KPT-9274 Inhibits Cellular NAD and Synergies with NAD Depleting Enzymes to Induce Cancer Cell Death

Christian Argueta, Trinayan Kashyap, Margaret Lee, Yosef Landesman, Sharon Shacham, William Senapedis, and Erkan Baloglu

Karyopharm Therapeutics Inc, Newton MA

Abstract #3016

Cross Reference Abstract #4827

Abstract

Nicotinamide adenine dinucleotide (NAD) is an essential metabolite and an important cofactor for several biological processes that undergo significant alterations during malignant transformation, including metabolism and genomic stability-processes. In cancer, NAD is rapidly turned over due to the high metabolic demands of rapidly proliferating cells and the increased activity of NAD consuming enzymes, such as sirtuin 1 (SIRT1) and poly ADP ribose polymerase 1 (PARP1). NAD can be generated de novo from tryptophan or regenerated by nicotinamide phosphoribosyl transferase (NAMPT) or nicotinate phosphoribosyl transferase 1 (NAPRT1) in NAD salvage pathways. However, cancer cells do not utilize the de novo or NAPRT1 pathways effectively. Instead, they rely on the NAMPT-dependent salvage pathway to generate NAD, making NAD depletion a promising anti-cancer therapy. We have previously described the PAK4 allosteric modulator, KPT-9274, a compound that also inhibits the enzymatic activity of NAMPT. KPT-9274 and other NAMPT inhibitors rapidly deplete cellular NAD levels, ultimately leading to ATP depletion and cell death. Similarly, several studies have shown that hyper activation of NAD consuming enzymes can lead to cell death. The purpose of this study is to determine whether co-administration of KPT-9274 with compounds that activate NAD depleting enzymes will enhance the cytotoxic effects of KPT-9274 in cancer cells.

Methods: Celltiter-Glo was used to measure ATP levels and viability of cells. NAD/NADH-Glo was used to measure total NAD levels in cells. Gene and protein expression was measured using quantitative PCR and western blot analysis, respectively. Protein knockdown was accomplished using RNAi.

Results: We have identified an orally bioavailable dual inhibitor of PAK4 and NAMPT, which demonstrated potent anti-cancer activity in a variety of cell lines both *in vitro* and *in vivo*. We have identified several mechanistic combinations that increase the anti-tumor activity of PAK4/NAMPT dual inhibitor, KPT-9274, through the activation of NAD consuming enzymes (e.g. SIRT1 and PARP1). Specifically, we found that SRT1720, (activator of SIRT1) synergizes with KPT-9274 to increase cancer cell death. In contrast, RNAi of SIRT1 diminishes the efficacy of KPT-9274. Activation of PARP1 by DNA damaging agents (e.g. gemcitabine) significantly enhances the effectiveness of KPT-9274 mediated cell death. Finally, we show that PARP activating DNA damage agents enhance the toxicity and anti-tumor properties of KPT-9274 in xenograft models.

Conclusions: Here we report that KPT-9274 synergizes with NAD depleting enzymes to induce cancer cell death *in vitro* and *in vivo*. This noteworthy enhancement to the anticancer activity of KPT-9274, together with its previously described PAK4 anti-tumor activity, support the continued development of this orally bioavailable small molecule in combinations with current therapies.

