Abstract #2442
AACR 2015

XPO1 is selinexor's prime target: validation by mutating cysteine 528 on both *XPO1* alleles using CRISPR/Cas9 genome editing

Neggers JE[1], Vercruysse T[1], Jacquemyn M[1], Vanstreels E[1], Baloglu E[2], Shacham S[2], Crochiere M[2], Senapedis W[2], Landesman Y[2], Daelemans D[1]

[1]Rega Institute for Medical Research, KU Leuven, 3000 Leuven, Belgium; [2]Karyopharm Therapeutics, Newton, MA 02459, USA

Background

Human exportin-1 (XPO1), or chromosome region maintenance 1 protein (CRM1), is a key nuclear-cytoplasmic transport protein. It exports a broad range of different cargo proteins out of the cell's nucleus to the cytoplasm. These cargo proteins include tumour suppressor and growth regulatory related proteins; therefore correct XPO1 function is key to normal cell homeostasis. In recent years, overexpression or dysfunction of XPO1 has commonly been observed in different types of cancer and alterations in XPO1 expression levels may cause subcellular mislocalization of tumour suppressor proteins and cell cycle regulators, resulting in uncontrolled cell growth and carcinogenesis. Recently, *N*-azolylacrylate small-molecule inhibitors of the XPO1-mediated nuclear export were further rationally optimized. These new inhibitors of nuclear export, called SINE, demonstrate potent activity against multiple types of cancer and induce apoptosis in different cancer models. Importantly, selinexor (KPT-330), the clinical candidate of these inhibitors, is currently enrolling for phase 2 clinical studies. Selinexor binds to the cysteine 528 residue of XPO1, causes accumulation of tumour suppressive proteins in the nucleus which is correlated with the induction of cell cycle arrest and apoptosis. However, the specificity of this drug target interaction in cancer cells is not validated.

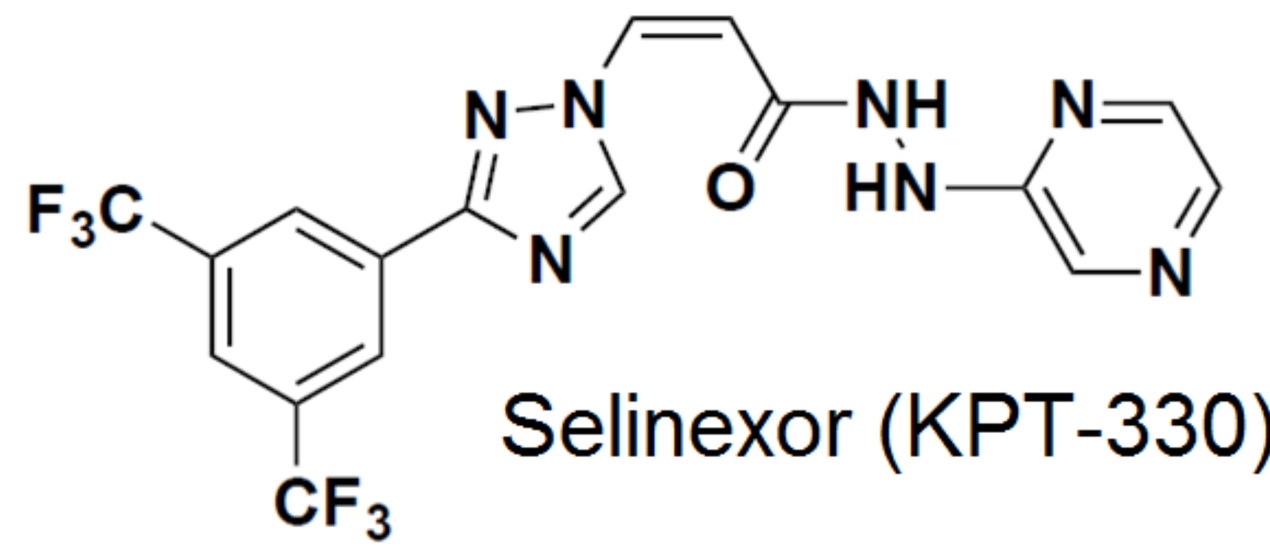
F3C
CF3
N-N
N
NH
HN
N
N
O

Selinexor (KPT-330)

Validation of drug-target interaction requires the introduction of resistant mutations into the wild-type background. However, until the advent of the CRISPR/Cas9 genome editing technique, this approach was not straightforward in mammalian cells. So far drug resistance against selinexor has not been raised, but we reasoned that genomic mutation of the cysteine 528 residue in XPO1 may cause resistance. In this study we applied CRISPR/Cas9 genome editing in combination with homology directed repair to introduce a single XPO1 C528S mutation in acute T cell leukaemia Jurkat cells in order to validate the drug-target interaction of selinexor with XPO1 in the living cell.

Reference

Neggers et al. *Chem Biol.* 2015 Jan 22;22(1):107-16.

Key Findings

Generation of a homozygous XPO1 C528S mutant Jurkat cell line

T-ALL Jurkat Clone E6-1 cells were transfected with DNA required for the CRISPR/Cas9 genome editing and homologous recombination. The CRISPR/Cas9 genome editing system was designed to target exon 14 of the *XPO1* genomic locus to facilitate repair by homologous recombination with a template containing the C528S missense mutation and 3 silent mutations (**figure 1**).

