Single cell longitudinal studies reveal cell cycle specific effects of selective inhibitors of nuclear export (SINE)

Russell T. Burke[1], Joshua M. Marcus[1], John A. DeSisto[1], Yosef Landesman[2], James D. Orth[1]

[1]University of Colorado - Boulder, Boulder, CO; [2]Karyopharm Therapeutics, Inc., Natick, MA

Abstract #1760

Background and Rationale

Anti-cancer responses to small molecule drugs or natural products are determined on the molecular and cellular scale. Understanding cell responses and fates following treatment using population average assays (e.g. immunoblotting), masks cell-cell variability and differences in timing, and discounts transient and rare responses. To more completely understand the complexity of drug response we must track molecular responses and cell fate choices simultaneously in individual cells in real time. The use of long-term longitudinal approaches to follow a given single cell or a cell population is a less common but very powerful approach that allows for the direct study of molecular response pathways, different phenotypes (e.g. cell death or cell division), observation of cell-to-cell variability within a population, and how these factors contribute to population response dynamics. XPO-1 inhibitors have been developed and are being evaluated as anti-cancer agents in the clinical setting. Termed selective inhibitor of nuclear export (SINE), these agents covalently bind XPO-1 and block XPO-1-dependent nuclear export of cargo proteins, including p53, p21 and p27. SINE effects on the cell cycle have been noted, but the effect(s) on the progression of individual cells, variability between cells and cell lines, and how these impact the overall response are unclear.

Abstract

Nuclear export of proteins is fundamental for cell growth and function. Selinexor is a SINE compound that is in clinical development for the treatment of different cancers. Selinexor forms a slowly reversible covalent bond to Exportin-1 (XPO1), preventing its association with protein cargos, thereby resulting in their nuclear retention. XPO1 cargos include the majority of tumor suppressor proteins (TSP) and cell cycle regulators such as p53, p21 and p27 that have key roles in cancer progression and drug response. It is unclear how selinexor affects cell cycle progression in individual cells and the subsequent stress and fate of those cells. To elucidate Selinexor action, we developed cell lines that stably express fluorescent ubiquitin cell cycle reporters (FUCCI), and followed individual cells longitudinally using continuous time-lapse microscopy for 72 hours. We report that in fibrosarcoma-derived HT-1080 cells that express wildtype p53 and p21, 20% of the initial cell population became arrested with >90% in G1- or S-phase and 35% died with nearly 70% from G1- or S-phase after a cell cycle delay or arrest. We also found that 45% of cells divided, but the progeny died or arrested in G1- or S-phase of the next cell cycle – often after cell cycle arrest or slowed cell cycle progression. Using FUCCI, we tracked the response of cells treated acutely in specific cell cycle stages. Cells treated in G1-phase most often arrested or died in G1- or S-phase, whereas cells treated in G2-phase usually progressed to cell division. As FUCCI revealed S-phase progression defects and associated cell death, we further characterized this phenotype. Using nucleotide incorporation with fluorescent detection, we find that as soon as 8 hours after Selinexor fewer cells are undergoing DNA replication and those that are, are doing so inefficiently as both the rate and maximal levels of nucleotide incorporation are significantly reduced. S-phase arrest and progression defects may manifest as DNA double-strand breaks. We find a strong association between S-phase status and DNA damage. In some cells, the damage occurs within hours of Selinexor treatment and appears as a striking cluster of foci. At 8 hours, nearly 50% of cells contain DNA damage clusters. Importantly, the damage clusters sometimes repair as determined by fixed cell time-course analysis and live-cell microscopy. We are exploring the nature of these DNA damage structures, the mechanisms of their formation and repair, and are also testing for redundancy of the cell cycle regulators in determining Selinexor responses. In summary, Selinexor is fast acting, shows cell cycle selectivity, and is highly effective at arresting cell growth and inducing apoptosis in tumor cells. These data suggest that Selinexor may exert anti-cancer effects even on slow growing tumors where the bulk of the cell mass presents a G1-like state and that it likely combines well with other cell cycle targeted therapeutics.

Monitoring cell cycle progression using the FUCCI system

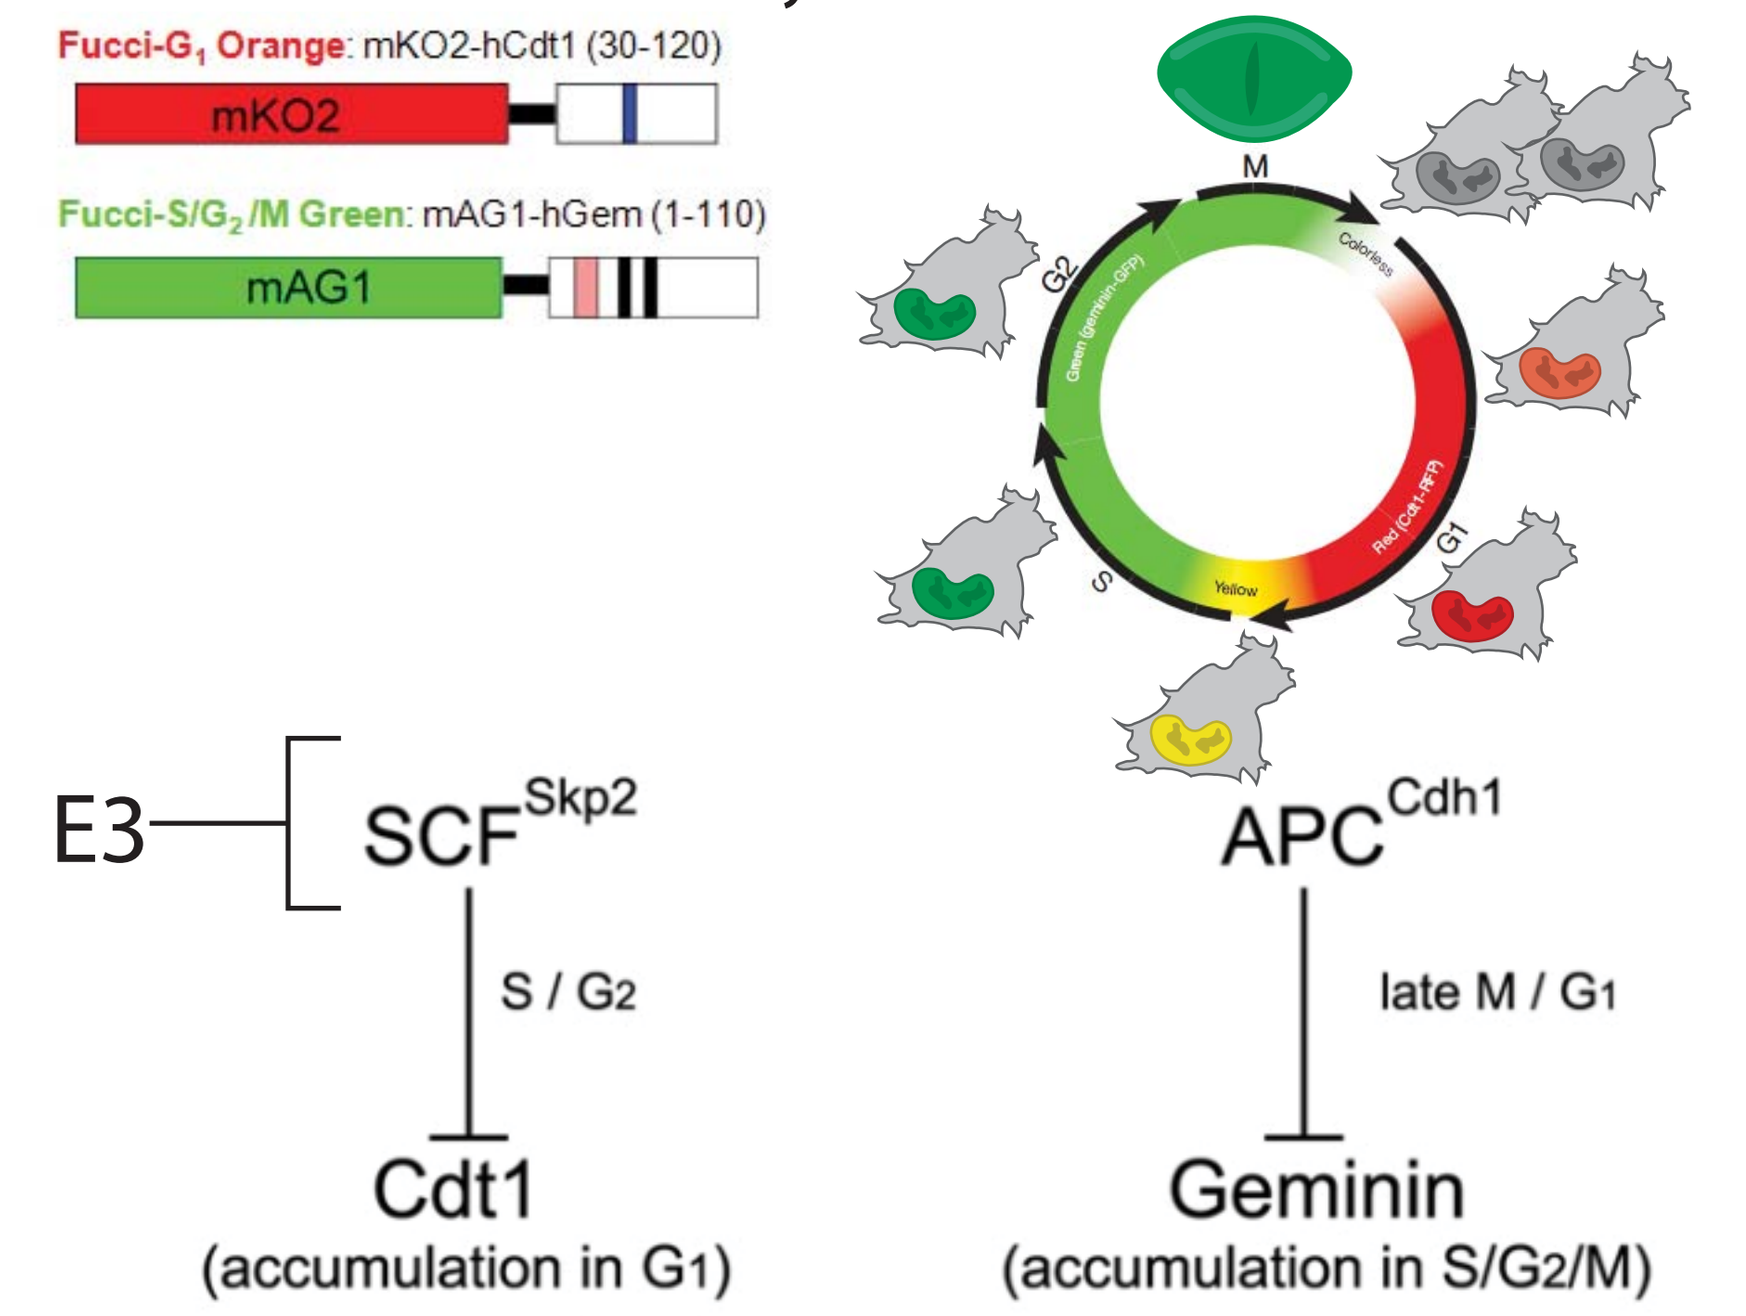
Fucci-G1 Orange: mKO2-hCdt1 (30-120)
mKO2
Fucci-S/G2/M Green: mAG1-hGem (1-110)
mAG1
M
G2
S
G1
E3
SCF^Skp2
S / G2
APC^Cdh1
late M / G1
Cdt1
(accumulation in G1)
Geminin
(accumulation in S/G2/M)

The FUCCI system consists of two fluorescent polypeptide sensors that are expressed and degraded in a cell cycle specific manner. In G1, the red sensor accumulates and remains elevated until progression into S-phase, where it is degraded. Upon progression into S-phase, the green sensor accumulates and remains elevated until cells progress to anaphase in mitosis. Note: when cells are born into G1 they are initially dark, and cells in early to mid-S-phase appear yellow.

Exportin-1 and Nuclear Export

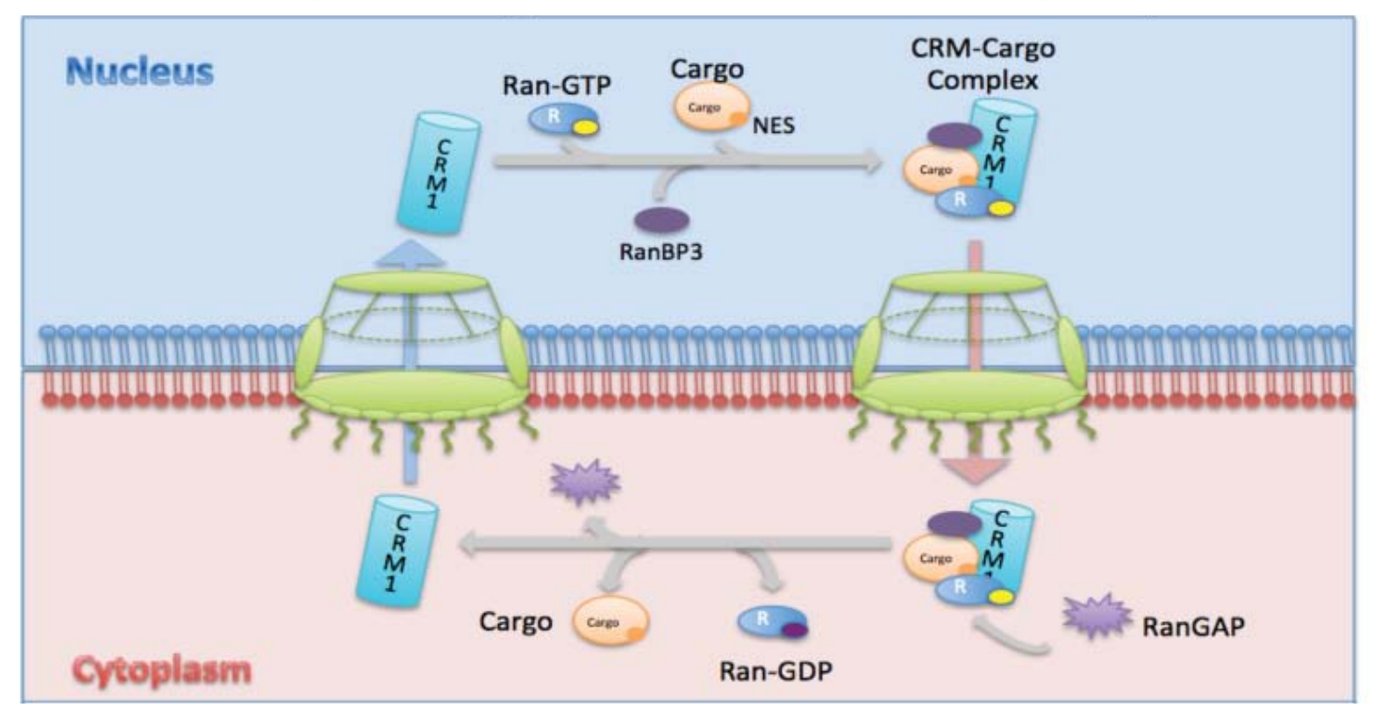
Nucleus
Ran-GTP
Cargo
NES
CRM-Cargo Complex
RanBP3
Cargo
Ran-GDP
RanGAP
Cytoplasm

Governs export of many proteins, including the tumor suppressors p53, p21^CIP1, p27.