KPT-9274: Dual Inhibitor of PAK4 and NAMPT

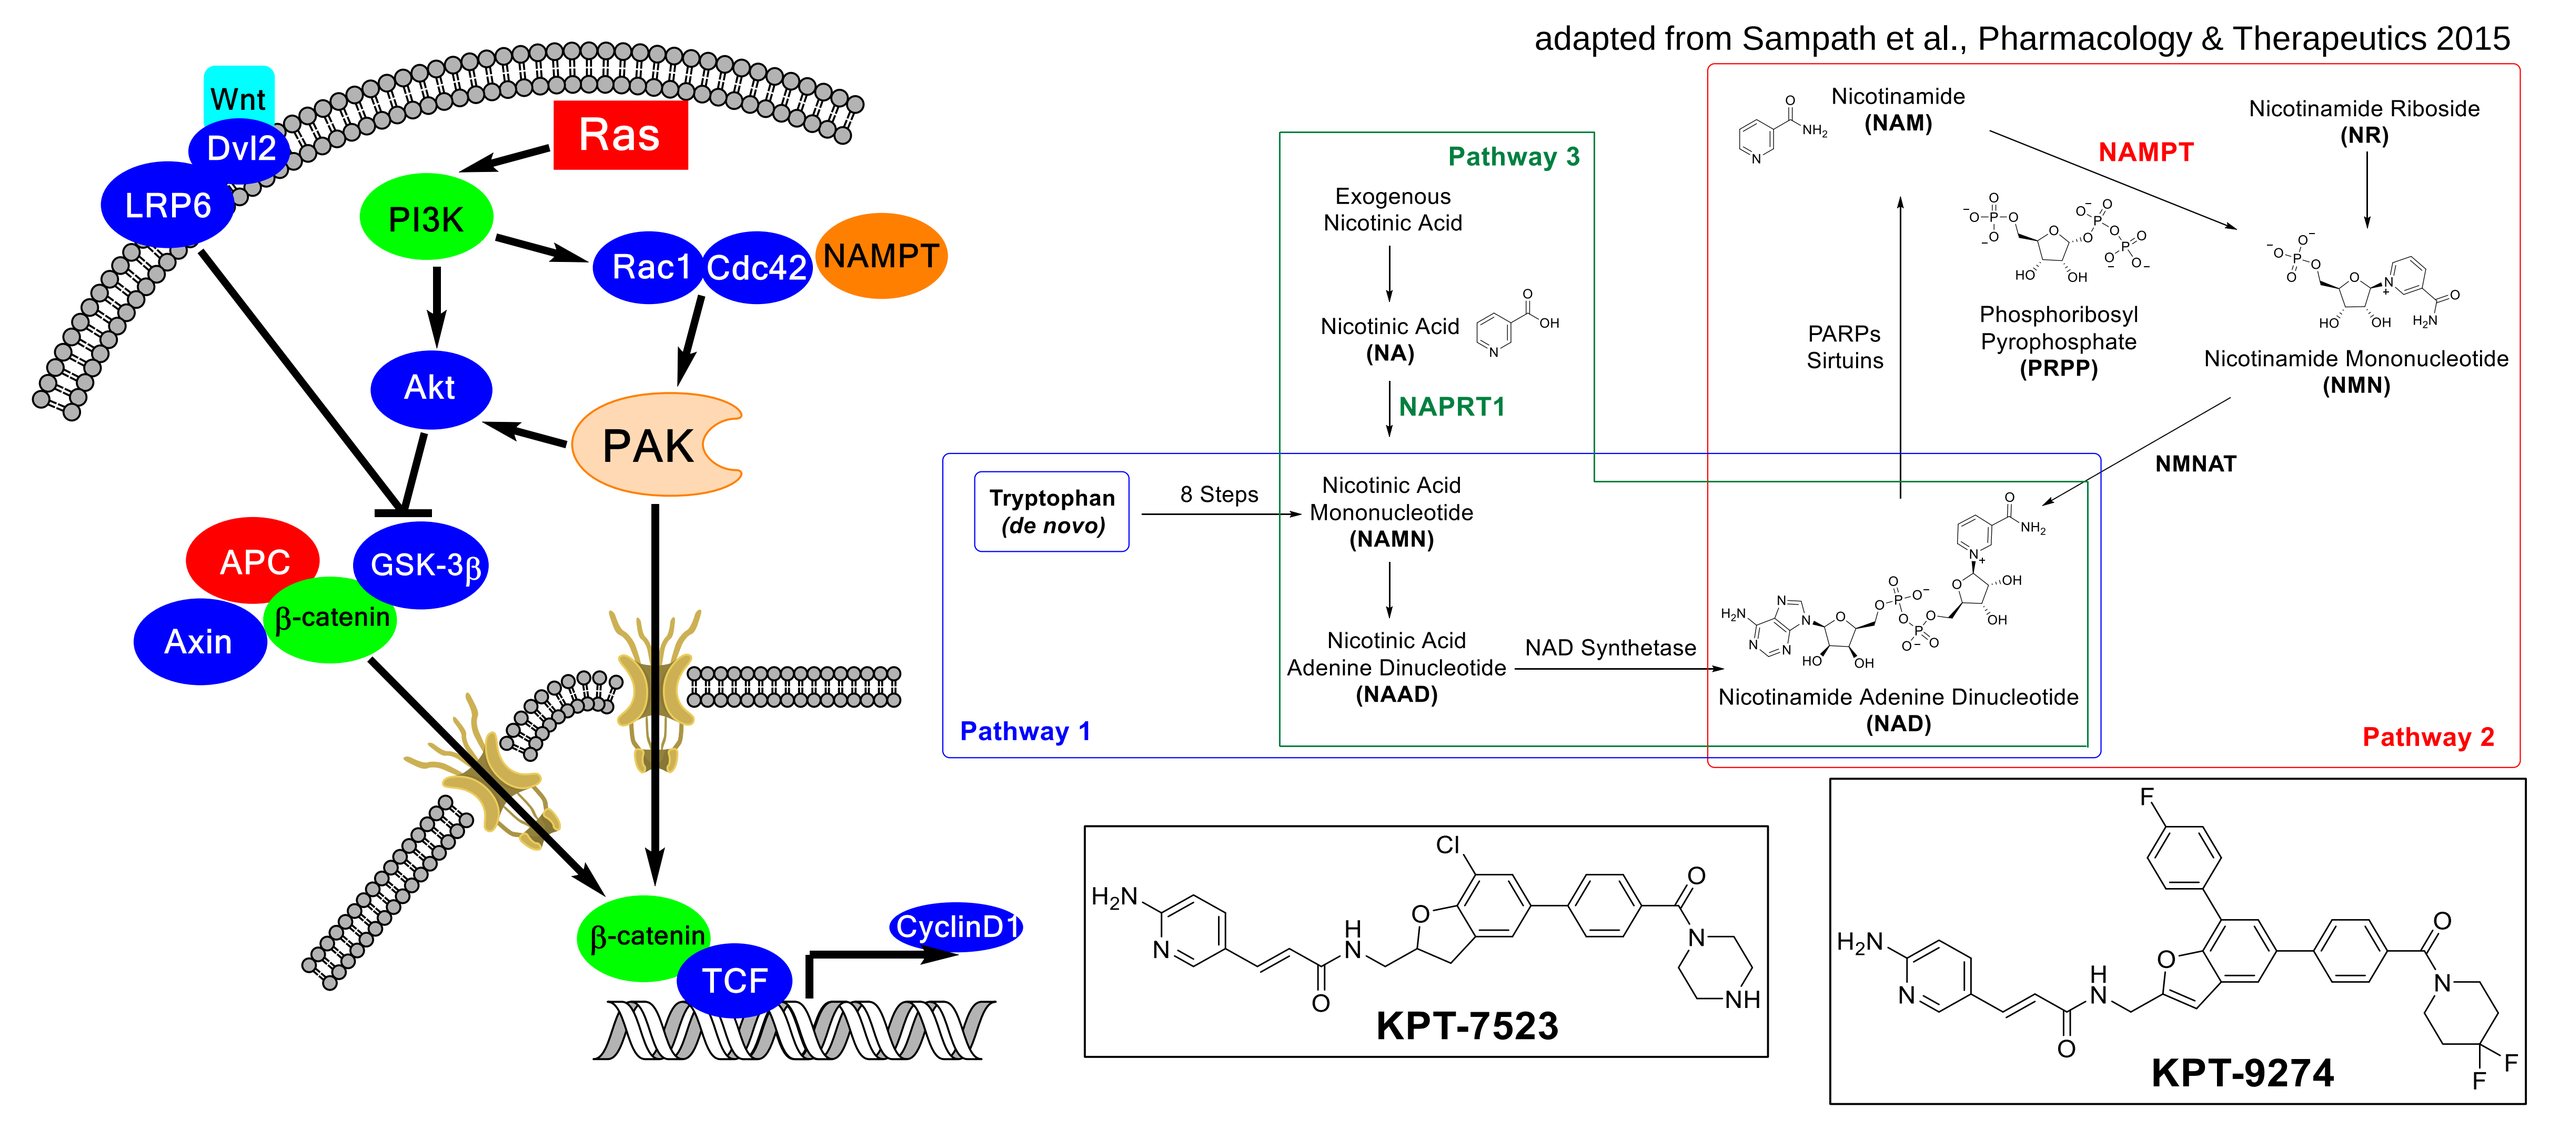

adapted from Sampath et al., Pharmacology & Therapeutics 2015
Wnt
Dvl2
LRP6
Ras
PI3K
Rac1
Cdc42
NAMPT
Akt
PAK
APC
GSK-3β
Axin
β-catenin
TCF
CyclinD1
Pathway 1
Pathway 2
Pathway 3
Tryptophan (de novo)
8 Steps
Nicotinamide (NAM)
Nicotinamide Riboside (NR)
Exogenous Nicotinic Acid
Nicotinic Acid (NA)
NAPRT1
Nicotinic Acid Mononucleotide (NAMN)
Nicotinic Acid Adenine Dinucleotide (NAAD)
NAD Synthetase
Nicotinamide Adenine Dinucleotide (NAD)
PARPs Sirtuins
Phosphoribosyl Pyrophosphate (PRPP)
Nicotinamide Mononucleotide (NMN)
NMNAT
KPT-7523
KPT-9274

Figure 1: KPT-9274 is a Dual Inhibitor of PAK4 and NAMPT. We have previously highlighted the anti-cancer properties of KPT-9274. In addition to specifically binding and targeting PAK4, KPT-9274 and analogues inhibit the catalytic activity of NAMPT.
(Cross reference with abstract #4827, April 20, 8 to 12 PM, Poster Session 20)

