Anti-tumor activity of selective inhibitors of XPO1/CRM1-mediated nuclear export in diffuse malignant peritoneal mesothelioma: The role of survivin

N. Zaffaroni[1] , M. A. De Cesare[1] , D. Cominetti[1] , V. Doldi[1] , A. Lopergolo[1] , M. Deraco[2] , P. Gandellini[1] , Y. Landesman[3] , S. Friedlander[3] , M.G. Kauffman[3] , S. Shacham[3] and M. Pennati[1].

[1]Dept. of Experimental Oncology & Molecular Medicine, Fondazione IRCCS Istituto Nazionale Tumori, Milano, Italy; [2] Dept. of Surgery, Fondazione IRCCS Istituto Nazionale Tumori, Milano, Italy; [3]Karyopharm Therapeutics Inc., Newton, MA, USA

BACKGROUND

Diffuse malignant peritoneal mesothelioma (DMPM)

- It is an uncommon and locally aggressive tumor that develops from mesothelial cells lining the peritoneal cavity, and accounts for approximately 25-30% of all mesotheliomas.
- It is a rapidly fatal disease characterized by a poor prognosis.
- Treatment of DMPM patients by palliative surgery, systemic/intraperitoneal chemotherapy and abdominal irradiation showed to be ineffective with a median survival of about one year.
- Loco-regional strategy combining aggressive cytoreductive surgery (CRS) with hyperthermic intraperitoneal chemotherapy (HIPEC) signficantly improve median survival up to 40-92 months in selected series of patients, although approximately 40-60% of patients still experience recurrence. For these patients, and for those who are not elegible to CRS+HIPEC, the prognosis remains severe due to lack of effective alternative treatment options.

The development of novel therapeutic strategies is urgently needed

- Dysregulation of the apoptotic pathway may play a role in DMPM resistance to chemotherapy.
- Survivin and other Inhibitors of Apoptosis Protein (IAP) family members may represent new therapeutic targets.

Survivin

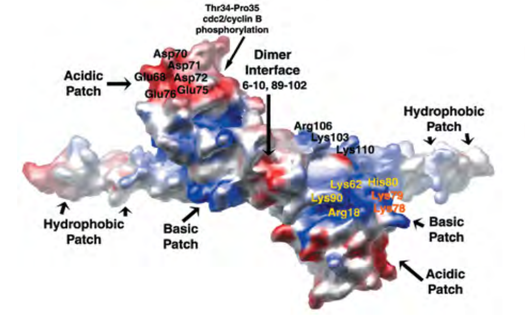

- It is a member of the Inhibitors of Apoptosis Protein (IAP) family.
- It is overexpressed in most human solid tumor types (including DMPM) and hematologic malignancies and it is absent or low in most terminally differentiated normal tissues with some exceptions.
- It plays an important role in the drug resistant phenotype of human cancer cells.
- It is a bifunctional protein that:
 - facilitates tumor cell evasion from apoptosis;
 - promotes mitotic progression.
- Its sub-cellular compartmentalization plays an essential role in determining its bifunctional role.

 Nuclear localization is mainly involved in spindle monitoring at mitosis, whereas cytoplasmic/mitochondrial survivin counteracts pro-apoptotic signals by preventing caspase-9 and caspase-3 activation.
- Its export from the nucleus back to the cytoplasm requires an interaction between the exportin-1/chromosome maintenance protein 1 (XPO1/CRM1) and the specific leucine-rich nuclear export signals (NES) within survivin.

 This interaction is accomplished via the RanGTP/GDP axis.

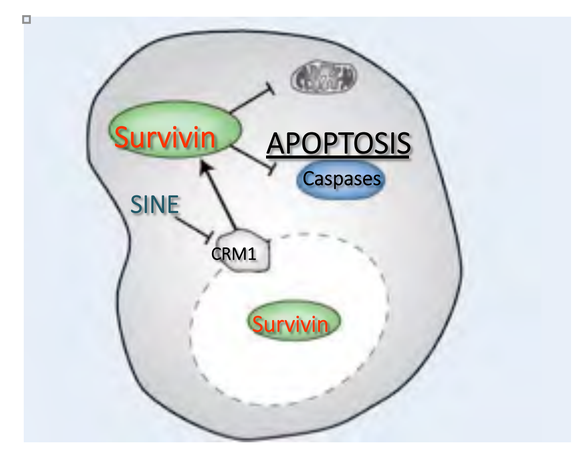
Survivin
APOPTOSIS
Caspases
SINE
CRM1
Survivin

Stauber RH et al, Cancer Res 2007

ChRomosome Maintenance protein 1 (CRM1)

- XPO1/CRM1 is a key member of the importin β superfamily of nuclear transport receptors that are involved in the nucleo-cytoplasmic active transport of over 200 proteins, including transcription factors, tumor suppressors, cell-cycle regulators and proteins involved in programmed cell death
- A novel, oral, bioavailable, small-molecule Selective Inhibitors of Nuclear Export (SINE) have been developed. These inhibitors bind specifically to the NES-binding groove of XPO1/CRM1 and prevent the interaction with its cargo proteins.
- These compounds have demonstrated anti-tumor activity in a variety of experimental models of solid and hematologic malignancies both *in vitro* and *in vivo*. Among those, selinexor (KPT-330) is the most advanced SINE with >500 hematologic and solid cancer patients treated to date in a number of Phase I/II clinical trials.
- Though six other members of the nuclear export protein family have been identified (XPO2-7), XPO1/CRM1 is the sole nuclear exporter for p53, CDKN1a and survivin.

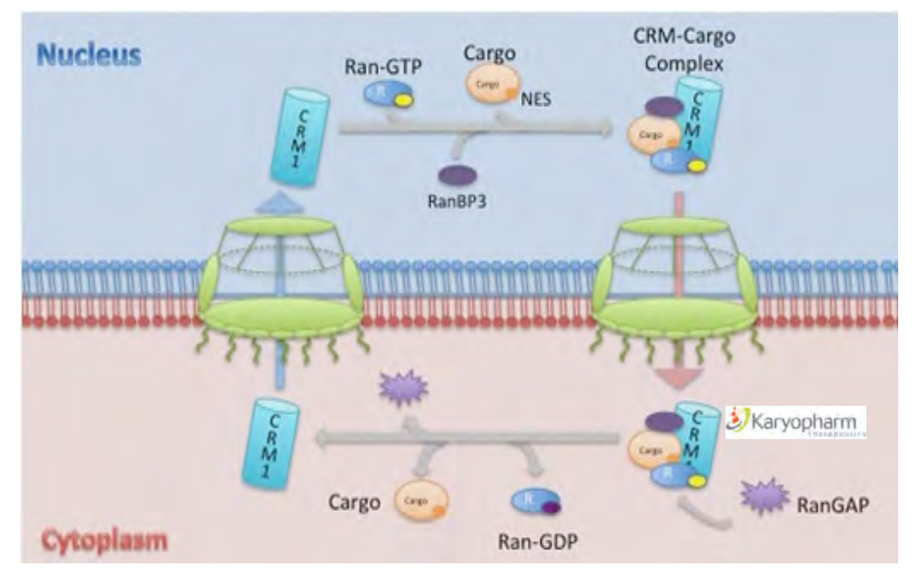
Nucleus
Ran-GTP
Cargo
NES
CRM-Cargo Complex
RanBP3
Cytoplasm
Cargo
Ran-GDP
RanGAP

MATERIALS & METHODS

Experimental Models

Human STO and MesoII DMPM cell lines

STO and MesoII cell lines were established from surgical specimens of patients who underwent surgery at Fondazione IRCCS Istituto Nazionale dei Tumori of Milan

TP53 Status

Cell Line	Exon	Aminoacid change	Protein Effect
STO	//	//	wild-type
MesoII	6	P223L	mutant
	8	V274F	

