Mitochondrial Priming of New Targeted Agents in Acute Myeloid Leukemia

THE UNIVERSITY OF TEXAS MD Anderson Cancer Center Making Cancer History®

Jo Ishizawa,[1] Kensuke Kojima,[1 2] Seshagiri R Duvvuri,[1] Teresa McQueen,[1] Vivian Ruvolo,[1] Graciela M. Nogueras-González,[3] Xuelin Huang,[3]
William Pierceall,[4] Michael Cardone,[4] Ryan Lena,[4] Camille Doykan,[4] Sharon Shacham,[5] Michael Kauffman,[5] Marina Konopleva,[1] Michael Andreeff[1]

[illegible]; [2]Hematology, Respiratory Medicine and Oncology, Department of Medicine, Saga University, Saga, Japan, and [illegible]ent of Hematopathology; [3]Department of Biostatistics, Division of Quantitative Sciences, the University of Texas MD Anderson Cancer Center; [4]Eutropics Pharmaceuticals, Cambridge, MA; [5]Karyopharm Therapeutics, Boston, MA

Abstract

As numerous molecularly targeted agents are entering clinical trials, predictive testing is highly desirable. We investigated if response to certain agents correlates with the recently reported method of BH3 profiling (Chonghaile TN. et al, Science, 2011), a functional assay developed by Letai's group that measures tumor cell mitochondrial priming by measuring mitochondrial outer membrane permeabilization (MOMP) following exposure to the peptide-mimicking BH3 domains of BH3-only proteins. Mitochondrial priming has been reported to be correlated with clinical responses to conventional chemotherapy in solid tumors and hematological malignancies (Vo TT, et al, Cell, 2012, Pierceall W et al, Mol Cancer Ther, 2013). Twenty-two AML lines were tested. Cells were permeabilized with digitonin and exposed to standardized doses of BH3 peptides (BIM, PUMA, NOXA, BAD, BMF, HRK, or PUMA2A). JC-1 was used for detection of MOMP and served as a measure of sensitivity to each peptide (reported as %[BH3 peptide]). Also, BCL-2, BCL-XL and MCL-1 expression levels were determined by Western blot. In addition to studies of untreated cells, treatment effects of different anti-leukemia drugs (AraC, Nutlin-3a, KPT-330 [Selinexor] and ABT-199) were determined over a wide dose range and denoted as ([specific apoptosis] = [(%Annexin V+ cells at each dose) - (%Annexin V+ cells at 0 μM)]/[100- (%Annexin V+ cells at 0 μM)]). Mixed linear models were used for analysis. As expected, ABT-199 sensitivity positively correlated with %[BAD]-%[HRK] (|b| = 3.22, p < 0.001), which is compatible with BCL-2 dependency of ABT-199 (while BAD is binding to BCL-2 and BCL-XL, HRK is binding to BCL-XL only and the difference is therefore BCL-2-specific). This was supported by the observed correlation between %[BAD]-%[HRK] and BCL-2 protein expression levels (r=0.619; p = 0.018). AraC sensitivity showed a similar correlation with %[BAD]-%[HRK] (|b| = 1.61, P < 0.05). Unexpectedly, Nutlin-3a activity did not correlate with any of the BH3 peptides. Results indicate that ABT-199, KPT-330 and Nutlin-3a show different modes of action in terms of BH3 peptide dependency, supporting potential combination effects of these agents. For ABT-199, increased MCL1 levels were associated with diminished cytotoxicity (r= 0.720; p < 0.005), as expected. For Ara-C, a similar correlation with MCL-1 was noted (r = 0.623; p < 0.05), but no correlations were observed for Nutlin-3a and KPT-330.

Nutlin-3a KPT-330 ABT-199

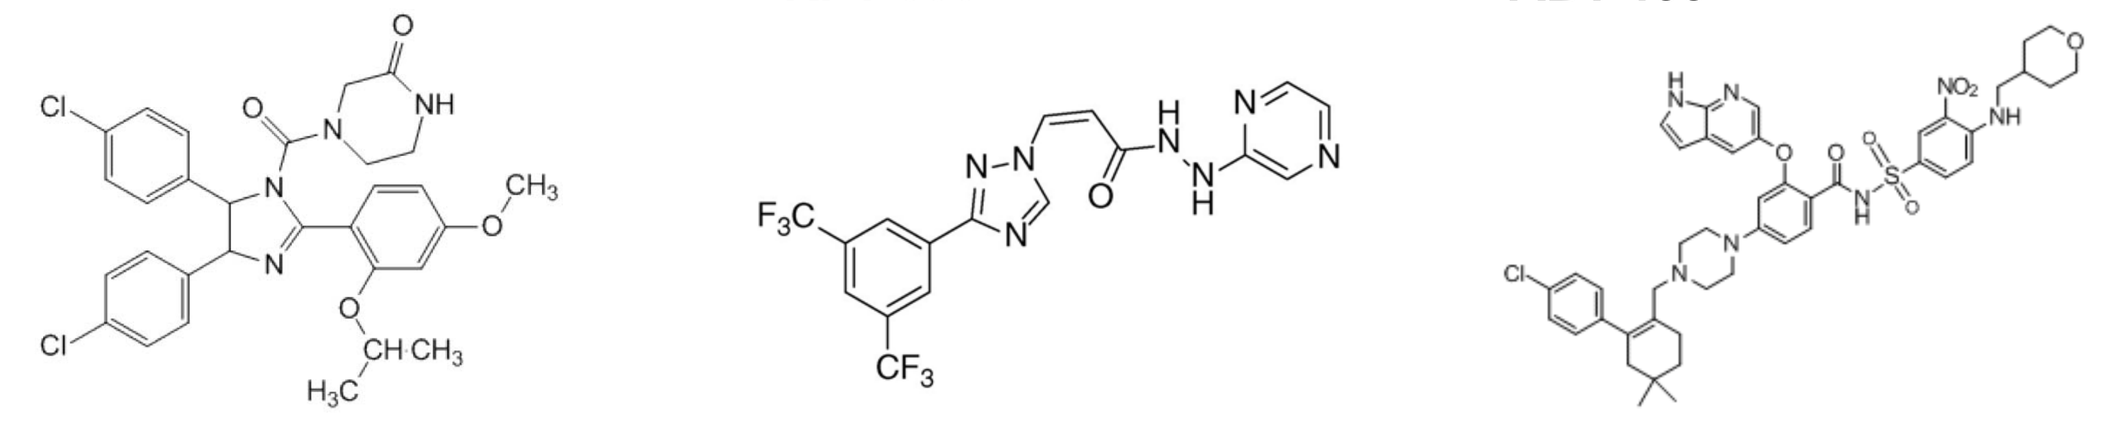

Methods

1. Reagents and cells: KPT-330 was synthesized at Karyopharm Therapeutics. (Natick, MA). MDM2 antagonist Nutlin-3a was purchased from Cayman Chemical Company (Ann Arbor, MI). BCL-2 inhibitor ABT-199 was purchased from Selleckchem. Total 22 AML cell lines, including OCI-AML3, MOLM-13 and MV4;11 cells were transduced with lentiviruses encoding either p53- or MCL-1- specific shRNA or scrambled shRNA, were used. BH3 profiling was conducted for all the lines. Independently, cells were treated with KPT-330, Nutlin3a, ABT-199 or Ara-C and apoptosis was assessed by Annexin V and PI staining.
2. Protein expression was assessed by Western blotting.
3. BH3 profilings: Cells were permeabilized with digitonin and exposed to BH3 peptides (BIM, PUMA, NOXA, BAD, BMF, HRK, or PUMA2A). JC-1 was used for detection of MOMP.
4. Statistical analysis: Student t test or Mann-Whitney (two-sided) results were considered significant for $P <$ 0.05. For analysis of the association between BH3 profiling and the sensitivity to each agent, Mixed linear models were used.

