The role of nuclear export in TDP43-mediated neurodegeneration

Hilary C. Archbold[1], Sharon Tamir[2], Xingli Li[1], Sami J. Barmada[1]

[1]Department of Neurology, University of Michigan, Ann Arbor, MI; [2]Karyopharm Therapeutics, Newton, MA; [3]Cellular & Molecular Biology Program, [4]Neuroscience Program, University of Michigan, Ann Arbor, MI

Cytoplasmic TDP43 mislocalization is found in the related neurodegenerative diseases ALS and FTD

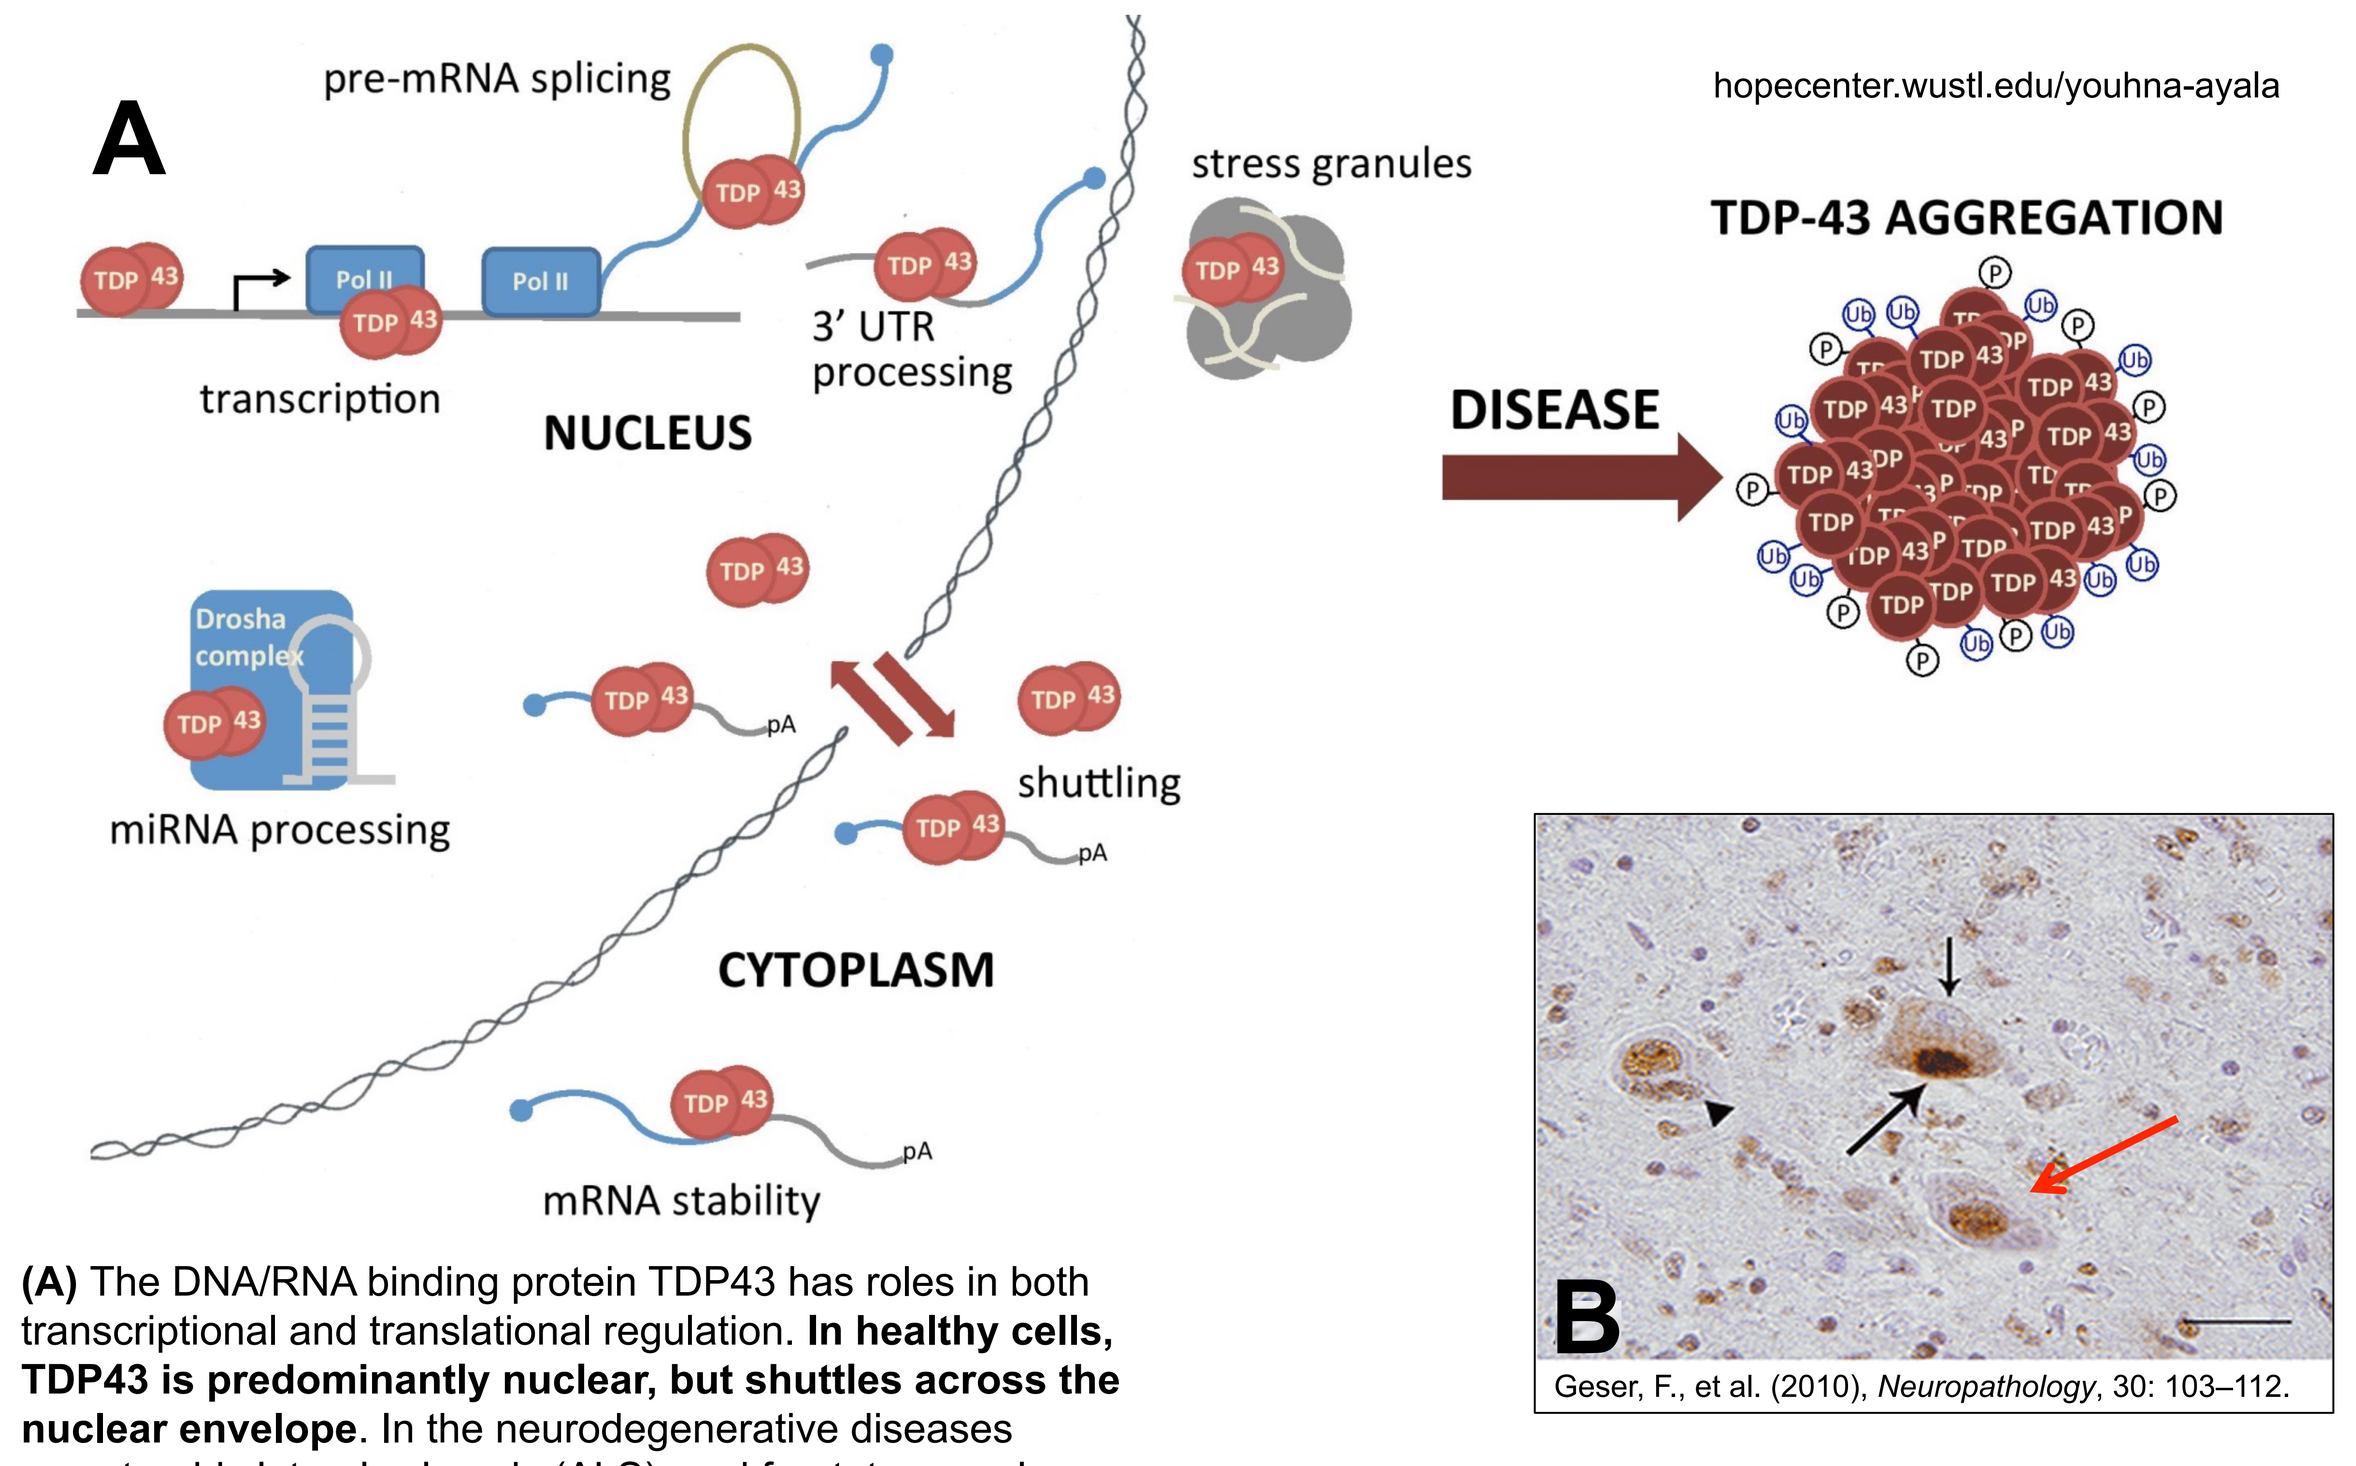
A
pre-mRNA splicing
transcription
3' UTR processing
stress granules
NUCLEUS
hopecenter.wustl.edu/youhna-ayala
TDP-43 AGGREGATION
DISEASE
Drosha complex
miRNA processing
shuttling
CYTOPLASM
mRNA stability
B

Geser, F., et al. (2010), *Neuropathology*, 30: 103–112.

(A) The DNA/RNA binding protein TDP43 has roles in both transcriptional and translational regulation. **In healthy cells, TDP43 is predominantly nuclear, but shuttles across the nuclear envelope**. In the neurodegenerative diseases amyotrophic lateral sclerosis (ALS), and frontotemporal dementia (FTD), accumulation and aggregation of TDP43 is seen in the cytoplasm of affected neurons (Image courtesy Dr. Youhna Ayala). **(B)** Post-mortem tissue section, stained for TDP43 (Geser et al., 2010). Red arrow, cell with normal nuclear TDP43 levels. Black arrowhead, cell with nuclear TDP43 plus cytoplasmic TDP43 aggregates. Black arrows, cell with depleted nuclear TDP43 (small arrow), and general cytoplasmic accumulation with aggregates (large arrow). In our primary neuron model of ALS, **increased cytoplasmic TDP43 is the strongest predictor of cell death**, and several disease associated mutations in TDP43 have been linked to increased cytoplasmic TDP43 levels. These observations led us to ask the question: **Can we slow disease progression and improve neuronal survival by blocking TDP43 export to the cytoplasm?**

