Selinexor (KPT-330) Radio-Sensitizes Non-Small Cell Lung Cancer Cells *In-Vitro* and *In-Vivo*

Tami Rashal[1], Sivan Elloul[1], Marsha Crochiere[1], Trinayan Kashyap[1], William Senapedis[1], Ryan George[2], Sharon Friedlander[1], Maya Ilouze[3], Yosef Landesman[1], Robert O. Carlson[1], Nir Peled[3], Michael Kauffman[1], Sharon Shacham[1], Yaacov Lawrence[3]

(1)Karyopharm Therapeutics Inc, Newton, MA; (2)SBH Sciences, Natick, MA; (3)Tel HaShomer Hospital, Ramat--Gan, Israel

ABSTRACT

The primary nuclear export protein, Exportin 1 (XPO1/CRM1), is overexpressed in most cancers and this overexpression is frequently correlated with poor prognosis. Selective Inhibitors of Nuclear Export (SINE) compounds are a family of small-molecule bioavailable drugs that bind covalently to XPO1 to inhibit nuclear export. This results in nuclear retention of major tumor suppressor proteins, such as p53, pRB, FOXO3A and other critical proteins in cancer biology, which leads to selective cancer cell death. Selinexor is the most advanced SINE with >500 cancer patients (hematological and solid tumors) treated to date in Phase I/II clinical trials. Blocking XPO1 function also blocks nuclear transport of key DNA damage repair (DDR) proteins and we therefore hypothesized that combination of selinexor with radiation therapy (RT) would lead to synergistic anti-tumor activity. Radiation-chemotherapy combination is an established therapeutic option for patients with medically inoperable non-small cell lung cancer (NSCLC), but its use is limited due to tolerability and adverse effects. Moreover, this treatment strategy typically has short-term benefits and the patients relapse with progressive disease.

Here we report on studies designed to test whether selinexor could interfere with DDR and synergize with radiation to enhance anti-tumor potency relative to either treatment alone. Cell cycle analysis and clonogenic assays were conducted in the presence or absence of selinexor in combination with escalating doses of RT on the radiation-sensitive and radiation-insensitive H1299 and A549 cell lines, respectively.

Interestingly, selinexor-treatment induced G1 cell cycle arrest in the p53-deficient cell line H1299 and a prominent G2 cell cycle arrest in p53-wild type A549 cells 24 hrs post treatment. Furthermore, clonogenic assays revealed that selinexor + RT treatment act synergistically to produce a dose-dependent growth inhibition in both cell lines. *In-vivo* combination treatment in the A549 NSCLC xenograft model with low doses of selinexor and RT displayed a strong synergistic effect for reduction of tumor volume. In addition, microscopic and immunohistochemical analysis of the resected tumors showed an overall reduction in tumor cell numbers, increased fibrosis and induction of apoptosis in the selinexor-RT treated tumors compared to controls. Finally, mechanistic studies revealed that selinexor does not induce DNA damage like RT, but downregulates CHK1, RAD51 and other DDR proteins.

Together, our results suggest that selinexor treatment sensitizes cells to RT by preventing single-stranded DNA break repair via downregulation of DDR protein expression. In contrast, selinexor induces cell cycle arrest at G1 in p53 deficient H1299 cells, allowing DNA damage accumulation and induction of apoptosis. These results provide a rationale basis for combining selinexor with RT in clinical trials studies.

INTRODUCTION

Exportin-1 (XPO1; Crm1) is the major nuclear exporter with >200 protein cargos, including proteins and some RNAs central to carcinogenesis, viral replication, and inflammation.

- XPO1 is overexpressed in several cancer indications and its levels correlate with poor prognosis.
- SINEs induce nuclear retention of proteins and RNAs to exert effects beneficial for a variety of disease states, including cancer, viral infection, brain damage and a variety of autoinflammatory conditions.
- Selinexor, the most advanced SINE, has been tested in >500 patients to date in three ongoing Phase 1 trial, with promising signs of efficacy, tolerability and safety.

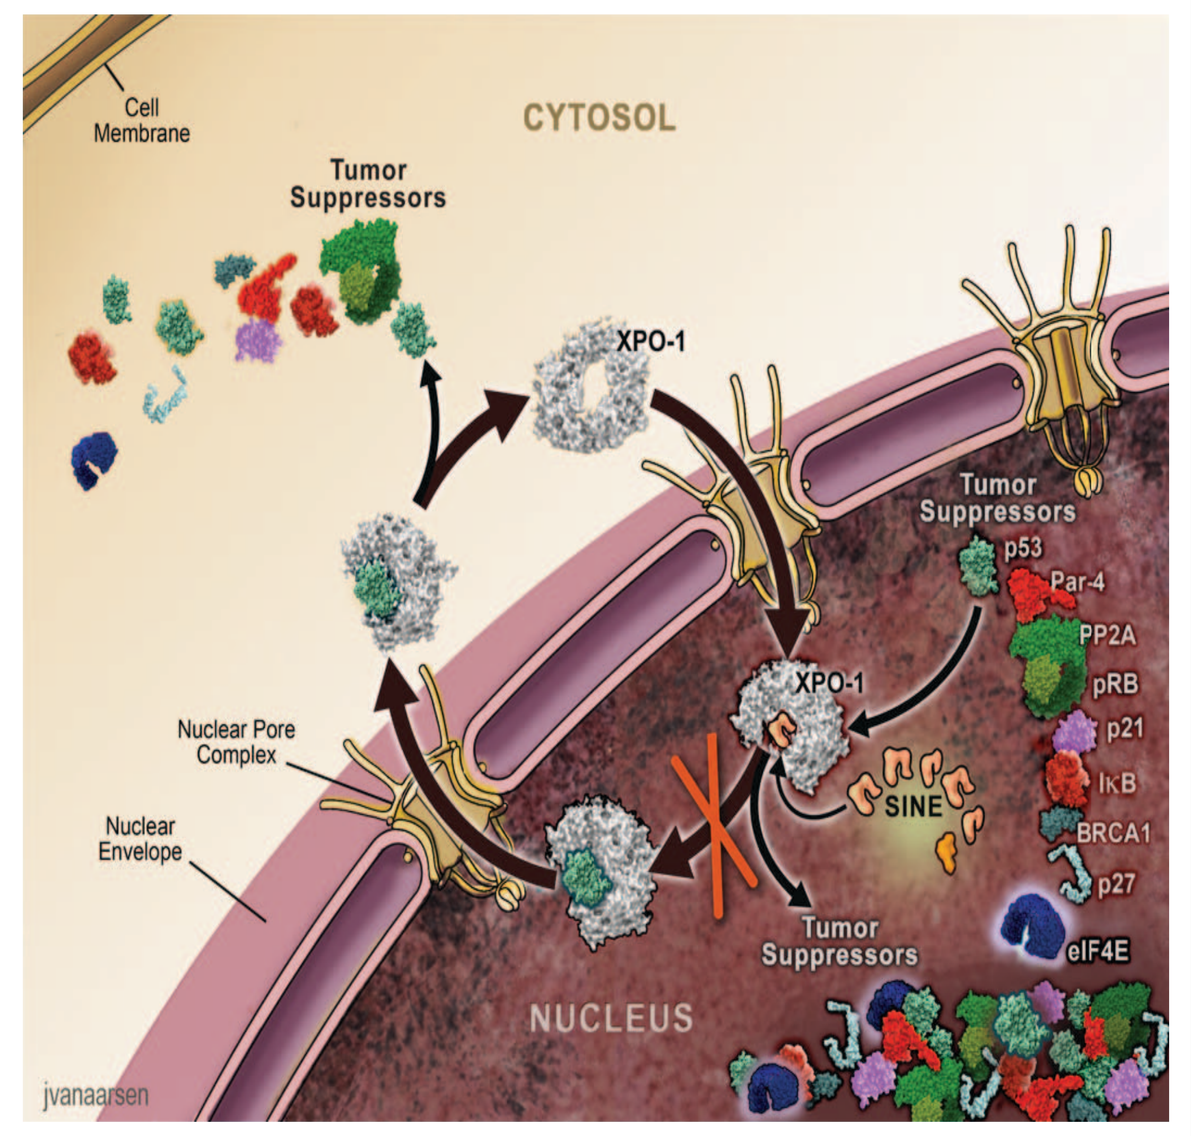
CYTOSOL
Cell Membrane
Tumor Suppressors
XPO-1
Tumor Suppressors
p53
Par-4
PP2A
pRB
p21
IkB
BRCA1
p27
eIF4E
XPO-1
SINE
Nuclear Pore Complex
Nuclear Envelope
Tumor Suppressors
NUCLEUS
jvanaarsen