Results

Induced apoptosis in each cell line by each agent

Nutlin-3a

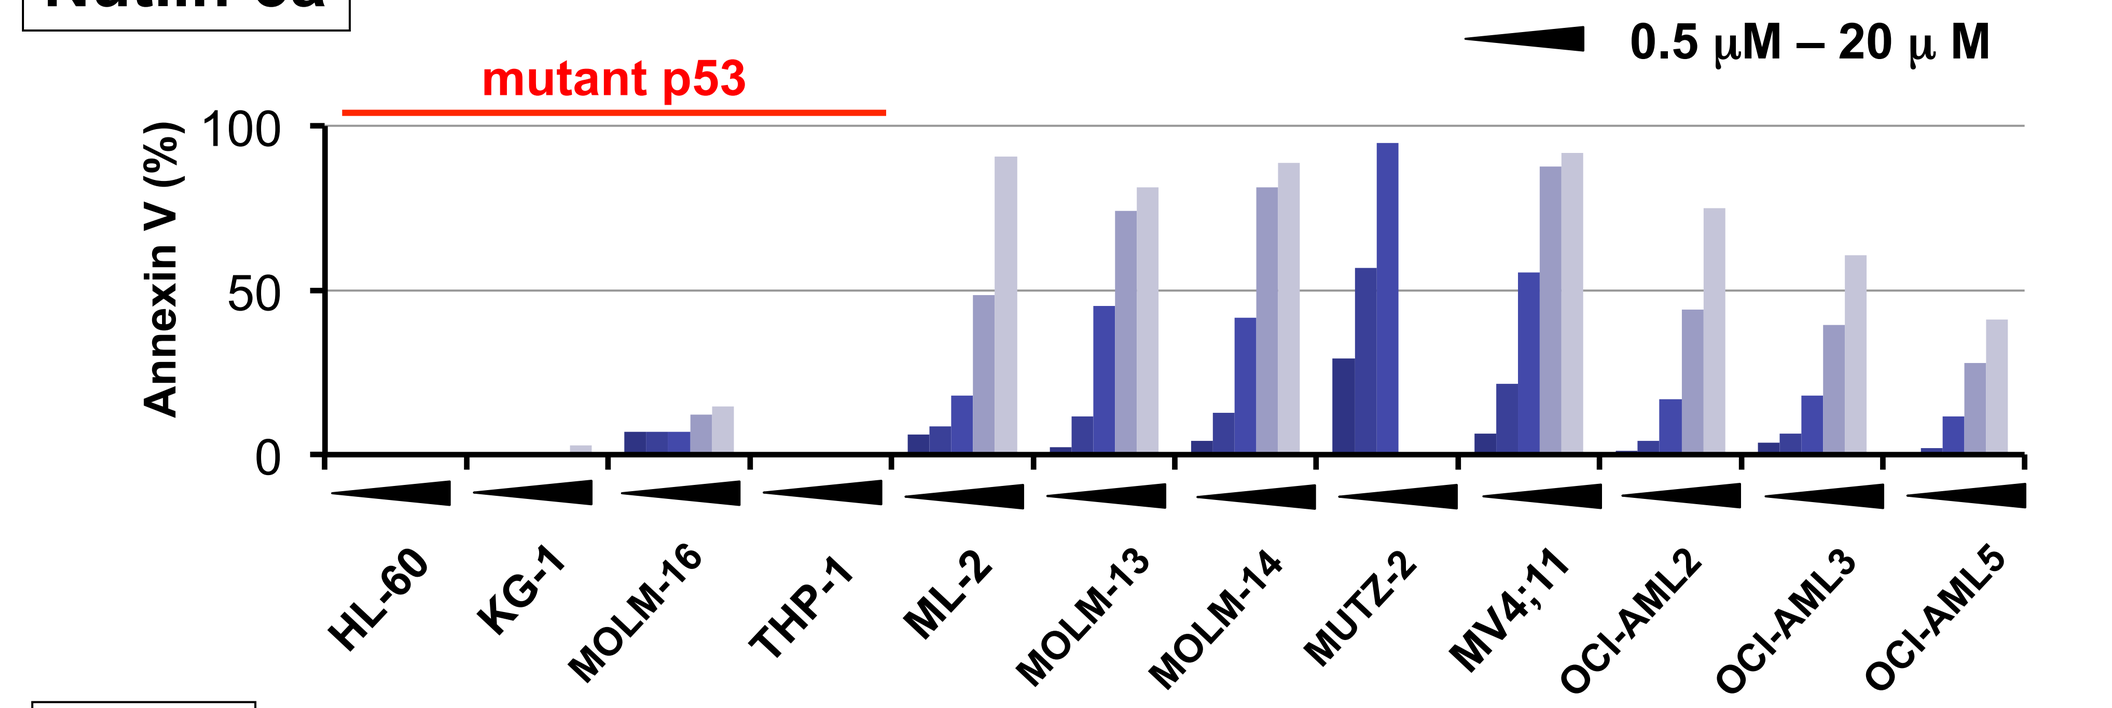
mutant p53
0.5 μM – 20 μ M
Annexin V (%)
HL-60 KG-1 MOLM-16 THP-1 ML-2 MOLM-13 MOLM-14 MUTZ-2 MV4;11 OCI-AML2 OCI-AML3 OCI-AML5

Ara-C

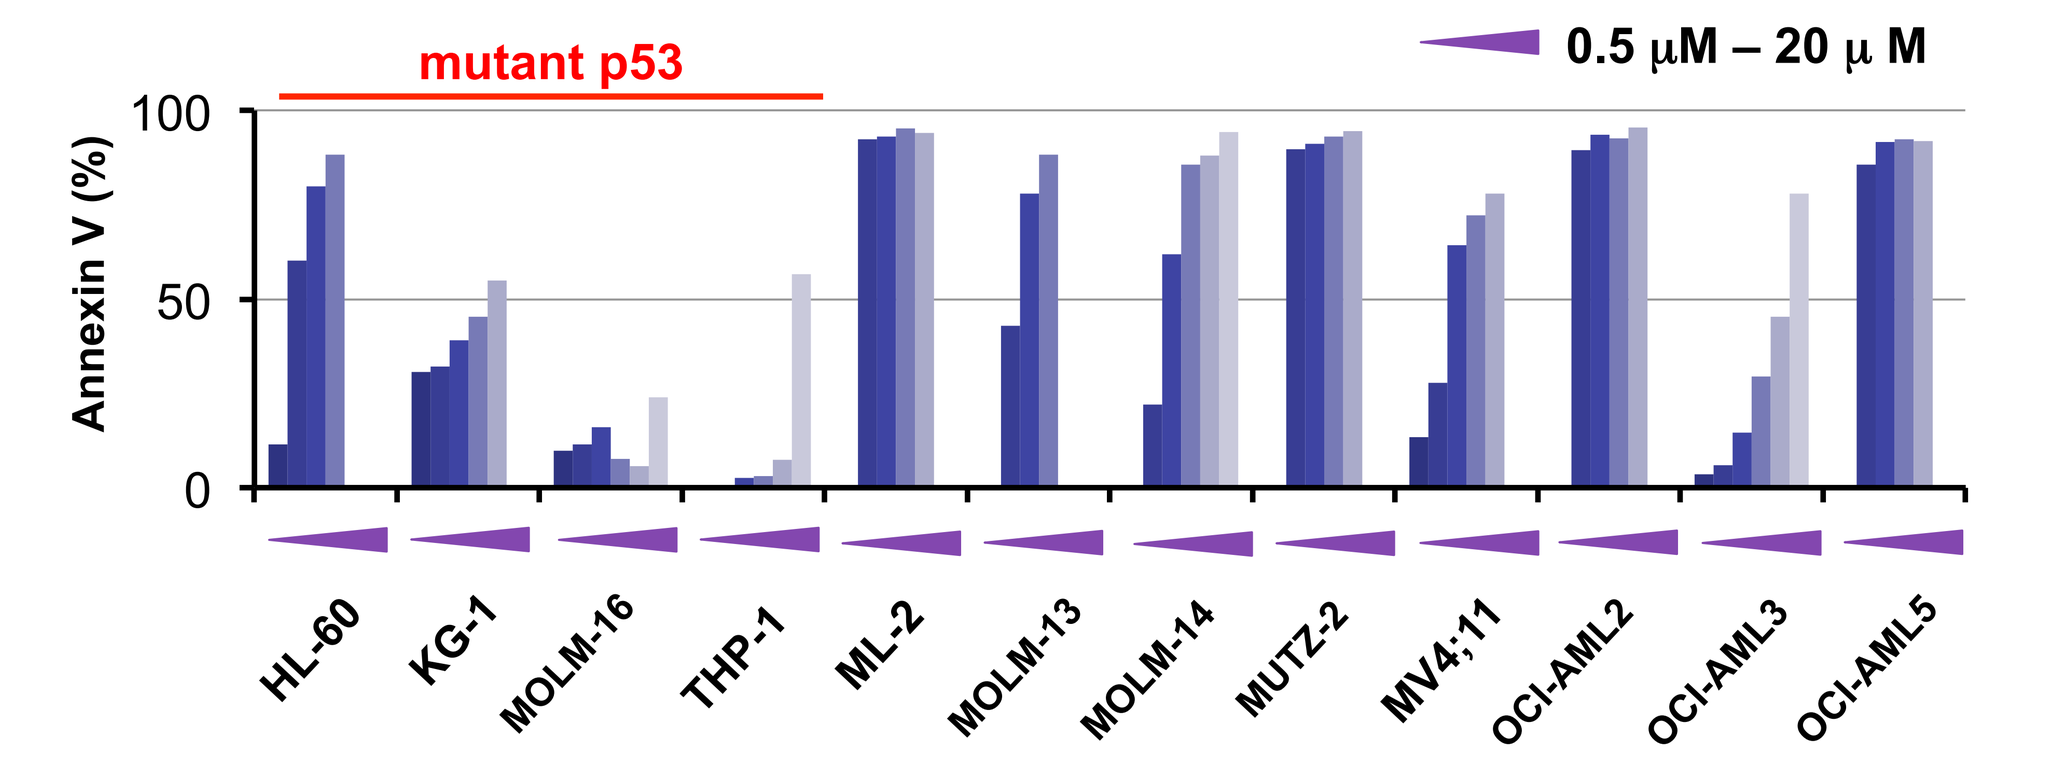
mutant p53
0.5 μM – 20 μ M
Annexin V (%)
HL-60 KG-1 MOLM-16 THP-1 ML-2 MOLM-13 MOLM-14 MUTZ-2 MV4;11 OCI-AML2 OCI-AML3 OCI-AML5