KPT-9274 Inhibits NAD Synthesis and Promotes Cancer Cell Death

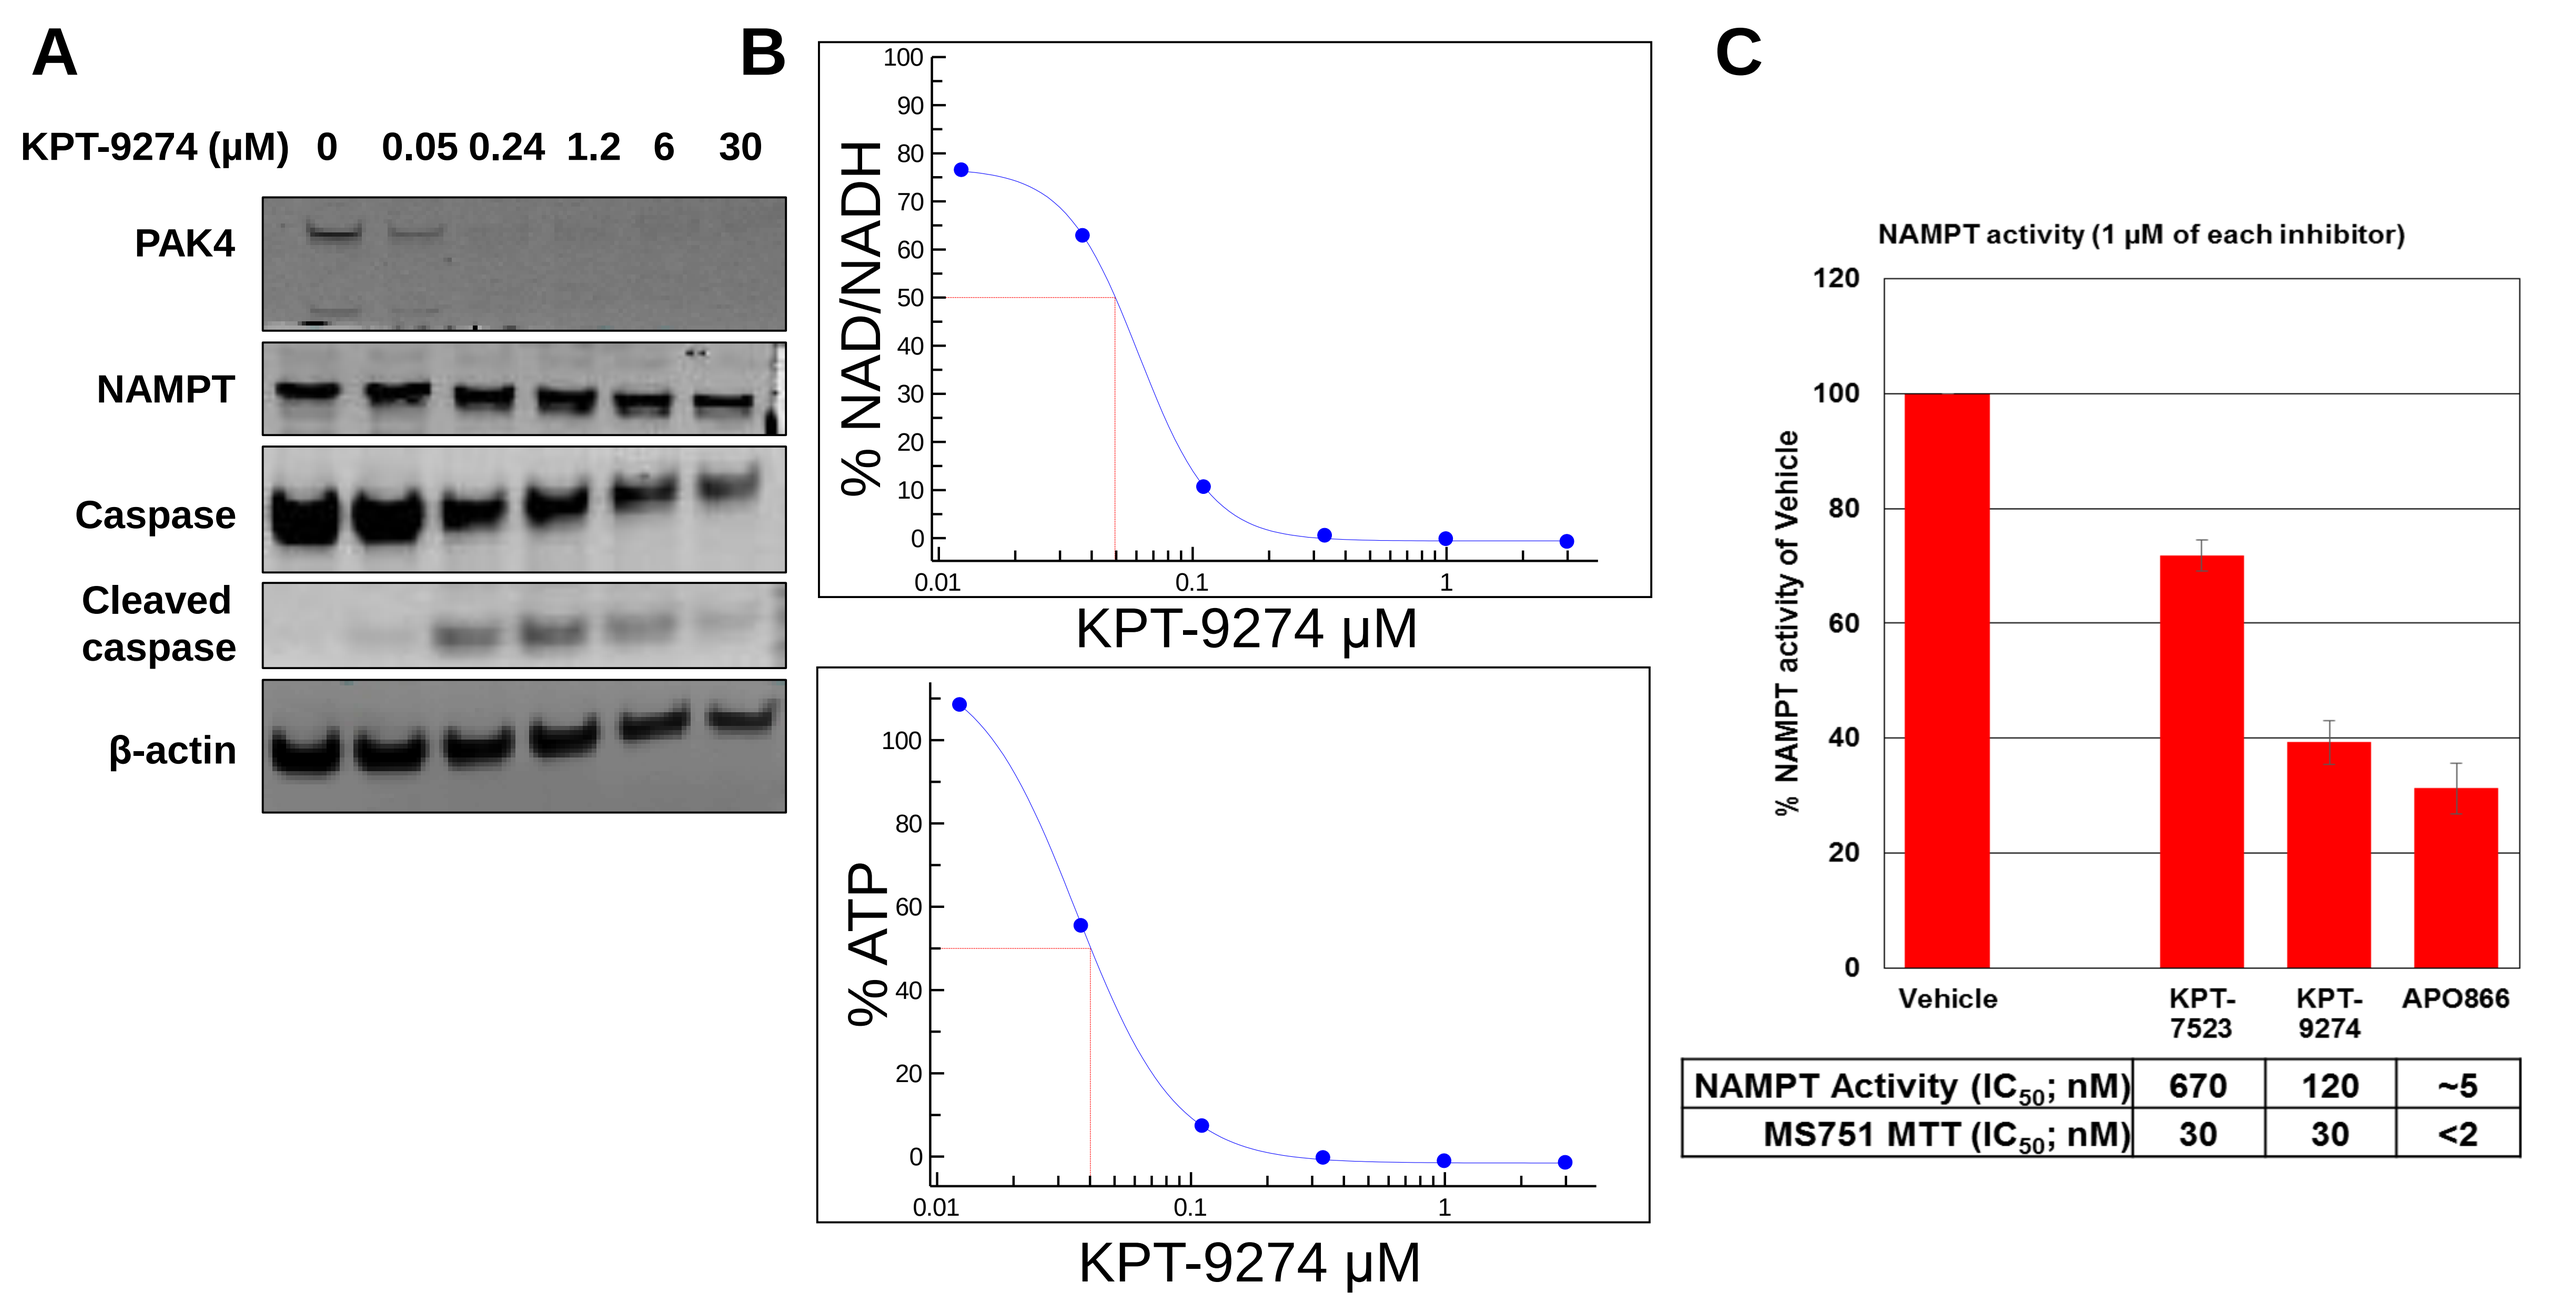

A
B
C
KPT-9274 (μM) 0 0.05 0.24 1.2 6 30
PAK4
NAMPT
Caspase
Cleaved caspase
β-actin
% NAD/NADH
% ATP
KPT-9274 μM
NAMPT activity (1 μM of each inhibitor)
% NAMPT activity of Vehicle
Vehicle
KPT-7523
KPT-9274
APO866

	KPT-7523	KPT-9274	APO866
NAMPT Activity (IC_{50}; nM)	670	120	~5
MS751 MTT (IC_{50}; nM)	30	30	<2

Figure 2: U-2 OS, COLO 205, and MS-751 cells were treated with different concentrations of the PAK4/NAMPT inhibitor KPT-9274 for 72 hours. (A) Western blot of COLO 205 cells. (B) U-2 OS cells were incubated with NAD/NADH-Glo or ATP-Glo reagent in order to measure NAD or ATP levels, respectively. (C) Cyclex NAMPT colorimetric assay was used to compare viability data in in MS-751 cells to *in vitro* enzyme data. KPT-9274 reduces PAK4, NAD and ATP resulting in cancer cell death.

SIRT1 Expression Affects KPT-9274 Cytotoxicity

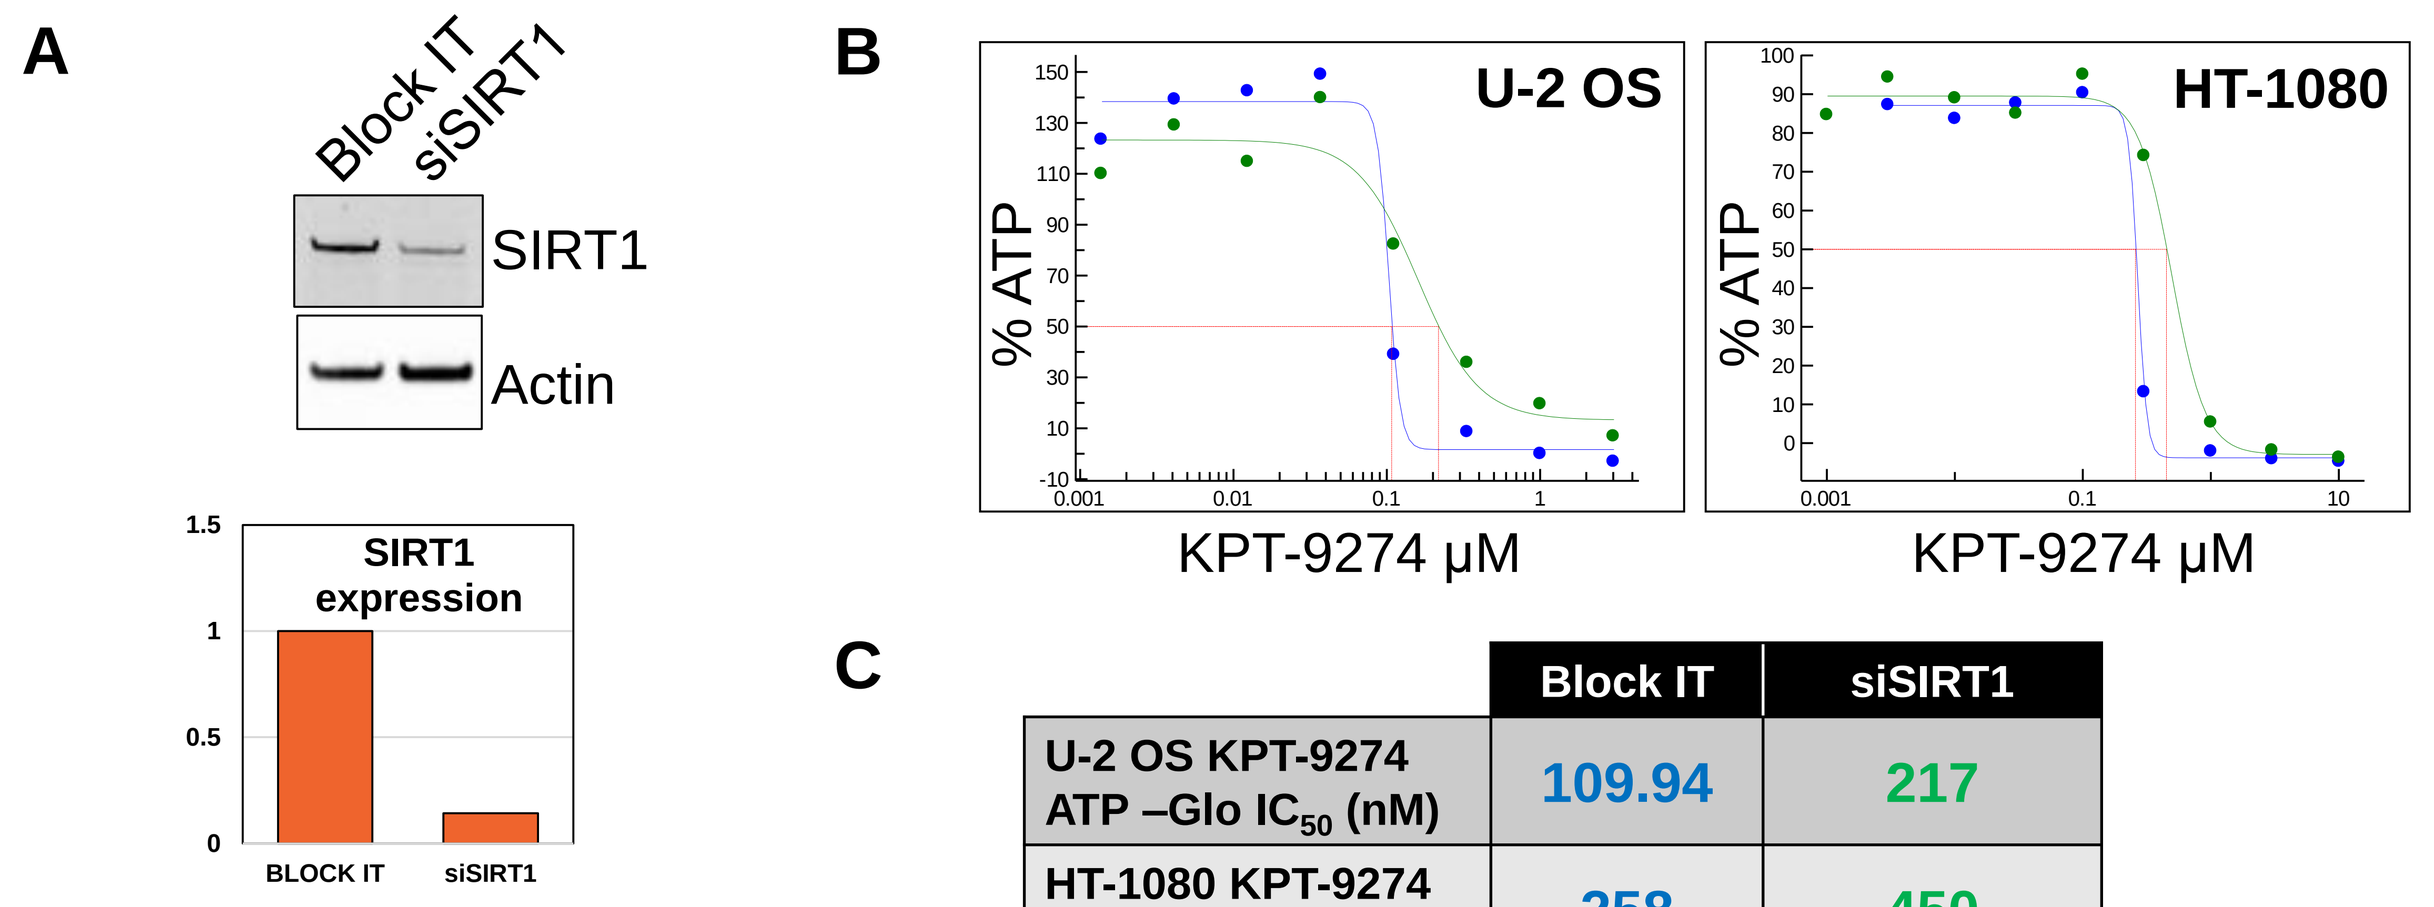

A
B
C
Block IT
siSIRT1
SIRT1
Actin
SIRT1 expression
BLOCK IT
siSIRT1
U-2 OS
HT-1080
% ATP
KPT-9274 μM