XPO1 is Expressed in All NSCLC Cells Tested

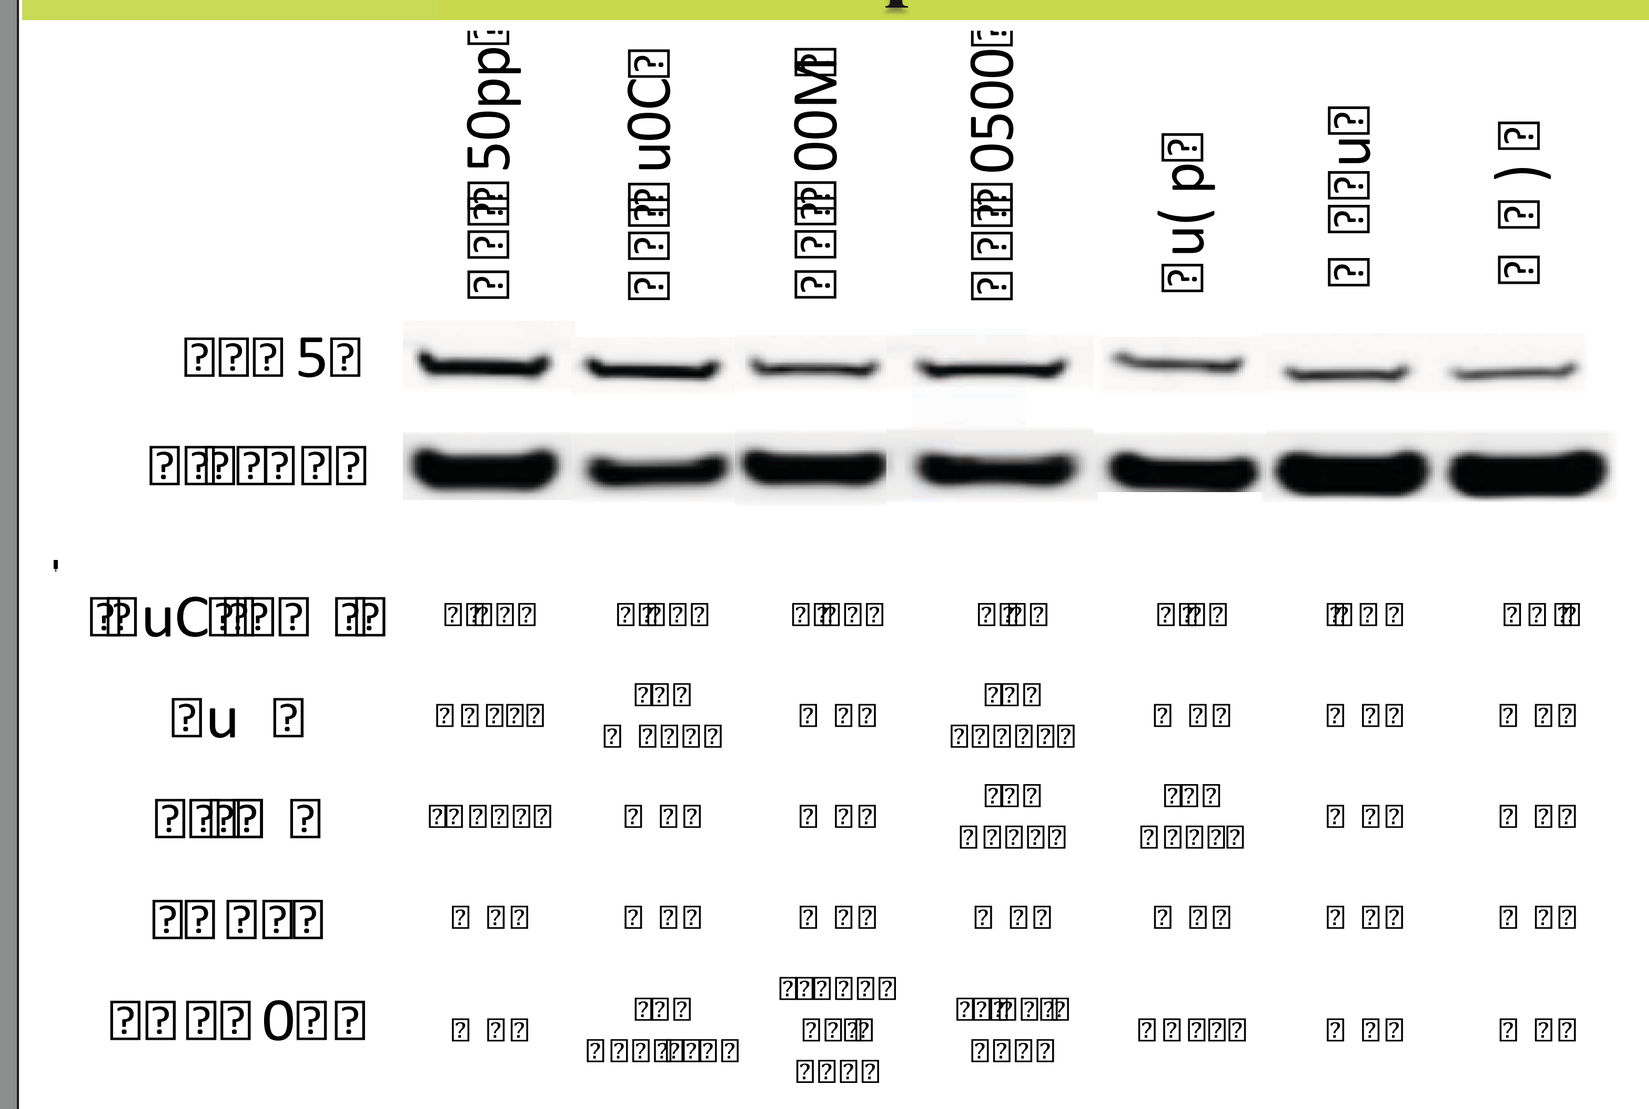

NSCLC cells express XPO1. Immunoblot of XPO1 expression in 5 NSCLC (NCIH1299, NCIH520, NCIH226, NCIH2122, A549) and 2 normal lung fibroblast cell lines (MRC5, WI38). Corresponding IC50 values (nM), calculated from a standard MTT assay, and genotype are listed in columns below. (WT = wild type, NULL = homozygous deletion, ND = not determined)

Selinexor Induces Phase-specific Cell Cycle Arrest in Different NSCLC Cell Lines. G1 Arrest in H1299 and G2 Arrest in A549

H1299 and A549 cells were treated with 1 and 5 uM selinexor, respectively, for 1 and 3 days, and were evaluated for BrdU incorporation and DNA content by flow cytometry. Applying selinexor to the more sensitive H1299 cells reduced but did not eliminate S phase, induced G1 arrest and increased the sub-G1 population. In contrast, A549 cells treated with selinexor eliminated S phase, induced both G1 and G2 arrest, and showed increased sub-G1 population, although not to the same extent as observed in selinexor treated H1299 cells.

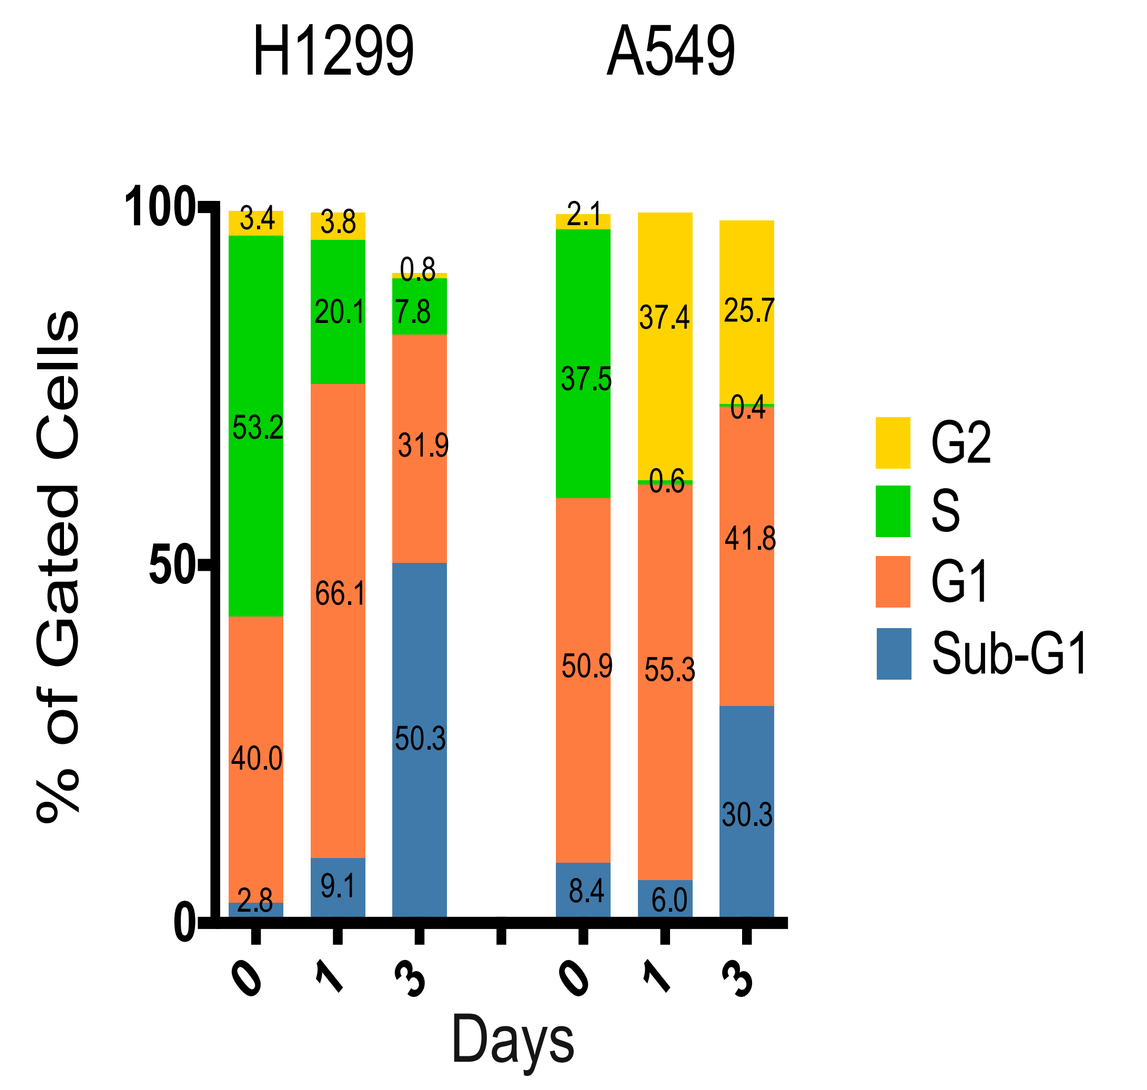
H1299
A549
% of Gated Cells
Days
G2
S
G1
Sub-G1

Synergistic Antitumor Effects of Selinexor and RT Combination on NSCLC Cells

Clonogenic assay - A549 (A&C), H1299 (B&D)