Exportin1 is a candidate transporter for TDP43

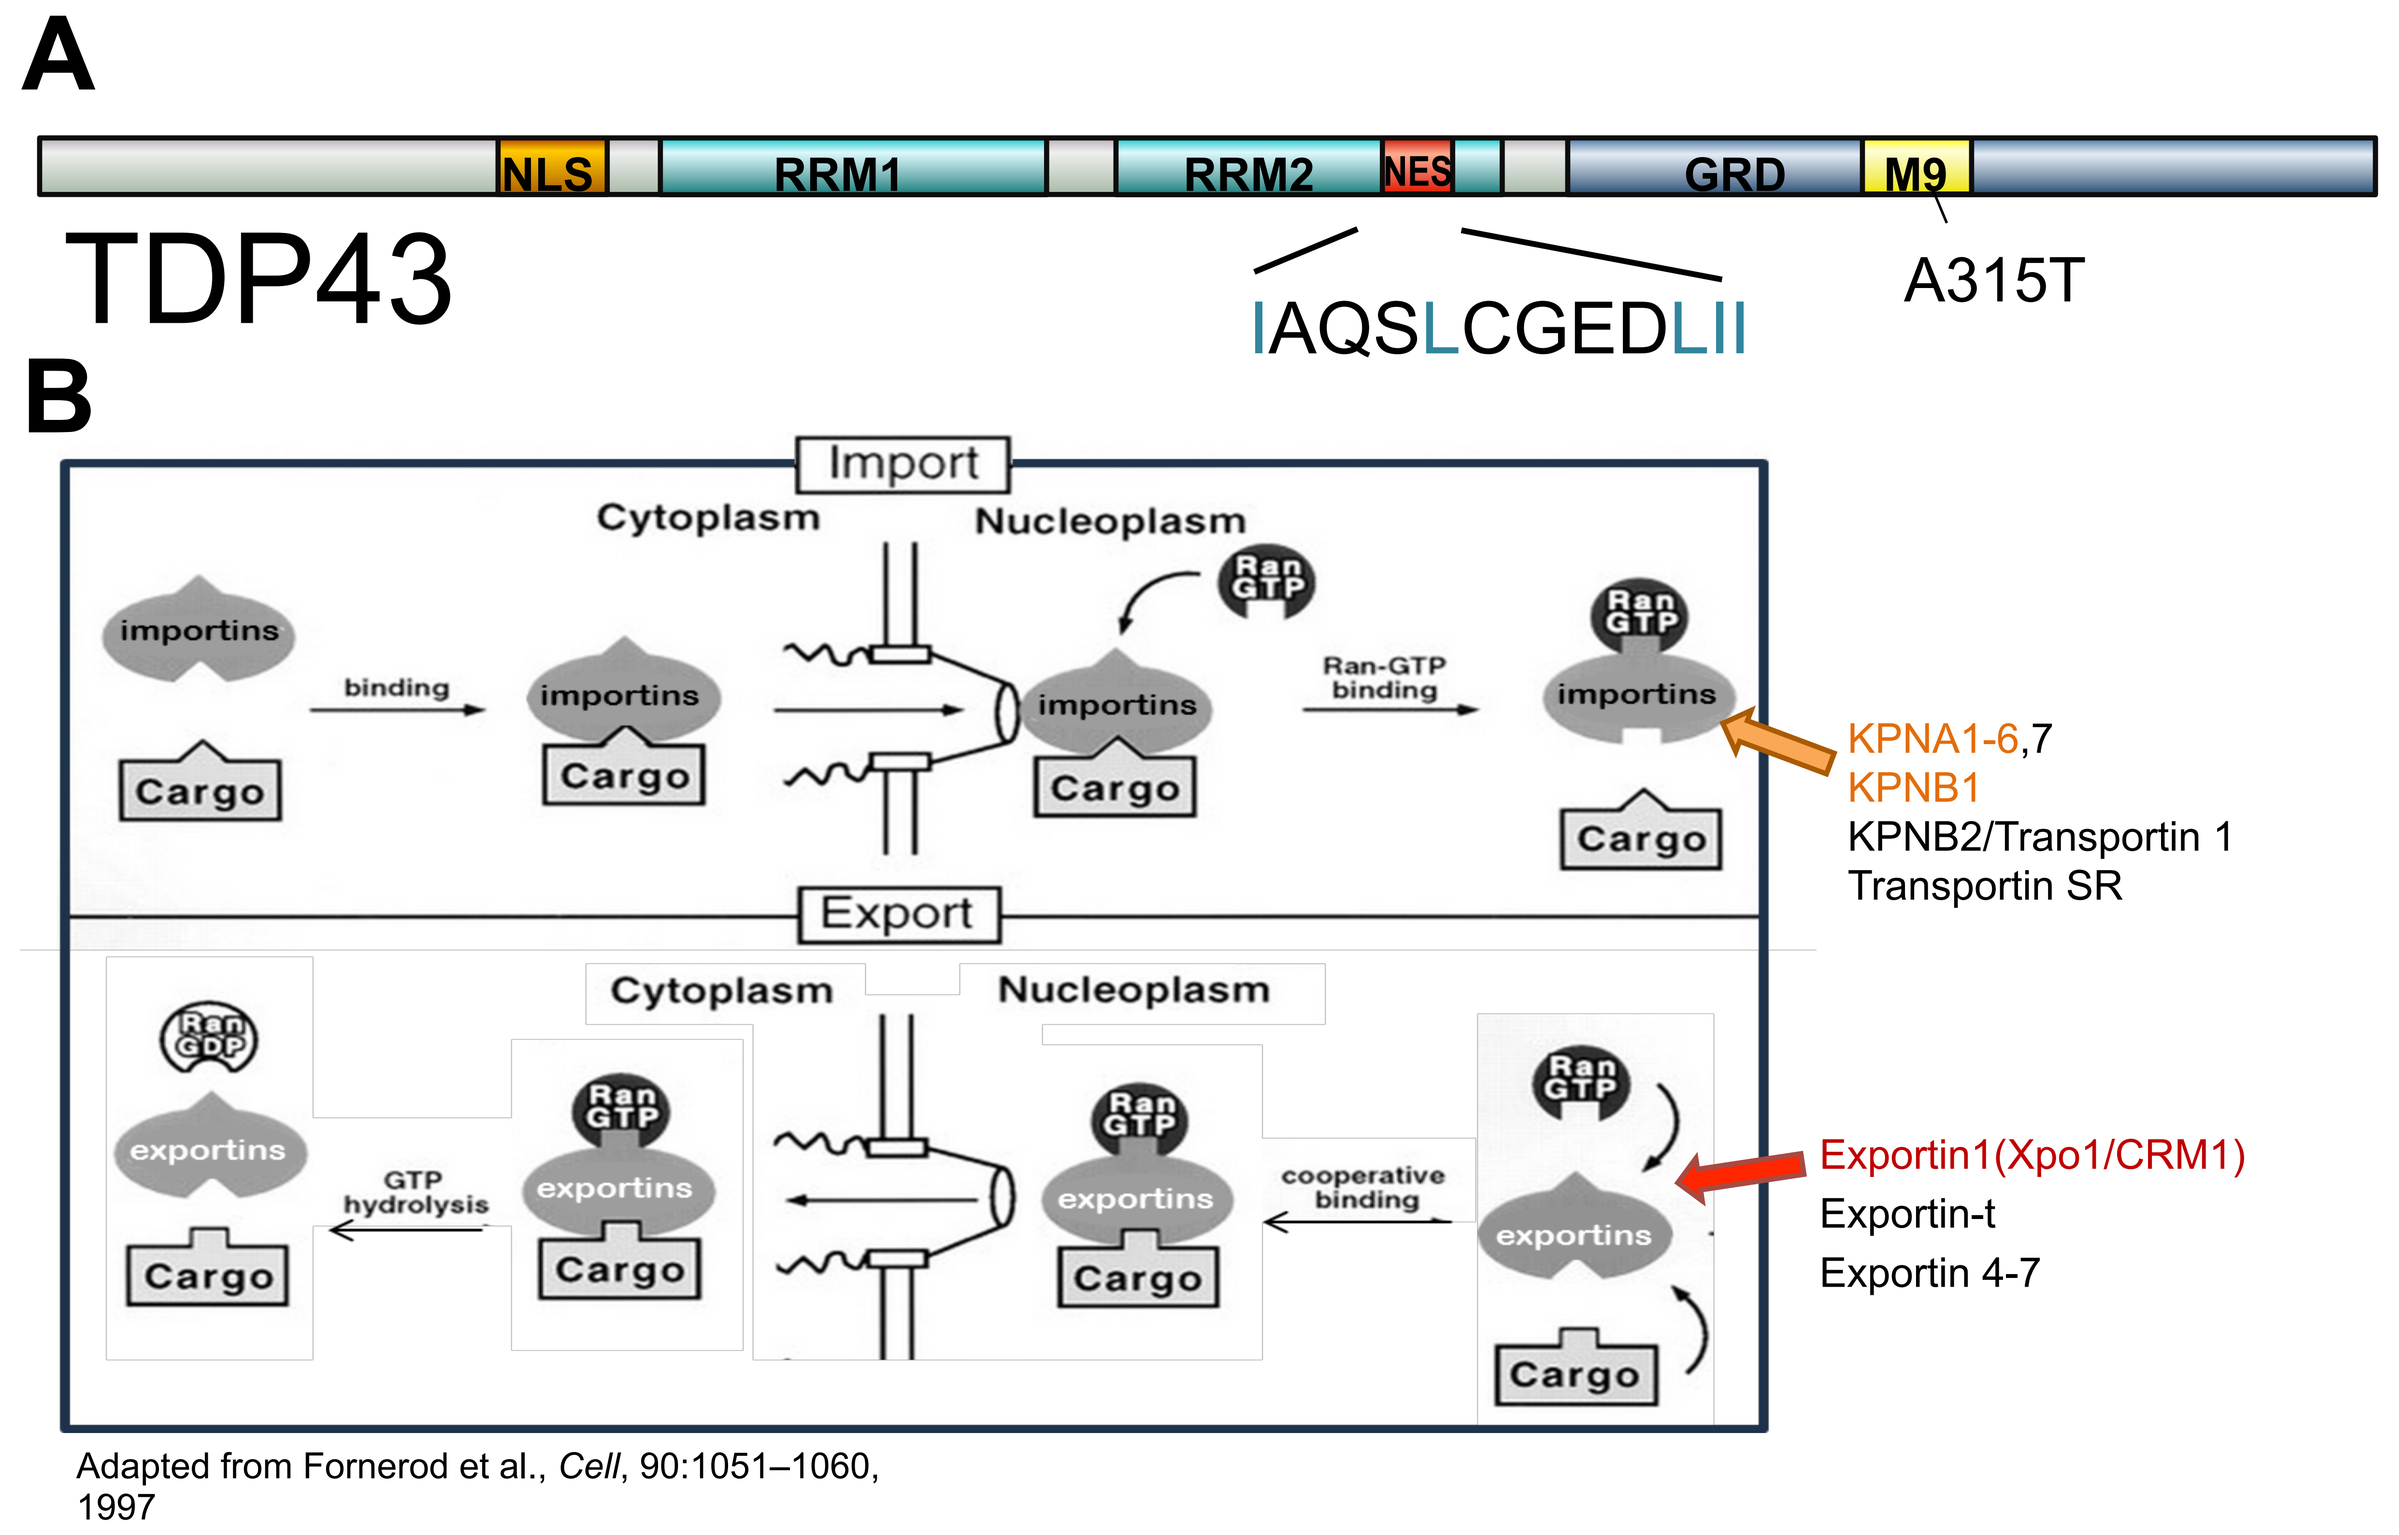
A
NLS
RRM1
RRM2
NES
GRD
M9
TDP43
IAQSLCGEDLII
A315T
B
Import
Cytoplasm
Nucleoplasm
importins
binding
Cargo
Ran-GTP binding
KPNA1-6,7
KPNB1
KPNB2/Transportin 1
Transportin SR
Export
exportins
GTP hydrolysis
cooperative binding
Exportin1(Xpo1/CRM1)
Exportin-t
Exportin 4-7

Adapted from Fornerod et al., *Cell*, 90:1051–1060, 1997

(A) TDP43 contains an isoleucine/leucine-rich Nuclear Export Signal (NES) motif (red), as well as a Nuclear Localization Signal (NLS) motif (orange), and an M9 domain (yellow), which has been shown to mediate bi-directional shuttling in the related RNA binding proteins, the hnRNPs. The familial ALS-associated mutation A315T, located in the M9 domain, has been experimentally shown to cause an increase in cytoplasmic localization of TDP43 (Barmada et al., *J Neurosci*., 2010). **(B)** The karyopherin family of transport proteins are the primary mediators of nucleocytoplasmic shuttling, and interact with their cargo by binding to either NLS, NES, or M9 motifs located on the cargo proteins. TDP43 has a well studied NLS which is recognized by multiple importin family members (orange). It is unknown which member or members of the exportin family are required for TDP43 export via the NES. **Exportin1 (red) mediates the majority of nuclear export events. It recognizes leucine rich NES motifs, leading us to hypothesize that exportin1 may transport TDP43.** Therefore, blocking its activity may have a positive impact on neuronal survival.

Selective inhibitor of nuclear export (SINE) compounds

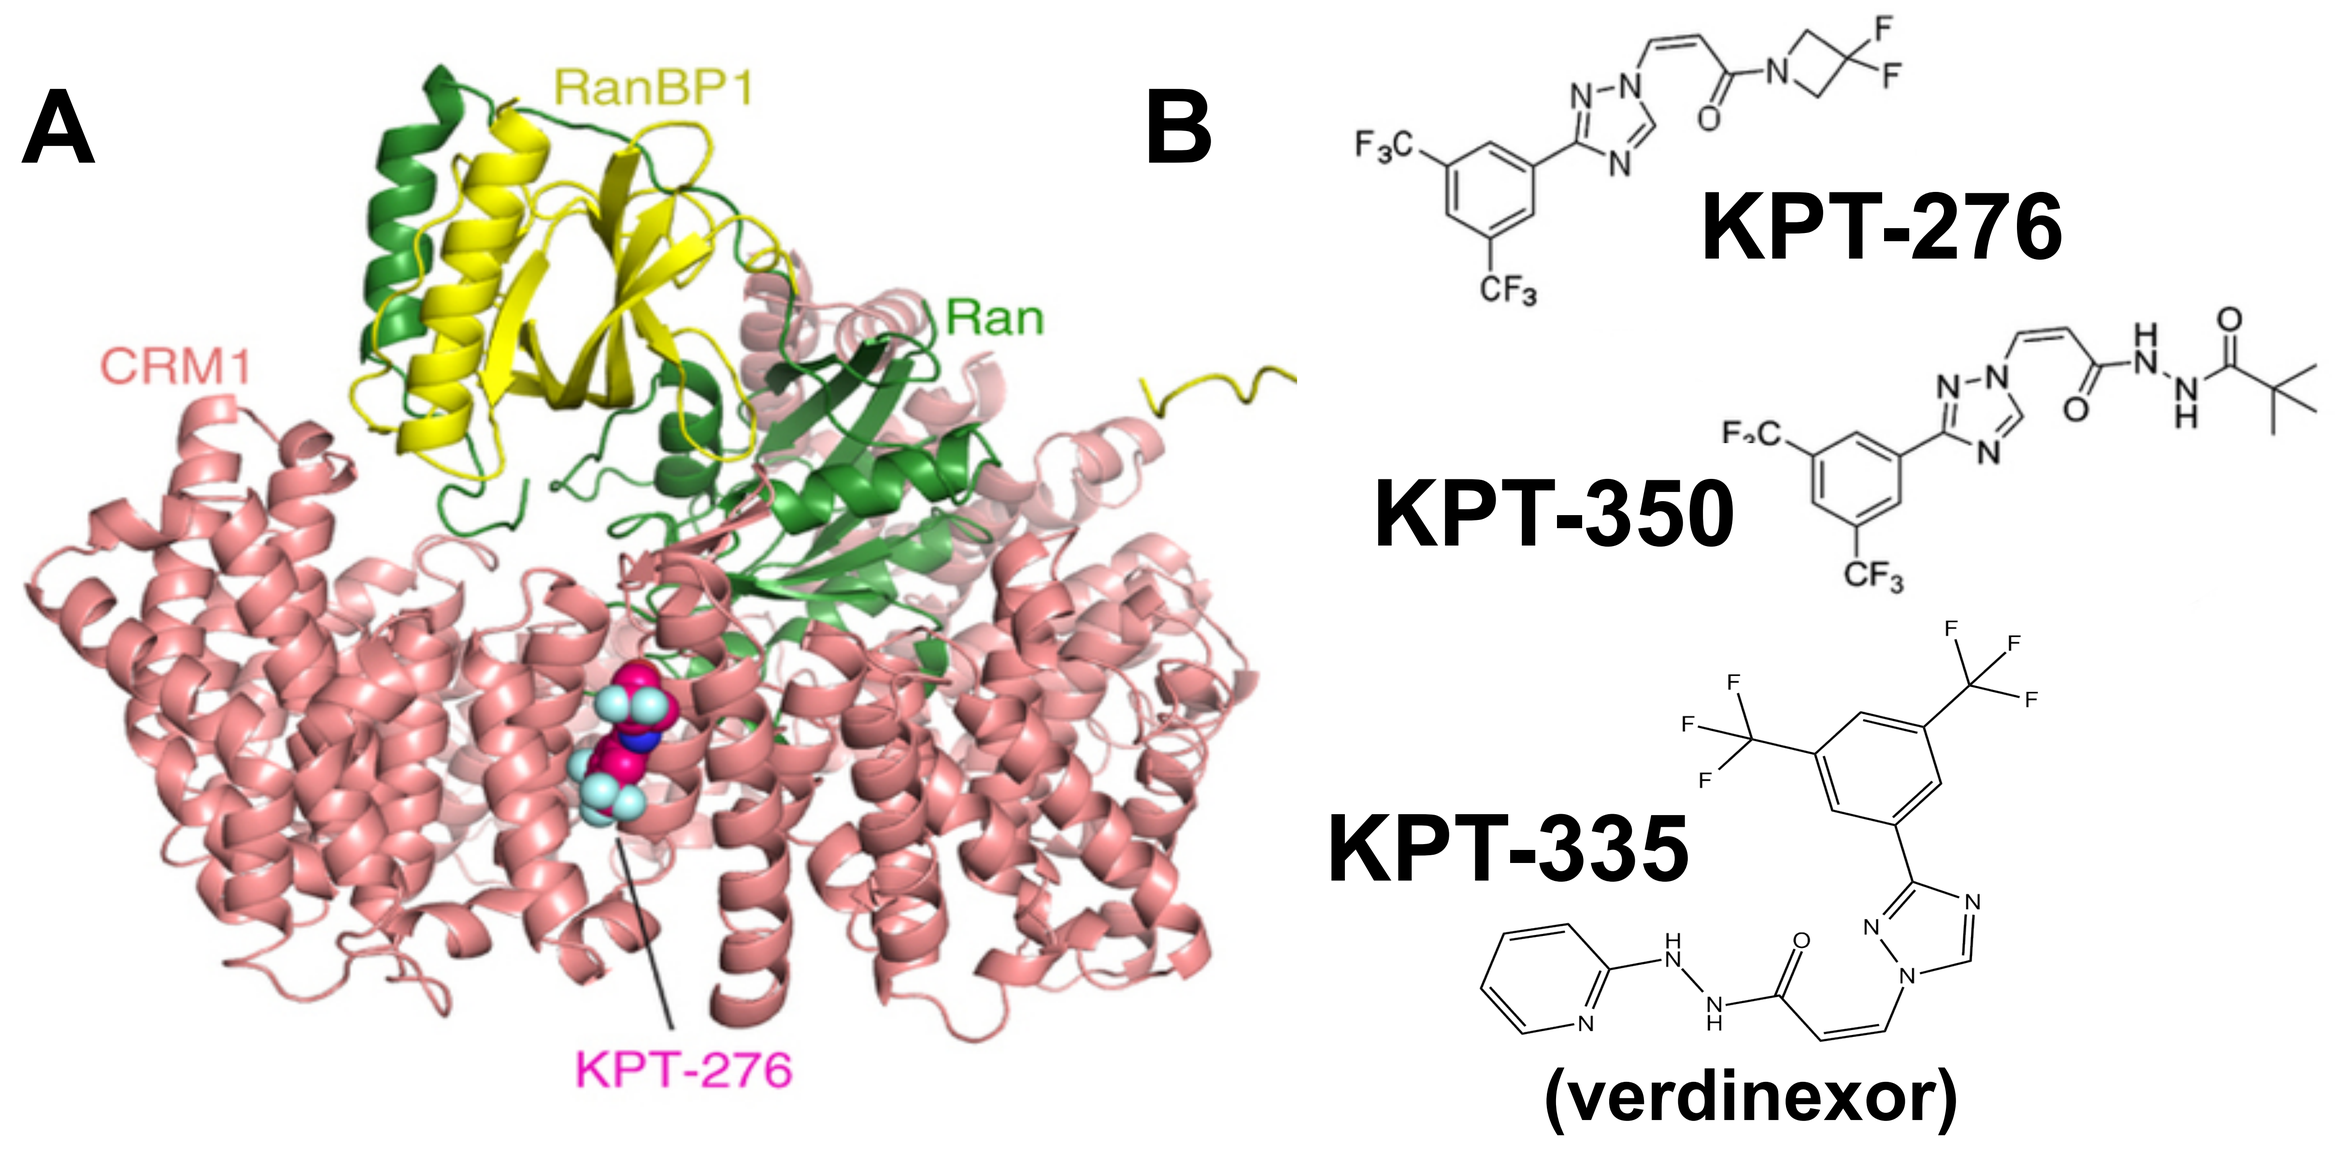
A
RanBP1
Ran
CRM1
KPT-276
B
KPT-276
KPT-350
KPT-335
(verdinexor)