KPT-330

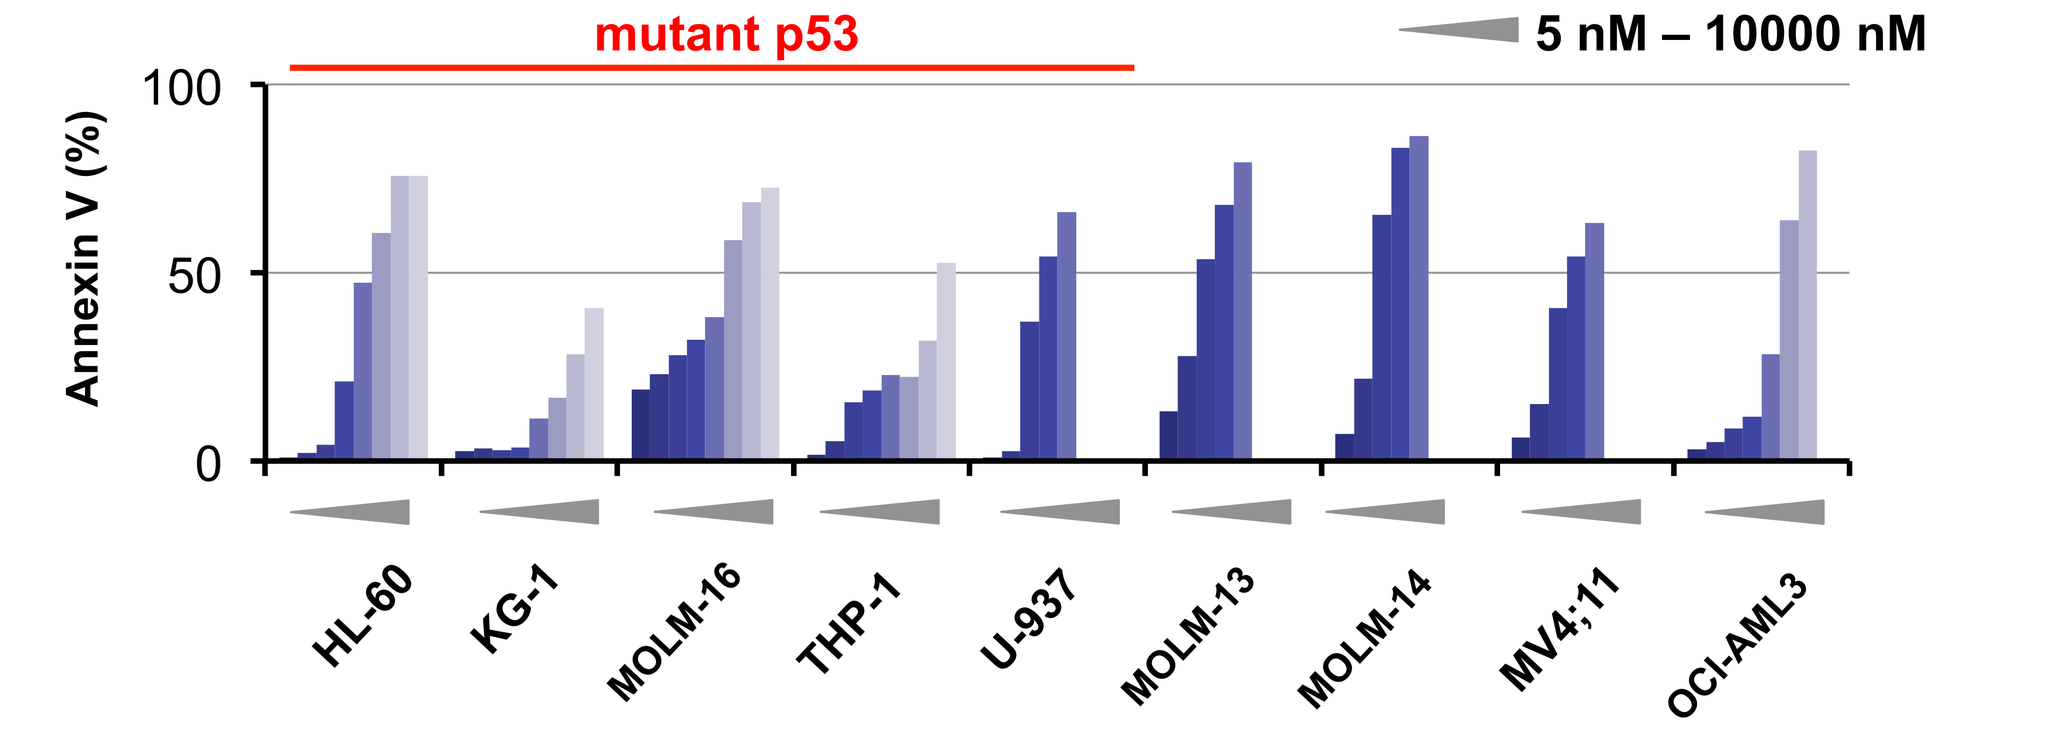
mutant p53
5 nM – 10000 nM
Annexin V (%)
HL-60 KG-1 MOLM-16 THP-1 U-937 MOLM-13 MOLM-14 MV4;11 OCI-AML3

ABT-199

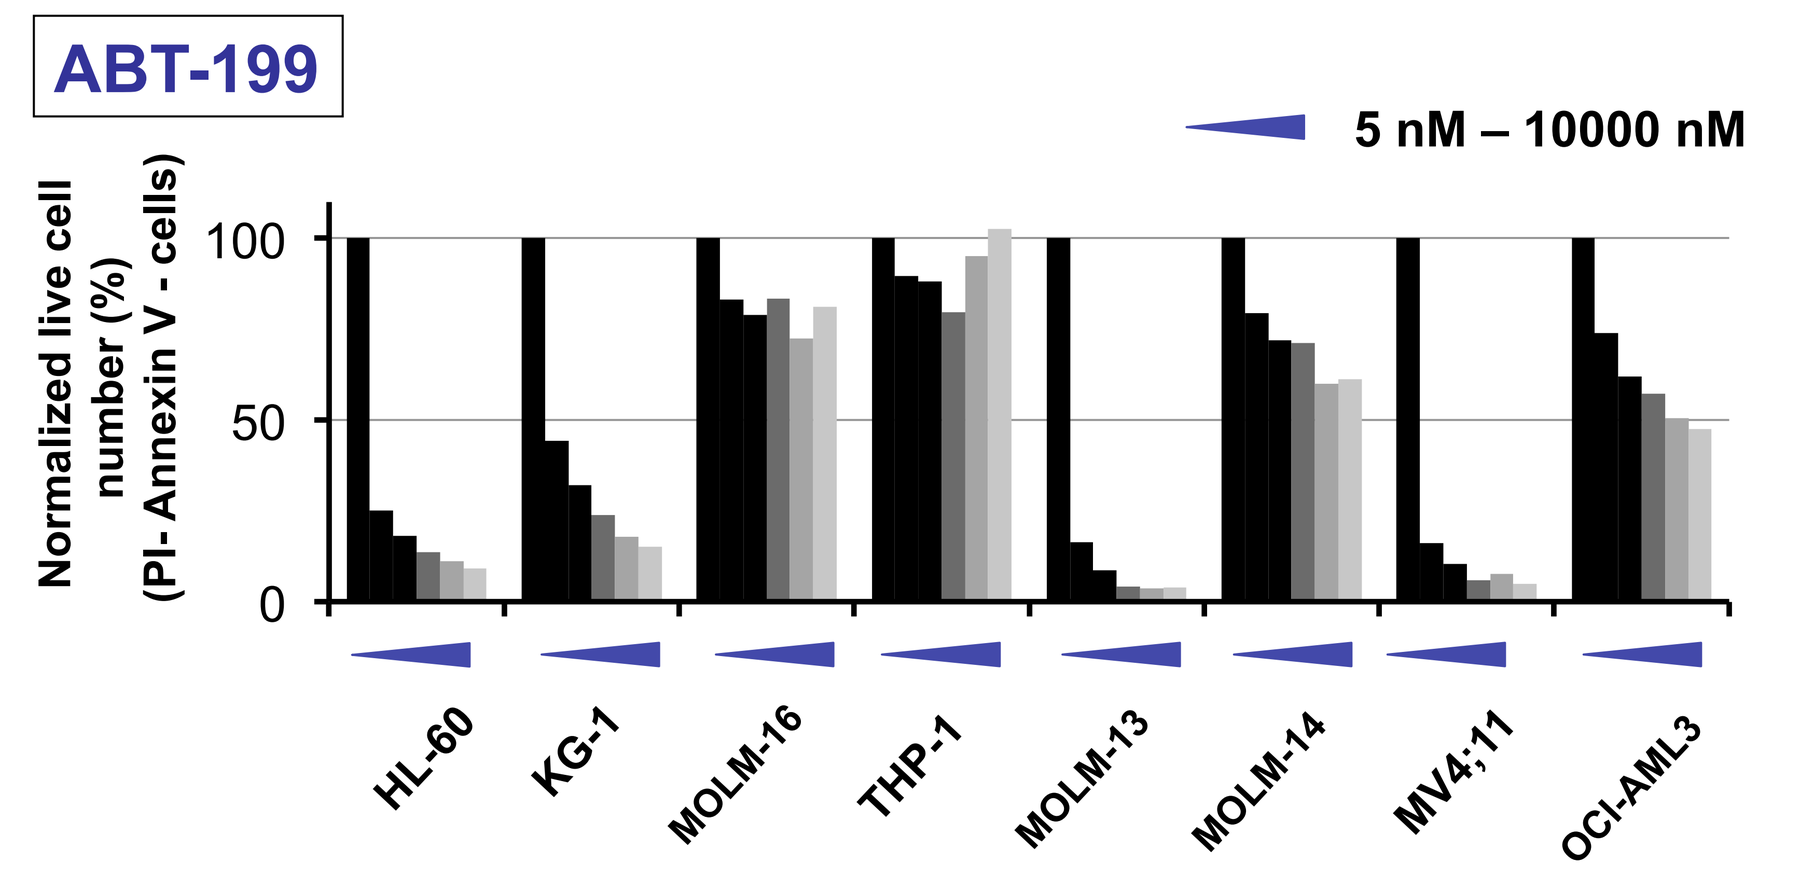
5 nM – 10000 nM
Normalized live cell number (%) (PI- Annexin V - cells)
HL-60 KG-1 MOLM-16 THP-1 MOLM-13 MOLM-14 MV4;11 OCI-AML3

p53-dependency in induced apoptosis by each agent

Apoptosis induced by Nutlin-3a and KPT-330 (partially) is p53-dependent.