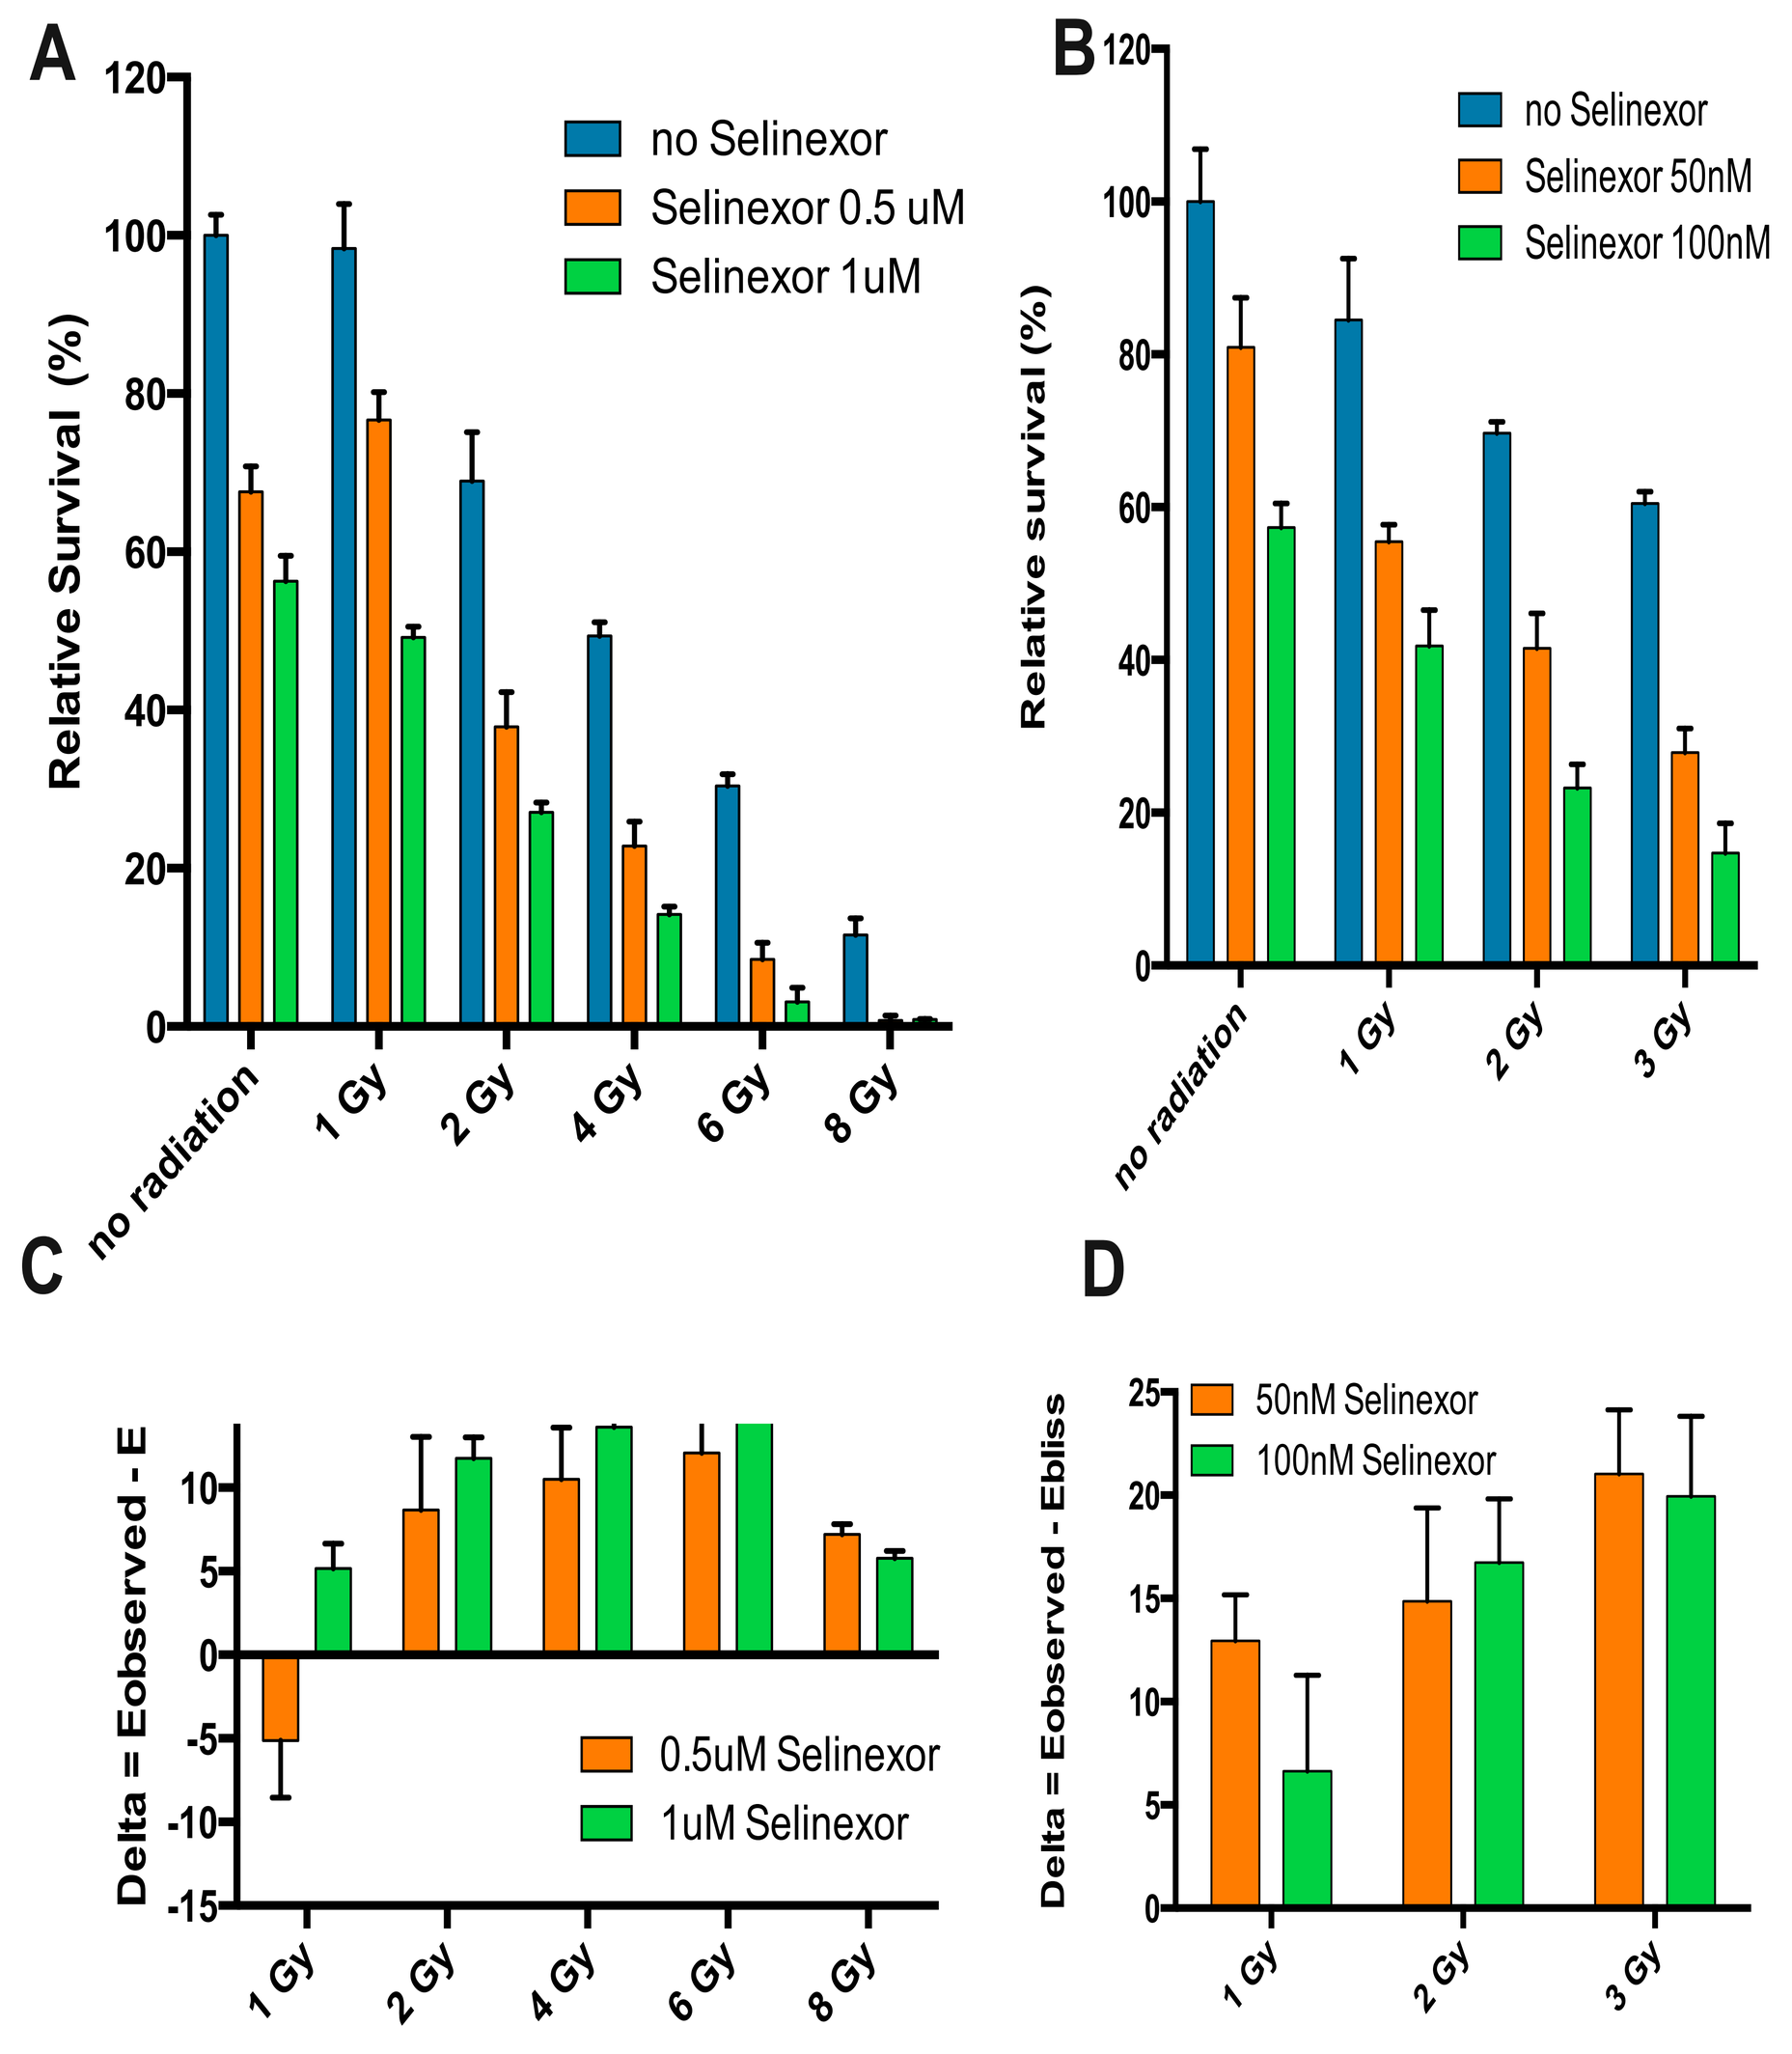
A
no Selinexor
Selinexor 0.5 uM
Selinexor 1uM
Relative Survival (%)
no radiation
1 Gy
2 Gy
4 Gy
6 Gy
8 Gy
B
no Selinexor
Selinexor 50nM
Selinexor 100nM
Relative survival (%)
3 Gy
C
Delta = Eobserved - E
0.5uM Selinexor
1uM Selinexor
D
Delta = Eobserved - Ebliss
50nM Selinexor
100nM Selinexor

Clonogenic assay showing percent survival of A549 (**A**) and H1299 (**B**) cells 8 days after treatment with selinexor and radiation therapy. The Bliss Additive Model : A549(**C**), H1299 (**D**)) was used to determine the synergistic, additive or antagonistic effects of combination therapy using the equation:

$E_{Bliss} = E_A + E_B - E_A{*}E_B$ in which EA and EB are the fractional inhibitions obtained by selinexor alone or radiation alone at specific concentrations.

Using this model, if Delta=Eobserved – EBliss is negative the effect is antagonistic, 0 is additive and positive is synergistic.