SINE compounds are slowly reversible covalent modifiers of Exportin1. (A) 1.8 Å crystal structure of XPO1/CRM1 bound to KPT-276, taken from Haines, JD et al., *Nature Neuroscience* **18**, 511–520 (2015). **(B)** Molecular structure of KPT 276 and the related compounds, KPT335 and KPT350, which we are testing for efficacy in our model of ALS and FTD.

Longitudinal Fluorescence Microscopy: tracking neuronal survival in a model of ALS/FTD

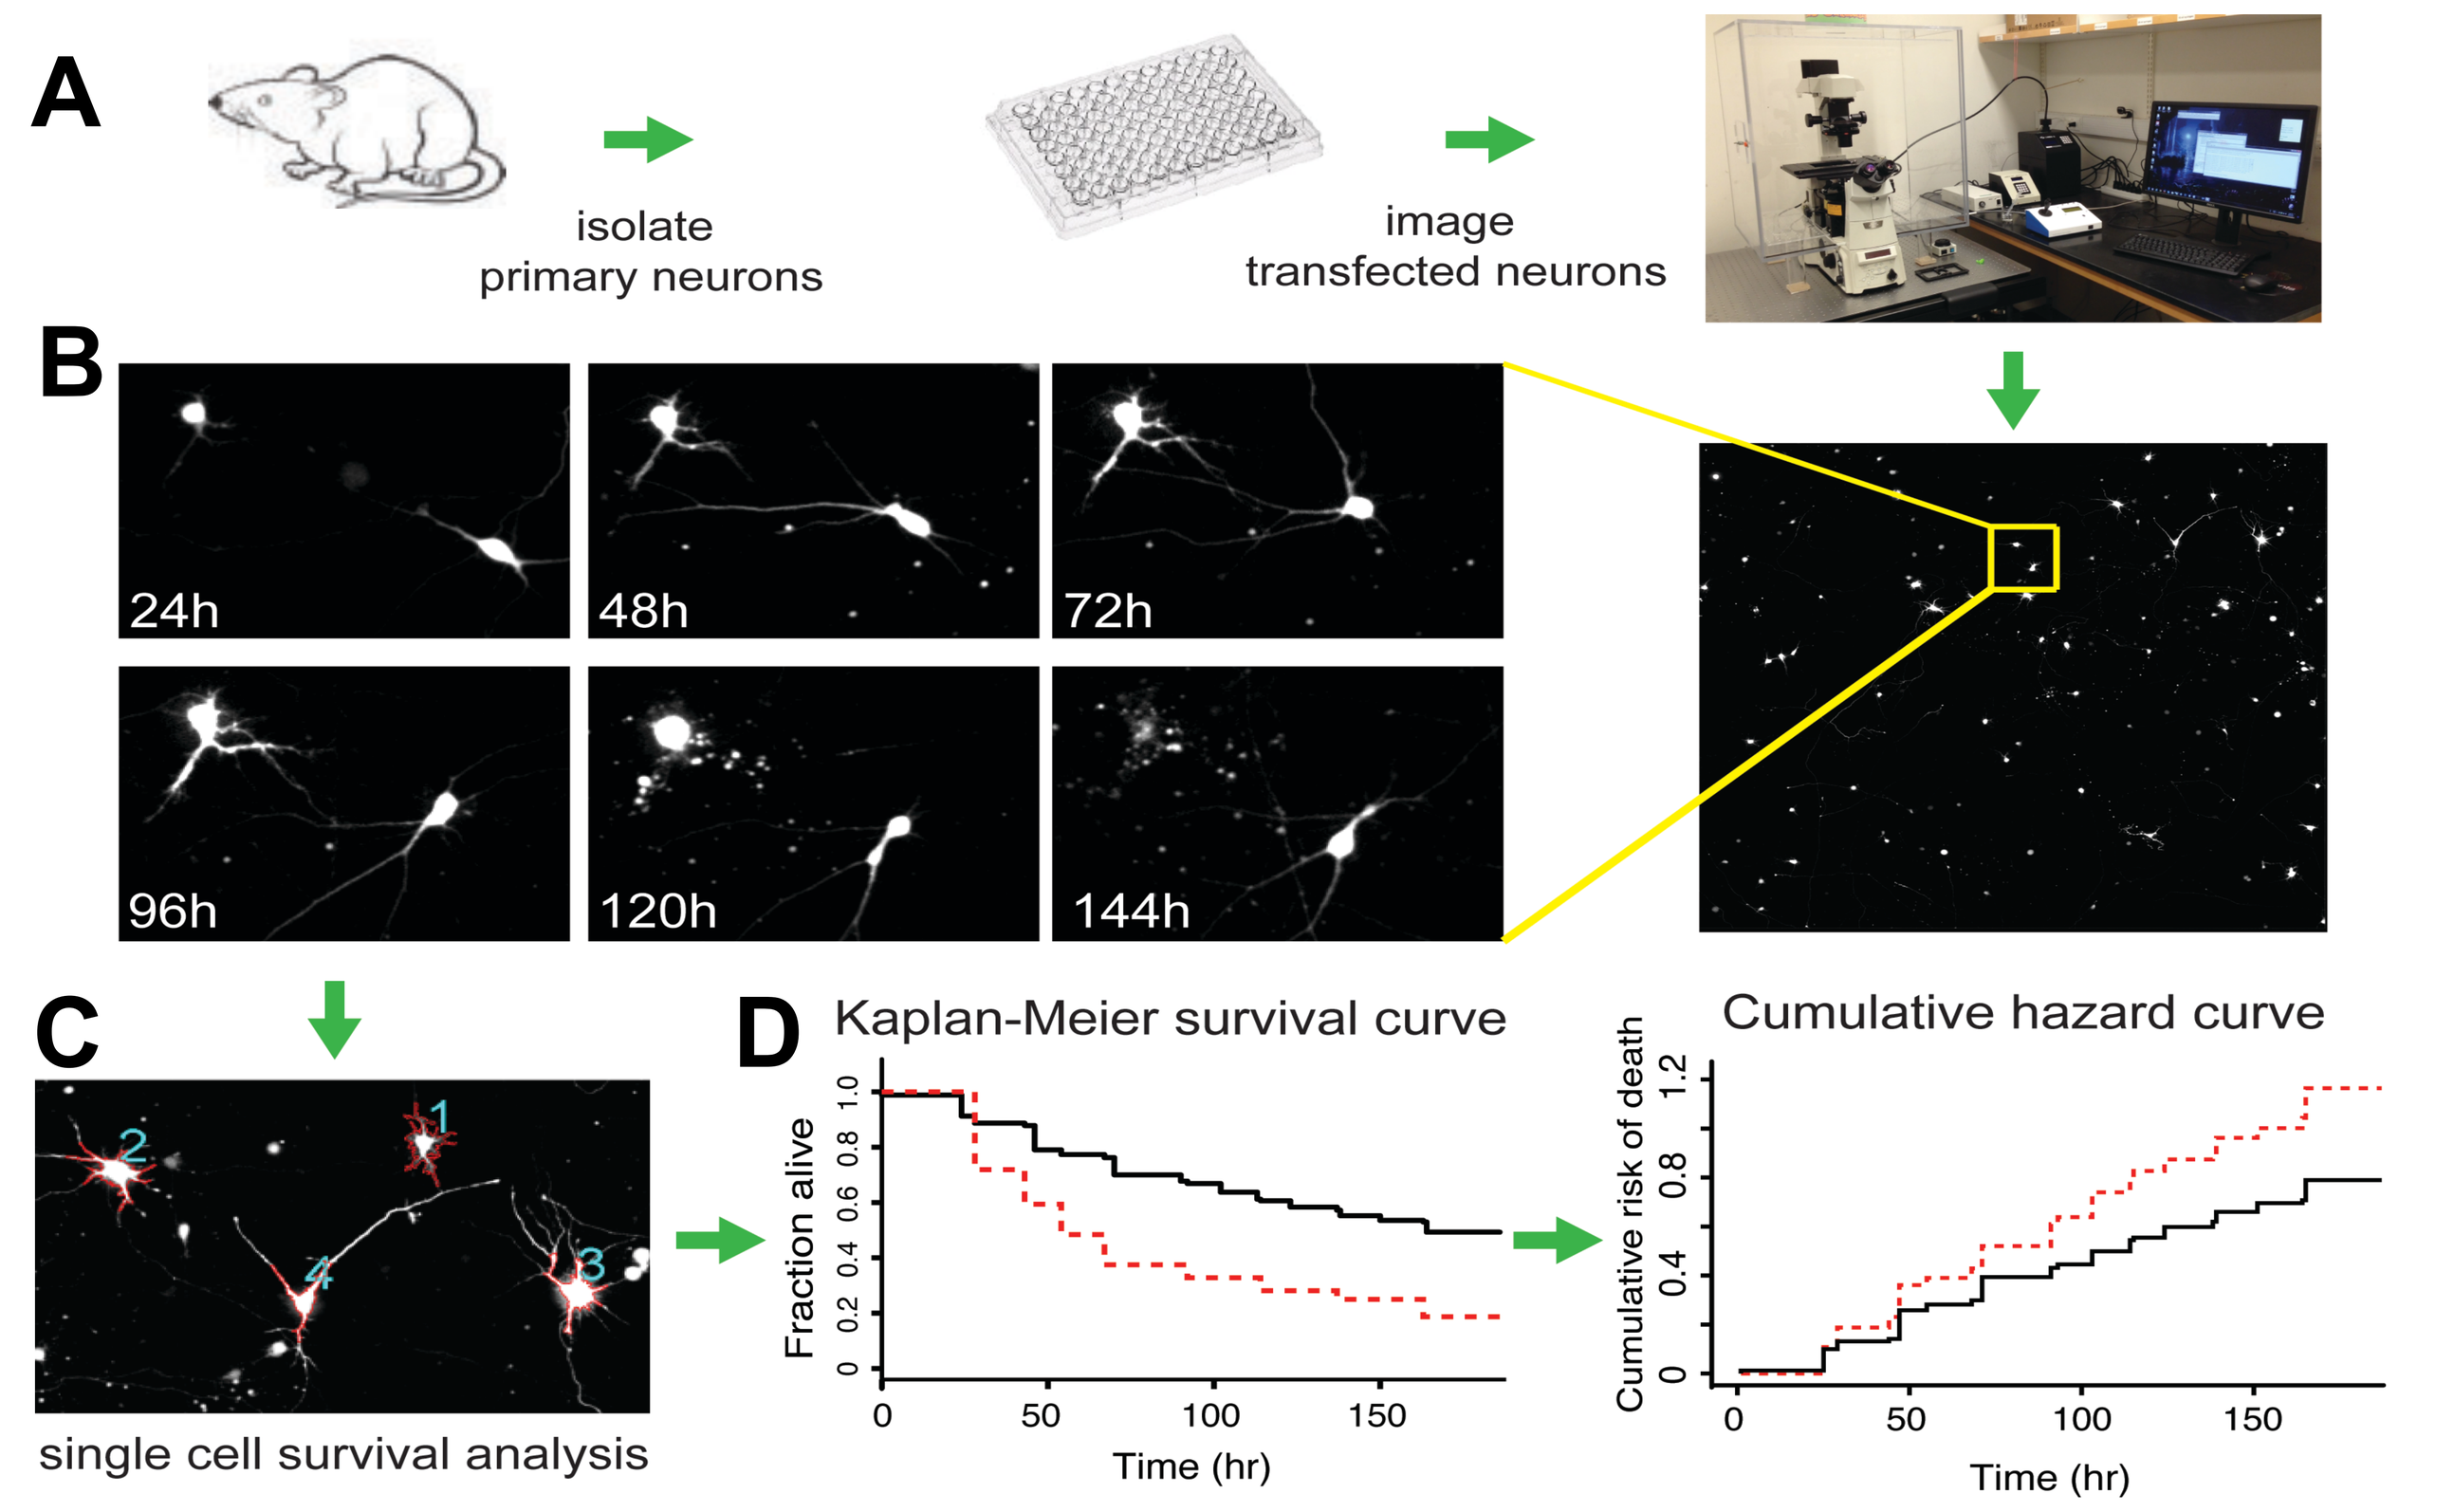
A
isolate primary neurons
image transfected neurons
B
24h
48h
72h
96h
120h
144h
C
single cell survival analysis
D
Kaplan-Meier survival curve
Fraction alive
Time (hr)
Cumulative hazard curve
Cumulative risk of death
Time (hr)