XPO-1 and Cancer

- Different cancers show elevated XPO-1, resulting in enhanced nuclear export and decreased function of many tumor suppressor proteins.
- These cancer cells maintain the ability to import XPO-1 cargos, they simply favor cytoplasmic localization.

Selective Inhibitors of Nuclear Export - SINE

SINE, including Selinexor, covalently bind Cys528 of XPO-1 and prevent cargo interaction, resulting in pharmacologically enforced nuclear sequestration and accumulation.

Nuclear Sequestration of RanBP1 following treatement with SINE

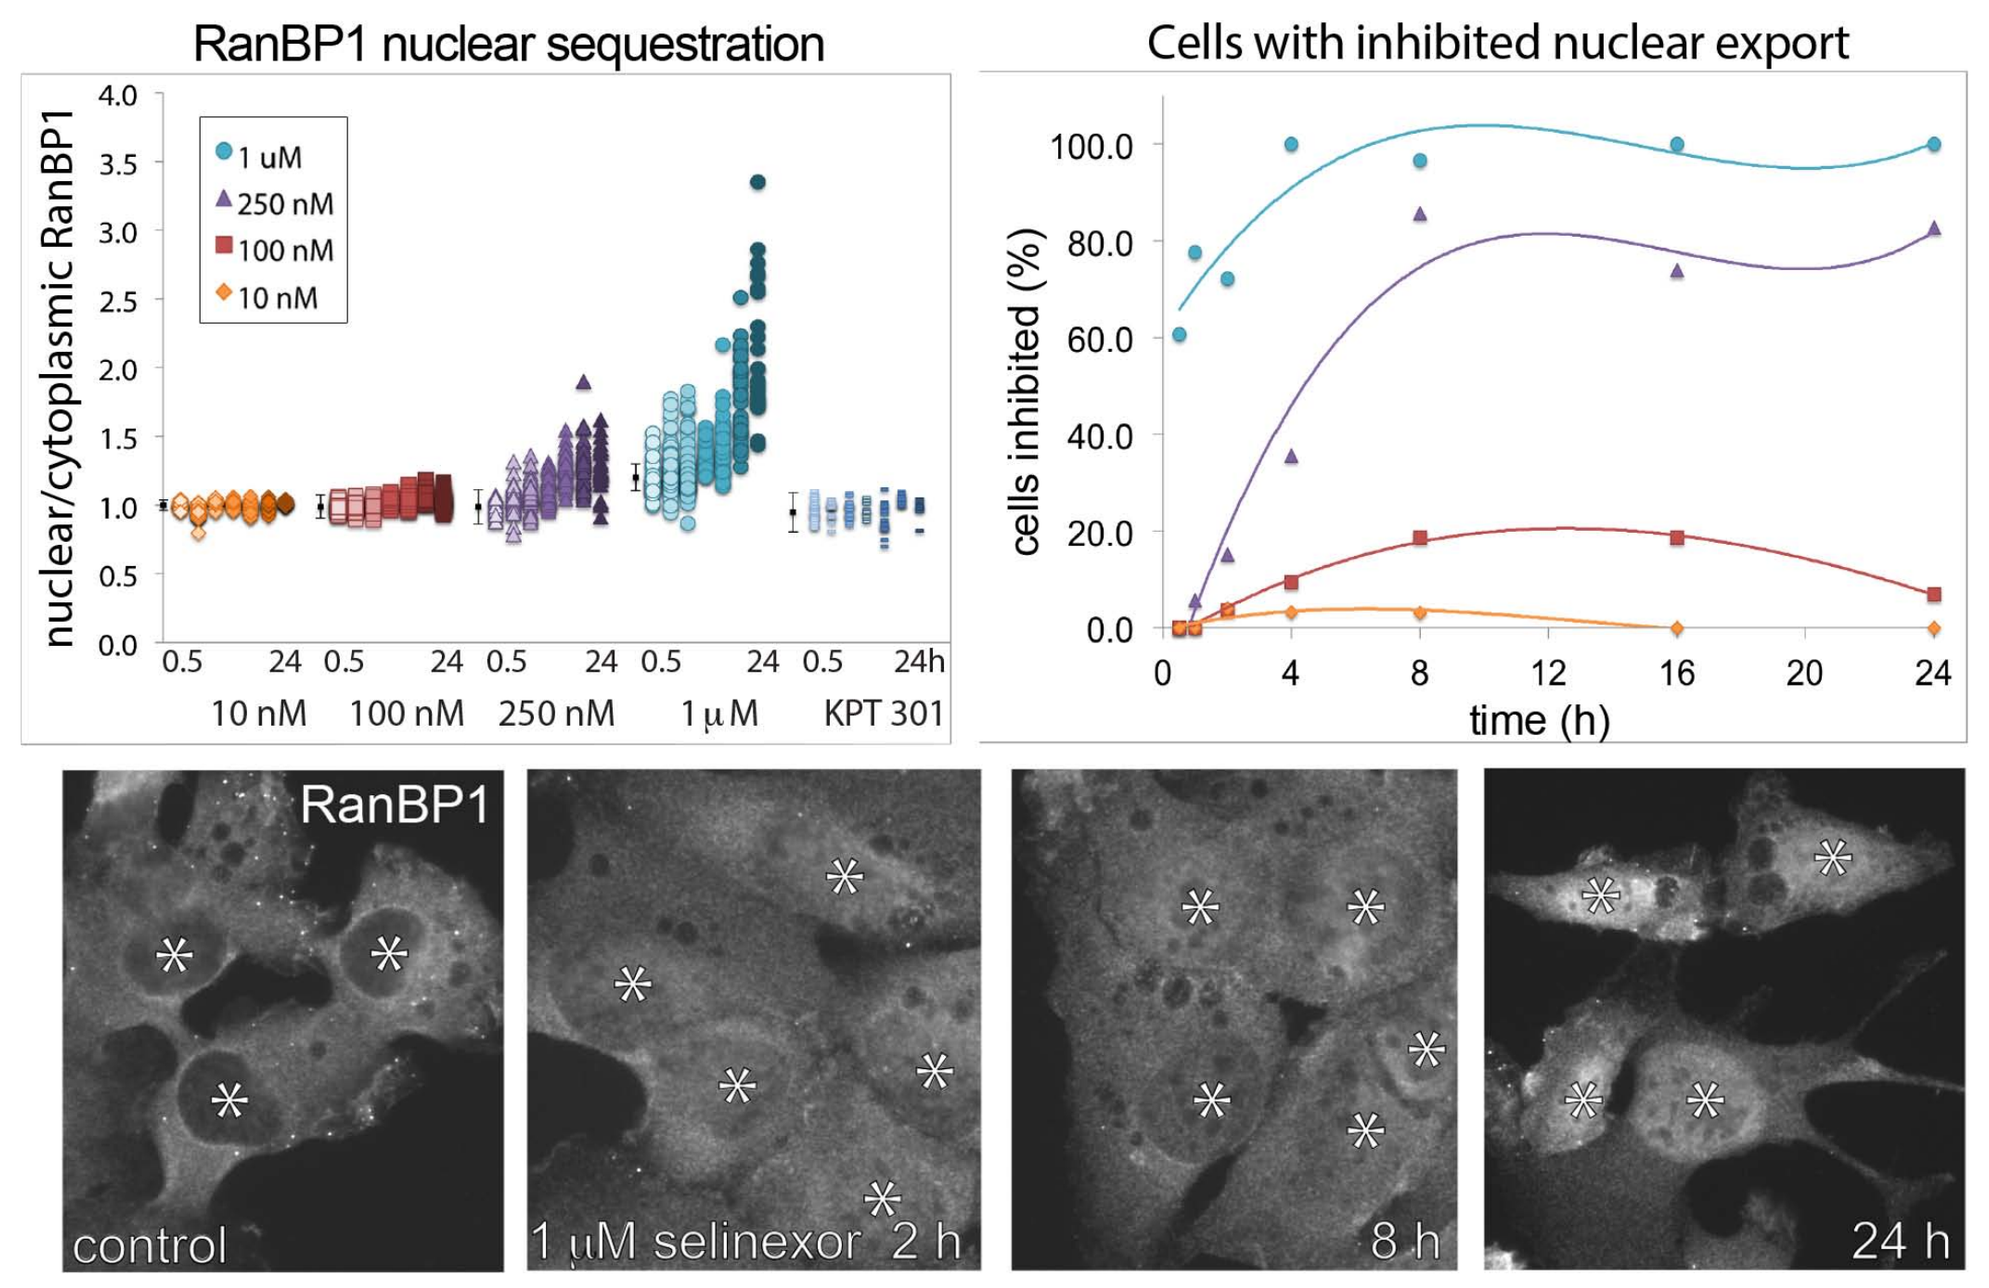
RanBP1 nuclear sequestration
nuclear/cytoplasmic RanBP1
1 uM
250 nM
100 nM
10 nM
0.5 24 0.5 24 0.5 24 0.5 24 0.5 24h
10 nM 100 nM 250 nM 1 µM KPT 301
Cells with inhibited nuclear export
cells inhibited (%)
time (h)
RanBP1
control
1 µM selinexor 2 h
8 h
24 h

Using FUCCI to monitor cells and cell cycle in real time

Figure 1. Untreated HT1080 - FUCCI cells observed from birth.