	Block IT	siSIRT1
U-2 OS KPT-9274 ATP –Glo IC_{50} (nM)	109.94	217
HT-1080 KPT-9274 ATP –Glo IC_{50} (nM)	258	450

Figure 3: U-2 OS and HT-1080 cells were incubated with control (Block IT) or SIRT1 siRNA for 24 hours. (A) SIRT1 knock down efficiency was confirmed with Western blot analysis. The cells were then treated with varying concentrations of KPT-9274. (B and C) Cell viability was measured using the ATP-Glo assay. Reducing SIRT1 expression reduces KPT-9274 potency in cancer cells.

SIRT1 NAD Dependent Activity Contributes to KPT-9274 Cytotoxicity

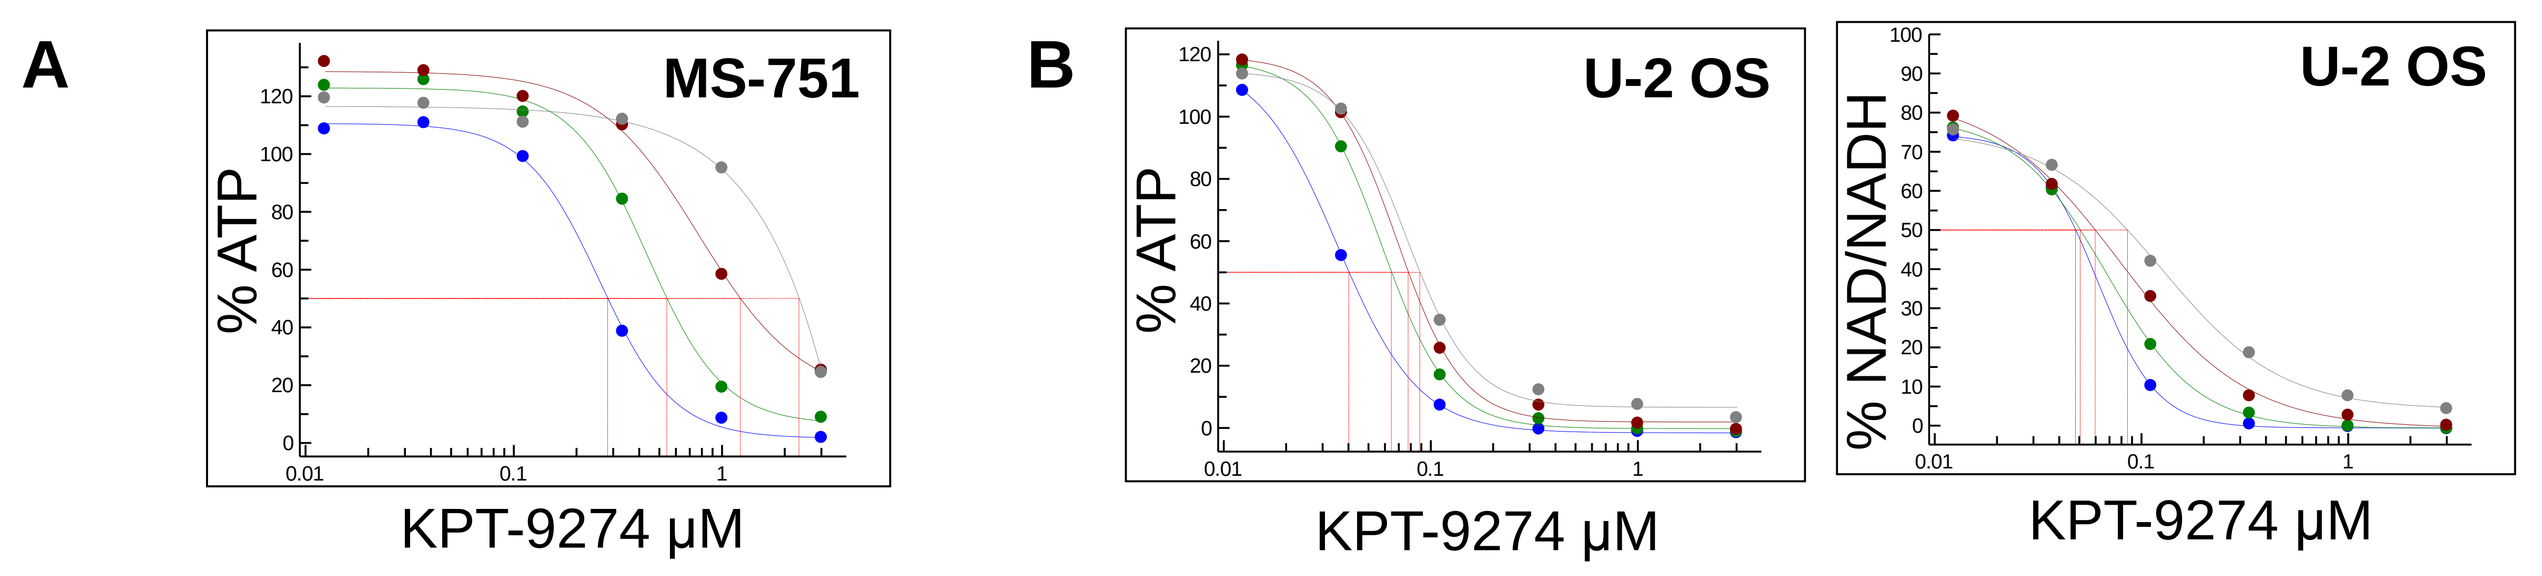

A
B
MS-751
U-2 OS
U-2 OS
% ATP
% NAD/NADH
KPT-9274 μM

Conc. of EX-527	KPT-9274 ATP-Glo IC_{50} (nM)
0.0 µM	282
0.1µM	542
1.0 µM	1225
10 µM	2338

Conc. Of EX-527	KPT-9274 ATP-Glo IC_{50} (nM)	KPT-9274 NADH IC_{50} (nM)
0.0 µM	40	47
0.1µM	64	50
1.0 µM	78	60
10 µM	88	86

Figure 4: (A) MS-751 and (B) U-2 OS cells were treated with different concentrations of KPT-9274 and the SIRT1 specific inhibitor EX-527 for 72 hours. The cells were incubated with ATP-Glo reagent or NAD/NADH-Glo reagent in order to measure ATP or NAD levels. Inhibiting the NAD dependent catalytic activity of SIRT1 reduces the potency of KPT-9274 by reducing the consumption of NAD by SIRT1.