Experimental Design

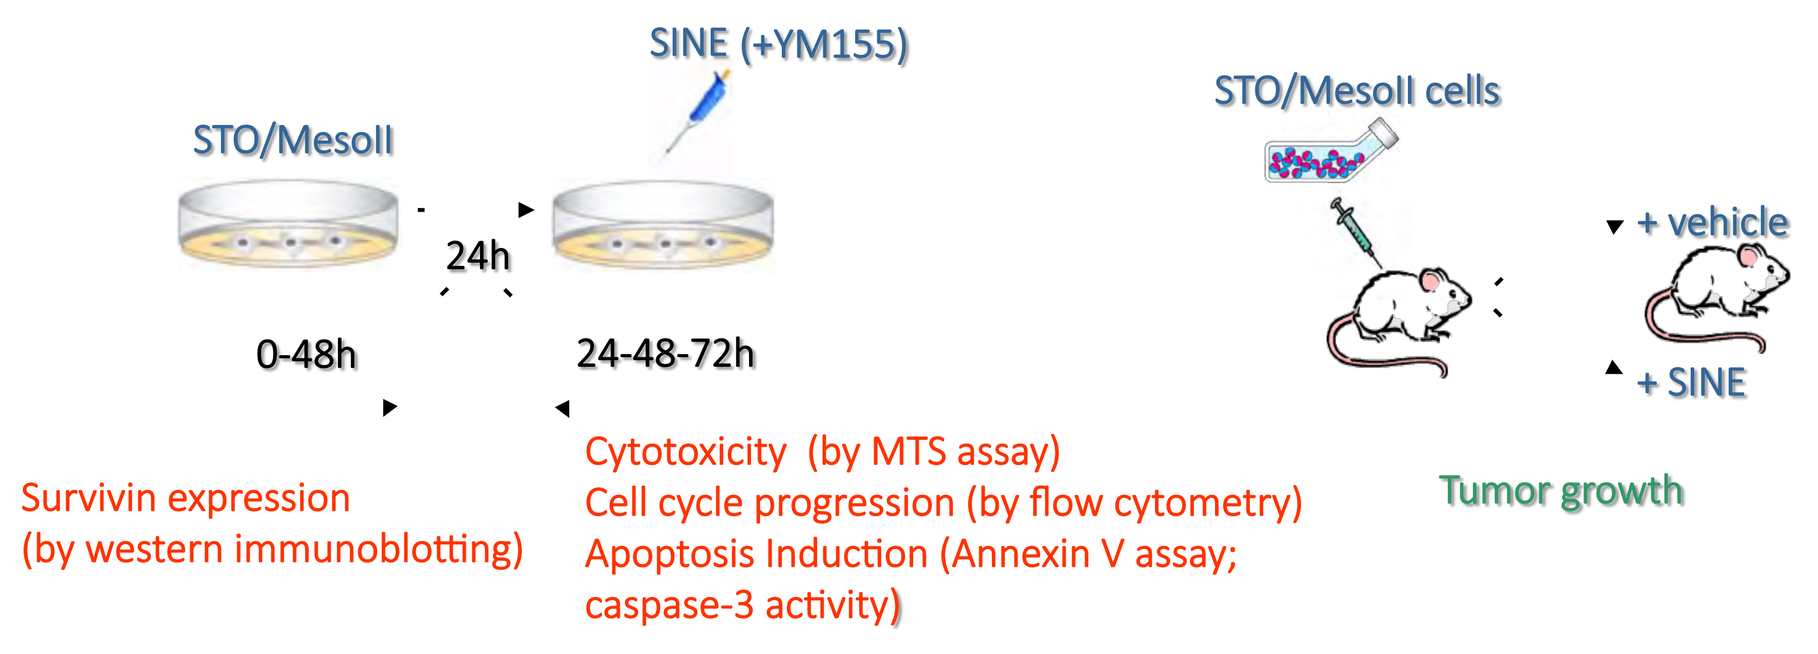
SINE (+YM155)
STO/MesoII
24h
0-48h
24-48-72h
Survivin expression
(by western immunoblotting)
Cytotoxicity (by MTS assay)
Cell cycle progression (by flow cytometry)
Apoptosis Induction (Annexin V assay; caspase-3 activity)
STO/MesoII cells
+ vehicle
+ SINE
Tumor growth

AIMS

In the present study, we explored the efficacy of Selective Inhibitors of Nuclear Export (SINE), KPT-251, KPT-276 and the orally available, clinical stage KPT-330 (selinexor), in DMPM preclinical models.

CONCLUSIONS

- Exposure to SINE induced dose-dependent inhibition of cell growth, cell cycle arrest at G1-phase and caspase-dependent apoptosis, which were consequent to a decrease of XPO1/CRM1 protein levels and the concomitant nuclear accumulation of its cargo proteins p53 and CDKN1a.
- Cell exposure to SINE led to a time-dependent reduction of cytoplasmic survivin levels. In addition, after an initial accumulation, the nuclear protein abundance progressively decreased, as a consequence of an enhanced ubiquitination and proteasome-dependent degradation.
- SINE and the survivin inhibitor YM155 synergistically cooperated in reducing DMPM cell proliferation.
- Orally administered SINE caused a significant anti-tumor effect in subcutaneous and orthotopic DMPM xenografts without appreciable toxicity.

Our study suggests SINE-mediated XPO1/CRM1 inhibition as a novel therapeutic option for DMPM