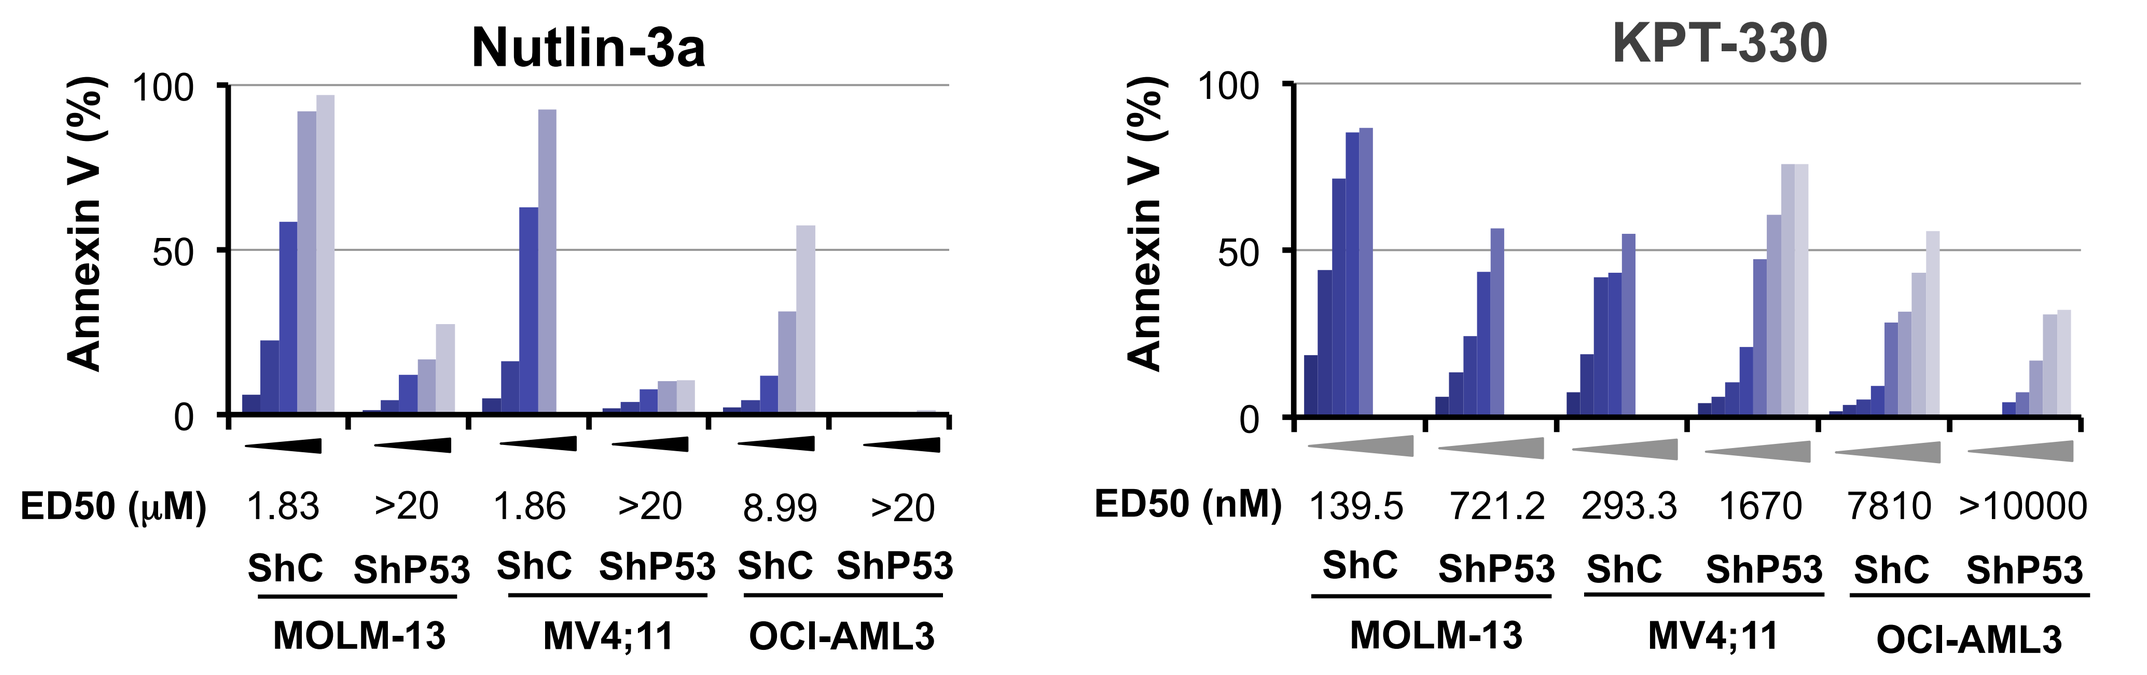
Nutlin-3a
KPT-330
Annexin V (%)
ED50 (μM) 1.83 >20 1.86 >20 8.99 >20
ED50 (nM) 139.5 721.2 293.3 1670 7810 >10000
ShC ShP53 ShC ShP53 ShC ShP53
MOLM-13 MV4;11 OCI-AML3

Apoptosis induced by Ara-C or ABT-199 is p53-independent.

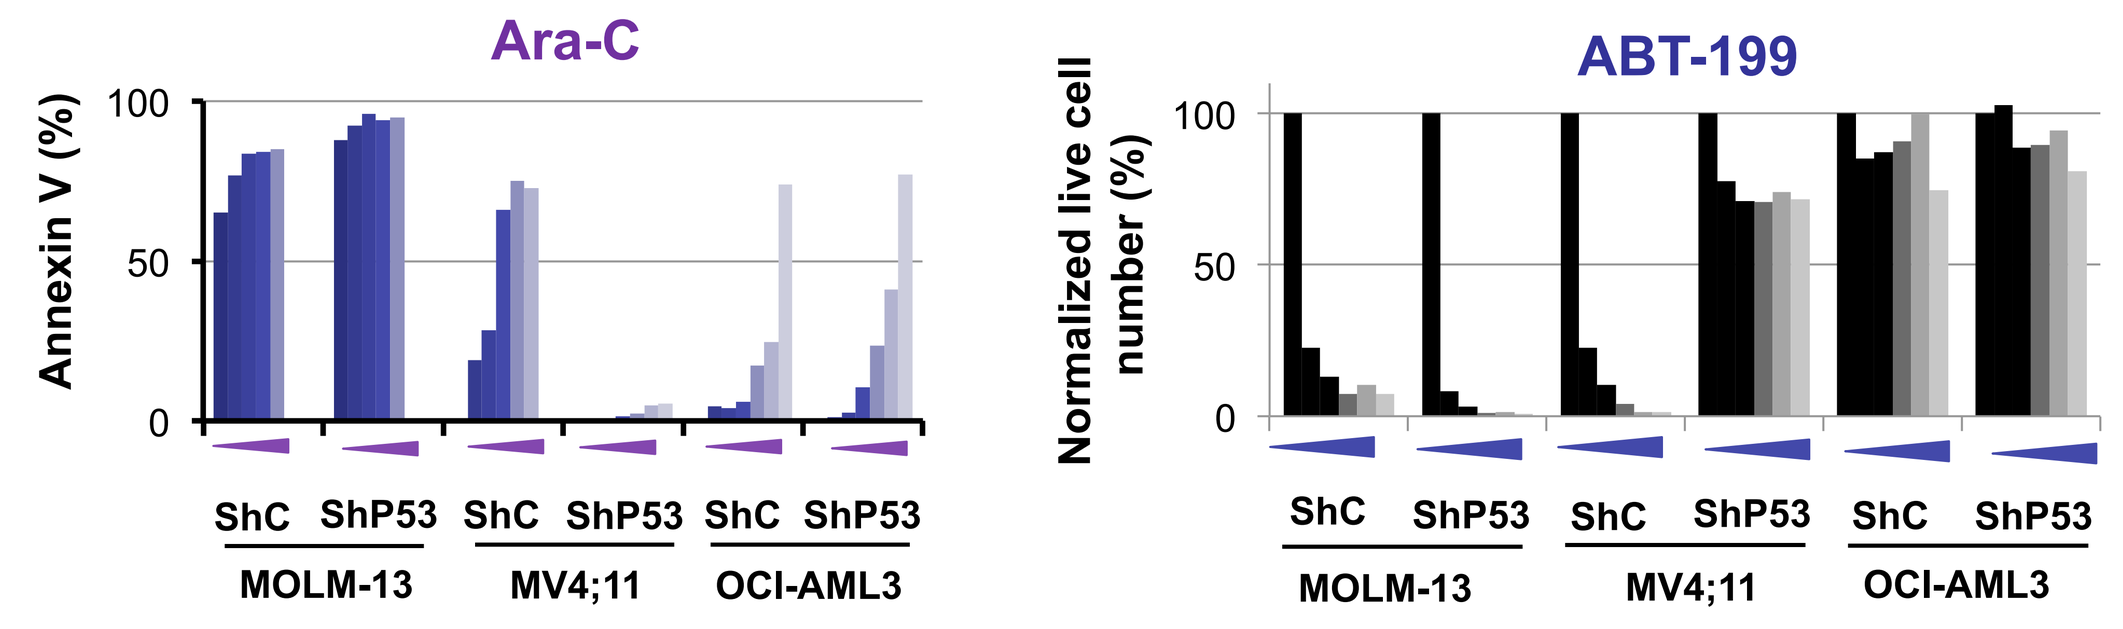
Ara-C
ABT-199
Annexin V (%)
Normalized live cell number (%)
ShC ShP53 ShC ShP53 ShC ShP53
MOLM-13 MV4;11 OCI-AML3

BH3 profiling and correlation with drug sensitivity

BH3 profiling in 22 AML cell lines.

