Phase I Trial of Selinexor (KPT-330), a First-In-Class Oral Selective Inhibitor of Nuclear Export (SINE) in Patients with Advanced Acute Myeloblastic Leukemia (AML)

Savona, M[1], Garzon R[2], Brown, P[3], Yee, K[4], Lancet, JE[5], Gutierrez, M[6], Gabrail, N[7], Mau-Sorensen, M[3], Baz, R[6], Byrd, JC[2], Kuruvilla, J[4], Siegel, DS[6], Shacham, S[8], Rashal, T[8], Yau, CYF[9], McCauley, D[8], Saint-Martin JR[8], McCartney, J[8], Landesman, Y[8], Klebanov B[8], Kashyap, T[8], Shacham, E[8], Pond, G[9], Oza, A[9], Kauffman, MG[8], Mirza, MR[3] and Stone, RM[10]

[1]Sarah Cannon Research Institute, Nashville, TN; [2]The Ohio State University, Columbus, OH; [3]Rigshospitalet, Copenhagen, Denmark; [4]Princess Margaret Hospital, Toronto, ON, Canada; [5]H. Lee Moffitt Cancer Center and Research Institute, Tampa, FL; [6]John Theurer Cancer Center, Hackensack, NJ; [7]Gabrail Cancer Center, Canton, OH; [8]Karyopharm Therapeutics Inc, Natick, MA; [9]Ozmosis Research Inc, Toronto, ON, Canada; [10]Dana Farber Cancer Institute, Boston, MA

ABSTRACT

Background: AML cells overexpress the nuclear exporter, Exportin 1 (XPO1/CRM1) and higher XPO1 levels correlate with poor outcome. The novel Selective Inhibitor of Nuclear Export (SINE), Selinexor (KPT-330), antagonizes XPO1 and shows potent cytotoxicity for AML and acute lymphoblastic leukemia (ALL) cells *in vitro*, independent of genotype. Mechanistic studies show that SINE induces nuclear localization and activation of multiple tumor suppressor proteins (TSP), leading to rapid apoptosis of AML cells, while sparing normal hematopoietic cells.

Methods: Patients (pts) with relapsed and refractory AML were dosed with oral KPT-330 (8-10 doses / 4-week cycle) in one arm of a Phase 1 trial in advanced hematological malignancies (NCT 01607892). Dosing was continued until progression or intolerance. Standard dose limiting toxicity (DLT) definition was used. Detailed pharmacokinetic (PK) and pharmacodynamic (PDn) analyses were done. Response evaluation was performed every cycle.

Results: 39 AML pts were enrolled in 8 sites in the US, Canada, and Denmark. Median age 68 yrs; ECOG PS 0/1: 9/30; median number of prior regimens: 2.6 [range 0-7]. Two patients had not received treatment specifically for AML, but had received hypomethylating agents and other therapy for myelodysplastic syndrome (MDS) prior to transformation to AML, and enrollment on the trial. Patients received KPT-330 across 5 dose levels (16.8 to 55 mg/m^2, with additional cohorts ongoing). There has been no DLT. Patients experienced drug-related grade 3/4 non-hematological Adverse Events (AEs) including anorexia (n=1), nausea (n=2), fatigue (n=3), vomiting (n=1), dehydration (n=1), hypotension (n=1), AST increased (n=1), and hypokalemia (n=2). The most common grade 1/2 toxicities were: nausea (20/38; 53%), anorexia (16/38; 42%), fatigue (15/38; 39%), vomiting (12/38; 31.5%), diarrhea (12/38; 31.5%), and weight loss (10/38; 26%). These AEs were manageable with supportive care. There were no clinically significant cumulative toxicities or major organ dysfunction. PK analysis demonstrated a proportional increase in C_{max} and AUC with increasing dose with no accumulation. Half-life was ~6-7 hours with rapid clearance of KPT-330. Of 33 pts who were evaluable for response, complete response (CR) with full hematological recovery was achieved in 4 patients (11%), and CR without hematological recovery (CRi) in 1 patient (3%). Partial Response (PR) was achieved in 2 patients (6%). Morphological leukemia free state was achieved in 1 patient (3%). Ten (29%) of the remaining patients have had stable disease for > 30 days, and 13 (34%) have had progressive disease.

Conclusions: Single agent oral KPT-330 treatment is generally well tolerated and has a favorable PK profile. Remissions and prolonged stable disease have been observed at doses below the maxium tolerated dose (MTD), and dose escalation is ongoing with current cohort being dosed at 55 mg/m^2 twice weekly.

