Quantification of Exportin-1 (XPO1) Occupancy by Selective Inhibitor of Nuclear Export / SINE™ Compounds 064

Marsha Crochiere, Boris Klebanov, Erkan Baloglu, Ori Kalid, Trinayan Kashyap, William Senapedis, Diego del Alamo, Sharon Tamir, Dilara McCauley, Robert Carlson, Michael Kauffman, Sharon Shacham, and Yosef Landesman

Karyopharm Therapeutics, Newton, MA, USA

Abstract

SINE™ compounds are a family of small-molecules that selectively inhibit nuclear export through covalent binding to Cysteine 528 (Cys528) in the cargo binding pocket of Exportin 1 (XPO1/CRM1). Such binding leads to forced nuclear retention and activation of major tumor suppressor proteins (TSPs) such as p53, FOXO, pRB and IkB and results in specific cancer cell death. Selinexor, an orally available SINE™ compound currently in human phase I and II clinical trials for advanced cancers (Clinicaltrials.gov), forms a slowly reversible covalent bond with XPO1. Oral selinexor demonstrates maximal pharmacokinetic exposure at 2-4 hours and increases in pharmacodynamic markers of XPO1 inhibition at 4 hours that last for 48 hours. The goal of this study was to develop a binding assay that would enable quantification of XPO1 occupancy in patients' PBMC and tumor cells following oral administration of selinexor.

Methods

To measure the binding of SINE™ compounds to XPO1, biotinylated leptomycin B (LMB) was utilized. Biotinylated LMB binds covalently and irreversibly to Cys528 in the cargo-binding site of free XPO1 and its activity was confirmed to be similar to that of unmodified LMB in cytotoxicity assays. To measure SINE™ compound binding to XPO1 in vitro, cancer cell lines and PBMCs from normal human donors were treated with SINE™ compounds prior to treatment with biotinylated LMB. In in-vivo studies, mice were given oral selinexor 0 - 10 mg/kg, then PBMCs were isolated and treated with biotinylated LMB. After incubation with biotinylated LMB, cells were harvested, lysed, and protein lysates were subjected to pull down experiments with streptavidin-conjugated beads followed by immunoanalysis of XPO1 and actin on Peggy Sue (Protein Simple) at SBH Sciences.

Results

To evaluate selinexor-XPO1 binding kinetics in vitro, MM.1s cells were treated with serial dilutions (0 - 10 µM) of SINE™ compounds and unbound XPO1 was pulled down from cell lysates treated with biotinylated LMB. Immunoanalysis showed that 50% of XPO1 occupancy with selinexor was achieved at 0.07 µM (i.e., EC_{50} 70 nM). Selinexor-XPO1 occupancy experiments using human PBMCs isolated from donor whole blood showed 50% XPO1 occupancy at 6.5 – 9.87 nM. XPO1 occupancy in vivo was measured in mice following oral administration of selinexor at 0.75 - 10 mg/kg for 4 hours resulting in XPO1 occupancy in PBMCs of 1.84 mg/kg. To measure the in vivo pharmacokinetic occupancy of selinexor over time, mice were treated with a single dose of selinexor ranging from 1.5 to 10 mg/kg and competition was evaluated in PBMCs isolated from these mice at time points ranging from 4 – 96 hours. The results showed a dose-dependent competition of XPO1 binding and significant XPO1 occupancy in vivo up to 72 hours post-treatment.

Conclusions

This new XPO1 occupancy assay can be used to evaluate drug exposure following treatment with oral selinexor. XPO1 occupancy will be determined in the ongoing clinical studies and its utility to predict response to selinexor will be evaluated.

Selinexor Mechanism of Action

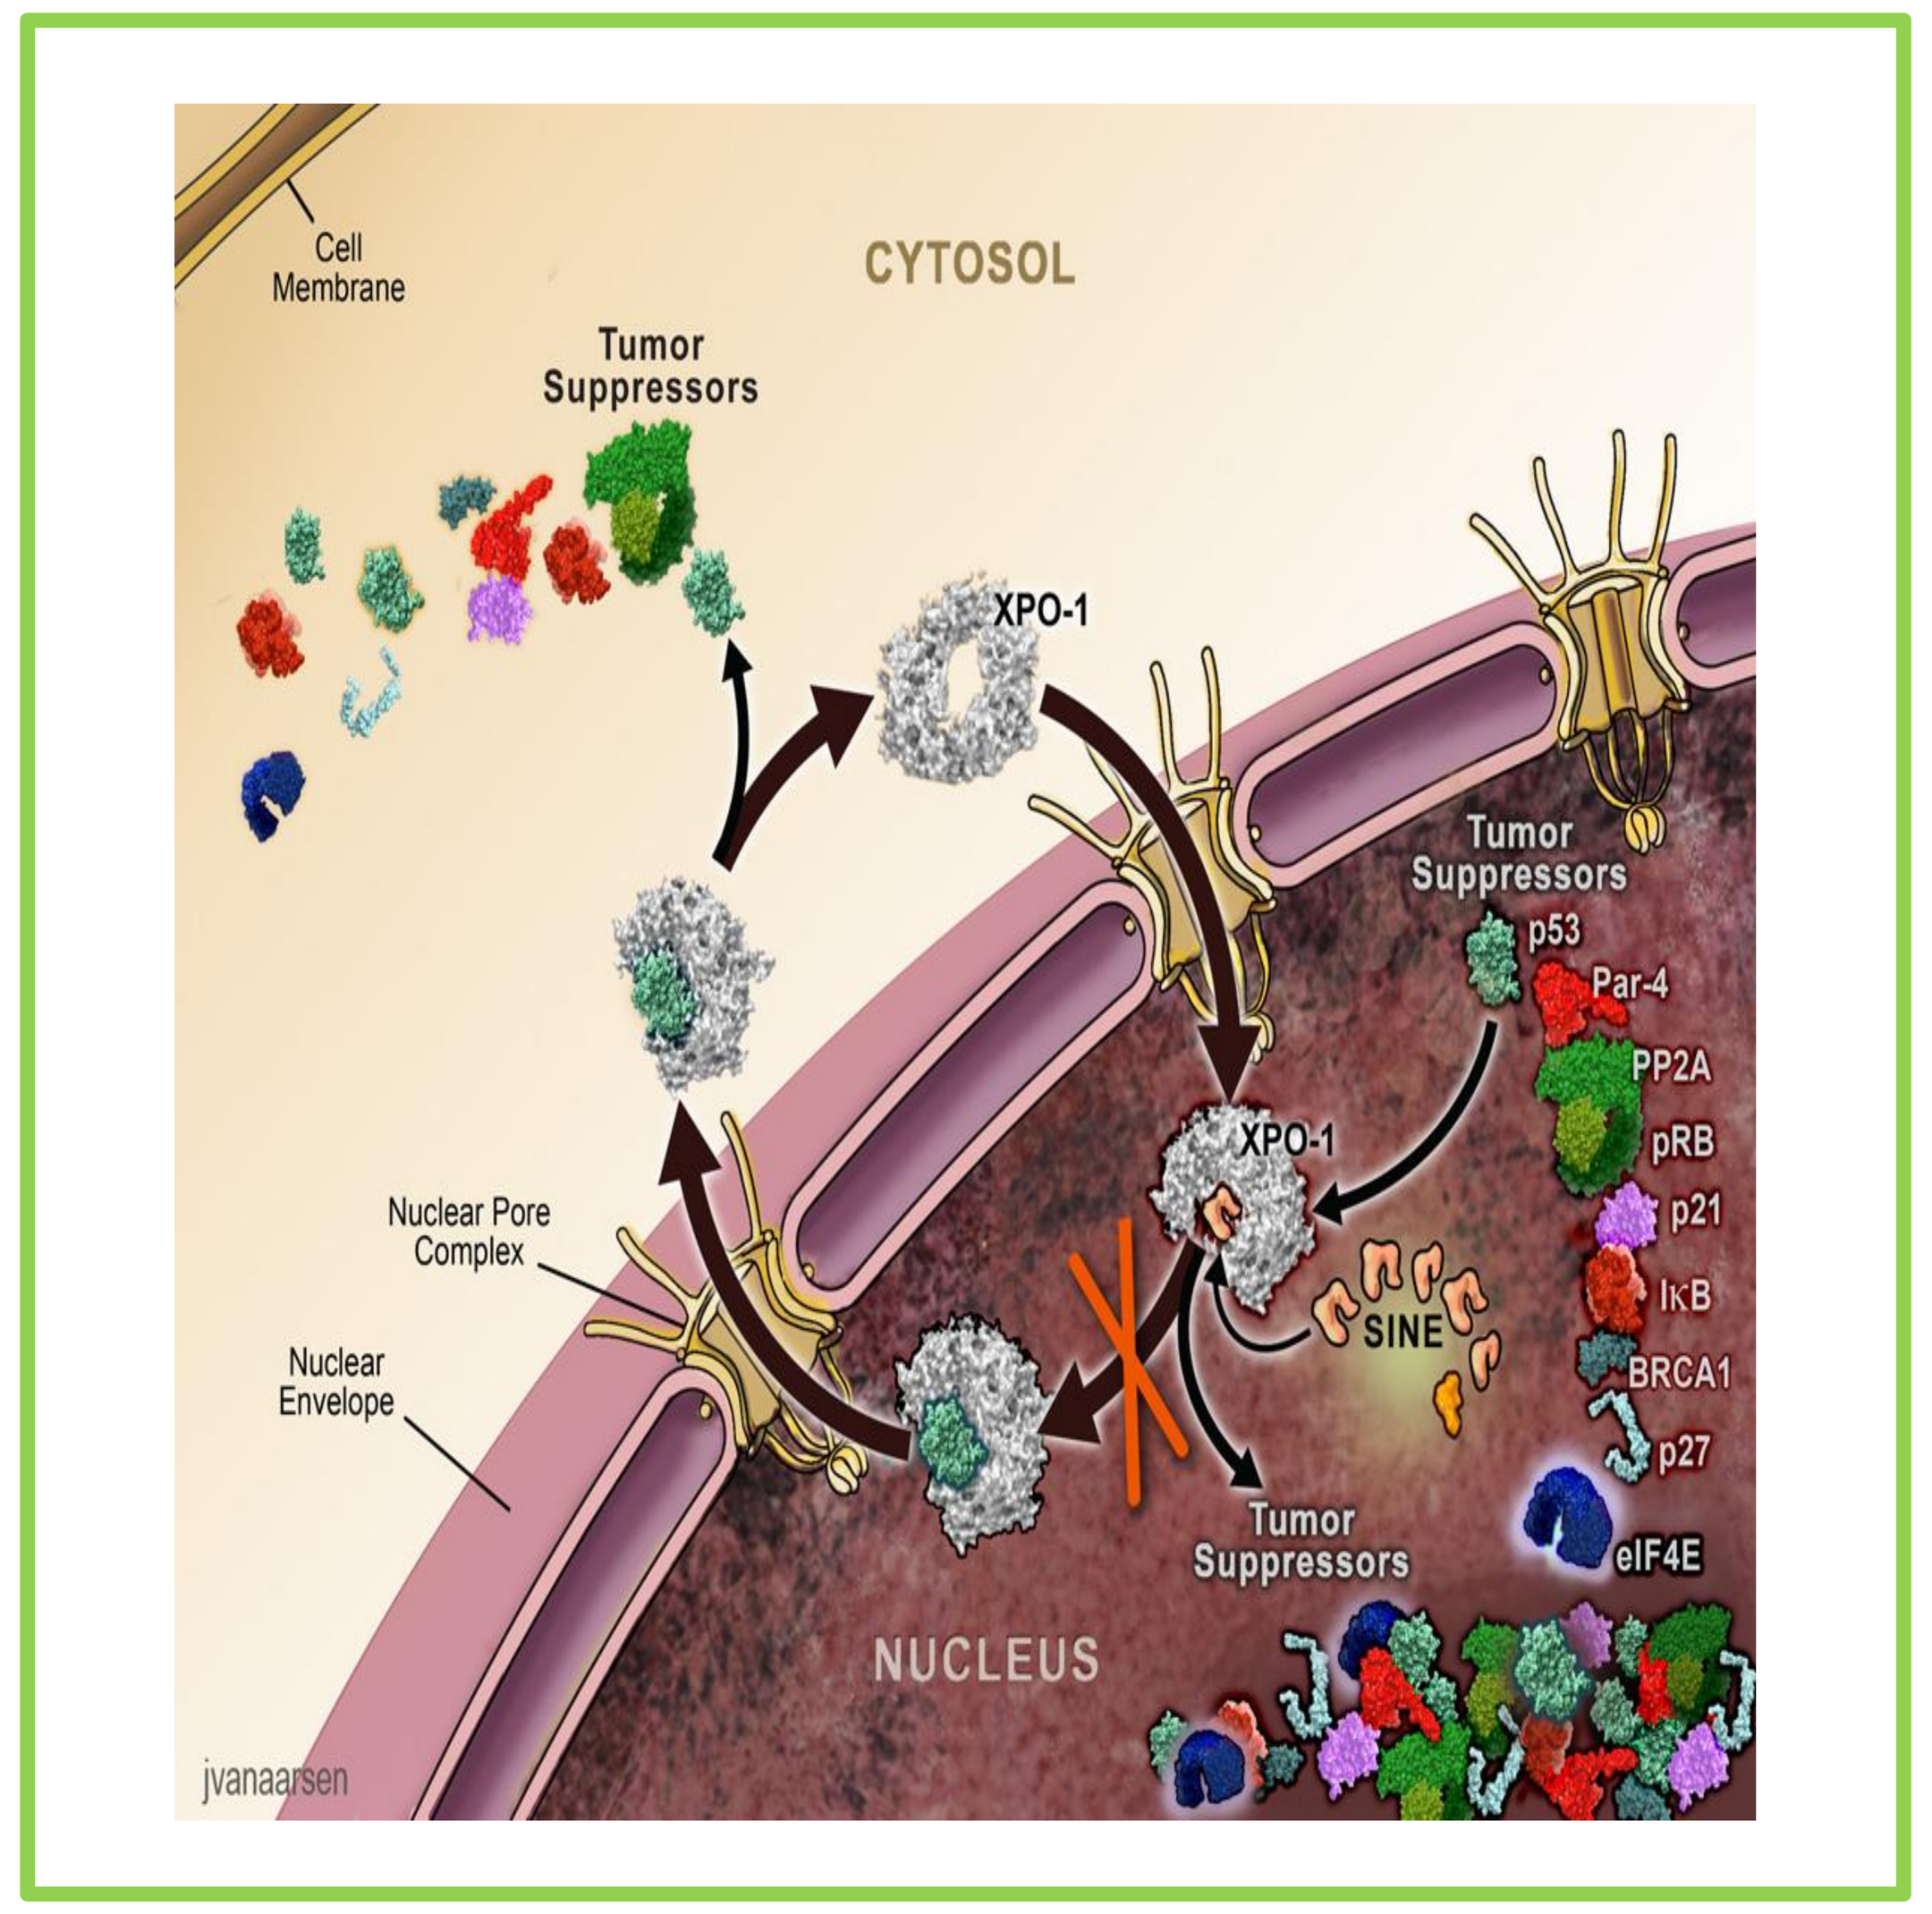
CYTOSOL
Cell Membrane
Tumor Suppressors
XPO-1
Tumor Suppressors
p53
Par-4
PP2A
pRB
p21
IkB
BRCA1
p27
eIF4E
XPO-1
SINE
Nuclear Pore Complex
Nuclear Envelope
Tumor Suppressors
NUCLEUS
jvanaarsen