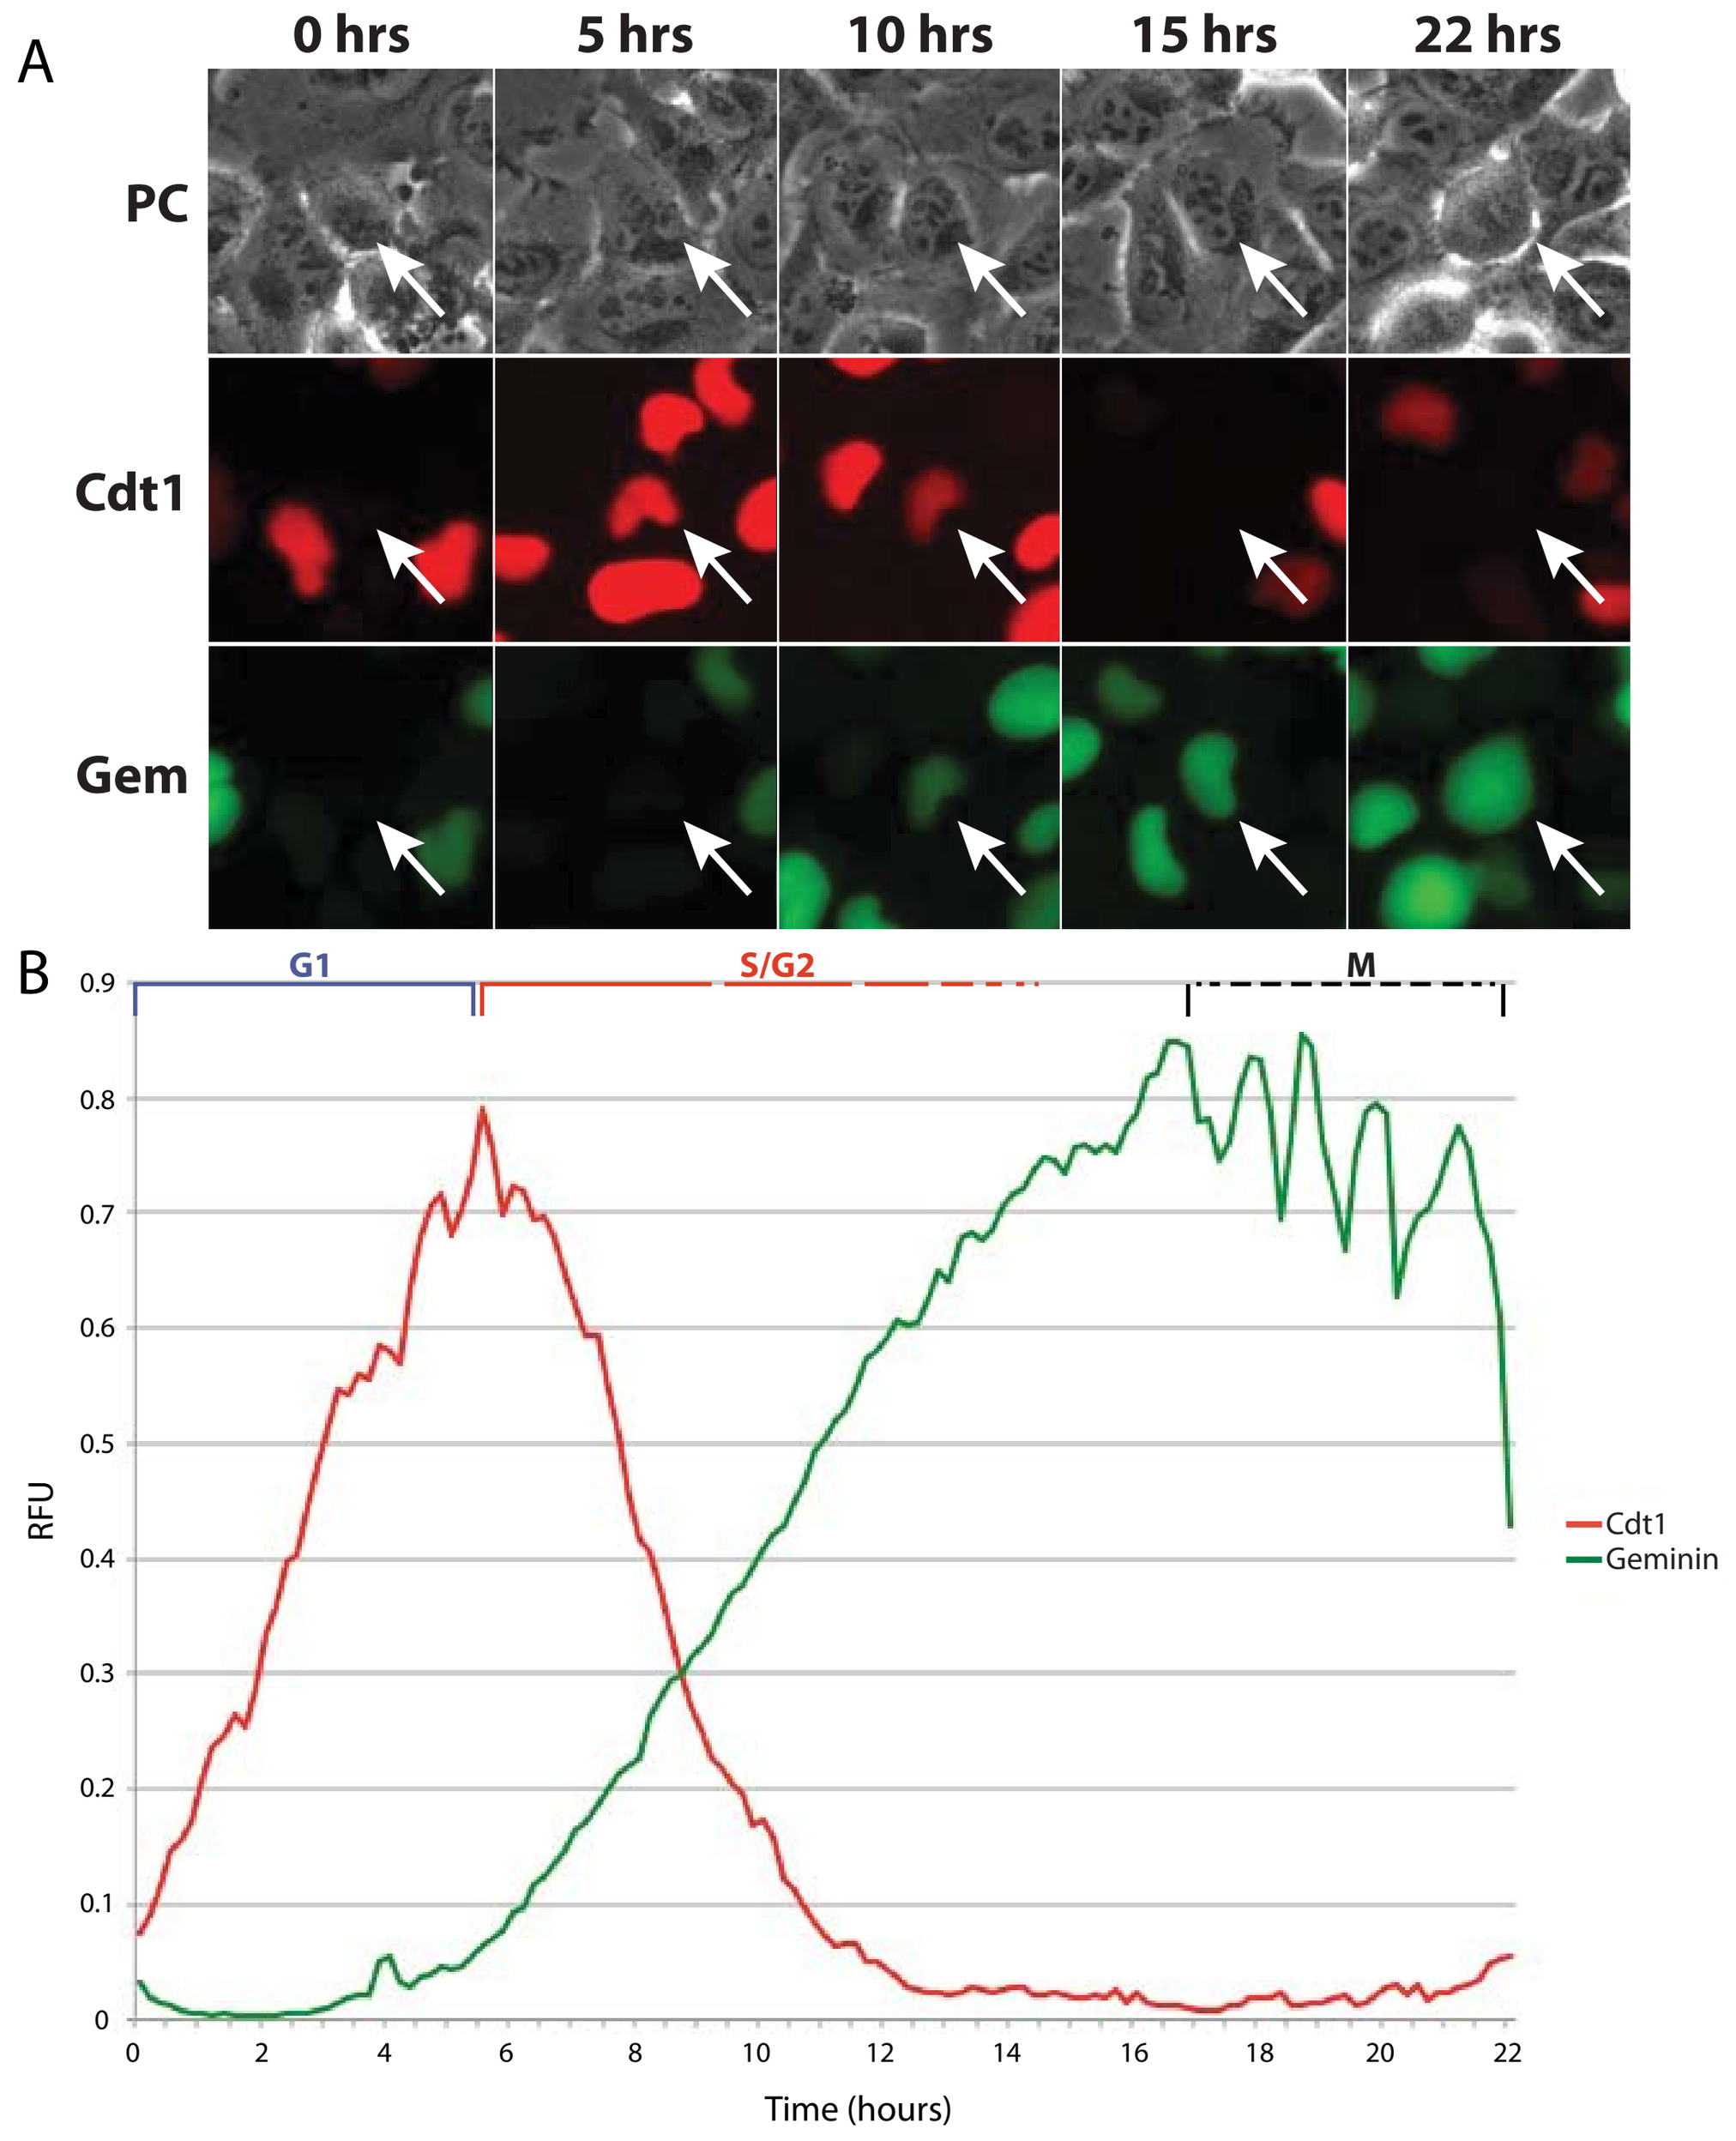
A
0 hrs
5 hrs
10 hrs
15 hrs
22 hrs
PC
Cdt1
Gem
B
G1
S/G2
M
RFU
Time (hours)
Cdt1
Geminin

HT1080 fibrosarcoma cells expressing FUCCI. These untreated cells were tracked from their birth. (A) Cells turned red as they progressed into G1 and the red declined when they progressed into S-phase, as indicated by the increase in green at ≈6 hours. Cells remained green through S-phase and G2, and turned dark upon anaphase onset in mitosis (indicated by the changes in RFU during mitosis). (B) Normal cell cycle times are ≈16-22 hours. Average time in mitosis is ≈1.5 hours. RFU = relative fluorescent units. Arrows indicate cell of interest. Graph is an average 10 cells. PC = phase contrast microscopy.

What are the cell fates and cell cycle responses after Selinexor treatment?

Figure 2. Selinexor results in strong, but highly variable cell cycle and cell death phenotypes.

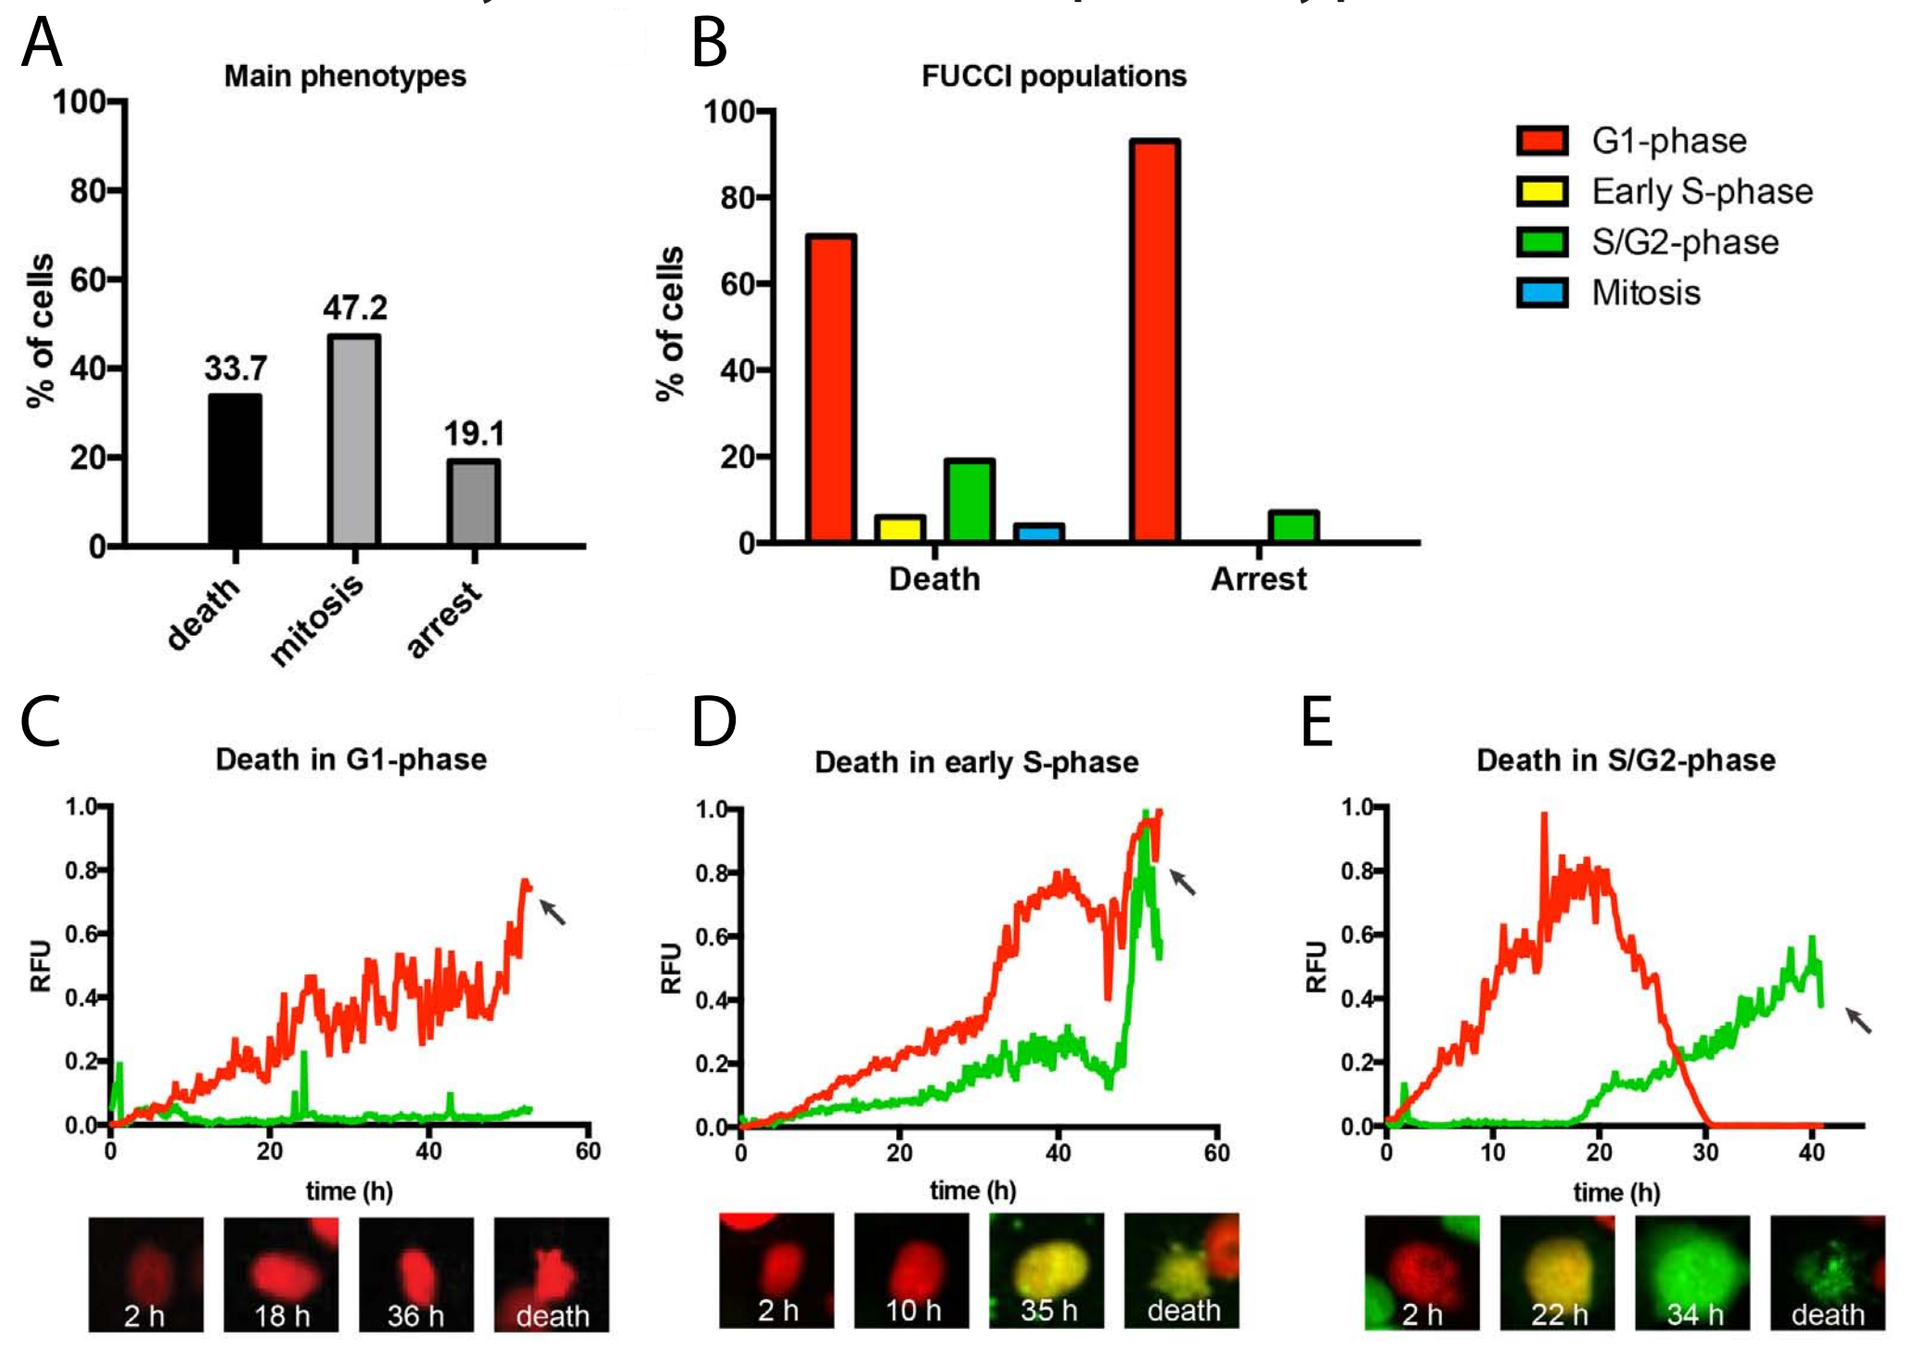
A
Main phenotypes
% of cells
33.7
47.2
19.1
death
mitosis
arrest
B
FUCCI populations
% of cells
G1-phase
Early S-phase
S/G2-phase
Mitosis
Death
Arrest
C
Death in G1-phase
RFU
time (h)
2 h
18 h
36 h
death
D
Death in early S-phase
RFU
time (h)
2 h
10 h
35 h
death
E
Death in S/G2-phase
RFU
time (h)
2 h
22 h
34 h
death