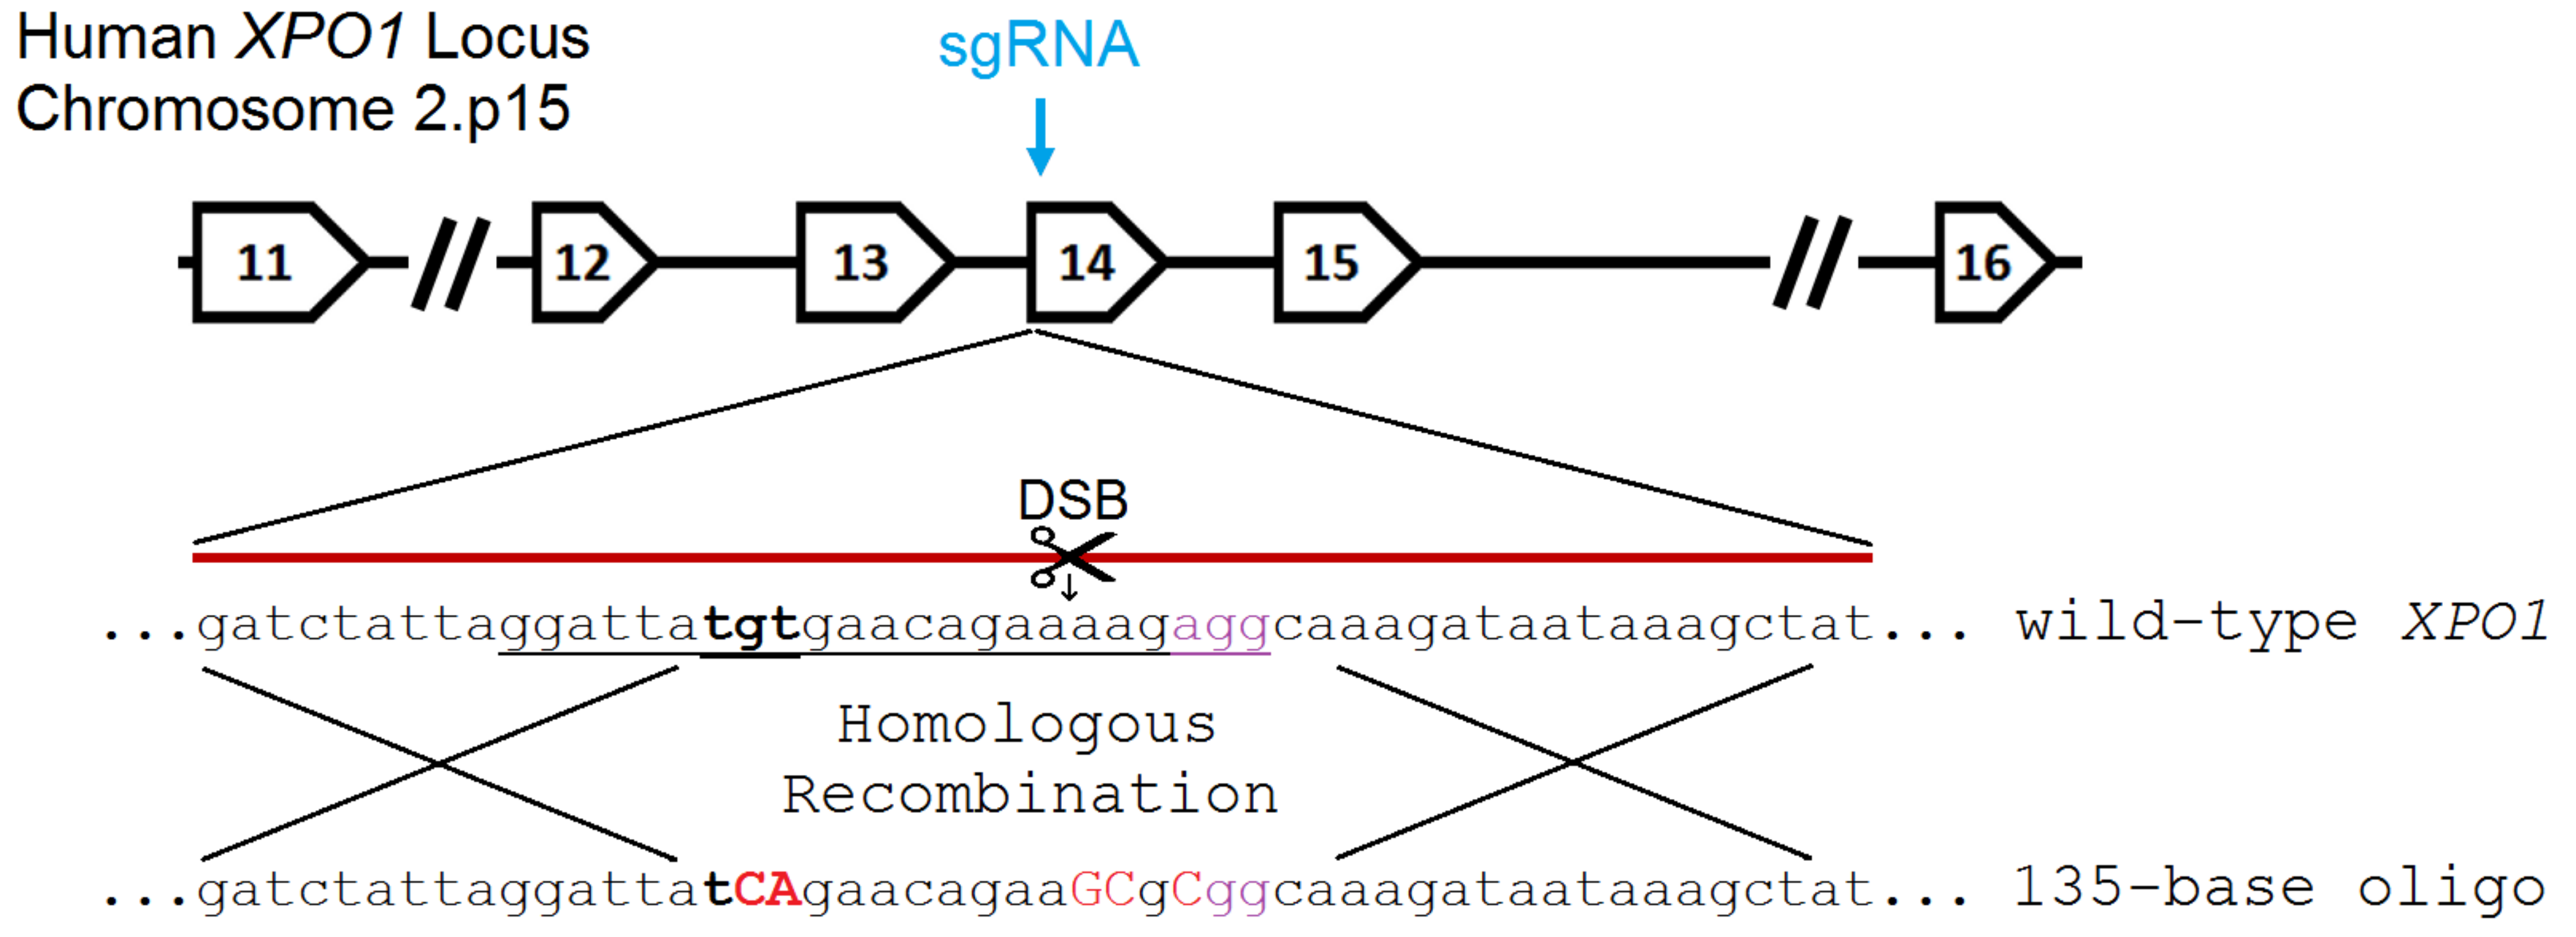
Human *XPO1* Locus
Chromosome 2.p15
sgRNA
11
12
13
14
15
16
DSB
...gatctattaggattatgtgaacagaaaagaggcaaagataataaagctat... wild-type *XPO1*
Homologous
Recombination
...gatctattaggattatCAgaacagaaGCgCggcaaagataataaagctat... 135-base oligo