Mechanism of Action

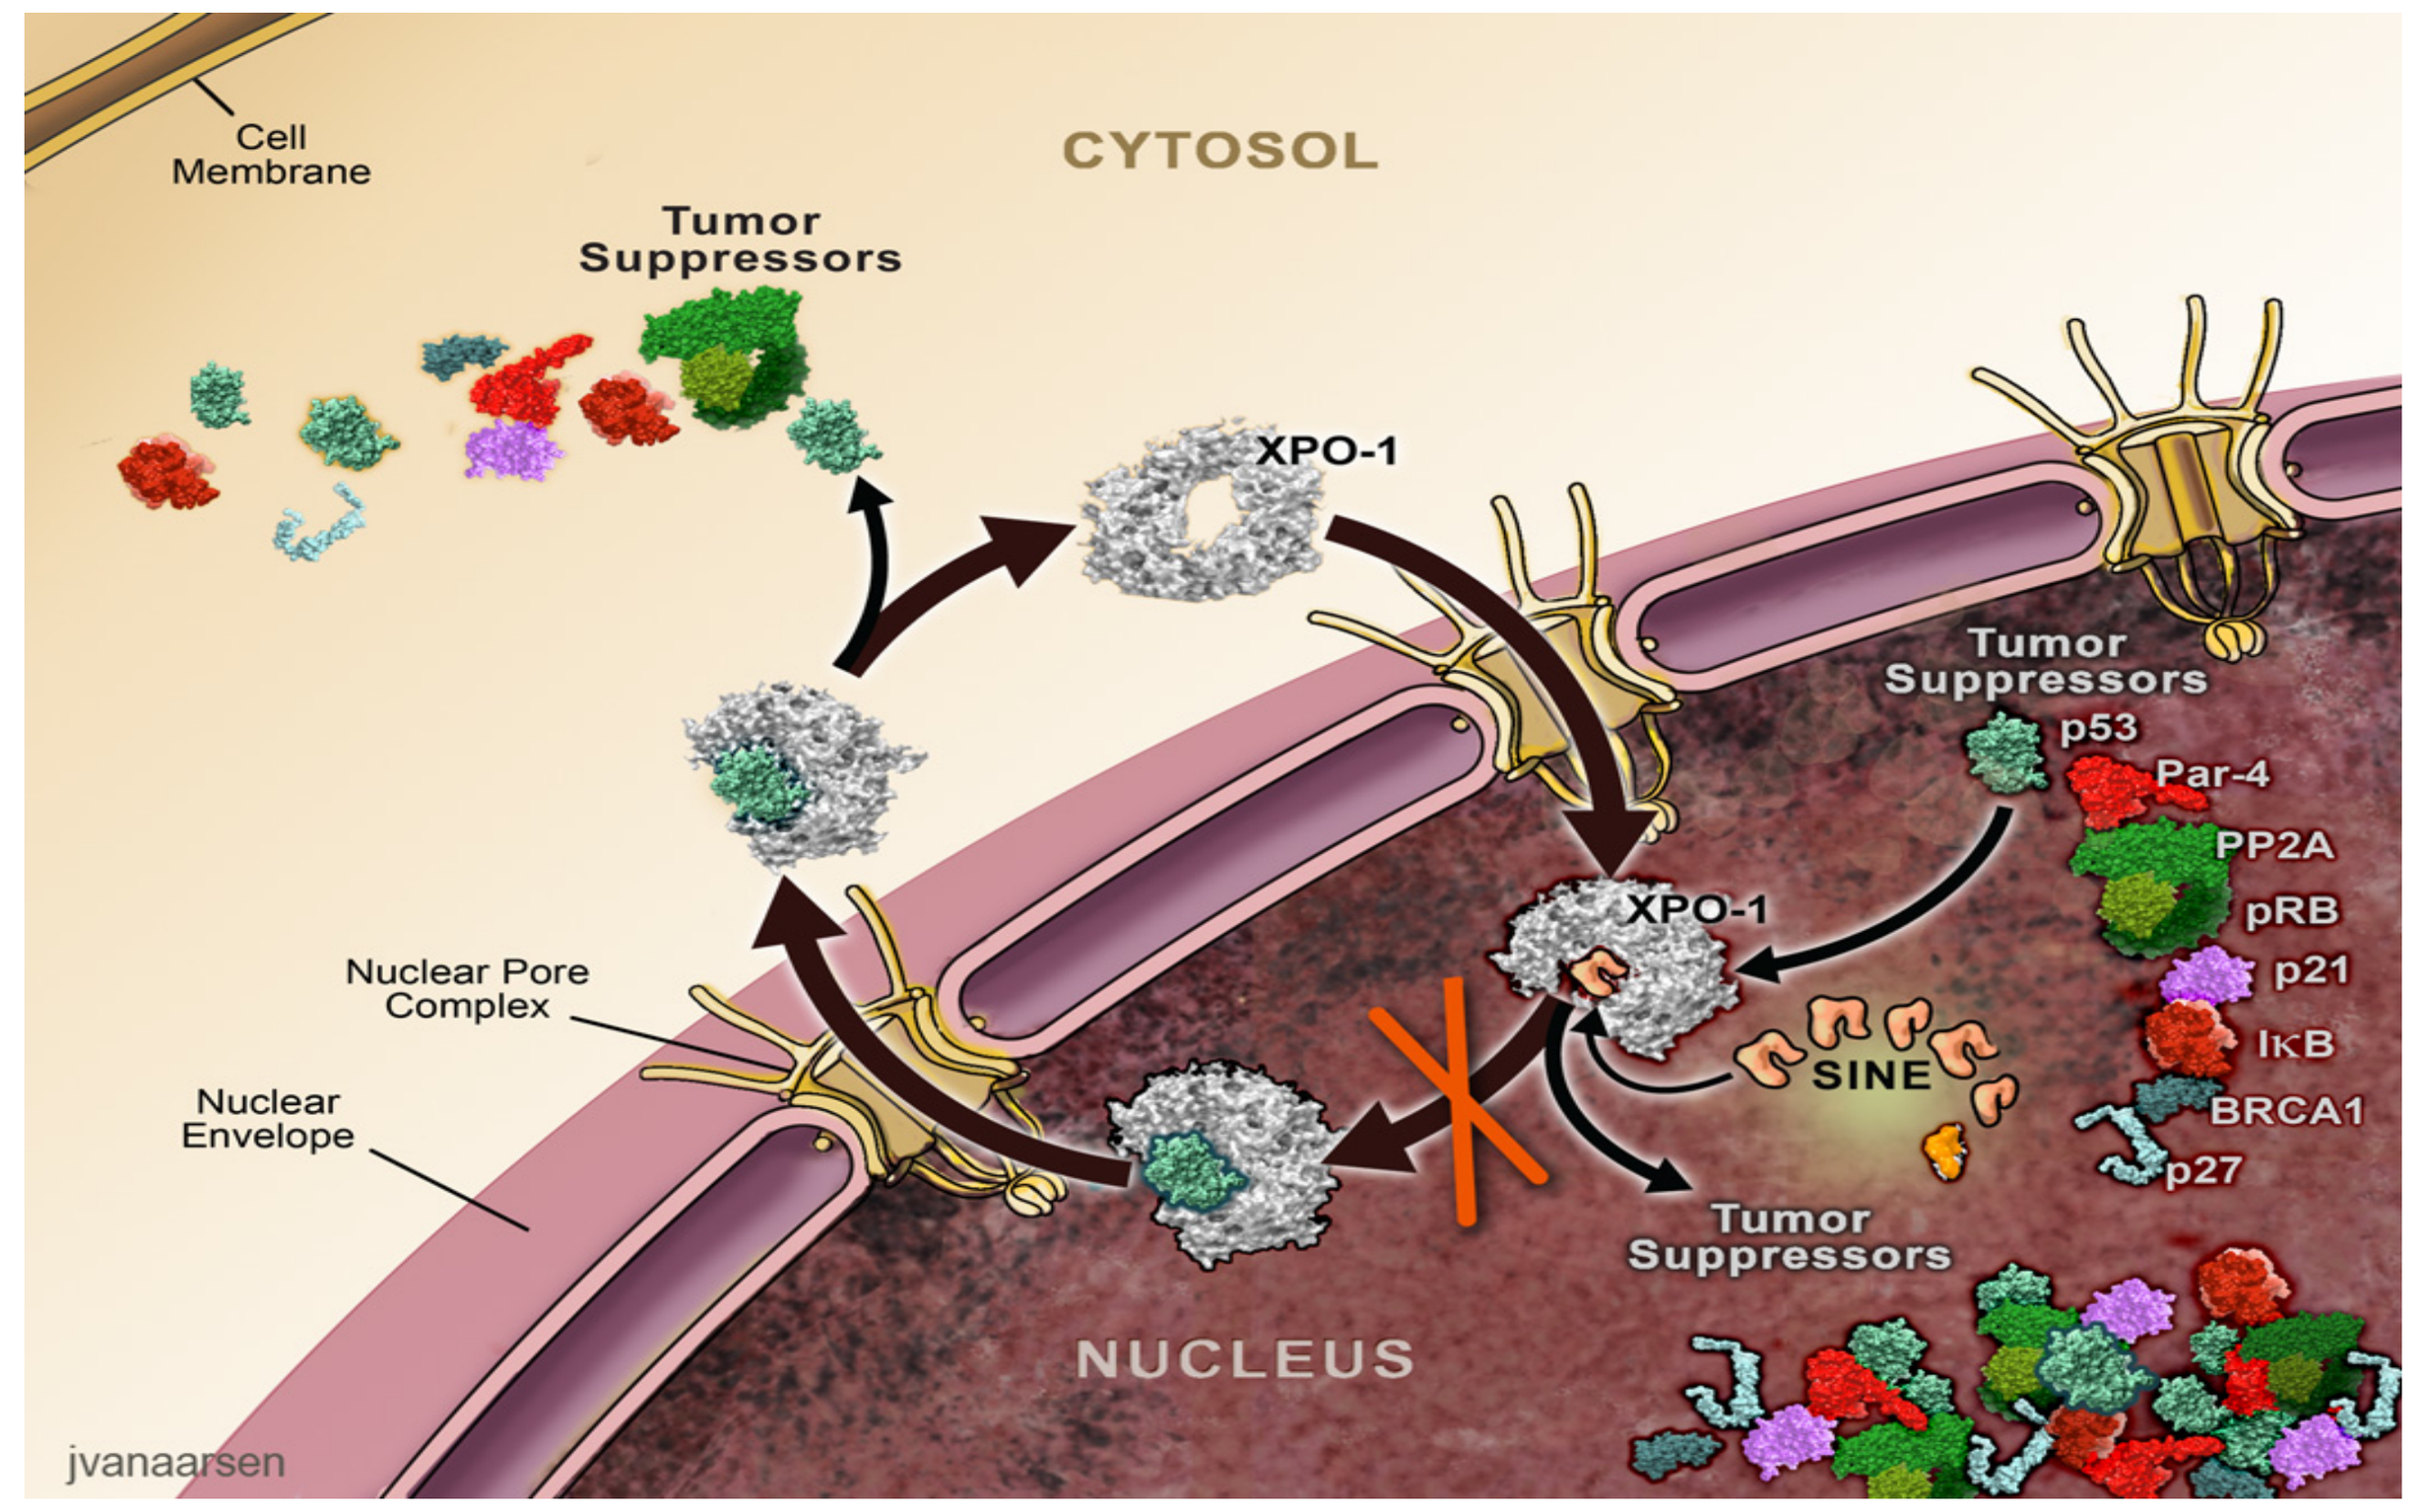

Cell Membrane
CYTOSOL
Tumor Suppressors
XPO-1
Tumor Suppressors
p53
Par-4
PP2A
pRB
p21
IκB
BRCA1
p27
XPO-1
SINE
Nuclear Pore Complex
Nuclear Envelope
Tumor Suppressors
NUCLEUS
jvanaarsen

- XPO1 (CRM1) is overexpressed in AML and its levels correlate with poor outcomes.
- XPO1 is the sole nuclear exporter of major TSP. XPO1 inhibition results in nuclear restoration and reactivation of TSP leading to selective induction of apoptosis of AML cells.
- NPM1 mutations (NPM1c) create an additional Nuclear Export Sequence (NES) leading to abnormal cytoplasmic localization of NPM1. XPO1 inhibition restores NPM1c to the nucleus.
- KPT-330 is a novel, potent, oral SINE in Ph1 studies in solid and hematological malignancies.
- KPT-330 showed potent anti AML activity in animal models of AML, significantly reducing tumor burden, sparing normal hematopoietic cells and increasing overall survival.
- KPT-330 also showed potent cytotoxicity against leukemic stem cells while sparing normal hematopoietic stem cell progenitors.