HT1080 FUCCI cells were treated with 1μM Selinexor and tracked longitudinally with time-lapse microscopy. (A) The distribution of cell fates after selinexor treatment, as determined through longitudinal, single cell analysis. Approximately half of cells arrest or die, while the other half divides in the presence of drug. (B) The distribution of FUCCI populations at time of death and when arrested, respectively. 60% of death occurs in G1-phase and >90% of arrested cells are in G1-phase. Death in early S-phase and late S/G2-phase are also observed. c-e) Representative examples of a death from different cell cycle phases. (C) Death in G1-phase. This cell is tracked from birth, progressed into G1-phase and dies at ~52 h, indicating the cell is arrested in G1-phase prior to death. (D) Death in early S- phase. This cell is tracked from birth, progresses to yellow state after ~10 h, and remains yellow until the cell dies at ~52 h. (E) Death in late S/G2-phase. This cell is tracked from birth. After an abnormally long G1/early S-phase lasting ~30 h, it transitions to green and die ~41 h after birth.

Summary of HT1080 response to Selinexor

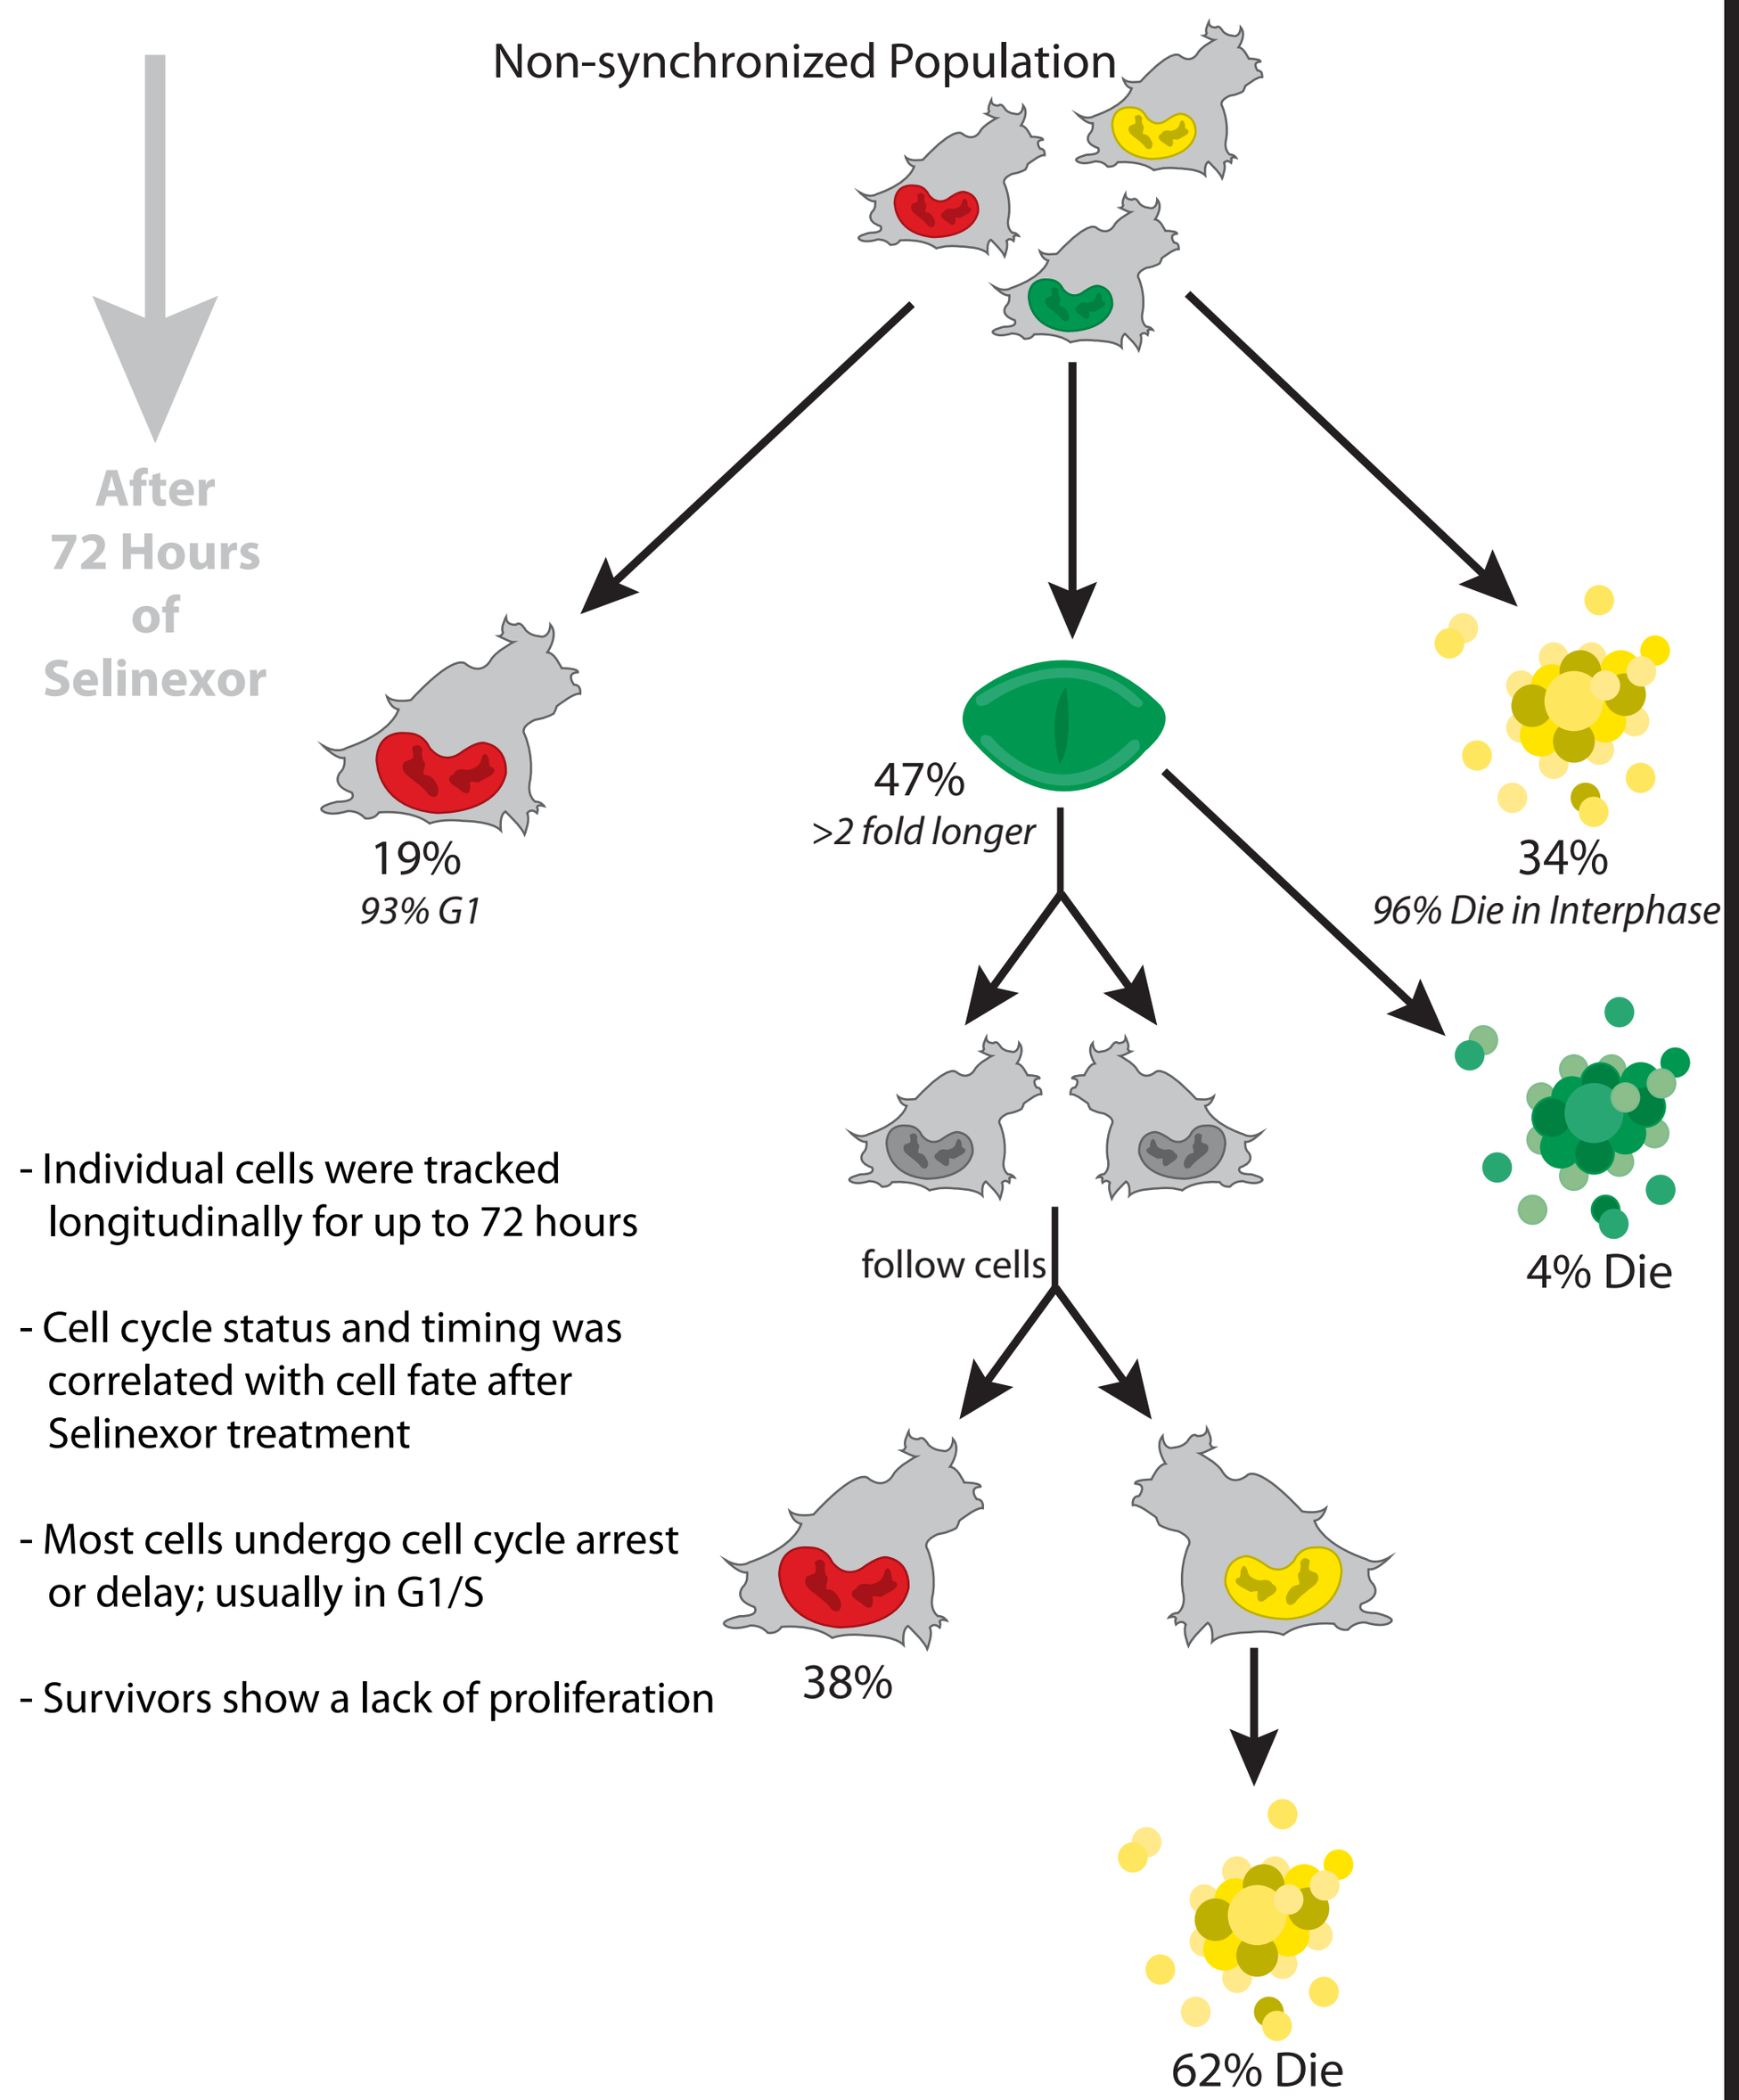
Non-synchronized Population
After 72 Hours of Selinexor
19%
93% G1
47%
>2 fold longer
34%
96% Die in Interphase
4% Die
follow cells
38%
62% Die
- Individual cells were tracked longitudinally for up to 72 hours
- Cell cycle status and timing was correlated with cell fate after Selinexor treatment
- Most cells undergo cell cycle arrest or delay; usually in G1/S
- Survivors show a lack of proliferation