Figure 1: schematic overview of CRISPR/Cas9 induced homologous recombination of XPO1

Clones were sequenced to screen for C528S mutations in both the DNA and mRNA and homozygous C528S mutants were obtained (**figure 2 A&B**). The XPO1 expression in this homozygous clone was compared to the wild-type by western-blotting of the XPO1 protein (**figure 2C**) and qPCR of the *XPO1* mRNA (**figure 2D**).

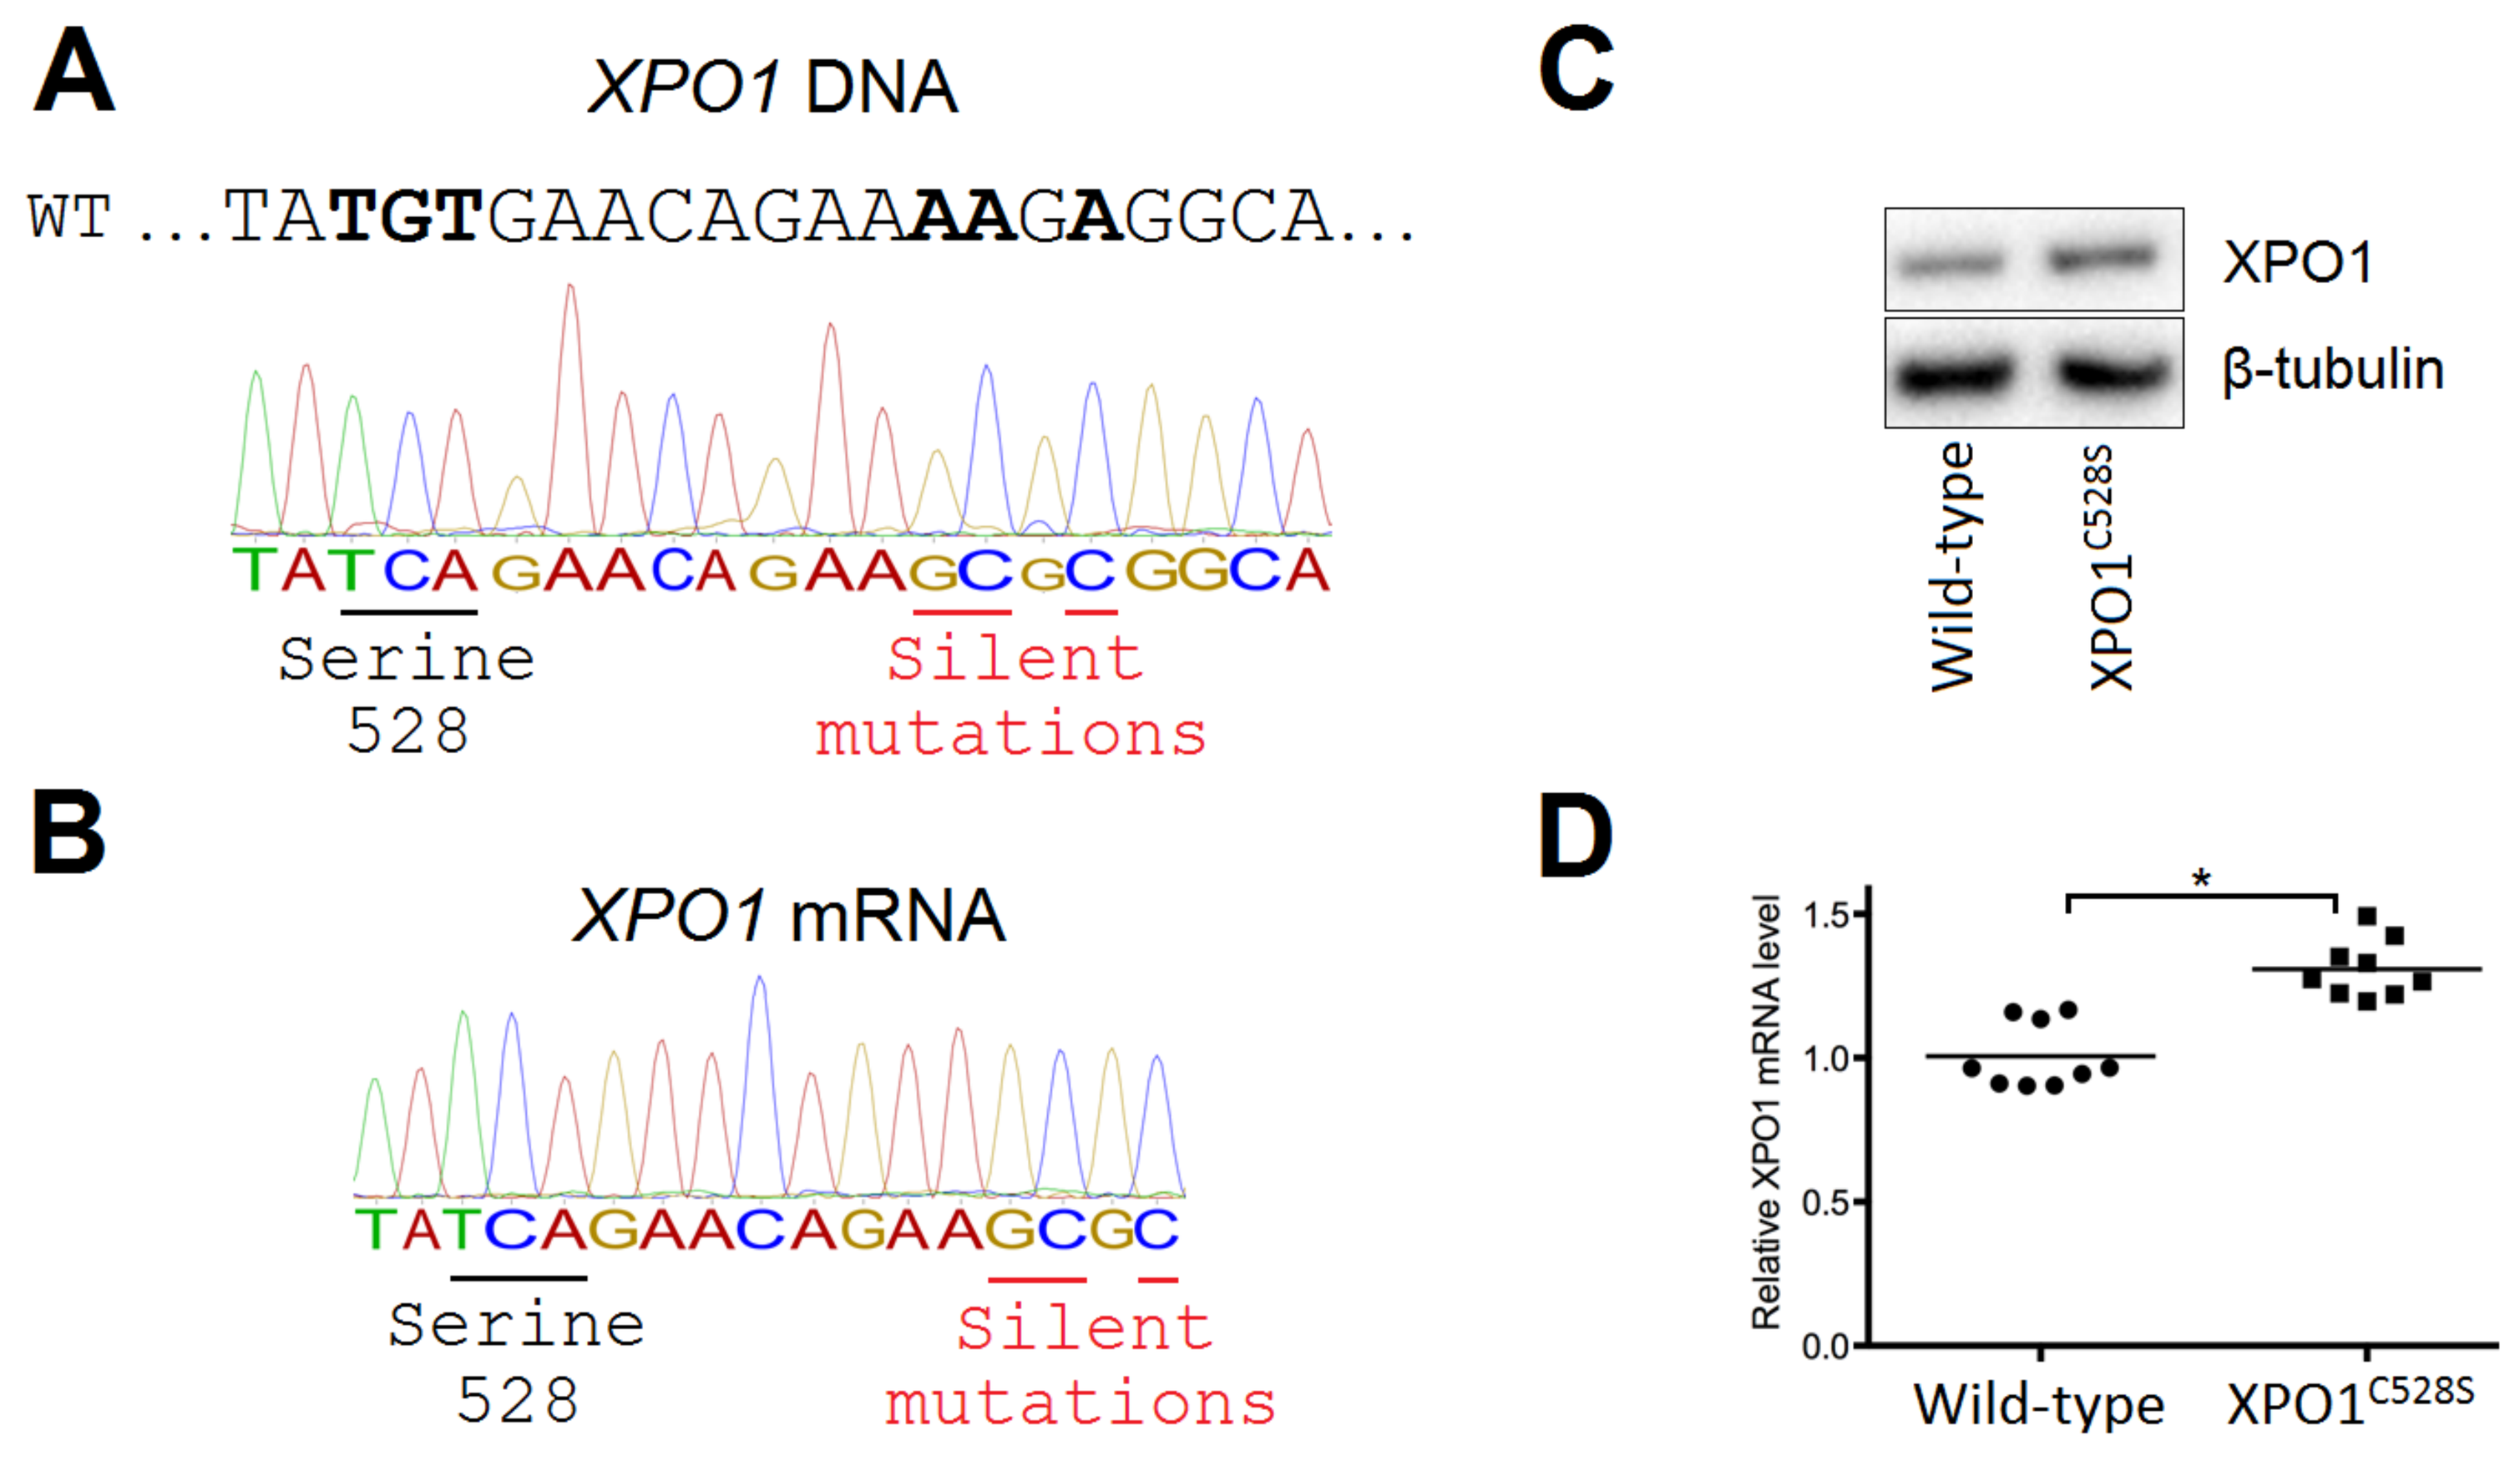
A
XPO1 DNA
WT ...TATGTGAACAGAAAAGAGGCA...
TATCAGAACAGAAGCGCGGCA
Serine
528
Silent
mutations
B
XPO1 mRNA
TATCAGAACAGAAGCGC
Serine
528
Silent
mutations
C
XPO1
β-tubulin
Wild-type
XPO1C528S
D
Relative XPO1 mRNA level
1.5
1.0
0.5
0.0
*
Wild-type
XPO1C528S