Contact Information: Dr. Michael Savona e-mail: msavona@tnonc.com

SINE Induced Nuclear Localization in AML Cells (*In-Vitro*)

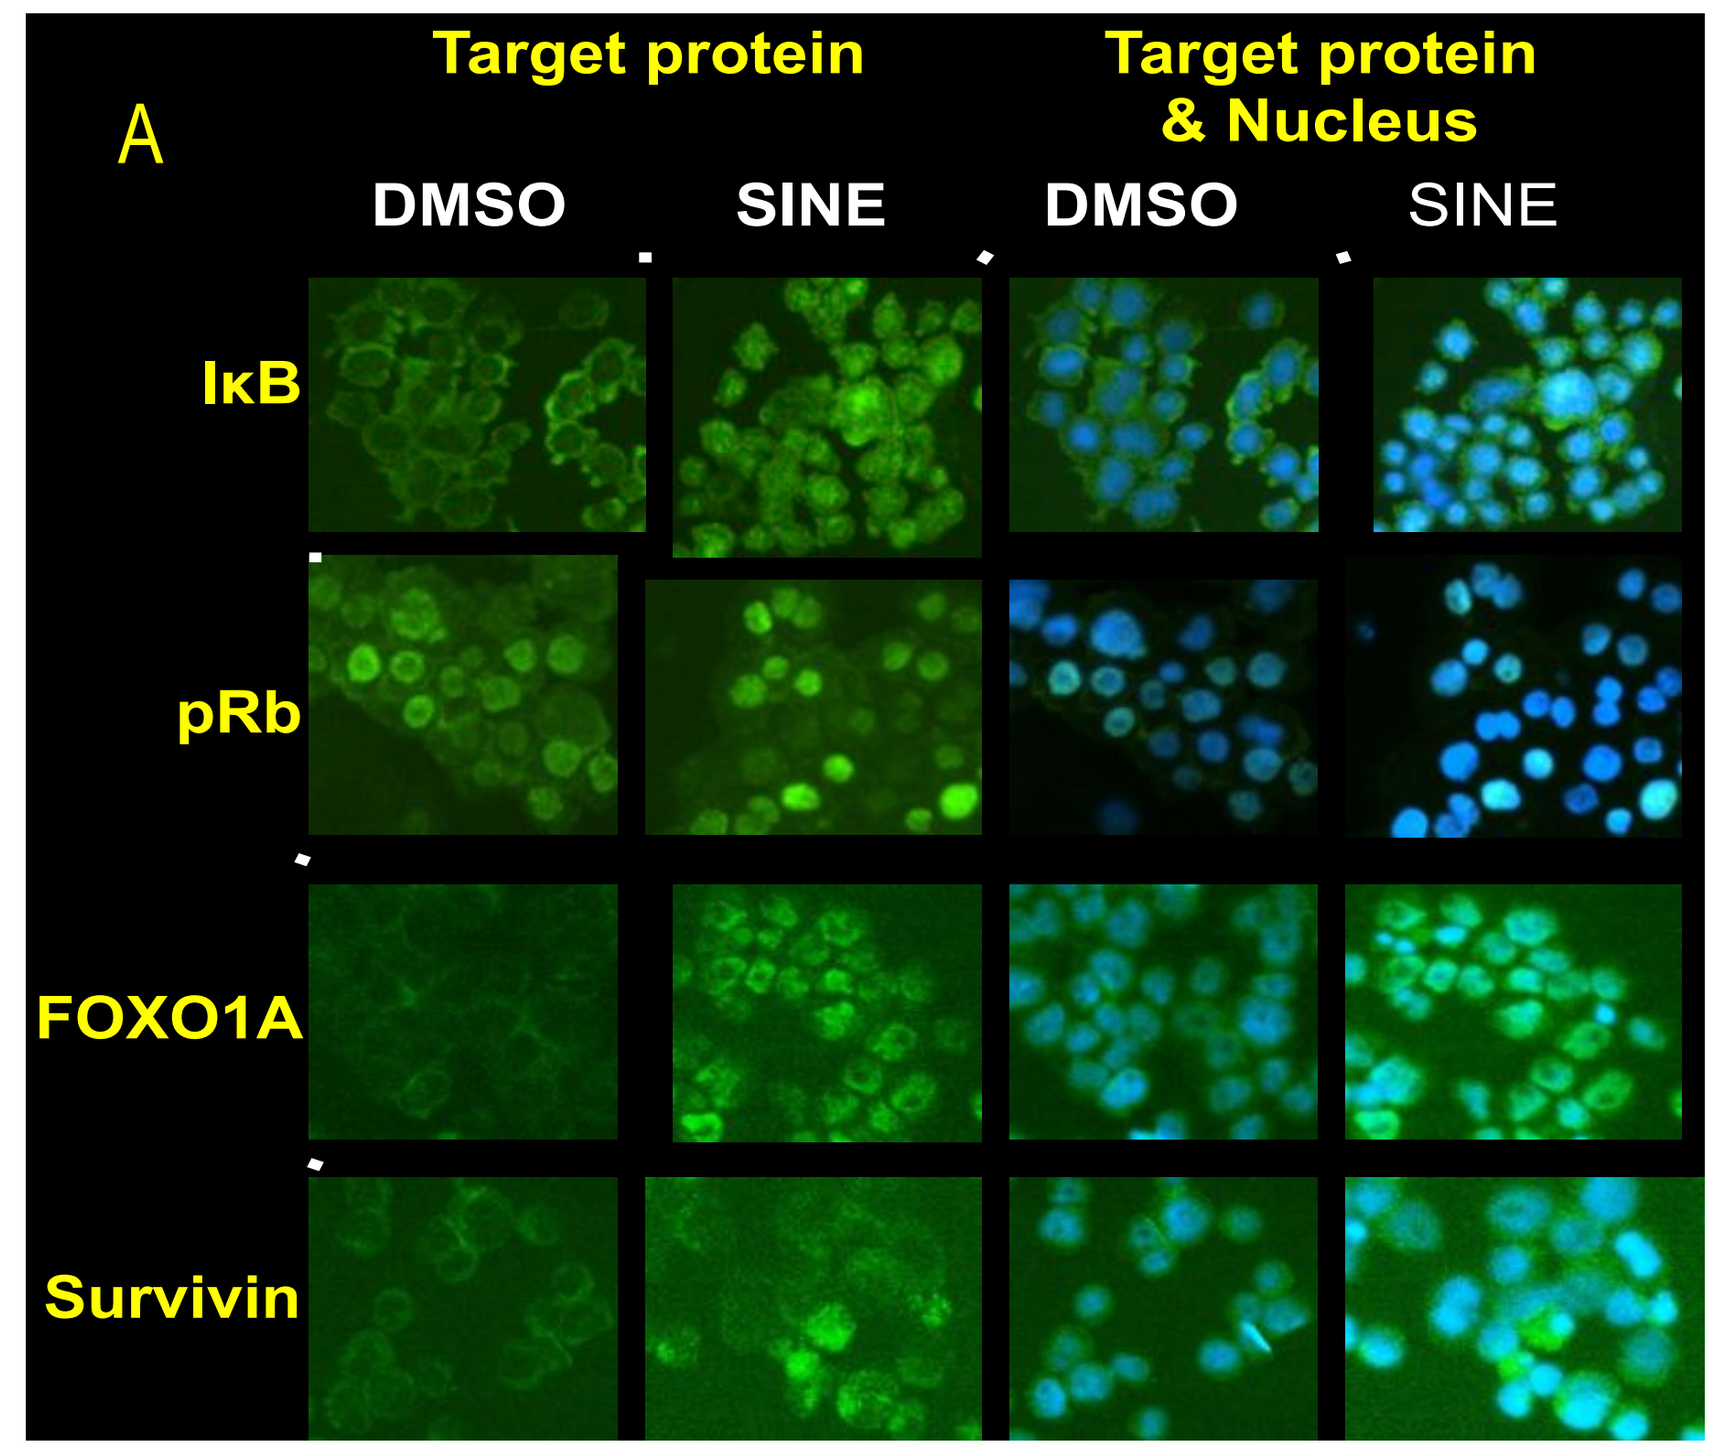

A
Target protein
Target protein & Nucleus
DMSO
SINE
DMSO
SINE
IκB
pRb
FOXO1A
Survivin