SIRT1 Activator SRT1720 Synergizes with KPT-9274 to Enhance Cancer Cell Death

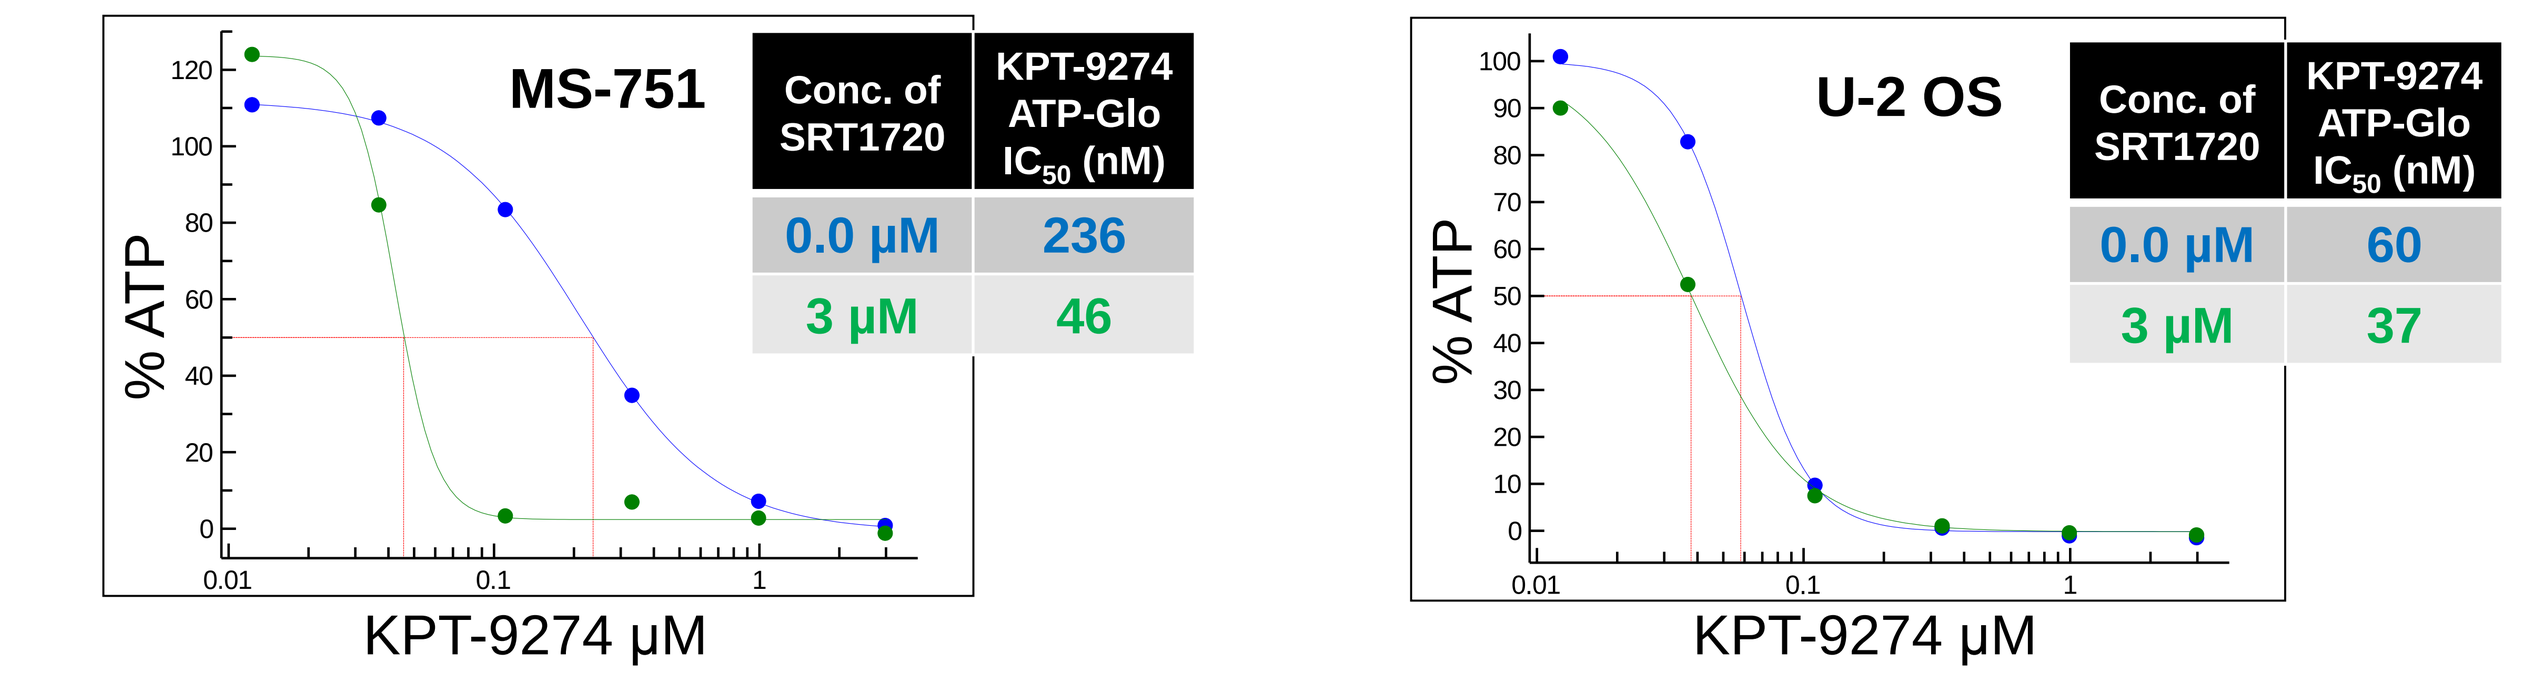

MS-751
U-2 OS
% ATP
KPT-9274 μM

MS-751:

Conc. of SRT1720	KPT-9274 ATP-Glo IC_{50} (nM)
0.0 µM	236
3 µM	46

U-2 OS:

Conc. of SRT1720	KPT-9274 ATP-Glo IC_{50} (nM)
0.0 µM	60
3 µM	37

Figure 5: U-2 OS and MS-751 cells were treated with different concentrations of KPT-9274 and a small molecule activator of SIRT1 (SRT1720) for 72 hours in 96-well plates. KPT-9274 and SRT1720 synergize to induce cell death by increasing the NAD consumption of SIRT1 and inhibiting the synthesis of NAD by NAMPT.

KPT-9274 Disrupts the Localization of Hexokinase II (HKII) and Induces the Formation of SIRT1 Foci in the Cytoplasm