(A) Experimental model of ALS and FTD. Rat primary cortical neurons are isolated, cultured, and transiently transfected with fluorescently tagged TDP43. Overexpression of TDP43 negatively impacts neuronal survival and recapitulates key pathological features of ALS. **(B)** Transfected neurons are imaged for 10 days, and an automated analysis program **(C)** identifies individual neurons and tracks their time of death using indicators such as morphology changes and fluorescence intensity loss. **(D)** Kaplan Meier survival curves (left) are a historically popular way to plot survival data. In these plots, the gradual decrease in surviving neurons is represented by a decrease in the y axis over time. Control population: black line, experimental population: dashed red line. However, an alternative is to **plot the cumulative risk of death using Cox hazard analysis** (right). In these plots, increased risk of death in an experimental population (red dashed line) is represented by an increase in the slope for the hazard curve, when compared to the control (black line), while inversely, a decrease in slope would indicate a condition which imparts a survival benefit on the experimental population (not shown). Figure courtesy Brittany Flores

SINE compounds are neuroprotective in a neuronal model of ALS and FTD

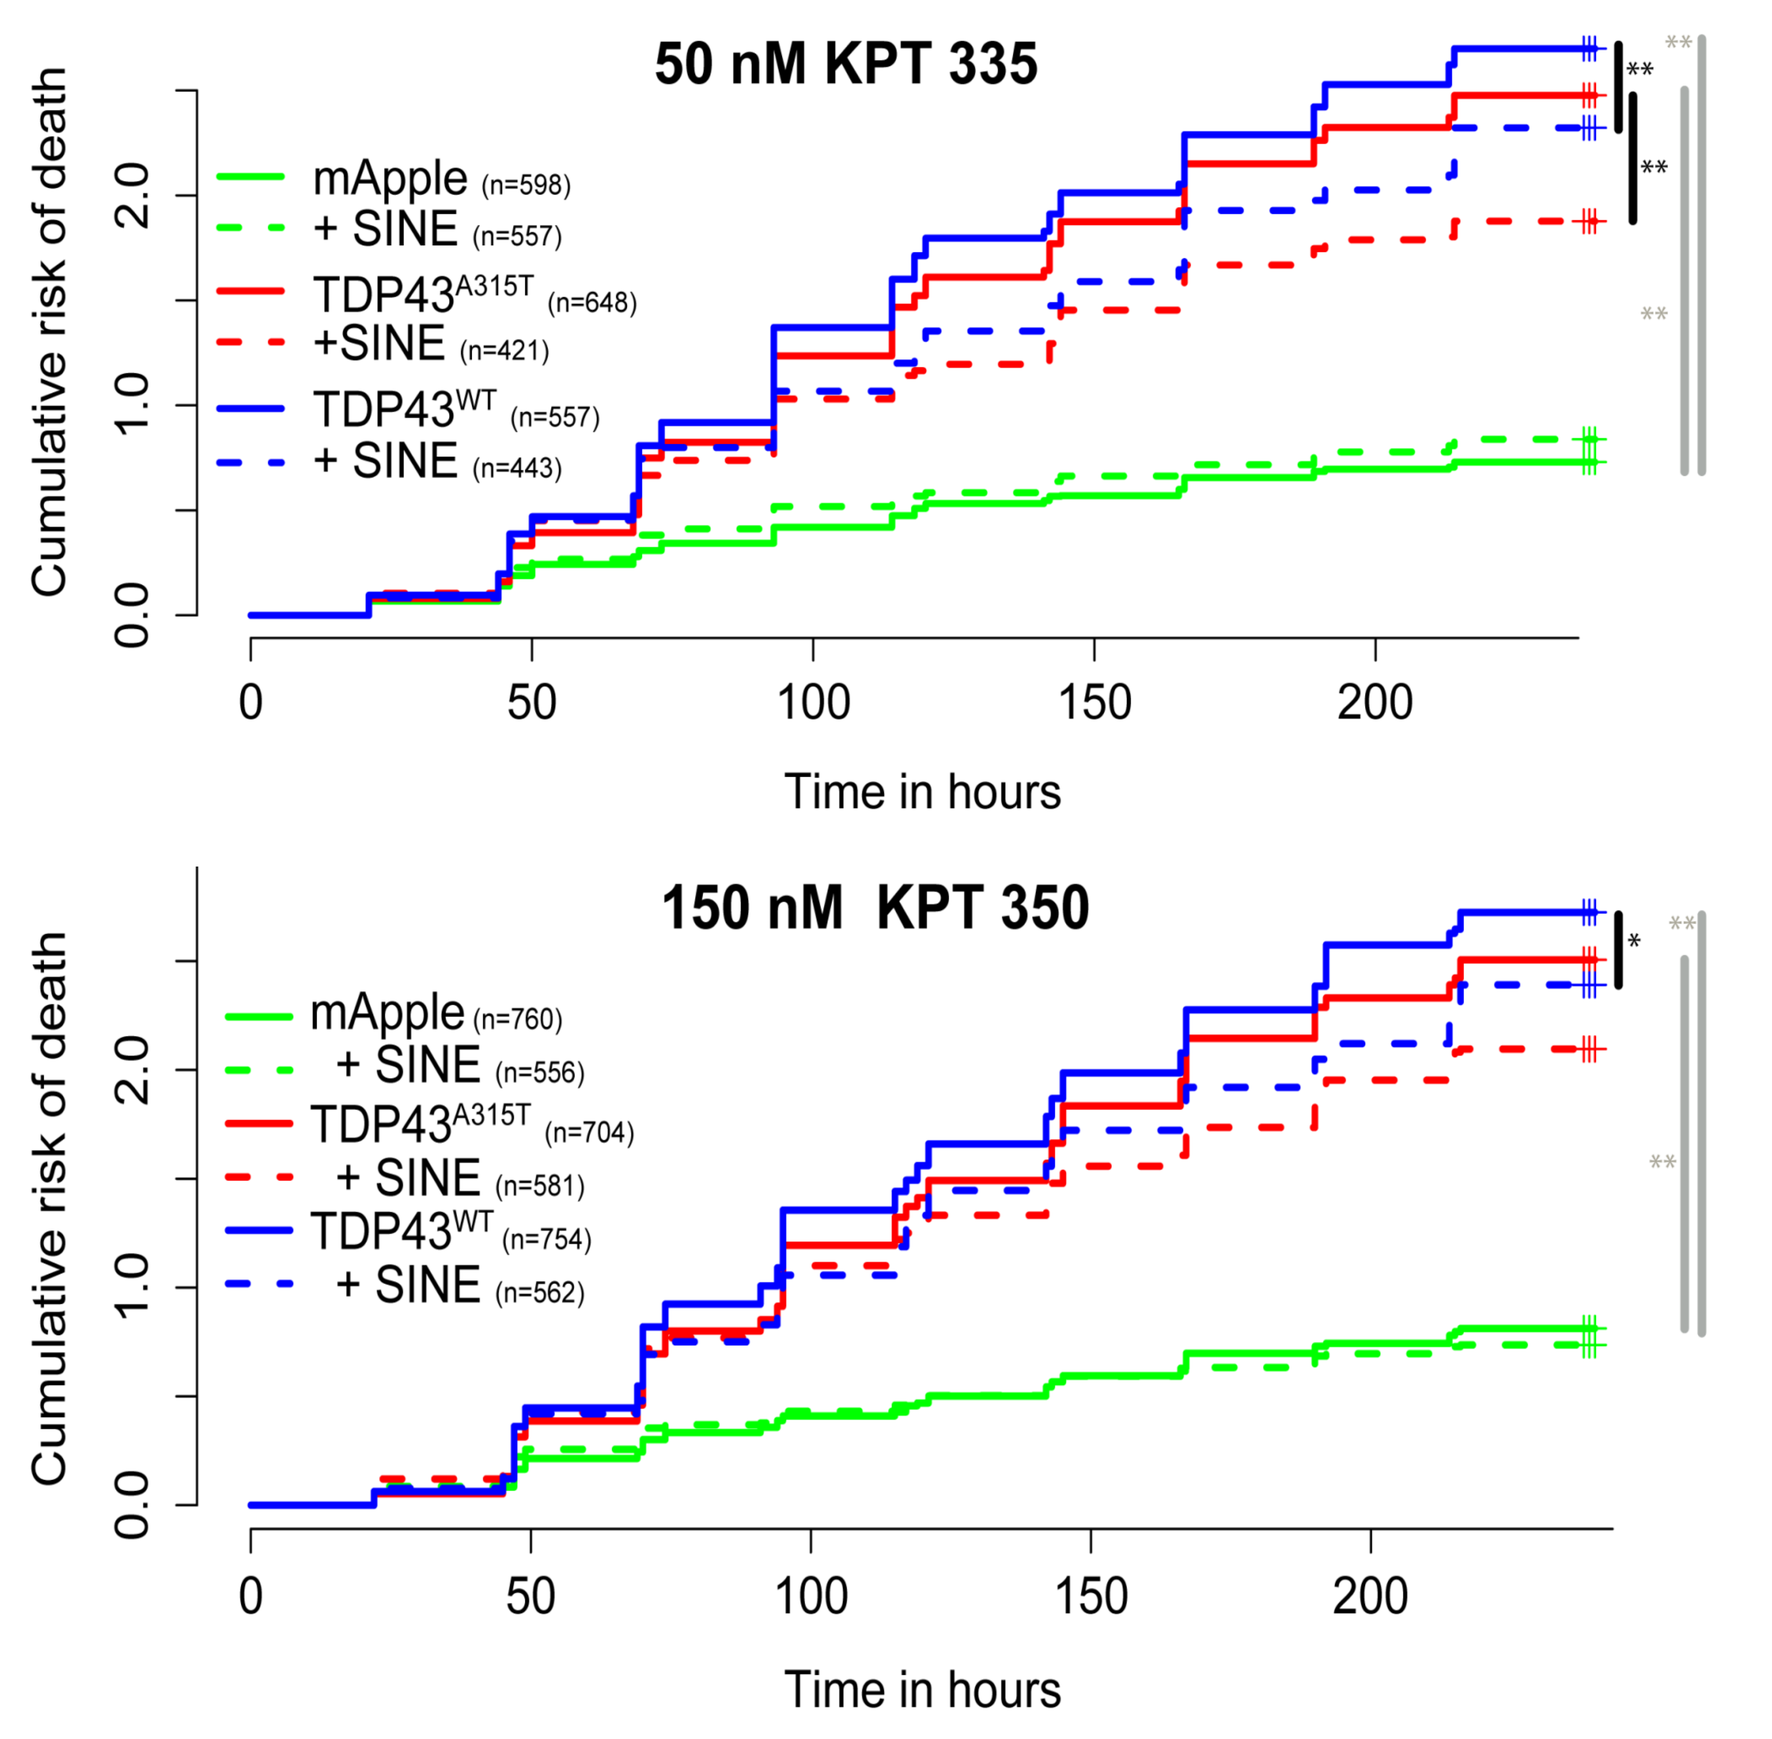
50 nM KPT 335
Cumulative risk of death
mApple (n=598)
+ SINE (n=557)
TDP43A315T (n=648)
+SINE (n=421)
TDP43WT (n=557)
+ SINE (n=443)
Time in hours
150 nM KPT 350
mApple (n=760)
+ SINE (n=556)
TDP43A315T (n=704)
+ SINE (n=581)
TDP43WT (n=754)
+ SINE (n=562)
Time in hours

(A and B) Overexpression of either mutant or WT TDP43 tagged with mApple, (red and blue lines, respectively) significantly increases the risk of death in our primary cortical neuron model of ALS/FTD when compared to control, shown in green (grey bars, ** = p<0.01). SINE compounds have no discernable effect on control neurons, panel A and B, dashed green line. However, **addition of SINE compounds has a significant neuroprotective effect in this model. (A)** Addition of 50 nM KPT 335 to neurons overexpressing WT TDP43-mApple (dashed blue line), or TDP43^{A315T}–mApple (dashed red line) decreased the probability of death (TDP43^{WT} Hazard Ratio (HR) = 0.8368, TDP43^{A315T} HR = 0.8310, ** = p<0.01). N = 24 wells/condition (3 expt, 8 wells each). **(B)** A similar effect is seen with KPT350 treatment, although the effect is significant only in the population expressing wild type TDP43 (TDP43^{WT} HR= 0.8633, * = p<0.05, TDP43^{A315T} HR=0.9218, p=0.17). N = 32 wells/condition (4 expt, 8 wells ea). We have found that the optimal dose for KPT350 is higher than for KPT335 (150 nM vs. 50 nM) (data not shown).