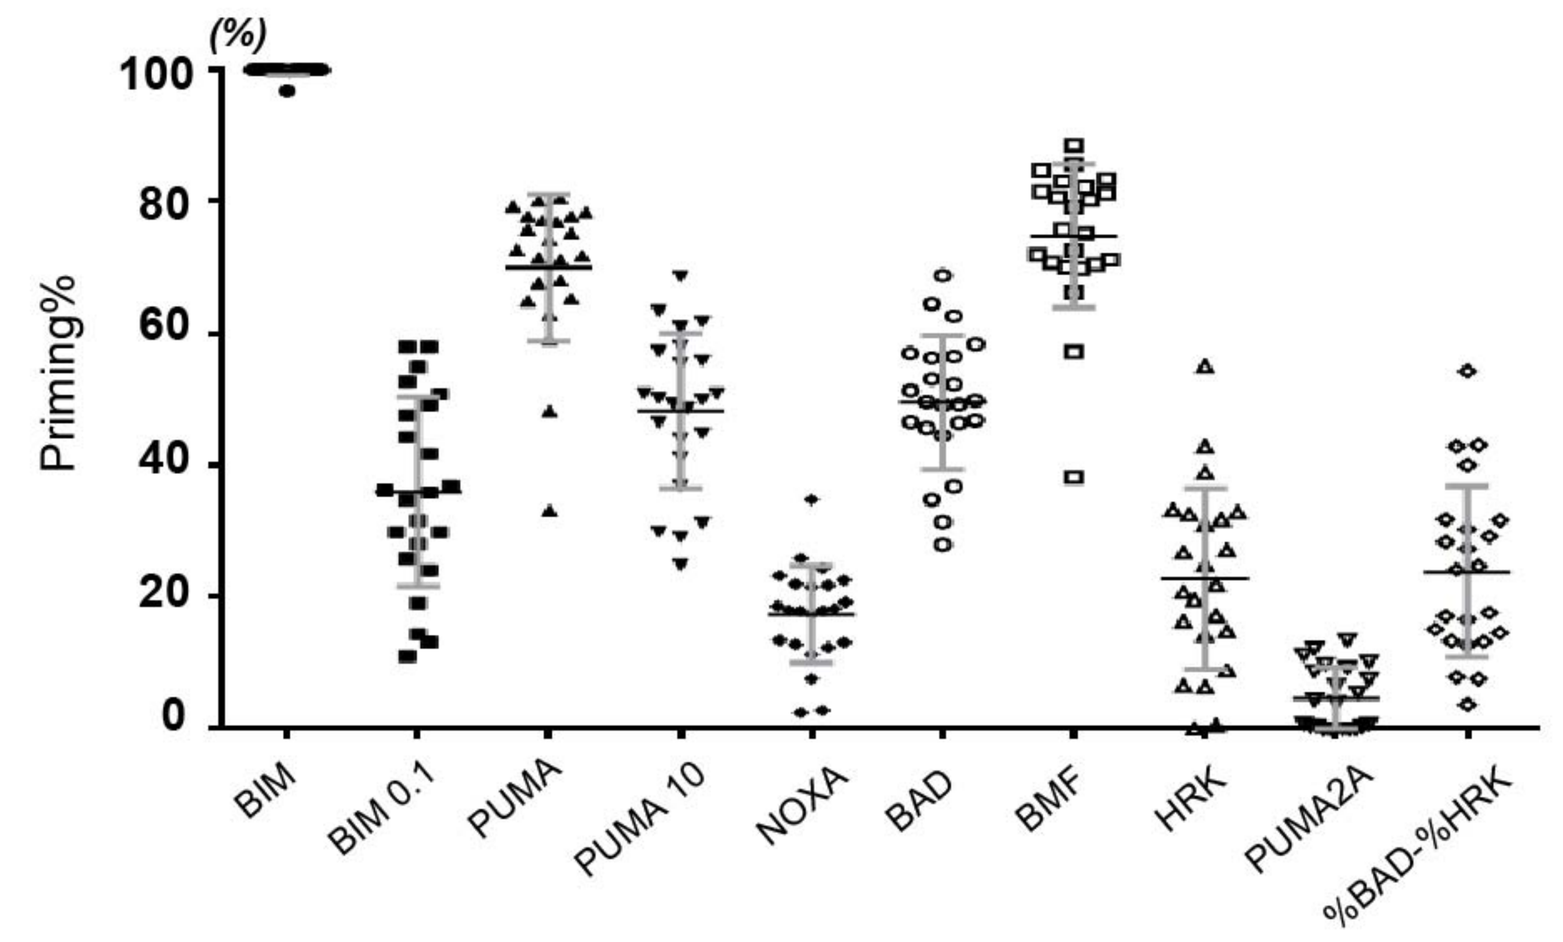
Priming%
BIM BIM 0.1 PUMA PUMA 10 NOXA BAD BMF HRK PUMA2A %BAD-%HRK

[%BAD - %HRK] can predict BCL-2 dependency (the sensitivity to ABT-199), and Ara-C-induced apoptosis also revealed BCL-2 dependency.

Table 1. Multivariate Mixed linear models

	Nutlin-3a		Ara-C		KPT-330		ABT-199*	
	β	p-value	β	p-value	β	p-value	β	p-value
%BIM 0.1	0.1	0.671	0.13	0.83	-0.04	0.921	-2.75	0.056
% PUMA 10	0.13	0.661	-0.34	0.642	0.78	0.088	0.38	0.734
% NOXA	0.34	0.555	0.95	0.506	1.04	0.124	2.39	0.249
% BAD	0.27	0.412	-0.26	0.756	0.08	0.875	-1.06	0.31
% BMF	0.34	0.199	-1	0.126	-0.54	0.453	1.98	0.271
% HRK	0.24	0.293	-1.05	0.056	0.49	0.22	0.76	0.366
% BAD - % HRK	-0.18	0.56	1.61	0.022	-0.88	0.109	3.22	<0.001

* normalized dead cell number

Baseline p53 levels do not affect mitochondrial priming.

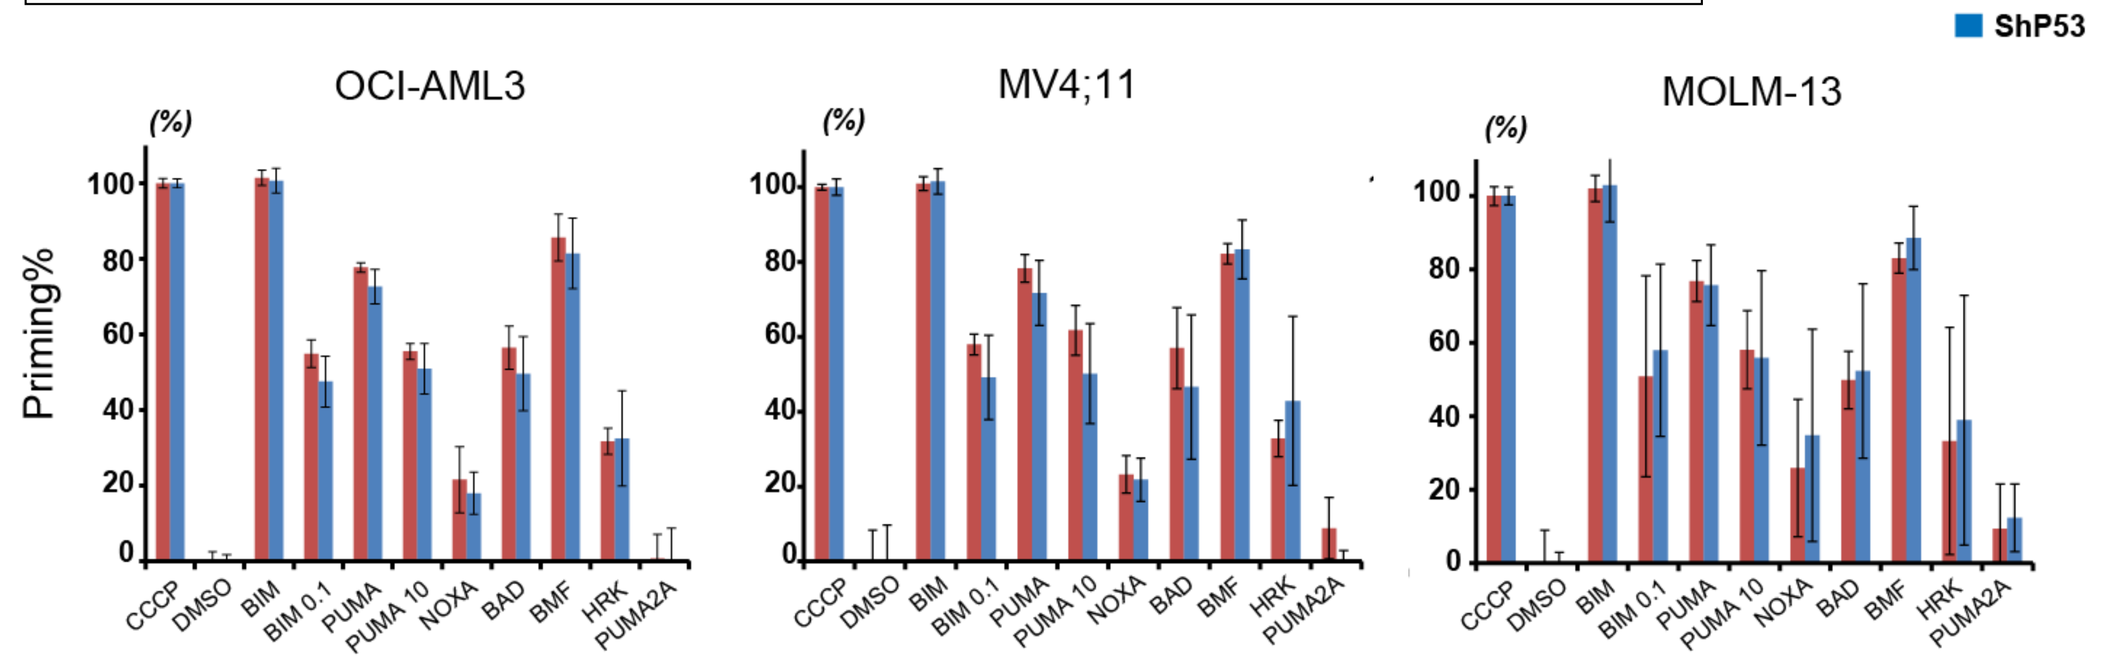
ShC
ShP53
OCI-AML3
MV4;11
MOLM-13
Priming%

BCL-2 levels are predictive for mitochondrial priming but do not always correlate with cell death in AML lines

