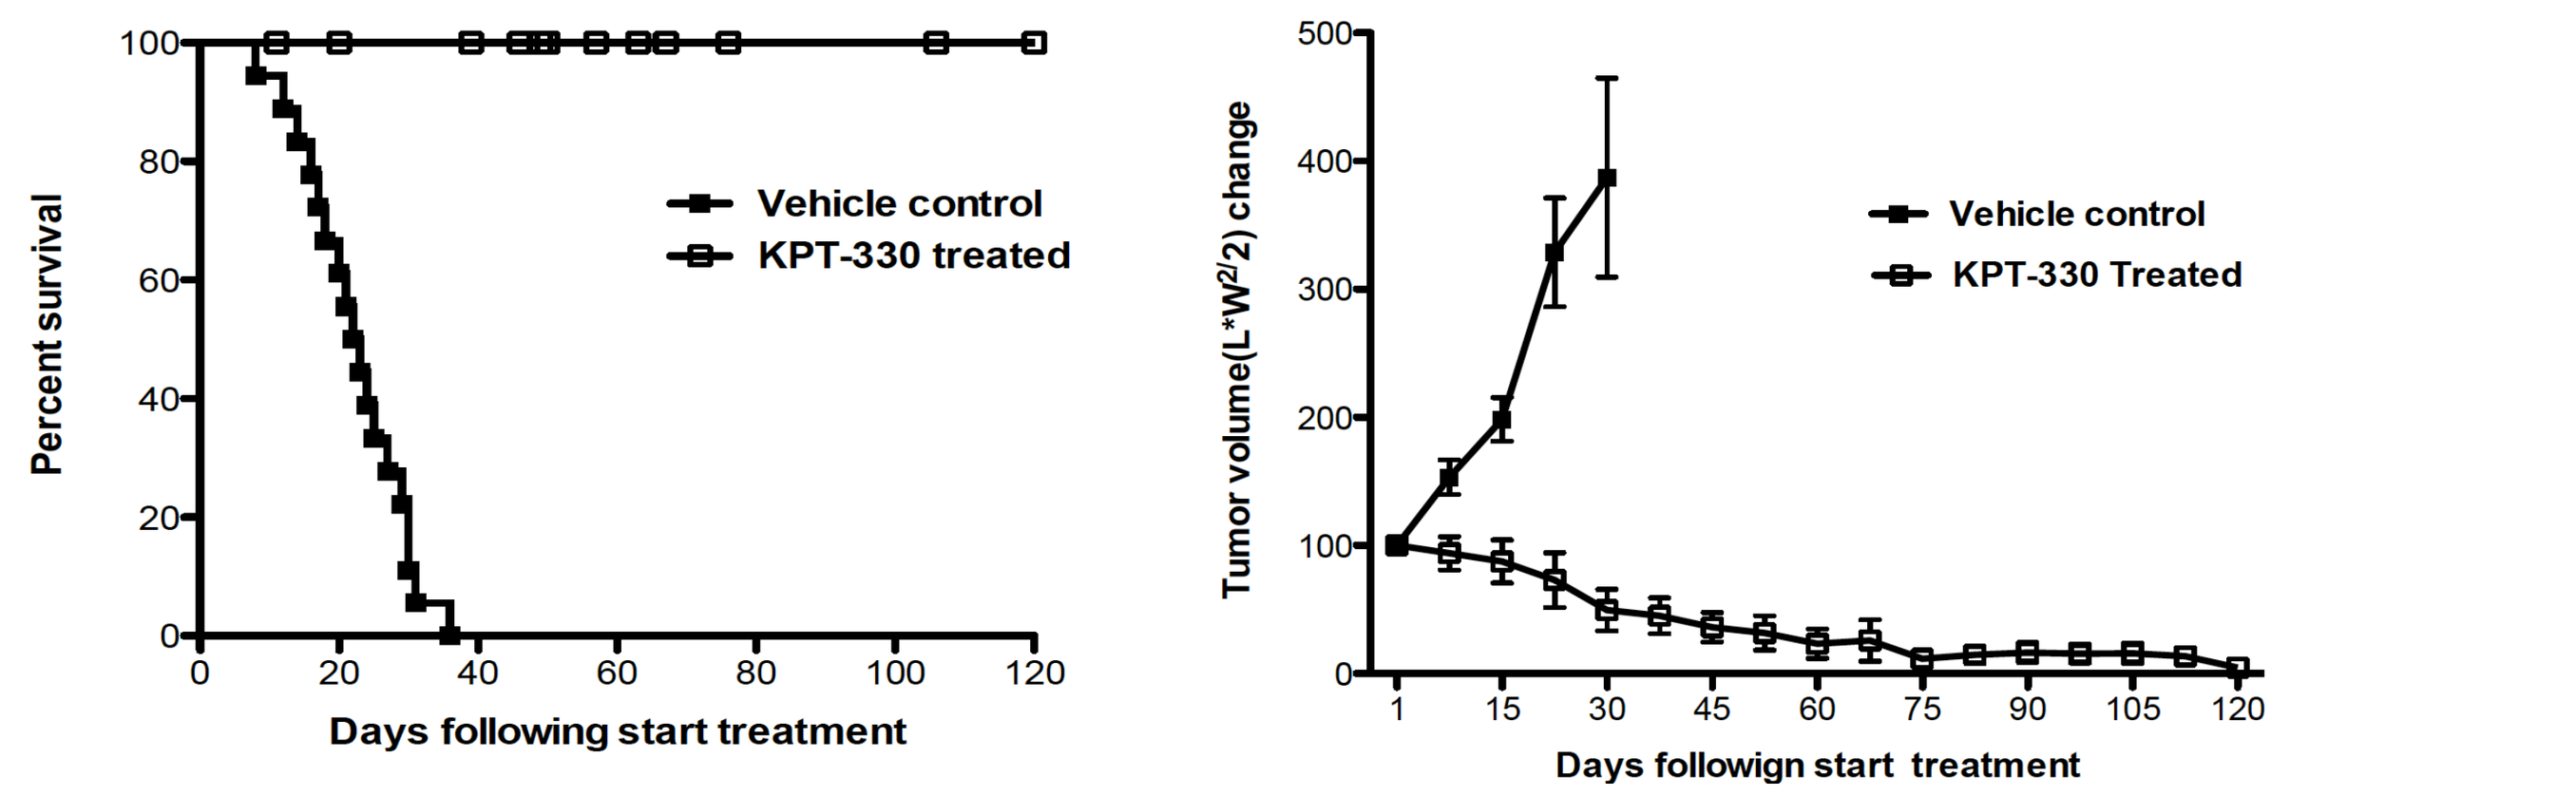
Percent survival
Vehicle control
KPT-330 treated
Days following start treatment
Tumor volume(L*W^2/2) change
Vehicle control
KPT-330 Treated
Days followign start treatment