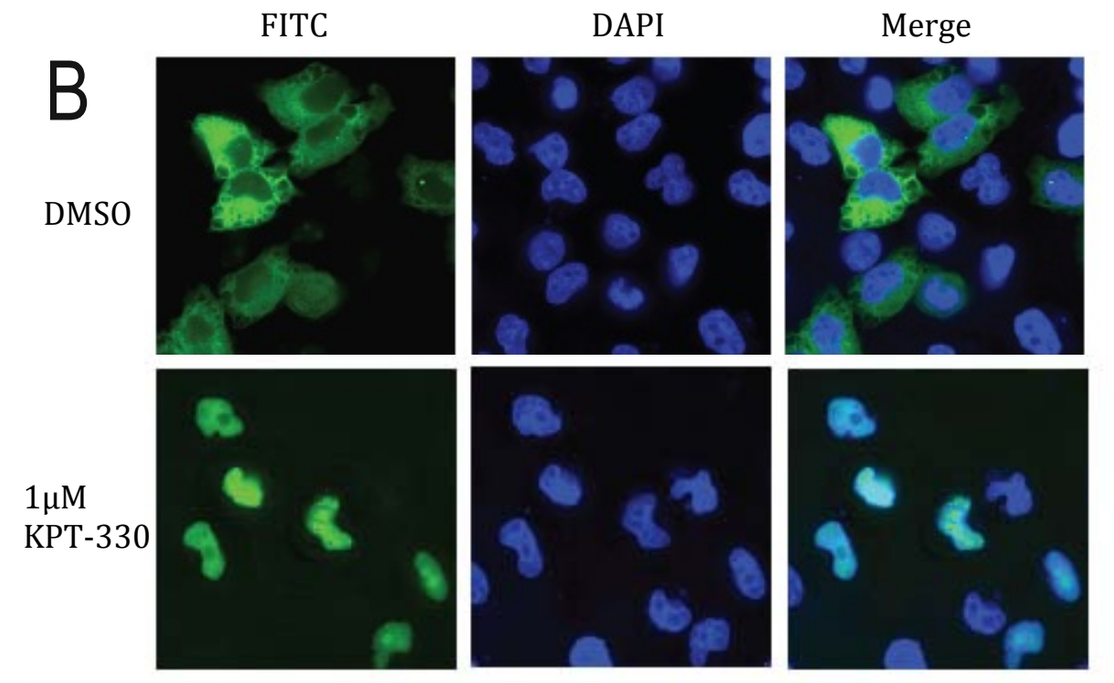

B
FITC
DAPI
Merge
DMSO
1μM KPT-330

A. AML cell lines THP-1 and Kasumi were treated with KPT-330 for 4-8h. Nuclear accumulation of IkB & pRb are shown in THP-1 cells. FOXO1 and Survivin are shown in Kasumi-1 cells.

B. KPT-330 induces nuclear accumulation of the AML associated mutant Nucleophosmin (mut NPM)

Phase 1 Study of KPT-330 in Hematological Malignancies

	Dose Escalation Cohorts	Dose Expansion Cohorts
Arm 1	Relapsed/Refractory B-Cell MM/WM, NHL, CLL	Relapsed/Refractory B-Cell MM/WM, DLBCL
Arm 2	Acute Myeloblastic Leukemia (AML)	AML

Patient Demographics

Characteristic N=39	
Median Age (Range)	68 (24-89)
Male to Female	20 Males : 19 Females
ECOG PS 0:1	9 : 30
Therapy Line for Disease in Study	
AML with prior MDS	2 (5%)
2nd Line AML	9 (23%)
3rd Line AML	10 (26%)
> 3rd Line AML	13 (33%)
Unknown	5 (13%)
AML Cytogentic Risk	
Good	4 (10%)
Intermediate	15 (39%)
Poor	13 (33%)
Unknown	7 (18%)

Pharmacokinetics and Pharmacodynamics of KPT-330

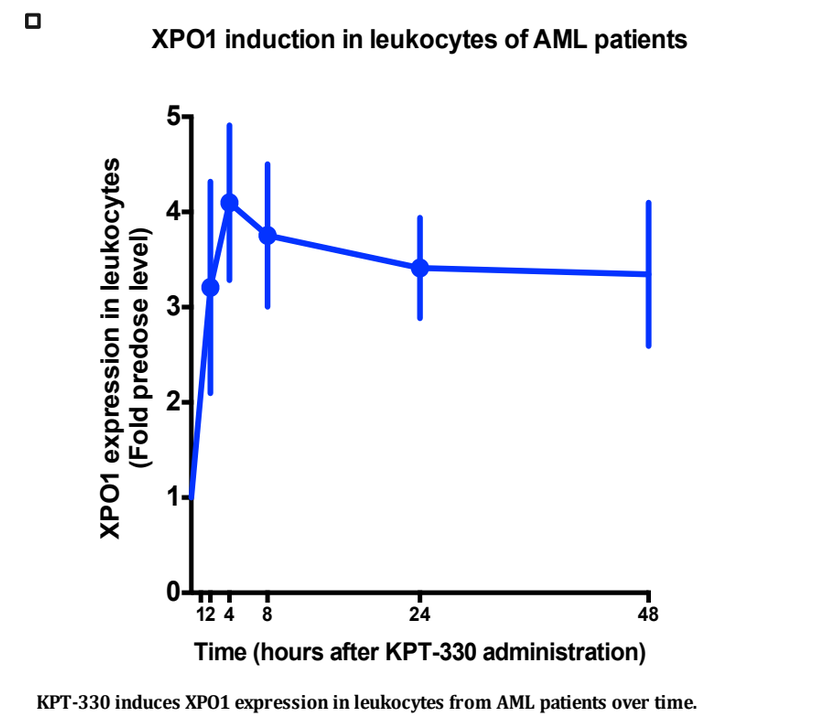

XPO1 induction in leukocytes of AML patients
XPO1 expression in leukocytes (Fold predose level)
Time (hours after KPT-330 administration)

KPT-330 induces XPO1 expression in leukocytes from AML patients over time.

Leukocytes were collected from AML patients over a timecourse of 48 h after oral administration of 30 mg/m^2 KPT-330. XPO1 expression was determined by qPCR from extracted RNA. Each point represents the mean and SEM for 3 – 8 samples.