Synergistic Tumor Growth Inhibition Effect of Selinexor and RT Combination Treatment on A549-derived Xenografts

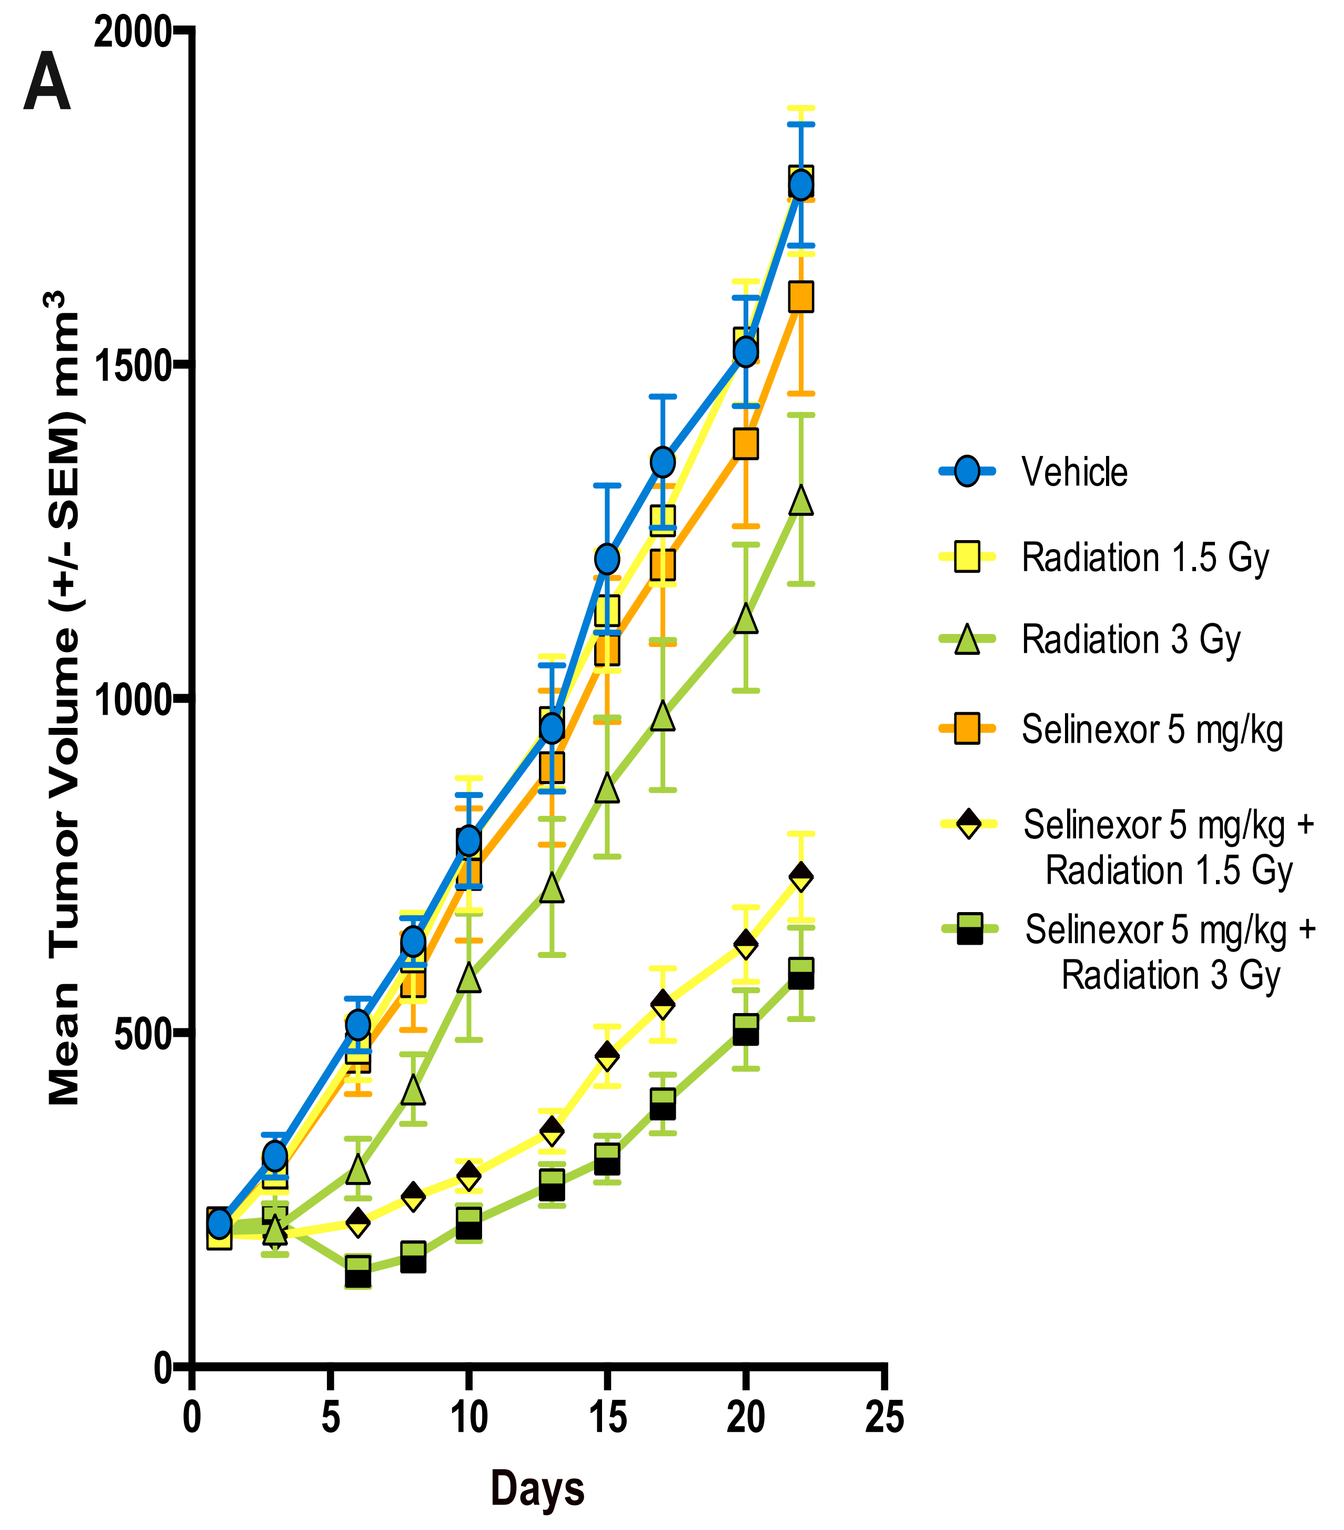
A
Mean Tumor Volume (+/- SEM) mm3
Days
Vehicle
Radiation 1.5 Gy
Radiation 3 Gy
Selinexor 5 mg/kg
Selinexor 5 mg/kg + Radiation 1.5 Gy
Selinexor 5 mg/kg + Radiation 3 Gy

Synergistic antitumor effects of selinexor and RT on NSCLC xenografts.

(**A**) Tumor volume of A549 xenografts in NOD/SCID mice treated with vehicle, selinexor (5 mg/kg ,PO), local radiation therapy (1.5 or 3 Gy) or combination selinexor and local radiation therapy for 22 days.

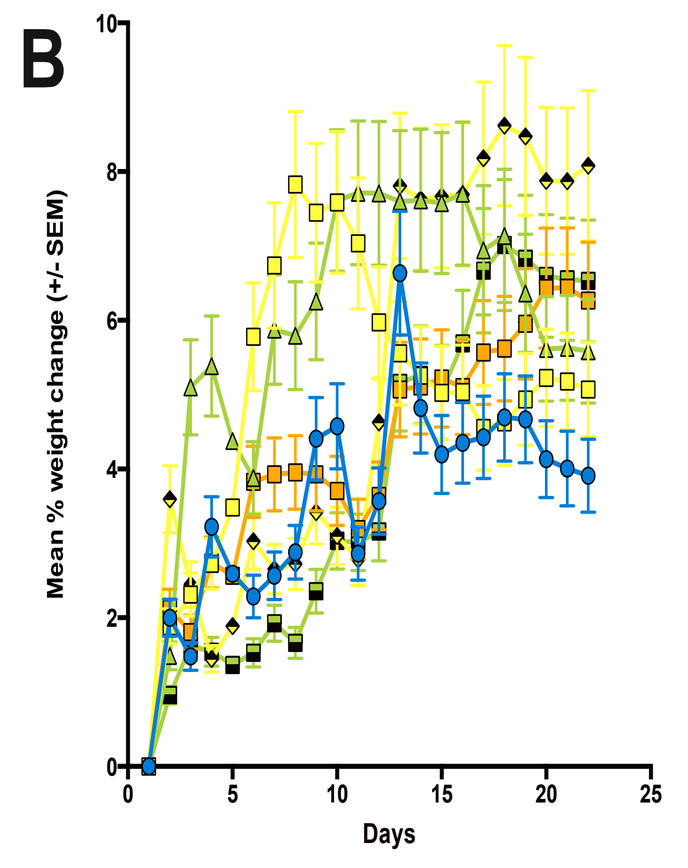
B
Mean % weight change (+/- SEM)
Days

(**B**) All treatments were well tolerated as indicated by weight gain.