A) Eight OvCa patient-derived tumors – 4 cisplatin resistant / 4 sensitive were implanted into the flanks of Rag1$^{-/-}$ mice. When tumor volume reached ~125 mm³, selinexor treatment at 10 mg/kg PO for 3x/wk started. No untreated mouse survived beyond 25 days while all selinexor treated mice had significantly improved survival when compared to controls (P<0.0001) and continued to survive beyond 147 days.

B

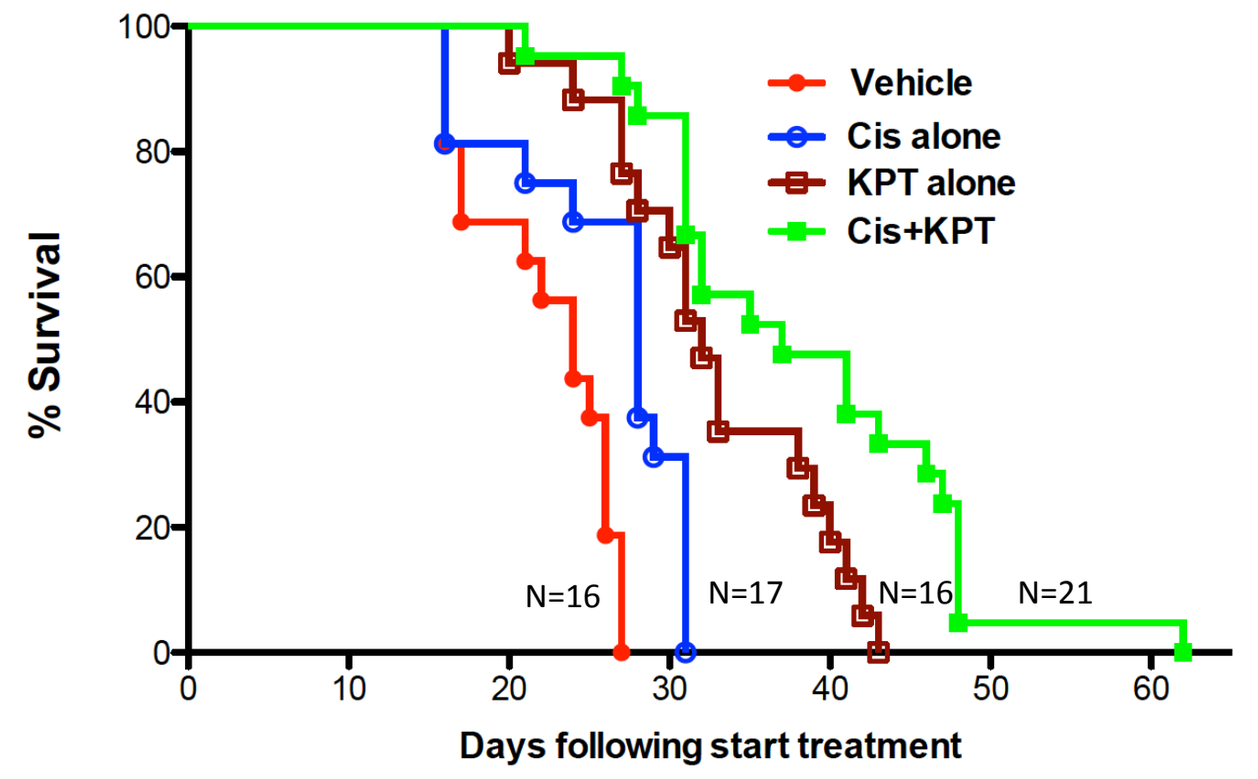
Vehicle
Cis alone
KPT alone
Cis+KPT
% Survival
N=16
N=17
N=16
N=21
Days following start treatment

B) Luciferase-positive CP70 cells were i.p inoculated into nude mice. Selinexor treatment was initiated by oral gavage at a dose of 15 mg/kg, 2xwk. All control (red line) and cisplatin–treated (blue) mice died within 32 days after initial treatment. Treatment with selinexor (brown) prolonged OS when compared to vehicle or cisplatin groups (P<0.001). Mice treated with a combination of selinexor and cisplatin had the greatest OS benefit, nearly doubling OS when compared to cisplatin alone.