Table 1.	Treatment: control	Treatment: selinexor
Cell fate determination (all cells)	**% of total population**	
Death	0%	34%
Mitosis	100%	47%
Arrest	0%	19%
FUCCI status - cells that die (all cells)	**% of death population**	
G1-phase (hCdt1 only)	-	71%
early S-phase (hCdt1 and hGem)	-	6%
mid-late S/G2-phase (hGem only)	-	19%
mitosis (hGem only)	-	4%
FUCCI status - cells that arrest (all cells)	**% of arrested poplulation**	
G1-phase (hCdt1 only)	-	93%
early S-phase (hCdt1 and hGem)	-	0%
S/G2-phase (hGem only)	-	7%
Cell fate determination (daughter cells)	**% of G1-phase cells**	
Death	0%	62%
Mitosis	100%	0%
Arrest	0%	38%
FUCCI status - cells that die (daughter cells)	**% of death population**	
G1-phase (hCdt1 only)	-	84%
early S-phase (hCdt1 and hGem)	-	3%
mid-late S/G2-phase (hGem only)	-	13%
mitosis (hGem only)	-	0%

Table 1.	Treatment: control	Treatment: selinexor
FUCCI status - cells that arrest (daughter cells)	**% of arrested poplulation**	
G1-phase (hCdt1 only)	-	96%
early S-phase (hCdt1 and hGem)	-	0%
S/G2-phase (hGem only)	-	4%
Fate determination - cells treated in G1-phase	**% of daughter cells**	
Death	0%	59%
Mitosis	100%	9%
Arrest	0%	32%
Fate determination - cells treated in early S-phase	**% of early S-phase cells**	
Death	0%	40%
Mitosis	100%	57%
Arrest	0%	3%
Fate determination - cells treated in S/G2/M-phase	**% of S/G2/M-phase cells**	
Death	0%	5%
Mitosis	100%	95%
Arrest	0%	0%
Average time observed per cell cycle phase	**time (hours)**	
G1-phase (hCdt1 only)	5	21
early S-phase (hCdt1 and hGem)	2	6
S/G2-phase (hGem only)	7	13

HT1080 FUCCI cells were treated with 1μM Selinexor and tracked longitudinally with time-lapse microscopy. The diagram is a graphical summary of cell fates observed after Selinexor treatment. The Table represents quantification of all observed responses of FUCCI cells in control conditions and after Selinexor treatment. Cell cycle stage upon death and when treated was correlated with response. The time spent in each FUCCI phase is also noted.

Figure 3. Selinexor treated cells show slowed DNA synthesis and accumulation of γH2A.X, indicating DNA double-strand breaks

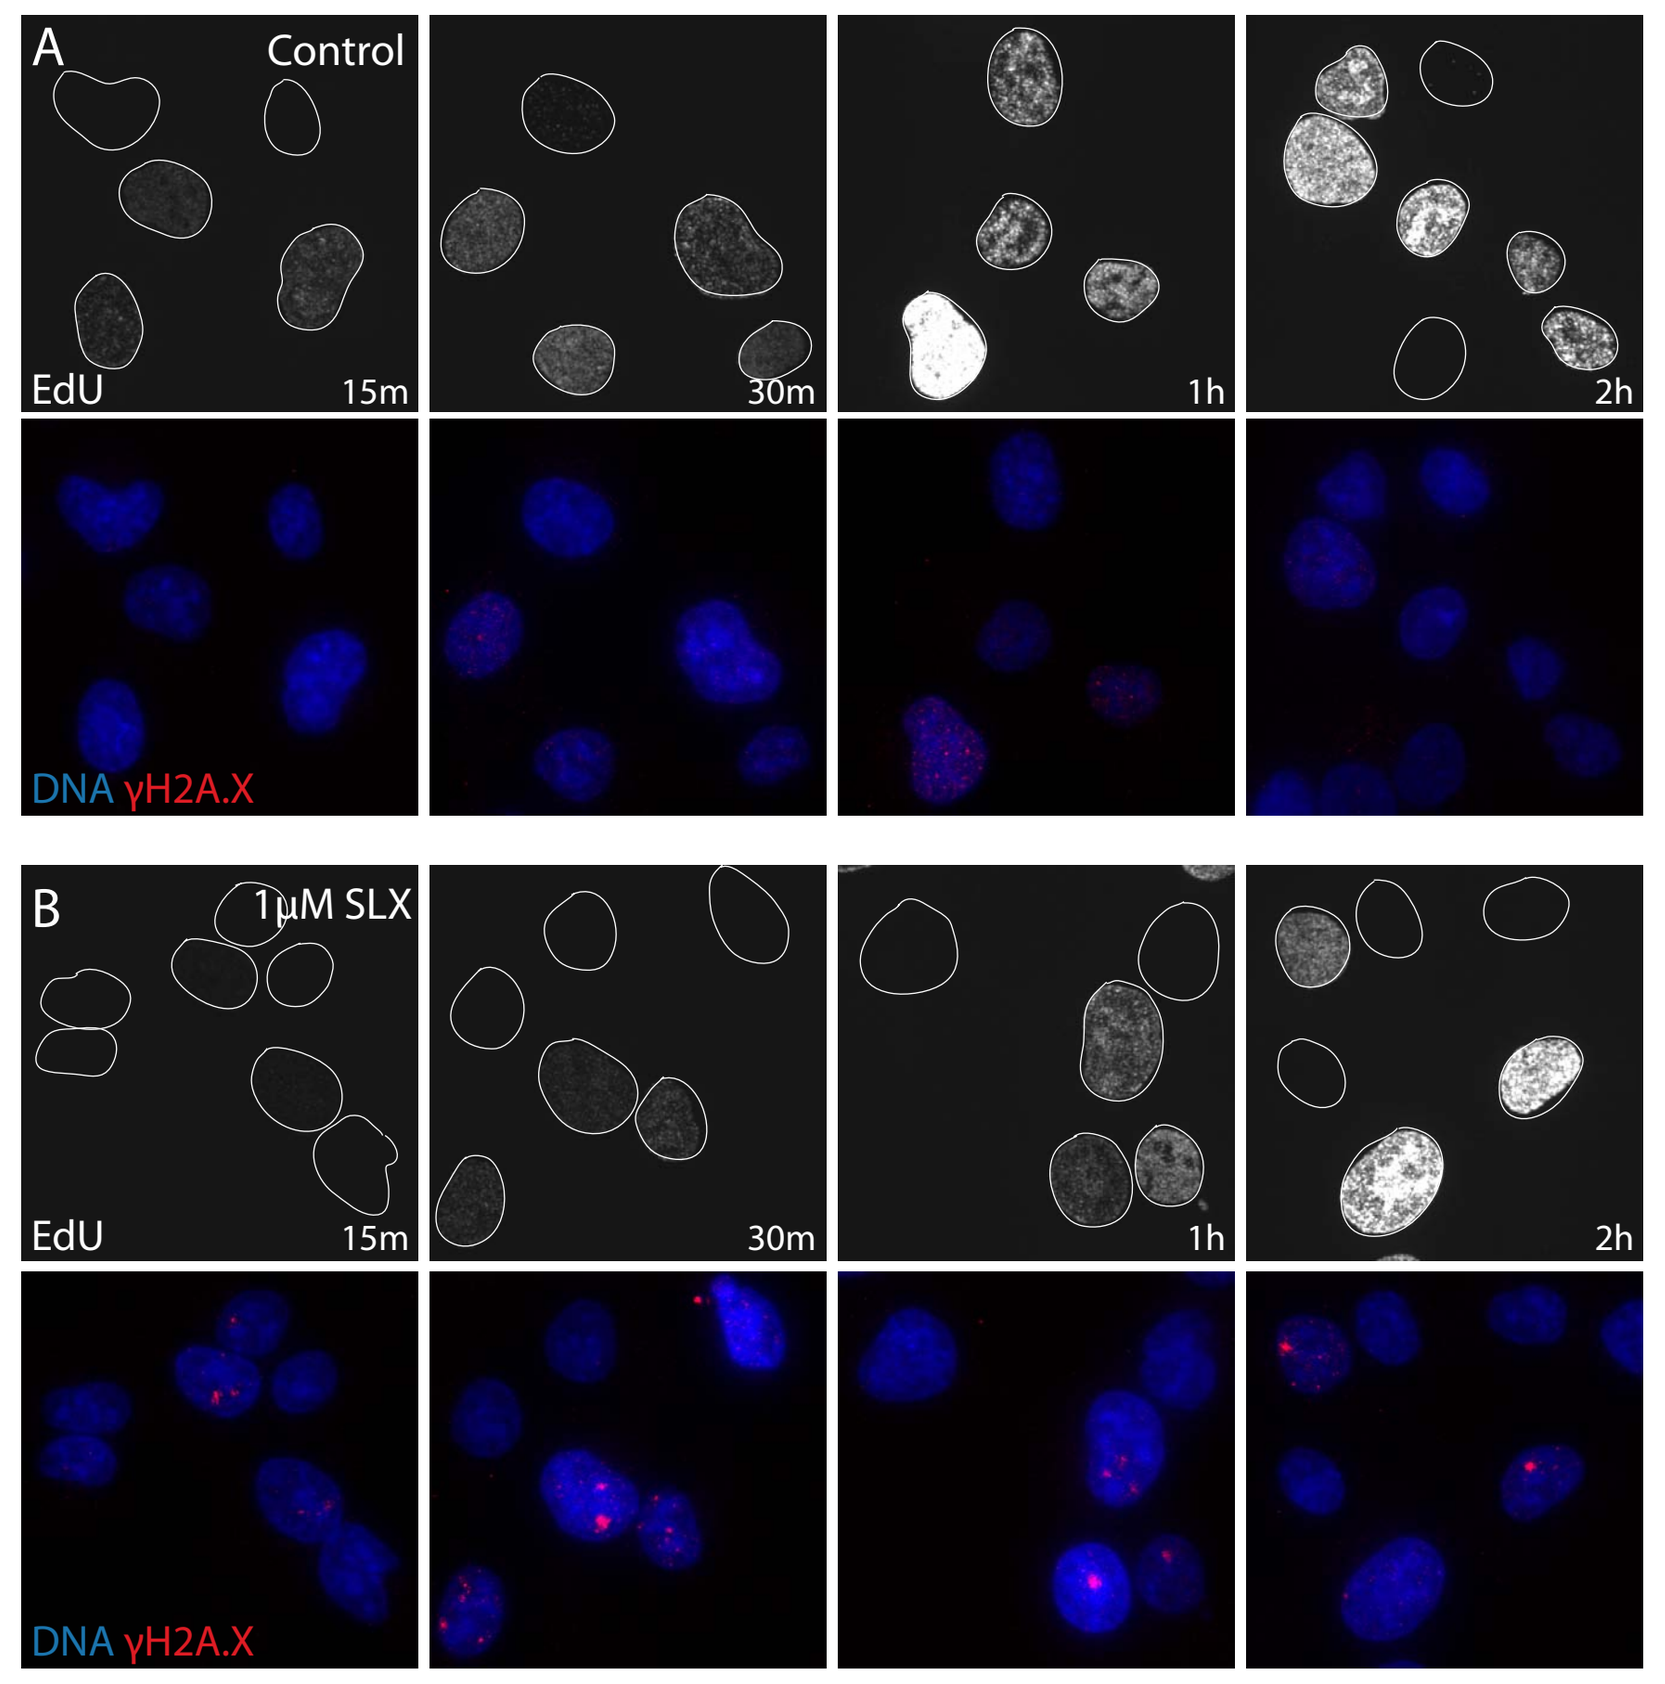
A
Control
EdU
15m
30m
1h
2h
DNA γH2A.X
B
1µM SLX
EdU
15m
30m
1h
2h
DNA γH2A.X