Selinexor and RT Combination Treatment on A549-derived Xenografts

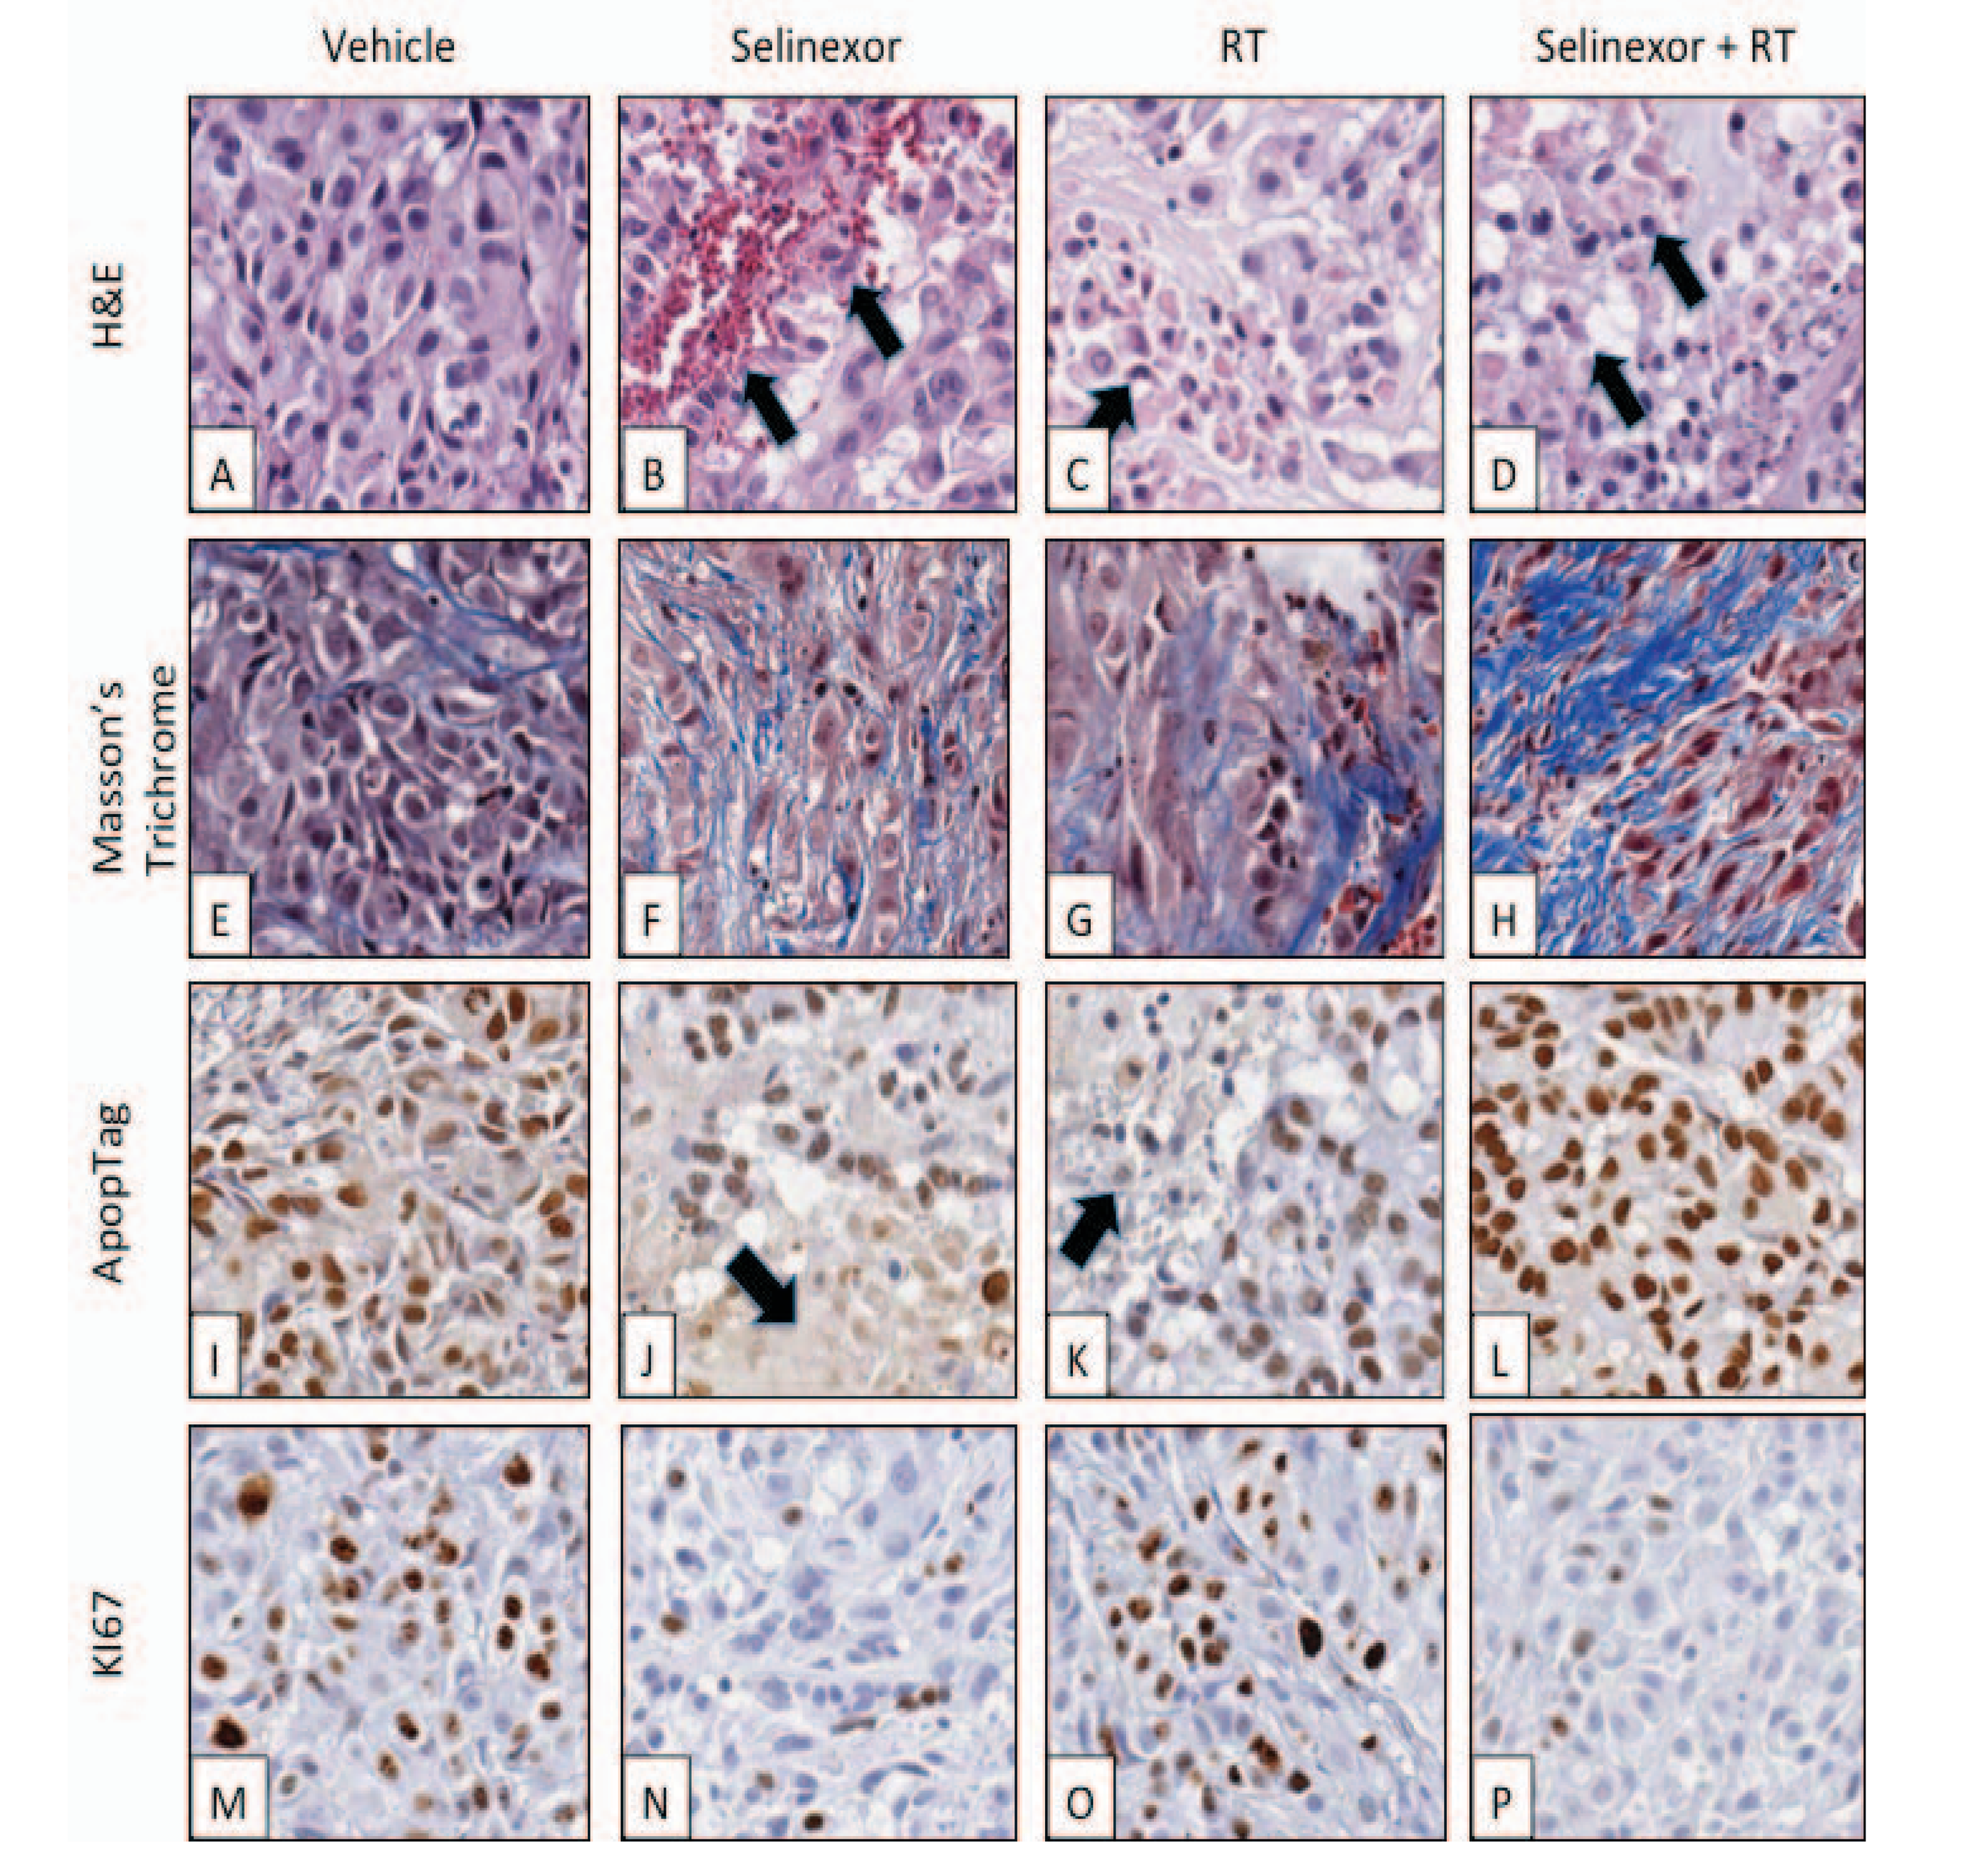
Vehicle
Selinexor
RT
Selinexor + RT
H&E
Masson's Trichrome
ApopTag
KI67

Selinexor-RT-treated xenografts exhibit reduction in tumor cell population, reduction in tumor cell proliferation, increased necrosis and apoptosis

(A-D) H&E staining. Increase in necrotic area (black arrows) and an overall reduction in tumor cell population is observed in all treatment groups compared to vehicle control (B-D Vs. A). (E-H) Masson's Trichrome staining show an increase in necrotic area (blue) in all treatment groups compared to vehicle control (F-H Vs. E), most predominantly in selinexor-RT combination-treated xenografts (H). (I-L) Apoptag staining. Increase in staining for the apoptosis marker Apoptag is observed in selinexor-RT combination-treated xenografts compared to each agent alone (L vs. J & K). Black arrows point to necrotic areas.

(M-P) KI67 staining. Reduction in number of positively stained cells is observed in selinexor-treated- and selinexor-RT combination-treated xenografts compared to vehicle and RT-treated-xenografts (N &P Vs. M & O, respectively)