Exportin 1 (XPO1) is the main nuclear export protein for tumor suppressor proteins. Selinexor (and other SINE™ compounds) bind to XPO1 and prevent it from leaving the nucleus leading to nuclear retention and activation of tumor suppressors and tumor cell apoptosis.

Maximum Selinexor Plasma Concentration (C_{max}) Occurs ~ 3 Hours Post-Dose

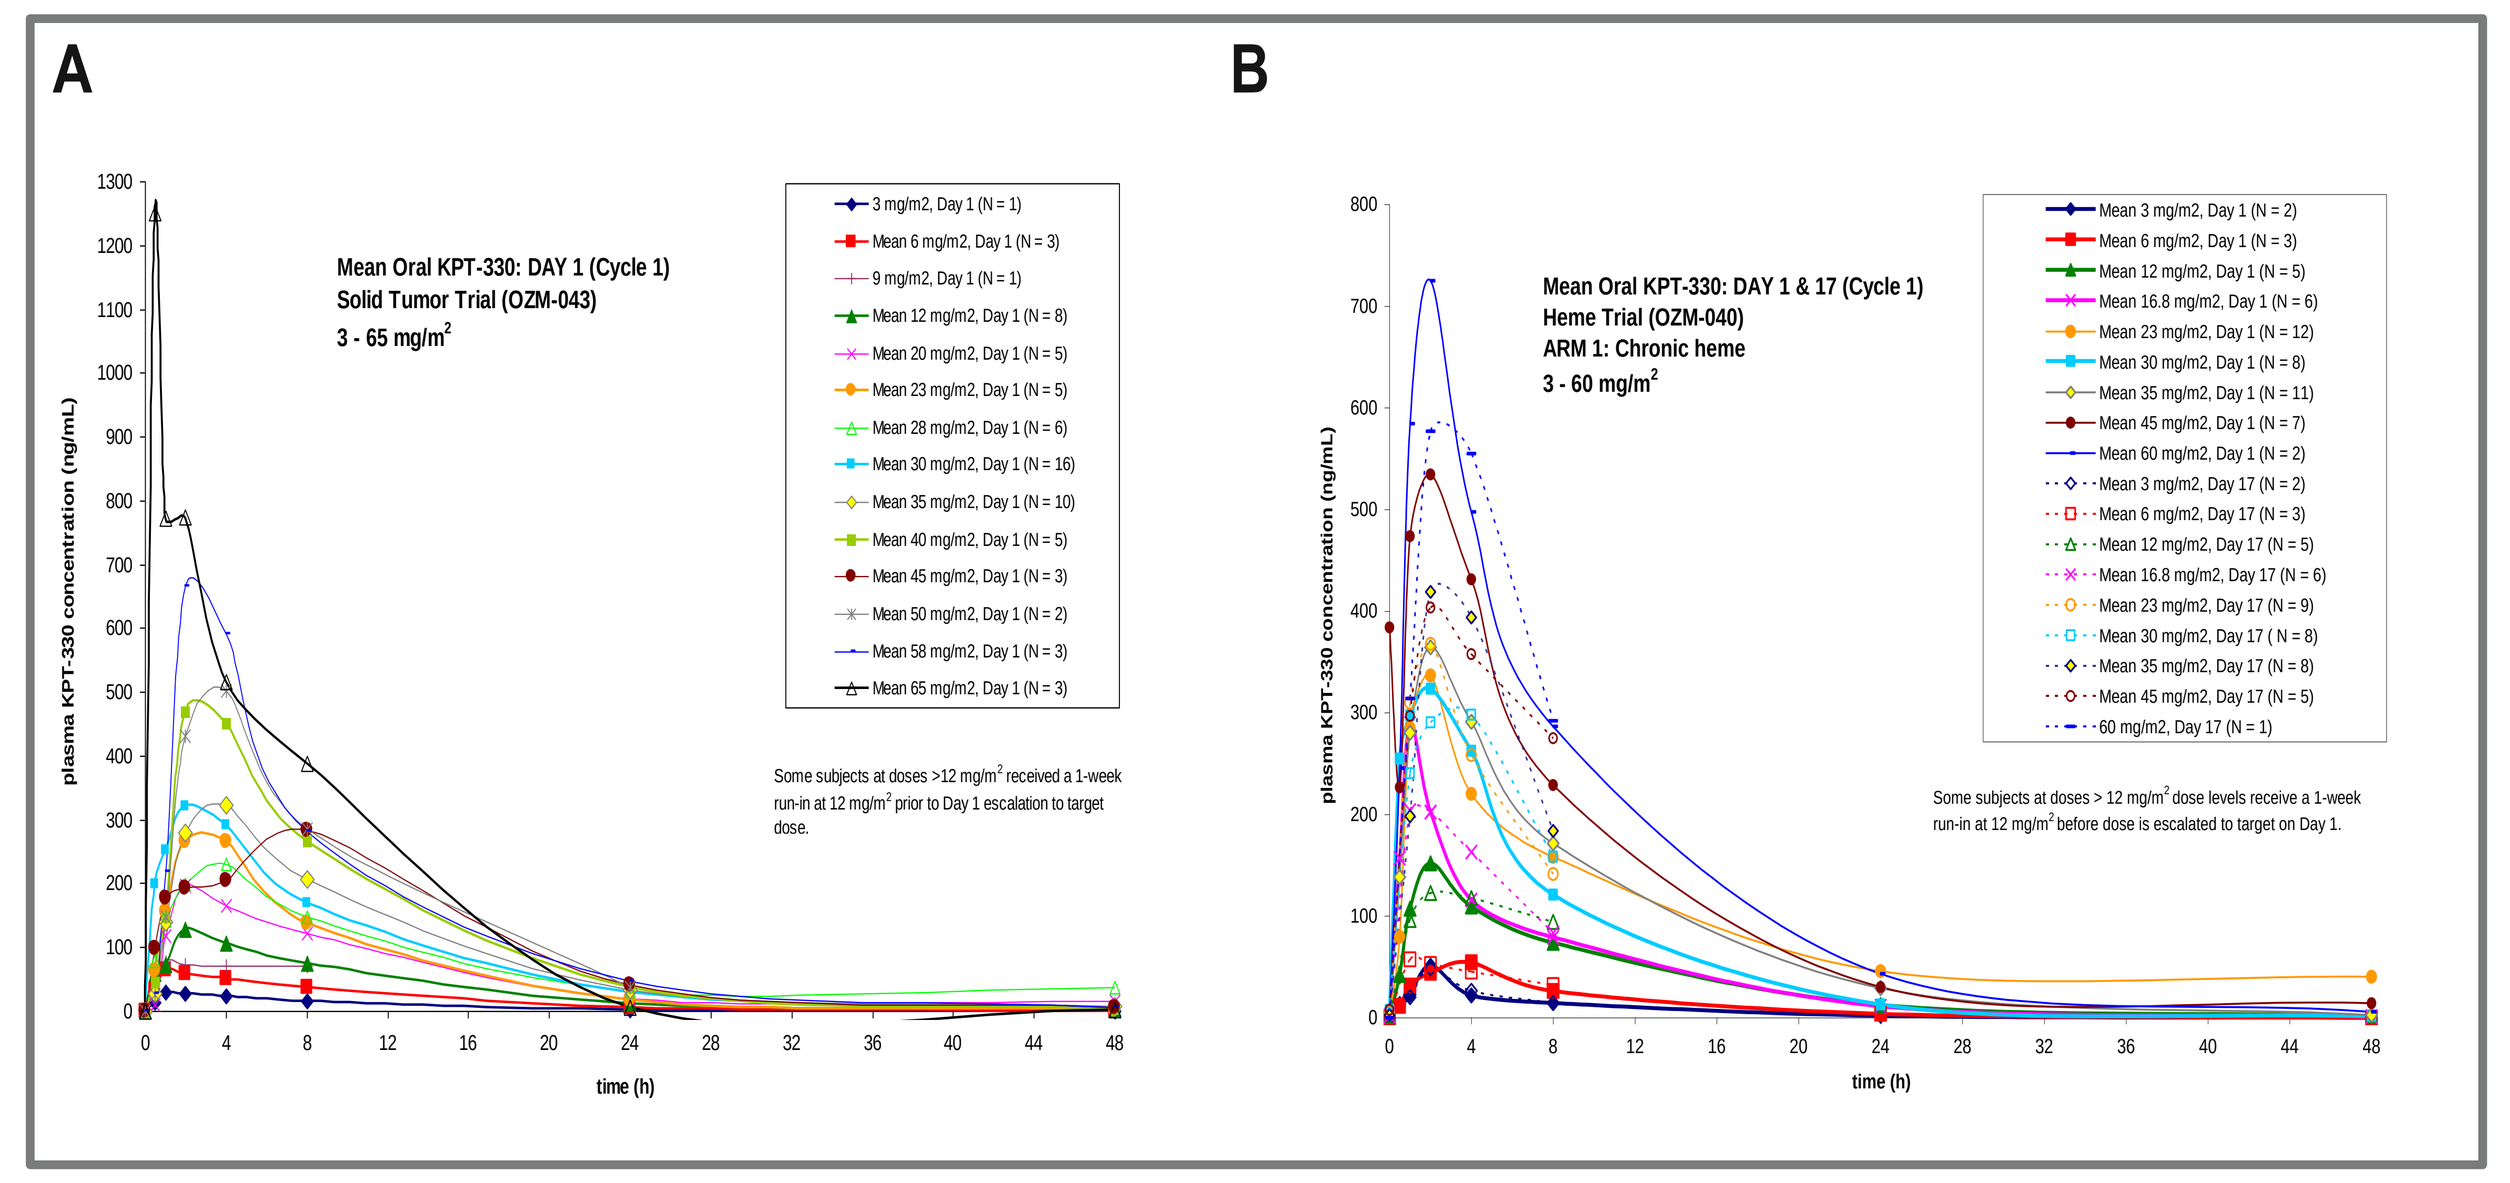
A
Mean Oral KPT-330: DAY 1 (Cycle 1)
Solid Tumor Trial (OZM-043)
3 - 65 mg/m²
plasma KPT-330 concentration (ng/mL)
time (h)
B
Mean Oral KPT-330: DAY 1 & 17 (Cycle 1)
Heme Trial (OZM-040)
ARM 1: Chronic heme
3 - 60 mg/m²
plasma KPT-330 concentration (ng/mL)
time (h)