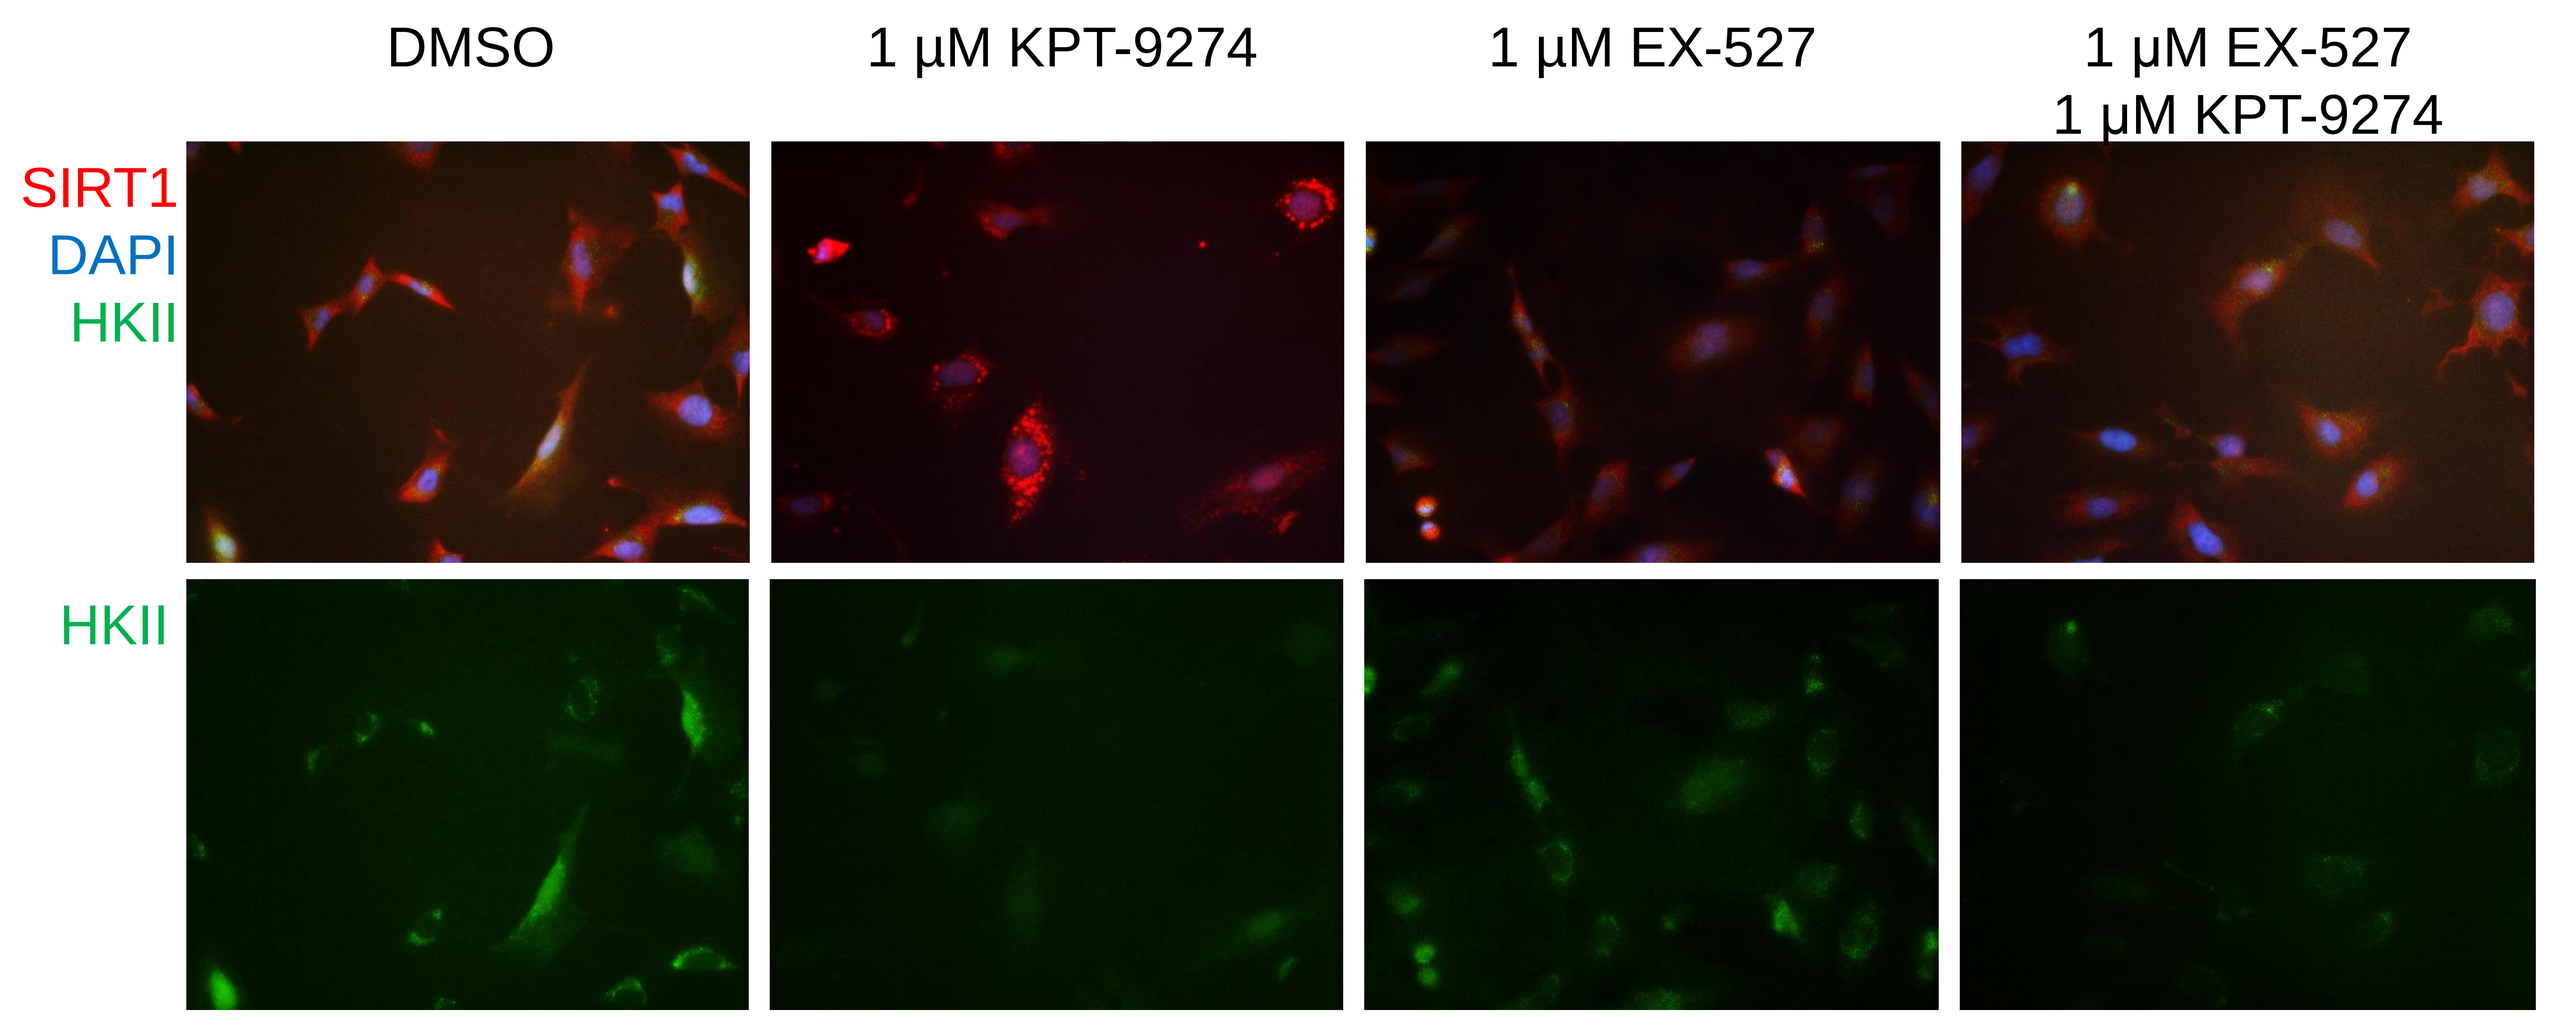

DMSO
1 μM KPT-9274
1 μM EX-527
1 μM EX-527
1 μM KPT-9274
SIRT1
DAPI
HKII
HKII

Figure 6: U-2 OS cells were plated onto glass coverslips and allowed to adhere overnight. The following day the cells were treated with 1 μM KPT-9274, 1 μM SIRT1 specific inhibitor EX-527, or both for 48 hours. KPT-9274 treatment disrupts the localization of the mitochondrial protein hexokinase II (HKII) and promotes the formation of SIRT1 foci in the cytoplasm. Disruption of foci may be an indication of loss of autophagy.

KPT-9274 Disrupts Mitochondrial Membrane Potential

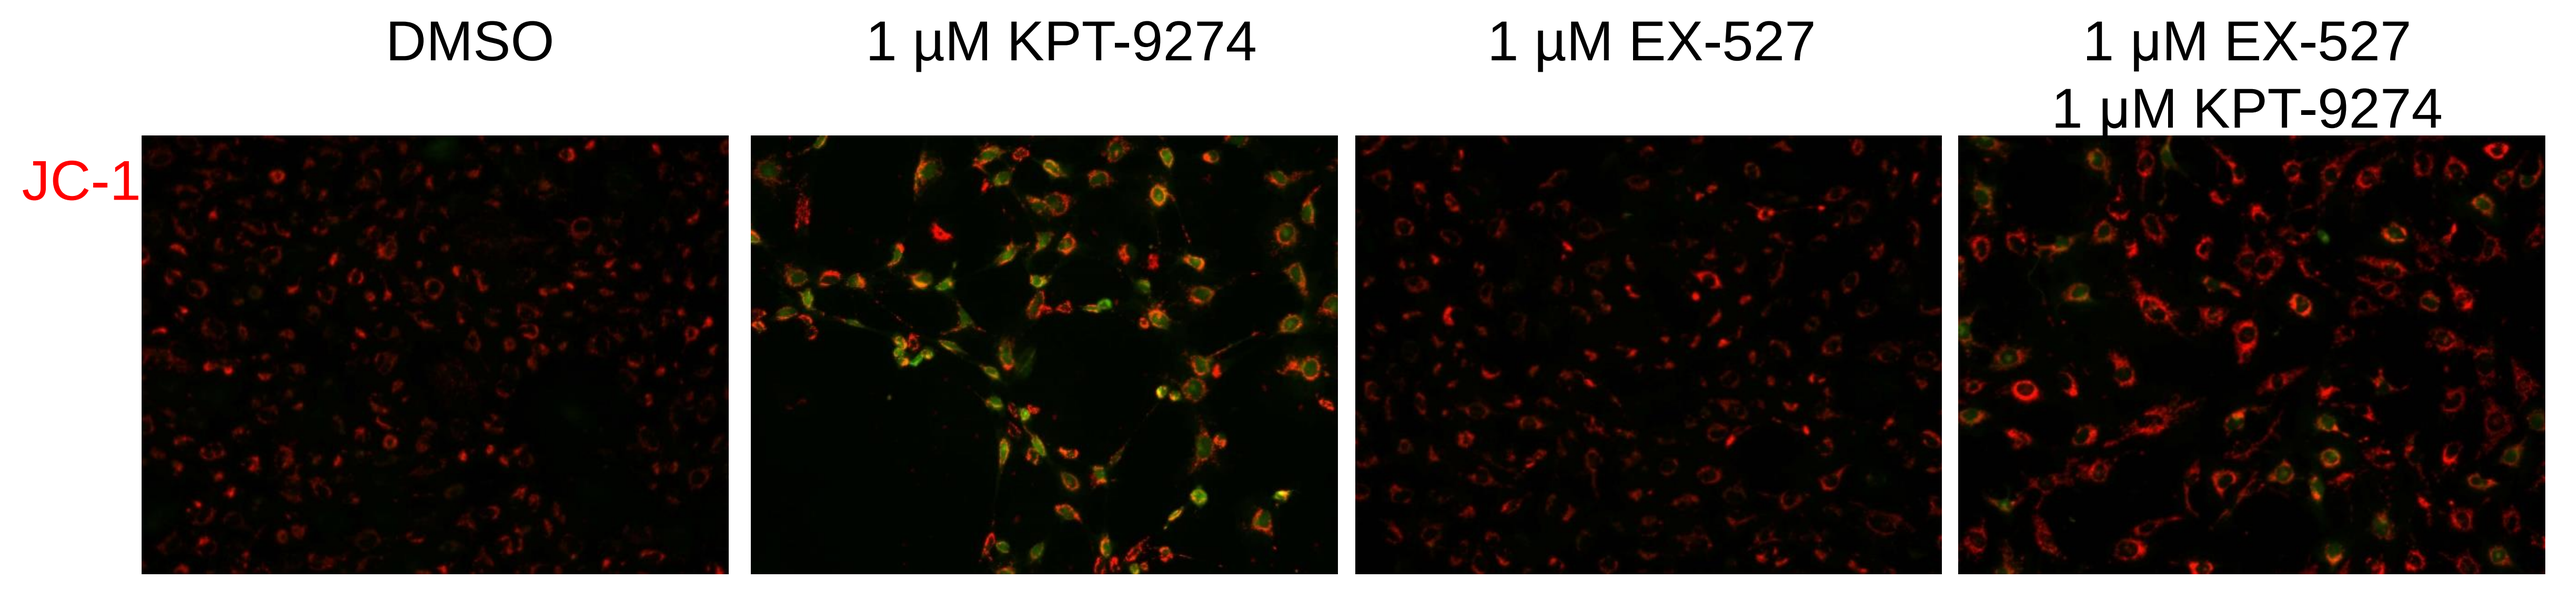

DMSO
1 μM KPT-9274
1 μM EX-527
1 μM EX-527
1 μM KPT-9274
JC-1

Figure 7: U-2 OS cells were plated in 6-well plates overnight then treated with 1 μM KPT-9274 and the 1 μM SIRT1 specific inhibitor EX-527 for 48 hours. The mitochondrial probe JC-1 was used for 30 minutes. JC-1 is incorporated into healthy mitochondria and emits a **red** signal. Unhealthy/ depolarized mitochondria stained with JC-1 emit a **green** signal. KPT-9274 treatment depolarized mitochondria, while inhibition SIRT1 diminished de-polarization of mitochondria by KPT-9274.