SINE compounds do not appear to alter the subcellular localization of endogenous TDP43

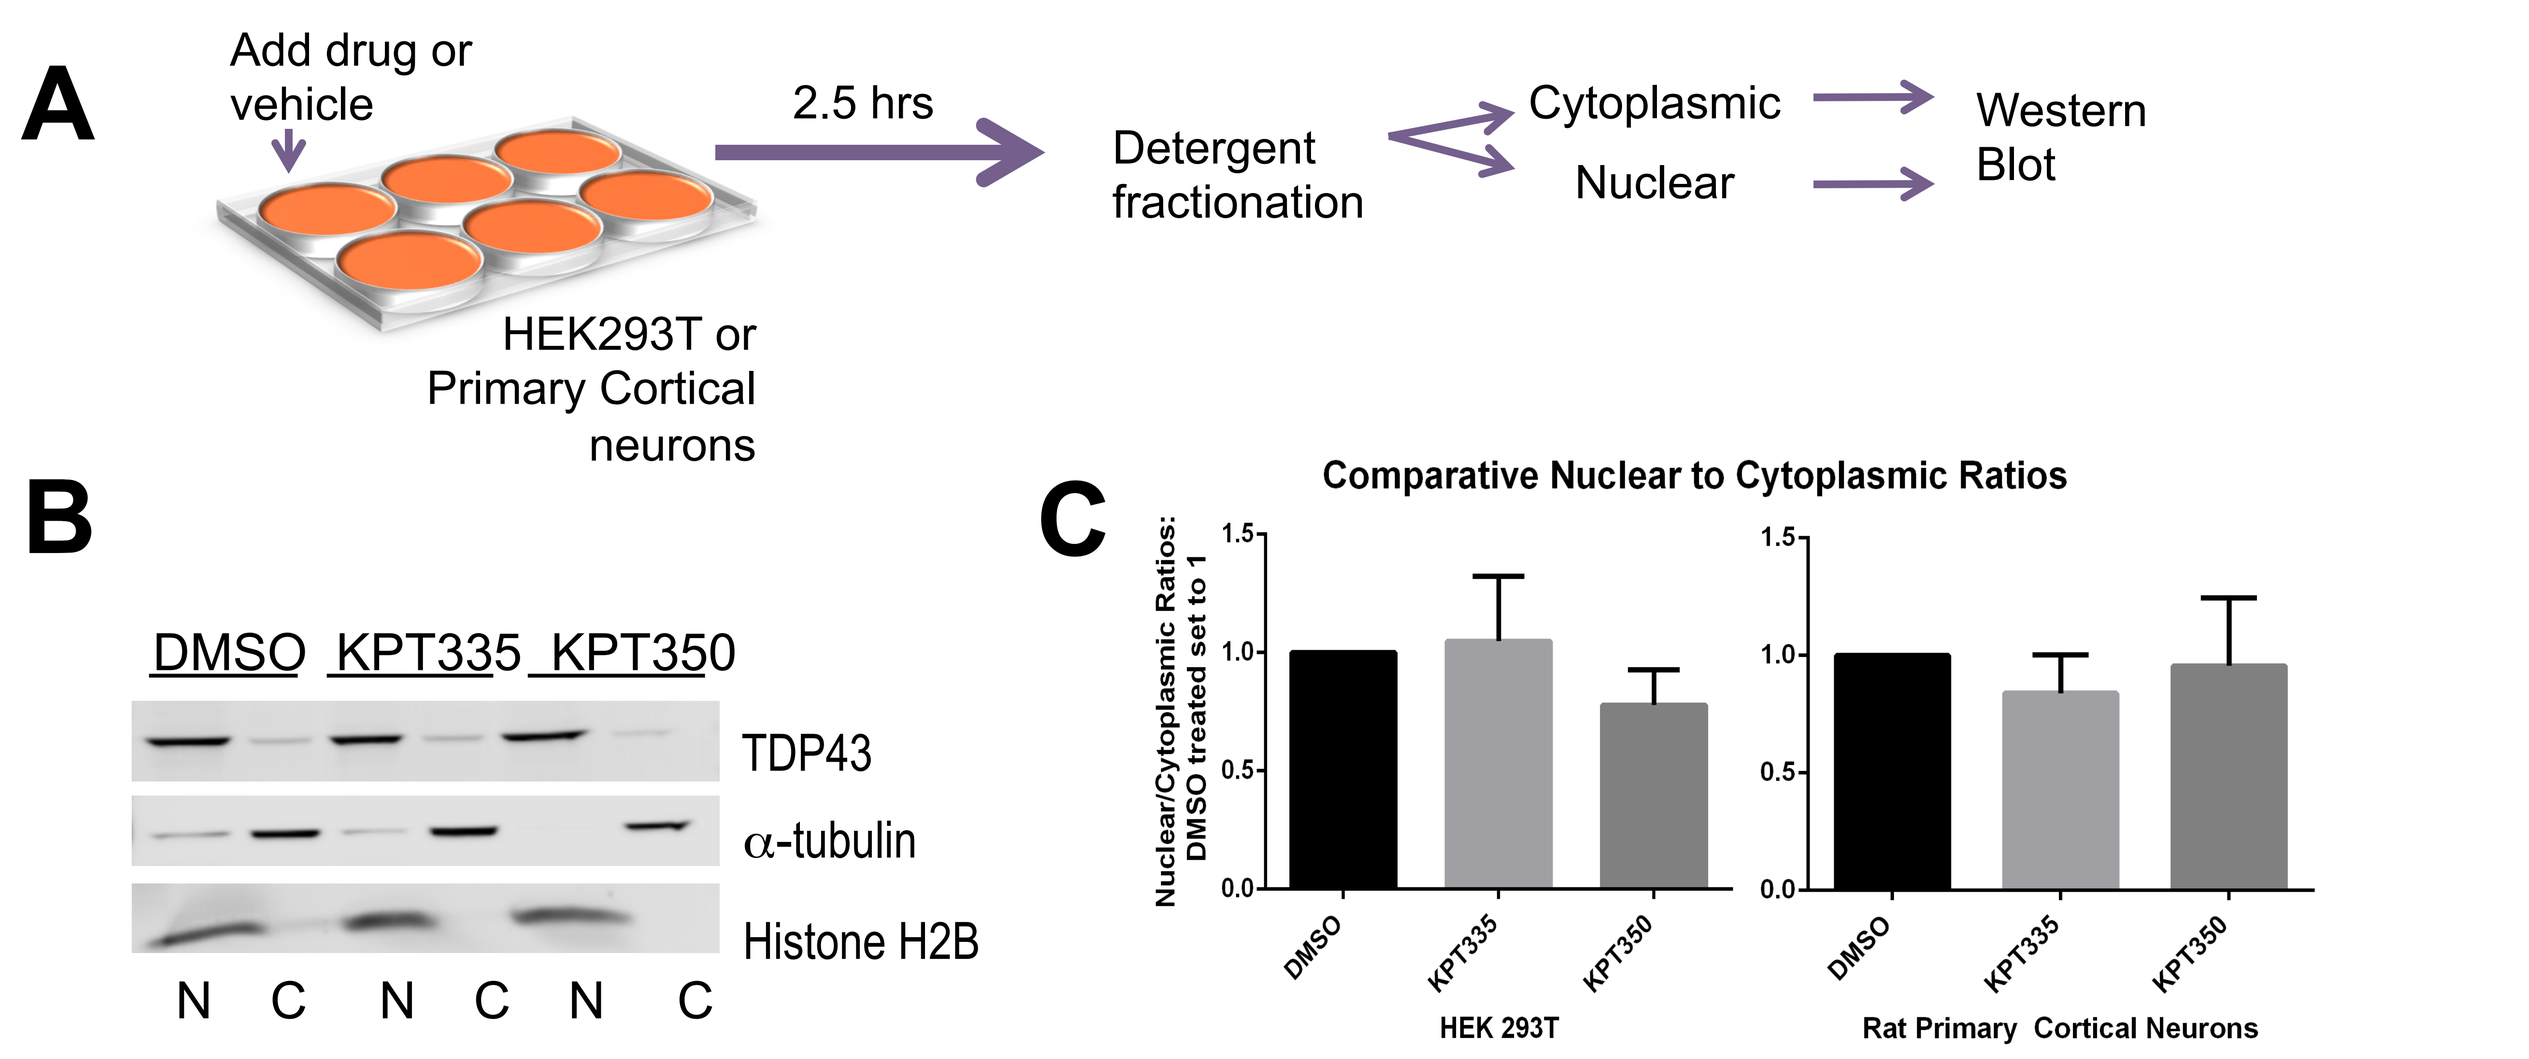
A
Add drug or vehicle
2.5 hrs
Detergent fractionation
Cytoplasmic
Nuclear
Western Blot
HEK293T or Primary Cortical neurons
B
DMSO KPT335 KPT350
TDP43
α-tubulin
Histone H2B
N C N C N C
C
Comparative Nuclear to Cytoplasmic Ratios
Nuclear/Cytoplasmic Ratios: DMSO treated set to 1
DMSO
KPT335
KPT350
HEK 293T
Rat Primary Cortical Neurons

(A) To assay the effect of SINE compounds on TDP43 subcellular localization, HEK293T cells or rat primary cortical neurons are treated with SINE compound or vehicle control (DMSO). 2.5 hours after treatment, the cells are harvested and subjected to a detergent fractionation. The nuclear and cytoplasmic fractions are probed for endogenous TDP43 by western blot, and visualized using a Licor infrared imaging system. **(B)** A representative blot of HEK293T cells. Nuclear and cytoplasmic fractions are indicated by N and C respectively. Histone H2B was used as a marker for the nuclear compartment, and α-tubulin for the cytoplasmic. In the control cells, TDP43 is predominantly nuclear, with a small amount of cytoplasmic TDP43, as expected. **We see no significant change in either nuclear or cytoplasmic levels of TDP43 in the presence of SINE compound,** and consequently, no change in nuclear/cytoplasmic ratios in HEK 293T cells or neurons **(C)**. SINE compounds are reported to be irreversibly bound to Xpo1 within 3 hours; however, we have not seen any reproducible differences in TDP43 localization from 1.5 to 46 hours post drug addition (data not shown).

Live imaging of exogenous TDP43 kinetics in response to SINE compounds shows no change in subcellular localization

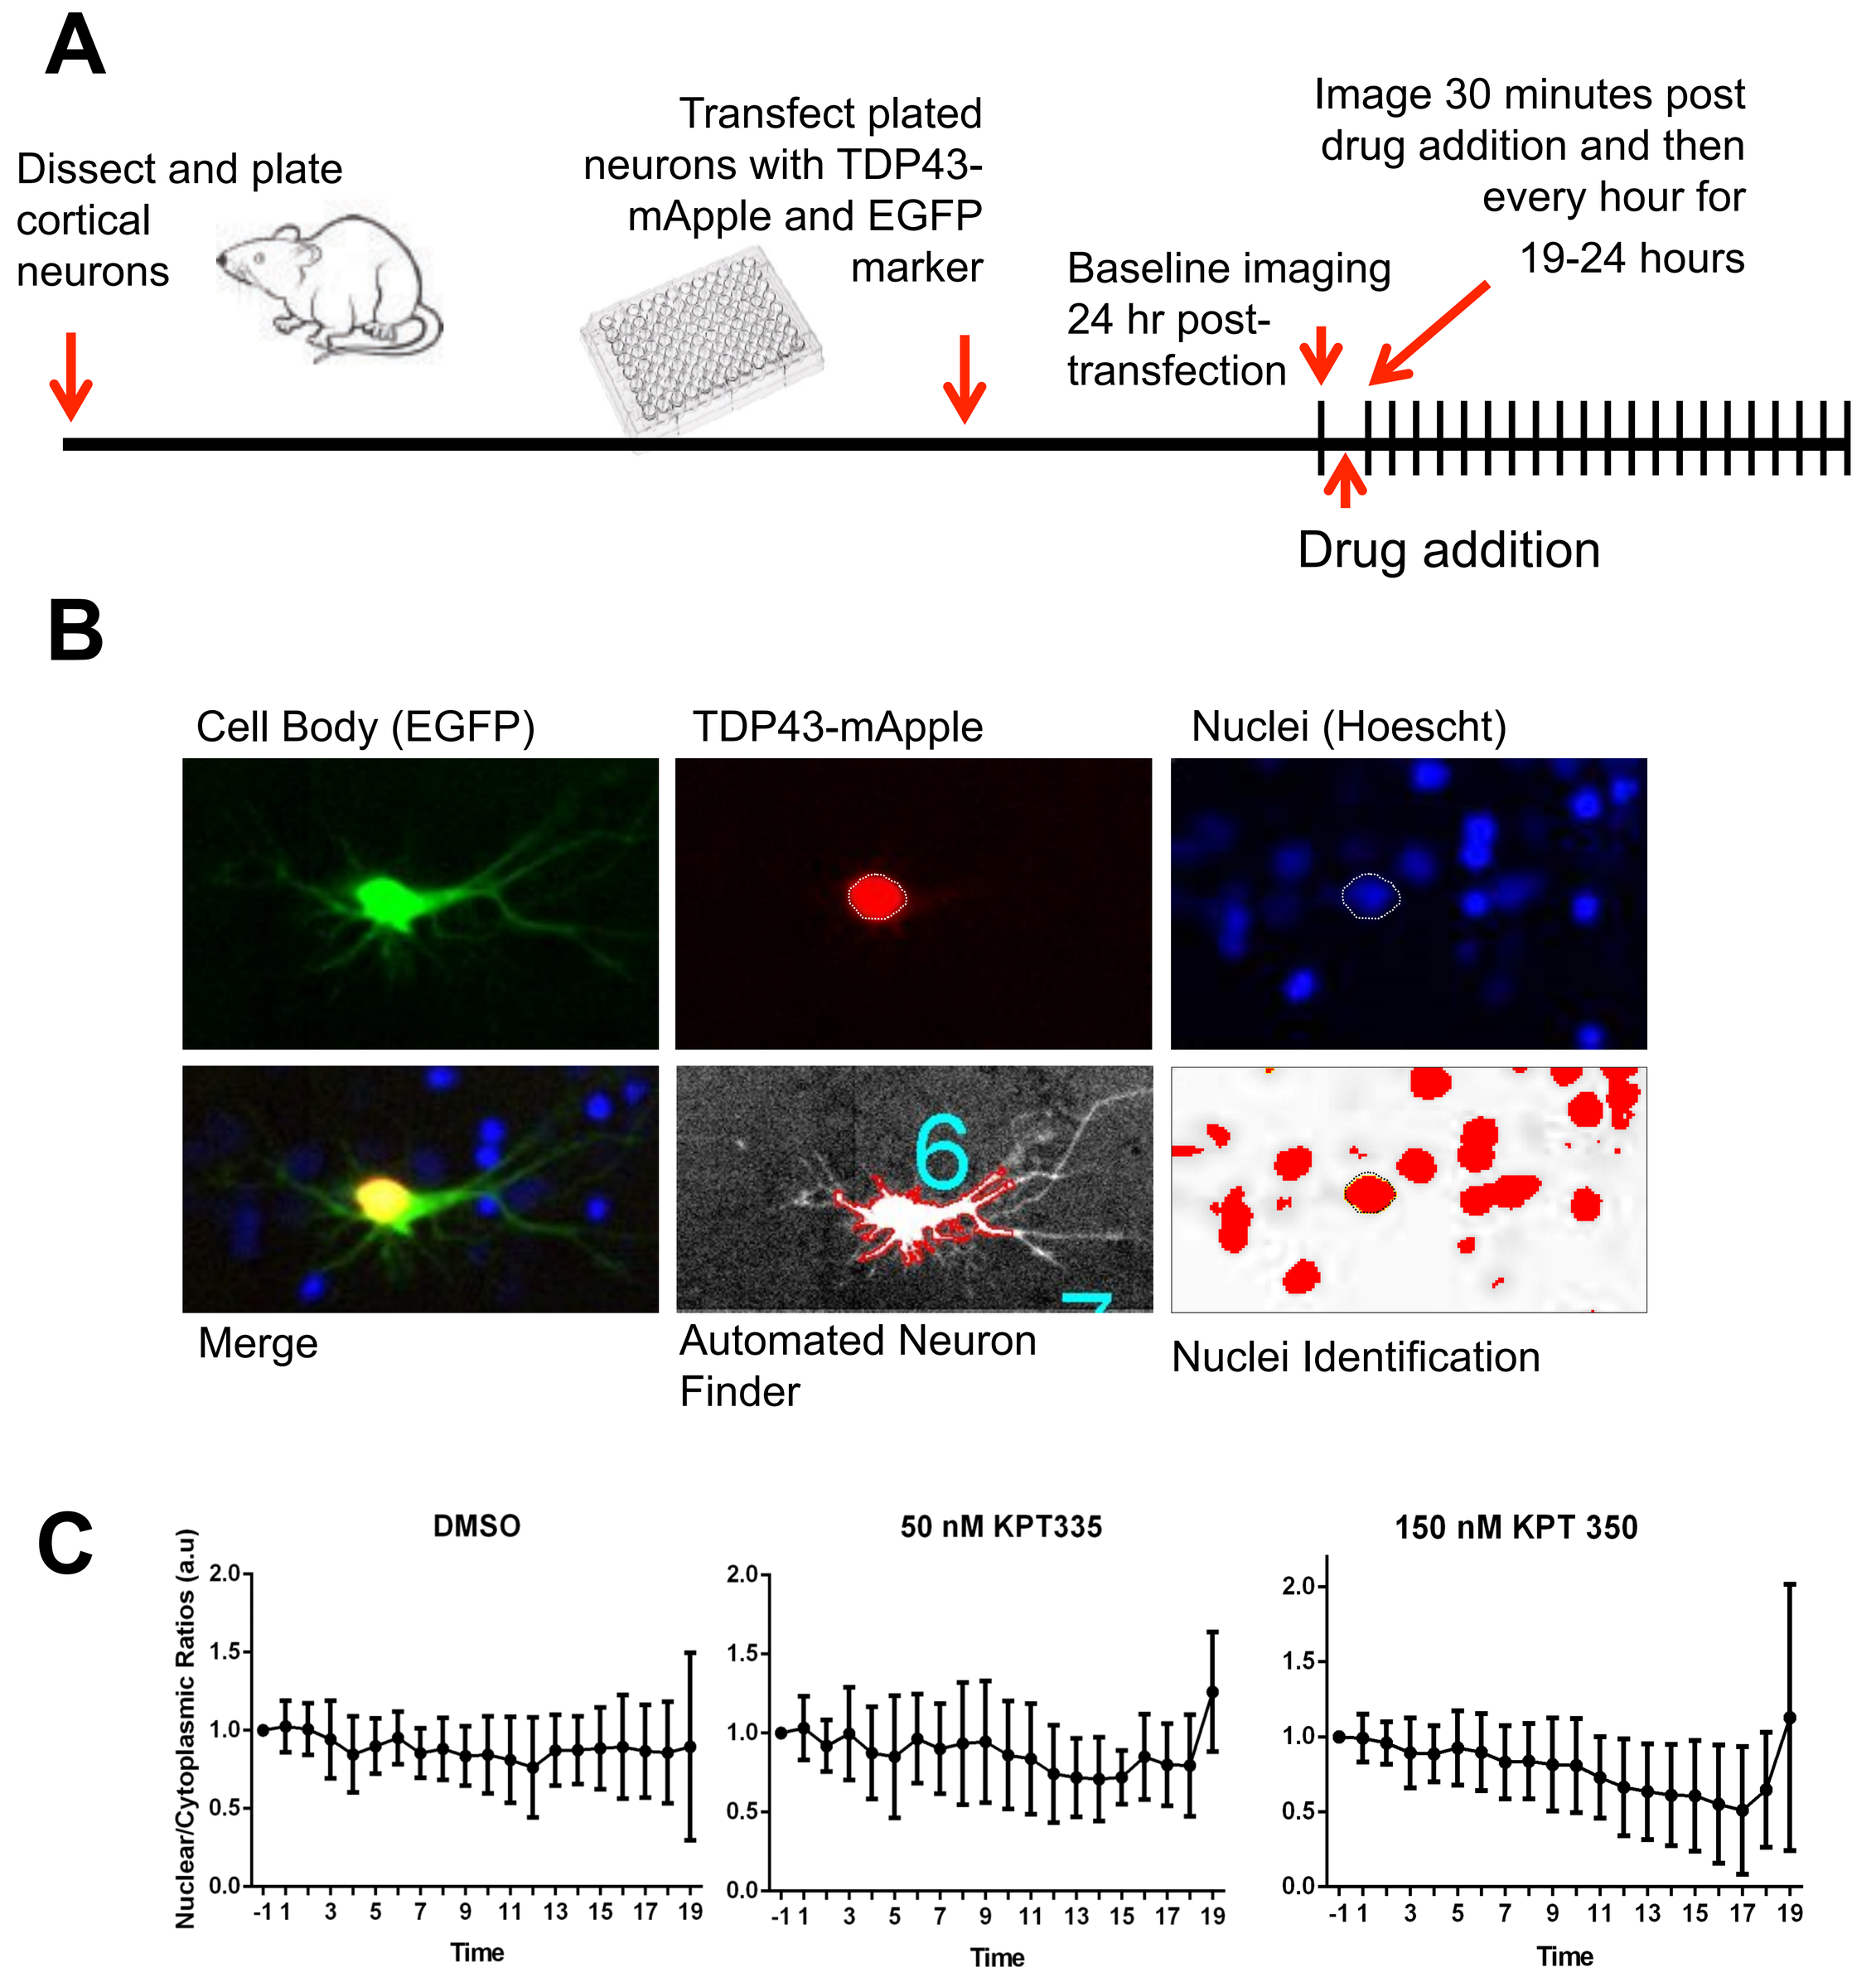
A
Dissect and plate cortical neurons
Transfect plated neurons with TDP43-mApple and EGFP marker
Baseline imaging 24 hr post-transfection
Image 30 minutes post drug addition and then every hour for 19-24 hours
Drug addition
B
Cell Body (EGFP)
TDP43-mApple
Nuclei (Hoescht)
Merge
Automated Neuron Finder
Nuclei Identification
C
DMSO
50 nM KPT335
150 nM KPT 350
Nuclear/Cytoplasmic Ratios (a.u)
Time