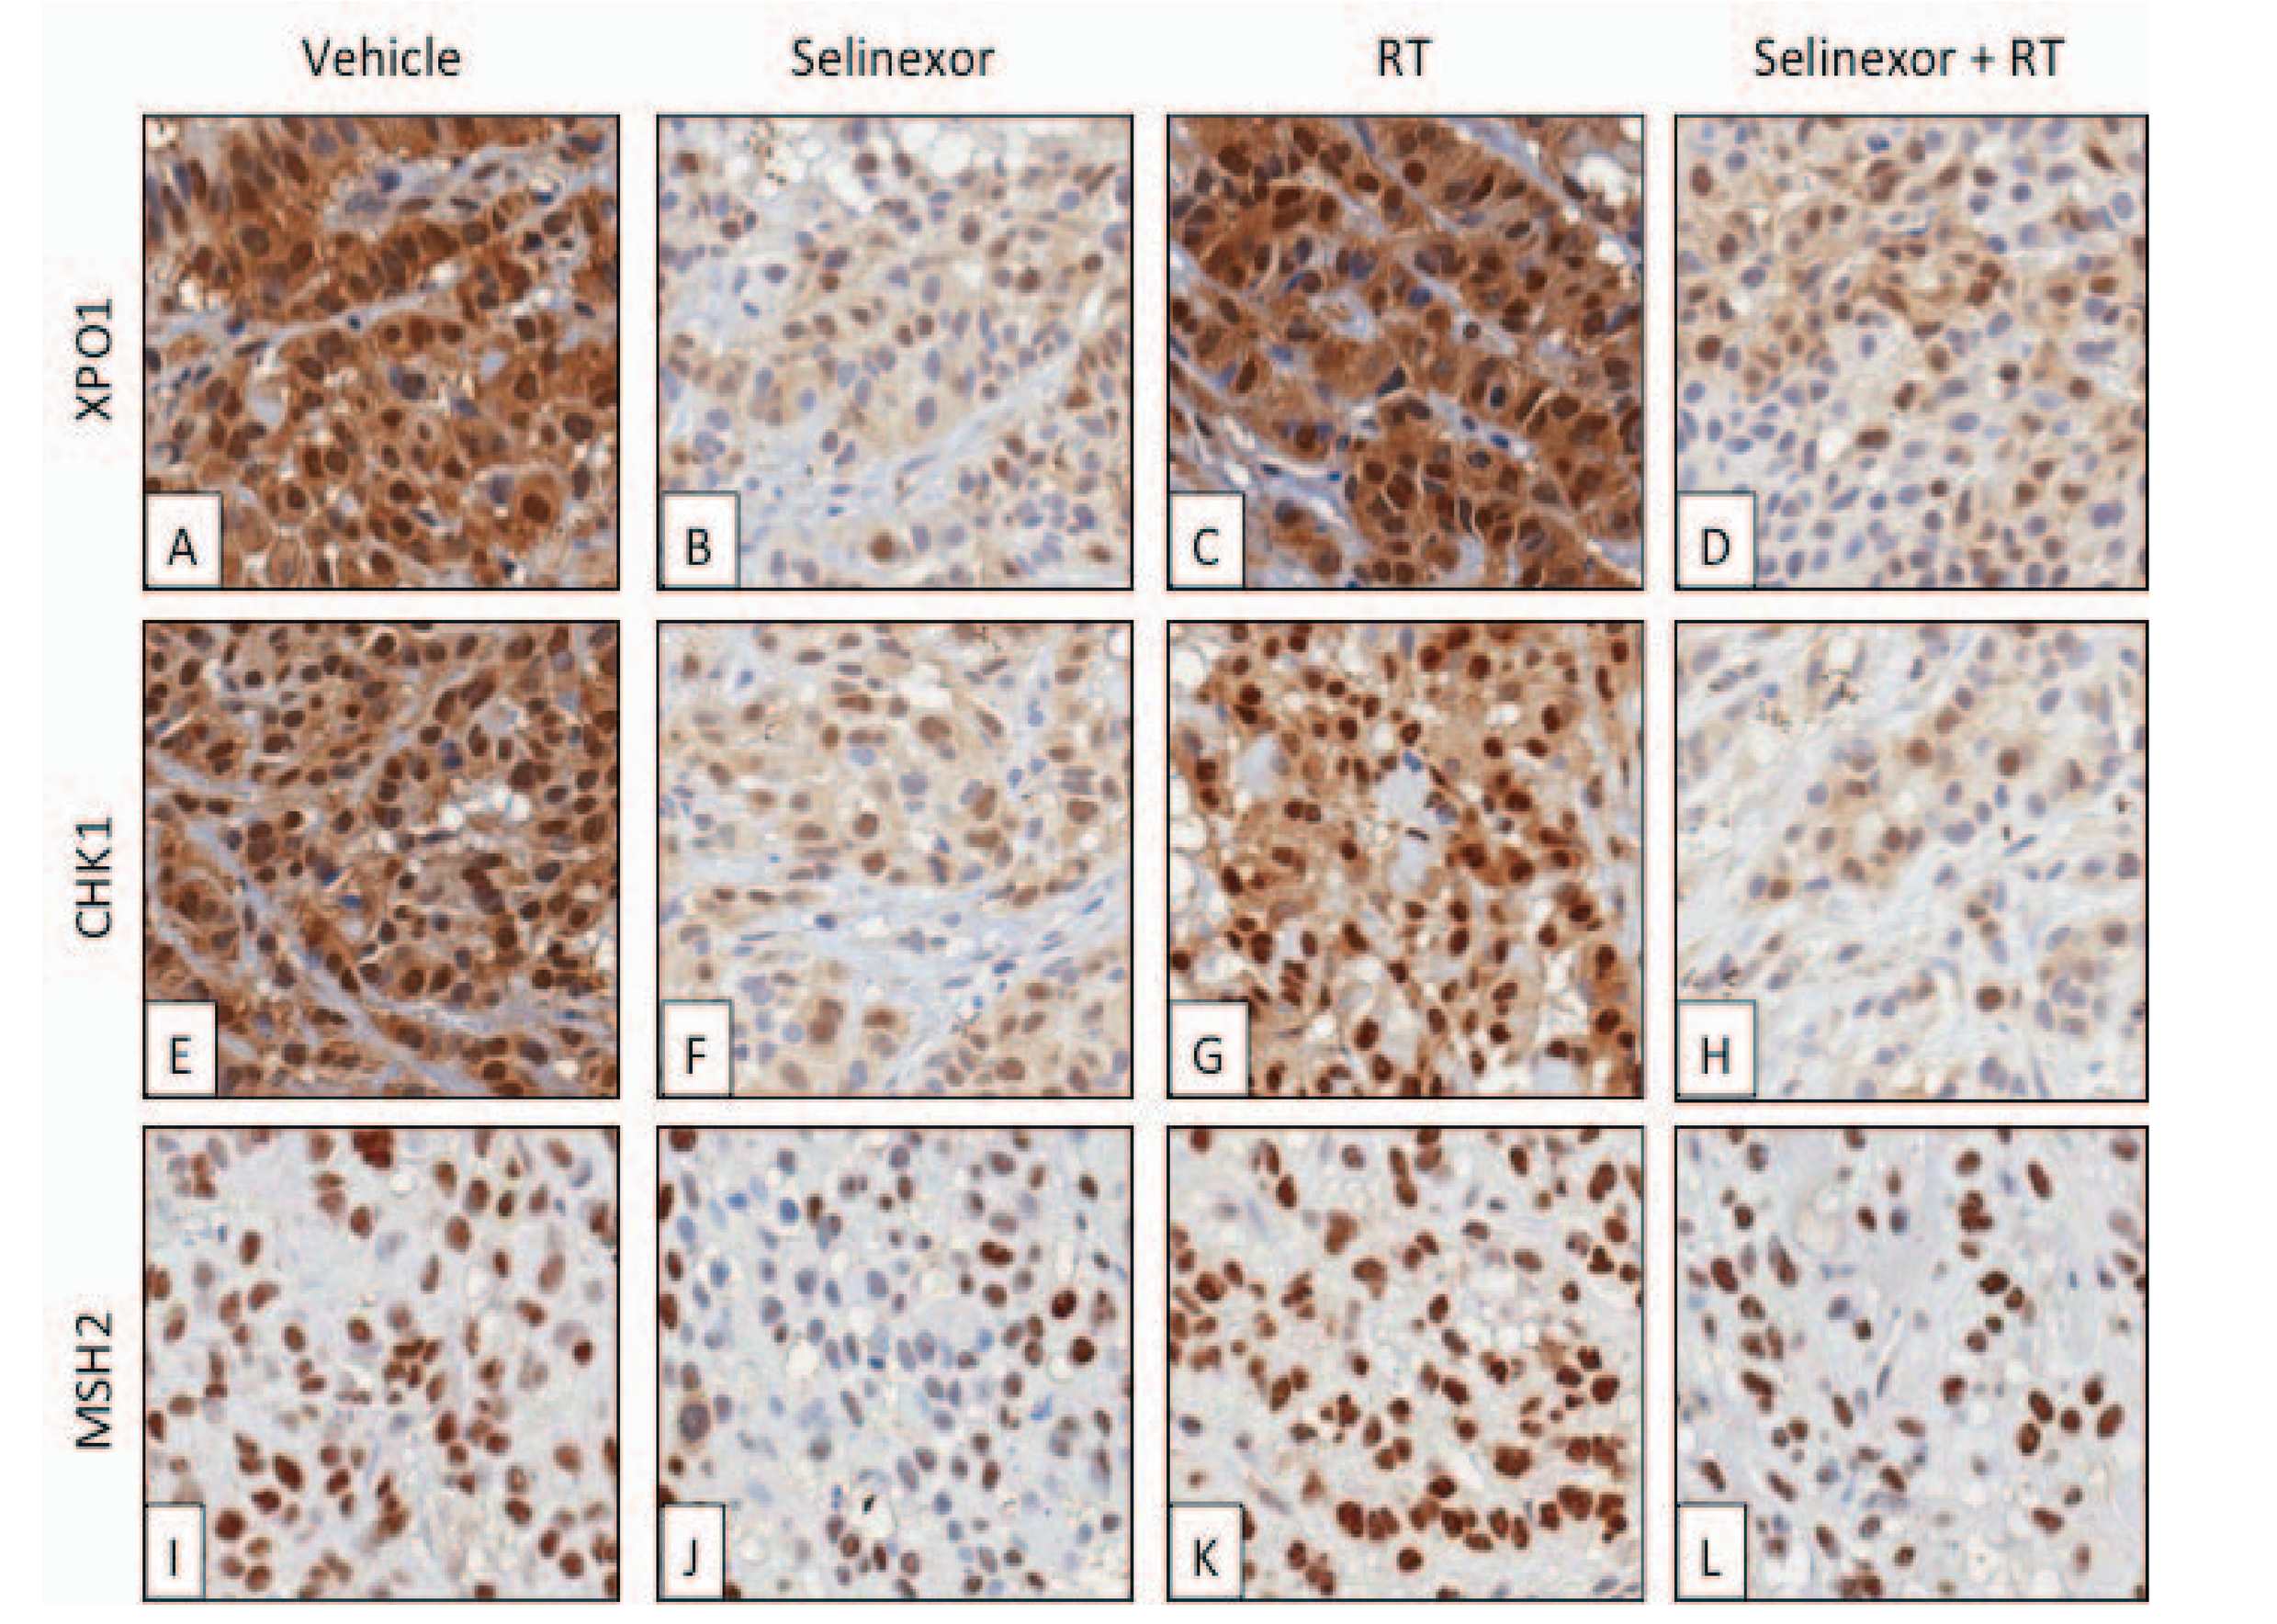
Vehicle
Selinexor
RT
Selinexor + RT
XPO1
CHK1
MSH2

Selinexor +/- RT treatment lead to reduction in XPO1 and the DNA damage response proteins CHK1 and MSH2 expression levels

(A-L) Reduction in number of positively stained cells for XPO1, CHK1 & MSH2 as well as reduced staining intensity is observed in selinexor-treated- and selinexor-RT combination-treated xenografts compared to vehicle and RT-treated-xenografts (B & D vs. A & C; F & H Vs. E & G; J &L Vs. I & K, respectively)

Selinexor Negatively Affects DNA Damage Response Proteins

Immunoblots on cell lysates from H1299 and A549 cultures treated with selinexor (0, 0.01, 0.03, 0.1, 0.3 or 1.0 uM) **(A),** or combination of selinexor and RT (4 Gy) **(B).**

Selinexor inhibits several components of the DNA damage response including CHK1, RAD51, MSH2 and MSH6. Inhibition of components of the DNA damage response is thought to limit DNA damage repair leading to apoptosis.