PARP Reduction Does Not Affect Cell Death Induced by KPT-9274

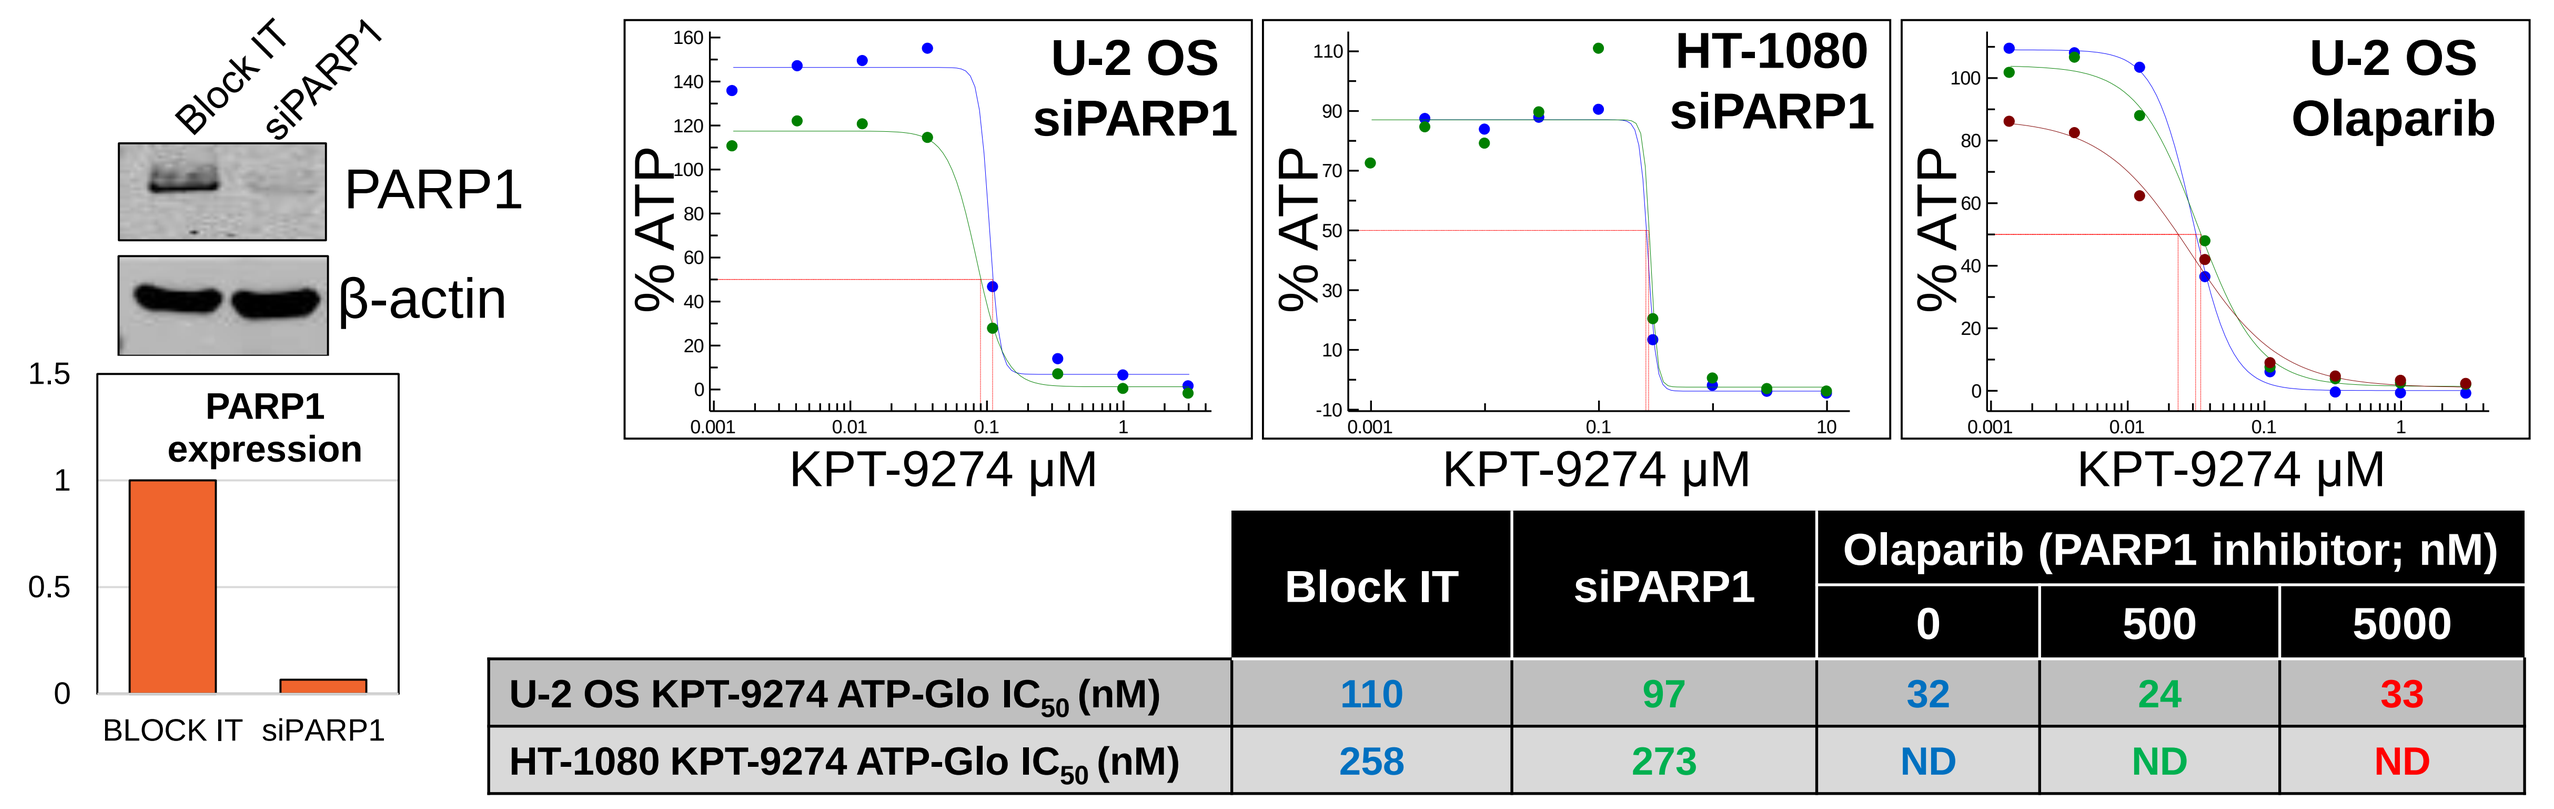

Block IT
siPARP1
PARP1
β-actin
PARP1 expression
BLOCK IT
siPARP1
U-2 OS siPARP1
HT-1080 siPARP1
U-2 OS Olaparib
% ATP
KPT-9274 μM

	Block IT	siPARP1	Olaparib (PARP1 inhibitor; nM) 0	500	5000
U-2 OS KPT-9274 ATP-Glo IC_{50} (nM)	110	97	32	24	33
HT-1080 KPT-9274 ATP-Glo IC_{50} (nM)	258	273	ND	ND	ND

Figure 8: U-2 OS cells were incubated with control (Block IT) or PARP1 siRNA for 24 hours. PARP1 knock down efficiency was confirmed with Western blot analysis. The cells were then treated with varying concentrations of KPT-9274 +/- olaparib. Cell viability was measured using the ATP-Glo assay. Reducing PARP1 expression did not have a significant effect on KPT-9274 toxicity in U-2 OS cells.

PARP1 Activation by Gemcitabine Enhances the Anti-cancer Activity of KPT-9274 *In vitro* and *In vivo*

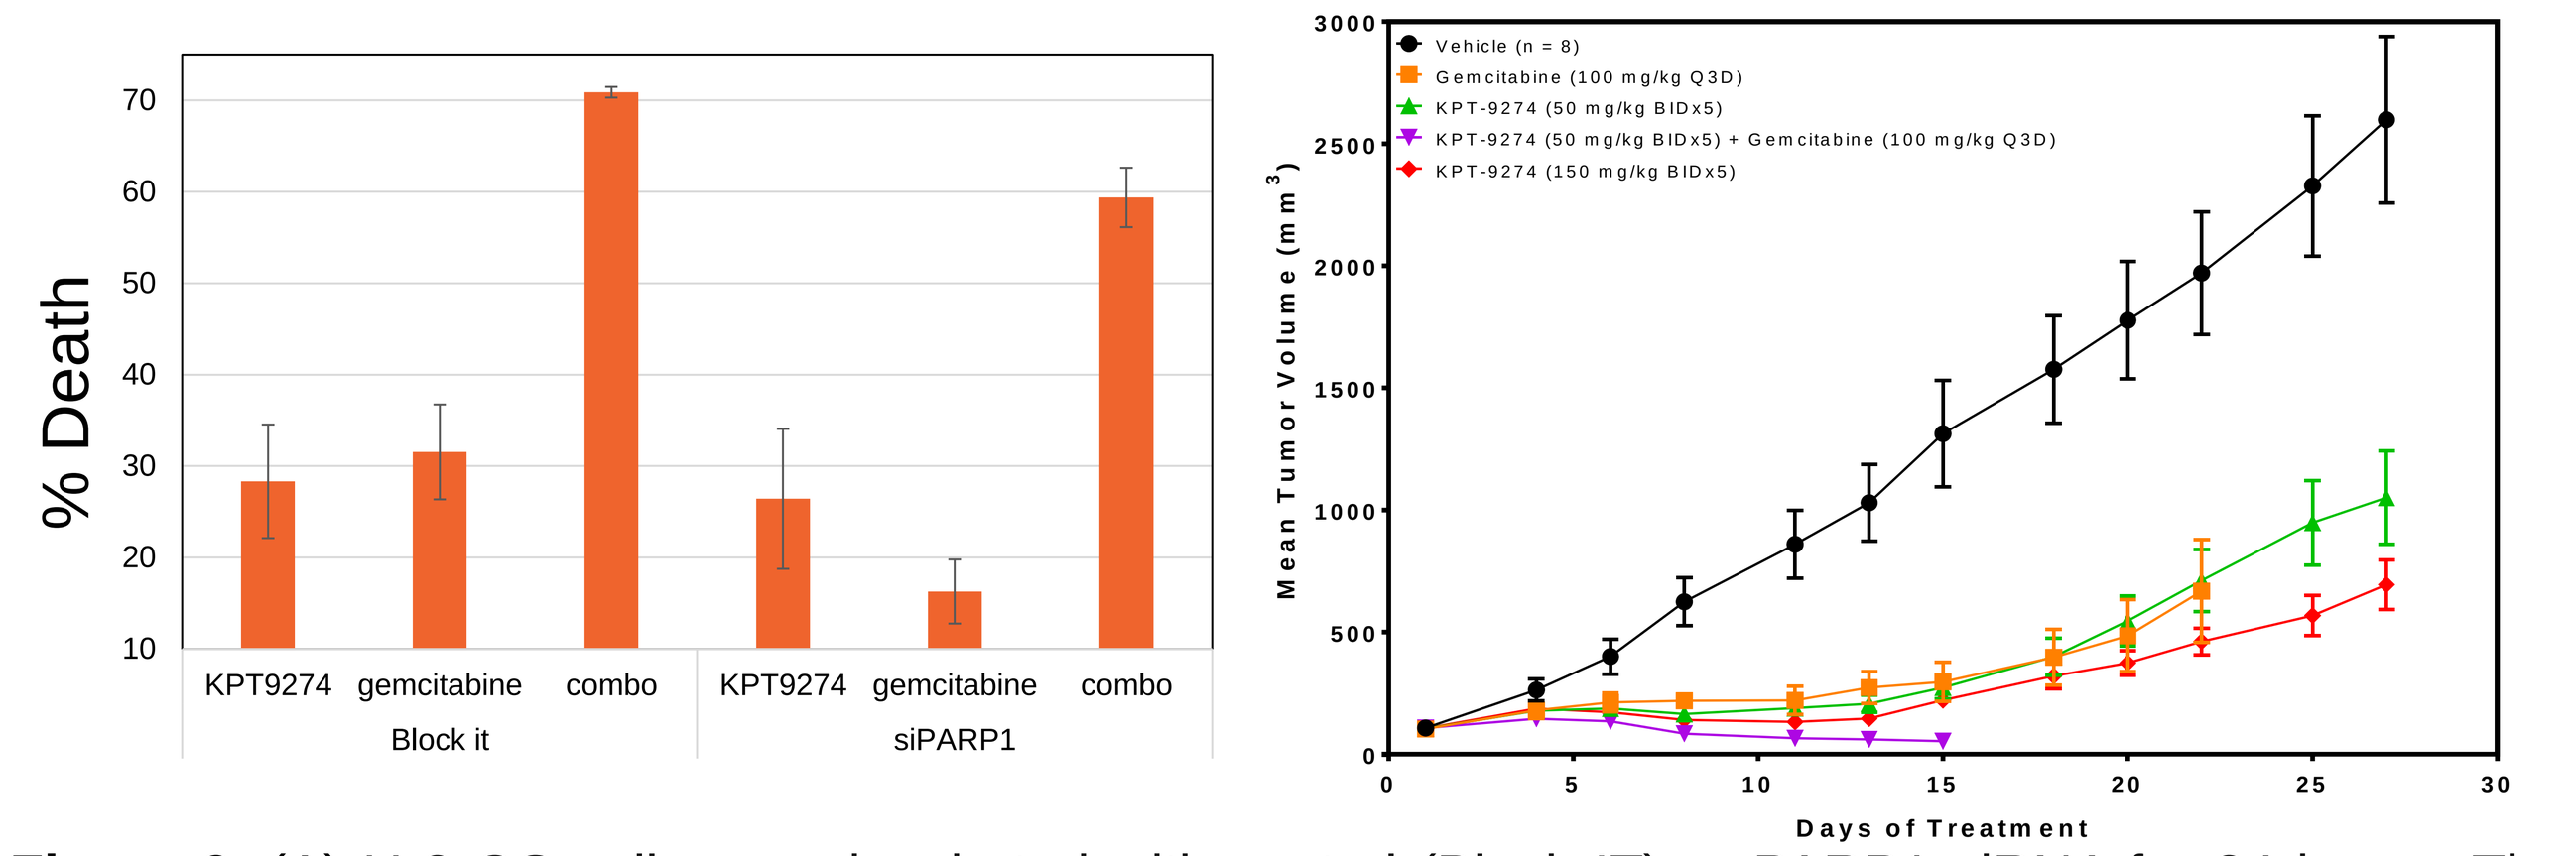

% Death
KPT9274
gemcitabine
combo
Block it
siPARP1
Vehicle (n = 8)
Gemcitabine (100 mg/kg Q3D)
KPT-9274 (50 mg/kg BIDx5)
KPT-9274 (50 mg/kg BIDx5) + Gemcitabine (100 mg/kg Q3D)
KPT-9274 (150 mg/kg BIDx5)
Mean Tumor Volume (mm3)
Days of Treatment