Representative selinexor clinical PK results from (A) solid tumor cancer Phase 1 (NCT) and (B) hematological cancer Phase 1 trial (NCT). For the data obtained from the trial collectively thus far, C_{max} and AUC were dose proportional across all cohort (3-85 mg/m²) and half-life (6.2 ± 1.8 hr, N=181) and T_{max} (3.0 ± 2.0 hr) were dose independent.

Inhibition of XPO1 Results in mRNA Induction of Specific Genes in Cancer Patient Leukocytes as Markers of Response to Selinexor

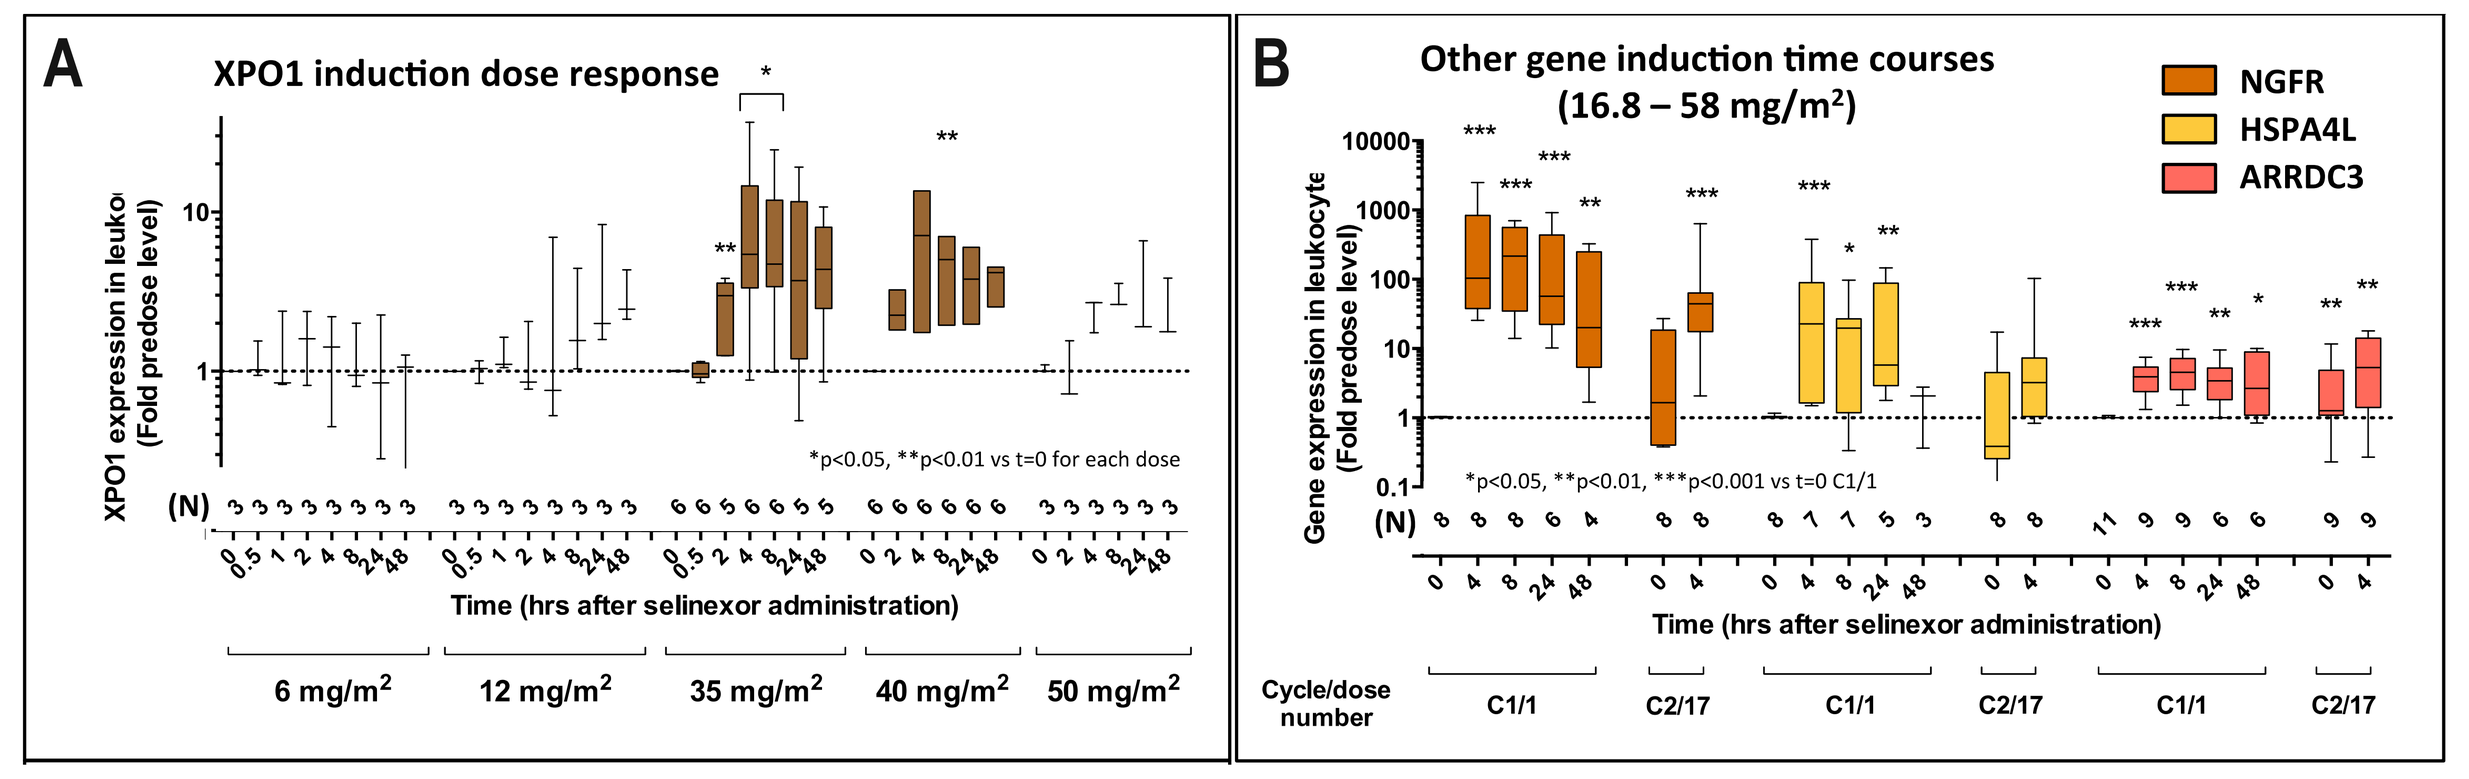
A
XPO1 induction dose response
XPO1 expression in leukocytes (Fold predose level)
*p<0.05, **p<0.01 vs t=0 for each dose
Time (hrs after selinexor administration)
6 mg/m²
12 mg/m²
35 mg/m²
40 mg/m²
50 mg/m²
B
Other gene induction time courses
(16.8 – 58 mg/m²)
NGFR
HSPA4L
ARRDC3
Gene expression in leukocyte (Fold predose level)
*p<0.05, **p<0.01, ***p<0.001 vs t=0 C1/1
Time (hrs after selinexor administration)
Cycle/dose number
C1/1
C2/17

Induction of gene expression in leukocytes from solid tumor patients following selinexor administration (QODX3/wk). Using qRT-PCR methods, expression of specific genes was measured in leukocytes taken from solid tumor patients at a variety of time points following selinexor administration over multiple cycles. Expression of (A) XPO1 and (B) NGFR, HSPA4L, and ARRDC3 expression in leukocytes as a function of time following the first dose received in Cycle 1 for 16.8-58 mg/m² selinexor, analyzed cumulatively. Mann Whitney test based statistical significance is listed on the graphs, relative to the mRNA levels prior to the first dose of the run-in week (at 12 mg/m² QODX3) or the first dose of Cycle 1.