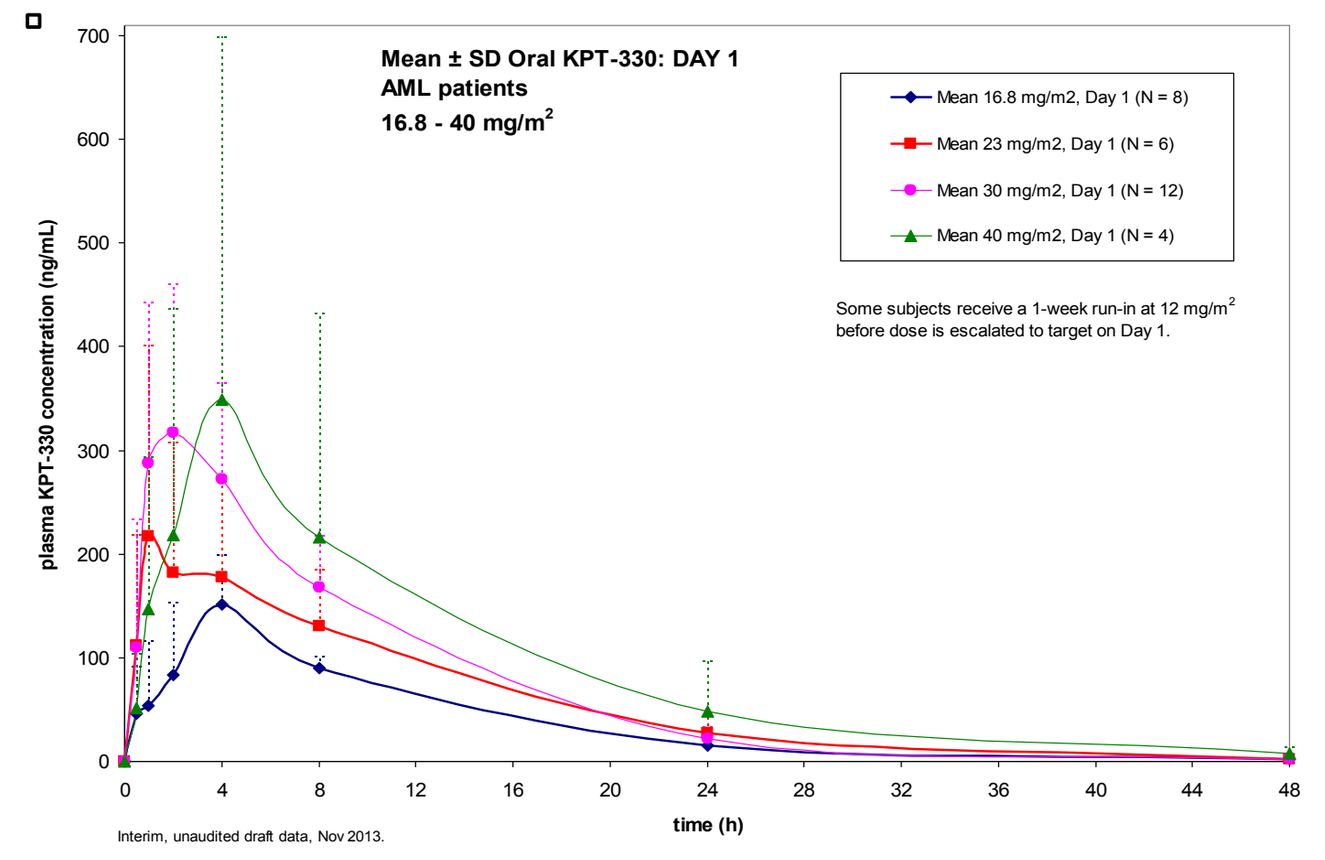

Mean ± SD Oral KPT-330: DAY 1
AML patients
16.8 - 40 mg/m2
Mean 16.8 mg/m2, Day 1 (N = 8)
Mean 23 mg/m2, Day 1 (N = 6)
Mean 30 mg/m2, Day 1 (N = 12)
Mean 40 mg/m2, Day 1 (N = 4)
Some subjects receive a 1-week run-in at 12 mg/m2 before dose is escalated to target on Day 1.
plasma KPT-330 concentration (ng/mL)
time (h)
Interim, unaudited draft data, Nov 2013.

- Oral absorption was rapid with median T_{max} from 2-4 hours.
- Exposure was dose proportional across 16.8 – 40 mg/m^2 doses.
- Half life was ~6 hours and dose independent.

KPT-330 Related Adverse Events in ≥ 3 AML Patients N=38

AE NAME	GRADE	KPT-330 Dose Levels (mg/m²) 16.8	23	30	40	Total All N=38
Gastrointestinal, Constitutional, and Others		N=10	N=6	N=14	N=8	
Nausea	Grade 1	3 (30%)	3 (50%)	2 (14%)	4 (50%)	31.5%
	Grade 2	2 (20%)	1 (17%)	4 (28.5%)	1 (12.5)%	21%
	Grade 3	1 (10%)	--	1 (7%)	--	5%
Anorexia	Grade 1	--	--	5 (36%)	1 (12.5)%	16%
	Grade 2	3 (30%)	3 (50%)	3 (21%)	1 (12.5)%	26%
	Grade 3	--	1 (17%)	--	--	3%
Fatigue	Grade 1	--	1 (17%)	1 (7%)	--	5%
	Grade 2	4 (40%)	3 (50%)	5 (38%)	1 (12.5%)	34%
	Grade 3	1 (10%)	--	1 (7%)	1 (12.5%)	8%
Vomiting	Grade 1	3 (30%)	3 (50%)	3 (21%)	2 (25%)	31.5%
	Grade 2	--	--	--	1 (12.5)%	3%
	Grade 3	--	--	1 (7%)	--	3%
Diarrhea	Grade 1	3 (30%)	1 (17%)	2 (14%)	2 (25%)	21%
	Grade 2	--	1 (17%)	2 (14%)	1 (12.5)%	10.5%
Weight Loss	Grade 1	2 (20%)	3 (50%)	2 (14%)	--	18%
	Grade 2	1 (10%)	1 (17%)	1 (7%)	--	8%
Dehydration	Grade 1	1 (10%)	--	1 (7%)	--	5%
	Grade 3	--	--	1 (7%)	--	3%
Taste Alteration	Grade 1	--	2 (33%)	2 (14%)	--	10.5%
	Grade 2	--	--	1 (7%)	--	3%
Asparate Aminotransferase Increase	Grade 1	--	1 (17%)	1 (7%)	--	5%
	Grade 3	1 (10%)	--	--	--	3%
Hypomagnesemia	Grade 1	--	--	2 (14%)	1 (12.5%)	8%
	Grade 2	1 (10%)	--	--	--	3%
Hypokalemia	Grade 1	--	1 (17%)	--	--	3%
	Grade 2	--	1 (17%)	--	--	3%
	Grade 3	2 (20%)	--	--	--	5%
Hypotension	Grade 1	1 (10%)	--	--	--	3%
	Grade 2	1 (10%)	--	--	--	3%
	Grade 3	1 (10%)	--	--	--	3%
Hyponatremia	Grade 1	2 (20%)	3 (50%)	--	--	13%
	Grade 3	1 (10%)	--	--	--	3%
Blurred Vision	Grade 1	--	2 (33%)	1 (7%)	--	8%
	Grade 2	--	--	--	1 (12.5%)	3%
Hematological						
Thrombocytopenia	Grade 4	1 (10%)	1 (17%)	1 (7%)	--	8%

KPT-330 Activity in Rel/Ref AML Patients (33 Evaluable Pts)

Responses in Arm 2 Acute Myeloid Leukemia Patients as of 4-Dec-2013

Number of Pts Evaluated	Total CRs, CR(i)s, PRs, and SD (%)	CR (%)	CR (i) (%)	PR (%)	MLFS (%)	SD (%)	PD	WC
33	18 (55%)	4 (12%)	1 (3%)	2 (6%)	1 (3%)	10 (30%)	11 (33%)	4 (12%)

CR=Complete Response With Hematological Recovery, CR(i)=Complete Response Without Hematological Recovery, PR=Partial Response, MFLS=Morphological Leukemia Free State, SD=Stable Disease, PD=Progressive Disease, WC=Withdrew Consent