(A) Cortical neurons are transfected 4 days after dissection with TDP43-mApple fusion protein, and a ubiquitously expressed GFP to mark the cell body, while Hoechst stain is added to visualize nuclei. Drug or vehicle control is added after taking baseline images of the neurons, the treated wells are then imaged thirty minutes after drug addition, and then every hour for 19-24 hours. **(B)** Our automated analysis system identifies the cell bodies (red outline, bottom center panel) using the GFP marker, and identifies the nucleus based on threshold intensity images of the Hoechst staining. The signal from TDP-mApple is then partitioned into nuclear and cytoplasmic regions. **(C) No obvious effects on localization of exogenous TDP43 are seen.** Single representative experiment (of three biological replicates) shown, (N=15-20 neurons per condition).

How do SINE compounds exert their protective effect?

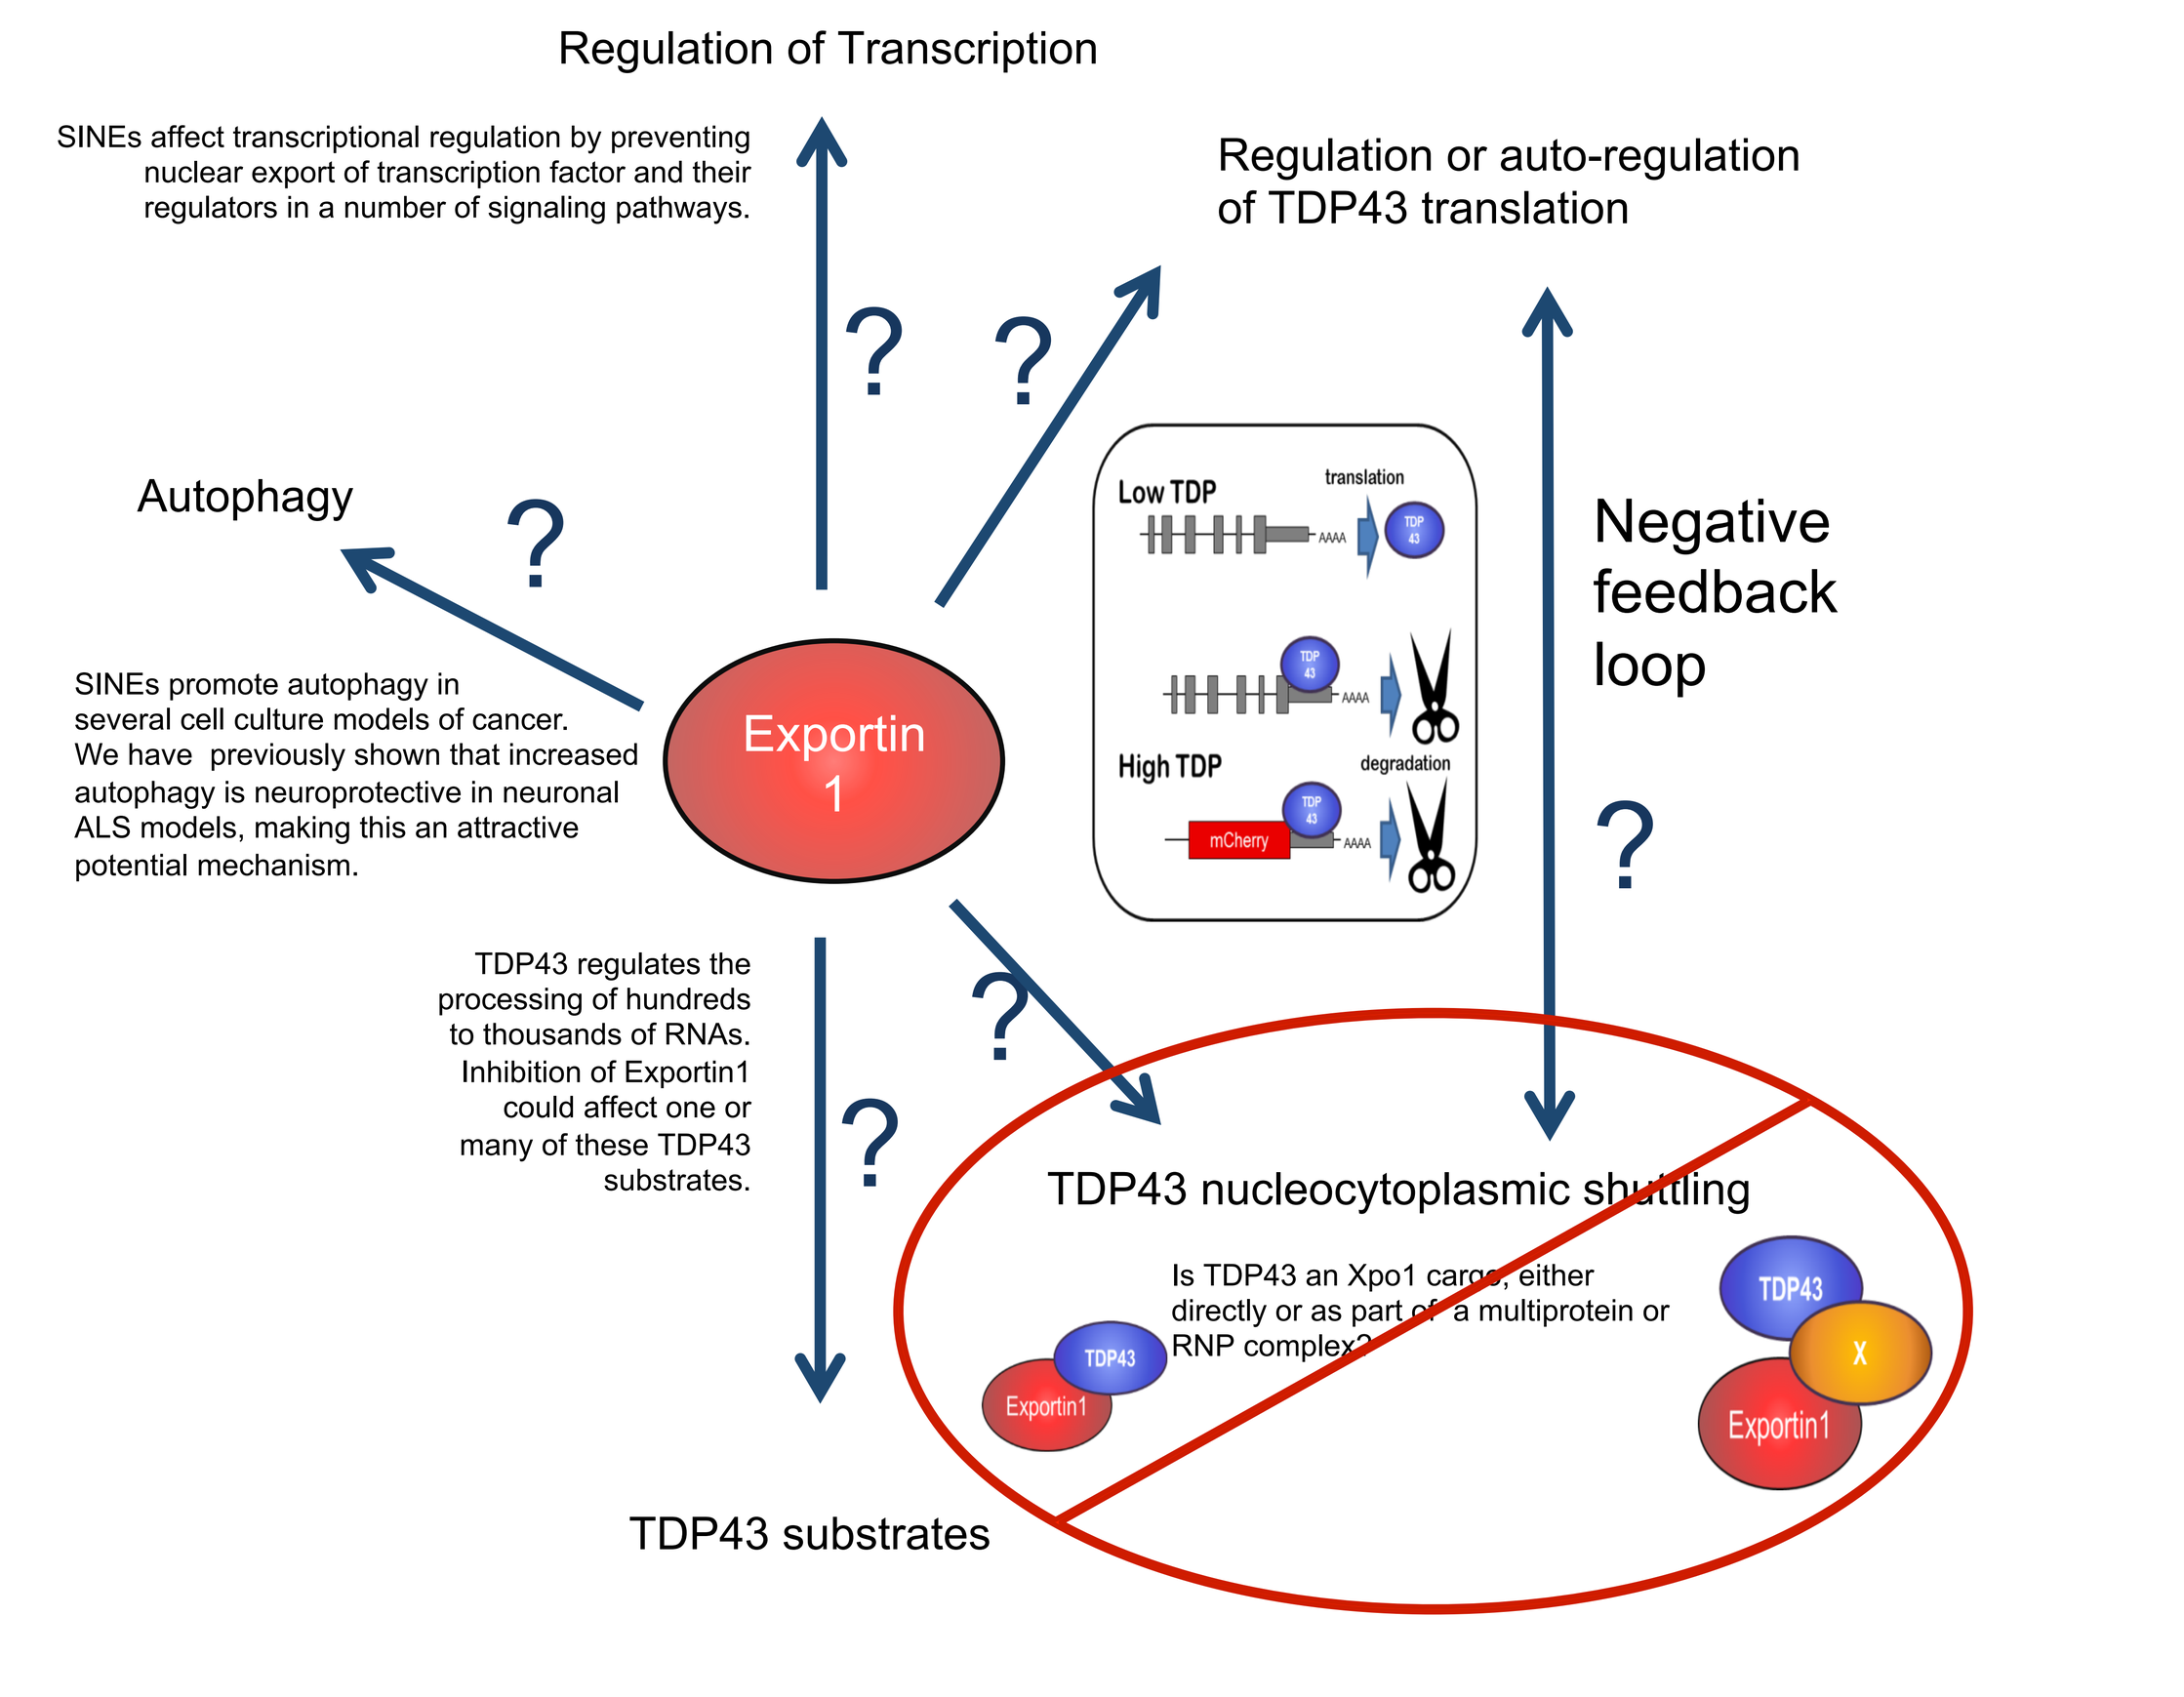
Regulation of Transcription
SINEs affect transcriptional regulation by preventing nuclear export of transcription factor and their regulators in a number of signaling pathways.
Regulation or auto-regulation of TDP43 translation
Autophagy
SINEs promote autophagy in several cell culture models of cancer. We have previously shown that increased autophagy is neuroprotective in neuronal ALS models, making this an attractive potential mechanism.
Exportin 1
Low TDP
High TDP
translation
degradation
mCherry
Negative feedback loop
TDP43 regulates the processing of hundreds to thousands of RNAs. Inhibition of Exportin1 could affect one or many of these TDP43 substrates.
TDP43 nucleocytoplasmic shuttling
Is TDP43 an Xpo1 cargo, either directly or as part of a multiprotein or RNP complex?
TDP43
Exportin1
X
TDP43 substrates