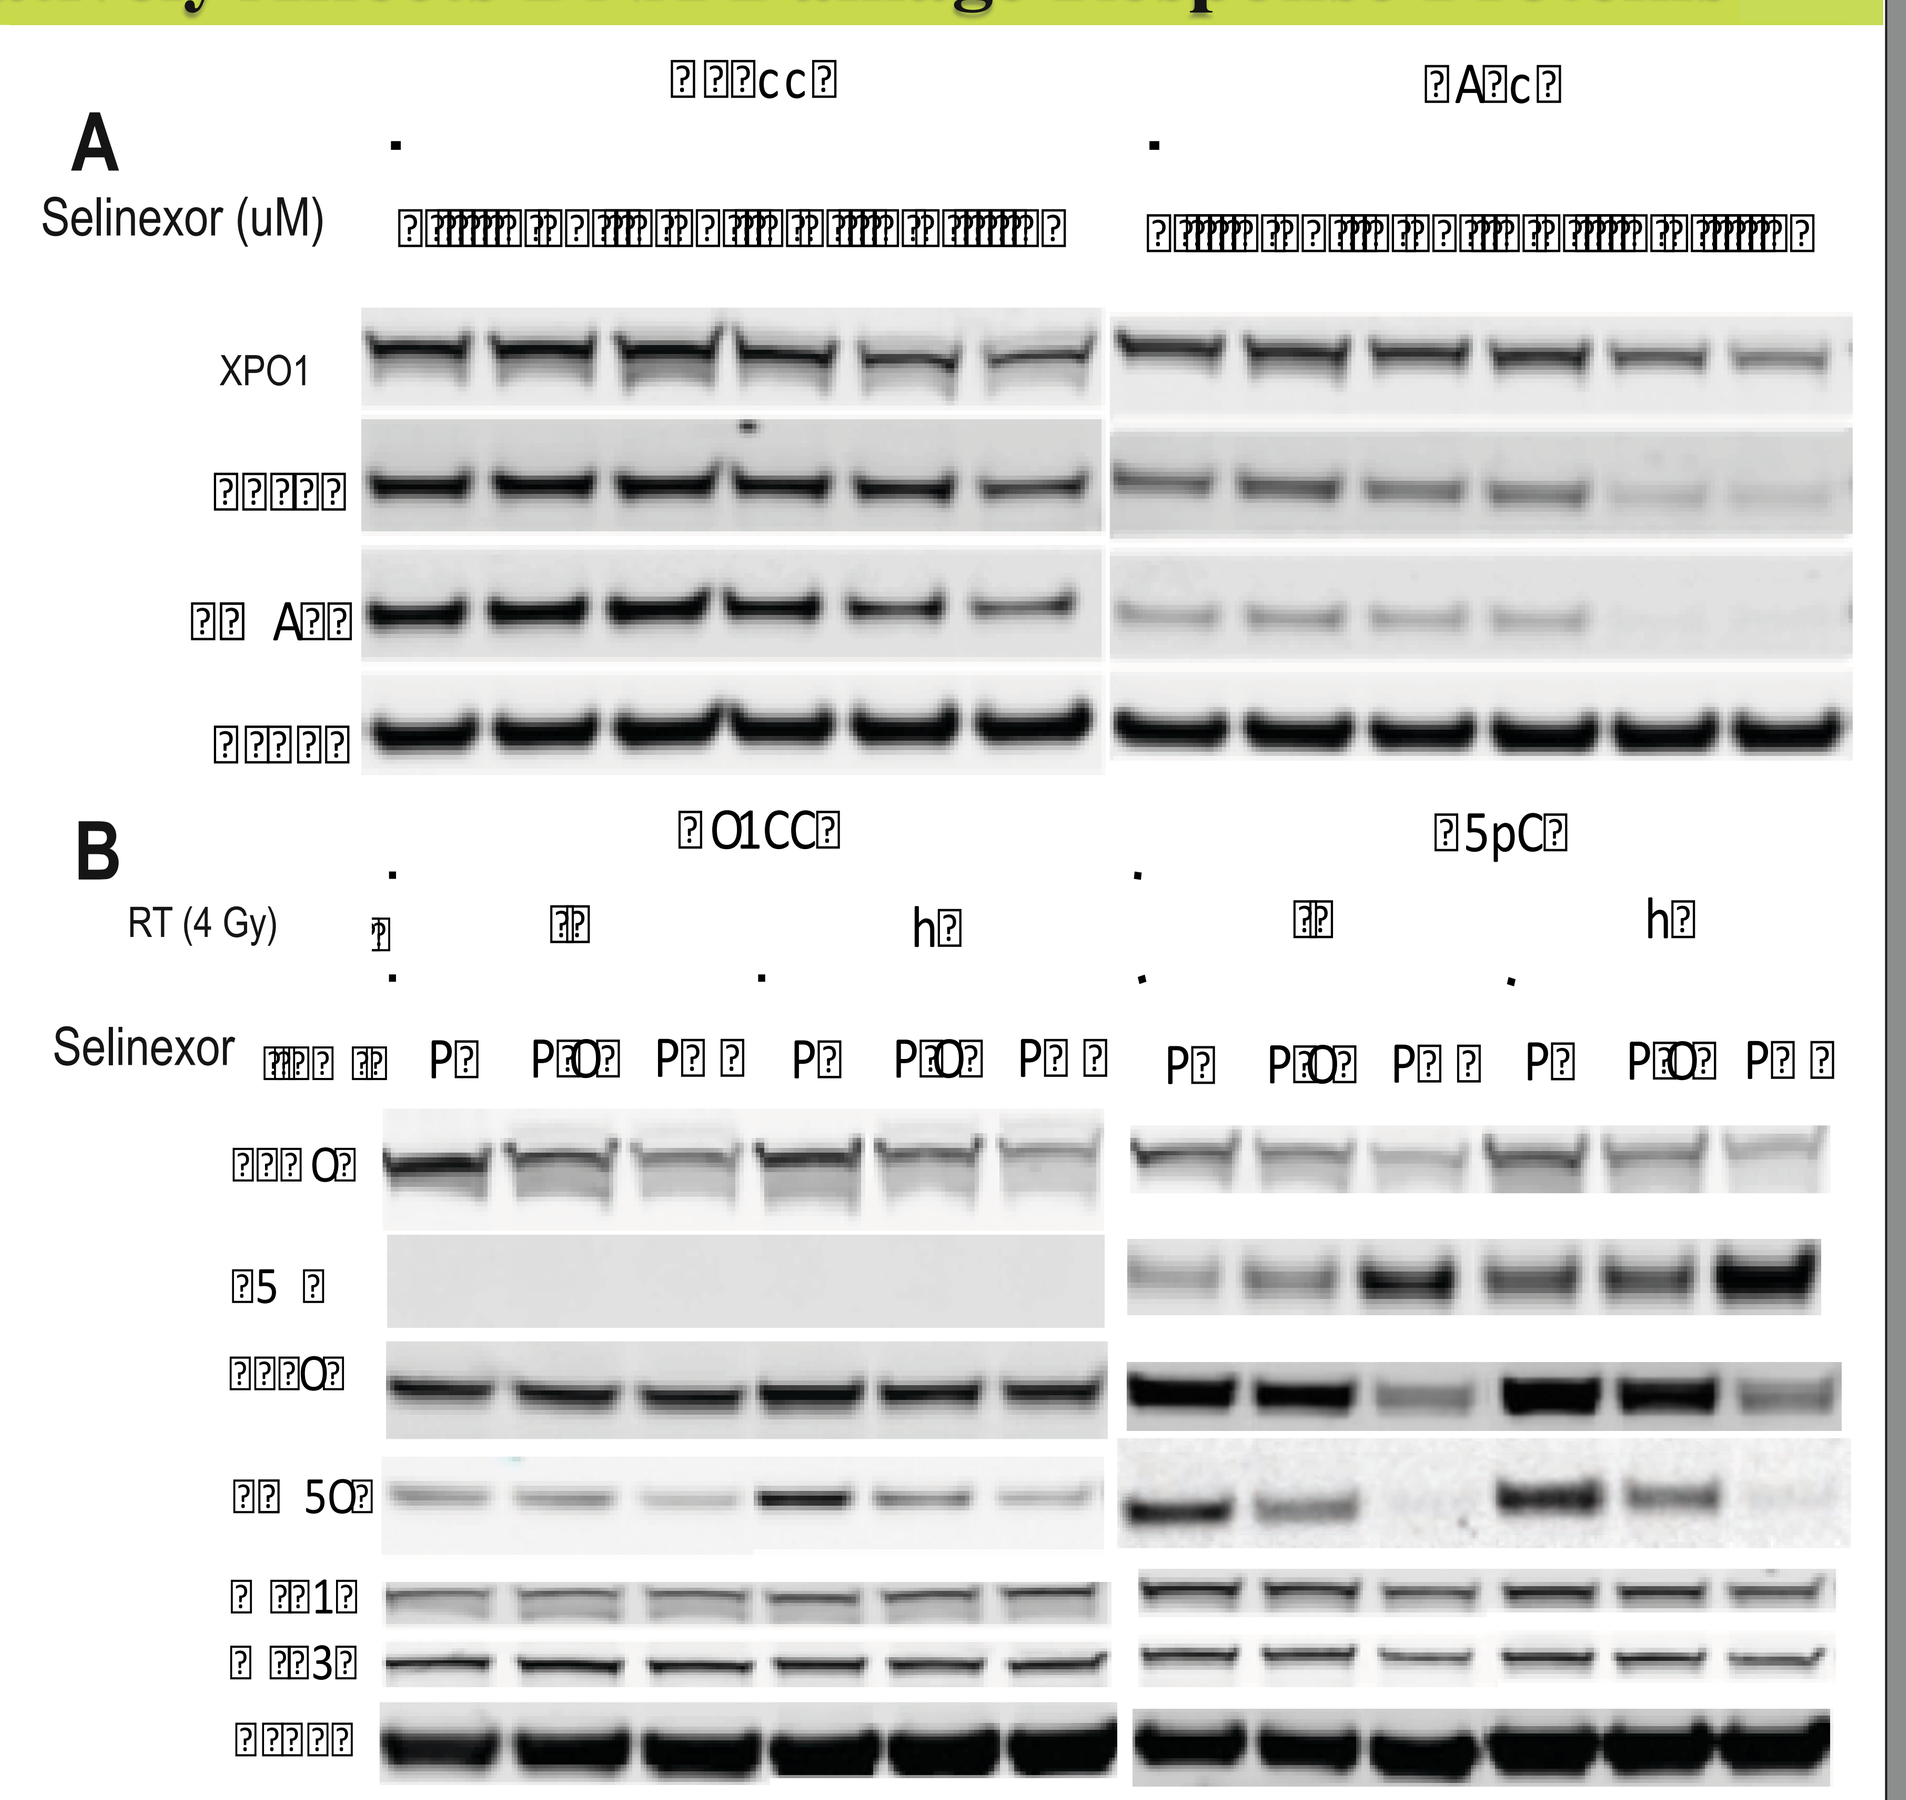
A
Selinexor (uM)
XPO1
B
RT (4 Gy)
Selinexor

Selinexor Reduces Cell Proliferation and Induces Nuclear Localization of Tumor Suppressor Genes in Human Lung Adeno-squamous Cell Carcinoma