C

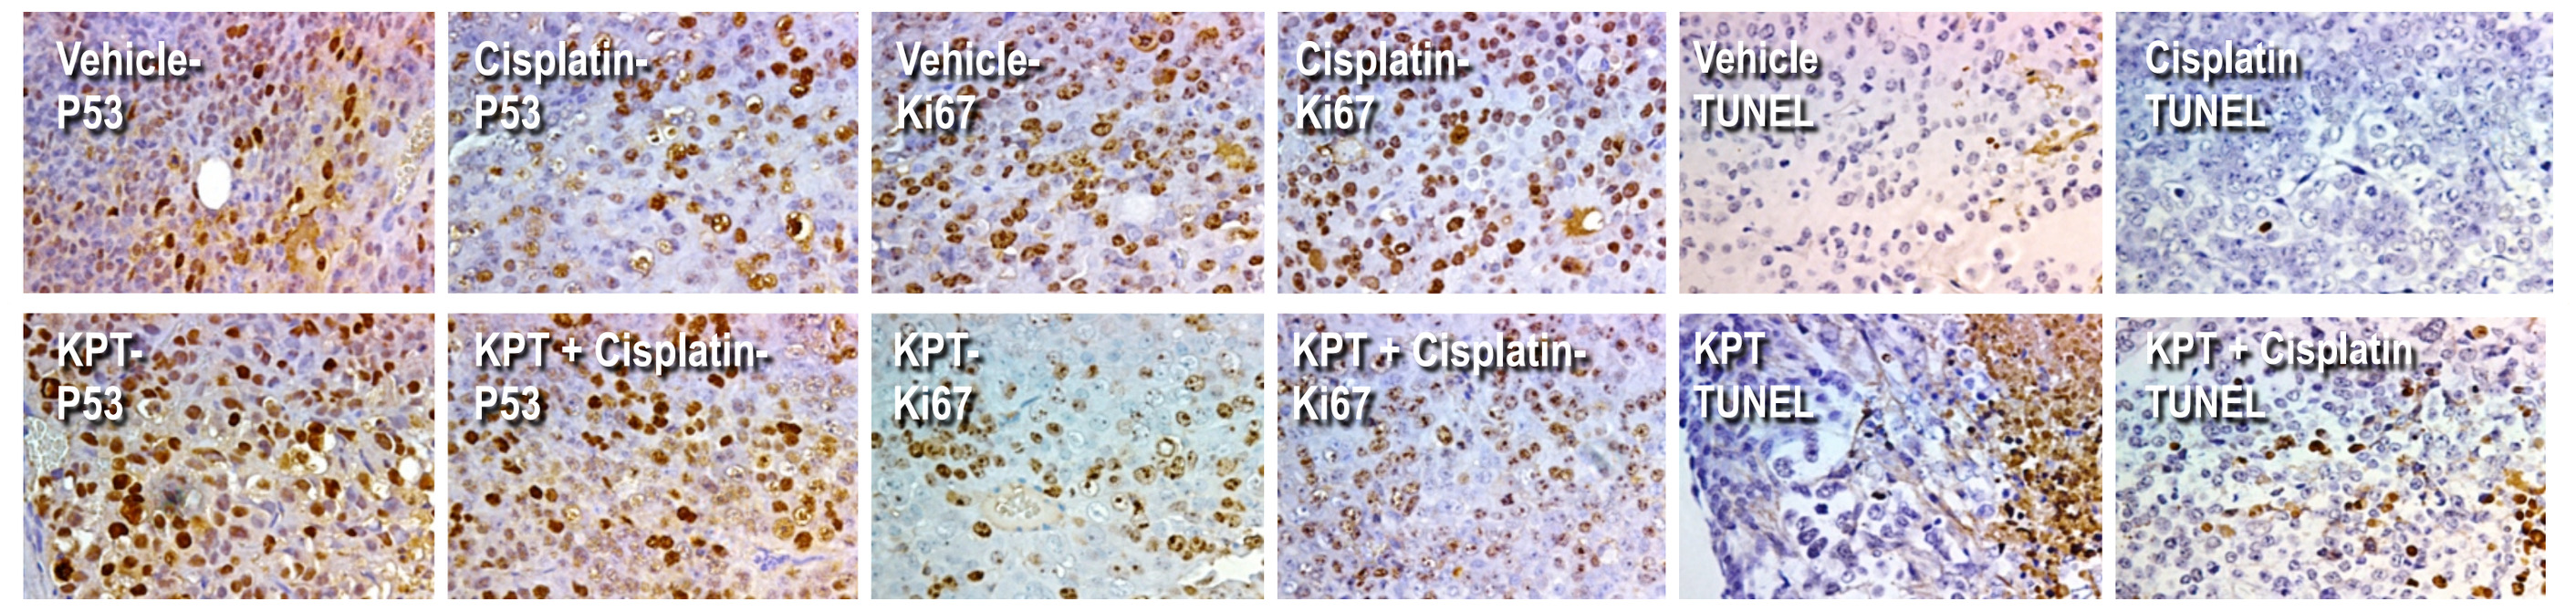
Vehicle-P53
Cisplatin-P53
Vehicle-Ki67
Cisplatin-Ki67
Vehicle TUNEL
Cisplatin TUNEL
KPT-P53
KPT+Cisplatin-P53
KPT-Ki67
KPT+Cisplatin-Ki67
KPT TUNEL
KPT+Cisplatin TUNEL

C) Tumor cells from selinexor treated CP70 tumor-bearing mice demonstrate increased TUNEL+ staining (lower panel) indicative of DNA breakage seen in apoptotic cells. Low levels of Ki67 and marked up-regulation of p53 *in situ* were also evident demonstrating that selinexor induces nuclear localization of these proteins.