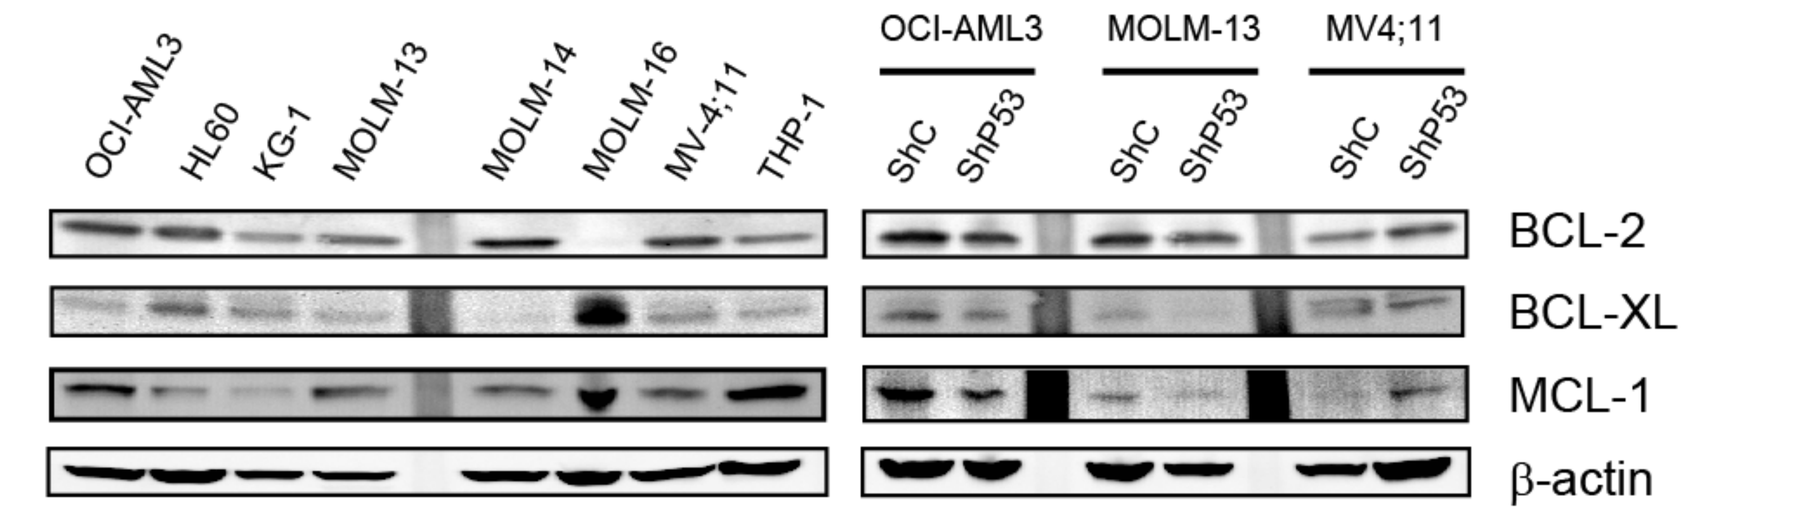
OCI-AML3 HL60 KG-1 MOLM-13 MOLM-14 MOLM-16 MV-4;11 THP-1
OCI-AML3 MOLM-13 MV4;11
ShC ShP53
BCL-2
BCL-XL
MCL-1
β-actin

BCL-2 protein expression correlated with %[BAD] - %[HRK], but does not significantly correlate with cell death.

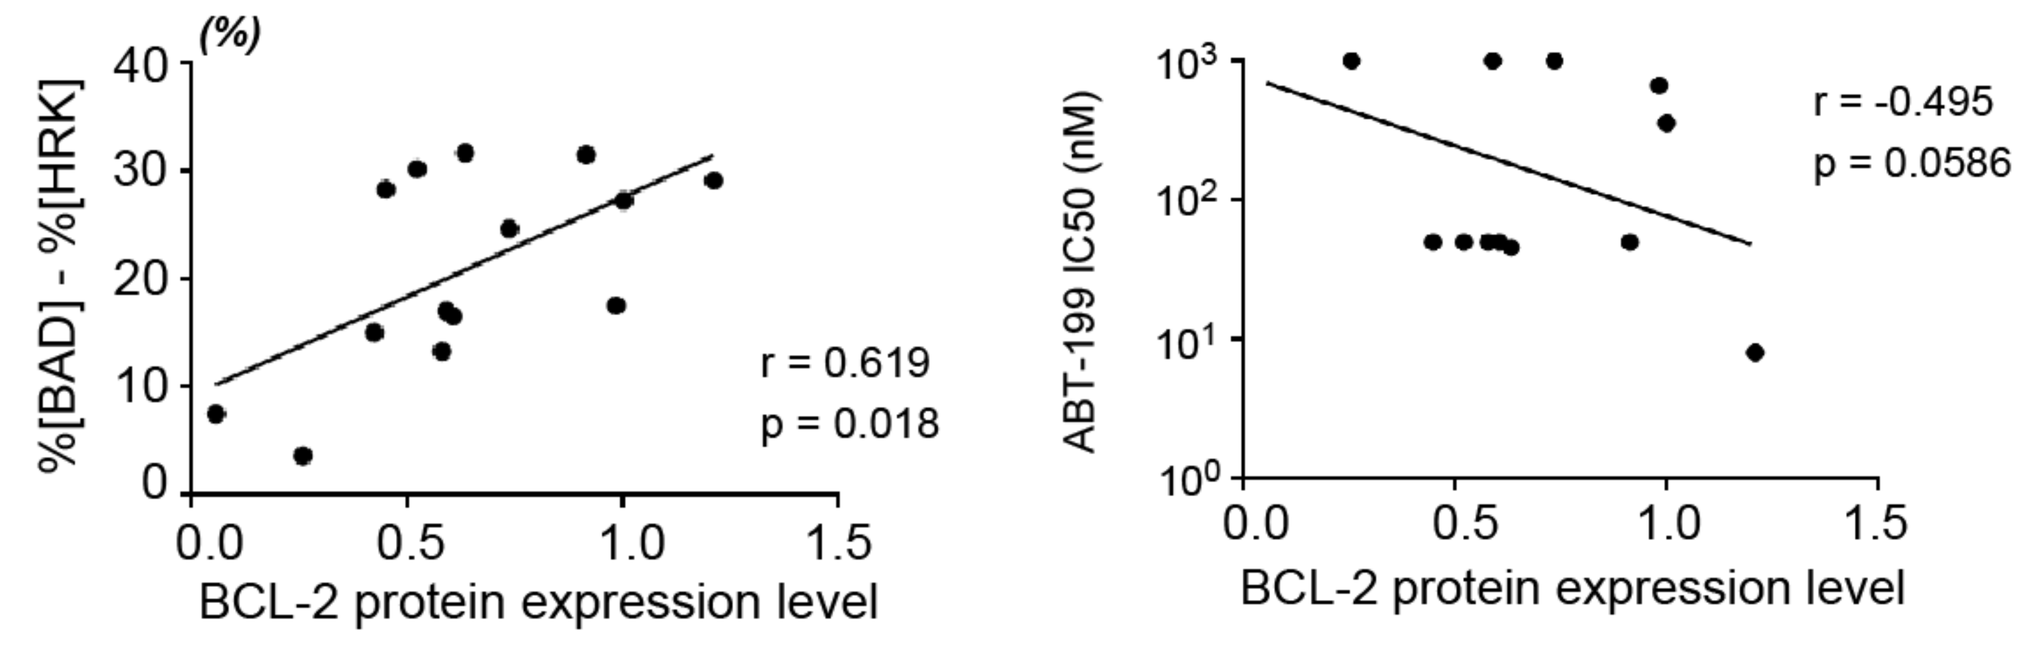
%[BAD] - %[HRK]
r = 0.619
p = 0.018
BCL-2 protein expression level
ABT-199 IC50 (nM)
r = -0.495
p = 0.0586
BCL-2 protein expression level

MCL-1 protein expression predicts resistance to ABT-199 and Ara-C.

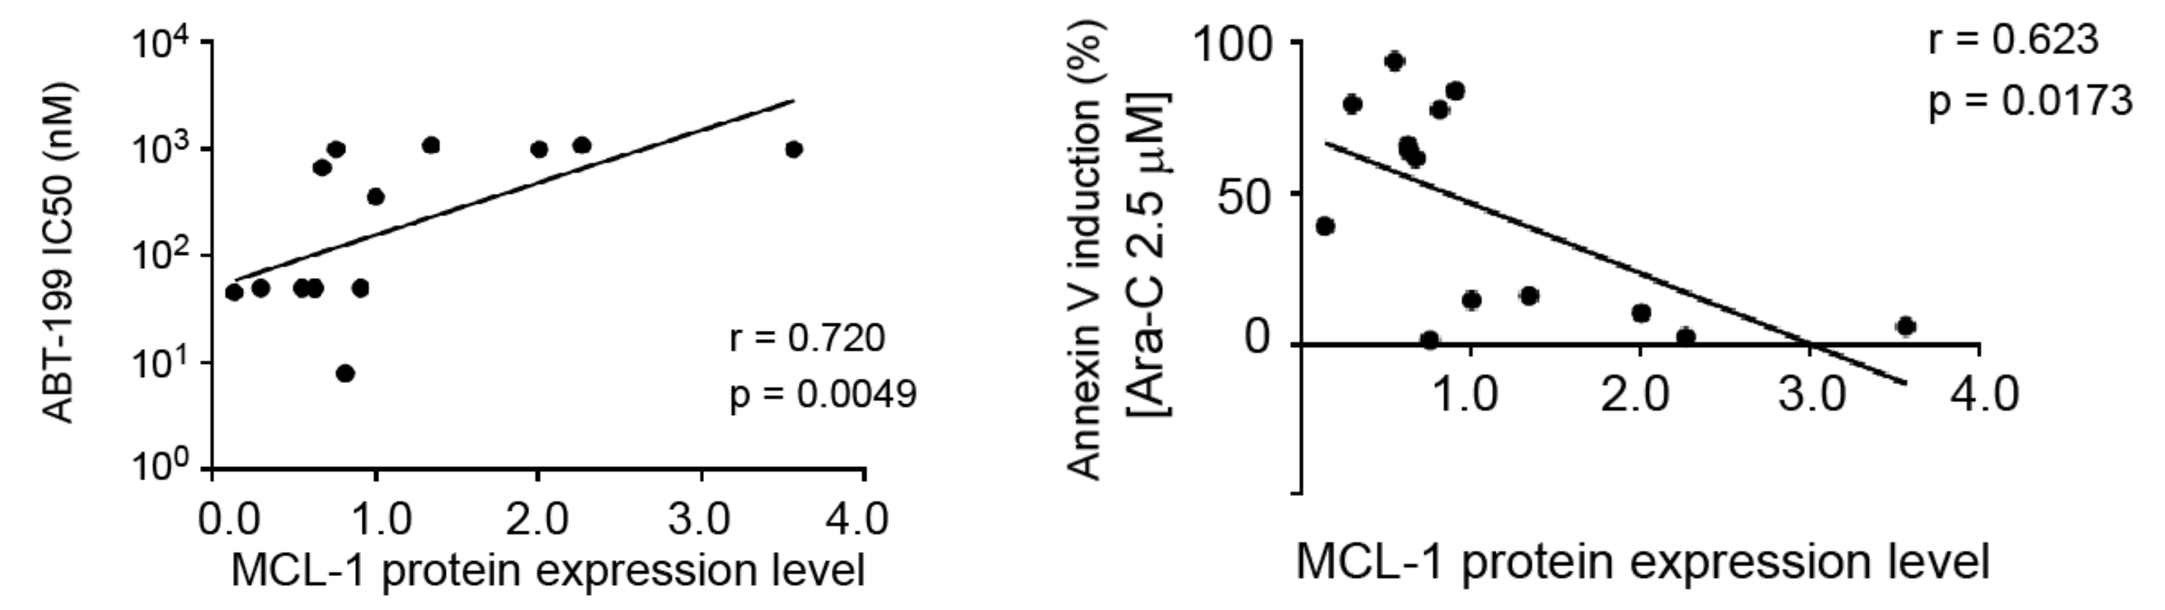
ABT-199 IC50 (nM)
r = 0.720
p = 0.0049
MCL-1 protein expression level
Annexin V induction (%) [Ara-C 2.5 μM]
r = 0.623
p = 0.0173
MCL-1 protein expression level

Changes in BCL-2 family protein expression variably alter the senstivity to each agent.