Pt 040-501 Achieved a Complete Response and Resolution of Leukemia Cutis

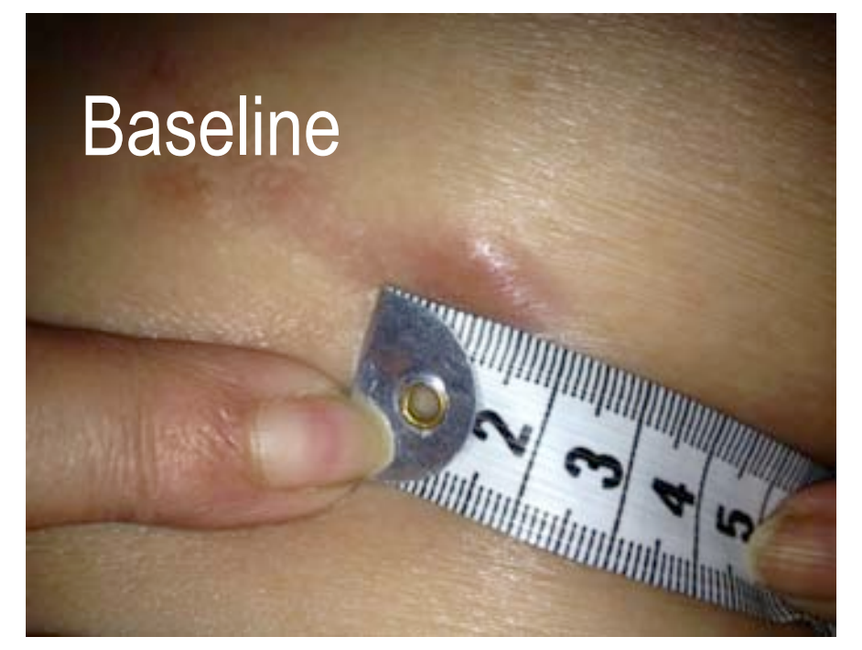

Baseline

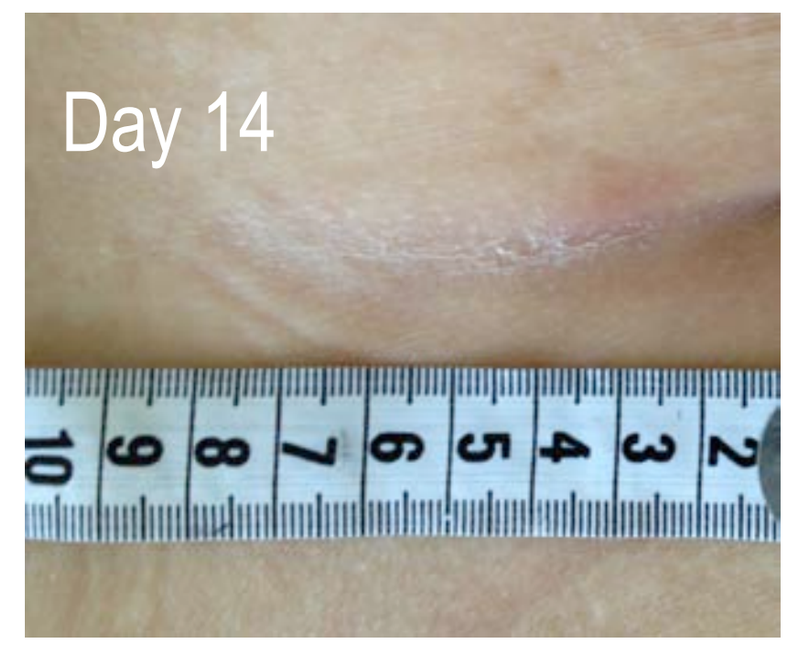

Day 14

69 year old with R/R AML with multiple skin lesions (leukemia cutis). After 14 days on KPT-330, leukemia cutis was nearly resolved and the patient achieved a CR in cycle 1. Patient progressed after 113 days of treatment.