Patient Time to Progression on Selinexor vs. Last Prior Therapy

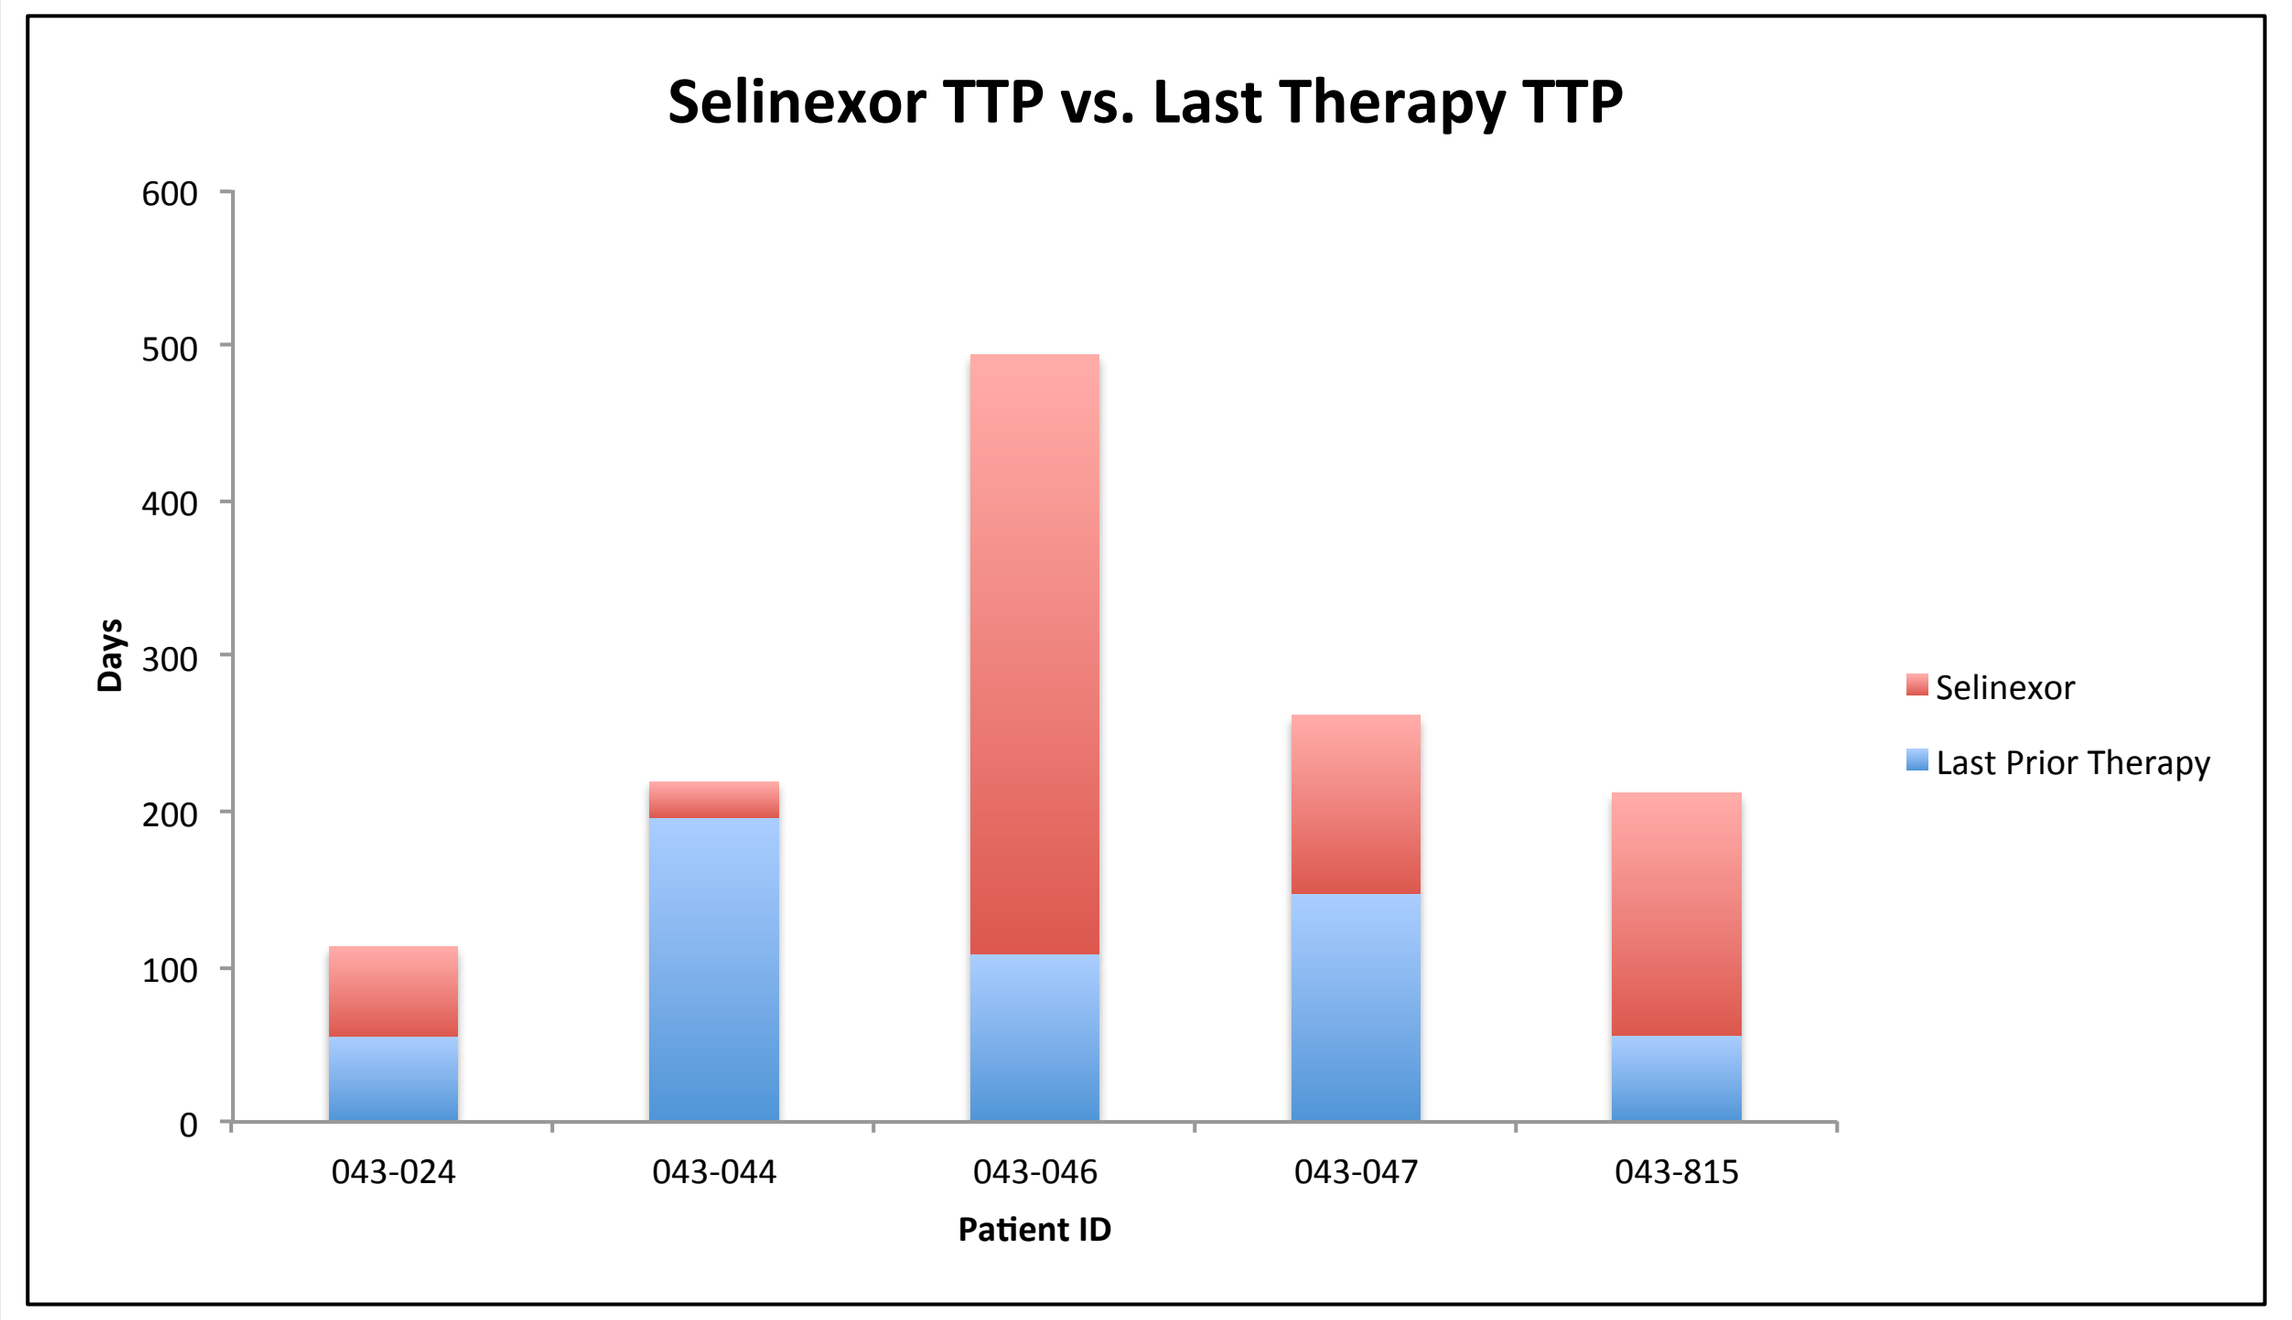
Selinexor TTP vs. Last Therapy TTP
Days
Selinexor
Last Prior Therapy
043-024
043-044
043-046
043-047
043-815
Patient ID

For patients that were evaluable, the time to progression (TTP) on selinexor was calculated (in days) versus the TTP of the last therapy prior to selinexor.