MCL-1 silencing with shRNA increases sensitivity to ABT-199 and Ara-C induced apoptosis.

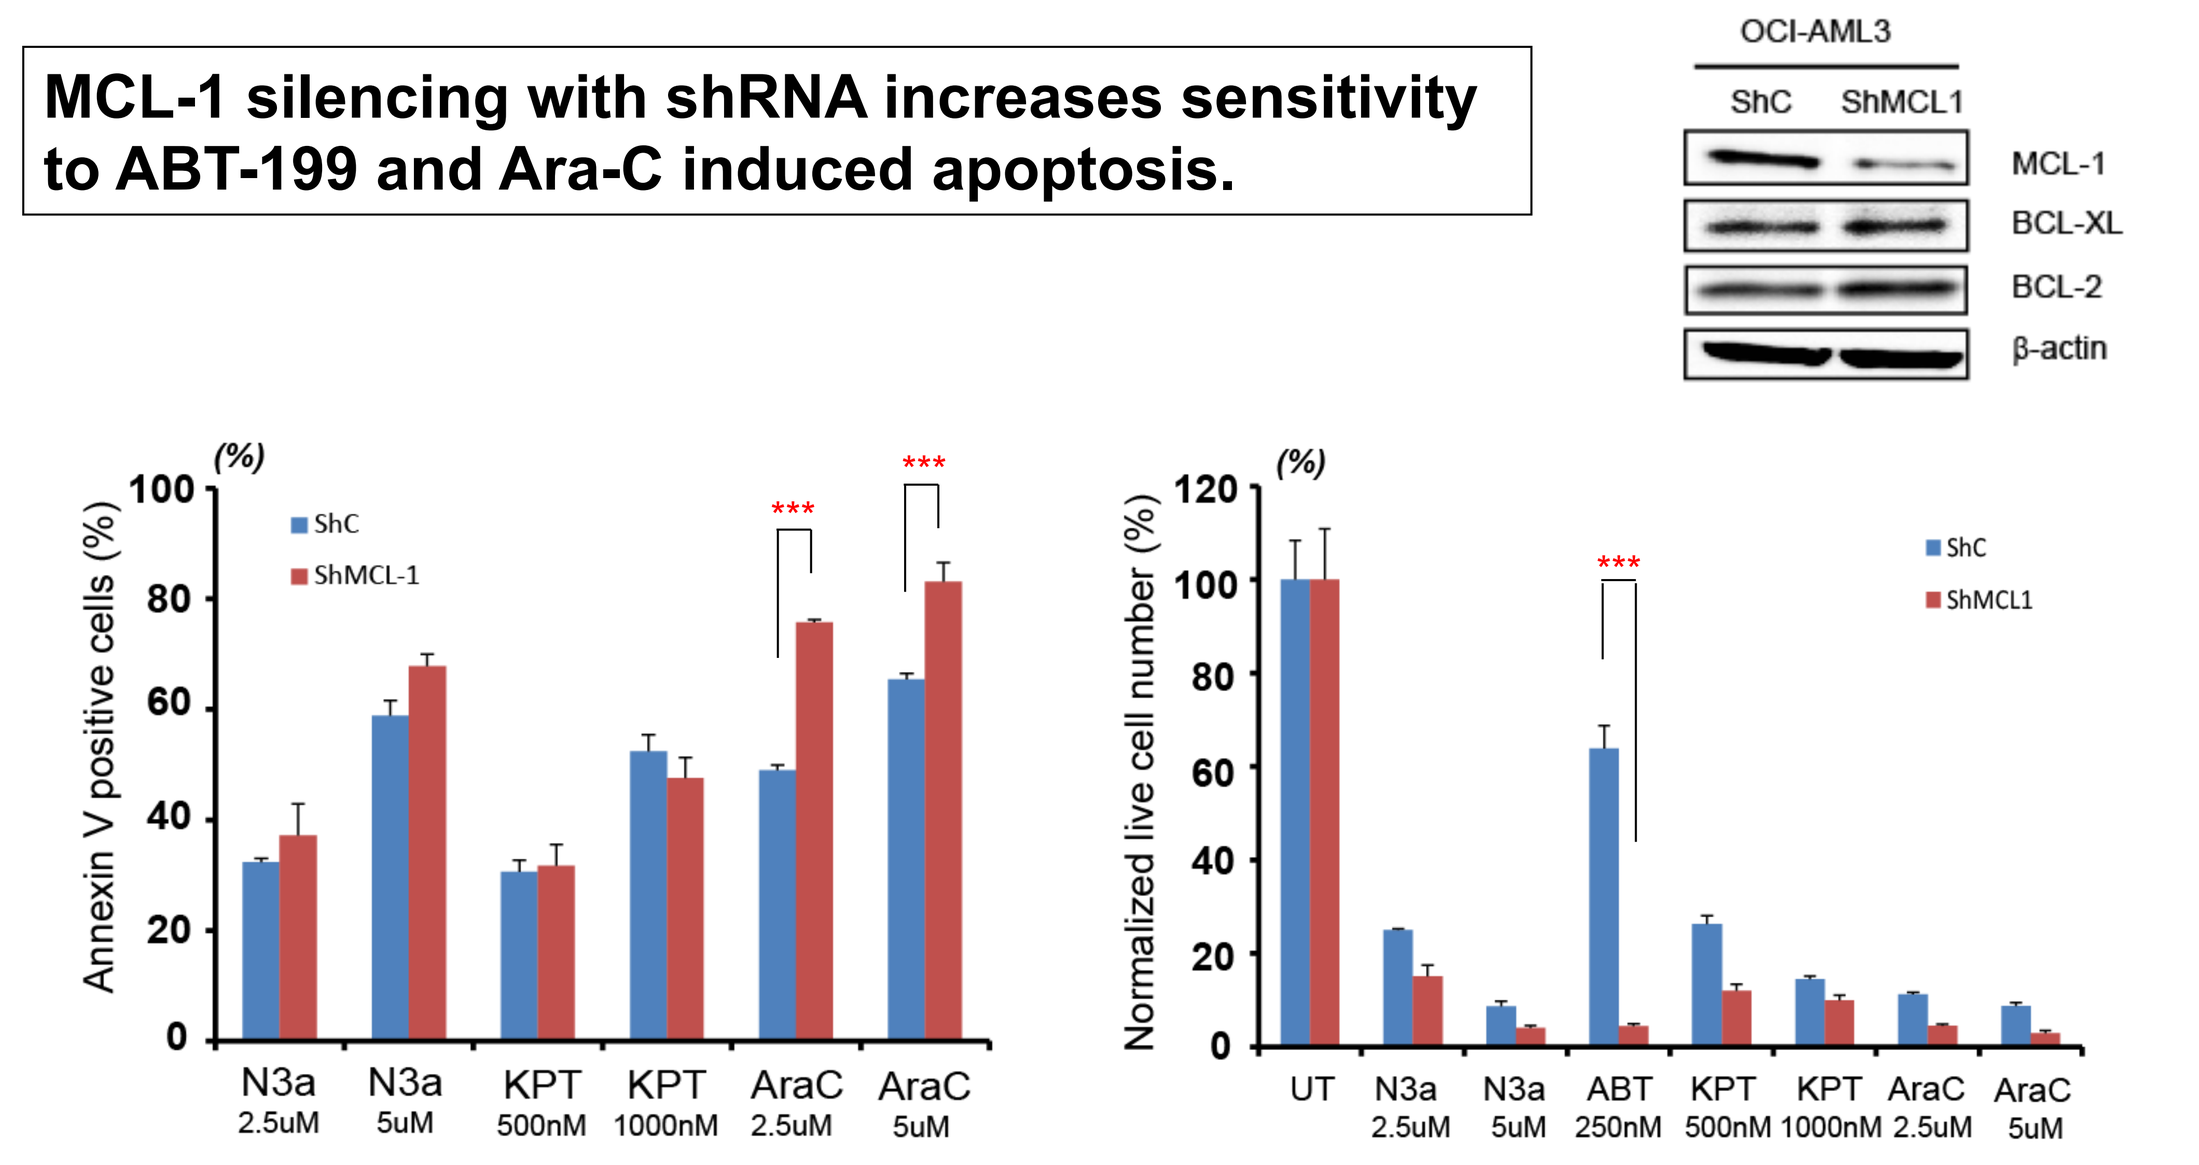
OCI-AML3
ShC ShMCL1
MCL-1
BCL-XL
BCL-2
β-actin
Annexin V positive cells (%)
Normalized live cell number (%)
UT N3a 2.5uM N3a 5uM ABT 250nM KPT 500nM KPT 1000nM AraC 2.5uM AraC 5uM