Anti-Cancer Activity of Biotinylated-LMB (b-LMB) is as Potent as Unmodified LMB

A

Compound	Designation
Leptomycin B	LMB
Biotinylated LMB (KPT-9159)	b-LMB
KPT-330	selinexor
KPT-335	KPT-335
KPT-350	KPT-350
KPT-8602	KPT-8602

B

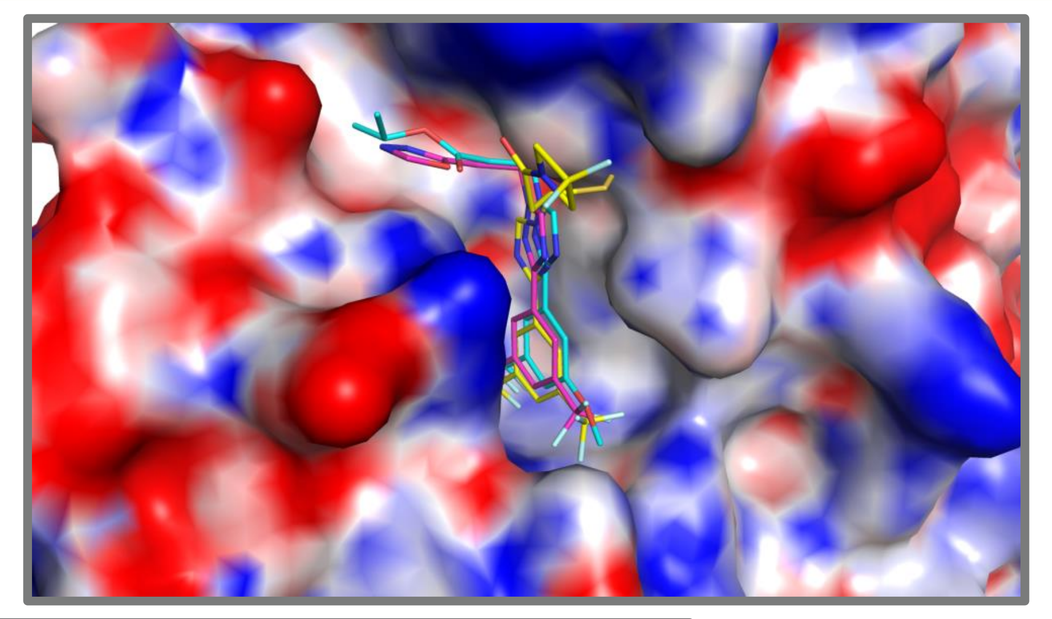

C

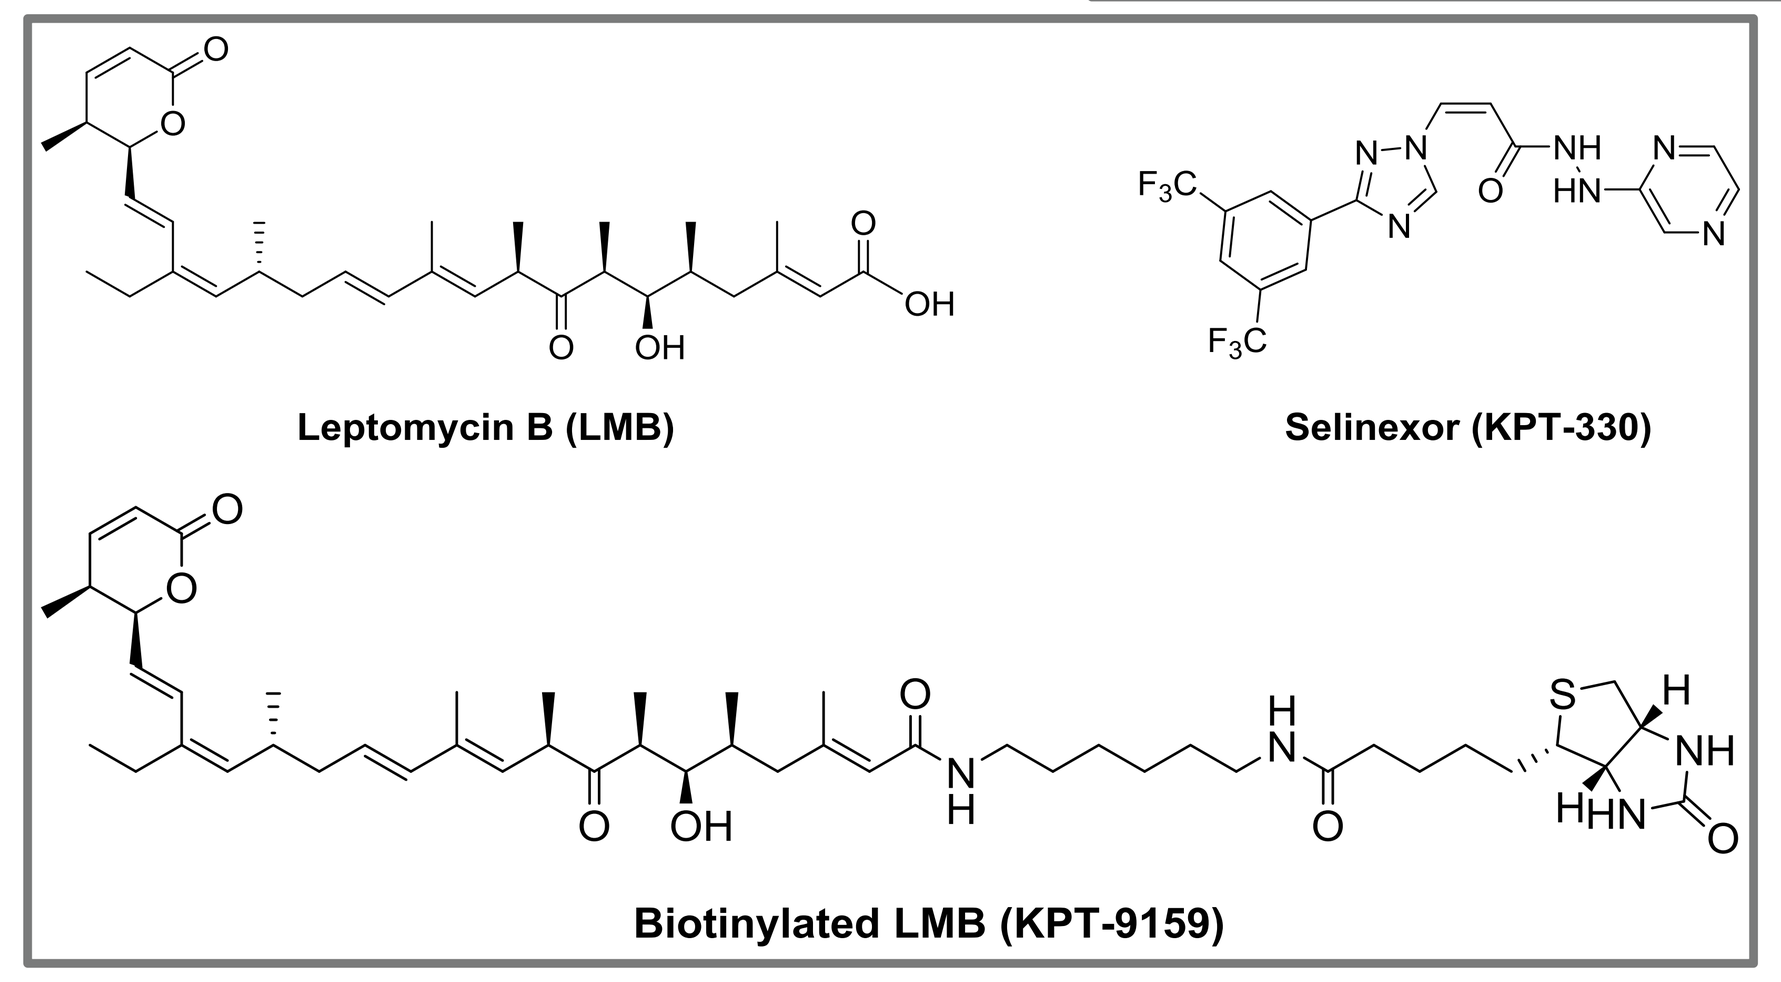
Leptomycin B (LMB)
Selinexor (KPT-330)
Biotinylated LMB (KPT-9159)

D

Compound	Rev-GFP (nM)
Selinexor	40
LMB	0.034
b-LMB	0.114

E

Cell Line	Selinexor IC_{50} (nM)	LMB IC_{50} (nM)	b-LMB IC_{50} (nM)
MM.1S	20	0.46	0.003
HCT-116	50	0.19	0.006
AML2	90	0.39	2.53
AML3	270	0.68	4.68

b-LMB is a novel tool compound to quantify the binding of SINE™ compounds to XPO1 protein. (A) Compounds and designations. (B) Space filling structure of XPO1 with SINE™ compounds bound XPO1 in the protein cargo binding (X-ray crystallography courtesy of Y. M. Chook). (C) Chemical structures of leptomycin B (LMB), selinexor (KPT-330), and b-LMB (KPT-9159). (D) b-LMB is a potent inhibitor of rev-GFP nuclear export. E. b-LMB is a potent inducer of cell death in several cell lines.