KPT-330 Induces a Reduction in AML Blast Counts

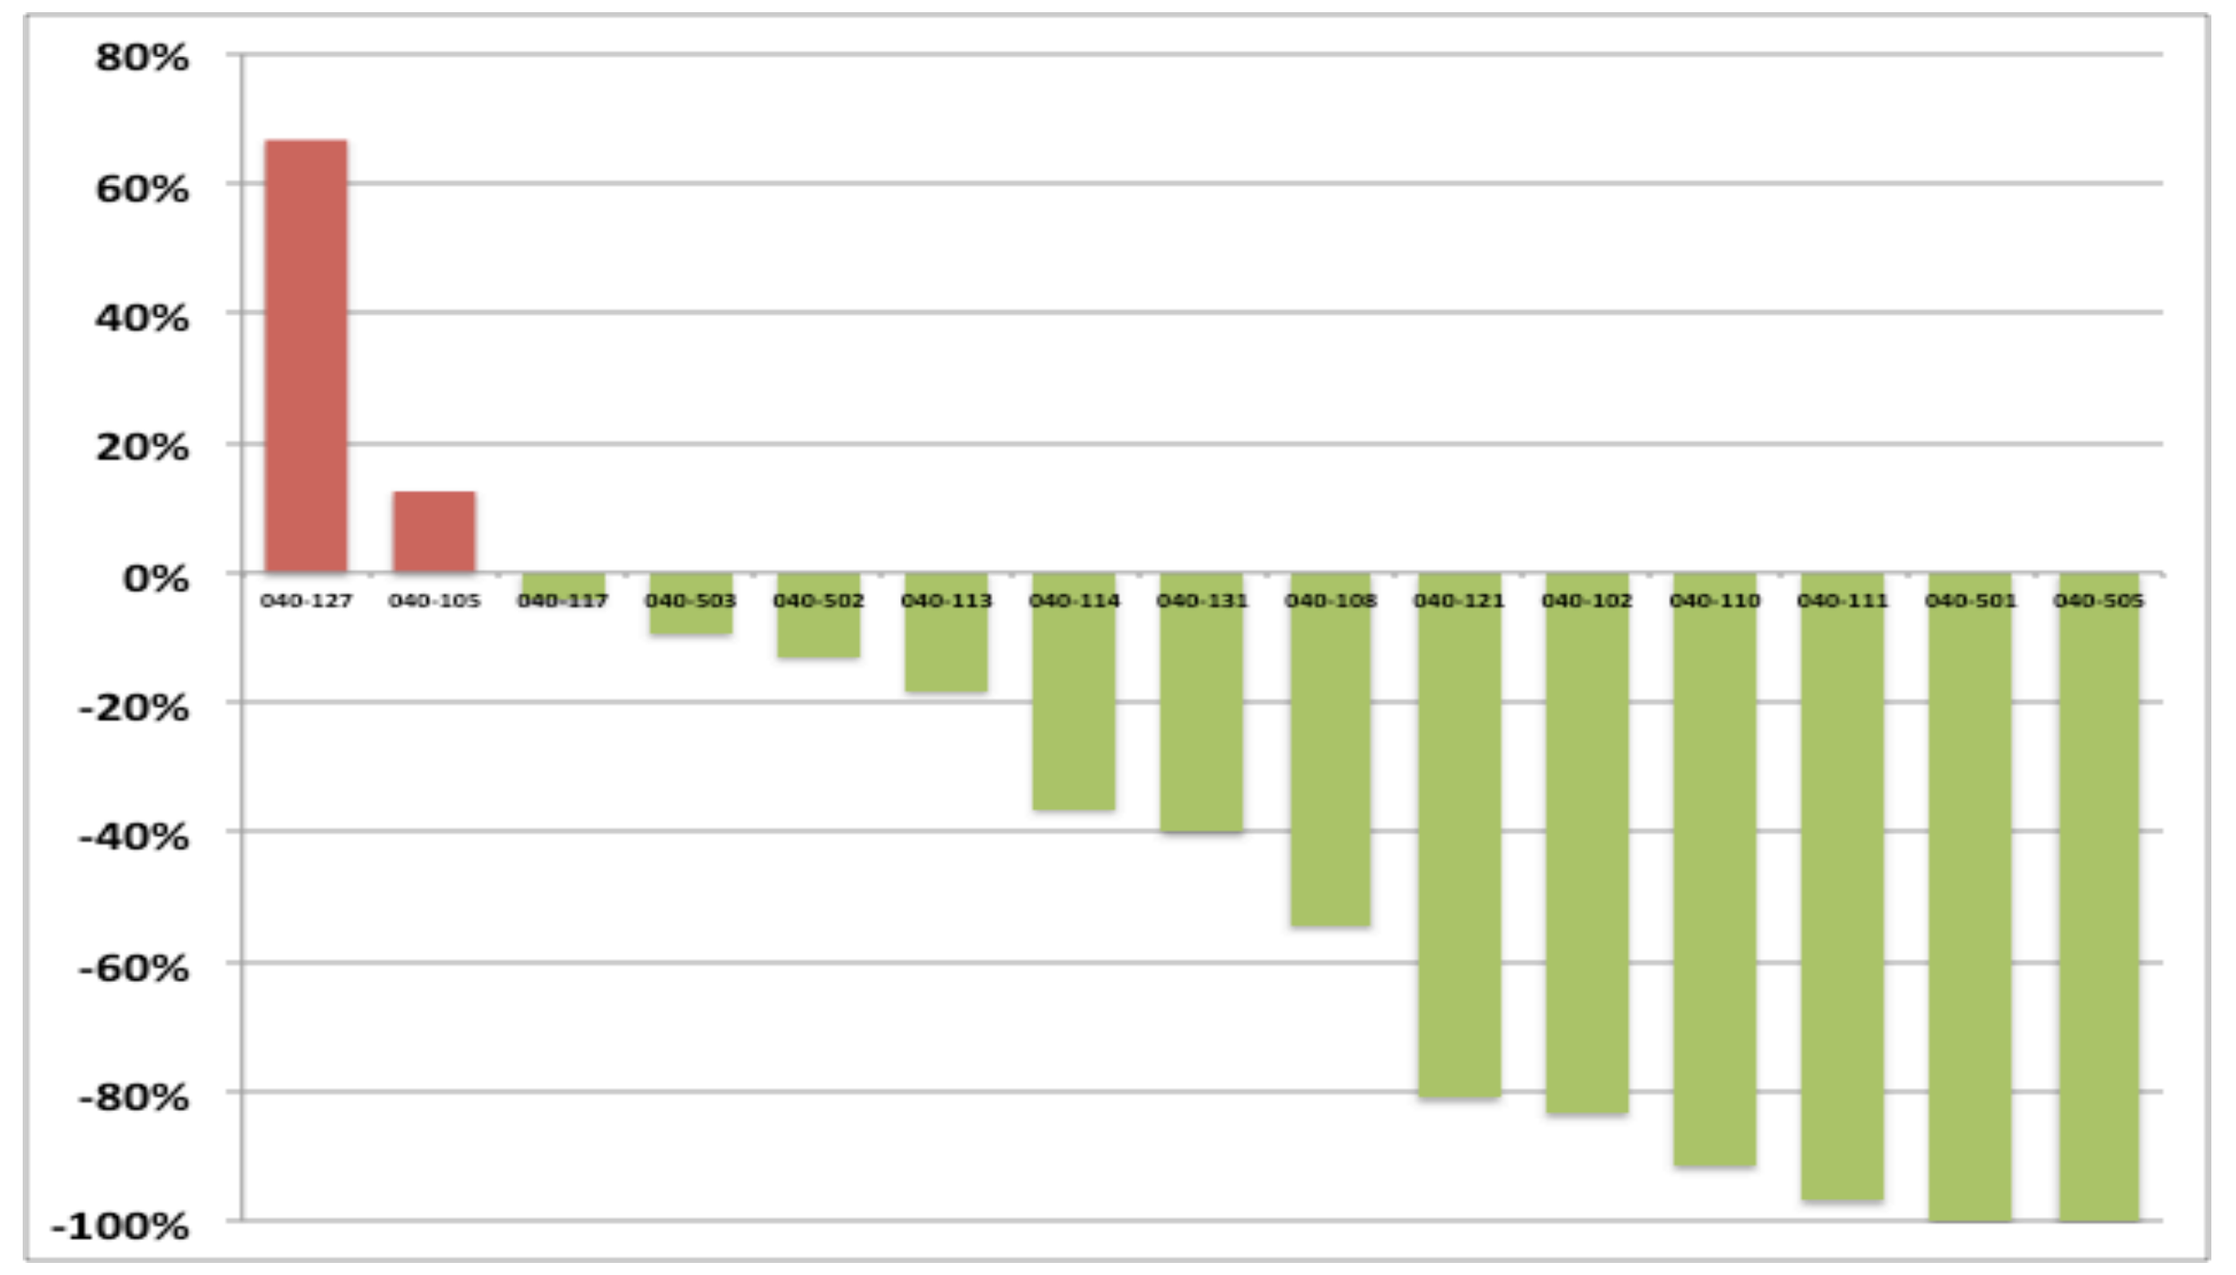

80%
60%
40%
20%
0%
-20%
-40%
-60%
-80%
-100%
040-127
040-105
040-117
040-503
040-502
040-113
040-114
040-131
040-108
040-121
040-102
040-110
040-111
040-501
040-505

BM Blast cells were evaluated at screening and at the end of each cycle. Maximal % change from baseline is shown