RESULTS

SINE impair cell growth, promote cell cycle arrest, and induce apoptosis in DMPM

SINE impair DMPM cell growth

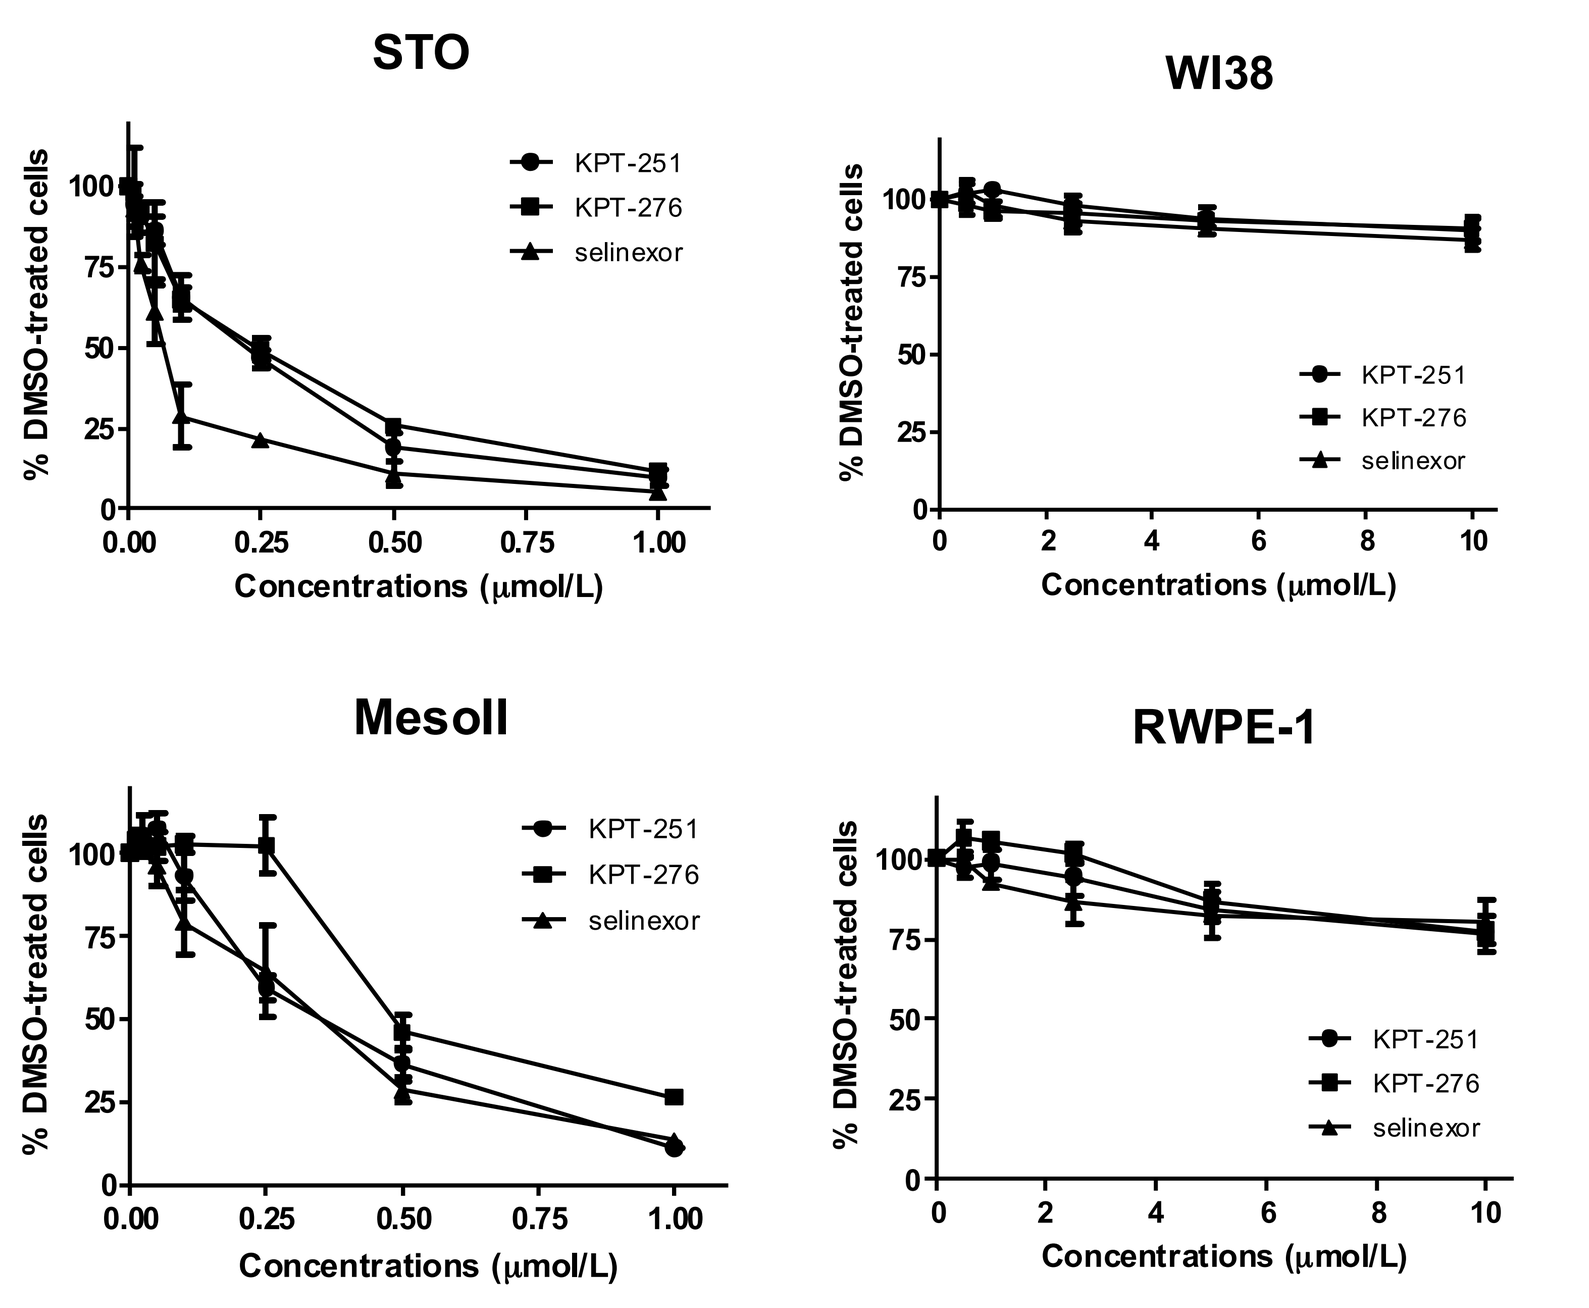

A dose- and time-dependent inhibition of cell growth was consistently observed in STO and MesoII cell lines after treatment with SINE. However, while STO cells showed a higher sensitivity to selinexor compared to KPT-251 and KPT-276, with IC_{50} values of 0.07±0.01, 0.23±0.05, 0.24±0.02 μmol/L respectively, MesoII cells showed a comparable sensitivity to all the compounds with IC_{50} values of 0.35±0.09, 0.36±0.04 and 0.47±0.04 μmol/L, respectively. In addition, at concentrations up to 10 μmol/L, SINE did not alter the growth of both normal human lung fibroblast (WI38) and normal adult human prostate (RWPE-1) cell lines.

SINE promote cell cycle arrest and induce a caspase-dependent apoptotic cell death in DMPM cells