b-LMB Binds XPO1 in a Dose-Dependent Manner in MM.1S Cells

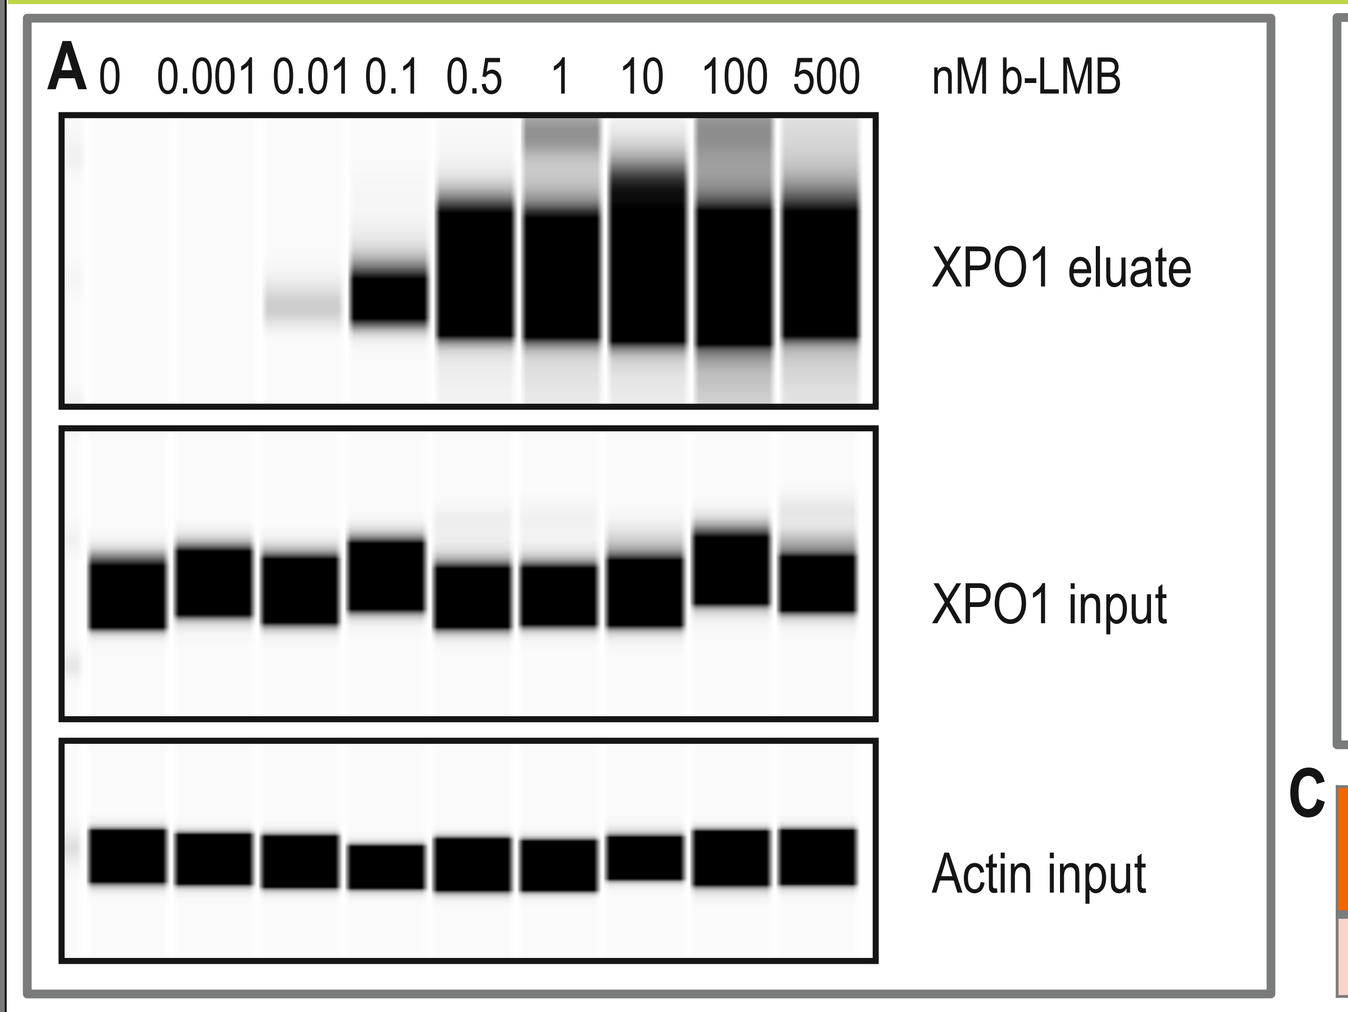
A 0 0.001 0.01 0.1 0.5 1 10 100 500 nM b-LMB
XPO1 eluate
XPO1 input
Actin input

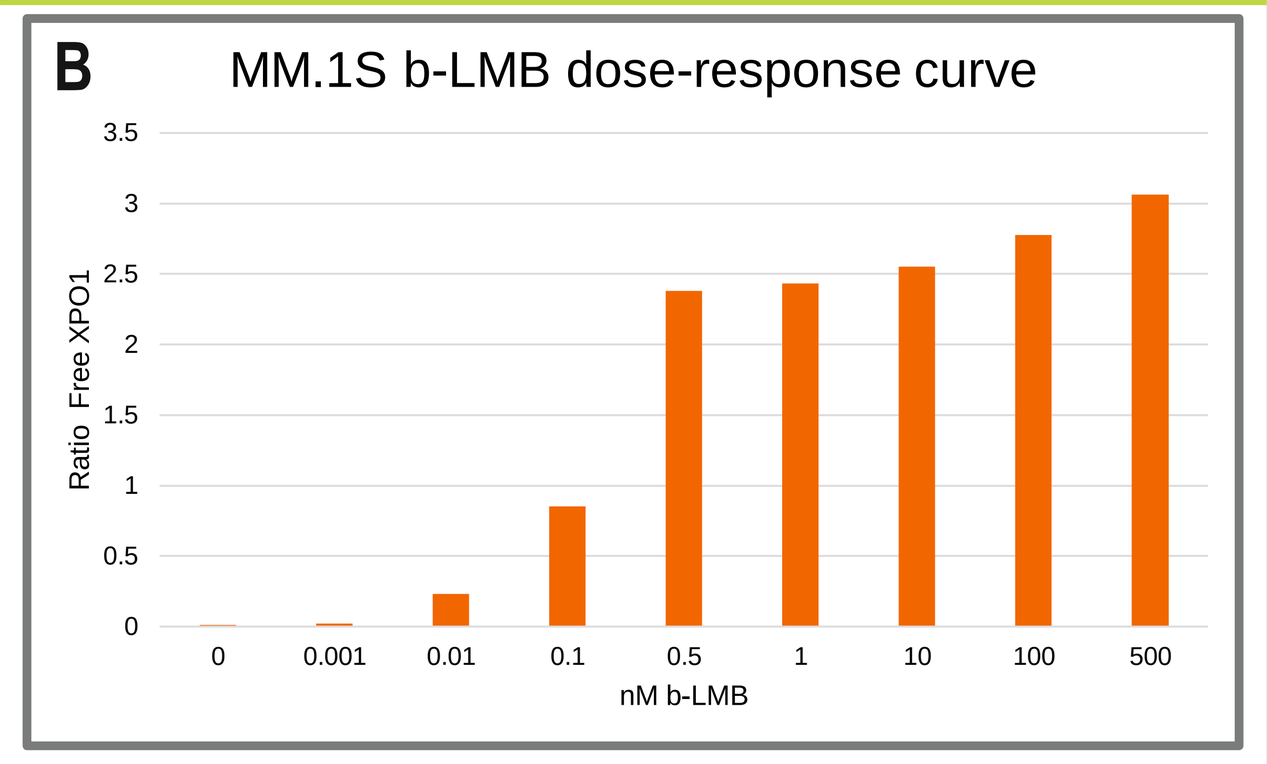
B
MM.1S b-LMB dose-response curve
Ratio Free XPO1
nM b-LMB

C

	50% Occupancy (nM)	90% Occupancy (nM)
MM.1S	0.20	4.32

Figure 4. MM.1S cells were serially dosed with b-LMB for 1.5 hours then subjected to the XPO1 occupancy assay. (A) Digital Western blot images representing XPO1 protein from eluates, and XPO1 and β-actin proteins from input material. (B) Ratio of free XPO1 equals the amount of b-LMB-bound XPO1 compared to the amount of total XPO1 for each dose of b-LMB. (C) 50% of XPO1 protein is occupied at 0.2 nM and 90% is occupied at 4.32 nM b-LMB.

SINE™ Compounds Compete Similarly with b-LMB for XPO1 Binding in a Dose-Dependent Manner in MM.1S Cells