Although nuclear sequestration of TDP43 by SINE compounds was an attractive hypothesis, we have seen no evidence of increased nuclear TDP43 levels after treatment using a variety of experimental methods. Therefore, **TDP43 is unlikely to be a cargo of Exportin1**. A potentially confounding issue is that TDP43 binds to its own mRNA, and negatively regulates translation by targeting the mRNA for degradation. Therefore, it is possible that nuclear sequestration of TDP43 could promote this negative feedback loop, resulting in overall lower TDP43 levels, rather than increasing nuclear localization, as predicted. It is also possible that Xpo1 inhibition acts to regulate TDP43 transcription or translation, without interacting directly with TDP43. However, based on our experimental evidence, it seems more likely that the beneficial effect of SINE compounds is TDP43-independent, perhaps by acting on a downstream target of TDP43, or by inducing broadly neuroprotective factors. **Given that related SINE compounds are currently undergoing evaluation in clinical trials for other indications, we feel that these SINE compounds represent a promising potential therapy. Identifying their mode of action is an important step in determining their potential to combat neurodegeneration.** Therefore, current research is aimed at identifying the cellular processes that may be targeted by SINE compounds.

SINE compounds are broadly neuroprotective

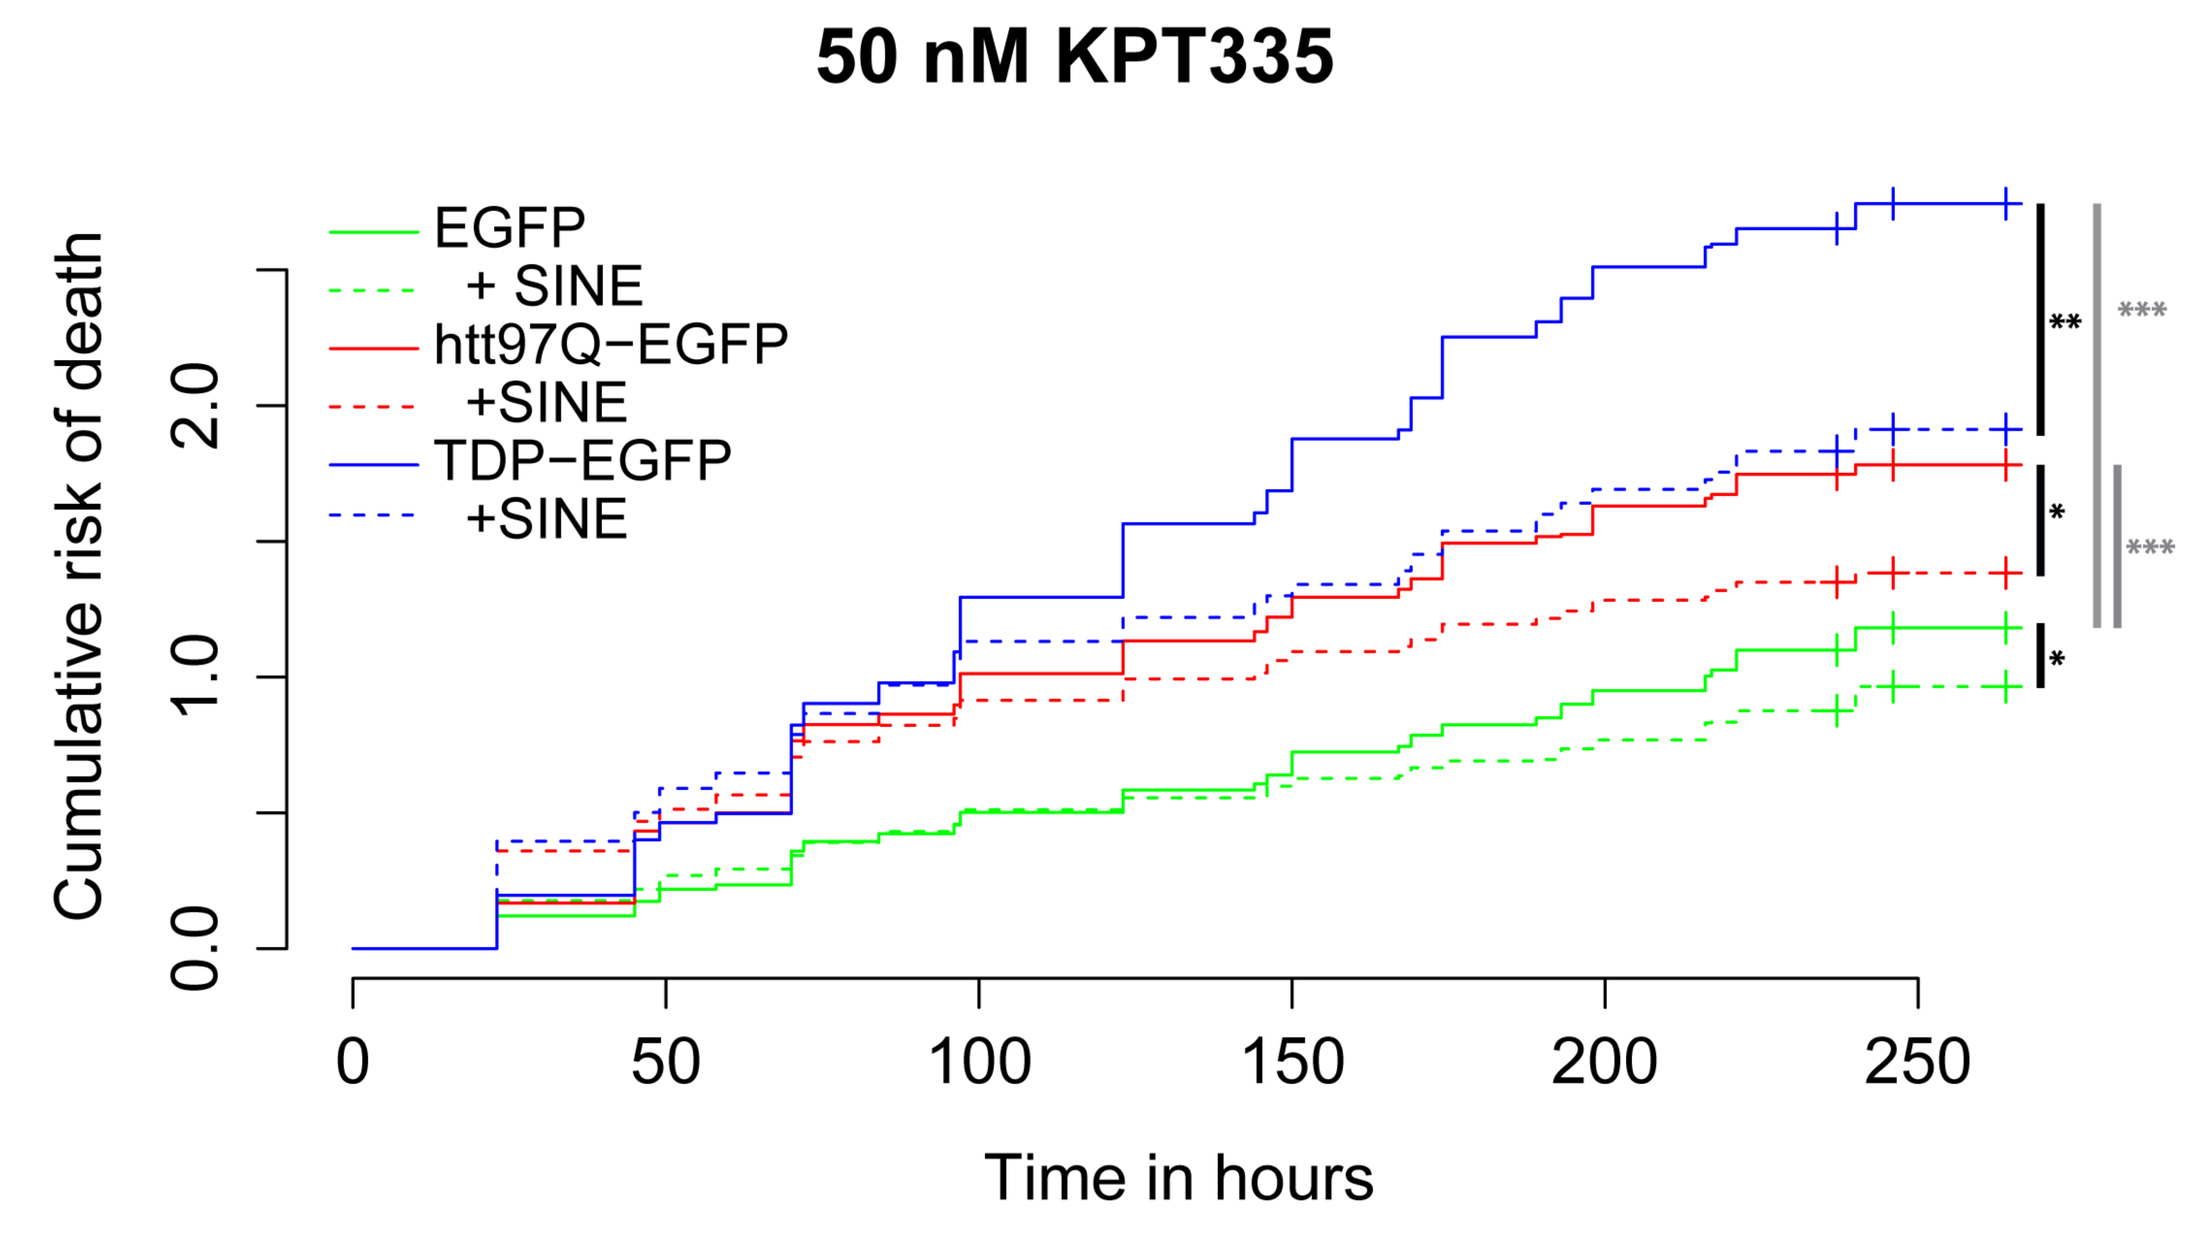
50 nM KPT335
Cumulative risk of death
EGFP
+ SINE
htt97Q–EGFP
+SINE
TDP–EGFP
+SINE
Time in hours

SINE compounds decrease the risk of death in response to multiple neuronal stressors. Overexpression of a fragment of the huntingtin protein containing a disease associated repeat expansion (htt97Q) is significantly toxic to cultured rodent primary cortical neurons (HR = 1.68), although not to the same extent as wildtype TDP-43 overexpression (HR = 2.22) (gray bars, ***p< 0.001). A single 50 nM dose of KPT335 significantly decreases risk of death in both models of neurodegeneration (HR$^{htt+SINE}$ = 1.46, HR$^{tdp+SINE}$ = 1.91) (black bars, ** p<0.01, * p<0.05) N= 40 wells/condition (Combined results of 3 experiments, 12-16 wells each).