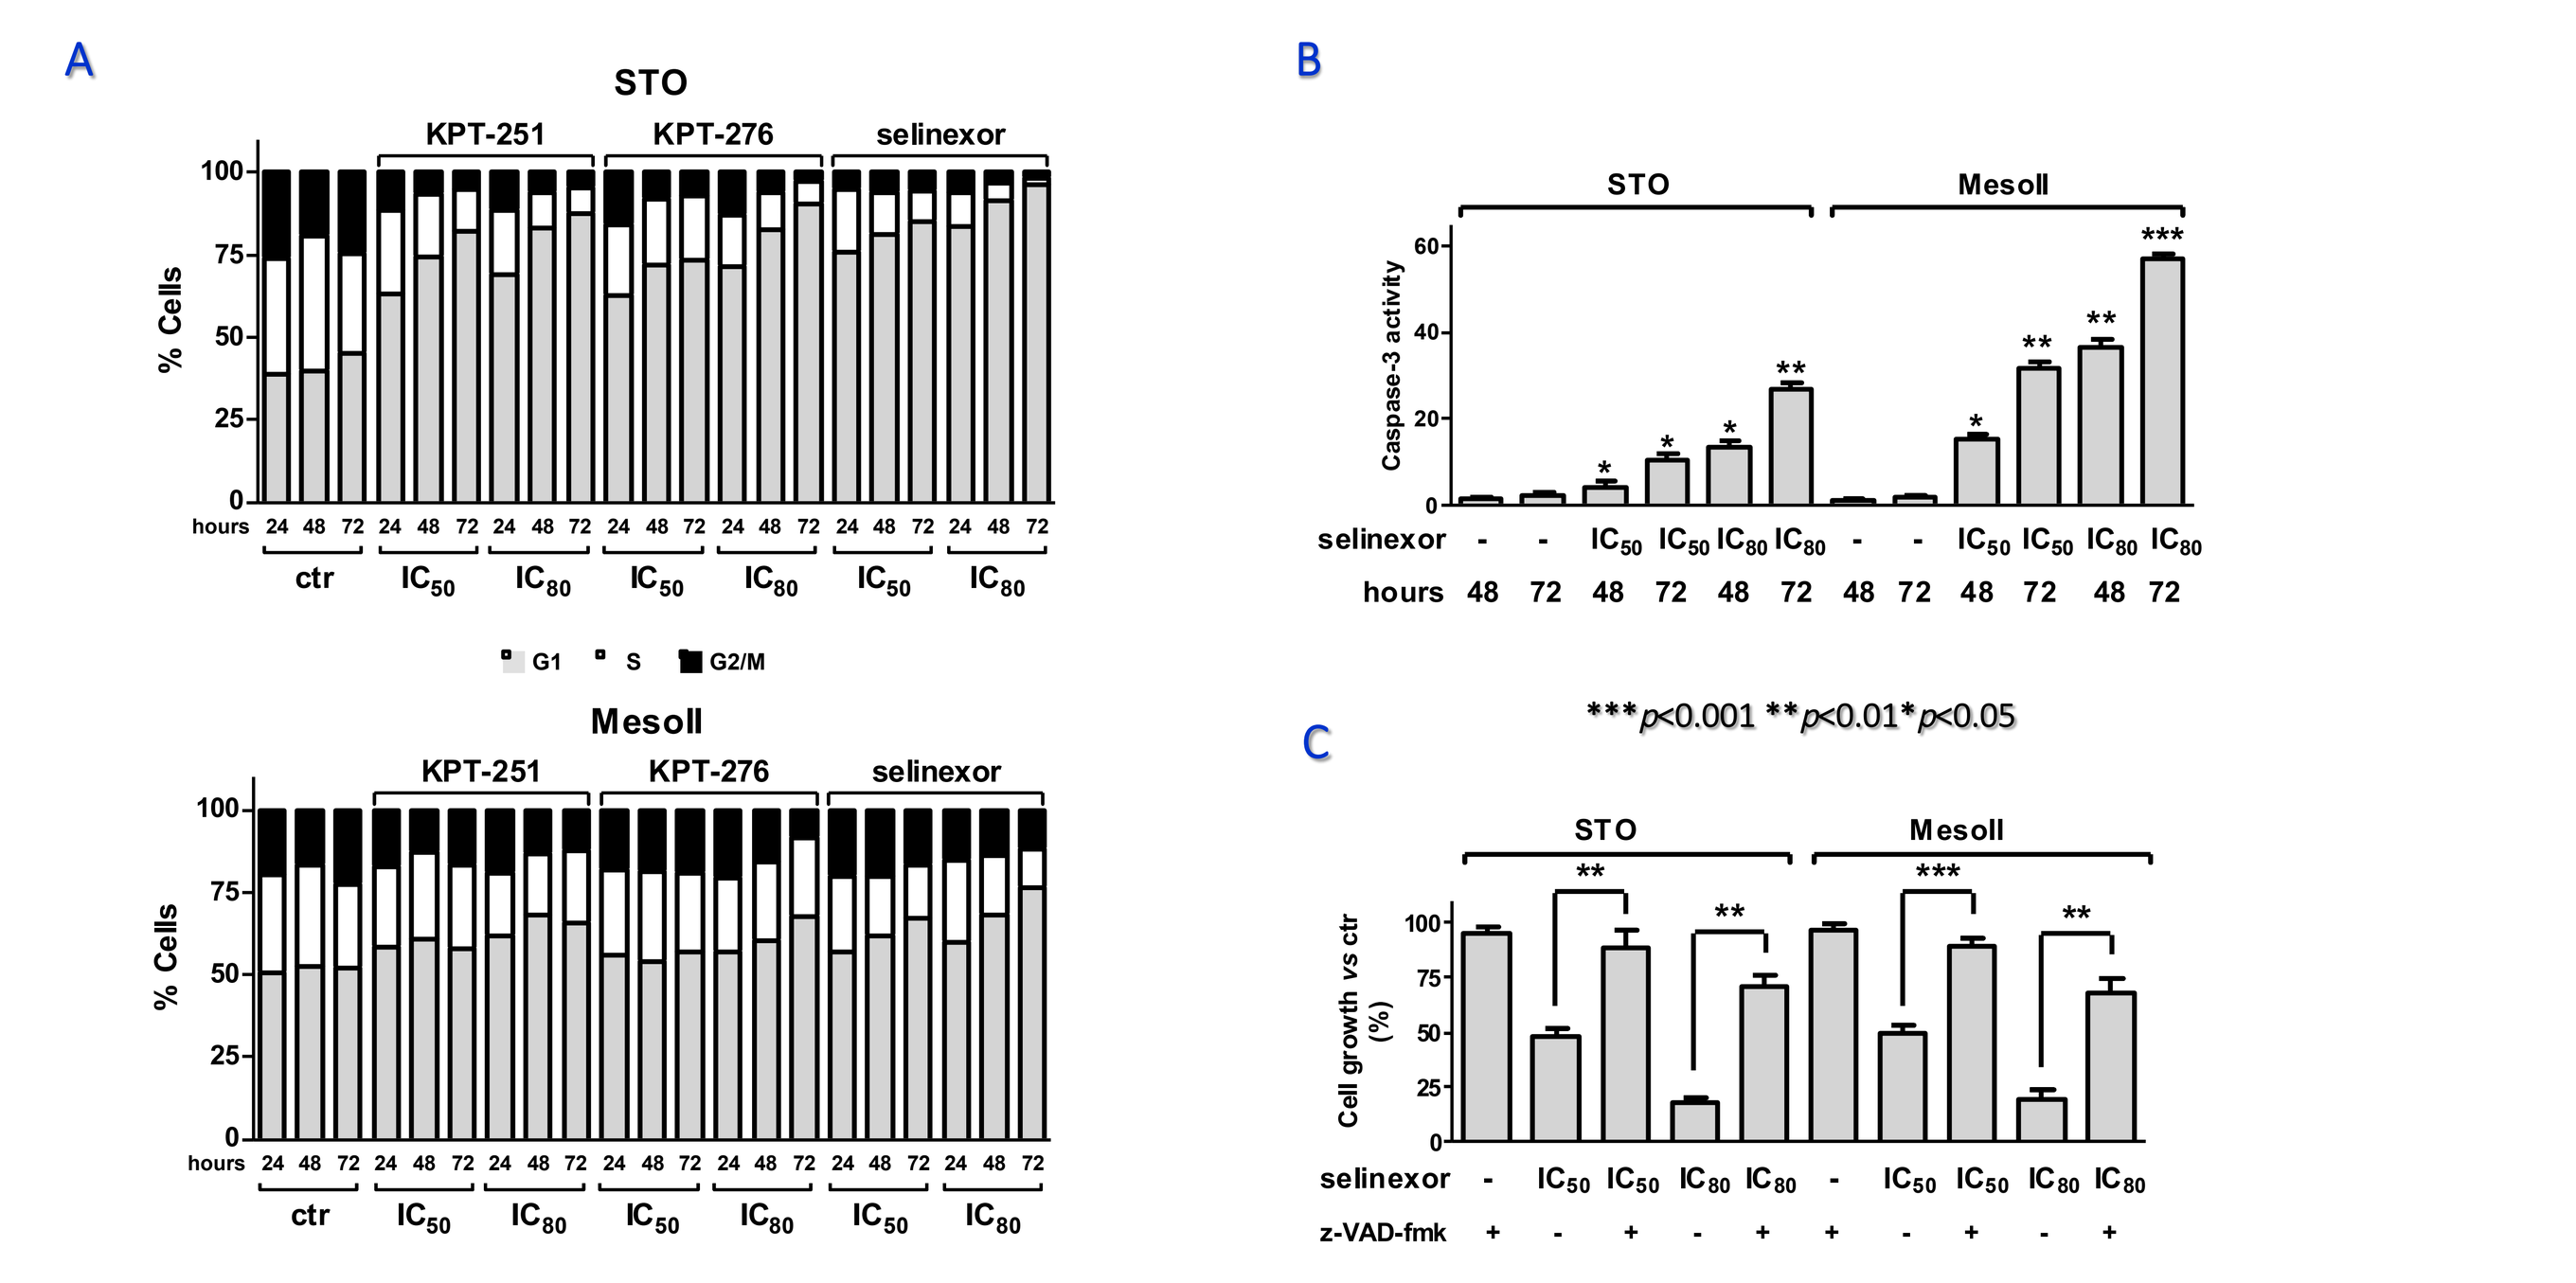

Flow cytometry profiles of nuclear DNA content revealed that 24-hour treatment of STO cells with SINE was sufficient to induce an accumulation of cells in G1 phase and a reduction in the percentage of cells in S and G2/M compartments (Fig.A). G1 phase accumulation markedly increased at 48 hours and reached a maximum 72 hours-post exposure to the highest doses of SINE (87.6±3.7%, 90.4±1.8% and 96.1±3.3% for KPT-251, KPT-276, and selinexor, respectively) (Fig.A). Although to a lesser extent compared to STO cells, an increase in the percentage of cells in G1 phase was appreciable following 72-hour exposure to the highest selinexor concentration in MesoII cells (Fig.A). In addition, a significant dose- and time-dependent increase in caspase-3 catalytic activity, as determined in vitro by the hydrolysis of the specific fluorogenic substrate, was found after treatment with each compound (Fig.B). Notably, the inhibitory effect of SINE on cell growth was almost completely reverted when DMPM cells were pretreated with the pan-caspase inhibitor z-Val-Ala-Asp-fluoromethylketone (z-VAD-fmk; Fig.C) -which by itself did not impair cell growth-, providing evidence that SINE induce a caspase-dependent apoptotic cell death in DMPM cells.

SINE inhibit XPO1/CRM1 functions, interfere with survivin subcellular distribution and promote its proteosome-dependent degradation

SINE modulate nuclear levels of XPO1/CRM1 and its cargo protein

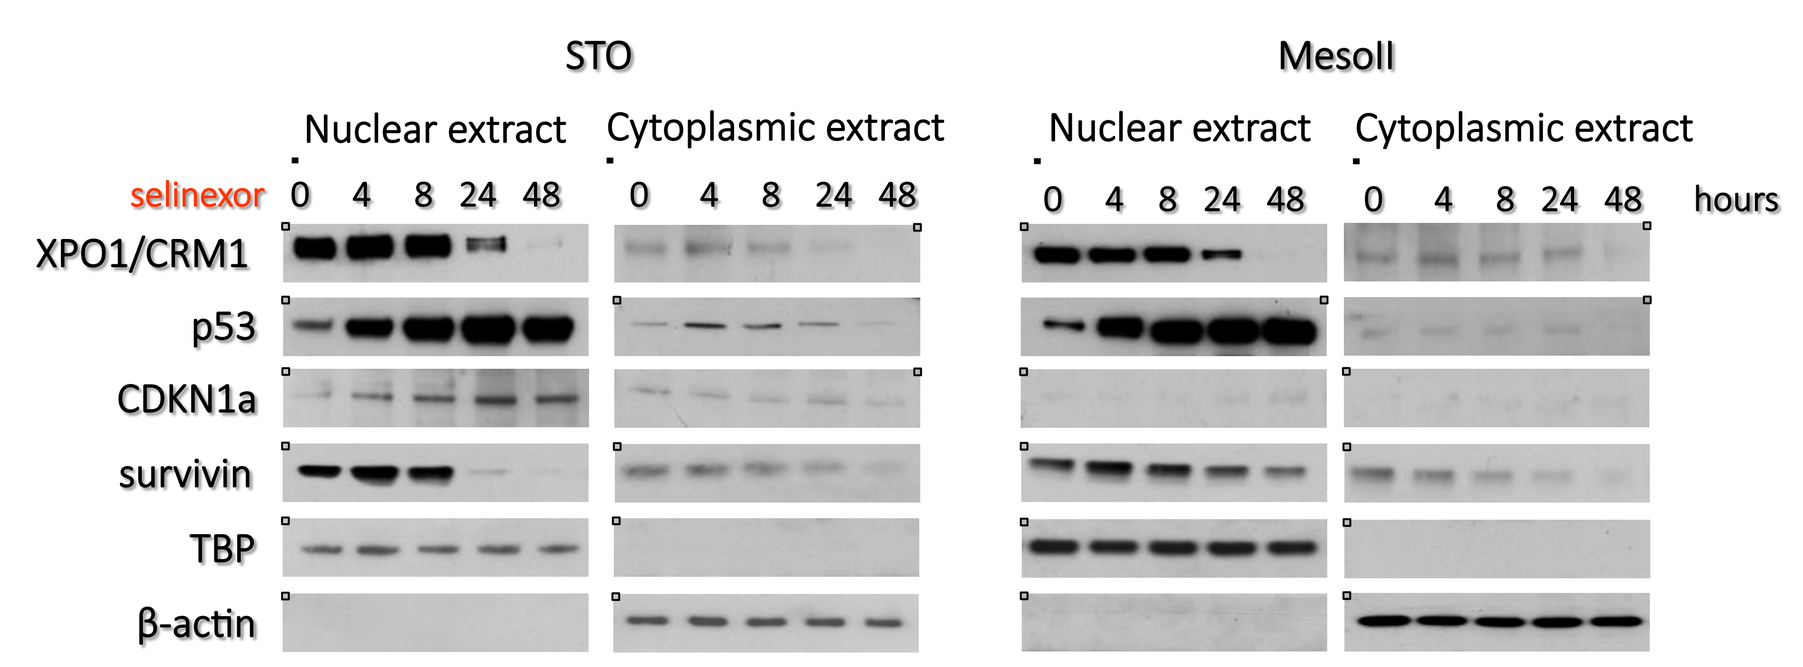

Immunoblotting analysis revealed that nuclear XPO1/CRM1 expression progressively decreased after SINE treatment. In addition, the compounds induced nuclear accumulation of p53 as early as 4 hours-post treatment initiation in both cell lines, whereas CDKN1a nuclear accumulation was observed only in STO cells.