Figure 9: (A) U-2 OS cells were incubated with control (Block IT) or PARP1 siRNA for 24 hours. The cells were then treated with different concentrations of KPT-9274 and single strand break inducing/PARP activating compound (gemcitabine) 72 hours in 96-well plates. The plates were then equilibrated to room temperature and incubated with ATP-Glo reagent. Gemcitabine enhances the anticancer activity of KPT-9274 in U-2 OS cells by activating the NAD consuming enzyme PARP1. (B) Female nu/nu mice were injected with NCI-H250 (non small cell lung) cells. When tumors reached ~200 mm^3, mice were treated with oral administration of vehicle, KPT-9274, and gemcitabine at the concentrations indicated above. Mean tumor volume in mice treated with KPT-9274 or gemcitabine was significantly reduced when compared to vehicle. The combination of KPT-9274 and gemcitabine further inhibited tumor growth.

Conclusions

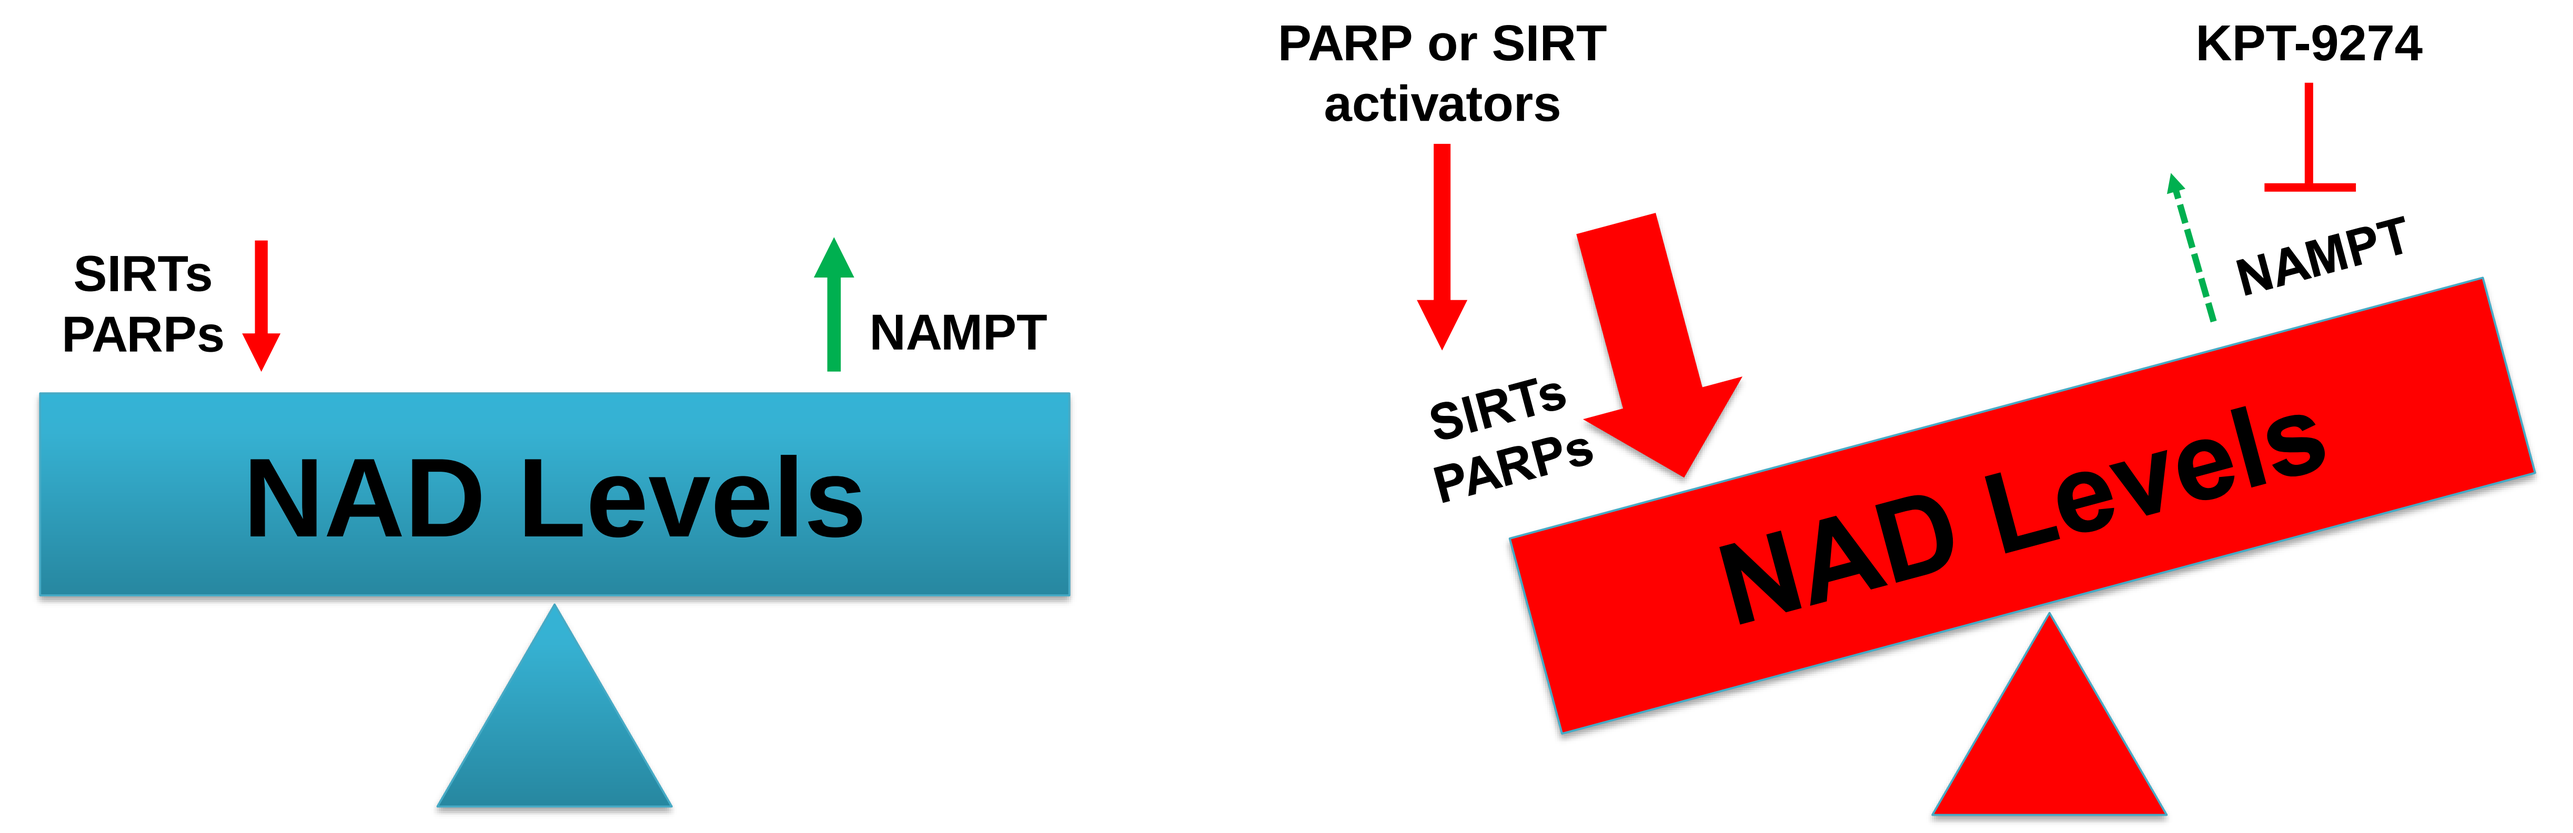

SIRTs
PARPs
NAMPT
NAD Levels
PARP or SIRT activators
KPT-9274
SIRTs
PARPs
NAMPT
NAD Levels

- KPT-9274, a dual inhibitor of PAK4 and NAMPT, significantly reduces cellular NAD levels in a dose dependent manner by reversibly inhibiting the catalytic activity of NAMPT. The combination of NAD depletion and PAK4 inhibition ultimately leads ATP depletion and cell death.
- NAD is a critical metabolite for ATP production and has additional roles in various process such as DNA damage repair, cell signaling and transcriptional regulation. NAMPT is an evolutionarily conserved enzyme that regulates the synthesis of NAD in cells.
- The NAD dependent deacetylase SIRT1 contributes to KPT-9274 mediated cell death. Silencing SIRT1 expression or reducing its activity reduces the efficacy of KPT-9274. In contrast, activing SIRT1 with the small molecule SRT1720 increases cell death when used in combination with KPT-9274.
- SIRT1, which also acts as an NAD sensor has been reported to regulate mitochondrial biogenesis and form cytoplasmic/perinuclear foci when cells are treated with KPT-9274. The observed SIRT1 foci coincides with the disruption of mitochondrial membrane polarity and the localization of mitochondrial proteins (e.g. HKII).
- PARP1 does not affect the cytotoxicity of KPT-9274 unless its activated through single strand breaks (e.g. gemcitabine treatment) both *in vitro* and *in vivo*.
- Together these data suggest that enhancing NAD depletion by activating NAD consuming enzymes in cells may enhance the anti-tumor activity of KPT-9274 and further development may lead to novel anti-cancer combination therapies in the future.

Contact Information: Dr. William Senapedis
e-mail: wsenapedis@karyopharm.com T: +1 617-658-0524