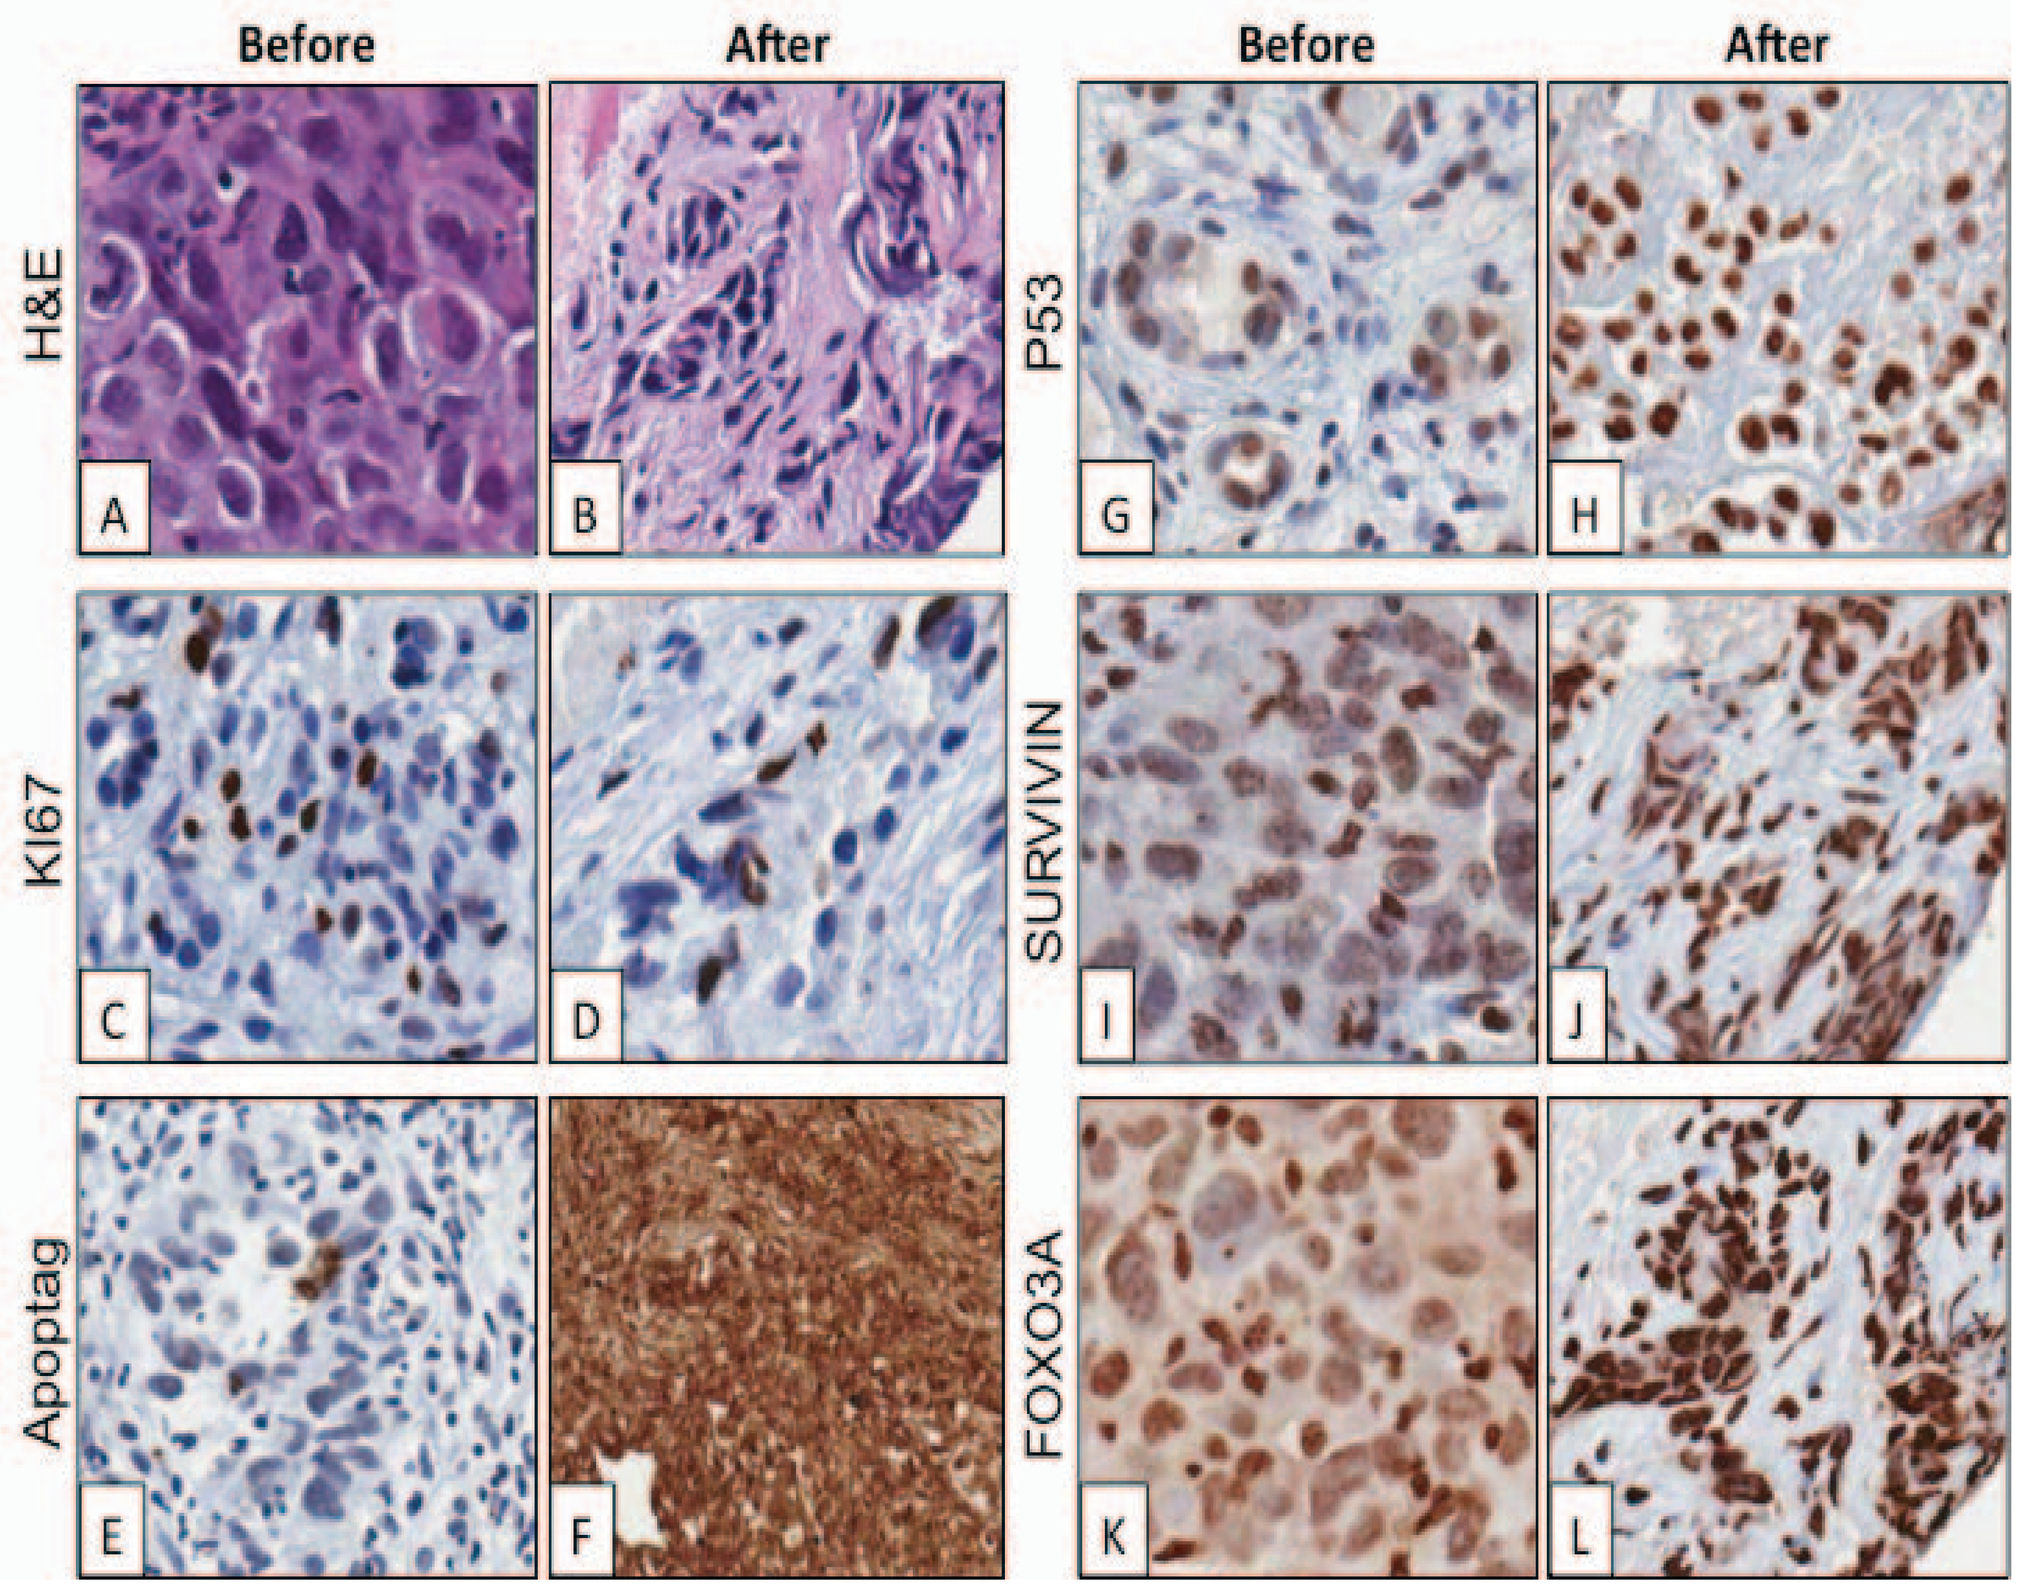
Before
After
H&E
KI67
Apoptag
P53
SURVIVIN
FOXO3A

Biopsies from a kidney lesion of lung adeno-SCC patient 043-813 were obtained prior ("before") and 3 weeks post ("after") selinexor treatment initiation.

(A-B) H&E IHC staining. Pre-treated tumor (A) is compromised of multiple foci of dense anaplastic/pleomorphic cells while post-treated tumor (B) has markedly fewer tumor cells surrounded with fibrosis

(C-D) KI67 staining shows reduction in number of positively stained nuclei in post-treated biopsy (D) vs. (C). (E-F) Apoptag staining shows increase in staining post-treatment (F) vs. (E). (G-L) Nuclear staining of XPO1 direct cargos and tumor suppressor genes: P53, SURVIVIN and FOXO3A (H,J & L) vs. (G, I & K) respectively, shows overall reduction in cytoplasmic staining and increase in nuclear staining intensity post treatment, indicating potent inhibition of XPO1 and suggestive of apoptosis signaling induction.

CONCLUSIONS

Our work suggests the SINE™ compound, selinexor, is a promising NSCLC therapeutic as a stand-alone agent and in combination with RT. We demonstrated that selinexor dose-dependently lowers the expression of several DDR proteins in NSCLC cell lines, including RAD51 (HR protein), CHK1, MSH2 and MSH6 (both belong to mismatch repair proteins). This reduction presumably limits the capacity of the cell to repair DNA insults induced by certain chemotherapeutics and RT, eventually leading to apoptosis. Selinexor is currently in Phase II clinical trials for the treatment of hematological malignancies and solid tumors. These clinical trials are using selinexor as a single therapy and combination studies with other anticancer drugs have yet to begin. Combination study with selinexor and radiation has recently started and enrolled the first patients.

Our work described here and the tolerability of selinexor observed in Phase I/II clinical trials, suggests that selinexor may be a promising therapeutic for the treatment of NSCLC.

Website: www.karyopharm.com contact: Sivan Elloul, email: sivan@karyopharm.com