Patient Outcomes and Time on Study

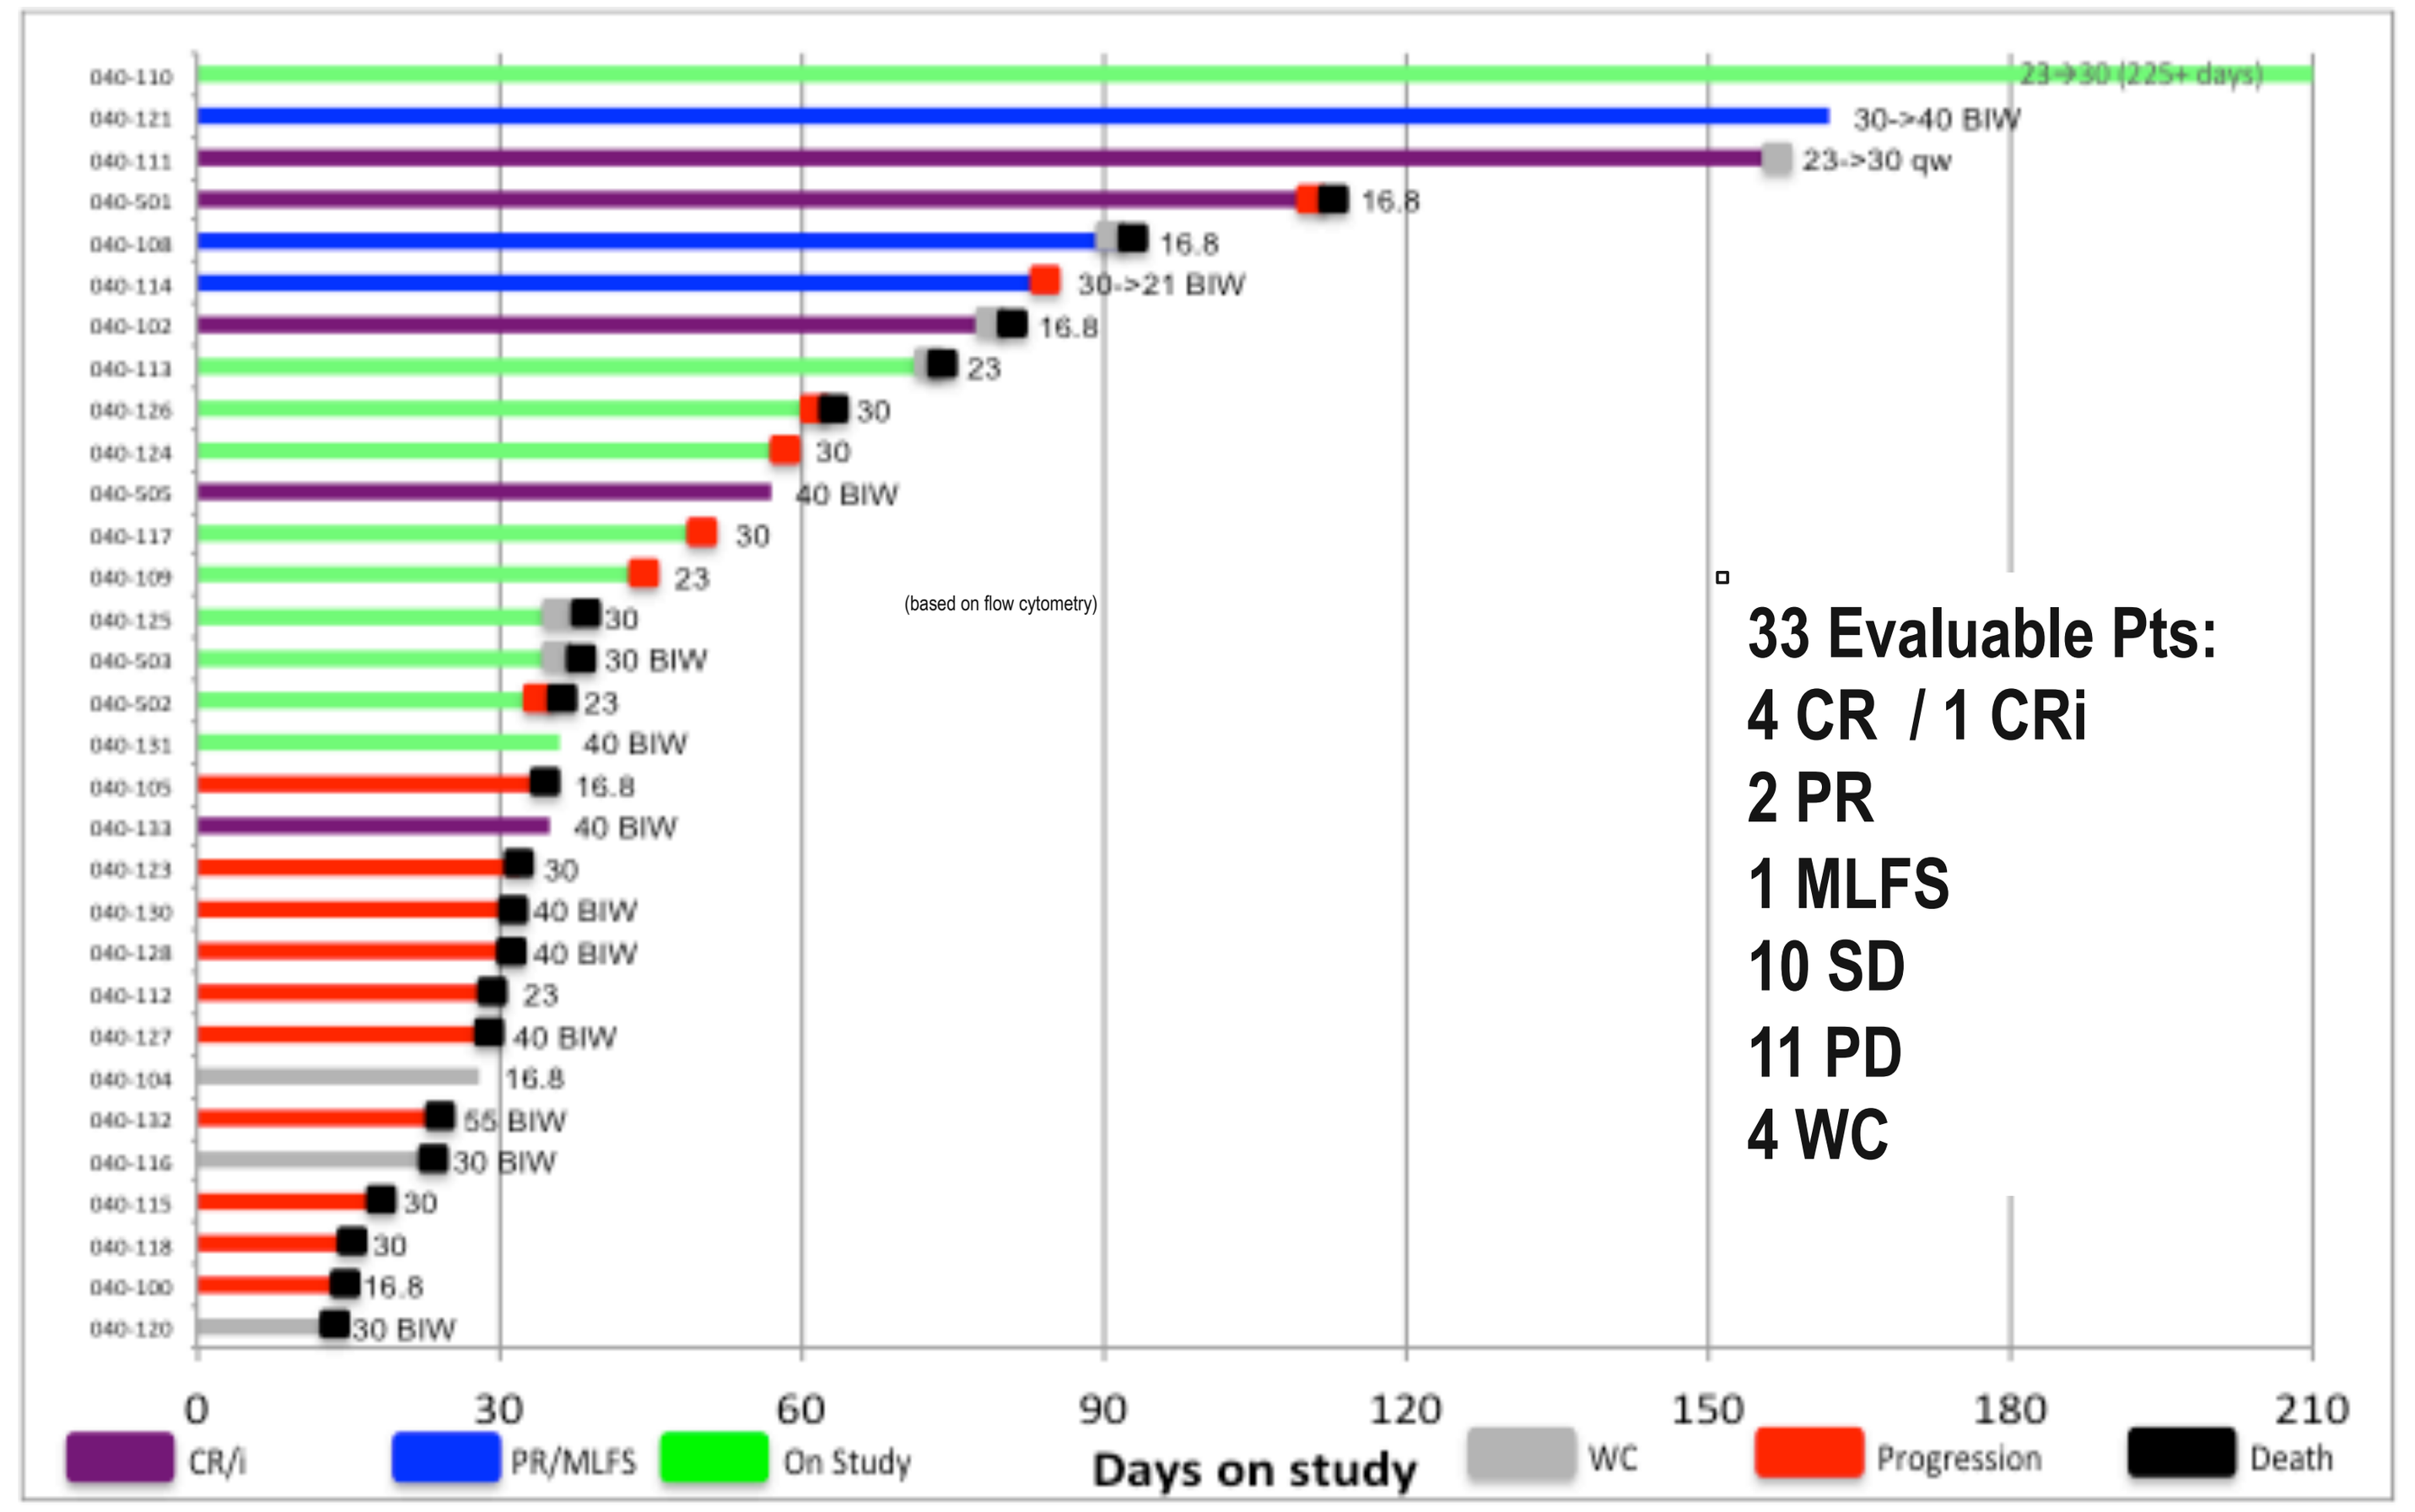

23->30 (225+ days)
30->40 BIW
23->30 qw
30->21 BIW
(based on flow cytometry)
33 Evaluable Pts:
4 CR / 1 CRi
2 PR
1 MLFS
10 SD
11 PD
4 WC
Days on study
CR/i
PR/MLFS
On Study
WC
Progression
Death

Conclusions

- XPO1 (CRM1) is the sole nuclear exporter of TSP (p53, Rb, etc). Previous studies have shown XPO1 is overexpressed in AML and its levels correlate with poor outcomes.
- XPO1 inhibition results in nuclear restoration and reactivation of TSP leading to selective induction of apoptosis of AML cells and leukemic stem cells, while sparing normal hematopoietic (stem) cells.
- Selinexor (KPT-330) is a first-in-class, potent, oral SINE that may induce responses and >3 month stable disease in patients with relapsed/refractory AML across multiple genotypes.
- Continued treatment with KPT-330 appears to allow recovery of normal hematopoietic cells.
- The most common side effects observed have been anorexia, nausea and fatigue, which have been significant but manageable with supportive care; no major organ toxicities have been observed to date.
- Additional studies with KPT-330 alone or in combination in patients with AML are planned.