Selinexor Related Adverse Events Occurring at Least Once in ≥ 2 Patients (N=7)

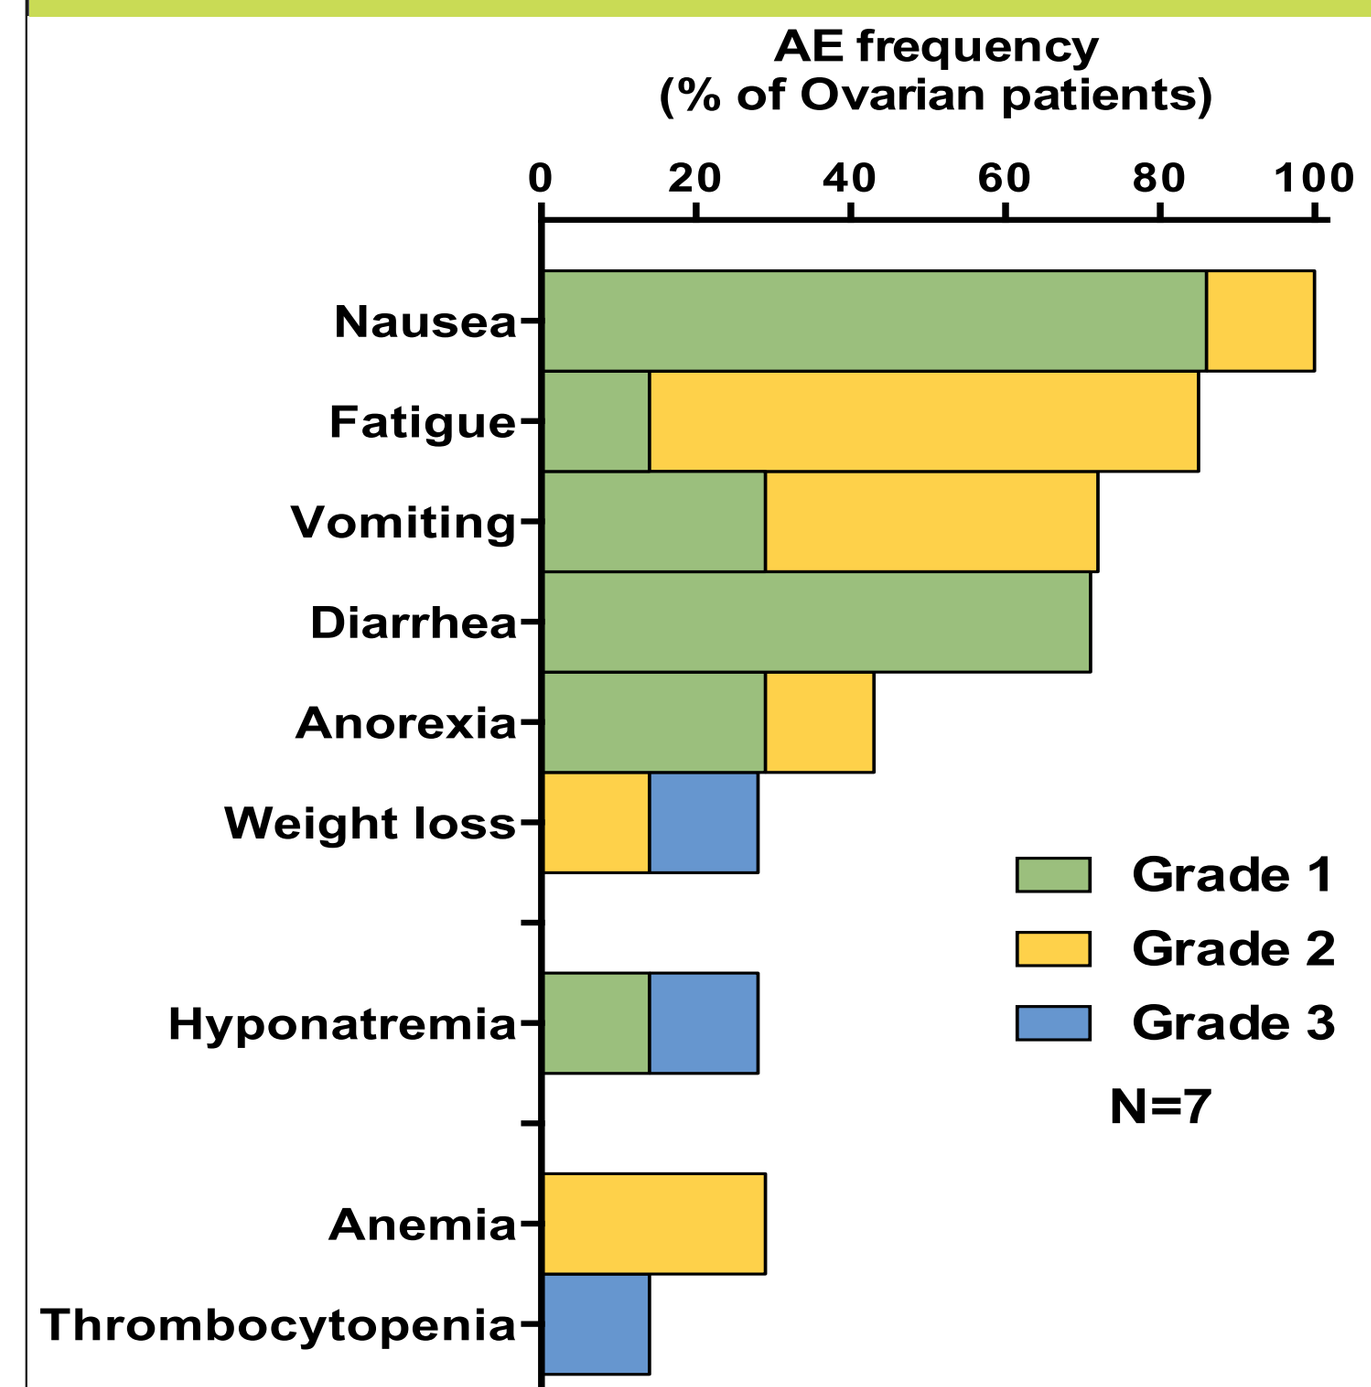
AE frequency
(% of Ovarian patients)
Nausea
Fatigue
Vomiting
Diarrhea
Anorexia
Weight loss
Hyponatremia
Anemia
Thrombocytopenia
Grade 1
Grade 2
Grade 3
N=7