Figure 2: identification of a homozygous C528S XPO1 mutant and it's XPO1 expression

C528S mutation of XPO1 confers resistance to selinexor treatment

To determine if selinexor interacts selectively with cysteine 528 of XPO1, homozygous mutants and wild-type cells were treated with selinexor for 3 days. Cytotoxicity was observed in wild-type cells with a CC50 of 41.0 ± 6.4 nM, while the $XPO1^{C528S}$ cells were highly resistant showing a CC50 of 10.3 ± 2.3 µM (**figure 3A**). To further examine drug-target selectivity, apoptosis induction by selinexor was measured by AnnexinV/PI flow cytometry (**figure 3B**) and western-blotting of cleaved caspase-3 and PARP (**figure 3C**).

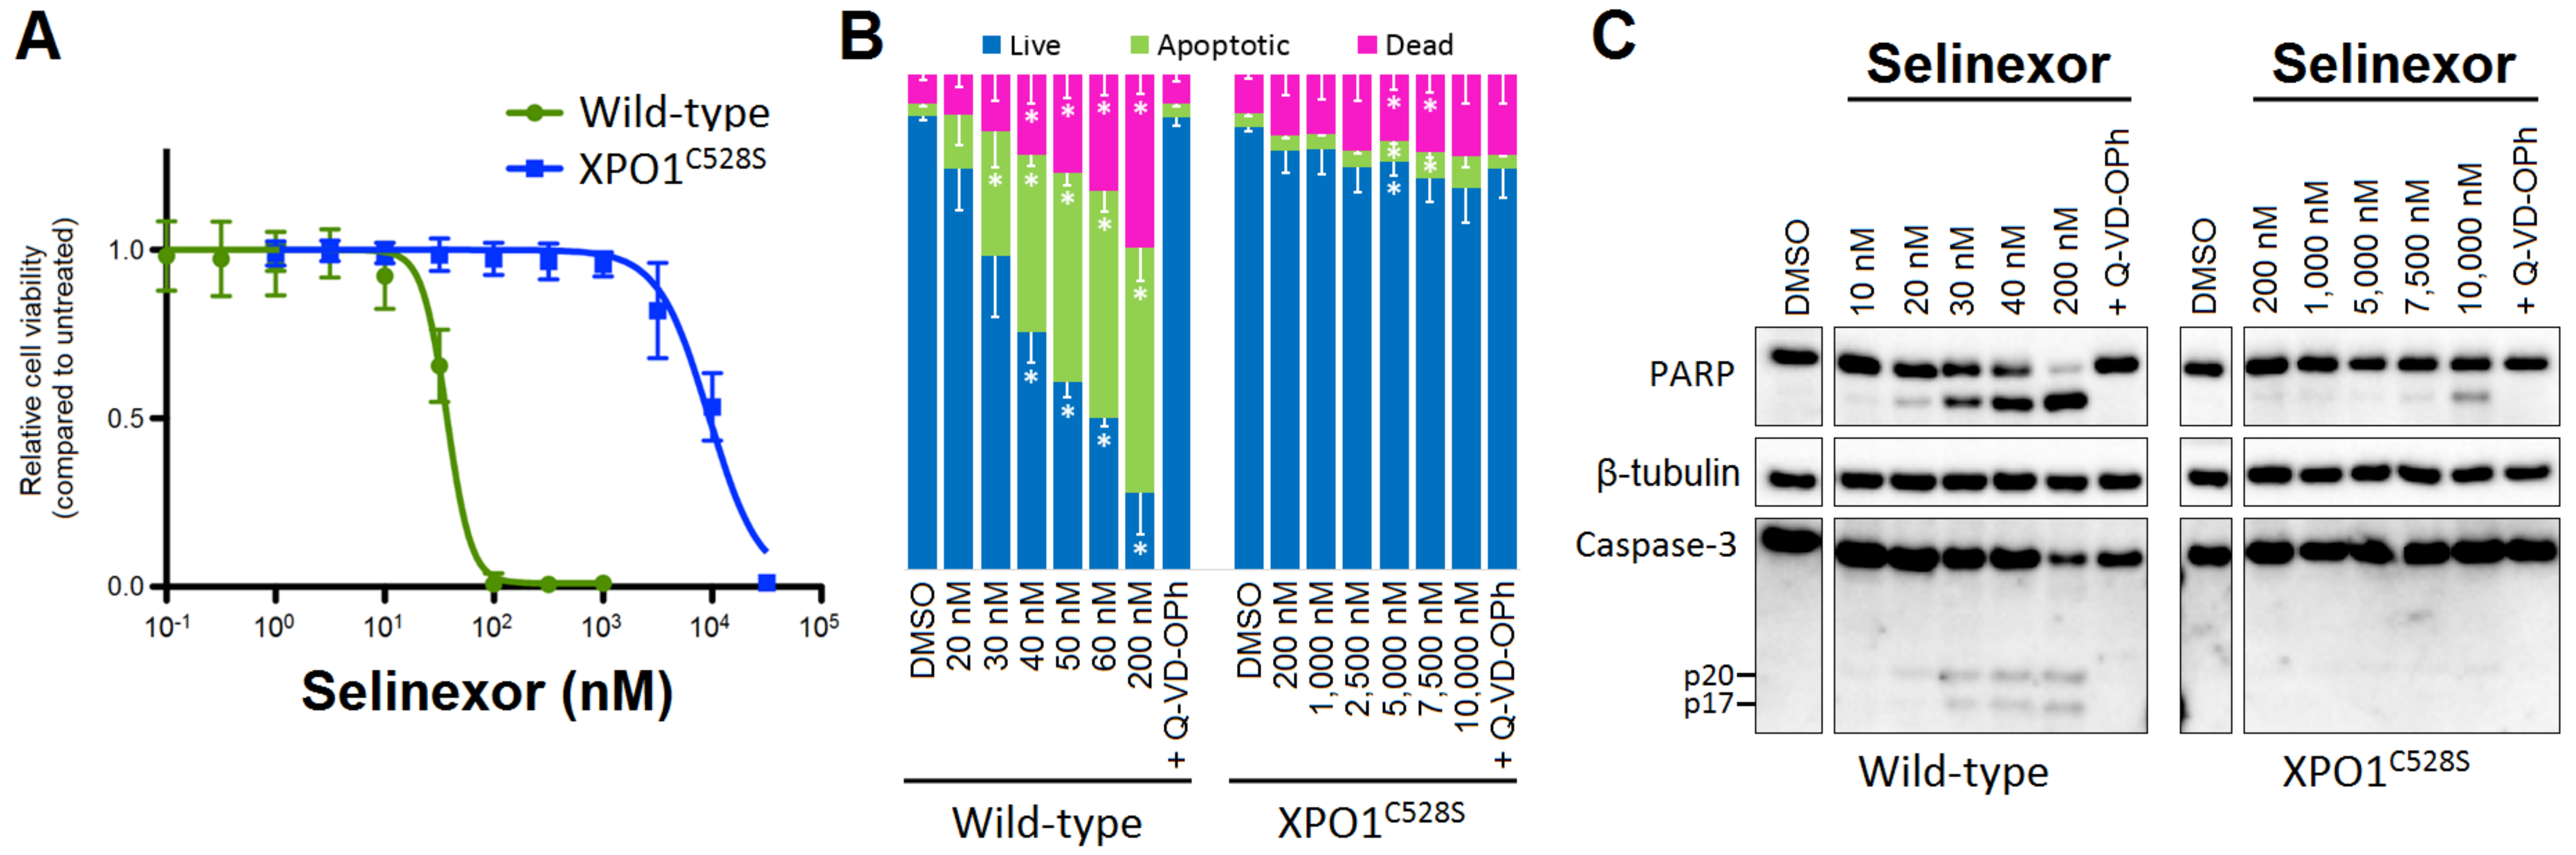
A
Wild-type
XPO1C528S
Relative cell viability (compared to untreated)
1.0
0.5
0.0
10-1
100
101
102
103
104
105
Selinexor (nM)
B
Live
Apoptotic
Dead
DMSO
20 nM
30 nM
40 nM
50 nM
60 nM
200 nM
+ Q-VD-OPh
DMSO
200 nM
1,000 nM
2,500 nM
5,000 nM
7,500 nM
10,000 nM
+ Q-VD-OPh
Wild-type
XPO1C528S
C
Selinexor
Selinexor
DMSO
10 nM
20 nM
30 nM
40 nM
200 nM
+ Q-VD-OPh
DMSO
200 nM
1,000 nM
5,000 nM
7,500 nM
10,000 nM
+ Q-VD-OPh
PARP
β-tubulin
Caspase-3
p20
p17
Wild-type
XPO1C528S