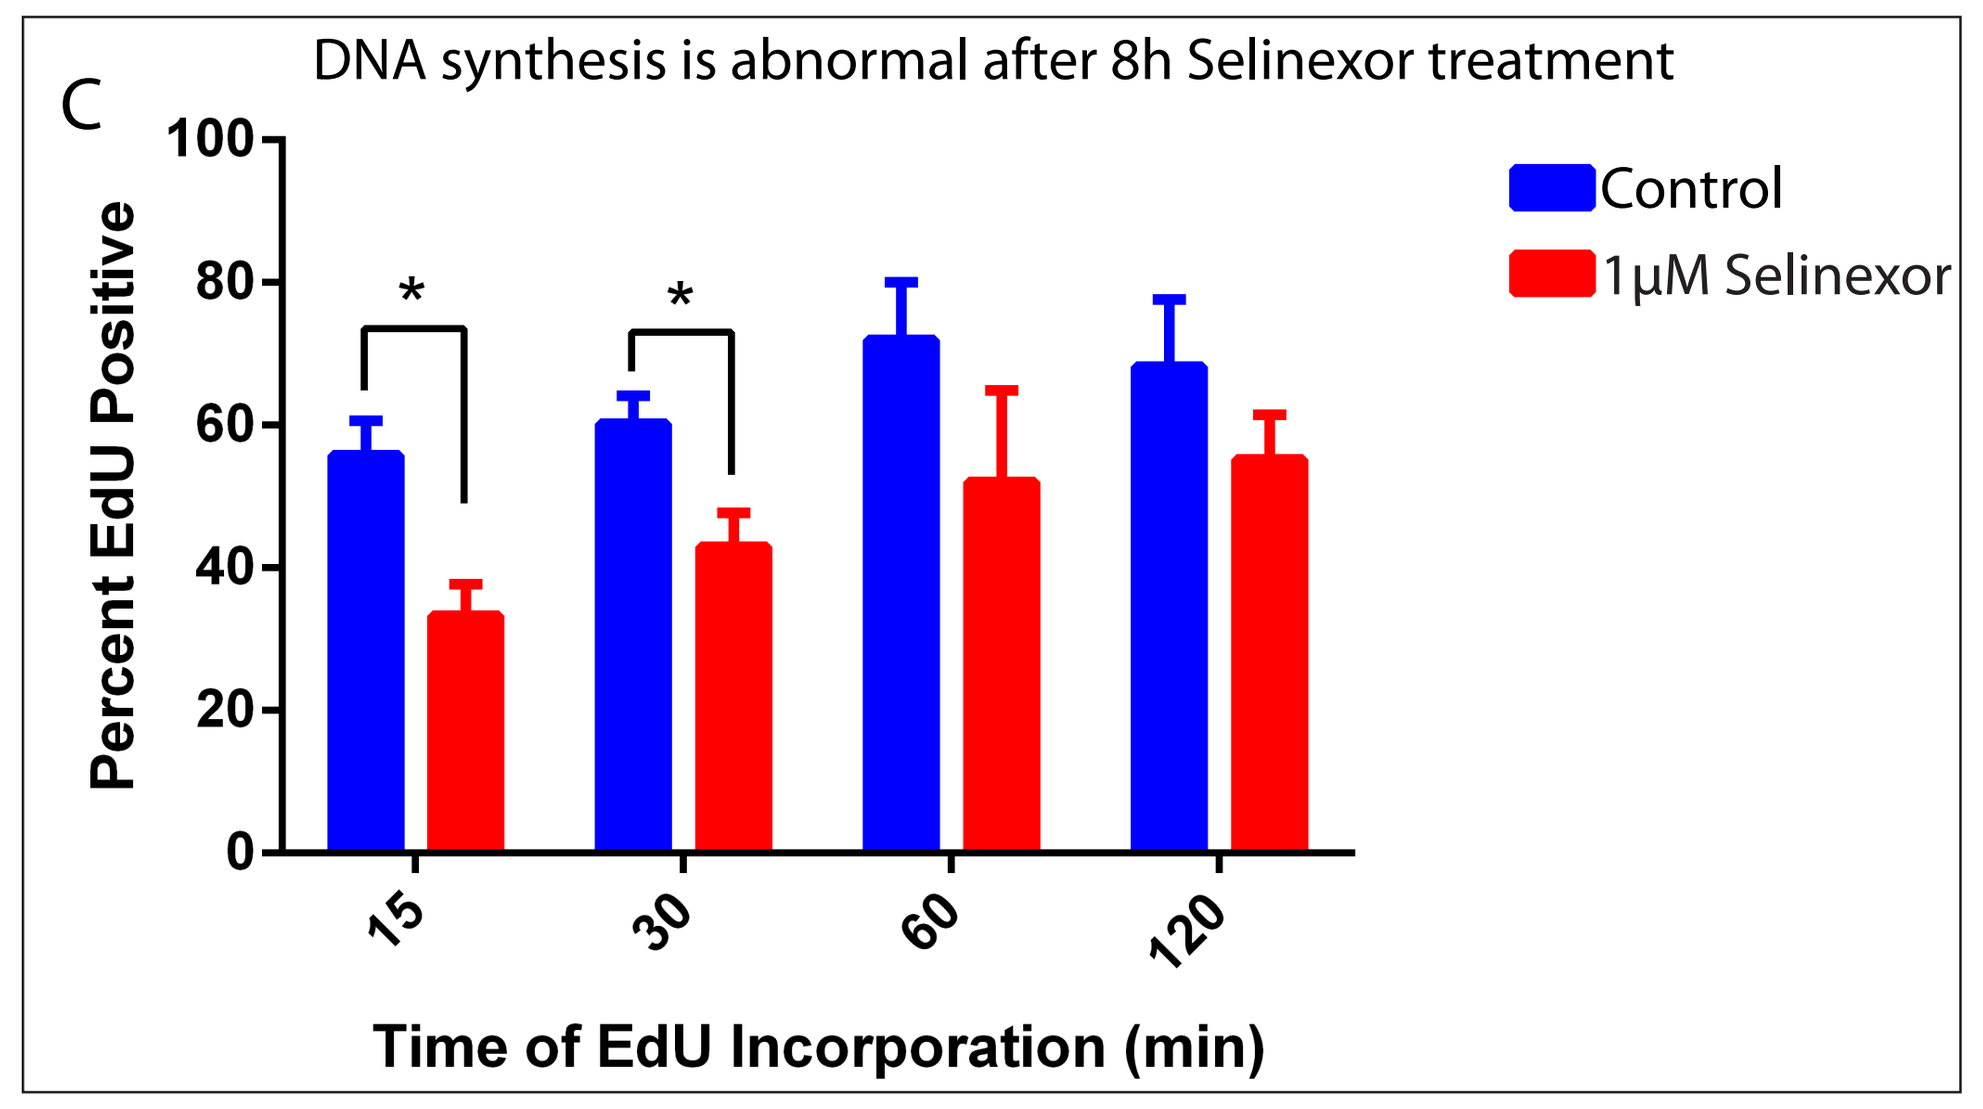
C
DNA synthesis is abnormal after 8h Selinexor treatment
Percent EdU Positive
Control
1µM Selinexor
15
30
60
120
Time of EdU Incorporation (min)

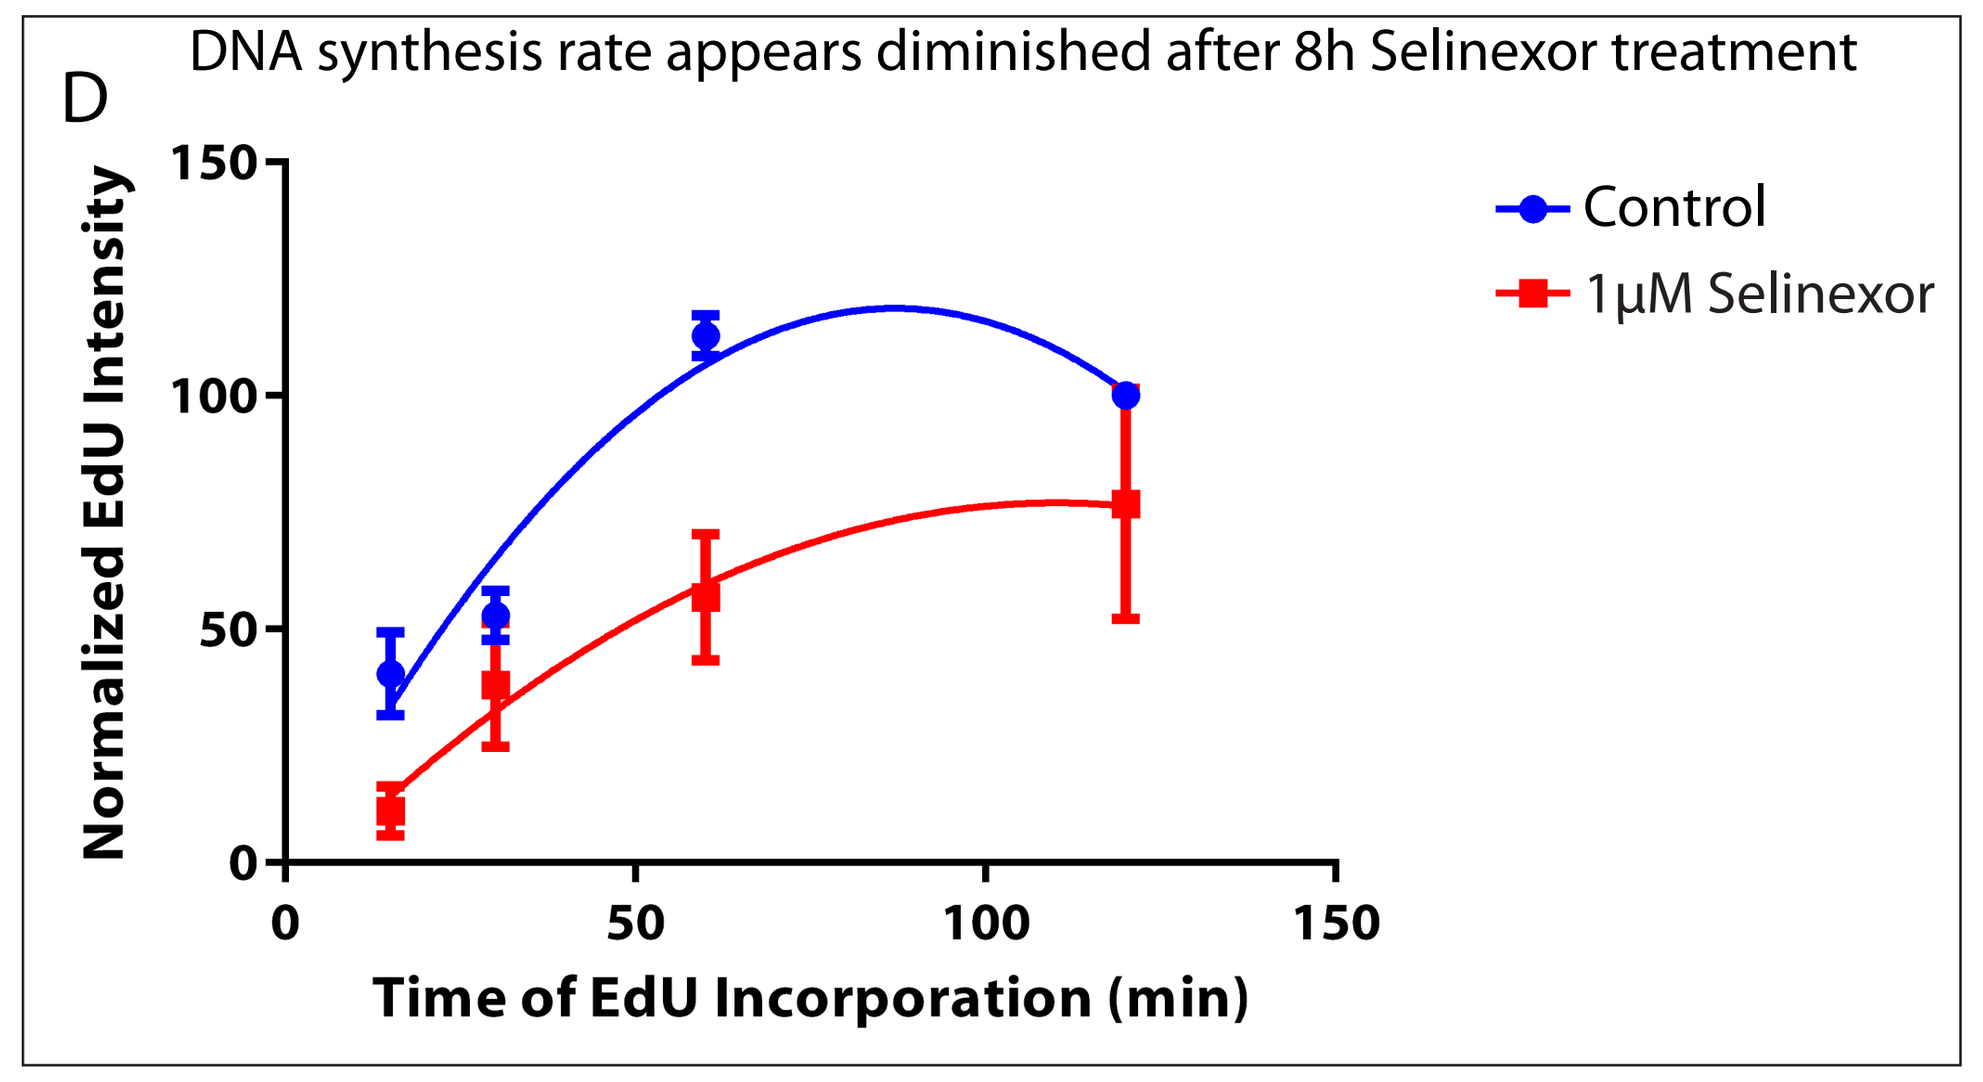
D
DNA synthesis rate appears diminished after 8h Selinexor treatment
Normalized EdU Intensity
Control
1µM Selinexor
Time of EdU Incorporation (min)

HT1080 cells were treated with Selinexor for 8h and then EdU was added to the media for varying amounts of time then pulse-labeled with EdU for varying amounts of time. Cells were fixed and stained for γH2A.X. (A, B) Representative images show EdU signal (white) and γH2A.X staining (red). Nuclei are indicated in the EdU images by white regions of interest. (C) Consistent with Figure 3D, the number of EdU positive cells after 8h Selinexor is decreased. * = p < 0.01. (D) The mean integrated EdU signal intensity per cell for three separate experiments in decreased after Selinexor treatment indicating that DNA synthesis is slowed and/or less efficient during XPO-1 inhibiition. Of note, cells with large γH2A.X foci are frequently positive for EdU demonstrating that Selinexor exerts an effect during S-phase.