SINE interfere with the subcellular localization of survivin and induce its downregulation through the ubiquitin/proteosome pathway

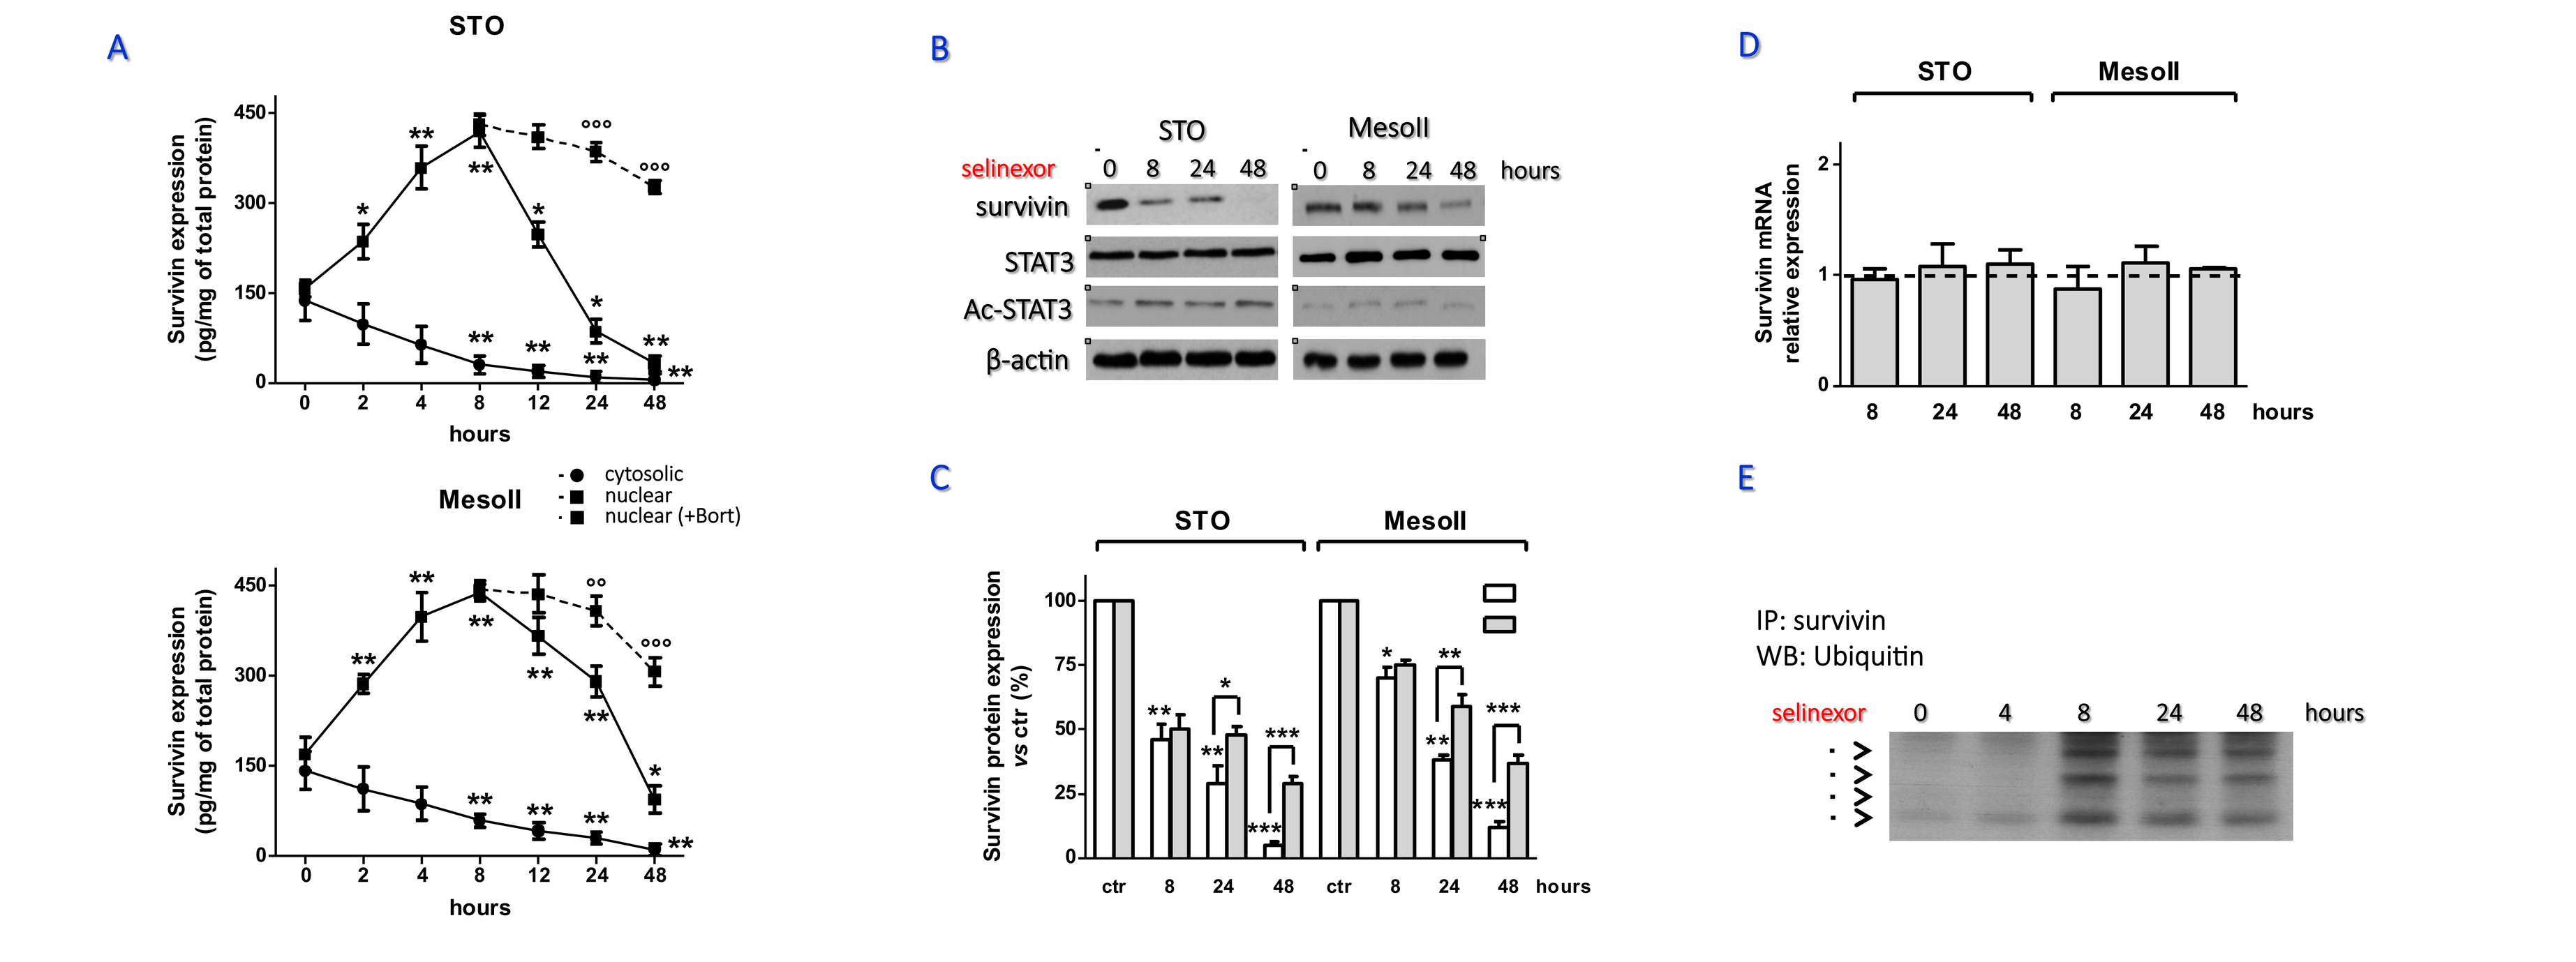

SINE treatment (at IC_{50}) induced nuclear accumulation of survivin concomitant with a time-dependent cytoplasmic reduction (Fig. A). Survivin nuclear accumulation was observed as early as 2 hours-post exposure to each compound and it reached a maximum 8 hours-post treatment initiation. Strikingly, starting from 12 hours-post treatment initiation, a progressive decrease in nuclear survivin protein abundance was observed (Fig. A), resulting in a significant and time-dependent reduction of total protein amount (Fig.B,C). To analyze the mechanism involved in survivin down-regulation, we assessed STAT3 protein expression and acetylation in DMPM cells following selinexor treatment by Western blot (Fig.B). However, no measurable effects on protein levels and acetylation status were observed. Our data suggest that the decrease of survivin protein abundance in DMPM cells is not related to post-translational modifications of its well-known transcriptional activator. Such a hypothesis is also corroborated by the evidence that exposure of DMPM cells to selinexor did not affect survivin mRNA expression (Fig.D). We then checked whether selinexor-mediated XPO1/CRM1 inhibition might lead to the ubiquitination of survivin nuclear