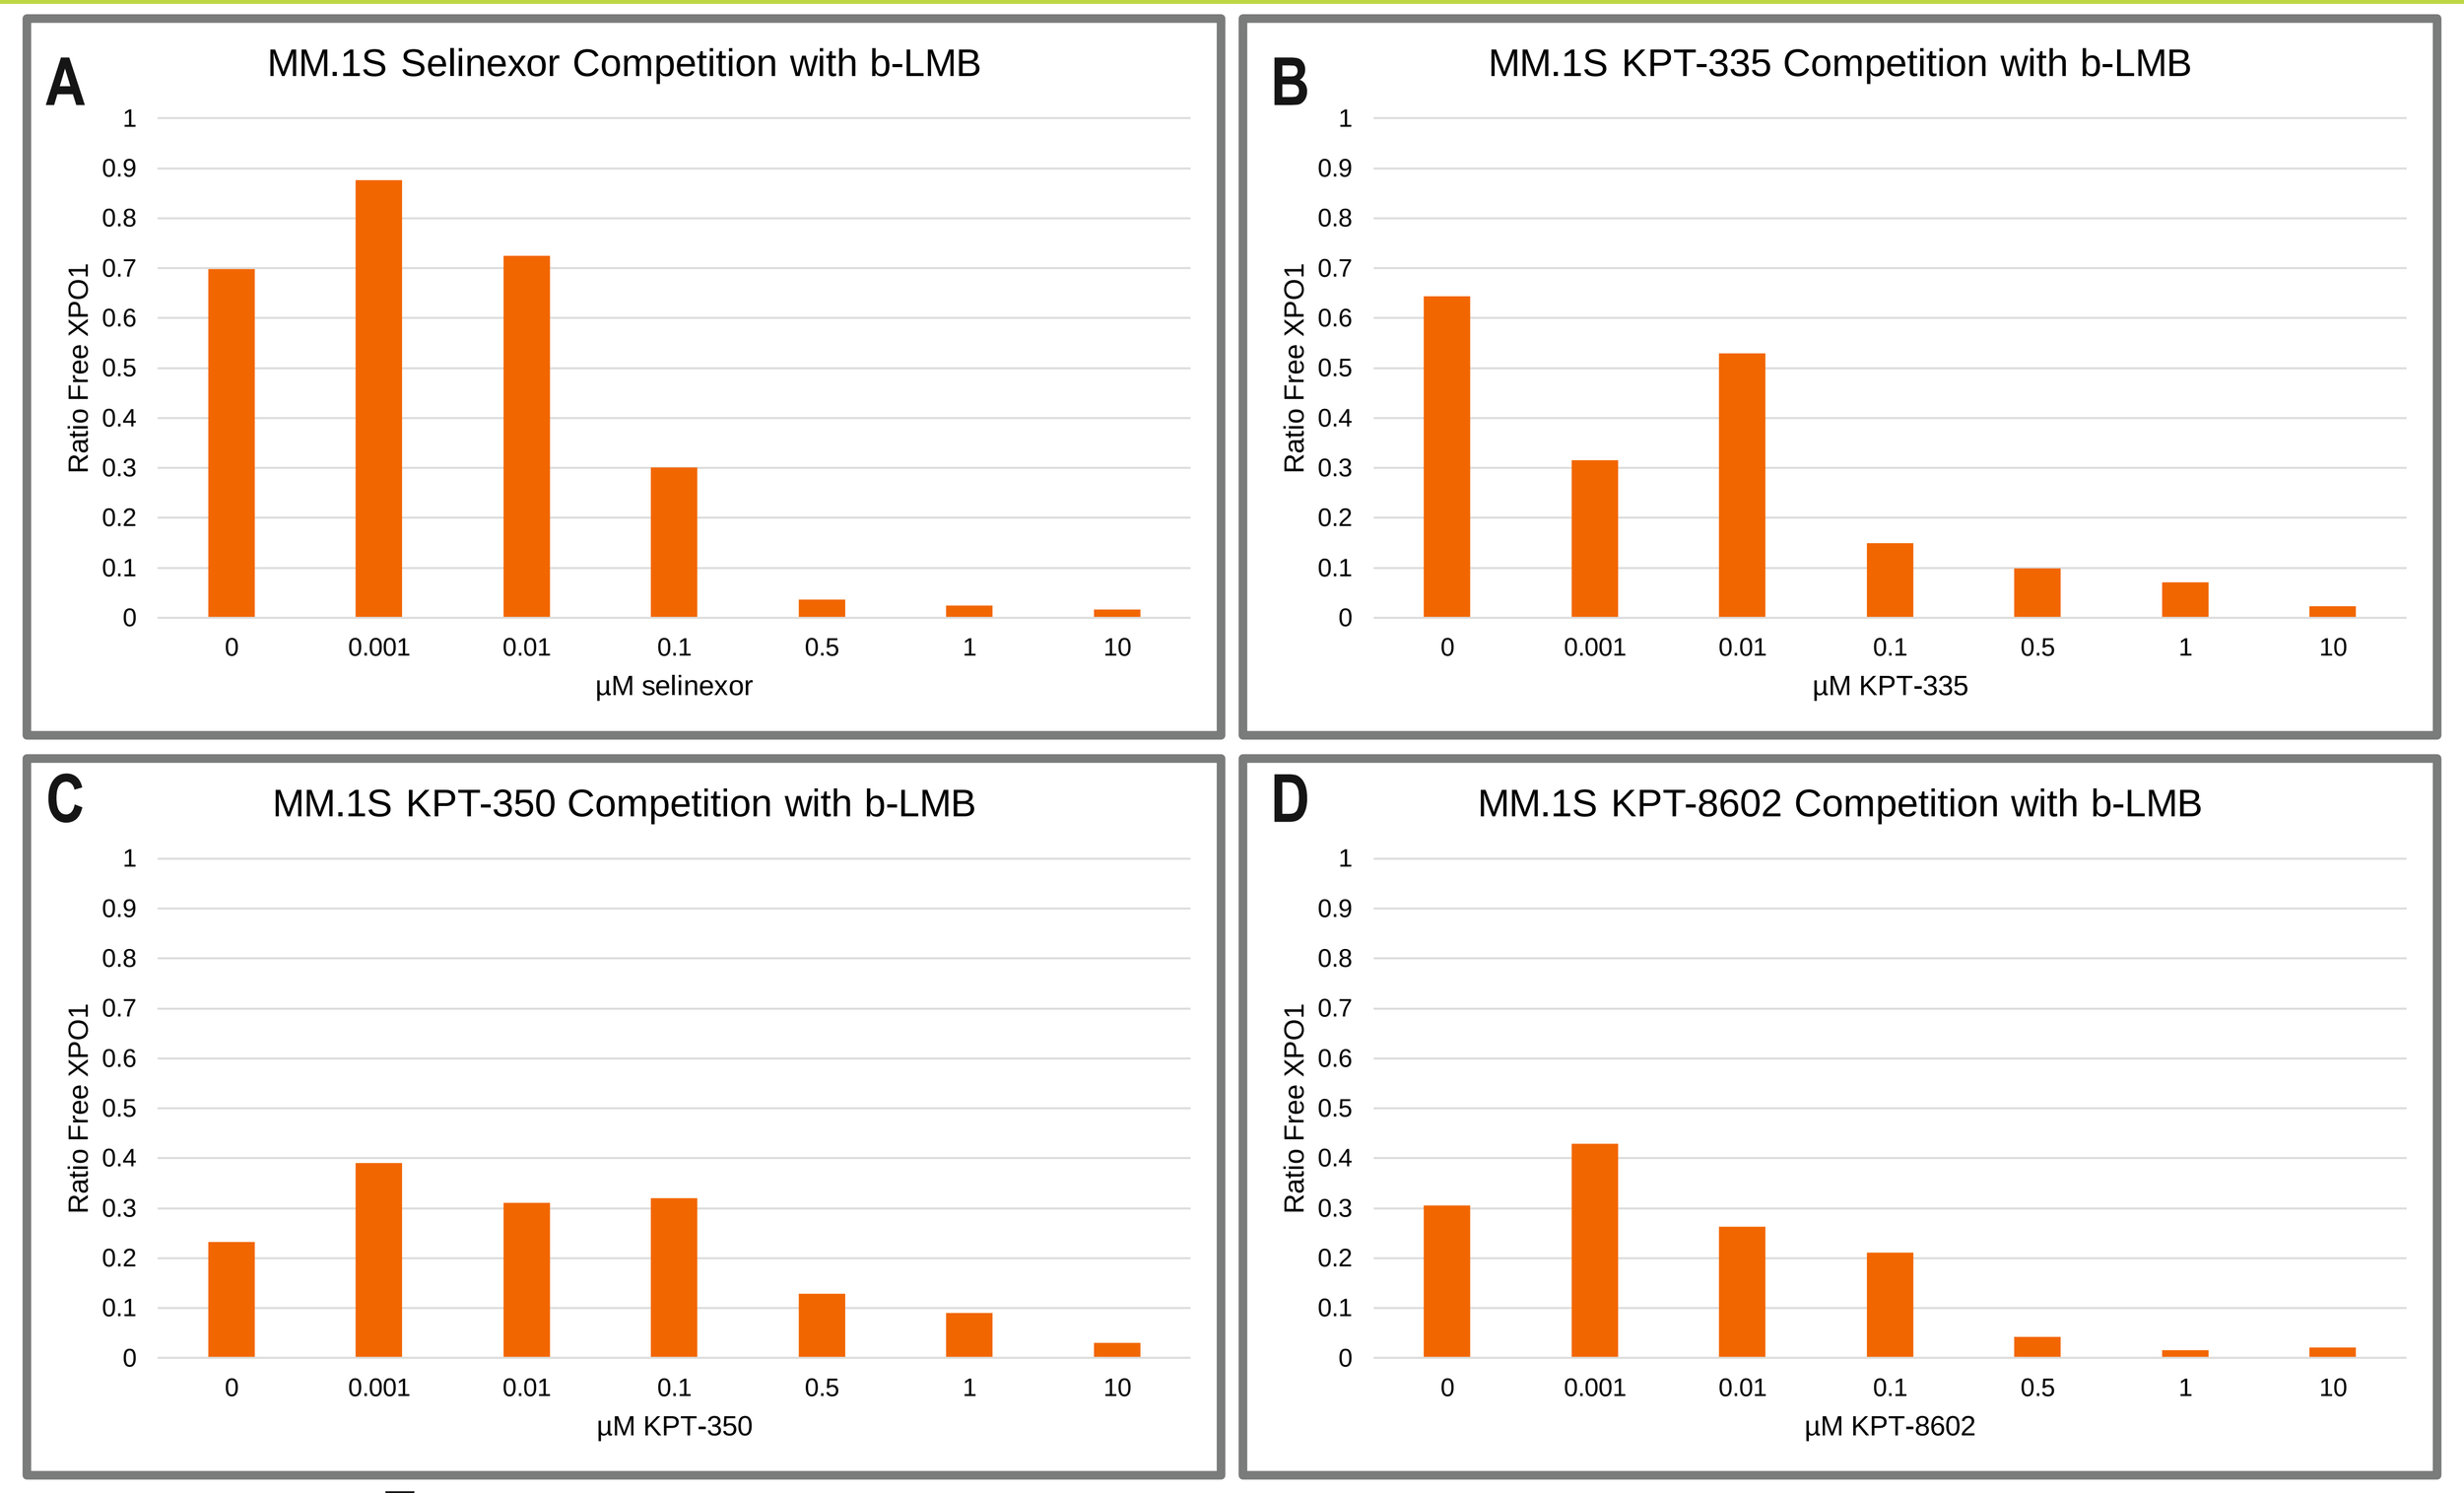
A
MM.1S Selinexor Competition with b-LMB
µM selinexor
B
MM.1S KPT-335 Competition with b-LMB
µM KPT-335
C
MM.1S KPT-350 Competition with b-LMB
µM KPT-350
D
MM.1S KPT-8602 Competition with b-LMB
µM KPT-8602
Ratio Free XPO1

E

SINE™ Compound	50% Occupancy (µM)	90% Occupancy (µM)
Selinexor	0.07	0.48
KPT-335	0.07	0.30
KPT-350	0.62	>10
KPT-8602	0.10	1.83

MM.1S cells were cultured for 1 hour with serial dilutions of SINE™ compounds followed by treatment with b-LMB for 1.5 hours prior to being subjected to the occupancy assay. MM.1S cells treated with (A) selinexor, (B) KPT-335, (C) KPT-350, and (D) KPT-8602. Graphs depict the ratio of free XPO1 compared to the amount of total XPO1 for each dose of SINE™ compound normalized to the drug-free control. (E) Concentrations of each SINE™ compound necessary to attain 50% and 90% occupancy of the XPO1 cargo binding site in the presence of b-LMB.

Selinexor Competes with b-LMB for XPO1 Binding in Human PBMCs

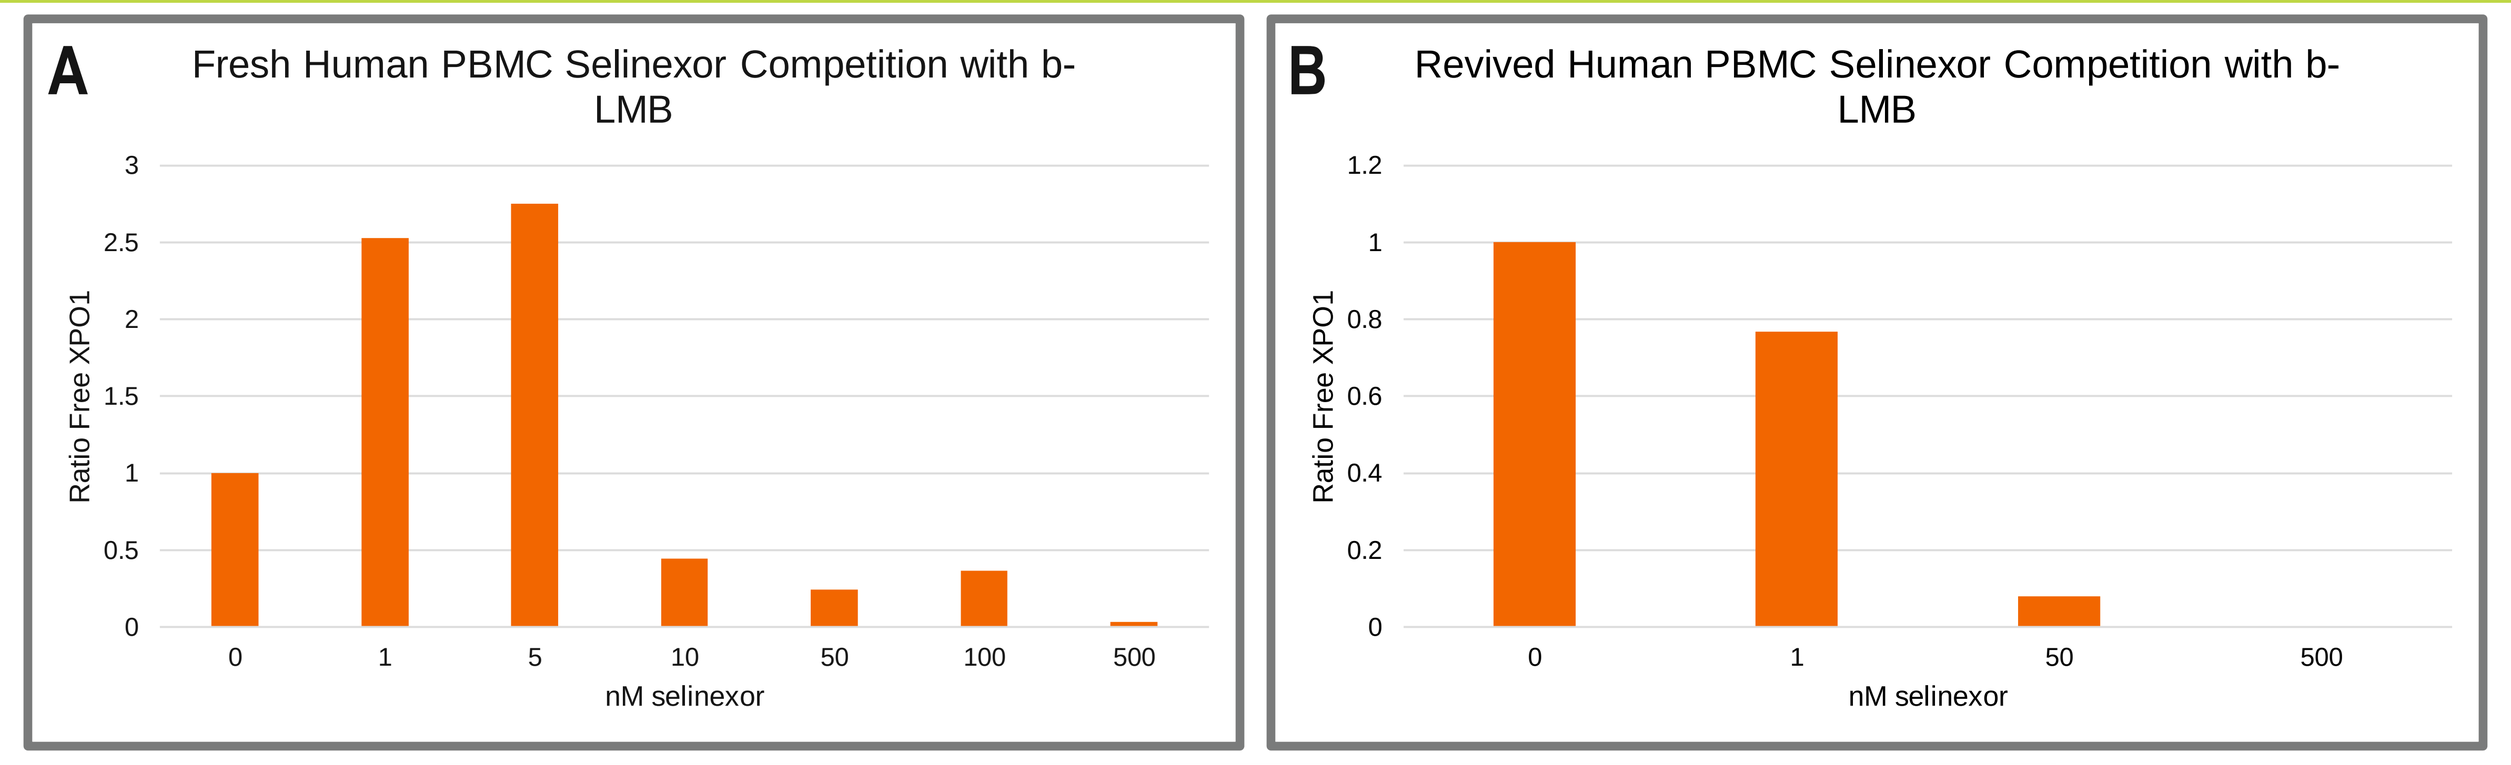
A
Fresh Human PBMC Selinexor Competition with b-LMB
B
Revived Human PBMC Selinexor Competition with b-LMB
Ratio Free XPO1
nM selinexor