SINE compounds activate autophagy

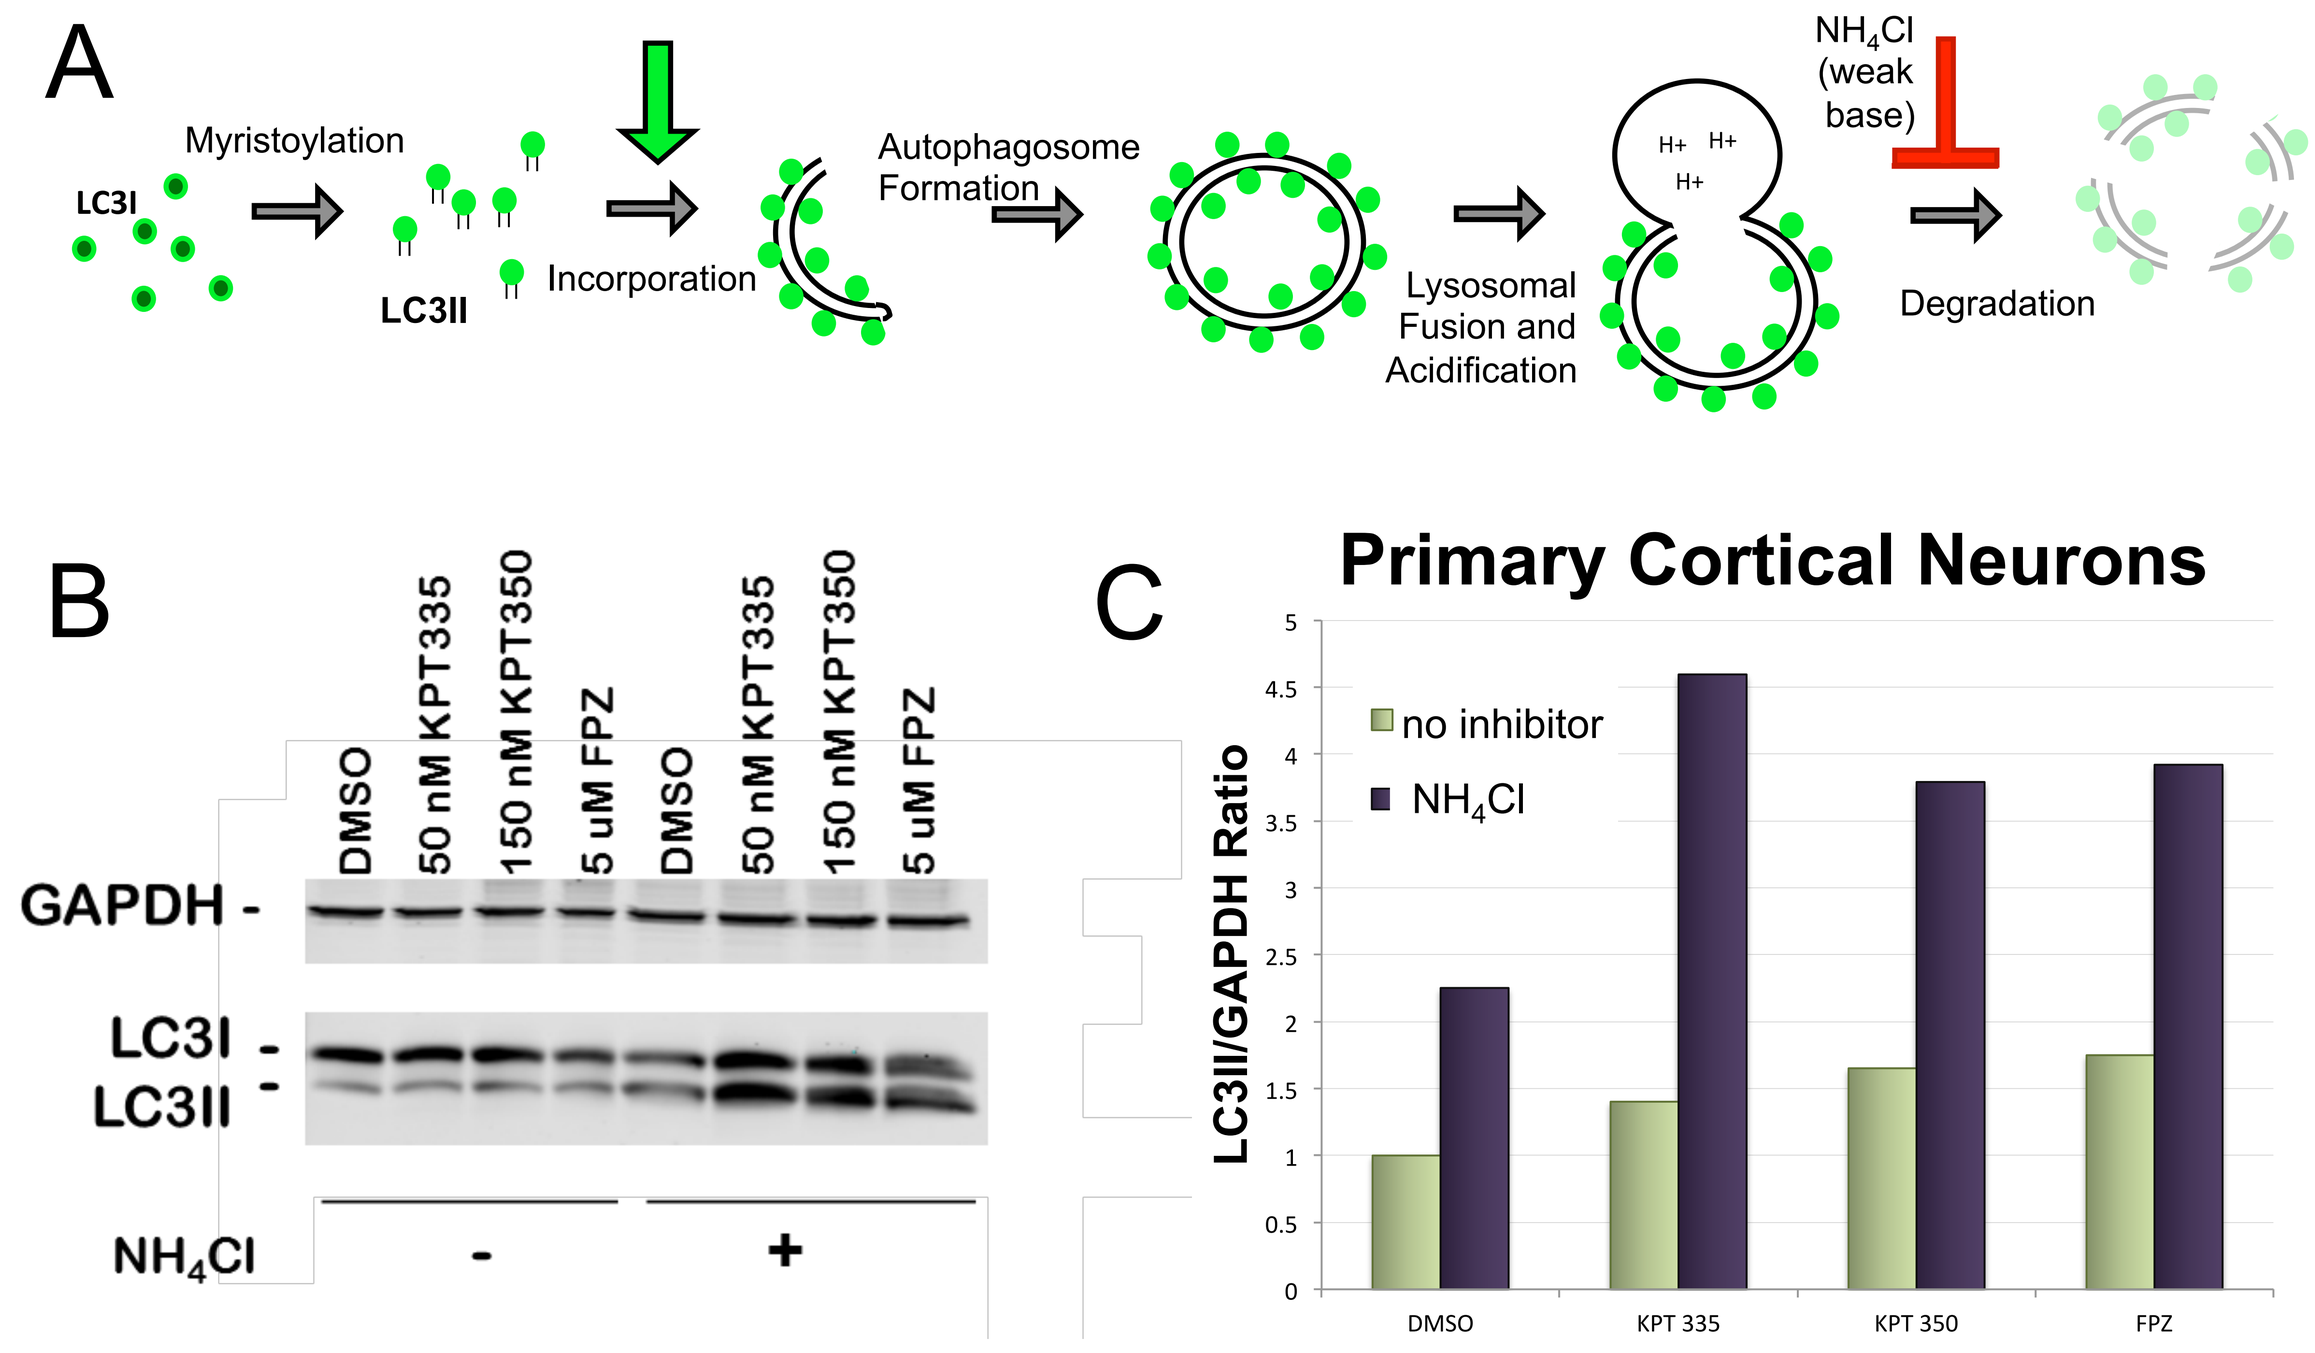
A
LC3I
Myristoylation
LC3II
Incorporation
Autophagosome Formation
Lysosomal Fusion and Acidification
NH4Cl (weak base)
Degradation
B
DMSO
50 nM KPT335
150 nM KPT350
5 uM FPZ
GAPDH
LC3I
LC3II
NH4Cl
C
Primary Cortical Neurons
LC3II/GAPDH Ratio
no inhibitor
NH4Cl
DMSO
KPT 335
KPT 350
FPZ

SINE compounds may exert a neuroprotective effect by activating autophagy. (**A**) LC3II levels are used to experimentally measure increases in autophagic activity, as it is both a component and a substrate of autophagic vesicles (AV). Increases in LC3II levels can be seen when vesicle formation is increased (green arrow), or when maturation and/or degradation of autophagolysosomes is blocked (in this example, by application of a weak base, NH_4Cl, red symbol). (**B**) Rat primary cortical neurons were treated with the indicated dose of vehicle control (DMSO), SINE compound, or fluphenazine (FPZ) — a potent neuronal autophagy inducer (Barmada *et al. Nat Chem. Biol.*, 2015) — on day 6 post dissection. A second dose was added 21 hours later, with or without 100 mM NH_4Cl, and after three hours cells were lysed and subjected to western blot analysis using LC3 and GAPDH antibodies. (**C**) Quantification of blot shown in (A), which is representative of a single trial of KPT350, and two trials of vehicle control, KPT335, and FPZ. LC3II levels were normalized to GAPDH, used as a loading control. An increase in LC3II levels in both the absence and presence of NH_4Cl indicates that the SINE compounds are promoting the formation of AV, rather than preventing their degradation.

SINE compounds and transcriptional regulation

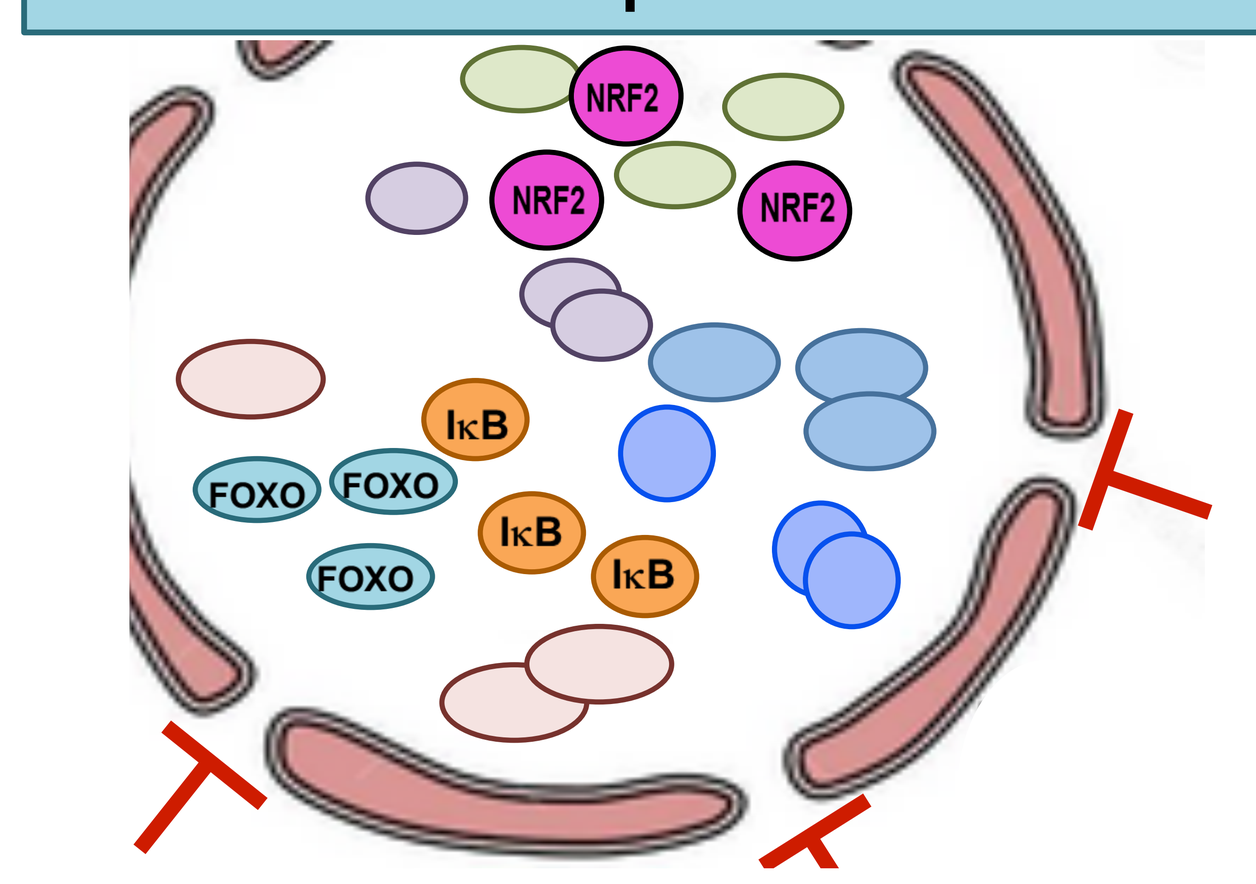
NRF2
IκB
FOXO

SINE mediated inhibition of Xpo1 leads to nuclear retention of multple transcription factors and transcriptional regulators

Inhibition of Xpol by SINE compounds results in nuclear retention of multiple transcription factors including AP-1, FoxO, p53, IκB, APC, PPARγ, RXRγ, and NRF2. We are particularly interested in factors which have been linked to neuroprotection in other model systems, including **FOXO** (downstream of the AKT/PI3K pathway) (Milosevic-Petrovic et al., 2014), the anti-oxidant response pathway factor **NRF2** (Zhang et al., 2013), and the NF-κB inflammatory response (Haines et al. 2015). Interestingly, the NF-κB inflammatory response is *inhibited* by the nuclear retention of **IκB** caused by SINE compounds, and pathological TDP43 has been linked to activation of the NF-κB response (Swarup et al. 2013). Also interesting is the link between FOXO activity and upregulation of autophagy (Warr et al., 2013). Future experiments will utilize reporter constructs to identify transcriptional pathways which are perturbed by SINE compounds in our model system, with the ultimate goal of identifying additional mediators of the neuroprotective effect seen in our model.

Acknowledgements

The Barmada Lab:
Sami Barmada, MD, PhD
Elizabeth Tank, PhD
Xingli Li, MD
Roberto Miguez
Brittany Flores
Kate Weskamp
Nathaniel Safren, PhD
Alex Bennett

Postdoctoral training committee:
Sami Barmada, MD, PhD
Peter Todd MD, PhD
Aaron Goldstrohm, PhD

UM Neurology Dept:
Denice Heckel, Howard Oishi
The Feldman, Shakkotai, Zou, Todd, Paulson, Dauer, and Parent Labs

LSU:
Klein Lab

Karyopharm Therapeutics, Inc.

Funding:
The ALS Therapy Alliance
NIH NINDS 1K08NS072233-01A1
Association for Frontotemporal Degeneration
UM Dept of Neurology Training Grant 5T32NS007222-33