fraction. Western blot experiments indicated that exposure of DMPM cells to selinexor resulted in multiple ubiquitination of survivin, which increased its molecular weight up to 100 kDa (Fig.E). These results suggest that in DMPM cells the reduction of survivin nuclear fraction by selinexor is ascribable at least in part to its proteasome-dependent degradation. Indeed, exposure of STO cells to the proteosome inhibitor Bortezomib partially restored nuclear survivin levels in selinexor treated cells (Fig.A).

Synergistic cytotoxic effect of selinexor/YM155 combinations

Synergistic cytotoxic effect of selinexor /YM155 combinations

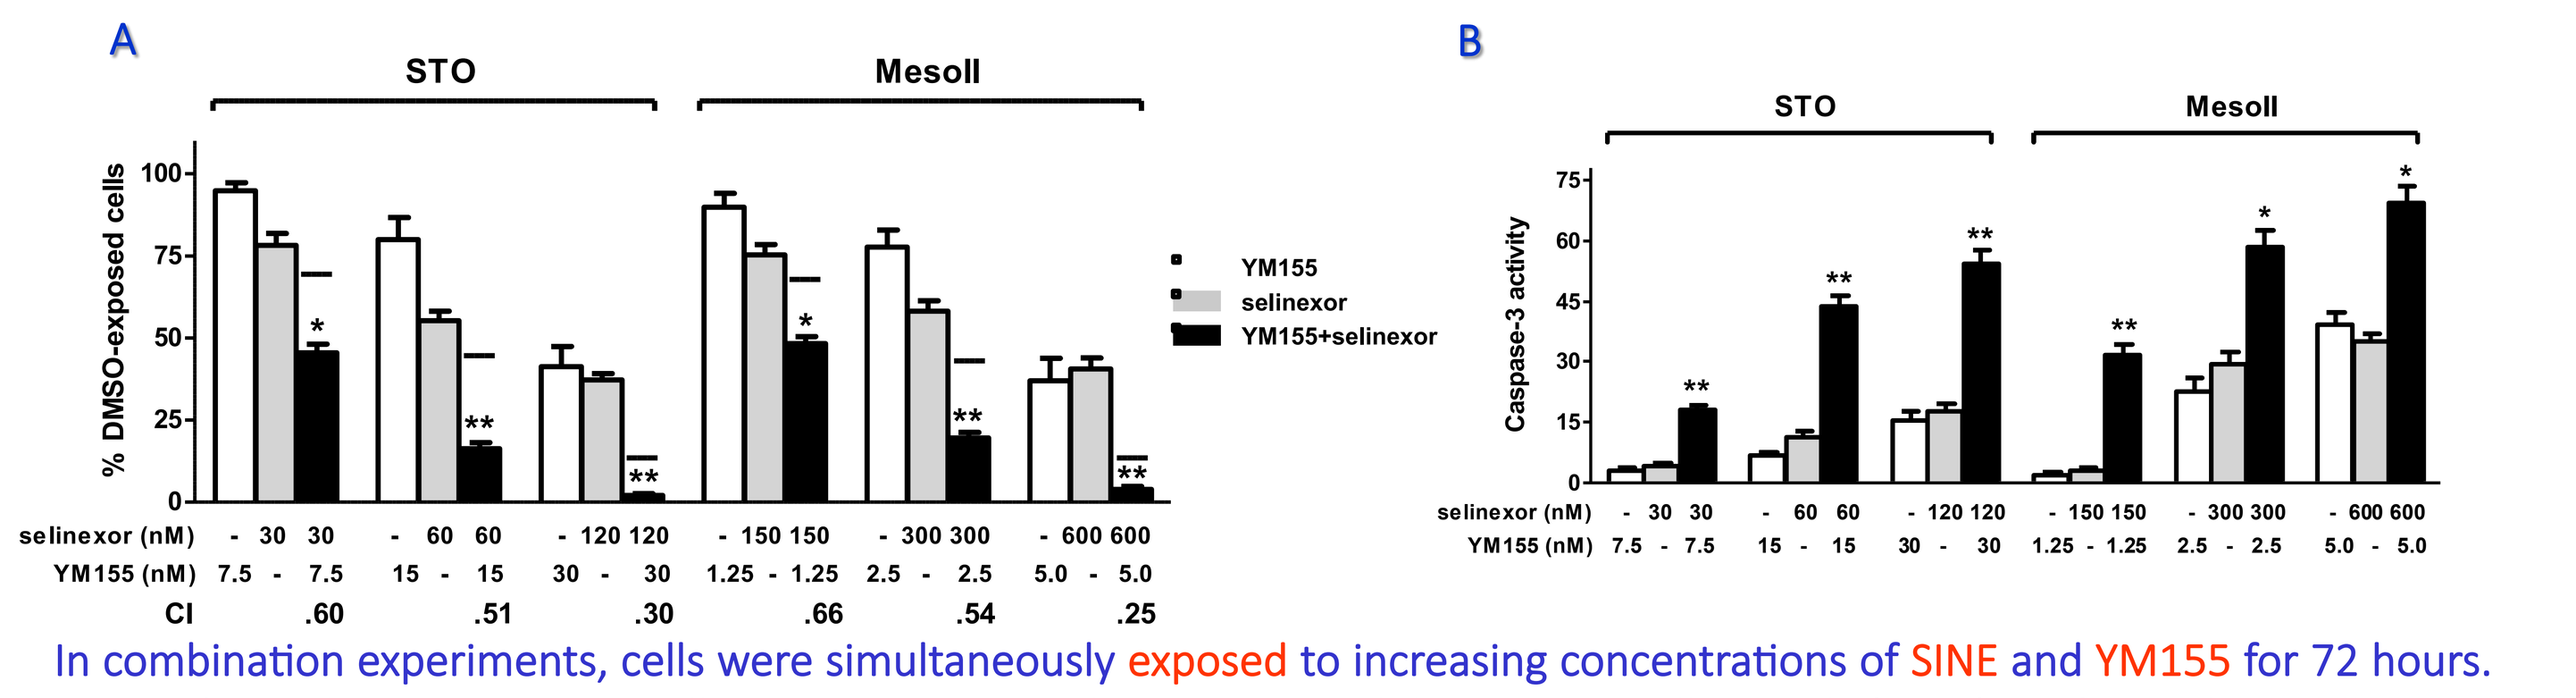

In combination experiments, cells were simultaneously exposed to increasing concentrations of SINE and YM155 for 72 hours. SINE effectively cooperated at all concentrations with YM155 to inhibit DMPM cell growth (Fig.A). In fact, when cells were treated with the drug combinations, the inhibition of cell proliferation was consistently greater than that expected by simple additivity of the effects of the individual drugs. Such a synergistic interaction was drug concentration-dependent, as indicated by the progressive decrease of combination index (CI) values. Moreover, caspase-3 catalytic activity was consistently and significantly higher in cells treated with the selinexor/YM155 combination than in cells exposed to single agents (Fig.B).