Selinexor related AEs occurring in ≥ 2 patients 30 mg/m² (2) or 35 mg/m² (5)

Selinexor was associated primarily with Grade 1/2 toxicities, with most adverse events gastrointestinal (GI) in nature. Grade 1/2 nausea (100%), fatigue (85%), vomiting (72%), diarrhea (71%), anorexia (43%), anemia (29%), weight loss (14%), and hyponatremia (14%) were the most common adverse events. Dose relationships were unclear, and supportive care generally diminished or eliminated many of these toxicities. Cumulative toxicities were rare, and no major organ dysfunction was noted.

Patient 043-815 Case Report

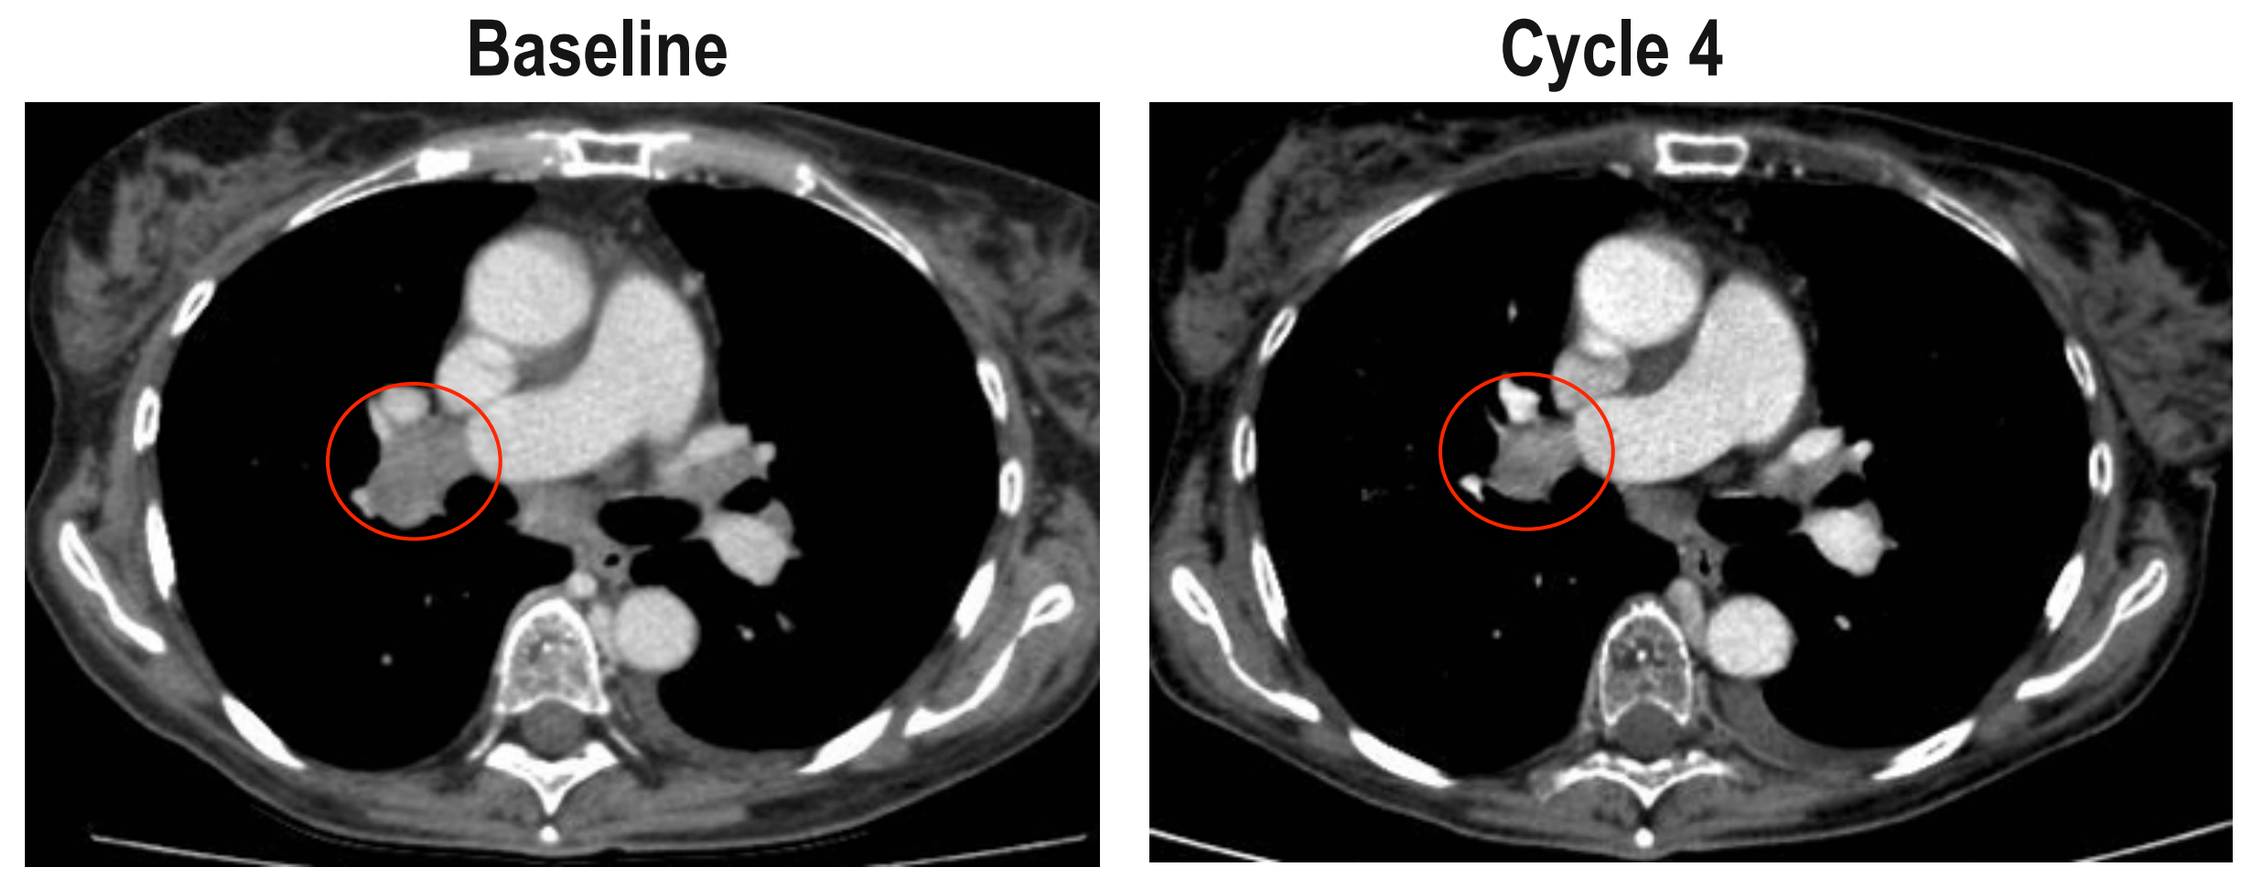
Baseline
Cycle 4

Patient Profile: 60 year old female with OvCa diagnosed in February 2000. She was treated with 8 prior regimens including: 1) Carbo/Taxol x 2; 2) Intraperitoneal Carbo x2 with cyclophosphamide; 3) Carbo/Docetaxel, Veliparib, Paclitaxel x2. The patient remained on single agent oral selinexor for 156 days before developing probable clinical progression with small bowel obstruction. Her CA-125 decreased from 540 at baseline to 240 U/mL at Cycle 4.

Selinexor Activity in OvCa Patients (16 September 2014)

Patient #	Prior Regimens	Best Response	Days on Study
043-815	Carbo/Taxol x2; IP Carbo x 2; Dox/Carbo; Veliparib; Paclitaxel	**PR**	156
043-046	Carbo/Taxotere; Doxil; Cis/Gemcitabine	**SD**	386
043-047	Carbo/Taxol x3; Trebananib/Temsiro.;Ganitumab/MEK inh.	**SD**	115