C

Human PBMCs	50% Occupancy (nM)	90% Occupancy (nM)
Fresh	9.87	>500
Revived	6.5	41.36

Selinexor competes with b-LMB for XPO1 binding in human PBMCs regardless of whether the PBMCs are treated after being freshly isolated or revived after being viably frozen. (A) Human PBMCs were isolated and treated immediately with selinexor for 1 hour, then with b-LMB for 1 hour, and then subjected to the occupancy assay. (B) Human PBMCs were isolated, viably frozen, revived, and treated with selinexor for 4 hours, then with b-LMB for 1 hour before being subjected to the occupancy assay. Graphs depict the ratio of free XPO1 compared to the amount of total XPO1 for each dose of selinexor normalized to the drug-free control. (C) Concentrations of selinexor necessary to attain 50% and 90% occupancy of the XPO1 cargo binding site in the presence of b-LMB in fresh versus revived PMBCs.

Selinexor Competes with b-LMB for XPO1 Binding in PBMCs From Mice Dosed with Selinexor

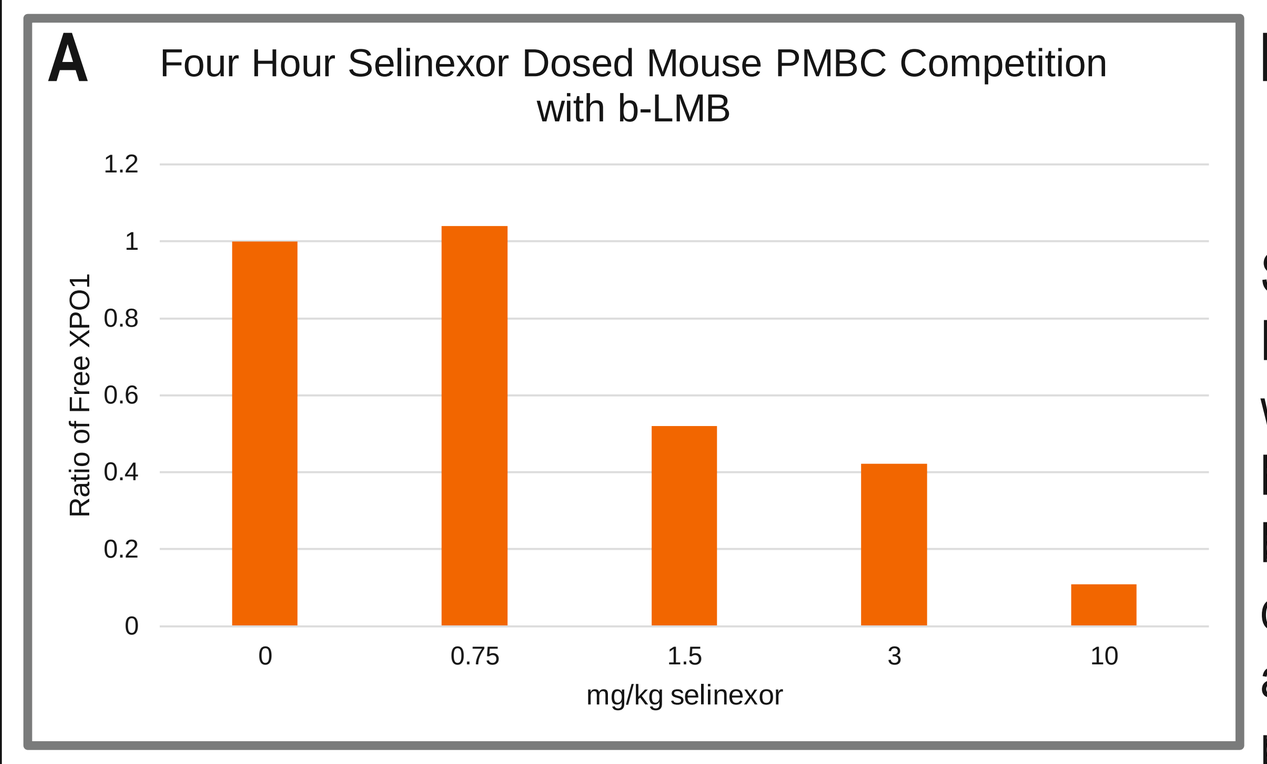
A
Four Hour Selinexor Dosed Mouse PMBC Competition with b-LMB
Ratio of Free XPO1
mg/kg selinexor

B

	50% Occupancy (mg/kg)	90% Occupancy (mg/kg)
Mouse PBMCs	1.84	>10

Selinexor competes with b-LMB for XPO1 binding mouse PBMCs in a dose-dependent manner. (A) Mice (4 per group) were dosed with 0, 0.75, 1.5, 3, or 10 mg/kg selinexor for 4 hours. PBMCs (pooled per group) were isolated, cultured with b-LMB for 1.5 hours, and the XPO1 occupancy was determined. (B) Amounts of selinexor necessary to attain 50% and 90% occupancy of the XPO1 cargo binding site in the presence of b-LMB after 4 hours of treatment.

Binding to XPO1 is Sustained For Up to 72 Hours in PBMCs From Mice Dosed with Selinexor