Efficacy of oral SINE against DMPM xenografts

Oral SINE show anti-tumor activity in DMPM xenografts

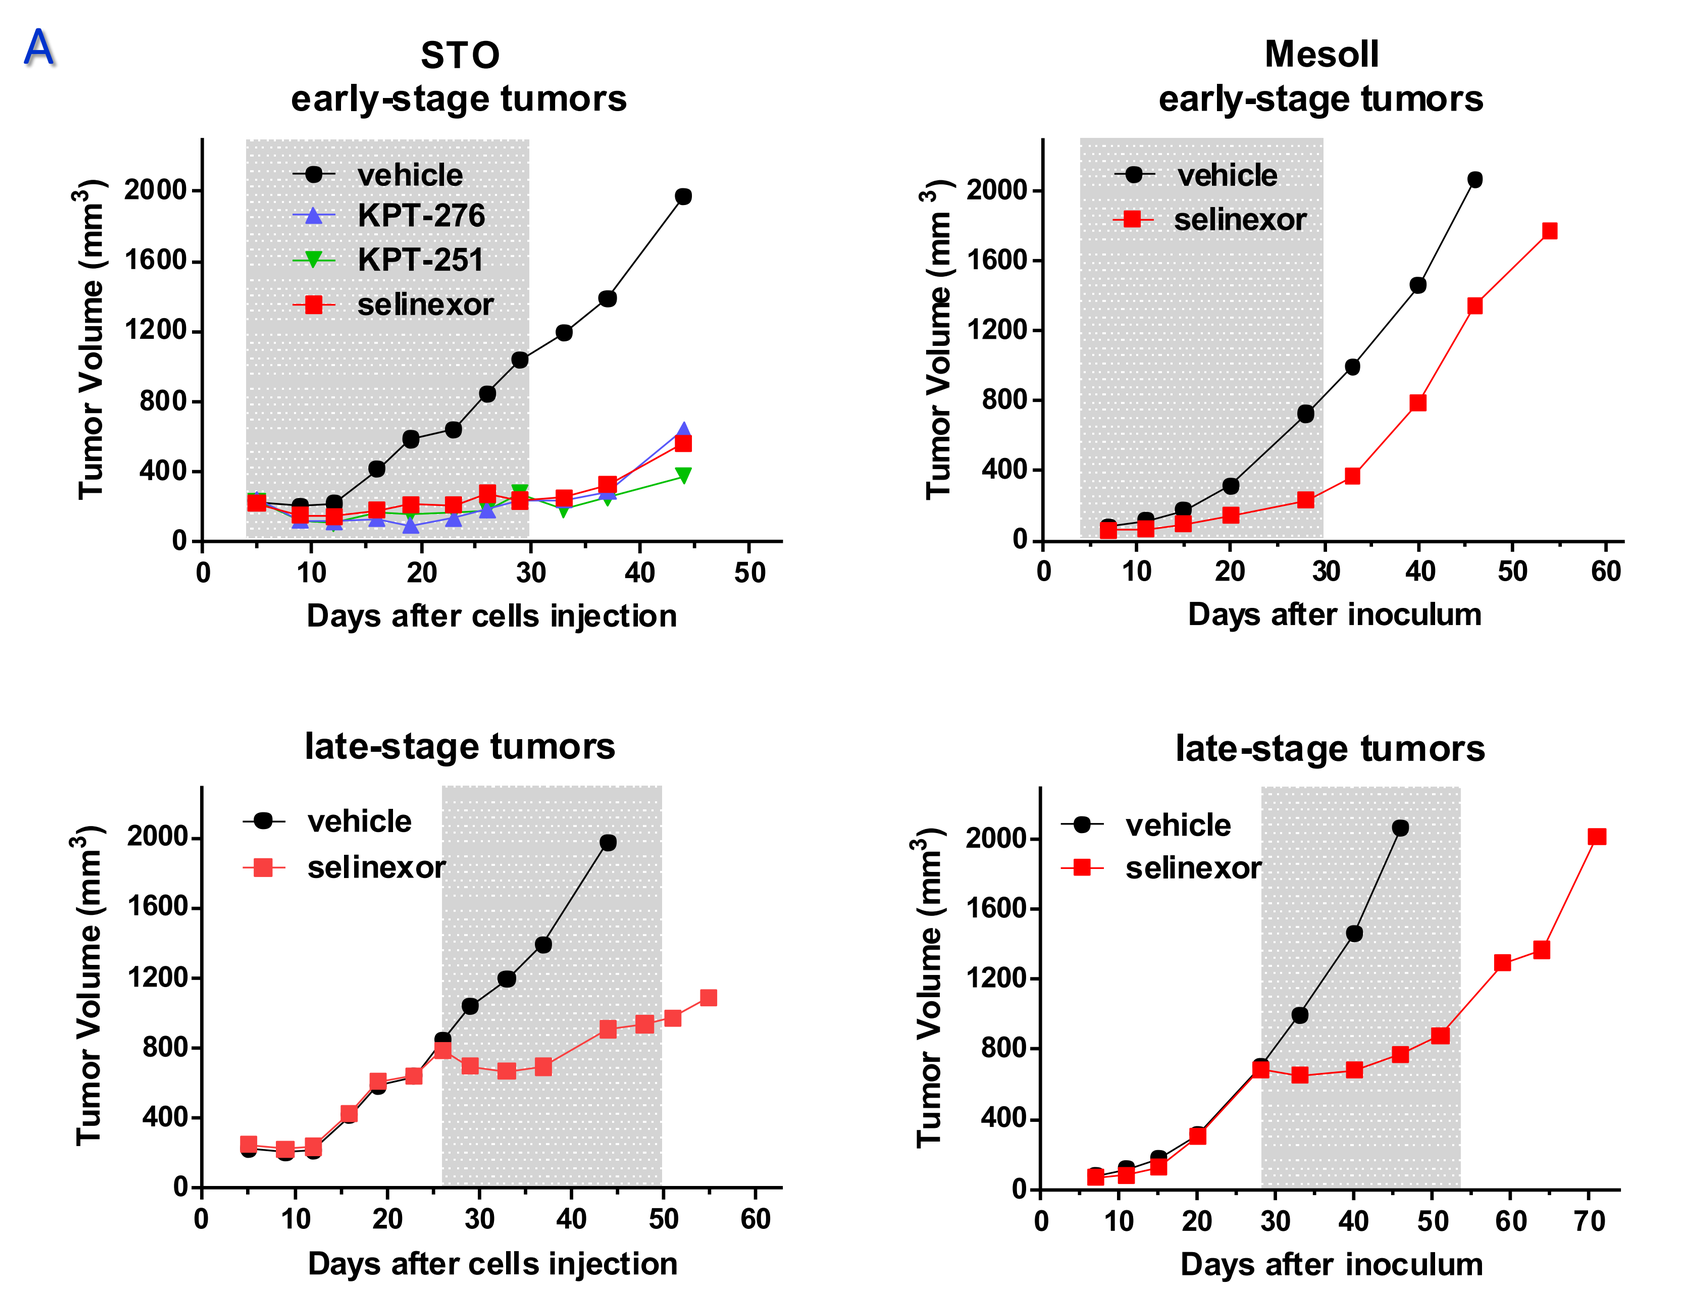

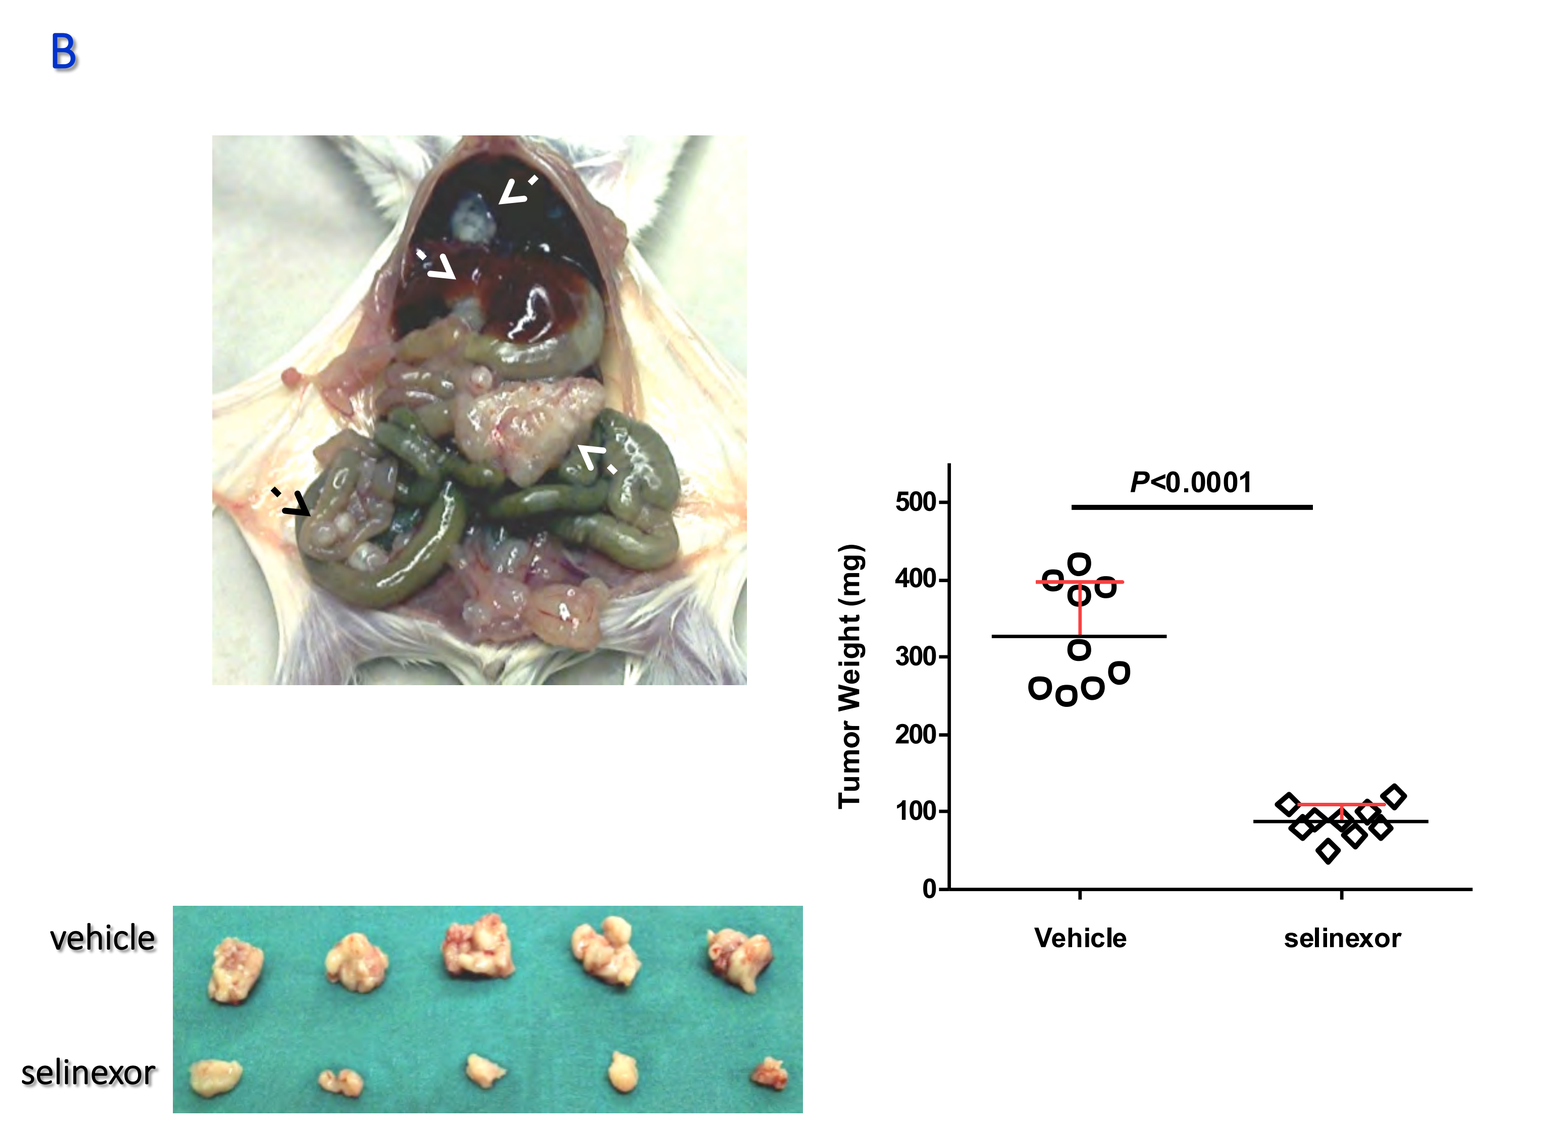

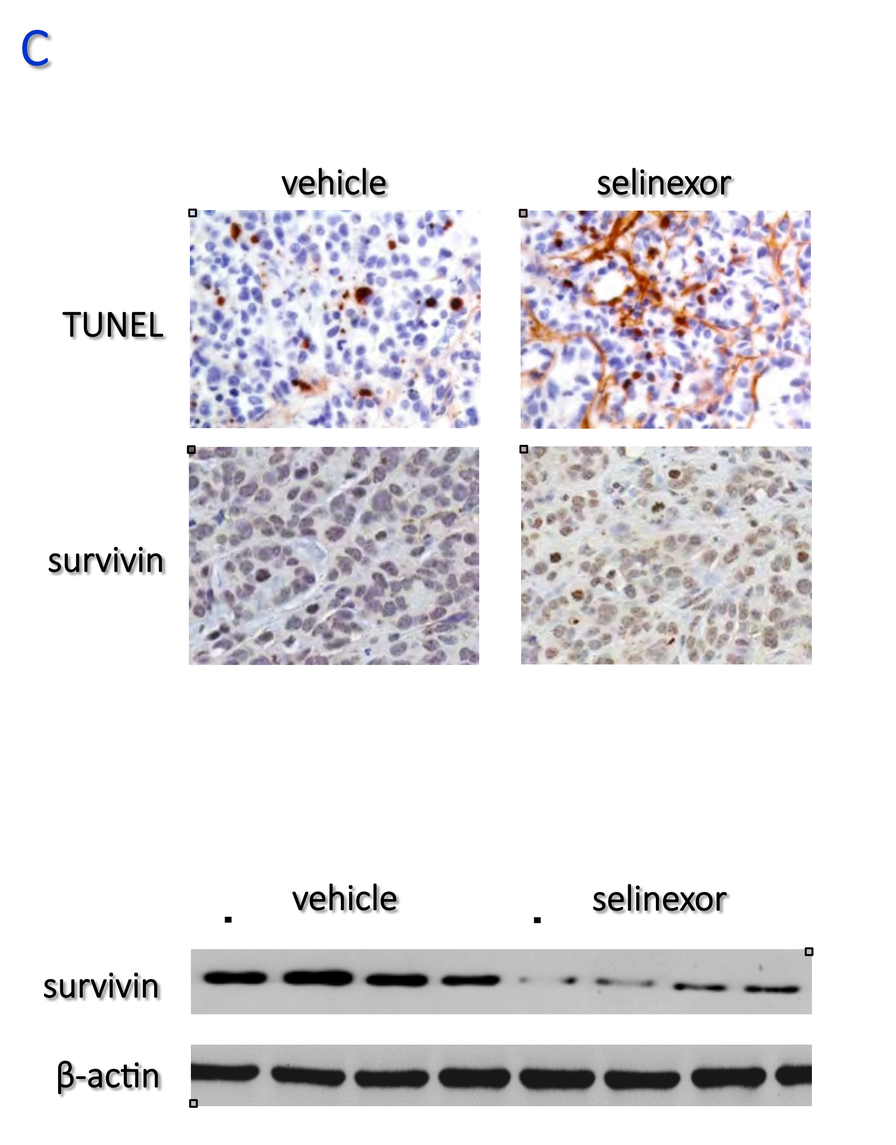

In vivo activity of KPT-251, KPT-276 and selinexor was initially tested against early-stage subcutaneous STO xenografts in nude mice. A remarkable and superimposable anti-tumor effect was observed after treatment with the different agents (Fig.A), and a stabilization of tumor volume was appreciable up to 2 weeks post drug withdrawal (FigA). Although to a lesser extent compared to early-stage tumors, the clinically available compound selinexor produced a significant tumor growth inhibition even in late-stage STO tumors (Fig.A). In addition, selinexor significantly inhibited the growth of both early- and late-stage subcutaneous MesoII tumors (Fig.A). Strikingly, in late-stage STO and MesoII tumors, the growth was dramatically slowed at the beginning of the treatment, and tumor volumes kept almost constant during the course of drug administration (Fig.A). The anti-tumor activity of selinexor was further investigated in STO cells orthotopically xenotransplanted into SCID mice. Twenty-five days after cells i.p. injection (i.e., 24 hours after the last treatment), mice were euthanized and tumors were removed. At necropsy, control (vehicle-treated) mice showed a large tumor mass at the site of cell injection mainly invading the peritoneum wall, and widespread small nodules in the peritoneum and attached to the diaphragm, liver and bowel (Fig.B), resulting in a tumor burden (average±SD mg) of 328±69 mg. In selinexor-treated animals, the size of the single residual tumor mass -which was adherent to the peritoneum, in the site of cell injection- was significantly reduced (88±21 mg) compared to control mice (Fig.B). In addition, TUNEL and survivin immunohistochemical staining of tumor sections obtained from orthotopic xenografts revealed increased apoptosis and reduced survivin expression at both nuclear and cytoplasmic cellular compartments (Fig.C) in selinexor-treated compared to control mice. These results were further corroborated by Western blot analysis performed on frozen tumor samples. SINE were well tolerated, with no toxic deaths and minimal weight loss (<5%). In addition, no gross pathology was observed at necropsy carried out at the end of each experiment. Images for one representative mouse per group are shown.