Is DNA damage repaired? How does damage affect cell fate?

Figure 4. Various cells fates are observed after DNA damage foci form during Selinexor treatment

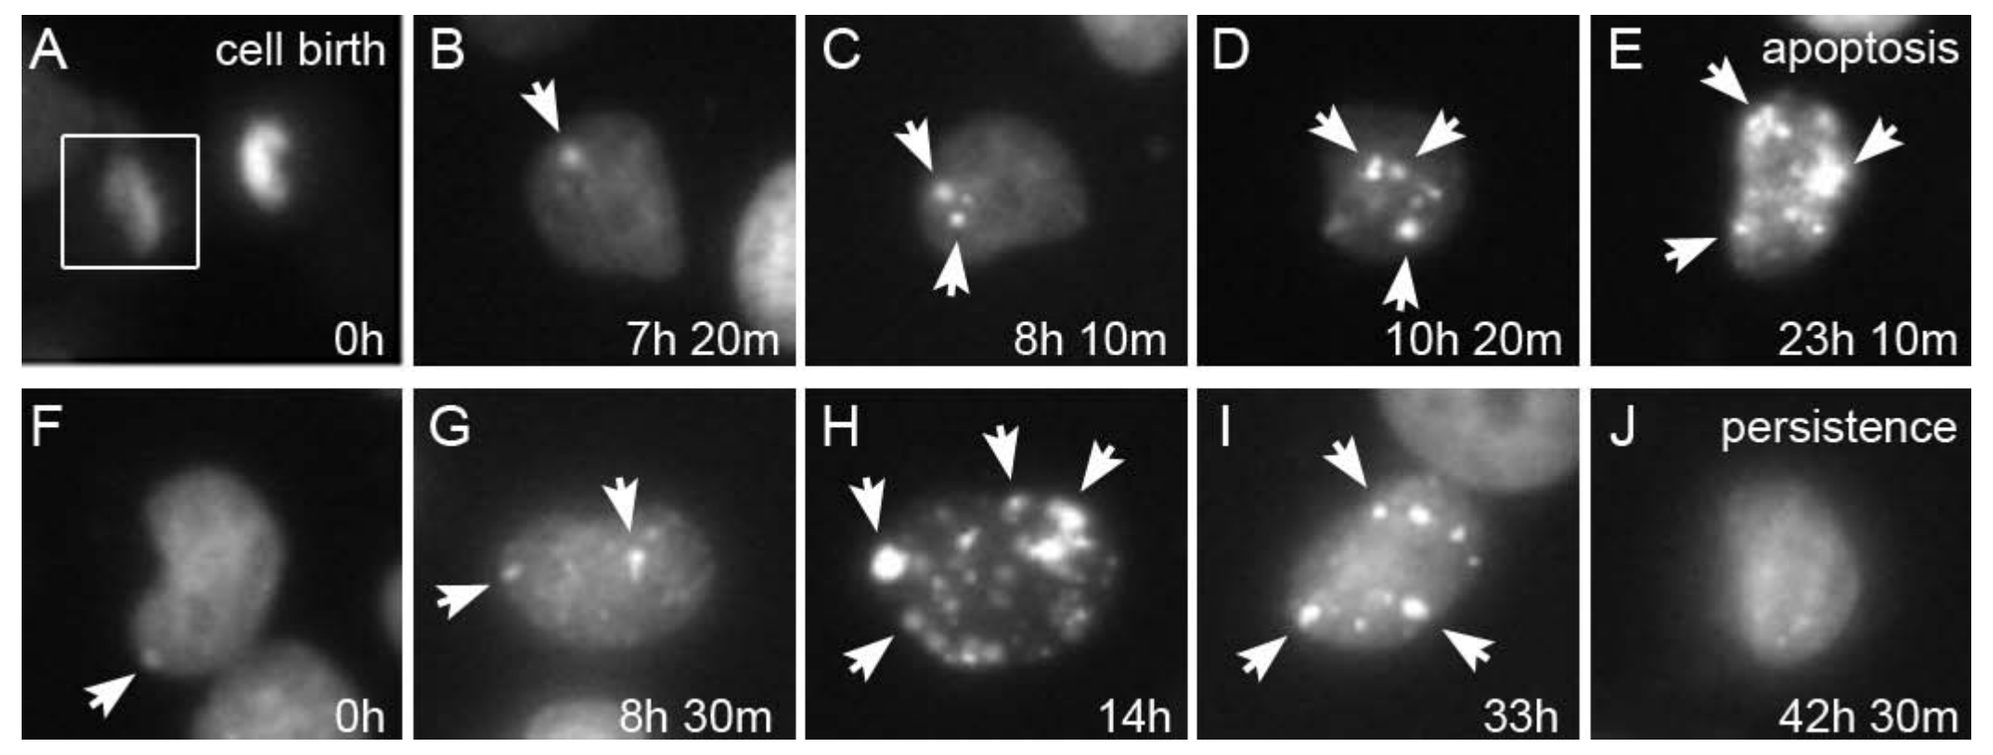
A
cell birth
0h
B
7h 20m
C
8h 10m
D
10h 20m
E
apoptosis
23h 10m
F
0h
G
8h 30m
H
14h
I
33h
J
persistence
42h 30m

(A) HT1080 expressing the mCherry-BP1-2 double-stranded DNA damage biosensor. This cell (box) was just born into the presence of 1μM Selinexor and showed no fluorescent foci. (B-E) The cell was directly tracked every ten minutes and DNA damage (foci, arrowheads) began forming after 7h and became more numerous over time until this cell died at 23h 10min. h = hours, m = minutes. (B) This cell also forms numerous foci, but appears to repair them and remains until the end of the time-lapse; we are now asking what the fate of these cells is.

How does EdU incorporation change over time?

Does DNA damage correlate with active DNA synthesis?

Figure 5. Active DNA synthesis decreases over time and correlates with DNA damage.

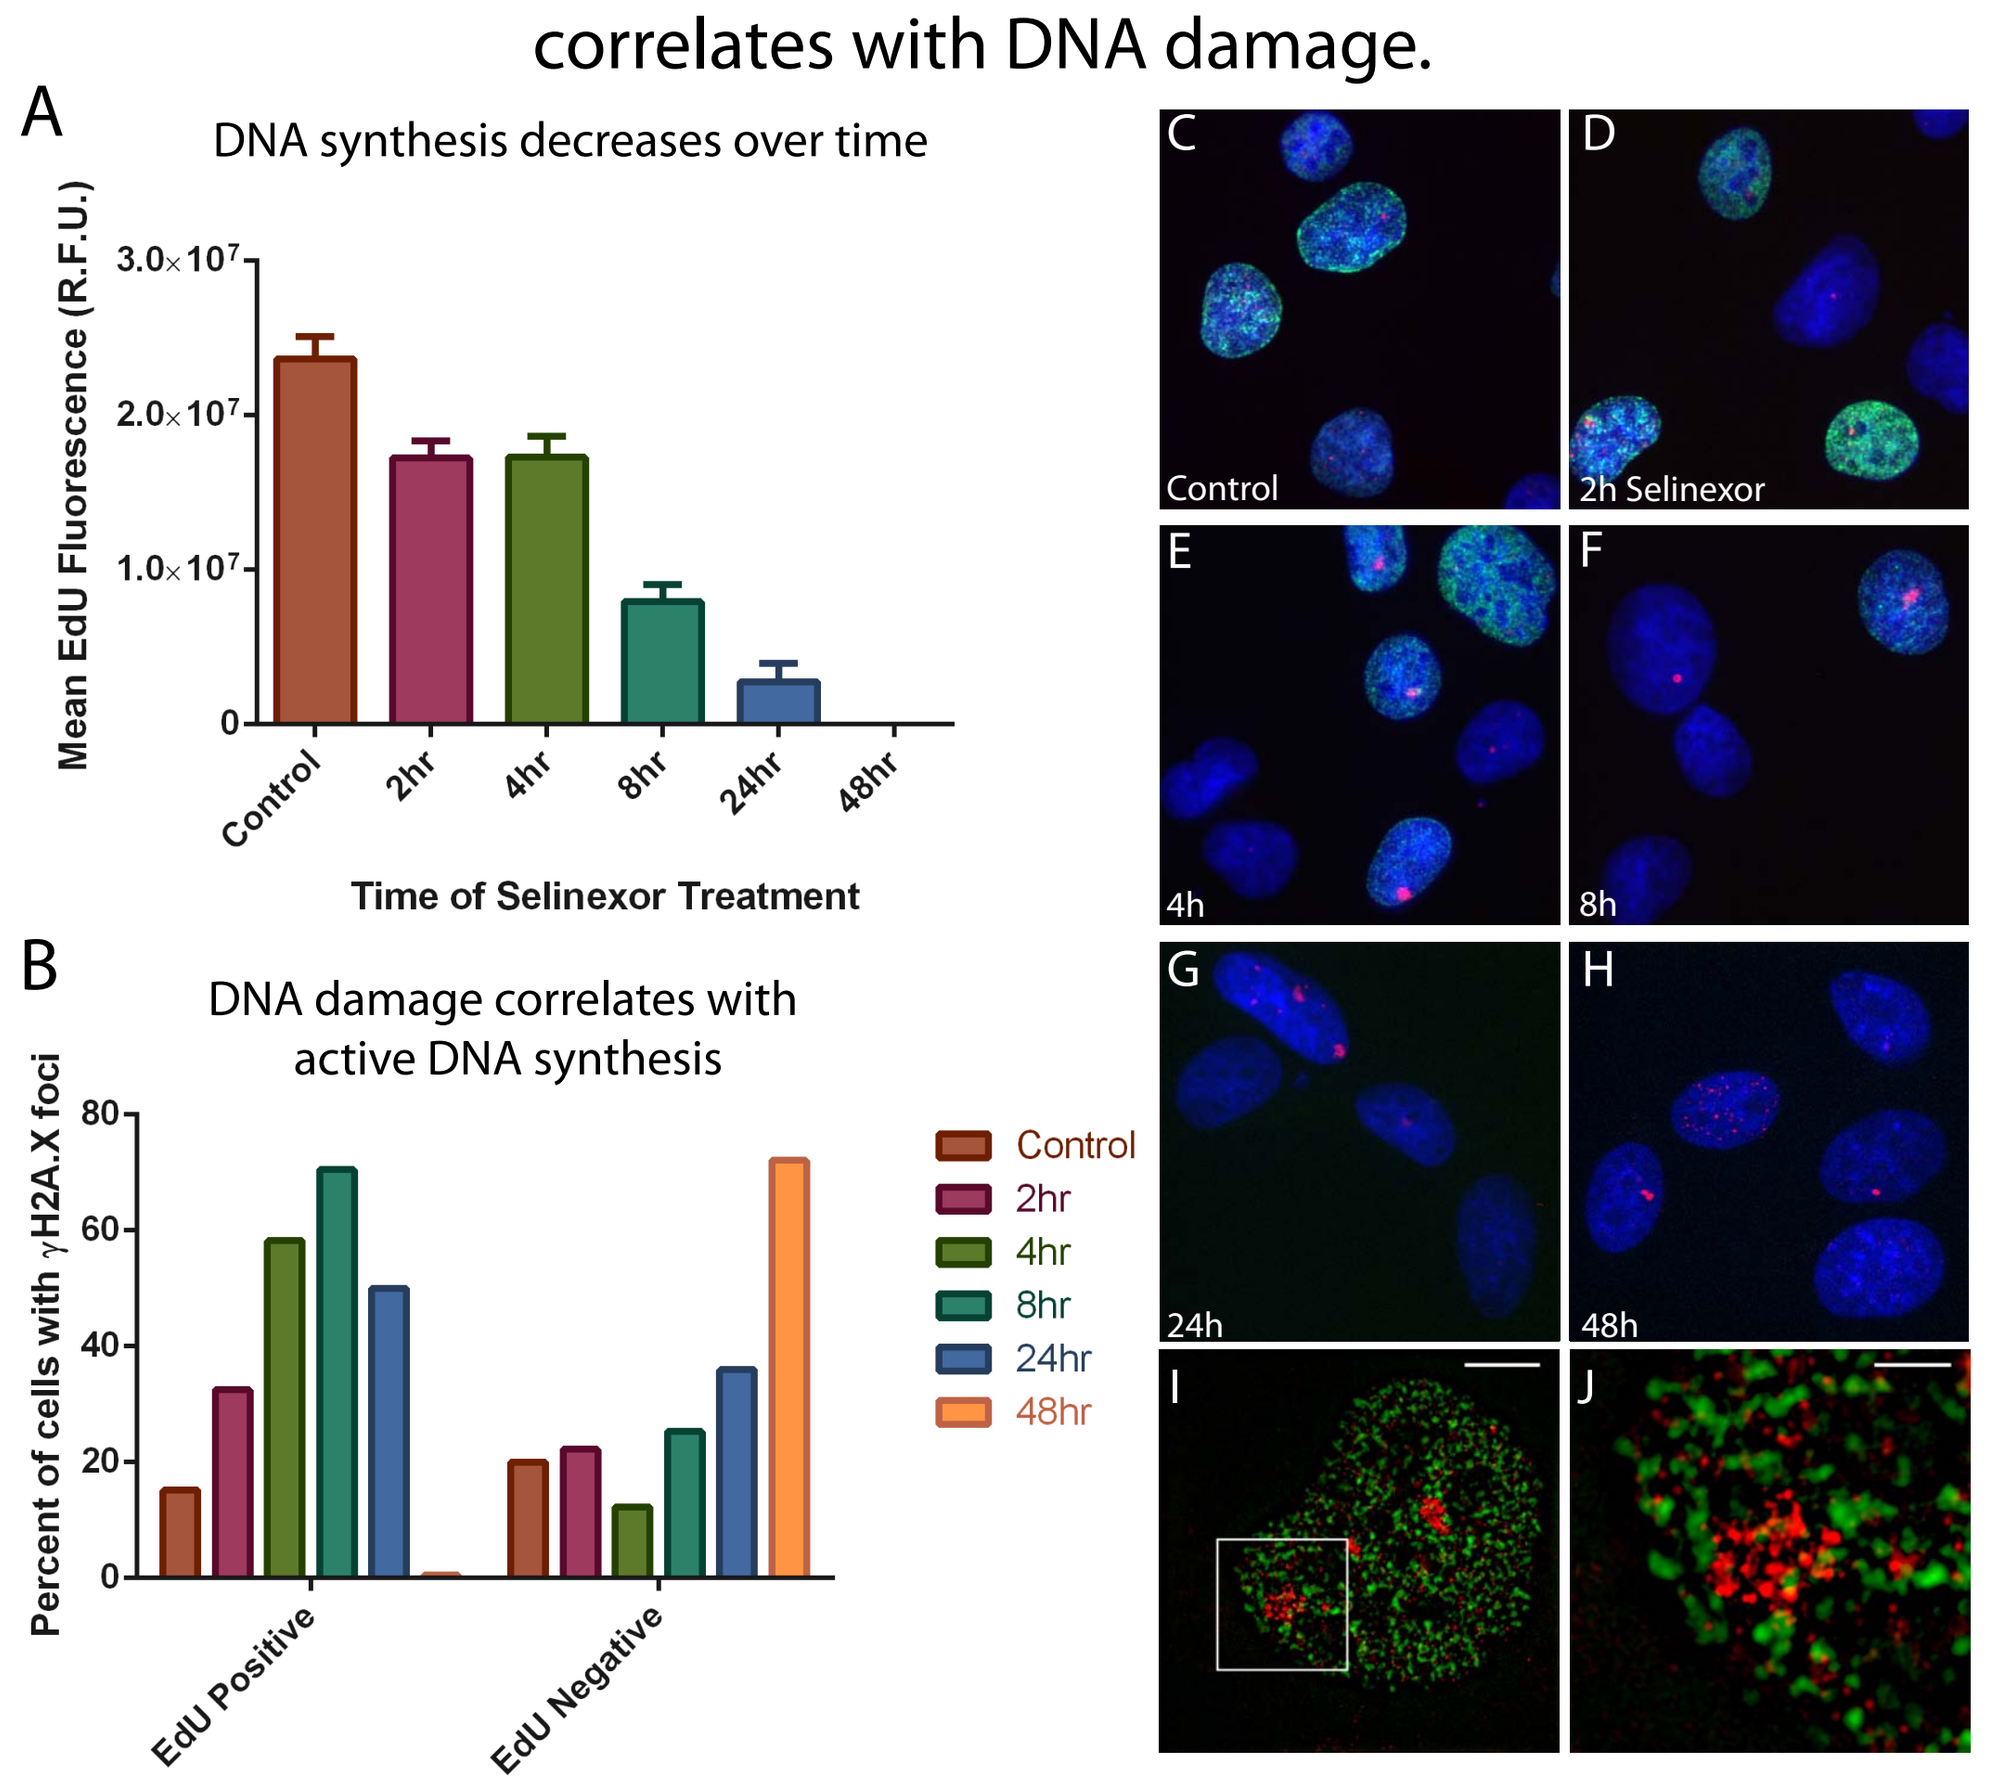
A
DNA synthesis decreases over time
Mean EdU Fluorescence (R.F.U.)
Control
2hr
4hr
8hr
24hr
48hr
Time of Selinexor Treatment
B
DNA damage correlates with active DNA synthesis
Percent of cells with γH2A.X foci
EdU Positive
EdU Negative
Control
2hr
4hr
8hr
24hr
48hr
C
Control
D
2h Selinexor
E
4h
F
8h
G
24h
H
48h
I
J