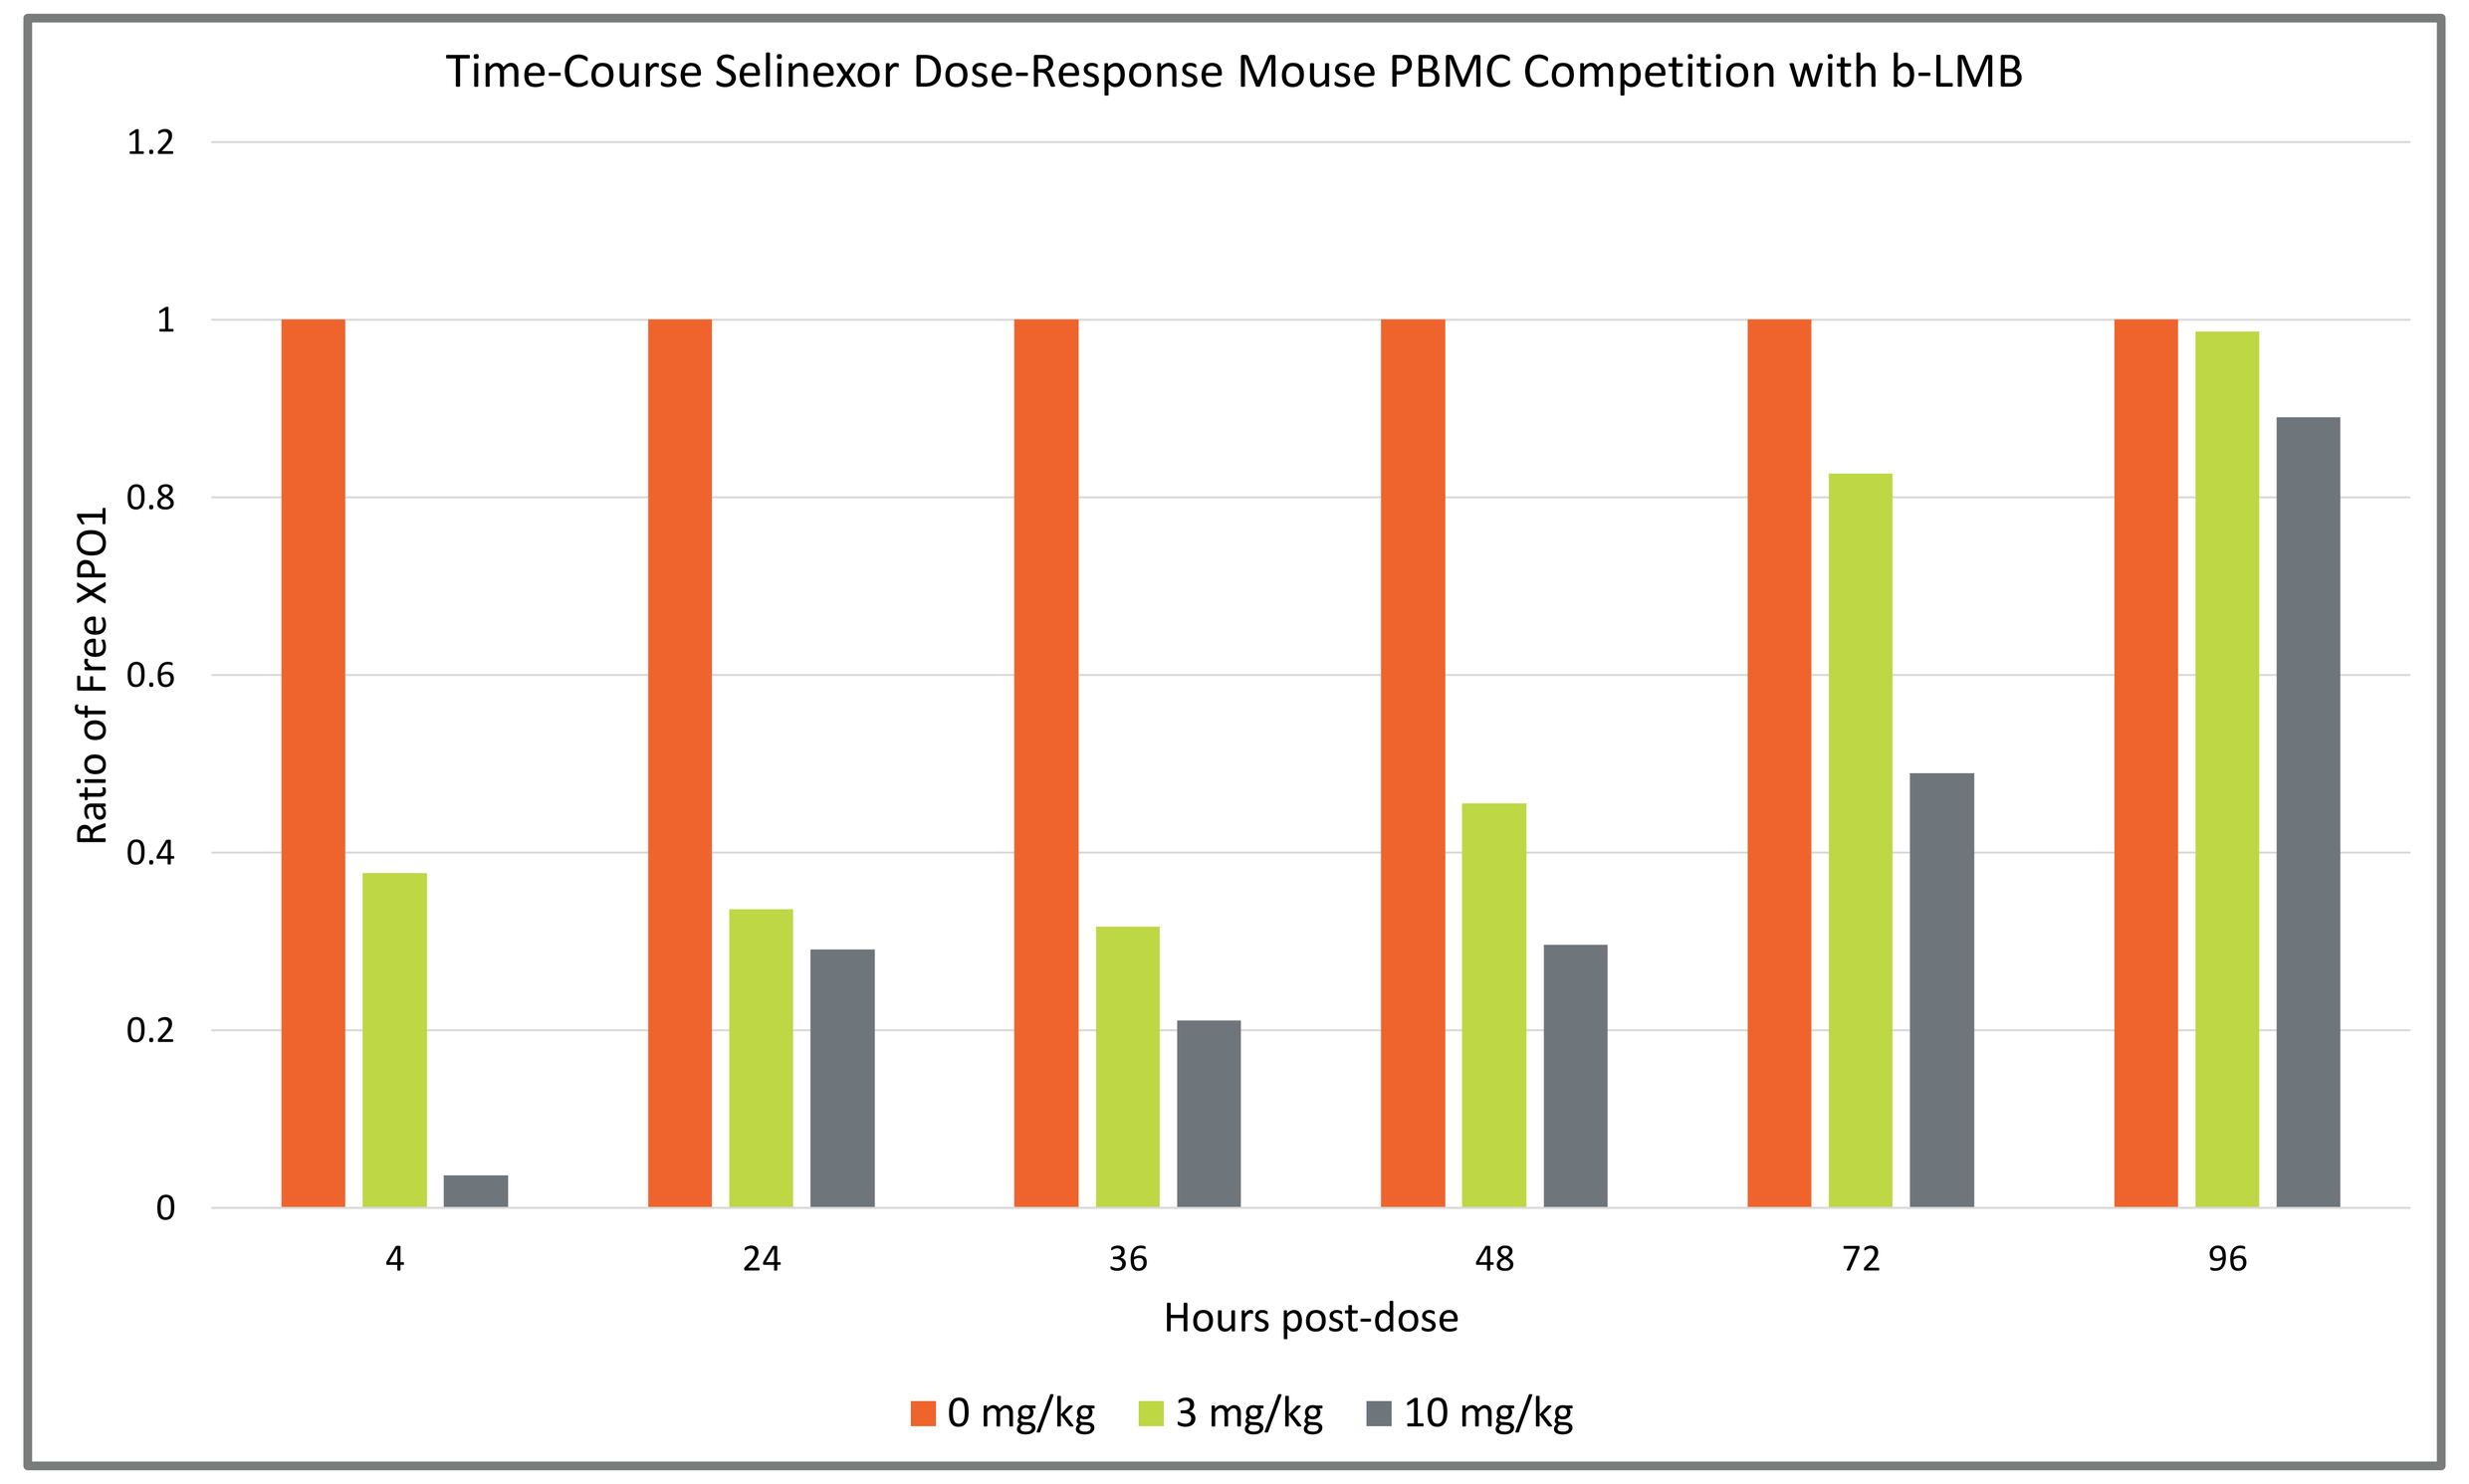
Time-Course Selinexor Dose-Response Mouse PBMC Competition with b-LMB
Ratio of Free XPO1
Hours post-dose
0 mg/kg
3 mg/kg
10 mg/kg

Selinexor competes with b-LMB for XPO1 binding from mouse PBMCs in a time-dependent manner. Mice (4 per group) were dosed once with either 0, 3, or 10 mg/kg selinexor. PMBCs (pooled per group) were isolated at 4, 24, 36, 48, 72, and 96 hours, cultured with b-LMB for 1.5 hours, and the XPO1 occupancy was determined. Selinexor competes with b-LMB for XPO1 at 4 through 72 hours post-dose. Selinexor no longer competes with b-LMB at 96 hours post-dose.

In Vitro Cell Lines Have Similar XPO1 Occupancy Values Regardless of Their Sensitivity to Selinexor

Cell Line	Selinexor IC_{50} (µM)	50% Occupancy selinexor (µM)	90% Occupancy selinexor (µM)
MM.1S	0.02	0.07	0.48
HCT-116	0.05	0.15	0.76
HT1080	0.07	0.44	1.35
AML2	0.09	0.01	0.22
AML3	0.27	0.04	0.46
HEL	0.35	0.12	0.31
Kasumi-6	0.53	0.14	0.57
HT1080 resistant	2.4	0.4	1.67
A549	3	0.09	0.56
UCH1	7	0.15	0.65
UCH2	30	0.27	0.89
LS174T	>10	0.11	0.38
ASPS-KY	>10	0.24	0.72

Summary

- b-LMB is a novel tool compound created by Karyopharm Therapeutics which shows similar nuclear export inhibitory activity and cytotoxicity compared to unmodified LMB.
- SINE™ compounds compete in a similar dose-response fashion with b-LMB for binding to XPO1 protein from cells in vitro.
- Selinexor effectively competes with b-LMB for XPO1 binding from PBMCs isolated from donor human blood regardless of whether the cells are fresh or recovered from freezing.
- In vivo experiments from mice dosed with selinexor show selinexor competes with b-LMB for XPO1 binding in PBMCs, with 50% of XPO1 binding occurring at 1.84 mg/kg and 90% of XPO1 binding occurring at >10 mg/kg, and that the competition is sustained up to 72 hours.
- Selinexor patient PK T_{max} is consistent with the optimal b-LMB – XPO1 occupancy of 4 hours observed in mice dosed with selinexor
- Changes in levels of selinexor-induced gene expression with time in patients is consistent with the degree of XPO1 occupancy by b-LMB over time in PMBCs from mice dosed with selinexor.
- Regardless of sensitivity to selinexor, competition between selinexor and b-LMB is similar in cell lines of various cancer indications in vitro.
- b-LMB can serve as a tool compound for the evaluation of drug exposure in patients by measuring the binding of selinexor to XPO1 in patient PBMCs ex vivo.

Contact Information: Marsha Crochiere marsha@Karyopharm.com +1-617-658-0544

TEMPLATE DESIGN © 2008
www.PosterPresentations.com