043-024	Carbo/Taxol; Topotecan; Linsitinib/Taxol; Temsiro./Trebananib; PI3K inh./IGF mAb	**PD**	58
043-044	Carbo/Taxol; Carbo/Taxol/PI3K inh.	**PD**	23
043-031	Carbo/Taxotere/Cisplatin/Gemzar/N3699G; Carbo/Gem/ Bevacizumab	**NE/WC**	9
043-023	Carbo/Taxol x2; Carbo/Gem/Bevaci.; Vintafolide/Dox; Paclitaxel/Cisplatin	**NE/WC**	22

PR = Partial Response, SD = Stable Disease, PD = Progressive Disease NE=Not evaluable WC= Withdrew consent

Conclusions

- In two different mouse models of OvCa, selinexor displayed significant efficacy and prolonged survival, including additivity/synergy with cisplatin. Histopathological analyses on tumor samples indicated that selinexor treatment reduced proliferation, induced nuclear localization of TSPs, and increased apoptosis
- As part of an all-comers Phase I solid tumor study, patients with platinum resistant/refractory OvCa, selinexor given orally 2-3 times per week is associated with rare Grade 3 events, with manageable Grade 1/2 GI toxicities (nausea, fatigue, vomiting, diarrhea) reduced with supportive care
- In patients with platinum resistant/refractory OvCa, single agent oral selinexor induced durable disease stabilization and tumor size reduction at tolerable doses
- Further evaluation of selinexor for the treatment of advanced gynaecologic malignancies as a single agent (NCT02025985) is currently on-going and combination studies are planned