Figure 3: the C528S mutation confers resistance to apoptosis induction by selinexor

Additional confirmation that cells expressing mutant XPO1 are resistant to selinexor was obtained by visualization of the RanBP1 cargo-protein with immunofluorescence (**figure 4A**). In steady-state RanBP1 is localized to the cytoplasm as this cargo-protein is actively exported from the nucleus by XPO1. However, in the presence of selinexor, a clear accumulation of RanBP1 is observed within the nucleus of wild-type cells, while no nuclear redistribution is observed at all in the mutant cells.

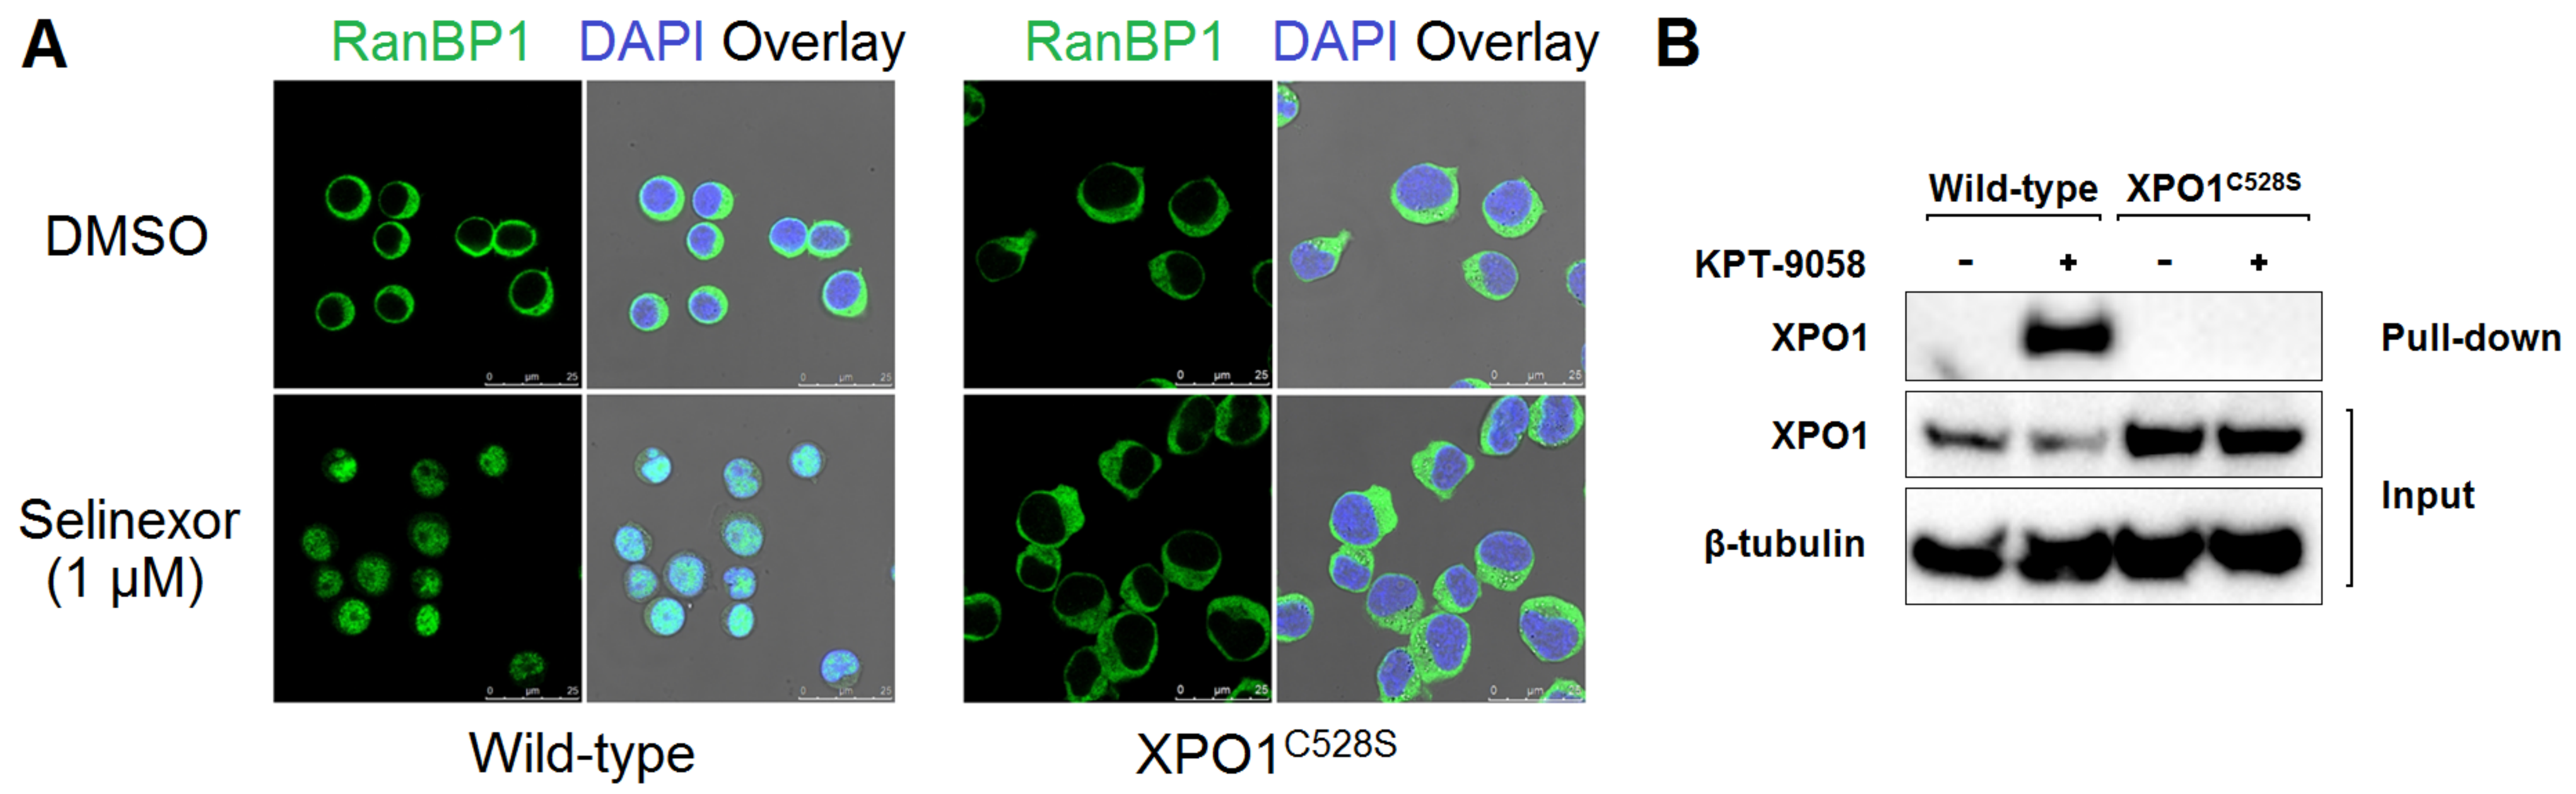
A
RanBP1
DAPI
Overlay
RanBP1
DAPI
Overlay
DMSO
Selinexor
(1 µM)
Wild-type
XPO1C528S
B
Wild-type
XPO1C528S
KPT-9058
-
+
-
+
XPO1
XPO1
β-tubulin
Pull-down
Input

Figure 4: selinexor cannot inhibit $XPO1^{C528S}$ mediated nuclear export and cannot bind to $XPO1^{C528S}$

Finally, direct binding of the drug to XPO1 was examined with a biotinylated analogue of selinexor, KPT-9058. Extraction of the biotin-labelled compound was performed with avidin coated beads and XPO1 was detected in the eluate by western-blotting (**figure 4B**). In wild-type cells a clear XPO1 extraction was observed, but no XPO1 was detected in the eluate of the treated $XPO1^{C528S}$ mutants.