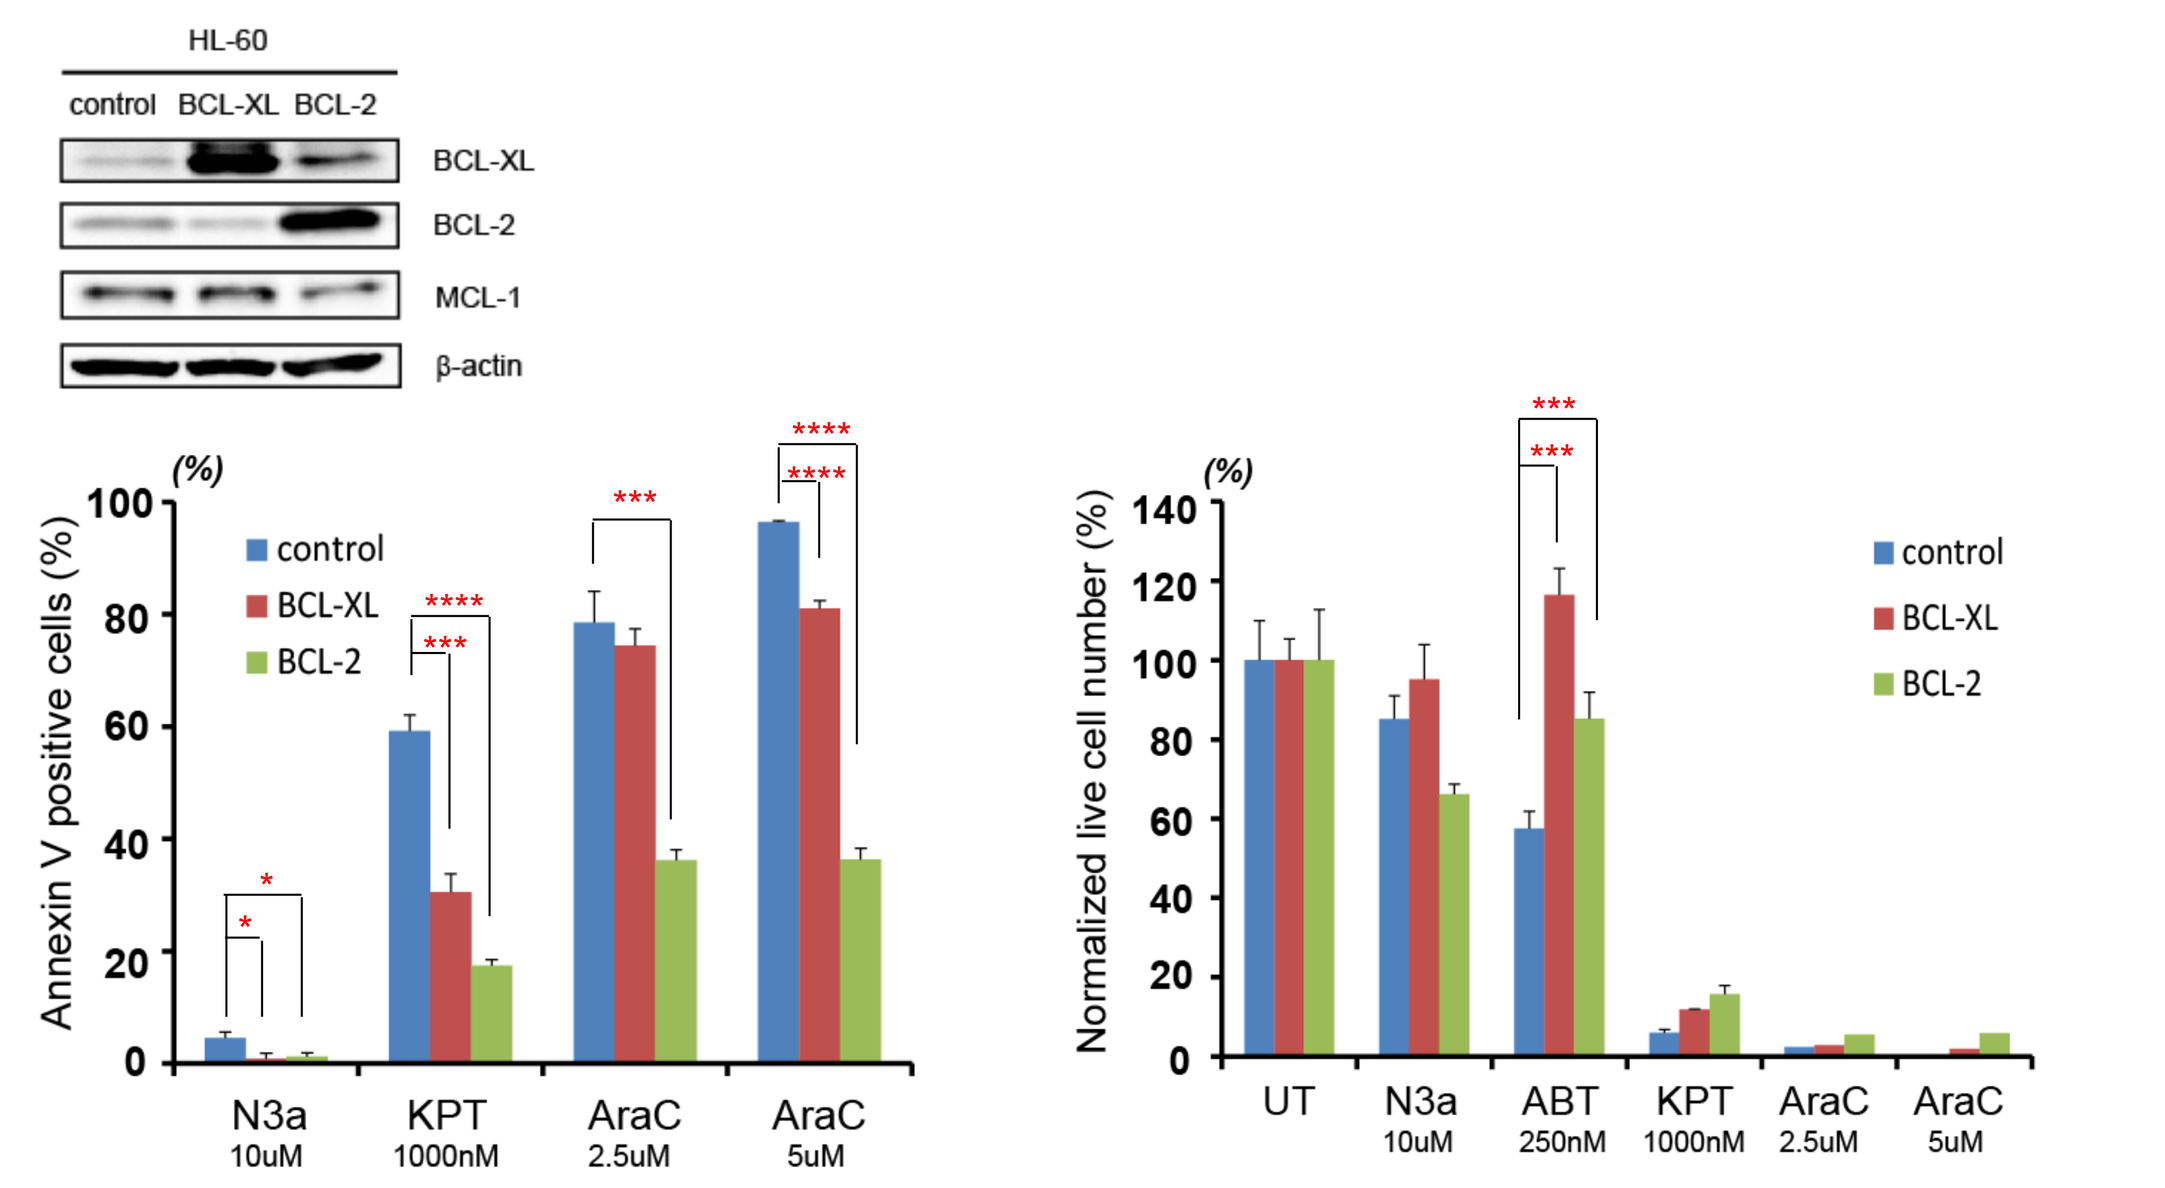
HL-60
control BCL-XL BCL-2
BCL-XL
BCL-2
MCL-1
β-actin
Annexin V positive cells (%)
control
BCL-XL
BCL-2
N3a 10uM KPT 1000nM AraC 2.5uM AraC 5uM
Normalized live cell number (%)
UT N3a 10uM ABT 250nM KPT 1000nM AraC 2.5uM AraC 5uM

BCL-2 overexpression is associated with Ara-C resistance and BCL-2/XL with resistance to KPT-330.

BCL-XL overexpression is associated with ABT-199 resistance.

Conclusion

1. **BH3 profiling (%[Bad]-%[HRK]) is highly predictive for cell sensitivity to BCL-2 dependent apoptosis by ABT-199.**
2. **Ara-C-induced apoptosis unexpectedly revealed BCL-2 dependency, similar to ABT-199.**
3. **BH3 profiling does not predict for Nutlin-induced p53-mediated apoptosis or KPT-induced apoptosis.**
4. **BCL-2 protein levels correlate with BCL-2 - dependent mitochondrial priming.**
5. **BH3 profiling is superior to BCL-2 protein level in terms of predicting cell sensitivity to BCL-2 dependent apoptosis by ABT-199.**
6. **MCL-1 protein expression predicts resistance to ABT-199 and Ara-C.**
7. **BCL-2 overexpression is associated with Ara-C or KPT-330 resistance, and BCL-XL overexpression confers resistance to KPT-330 and ABT-199.**

BH3 profiling in conjunction with BCL-2 family proteins expression is a promising tool to predict the BH3 peptide dependency of chemotherapeutic agents.

Acknowledgements

This study was supported in part by grants from National Institutes of Health Lymphoma SPORE (CA136411), Leukemia SPORE (CA100632), P01 "The Therapy of AML" (CA55164), Cancer Center Support Grant (CA16672) and the Paul and Mary Haas Chair in Genetics (to MA).
It is also partially supported by Japan Society for the Promotion of Science, Postdoctoral Fellowship For Research Abroad (to JI).

Conflict of Interest

WP, MC, RL and CD are [illegible] Eutropic[illegible]SS and MK are employees of Karyopharm. [illegible] has research support from Karyopharm and is a member of [illegible] Eutropics.