HT1080 FUCCI cells were treated treated with Selinexor for increasing times. EdU was pulsed for 15min at the end of treatment. Cells were then fixed and stained for γH2A.X. (A) Mean fluorescence for EdU was quantified for each time point. Error bars represent SEM. (B) Cells were scored at each time point as positive or negative for EdU. γH2A.X foci were identified using Find Maxima in ImageJ. The percent cells over time with 1 or more foci are indicated. (C-H) Representative images of cells treated with control, 2h, 4h, 8h, 24h and 48h Selinexor, respectively. Cells were stained for γH2A.X (red), EdU (green) and DNA (blue). (I-J) Structured illumination microscopy of a cell treated with 1μM Selinexor for 8hr and stained for γH2A.X (red) and EdU (green). Scale bars represent 6μm (I) and 2μm (J).

Figure 6. Cell recovery is significantly impaired after 8 hours of Selinexor treatment

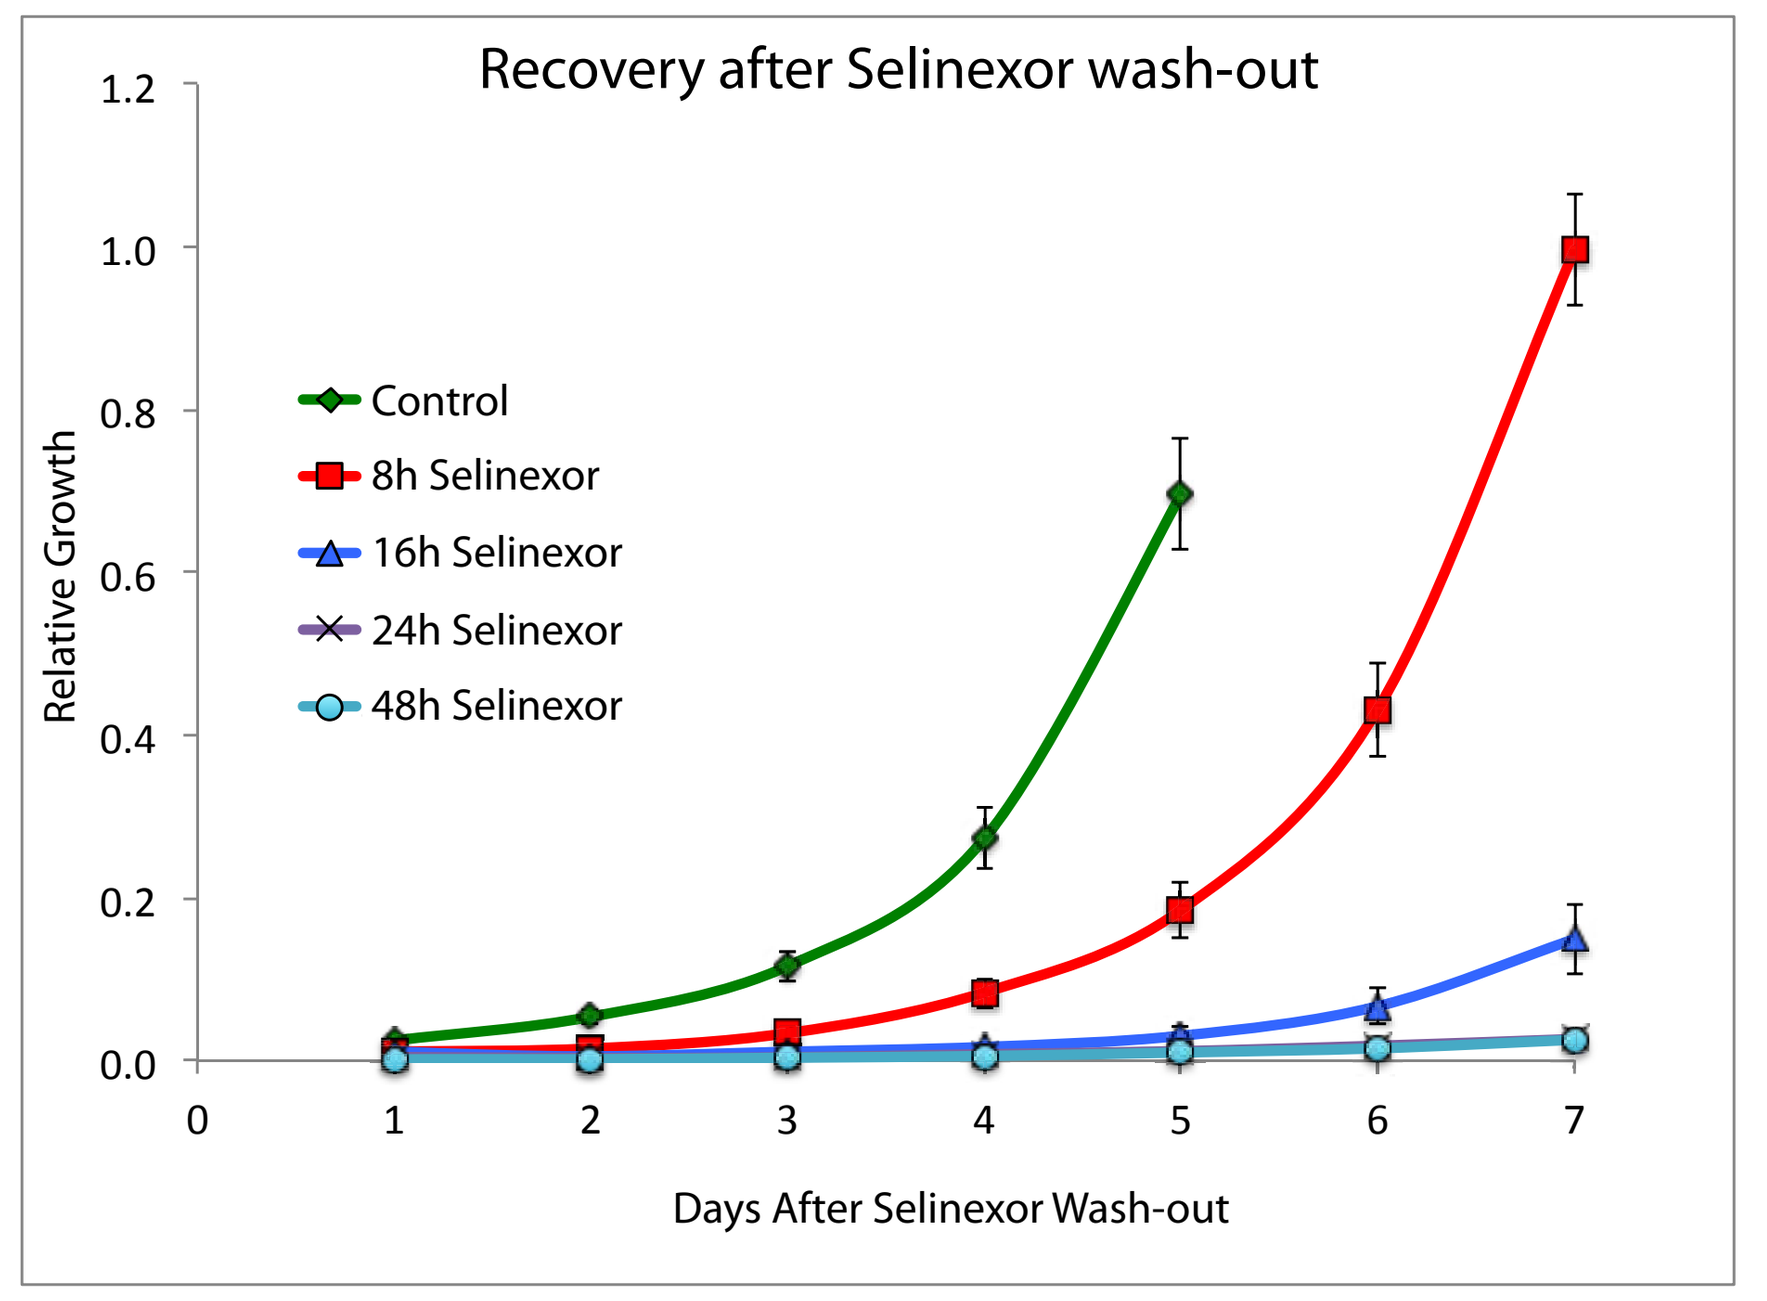
Recovery after Selinexor wash-out
Relative Growth
Control
8h Selinexor
16h Selinexor
24h Selinexor
48h Selinexor
Days After Selinexor Wash-out

HT1080 FUCCI cells were treated treated with Selinexor for increasing times. Drug was washed out and 10,000 cells from each condition were seeded into normal growth medium in gridded glass-bottom dishes. Twelve random locations in each dish were imaged using phase-contrast and fluorescent microscopy every 24 hours during recovery. Cells at each location were scored and normalized to 450 cells, which defines healthy confluency. * = All control locations were confluent after 5 and overgrown by 6 days. Means with standard errors are plotted.

Summary

Selinexor treatment results in cell cycle arrest and S-phase progression defects associated with striking γH2A.X foci. In agreement with the timing of S-phase defects and apparent DNA damage, cells treated for only eight hours show significantly impaired growth after drug removal. Cells treated for longer times show dramatically reduced recovery, indicating the importance of maintaining Selinexor for longer times to maximize it's anti-cancer effects. We are extending on these data using fluorescent p53 and DNA damage reporters expressed in the same cells to establish relationships between nuclear p53 retention, p53 dynamics, DNA damage, and cell fate decisions. We are also quantifying DNA damage and repair in live cells to establish its contribution to different cell fates. Given this action of Selinexor, it is possible it may remain effective on cancer cells regardless of TP53 and/or p21^CIP1 status. This is extremely important, as these are dysfunctional in many cancers.

Molecular, Cellular, and Developmental Biology
UNIVERSITY OF COLORADO BOULDER

Light Microscopy Core Facility
UNIVERSITY OF COLORADO BOULDER